SERALUTINIB FOR THE TREATMENT OF PULMONARY ARTERIAL HYPERTENSION: RESULTS FROM THE PHASE 3 PROSERA TRIAL

H-A Ghofrani*[1], VV McLaughlin*[2], G Meyer[3], RL Benza[4], RN Channick[5], KM Chin[6], RP Frantz[7], AR Hemnes[8], LS Howard[9], RT Zamanian[10], G Kopeć[11], A Skride[12], C Jerjes-Sánchez[13], AJ Lescano[14], MP Escribano Subías[15], RJ White[16], T Pulido[17], ER Perna[18], S Rosenkranz[19], R Badagliacca[20], L Scelsi[21], MA Lima[22], R Sulica[23], C Miller[24], H Al-Hiti[25], F Campos[26], M Zagolin[27], RF Roscigno[28], D Mottola[28], R Aranda[28], M Cravets[28], C Tompkins[28], E Parsley[28], E Elman[28], LS Zisman[28], J-L Vachiéry**[29], O Sitbon**[30]

[1]Universities of Giessen and Marburg Lung Center (UGMLC), Institute for Lung Health (ILH), Cardio-Pulmonary Institute (CPI), Member of the German Center for Lung Research (DZL), Giessen, Germany; [2]University of Michigan, Ann Arbor, MI, USA; [3]Irmandade Da Santa Casa de Misericordia de Porto Alegre, Porto Alegre, Brazil; [4]Eastern Virginia Medical School, Macon and Joan Brock Virginia Health Sciences at Old Dominion University, Sentara Health, Norfolk, VA, USA; [5]University of California Los Angeles, UCLA Medical Center, Los Angeles, CA, USA; [6]UT Southwestern Medical Center, Dallas, TX, USA; [7]Mayo Clinic, Rochester, MN, USA; [8]Vanderbilt University Medical Center, Nashville, TN, USA; [9]Imperial College Healthcare NHS Trust, Hammersmith Hospital, London, UK; [10]Stanford University School of Medicine, Stanford, CA, USA; [11]Krakowski Szpital Specjalistyczny im. św. Jana Pawla II, Krakow, Poland; [12]P. Stradins Clinical University Hospital, Riga, Latvia; [13]Unidad de Investigación Clínica En Medicina SC, Monterrey, NL, Mexico; [14]Cardiología Palermo, Buenos Aires, Argentina; [15]Hospital Universitario 12 de Octubre, Madrid, Spain; [16]University of Rochester, Rochester, NY, USA; [17]Instituto Nacional de Cardiología Ignacio Chávez, Mexico City, Mexico; [18]Instituto de Cardiologia de Corrientes Juana Francisca Cabral, Corrientes, Argentina; [19]Klinikum der Universität zu Köln, Herzzentrum, PAH-Ambulanz, Cologne, Germany; [20]Azienda Ospedaliero Universitaria Policlinico Umberto I, Rome, Italy; [21]Fondazione IRCCS Policlinico San Matteo, Pavia, Italy; [22]Hospital Dia do Pulmão, Blumenau – SC, Brazil; [23]NYU Langone Health, New York, NY, USA; [24]Piedmont Healthcare, Atlanta, GA, USA; [25]Institut Klinické A Experimentlni Medíciny, Prague, Czechia; [26]Hospital Madre Teresa, Belo Horizonte – MG, Brazil; [27]Enroll SpA – PPDS, Santiago, Chile; [28]Gossamer Bio, Inc., San Diego, CA, USA; [29]Université Libre de Bruxelles, HUB – Hôpital Erasme, Bruxelles, Belgium; [30]Université Paris–Saclay, Hôpital Bicêtre (AP-HP), Le Kremlin-Bicêtre, France.

Presented at the
ATS International Conference
Orlando, FL, USA
May 15–20, 2026

BACKGROUND

- Vascular remodeling in pulmonary arterial hypertension (PAH) is driven by smooth muscle proliferation, inflammation, and fibrosis—processes mediated in part through PDGFRα/β, CSF1R, and c-KIT[1,2]
- Seralutinib is an investigational, inhaled TKI that selectively inhibits these pathways and restores BMPR2[3]
- In the phase 2 TORREY study, seralutinib reduced PVR in a prevalent PAH population on standard-of-care therapy; greater benefit was observed in Functional Class (FC) III and in patients with REVEAL 2.0 risk score ≥6;[4] and informed the enrichment strategy for PROSERA

METHODS

- The phase 3 PROSERA study evaluated the effectiveness and safety/tolerability of seralutinib in patients with WHO Group 1 PAH, FC II-III, on stable background therapy (**Figure 1**)

Figure 1. PROSERA study schema

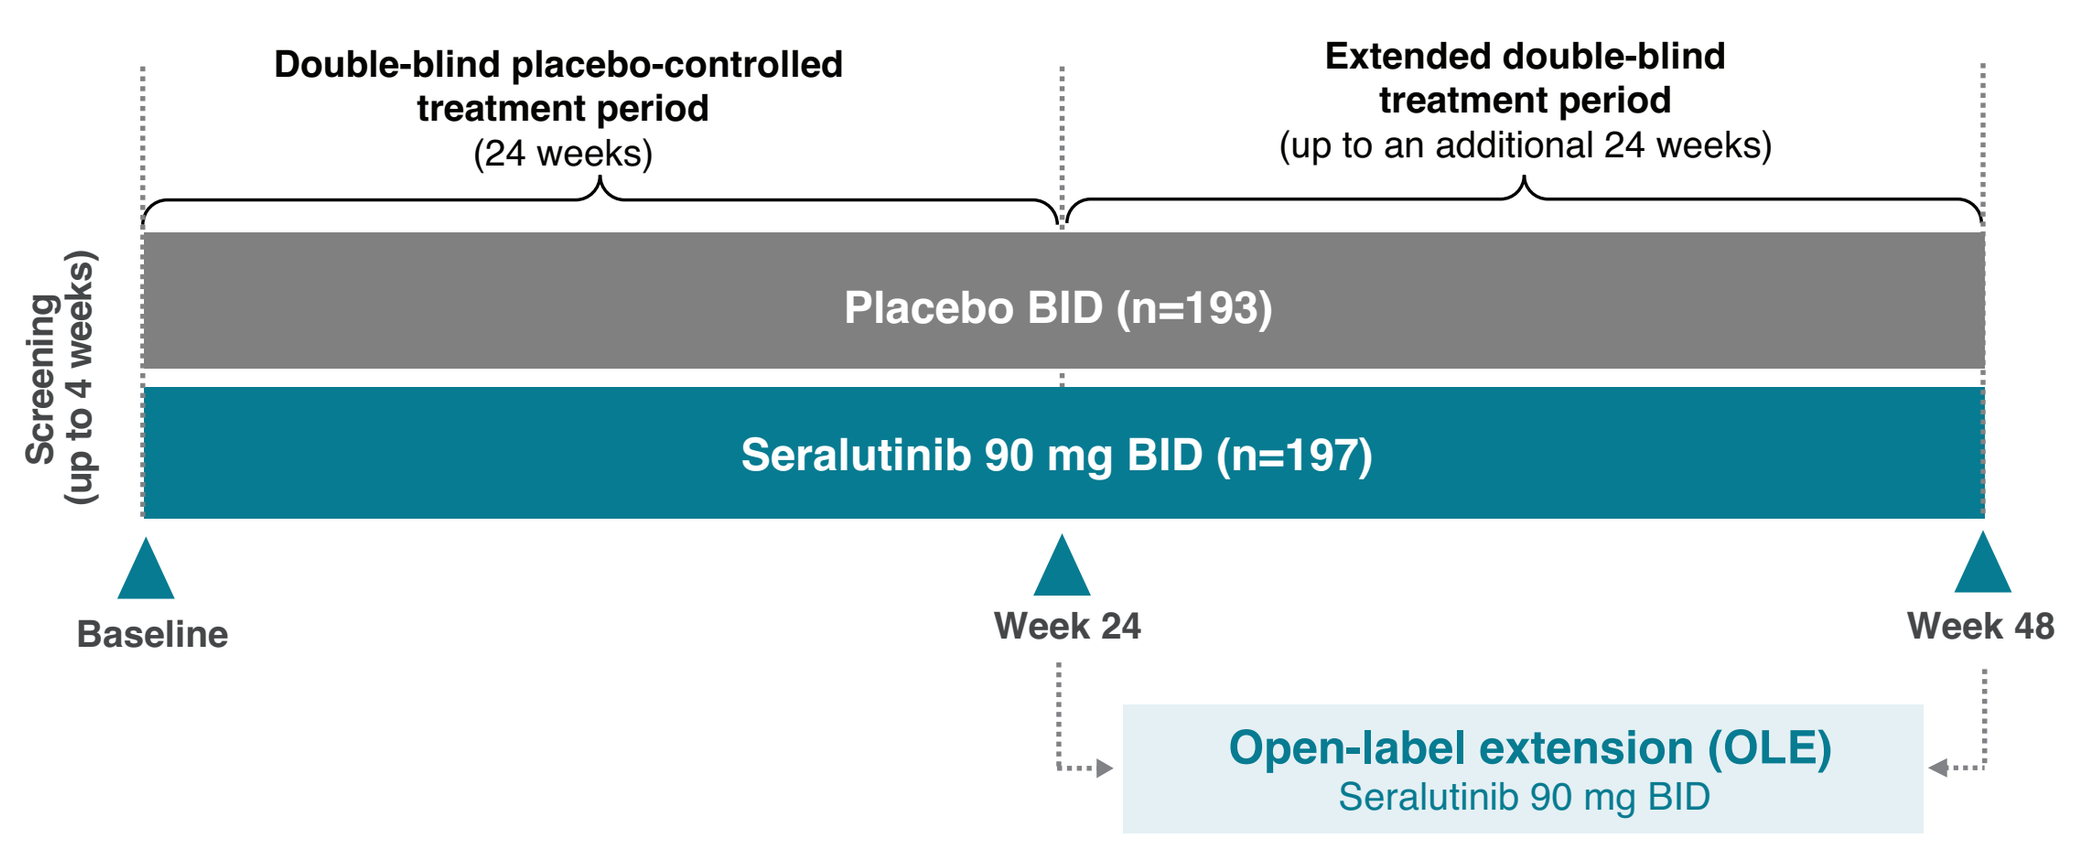
Double-blind placebo-controlled treatment period
(24 weeks)
Extended double-blind treatment period
(up to an additional 24 weeks)
Screening (up to 4 weeks)
Placebo BID (n=193)
Seralutinib 90 mg BID (n=197)
Baseline
Week 24
Week 48
Open-label extension (OLE)
Seralutinib 90 mg BID

