P920

SERALUTINIB IN PULMONARY ARTERIAL HYPERTENSION (PAH): EXPLORING MECHANISMS OF REVERSE REMODELING VERSUS VASODILATION

Ravikumar Sitapara[1], Fenja Knöpp[2], Muchen Li[2], Robin Osterhout[1], Robert F. Roscigno[1], Lawrence S. Zisman[1], Richard Aranda[1], Jean-Marie Bruey[1], Natascha Sommer[2]

[1]Gossamer Bio, Inc., San Diego, CA, USA; [2]Excellence Cluster Cardio-Pulmonary Institute, University of Giessen and Marburg Lung Center (UGMLC), Member of the German Center for Lung Research (DZL), Justus-Liebig-University Giessen, Giessen, Germany

JUSTUS-LIEBIG-UNIVERSITÄT GIESSEN

Presented at the
ATS International Conference
San Francisco, CA, USA
May 16-21, 2025

BACKGROUND

- Seralutinib is an inhaled tyrosine kinase inhibitor targeting PDGFRα/β, CSF1R, and c-KIT, with anti-inflammatory, anti-proliferative, and anti-fibrotic properties[1] (**Figure 1**)
- Computed tomography (CT) imaging, biomarker, and preclinical studies of seralutinib consistently support a reverse remodeling of the pulmonary vasculature in PAH[2-4]
- TORREY open-label extension (NCT04816604) data corroborate the potential for a reverse remodeling effect, as demonstrated by continual improvement in pulmonary vascular resistance and changes in vascular biomarkers[5,6]
- PDGFR signaling can regulate calcium flux, which may modulate vascular tone *in vivo*[7]
- We sought to further inform the mechanism of action of seralutinib by investigating its effect on calcium signaling and its acute effects in preclinical models of vasoconstriction

Figure 1. Seralutinib mechanism of action

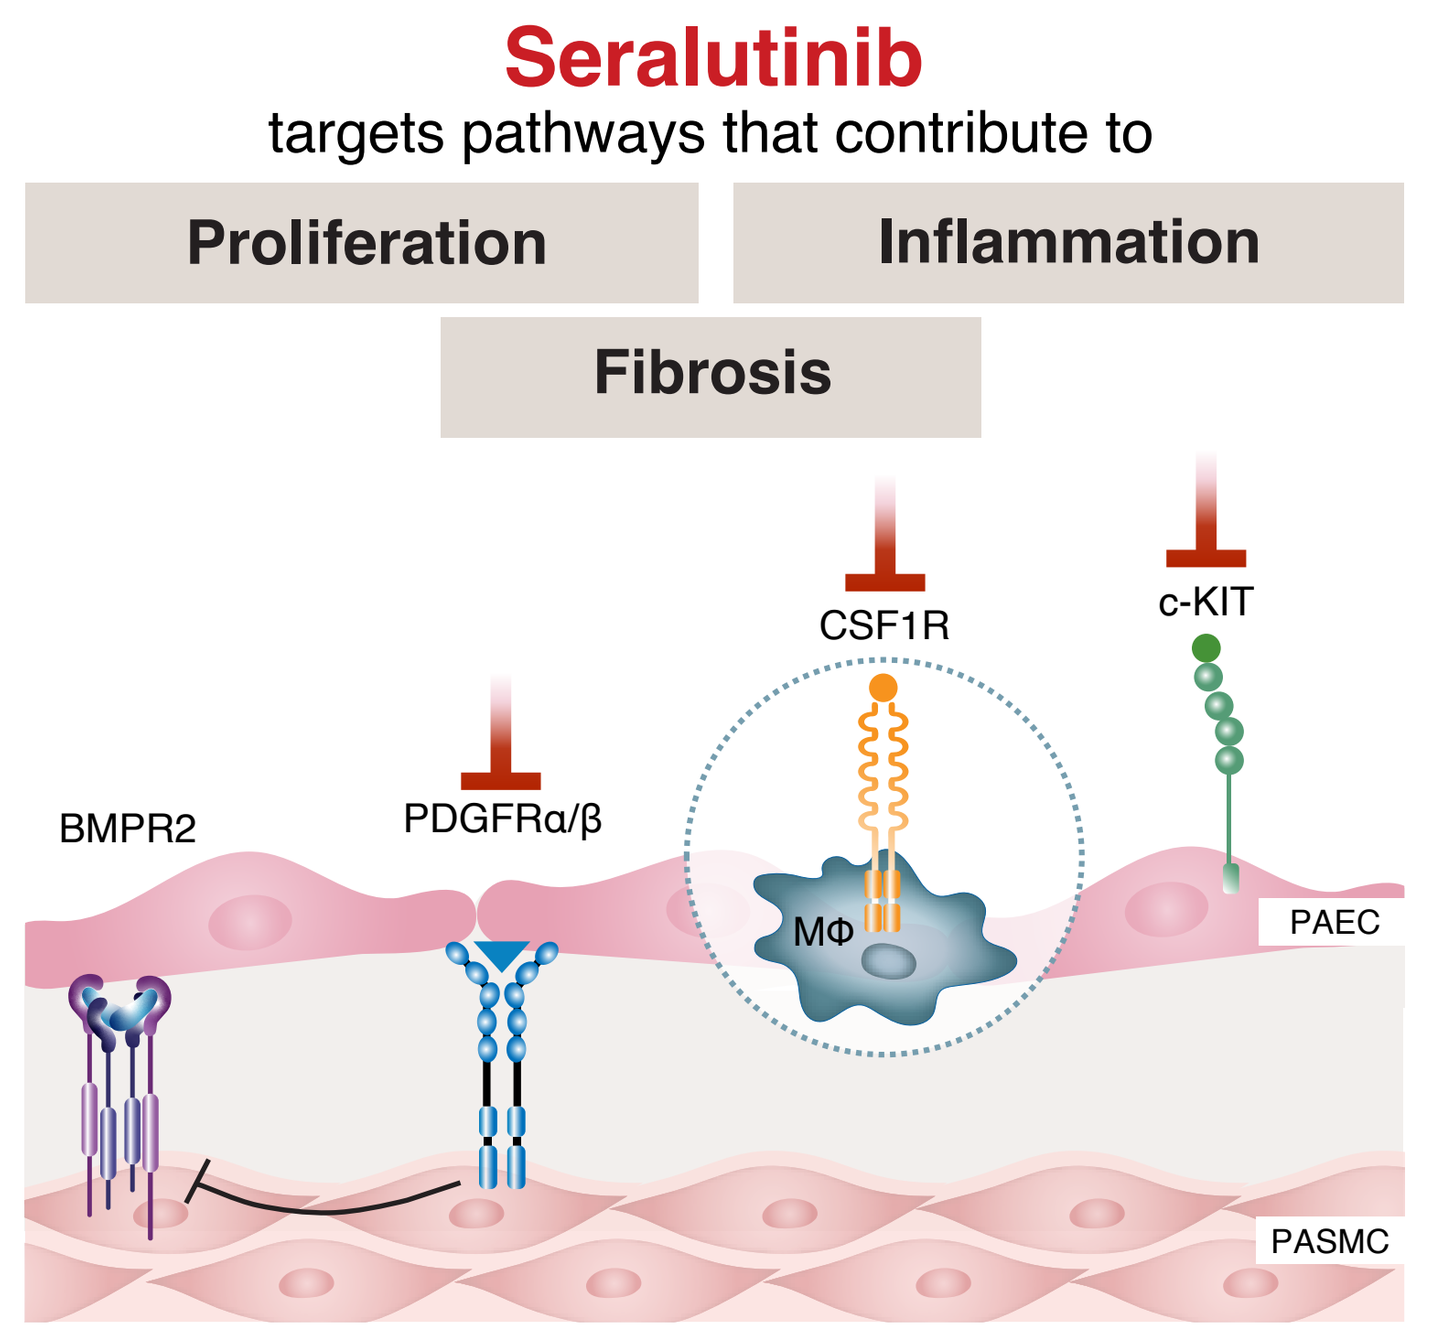
Seralutinib
targets pathways that contribute to
Proliferation
Inflammation
Fibrosis
CSF1R
c-KIT
BMPR2
PDGFRα/β
MΦ
PAEC
PASMC