- **Treatment period:** Up to a maximum of 48 weeks or until the last subject completed Week 24, whichever occurred first
- **Entry into OLE study:** After completion of Week 48 visit OR adjudicated clinical worsening event after Week 24 OR last study subject completed Week 24

INCLUSION CRITERIA

- Age ≥18 and ≤75 years old
- WHO Group 1 PH, FC II or III
- Baseline 6MWD 150–475 m
- Either REVEAL Lite 2 risk score ≥5 OR NT-proBNP ≥300 ng/L OR PVR ≥800 dyne·s/cm[5]
- Stable treatment with ≥1 SOC PAH therapy (≥12 wk. prior to screening and throughout screening period [6 mo. for sotatercept])

PRIMARY ENDPOINT (WEEK 24)

- Δ in 6MWD (prespecified statistical α=0.025)

KEY SECONDARY ENDPOINTS

- Δ in NT-proBNP
- Reduction of REVEAL Lite 2 risk score
- Time to clinical worsening (TTCW)
- Clinical improvement

Table 1. Selected baseline demographics and disease characteristics. Treatment arms were well balanced.

Characteristic	Placebo (n=193)	Seralutinib (n=197)
Age, mean (SD), years	49.9 (13.9)	50.1 (13.9)
Sex, female, n (%)	164 (85.0)	170 (86.3)
PAH classification, n (%)		
Idiopathic / Heritable	113 (58.5) / 25 (13.0)	126 (64.0) / 13 (6.6)
Associated with CTD	42 (21.8)	45 (22.8)
Other*	13 (6.7)	13 (6.6)
Time from PAH diagnosis, median (min, max), years	4.7 (0.2, 35.5)	5.8 (0.1, 38.4)
WHO FC II / III at screening, n (%)	49 (25.4) / 144 (74.6)	51 (25.9) / 146 (74.1)
6MWD, median, m	389.0	396.0
NT-proBNP, median, ng/L	422.0	451.0
Number of background PAH-specific medications, n (%)		
1 / 2	9 (4.7) / 76 (39.4)	13 (6.6) / 78 (39.6)
≥3	108 (56.0)	106 (53.8)
Prostacyclin/PRA, n (%)	120 (62.2)	117 (59.4)
Inhaled / oral	13 (6.7) / 50 (25.9)	14 (7.1) / 49 (24.9)
Parenteral	59 (30.6)	56 (28.4)

*PAH associated with anorexigen use, methamphetamine use, or congenital heart disease with corrected simple systemic-to-pulmonary shunt.

RESULTS

Figure 2. Primary endpoint: Δ6MWD at Week 24. Seralutinib increased 6MWD from baseline to Week 24 in the overall population by 28.2 m vs 13.5 m in the placebo group, yielding a placebo-corrected effect of 13.3 m (p=0.032), which did not meet the prespecified α=0.025 (Aligned-Rank Stratified Wilcoxon [ARSW] with Multiple Imputation [MI]; Intention-to-Treat [ITT] population).

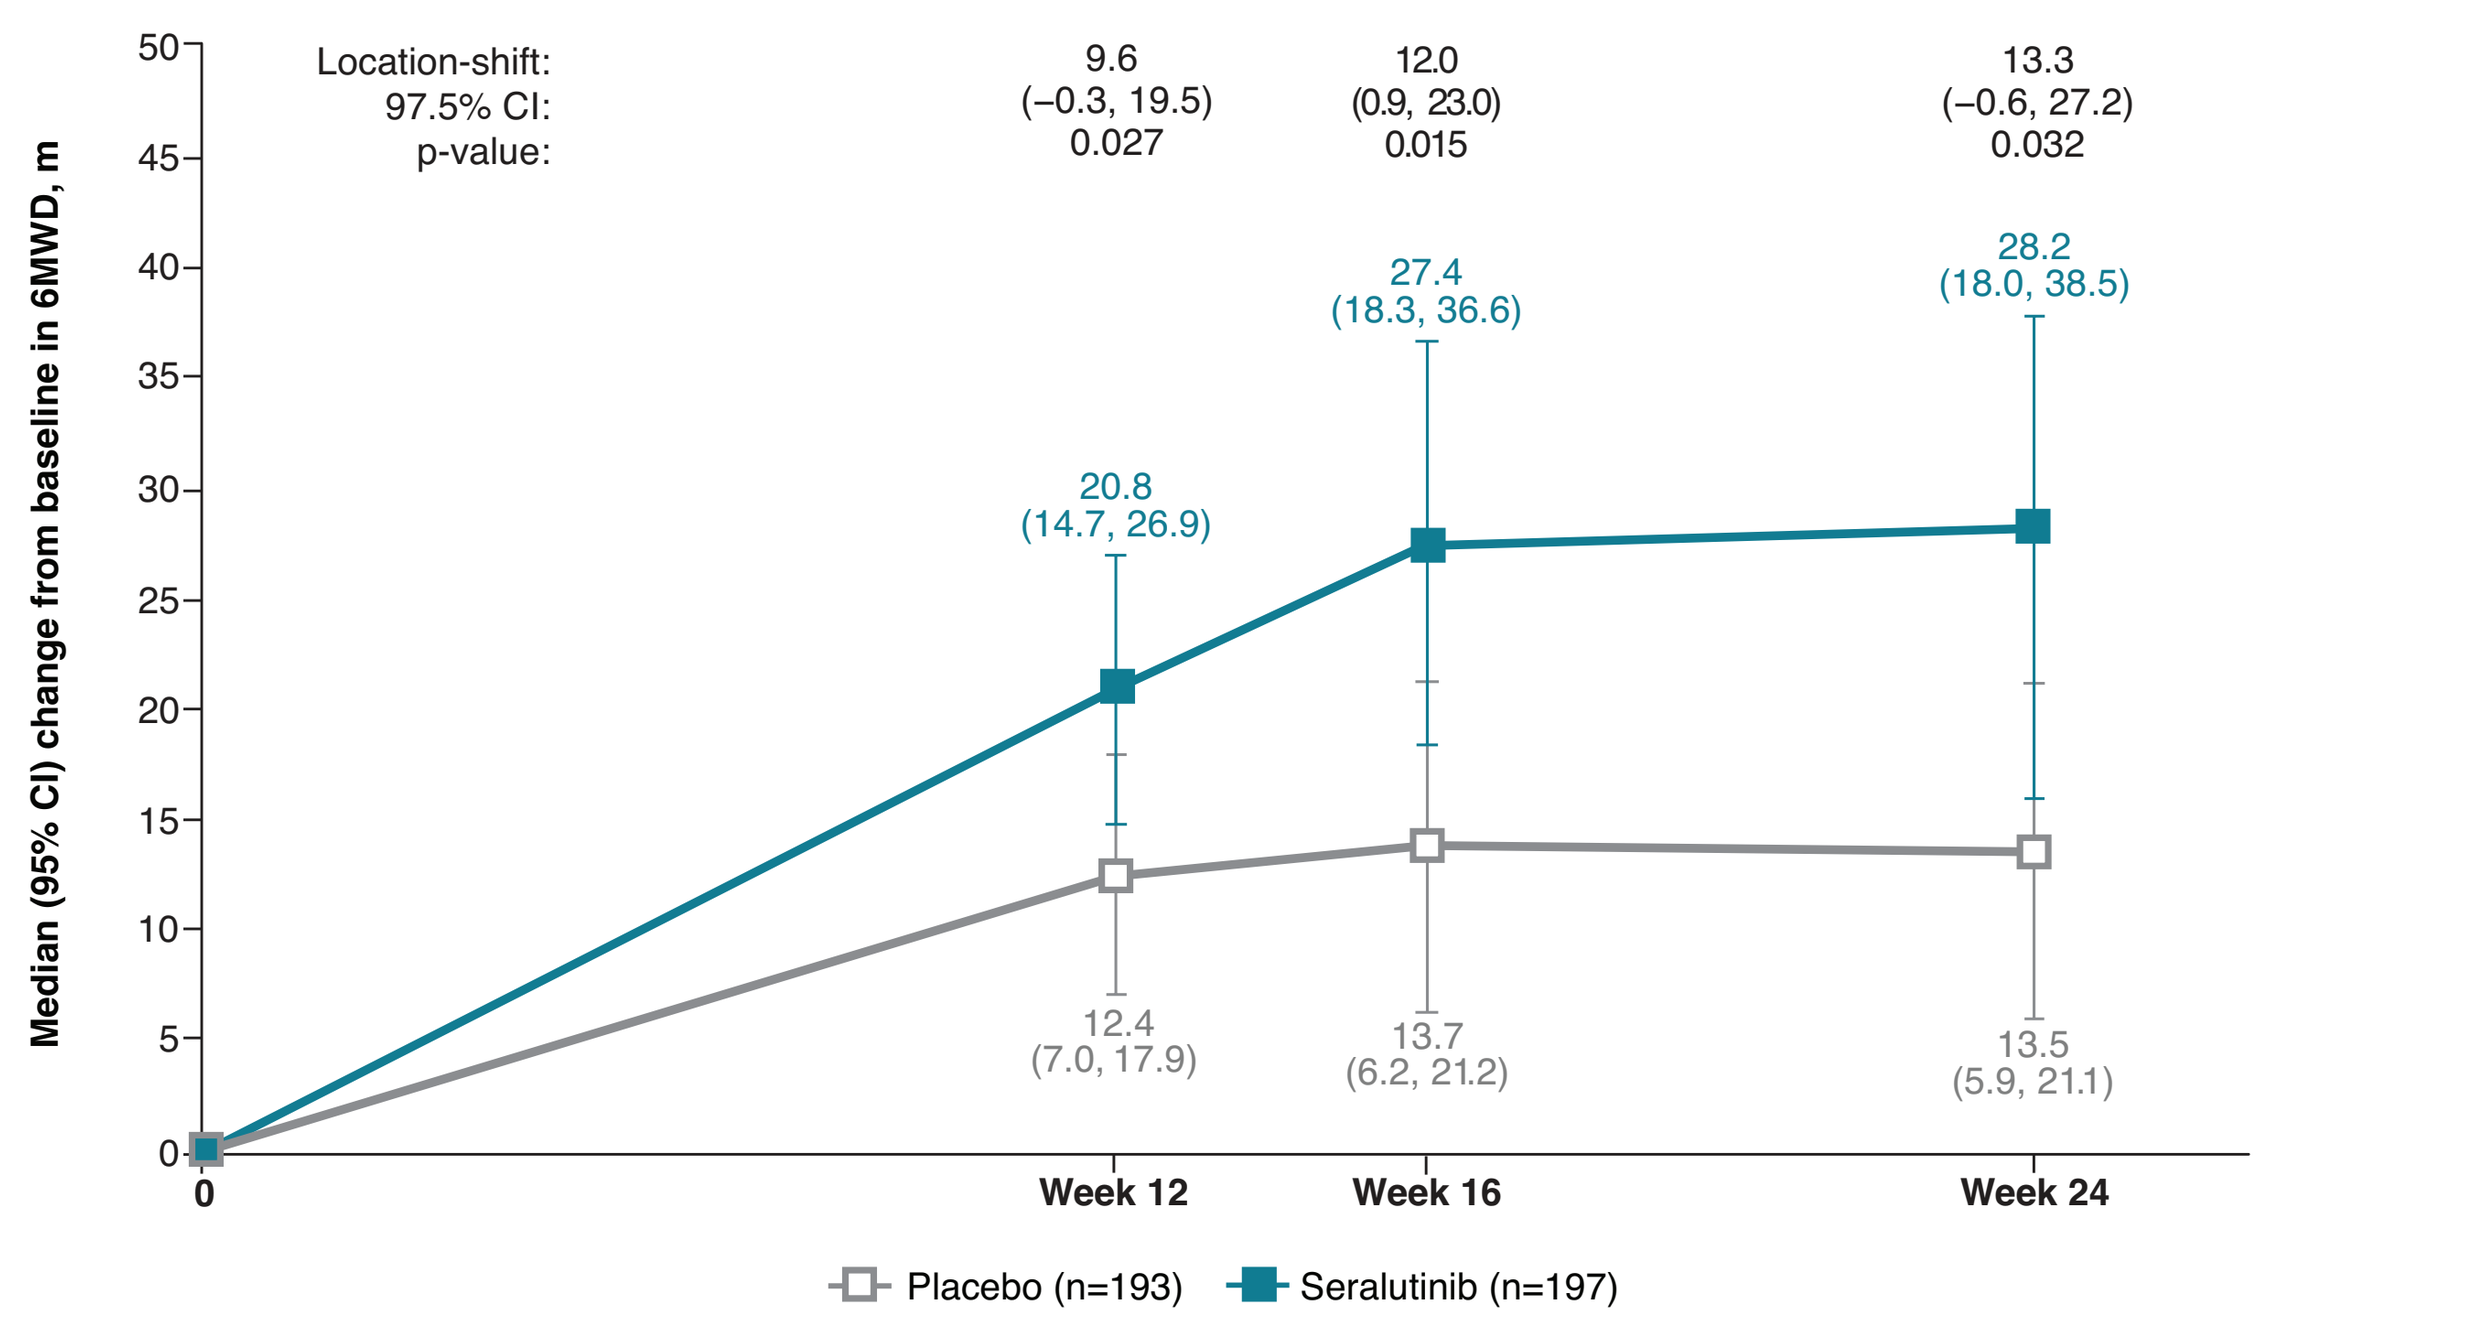
Location-shift:
97.5% CI:
p-value:
9.6
(−0.3, 19.5)
0.027
12.0
(0.9, 23.0)
0.015
13.3
(−0.6, 27.2)
0.032
20.8
(14.7, 26.9)
27.4
(18.3, 36.6)
28.2
(18.0, 38.5)
12.4
(7.0, 17.9)
13.7
(6.2, 21.2)
13.5
(5.9, 21.1)
Median (95% CI) change from baseline in 6MWD, m
0
Week 12
Week 16
Week 24
Placebo (n=193)
Seralutinib (n=197)

Figure 3. Change in 6MWD by prespecified subgroups (ARSW with MI; ITT population).