Blunted arrows indicate inhibition. BMPR2, bone morphogenetic protein receptor type 2; c-KIT, mast/stem cell growth factor receptor kit; CSF1R, colony stimulating factor 1 receptor; MΦ, macrophage; PAEC, pulmonary artery endothelial cell; PASMC, pulmonary artery smooth muscle cell; PDGFR, platelet-derived growth factor receptor.

METHODS

Calcium flux in human pulmonary artery smooth muscle cells (hPASMCs)

- hPASMCs were pretreated with seralutinib or bosentan, and stimulated with PDGF-BB or endothelin-1, respectively
- Cytosolic Ca^{2+} levels were measured using the Fura-2 assay; cells were loaded with Fura-2 dye, and extracellular dye was removed by washing

Vascular tone in mouse pulmonary artery (PA) rings

- C57BL/6 mice were euthanized and extrapulmonary arteries dissected
- Vessel segments were mounted in wire myograph chambers to measure vessel tension (**Figure 2**)
- Effects of PDGF-BB on vessel tension were evaluated with or without seralutinib

Mean pulmonary artery pressure (mPAP) and mean systemic arterial pressure (mSAP) in an acute hypoxia-induced pulmonary vasoconstriction rat model

- Male Sprague-Dawley rats (275–300 g) were anesthetized, intubated, and placed on a ventilator
- A catheter was inserted into pulmonary and femoral arteries for continuous measurement of mPAP and mSAP, respectively
- Hypoxia was induced by reducing the inspired oxygen concentration to ~10%
- Once stable 60% pO_2 was established, seralutinib, treprostinil, or vehicle was administered via the intratracheal route using a microsprayer
- Hemodynamic variations were monitored for 45 minutes