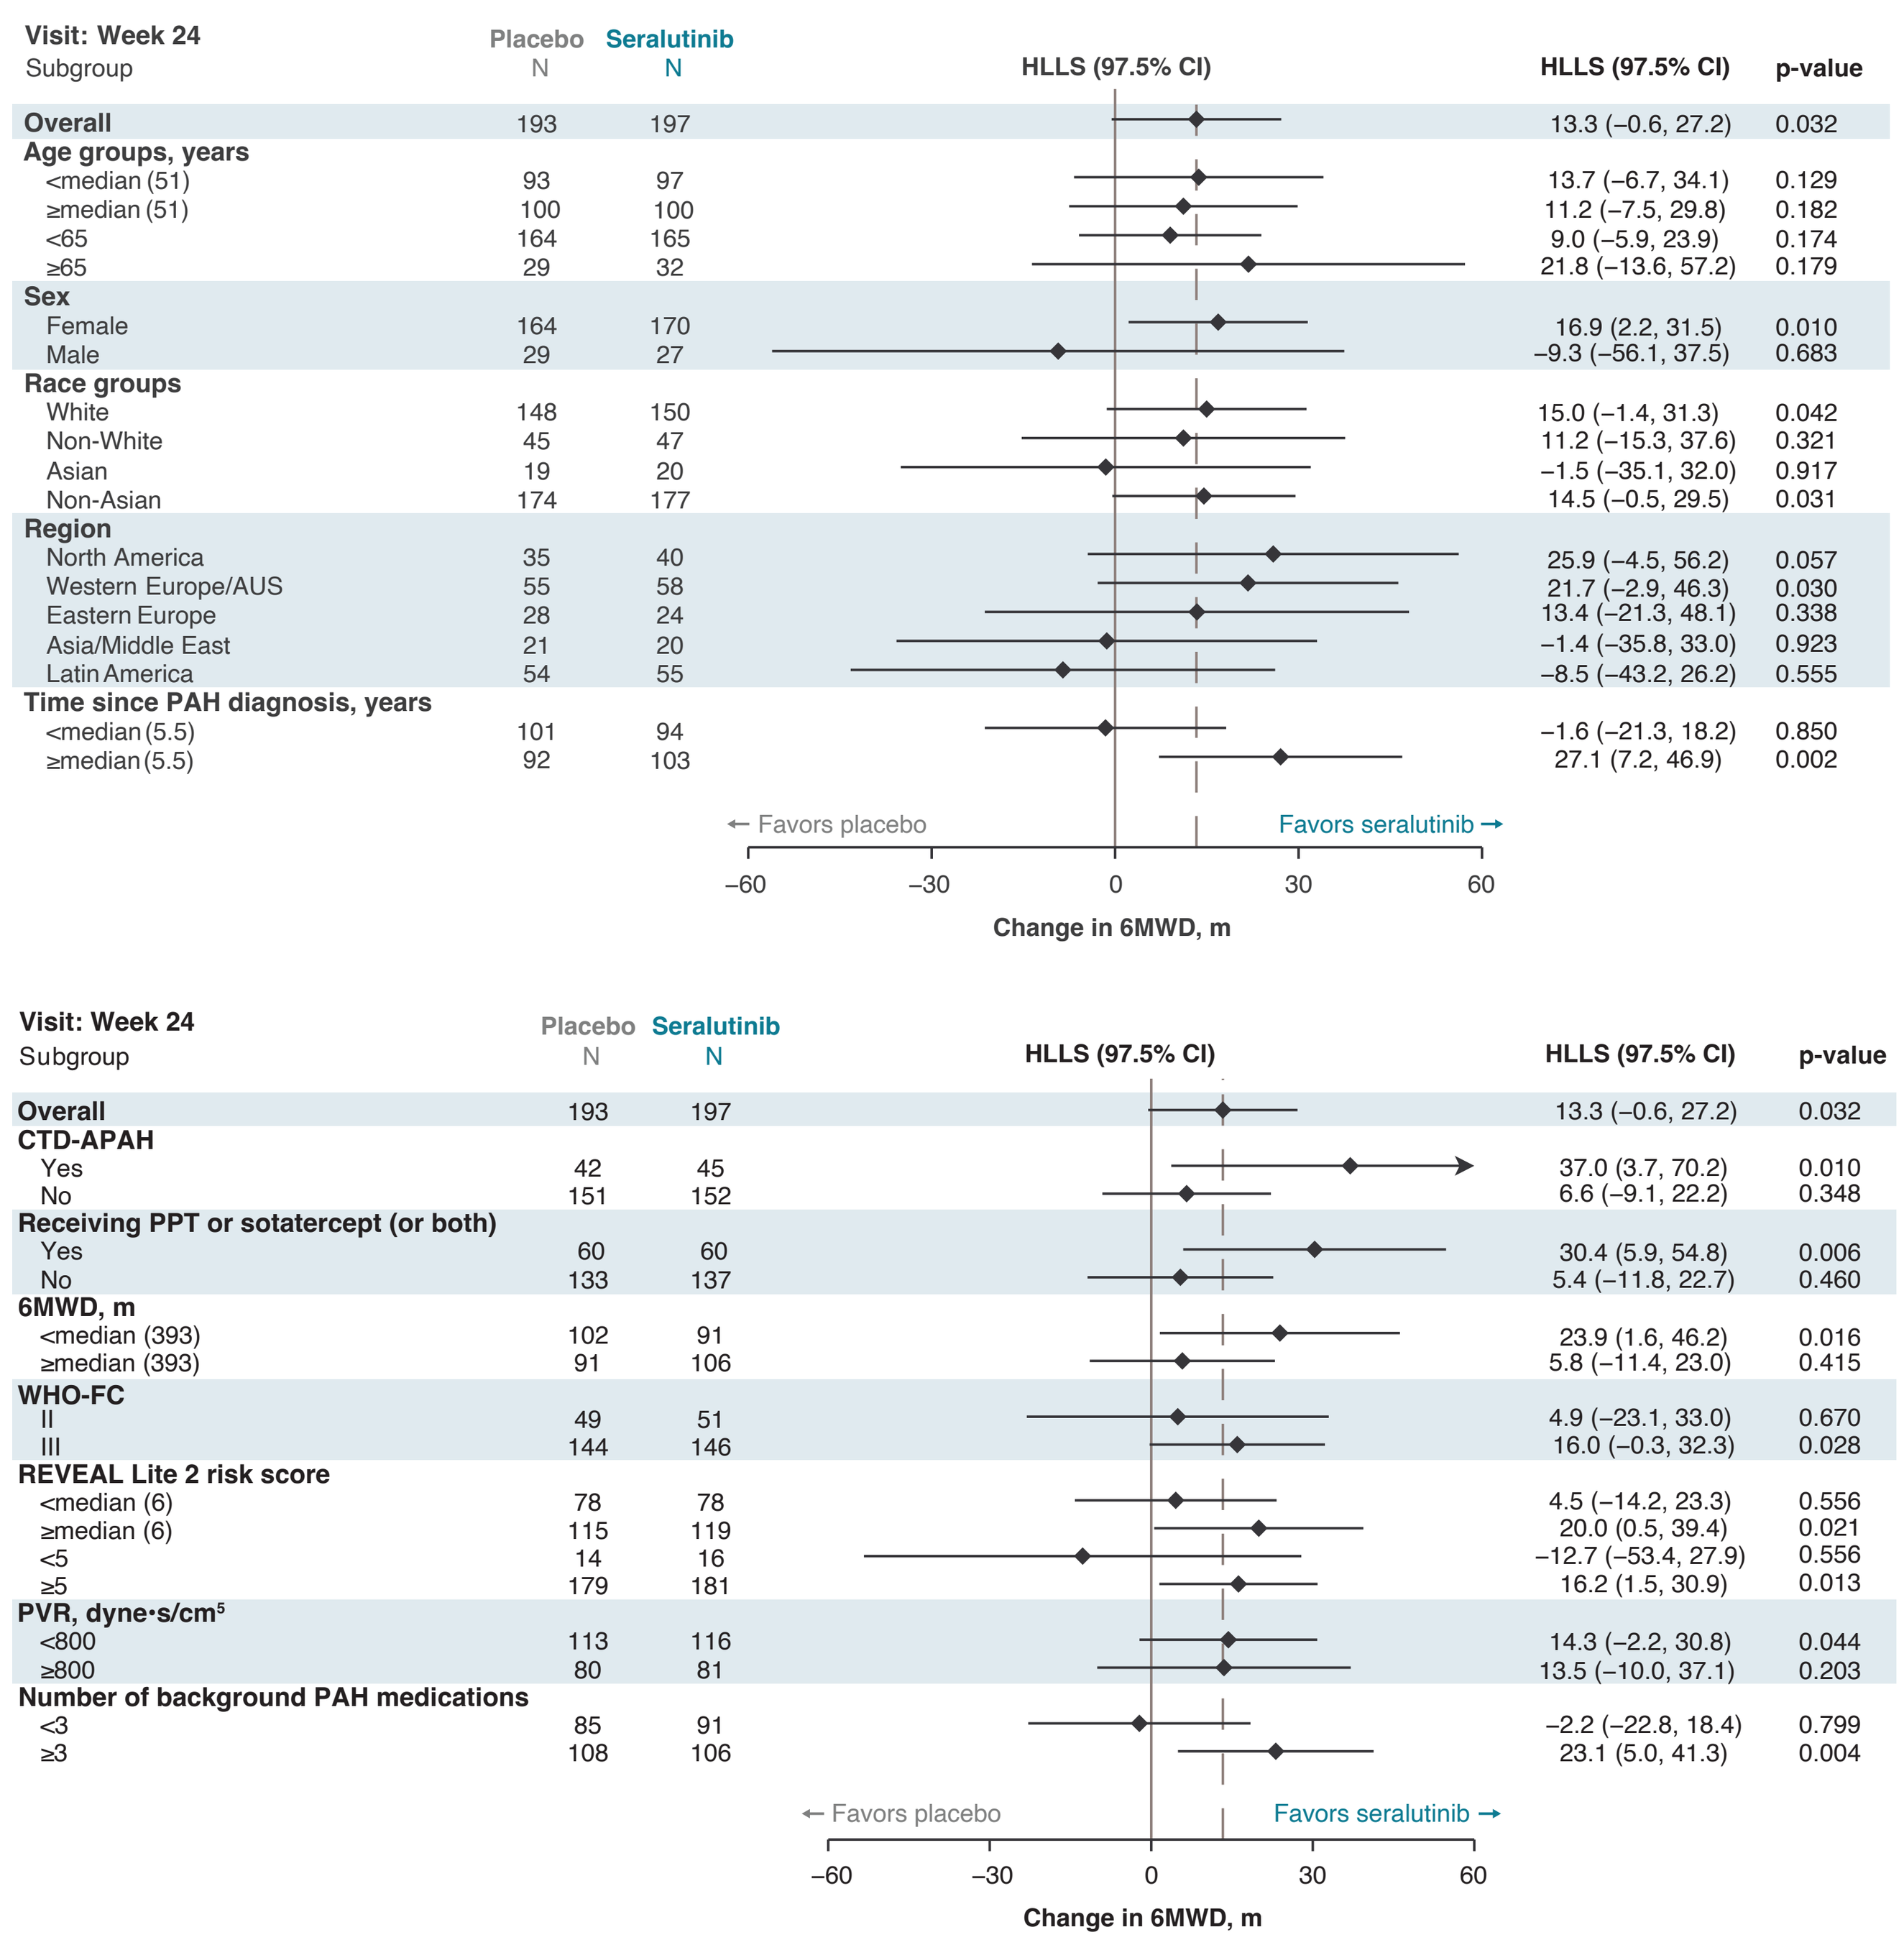
Visit: Week 24
Subgroup | Placebo N | Seralutinib N | HLLS (97.5% CI) | p-value
Overall | 193 | 197 | 13.3 (−0.6, 27.2) | 0.032
Age groups, years
<median (51) | 93 | 97 | 13.7 (−6.7, 34.1) | 0.129
≥median (51) | 100 | 100 | 11.2 (−7.5, 29.8) | 0.182
<65 | 164 | 165 | 9.0 (−5.9, 23.9) | 0.174
≥65 | 29 | 32 | 21.8 (−13.6, 57.2) | 0.179
Sex
Female | 164 | 170 | 16.9 (2.2, 31.5) | 0.010
Male | 29 | 27 | −9.3 (−56.1, 37.5) | 0.683
Race groups
White | 148 | 150 | 15.0 (−1.4, 31.3) | 0.042
Non-White | 45 | 47 | 11.2 (−15.3, 37.6) | 0.321
Asian | 19 | 20 | −1.5 (−35.1, 32.0) | 0.917
Non-Asian | 174 | 177 | 14.5 (−0.5, 29.5) | 0.031
Region
North America | 35 | 40 | 25.9 (−4.5, 56.2) | 0.057
Western Europe/AUS | 55 | 58 | 21.7 (−2.9, 46.3) | 0.030
Eastern Europe | 28 | 24 | 13.4 (−21.3, 48.1) | 0.338
Asia/Middle East | 21 | 20 | −1.4 (−35.8, 33.0) | 0.923
Latin America | 54 | 55 | −8.5 (−43.2, 26.2) | 0.555
Time since PAH diagnosis, years
<median (5.5) | 101 | 94 | −1.6 (−21.3, 18.2) | 0.850
≥median (5.5) | 92 | 103 | 27.1 (7.2, 46.9) | 0.002