Figure 2. Wire myography experimental setup

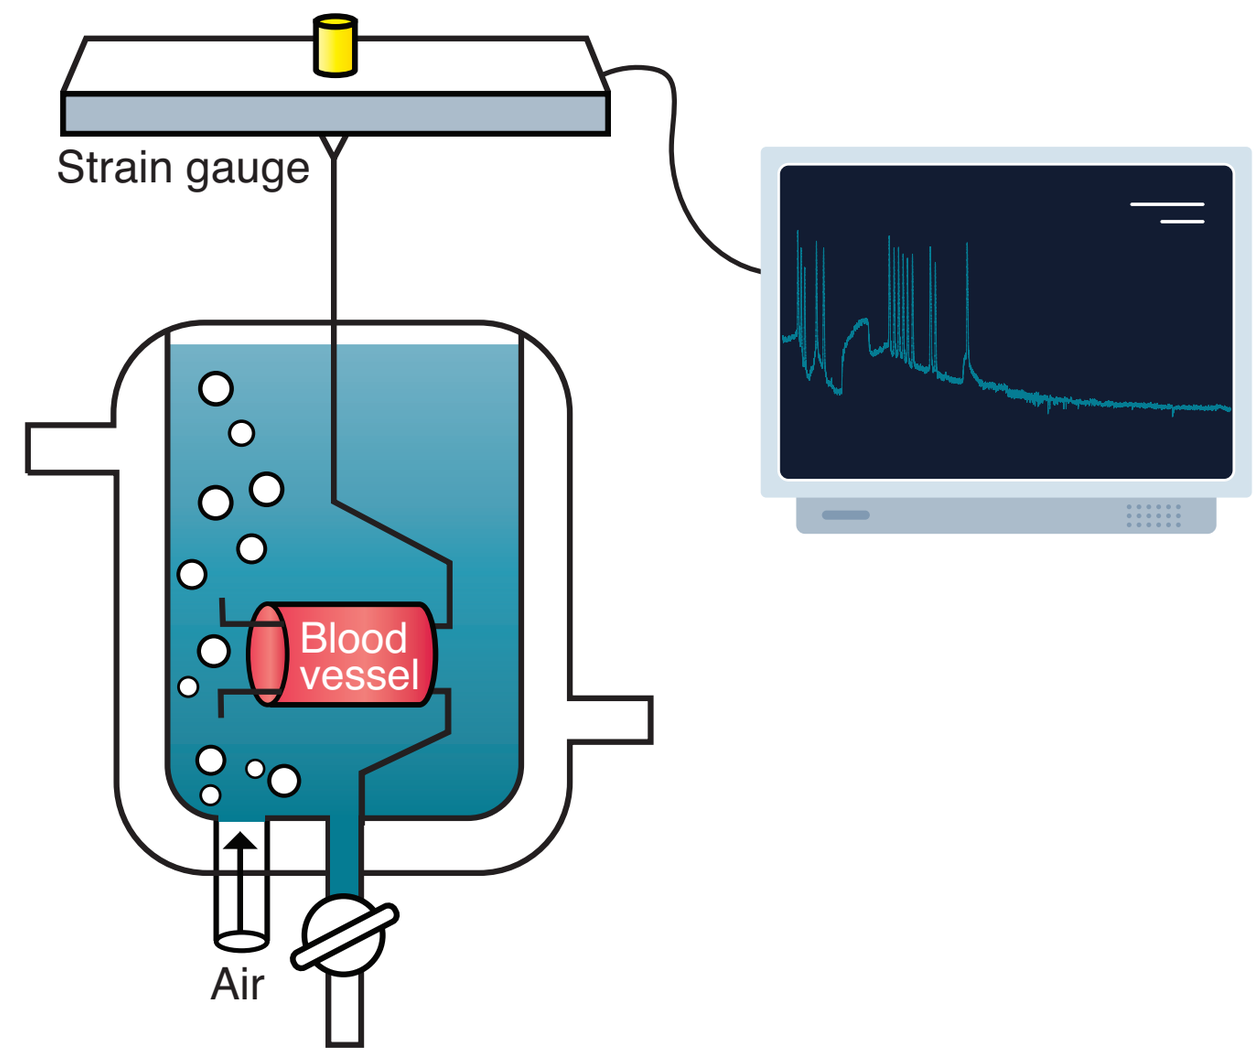
Strain gauge
Blood vessel
Air

Isolated mouse PA rings were exposed to agents, and change in vascular tension was measured.

RESULTS

Figure 3. Seralutinib and bosentan inhibit PDGF-BB– and endothelin-induced calcium (Ca^{2+}) influx, respectively, in hPASMCs

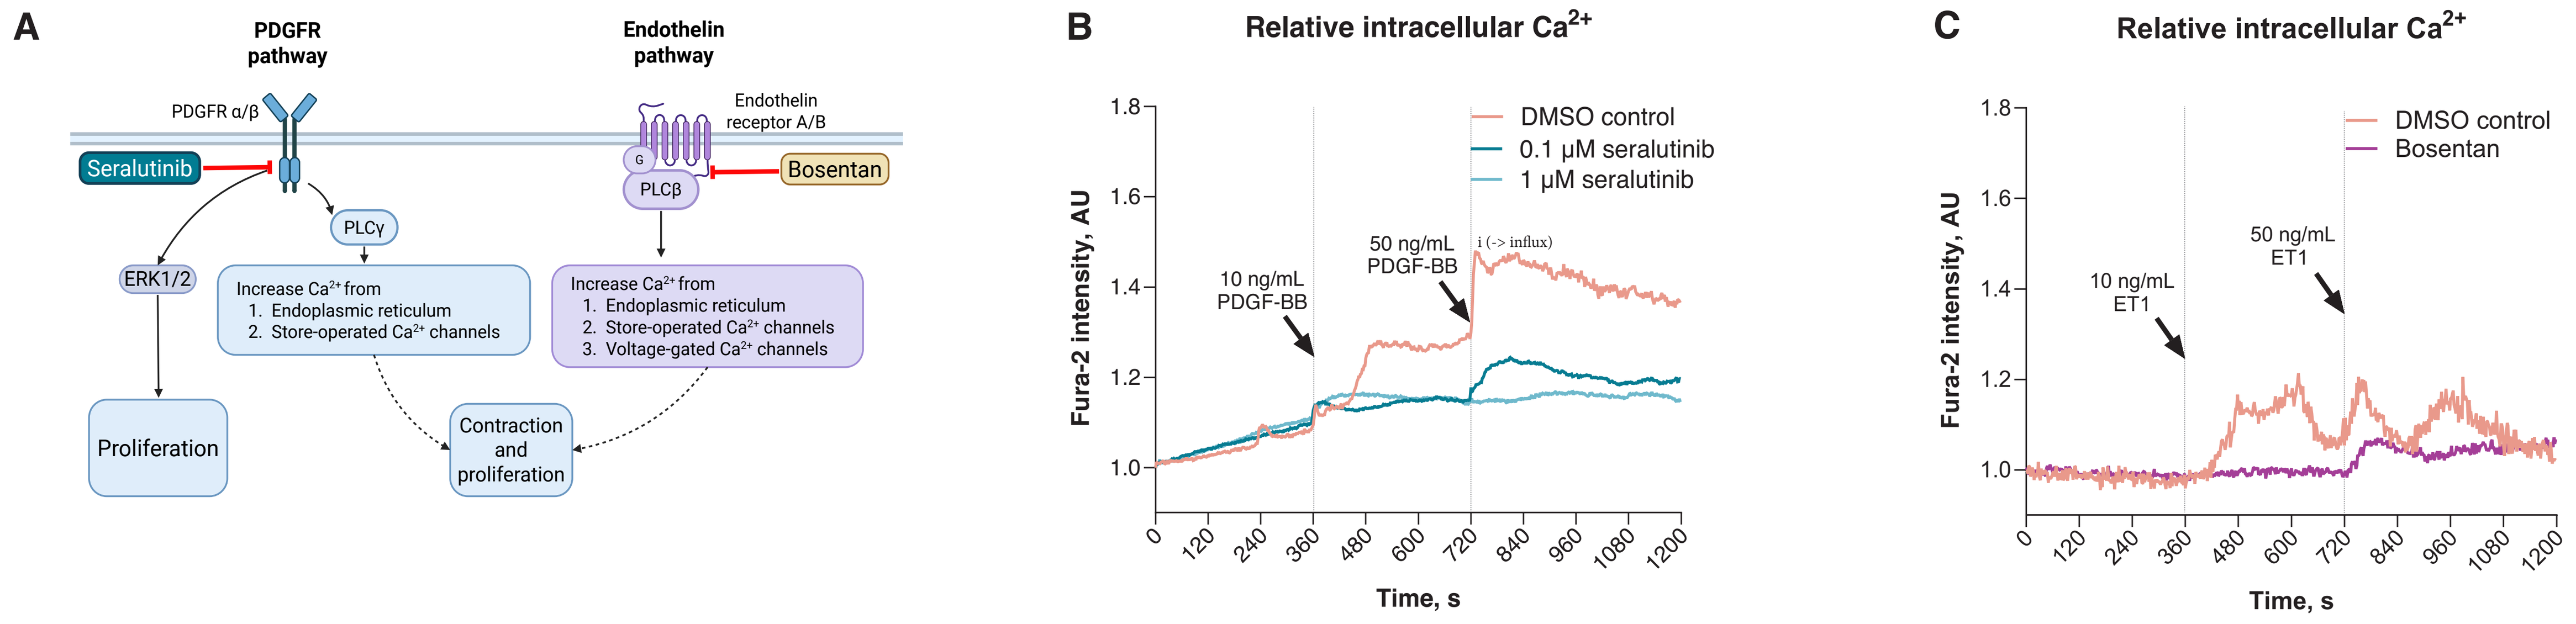
A
PDGFR pathway
Endothelin pathway
PDGFR α/β
Endothelin receptor A/B
Seralutinib
Bosentan
PLCγ
PLCβ
G
ERK1/2
Increase Ca2+ from
1. Endoplasmic reticulum
2. Store-operated Ca2+ channels
Increase Ca2+ from
1. Endoplasmic reticulum
2. Store-operated Ca2+ channels
3. Voltage-gated Ca2+ channels
Proliferation
Contraction and proliferation
B
Relative intracellular Ca2+
DMSO control
0.1 μM seralutinib
1 μM seralutinib
10 ng/mL PDGF-BB
50 ng/mL PDGF-BB
Fura-2 intensity, AU
Time, s
C
Relative intracellular Ca2+
DMSO control
Bosentan
10 ng/mL ET1
50 ng/mL ET1
Fura-2 intensity, AU
Time, s

(**A**) Diagram of Ca^{2+} influx induced by PDGF-BB and endothelin-1, and downstream pathways. (**B**) The effect of PDGF-BB, with or without seralutinib pre-treatment, on intracellular Ca^{2+} levels was assessed by measuring Fura-2 intensity (ratio 340/380 nm vs baseline). (**C**) The effect of endothelin-1, with or without bosentan pre-treatment, on intracellular Ca^{2+} levels was assessed by measuring Fura-2 intensity (ratio 340/380 nm vs baseline). Data are expressed as mean (n=7-9 for PDGF-BB and seralutinib; n=4 for ET1 and bosentan). AU, arbitrary units; Ca^{2+}, calcium; DMSO, dimethyl sulfoxide; ERK, extracellular signal-regulated kinase; ET1, endothelin-1; G, g protein-coupled receptor; hPASMC, human pulmonary artery smooth muscle cell; PDGF(R), platelet-derived growth factor (receptor); pERK, phosphorylated extracellular signal-regulated kinase; PLC, phospholipase C.