← Favors placebo
Favors seralutinib →
Change in 6MWD, m
Visit: Week 24
Subgroup | Placebo N | Seralutinib N | HLLS (97.5% CI) | p-value
Overall | 193 | 197 | 13.3 (−0.6, 27.2) | 0.032
CTD-APAH
Yes | 42 | 45 | 37.0 (3.7, 70.2) | 0.010
No | 151 | 152 | 6.6 (−9.1, 22.2) | 0.348
Receiving PPT or sotatercept (or both)
Yes | 60 | 60 | 30.4 (5.9, 54.8) | 0.006
No | 133 | 137 | 5.4 (−11.8, 22.7) | 0.460
6MWD, m
<median (393) | 102 | 91 | 23.9 (1.6, 46.2) | 0.016
≥median (393) | 91 | 106 | 5.8 (−11.4, 23.0) | 0.415
WHO-FC
II | 49 | 51 | 4.9 (−23.1, 33.0) | 0.670
III | 144 | 146 | 16.0 (−0.3, 32.3) | 0.028
REVEAL Lite 2 risk score
<median (6) | 78 | 78 | 4.5 (−14.2, 23.3) | 0.556
≥median (6) | 115 | 119 | 20.0 (0.5, 39.4) | 0.021
<5 | 14 | 16 | −12.7 (−53.4, 27.9) | 0.556
≥5 | 179 | 181 | 16.2 (1.5, 30.9) | 0.013
PVR, dyne·s/cm5
<800 | 113 | 116 | 14.3 (−2.2, 30.8) | 0.044
≥800 | 80 | 81 | 13.5 (−10.0, 37.1) | 0.203
Number of background PAH medications
<3 | 85 | 91 | −2.2 (−22.8, 18.4) | 0.799
≥3 | 108 | 106 | 23.1 (5.0, 41.3) | 0.004
← Favors placebo
Favors seralutinib →
Change in 6MWD, m