Figure 4. Neither PDGF-BB, nor seralutinib, affects vascular tone in isolated mouse PA rings

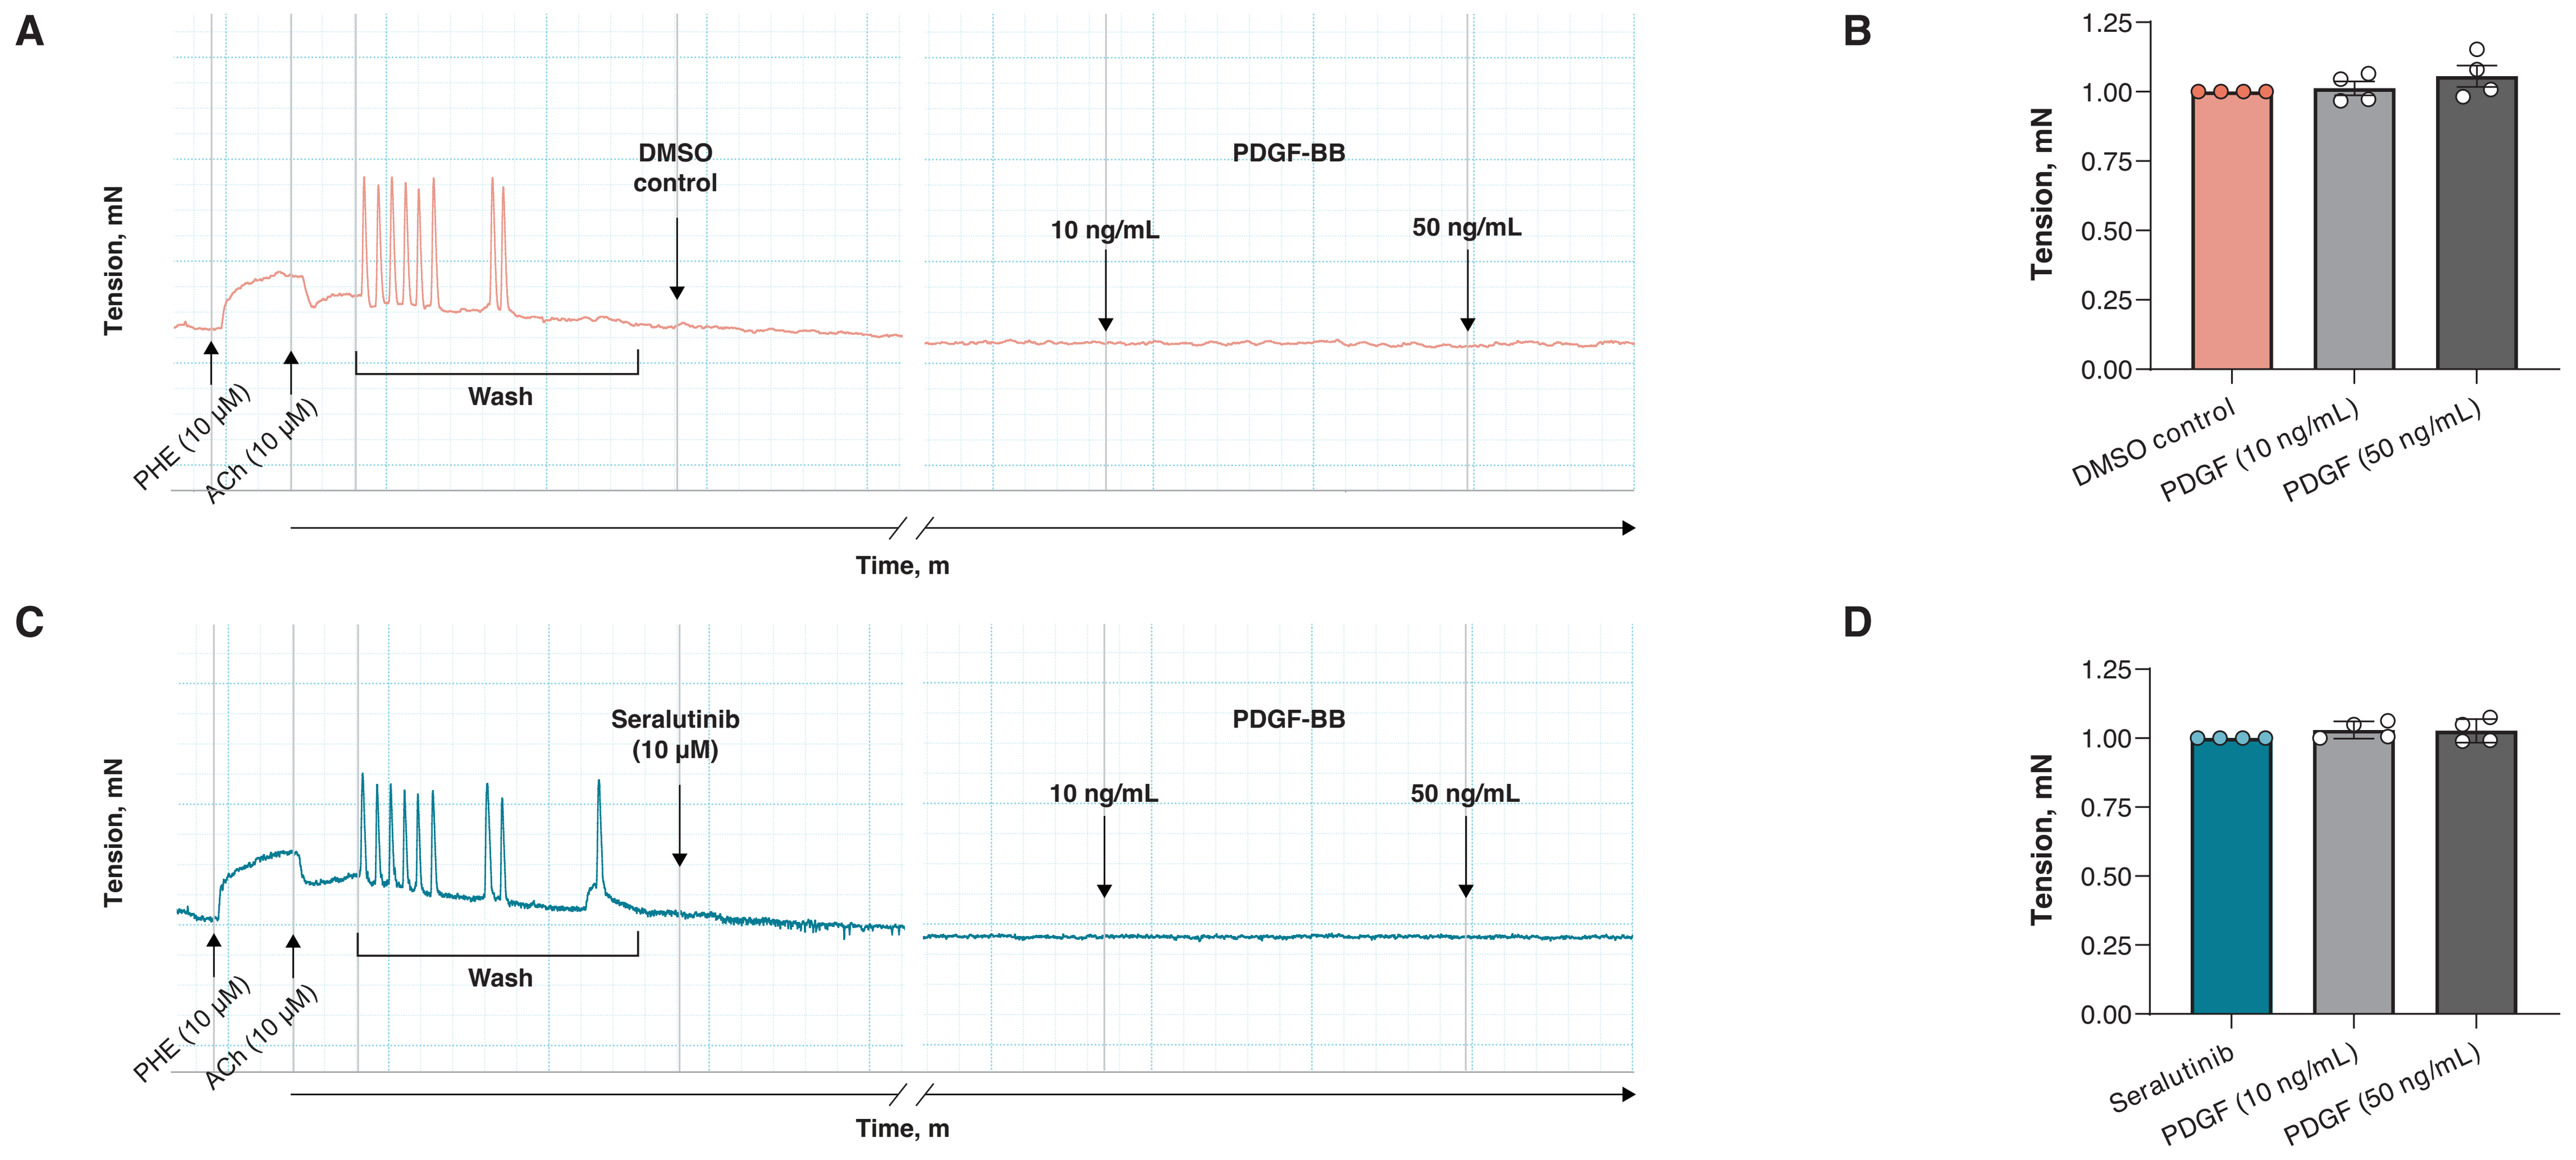
A
Tension, mN
DMSO control
PDGF-BB
10 ng/mL
50 ng/mL
PHE (10 μM)
ACh (10 μM)
Wash
Time, m
B
Tension, mN
DMSO control
PDGF (10 ng/mL)
PDGF (50 ng/mL)
C
Tension, mN
Seralutinib (10 μM)
PDGF-BB
10 ng/mL
50 ng/mL
PHE (10 μM)
ACh (10 μM)
Wash
Time, m
D
Tension, mN
Seralutinib
PDGF (10 ng/mL)
PDGF (50 ng/mL)

Change in tension in pulmonary vessels following PDGF-BB treatment, without (**A, B**) and with (**C, D**) seralutinib. The functional state of PA rings was assured by PHE and ACh pre-treatment. Data are expressed as mean ± SEM (n=2). Statistical analysis was performed using one-way ANOVA with Dunnett's multiple comparison test. ACh, acetylcholine; ANOVA, analysis of variance; DMSO, dimethyl sulfoxide; mN, milliNewton; PA, pulmonary artery; PDGF, platelet-derived growth factor; PHE, phenylephrine.

Figure 5. Seralutinib does not exhibit vasodilatory effects in a rat model of acute hypoxia-induced pulmonary vasoconstriction

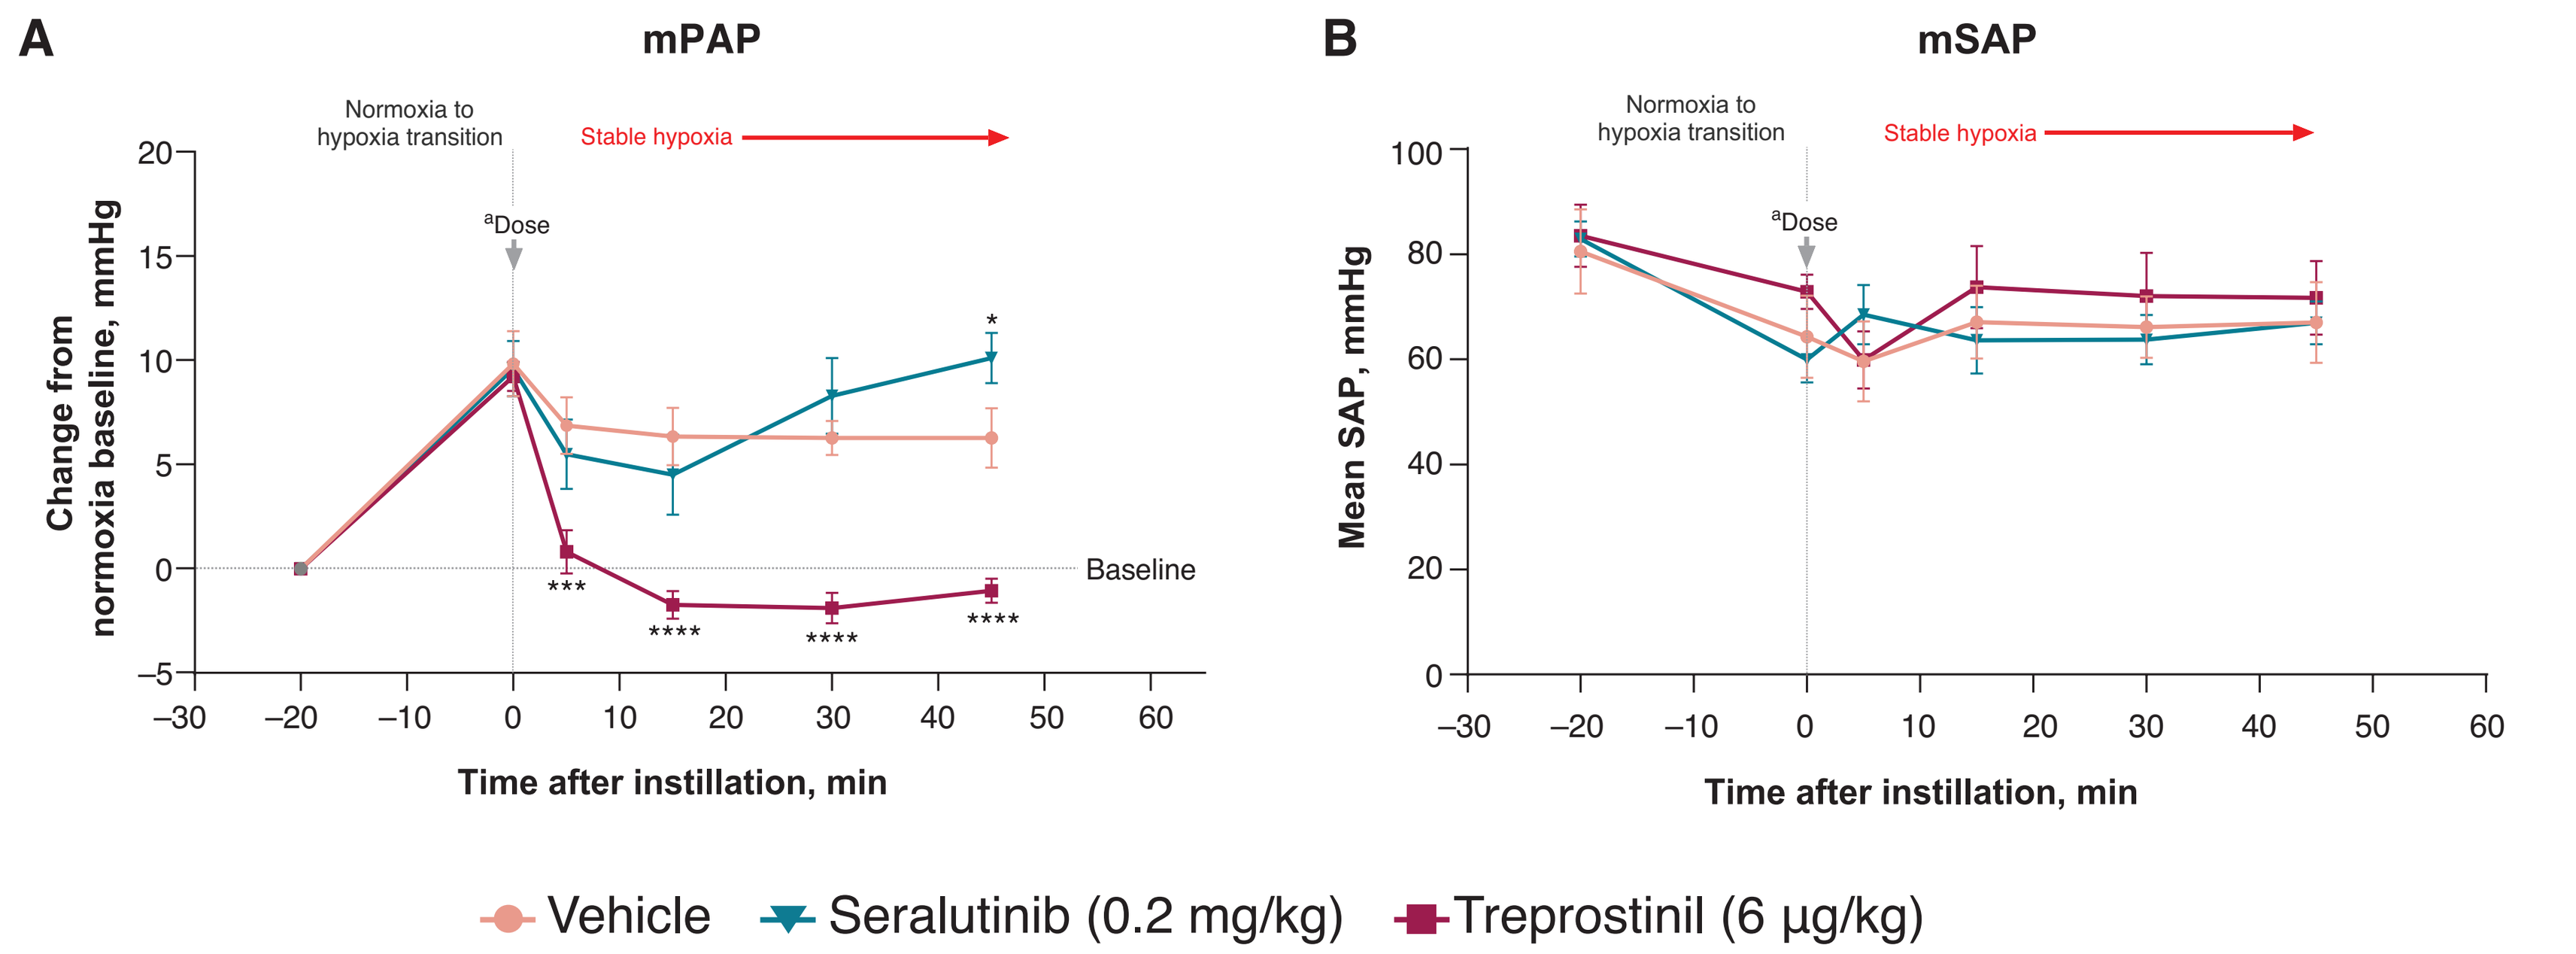
A
mPAP
Normoxia to hypoxia transition
Stable hypoxia
aDose
Baseline
Change from normoxia baseline, mmHg
Time after instillation, min
B
mSAP
Normoxia to hypoxia transition
Stable hypoxia
aDose
Mean SAP, mmHg
Time after instillation, min
Vehicle
Seralutinib (0.2 mg/kg)
Treprostinil (6 μg/kg)