Figure 4. Key secondary endpoints. A. Seralutinib demonstrated a robust reduction in NT-proBNP at Week 24 vs placebo (−120 ng/L, nominal p=0.0002), with separation between groups evident as early as Week 4. **B.** Two of three additional key secondary endpoints numerically favored seralutinib over placebo. (Panels show data for ARSW with MI; ITT population.)

A.

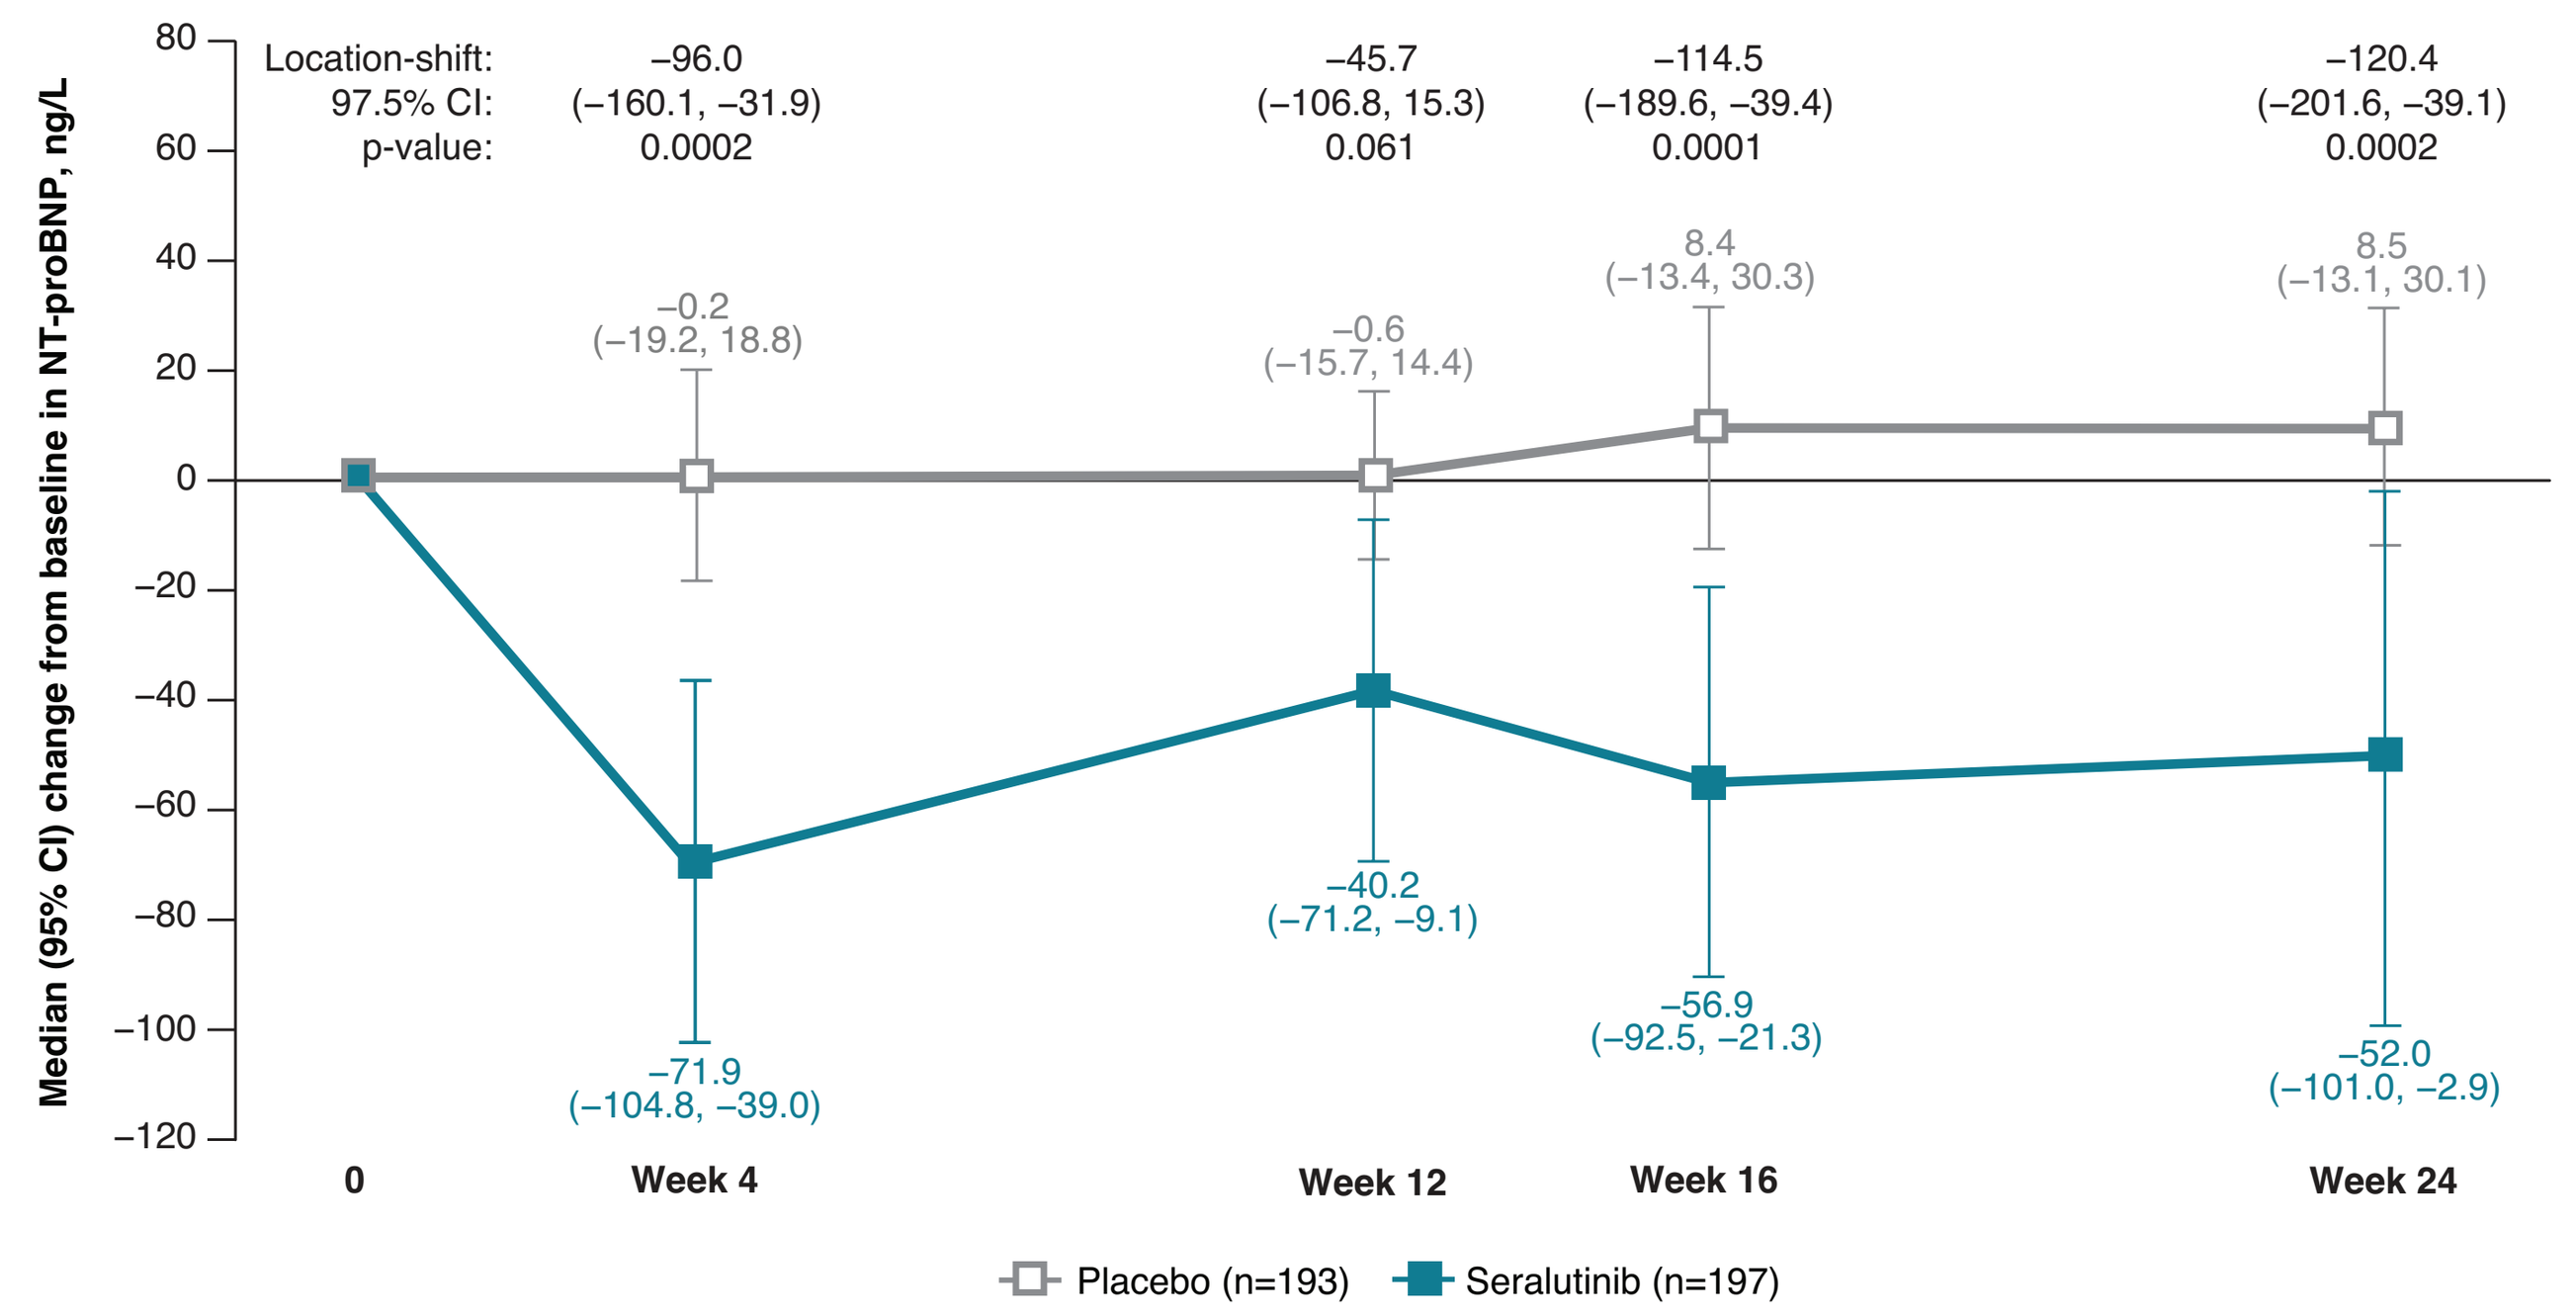
Location-shift:
97.5% CI:
p-value:
−96.0
(−160.1, −31.9)
0.0002
−45.7
(−106.8, 15.3)
0.061
−114.5
(−189.6, −39.4)
0.0001
−120.4
(−201.6, −39.1)
0.0002
−0.2
(−19.2, 18.8)
−0.6
(−15.7, 14.4)
8.4
(−13.4, 30.3)
8.5
(−13.1, 30.1)
−71.9
(−104.8, −39.0)
−40.2
(−71.2, −9.1)
−56.9
(−92.5, −21.3)
−52.0
(−101.0, −2.9)
Median (95% CI) change from baseline in NT-proBNP, ng/L
0
Week 4
Week 12
Week 16
Week 24
Placebo (n=193)
Seralutinib (n=197)