Effects of seralutinib and treprostinil on (**A**) mPAP change from normoxia baseline and (**B**) mSAP. Data are represented as mean (n=5). Statistical analysis was performed using two-way ANOVA with Dunnett's multiple comparison test. *$p<0.05$, ***$p<0.001$, and ****$p<0.0001$ versus vehicle. [a]Animals were taken off the ventilator and exposed to normoxia during intratracheal dosing. mPAP, mean pulmonary artery pressure; mSAP, mean systemic arterial pressure.

CONCLUSIONS

- CT imaging, biomarker, and preclinical studies previously demonstrated a reverse remodeling effect of seralutinib
- Our current findings substantiate that the primary mechanism of action of seralutinib is reverse remodeling, rather than vasodilation
 - Seralutinib inhibited PDGF-BB–induced Ca^{2+} flux, which is known to contribute to proliferation, and did not display significant vasodilatory effects
 - Seralutinib or PDGF-BB signaling did not modulate tension in PA rings
 - Seralutinib did not reverse acute hypoxia-induced vasoconstriction
- Seralutinib is an anti-proliferative and anti-fibrotic agent that is differentiated from traditional vasodilator therapies

References: **1** Galkin A, et al. *Eur Respir J*. 2022;60(6):2102356. **2** Zamanian RT, et al. *Eur Respir J*. 2023;62(suppl 67):OA742. **3** Osterhout R, et al. *Eur Respir J*. 2024;64(suppl 68):OA1872. **4** Schlüter B, et al. *Eur Respir J*. 2024;64(suppl 68):PA1613. **5** Sitbon O, et al. *Am J Respir Crit Care Med*. 2024;209:A1011. **6** McLaughlin VV, et al. *Eur Respir J*. 2024;64(suppl 68):AO1871. **7** Rieg AD, et al. *Respir Res*. 2018;19(1):120.

Research supported by: Gossamer Bio, Inc.