B.

Overall study population (placebo, n=193; seralutinib, n=197)	OR/HR	97.5% CI	Nominal p-value
Clinical improvement	OR 1.64	0.9, 3.1	0.081
≥1-point reduction in REVEAL Lite 2	OR 1.42	0.9, 2.2	0.088
TTCW	HR 0.83	0.4, 1.7	0.562

HR, hazard ratio; OR, odds ratio.

Figure 5. Primary and key secondary endpoints for prespecified subgroups. Seralutinib demonstrated clinically meaningful treatment effects in 6MWD in **A.** intermediate- and high-risk patients (REVEAL Lite 2 risk score ≥median (median=6); placebo-adjusted Δ6MWD +20.0 m, nominal p=0.021) and **B.** patients with connective tissue disease-associated PAH (CTD-PAH; placebo-adjusted Δ6MWD +37.0 m, nominal p=0.01). **C.** Key secondary endpoints for REVEAL Lite 2 risk score ≥6 and CTD-PAH subgroups. (Panels show data for ITT population.)

A. REVEAL Lite 2 risk score ≥6 **B. CTD-PAH**

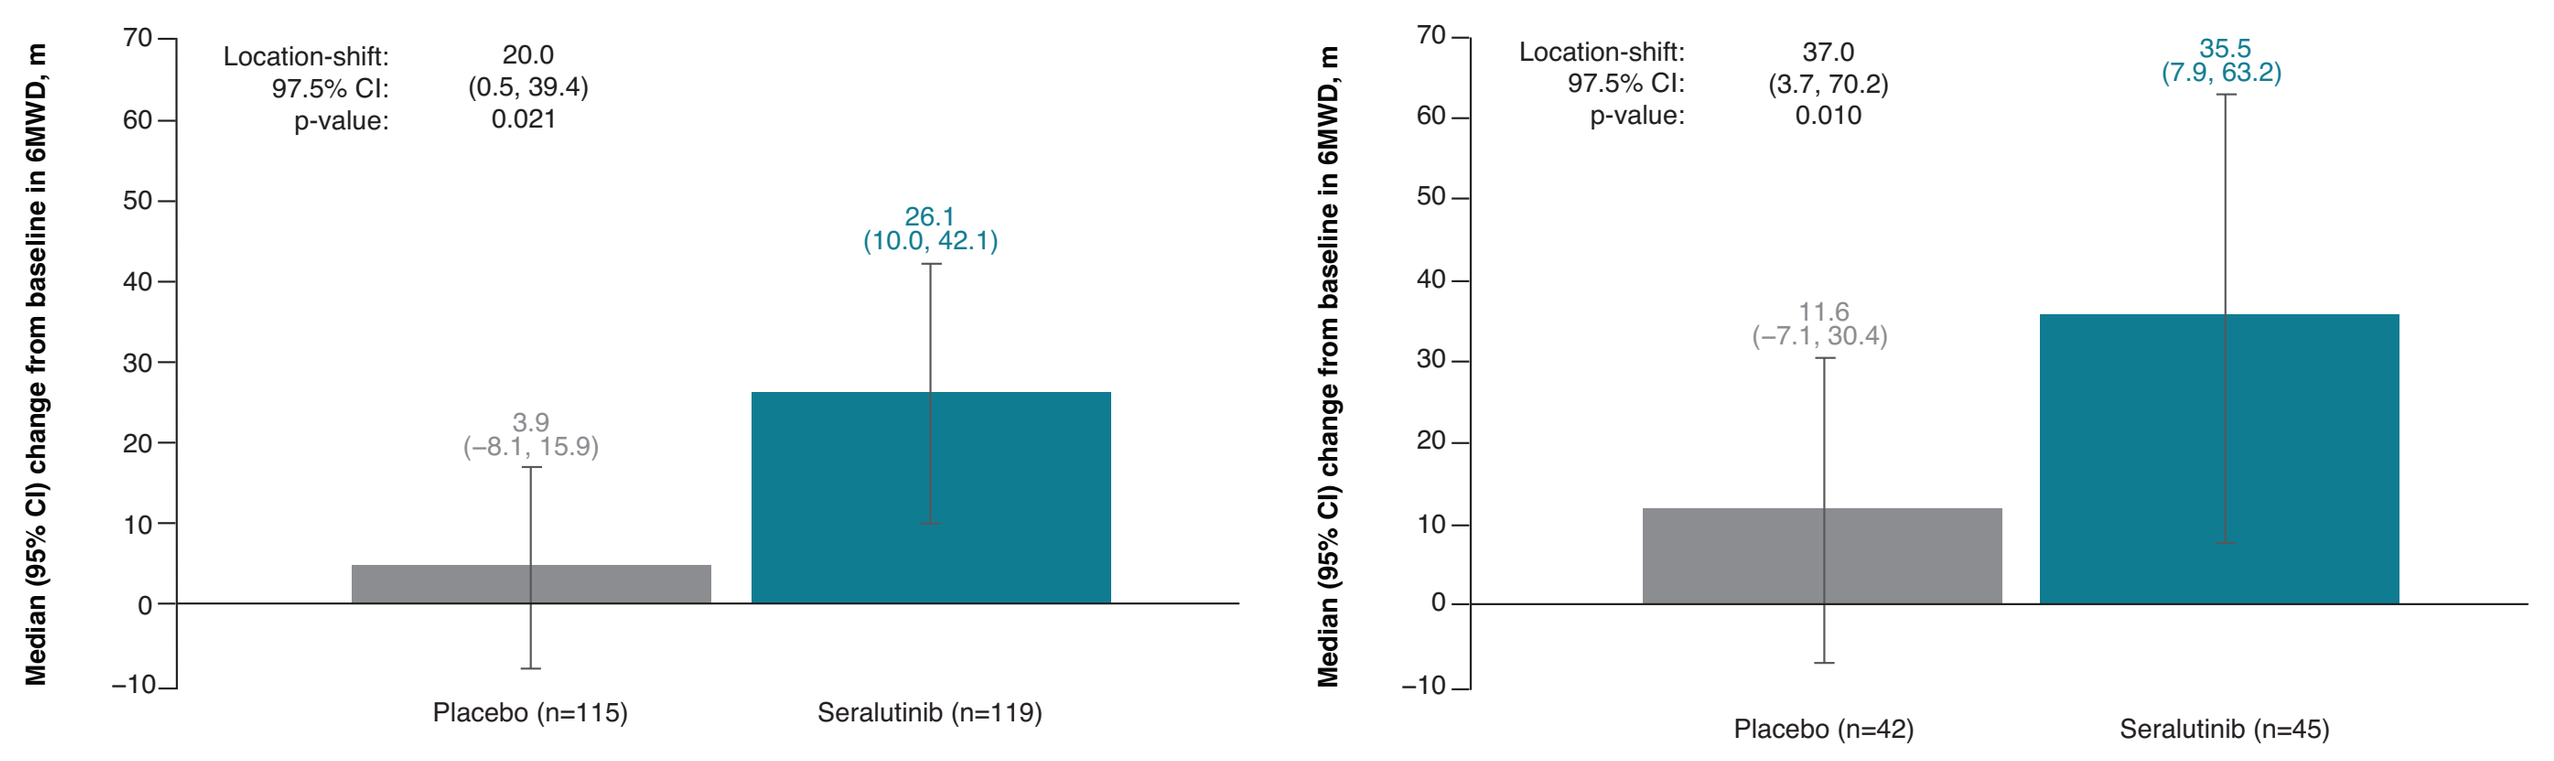
Location-shift:
97.5% CI:
p-value:
20.0
(0.5, 39.4)
0.021
26.1
(10.0, 42.1)
3.9
(−8.1, 15.9)
Median (95% CI) change from baseline in 6MWD, m
Placebo (n=115)
Seralutinib (n=119)
Location-shift:
97.5% CI:
p-value:
37.0
(3.7, 70.2)
0.010
35.5
(7.9, 63.2)
11.6
(−7.1, 30.4)
Median (95% CI) change from baseline in 6MWD, m
Placebo (n=42)
Seralutinib (n=45)

C.

	LS/OR/HR	97.5% CI	Nominal p-value
REVEAL Lite 2 ≥6 (placebo, n=115; seralutinib, n=119)			
NT-proBNP	LS −265.82	−446.2, −85.5	0.0002
Clinical improvement	OR 3.32	1.2, 9.4	0.010
≥1-point reduction in REVEAL Lite 2	OR 2.03	1.1, 3.7	0.008
TTCW (through Week 48)	HR 0.74	0.3, 1.7	0.436
CTD-PAH (placebo, n=42; seralutinib, n=45)			
NT-proBNP	LS −74.29	−289.8, 141.1	0.399
Clinical improvement	OR 1.58	0.3, 8.9	0.550
≥1-point reduction in REVEAL Lite 2	OR 1.98	0.7, 5.4	0.128
TTCW (through Week 48)	HR 0.25	0.03, 1.2	0.082

HR, hazard ratio; LS, location shift; OR, odds ratio.

Table 2. Treatment-emergent adverse events. Overall TEAE and SAE rates were comparable between groups (safety population).

Category, n (%)	Placebo (n=190)*	Seralutinib (n=200)*
Number of patients with at least one:		
TEAE	153 (80.5)	173 (86.5)
TEAE leading to discontinuation of study drug	11 (5.8)	30 (15.0)
AE of special interest	12 (6.3)	41 (20.5)
Major bleeding event	1 (0.5)	4 (2.0)
Cardiac effect	0	1 (0.5)
Hepatic effect[a]	11 (5.8)	36 (18.0)
SAE	36 (18.9)	32 (16.0)
Fatal TEAE[b]	3 (1.6)	4 (2.0)
Number of SAEs	67	53

*Three placebo patients received a seralutinib kit and are treated as exposed; per the SAP, they are counted in the seralutinib safety group. ITT: 193 placebo, 197 seralutinib; Safety: 190 placebo, 200 seralutinib. [a]Including Metabolism & Nutrition Disorders, GI Disorders, Hepatobiliary Disorders, Investigations; [b]Per IDMC, fatal TEAEs were not drug related. AE, adverse event; S, serious; TE, treatment-emergent.

Table 3. Incidence of TEAEs by preferred term: ≥10% in seralutinib group. Seralutinib was generally well tolerated and overall TEAE and SAE rates were comparable between groups. Transaminase elevations were not symptomatic, without clinical sequelae, and resolved with treatment withdrawal (safety population).

Preferred term, n (%)	Placebo (n=190)*	Seralutinib (n=200)*
Number of subjects with a TEAE	153 (80.5)	173 (86.5)
Cough	26 (13.7)	74 (37.0)
Headache	27 (14.2)	32 (16.0)
ALT increased	1 (0.5)	29 (14.5)
AST increased	1 (0.5)	28 (14.0)
ALT or AST >5x ULN	0	10 (5.0)
Nausea	20 (10.5)	24 (12.0)
Diarrhea	26 (13.7)	23 (11.5)

*Three placebo patients received a seralutinib kit and are treated as exposed; per the SAP, they are counted in the seralutinib safety group. ITT: 193 placebo, 197 seralutinib; Safety: 190 placebo, 200 seralutinib.

CONCLUSIONS

- **Primary Endpoint:** Seralutinib increased 6MWD from baseline to Week 24 in the overall population by 28.2 m compared with 13.5 m in the placebo group, yielding a placebo-adjusted treatment effect of 13.3 m (p=0.032; prespecified α=0.025)
- **Secondary Endpoints:** Results favored seralutinib vs placebo at Week 24, including change in NT-proBNP, clinical improvement, and ≥1-point REVEAL Lite 2 risk score reduction
- **Prespecified intermediate- and high-risk subgroups (REVEAL Lite 2 risk score ≥6):** Greater treatment effect on 6MWD vs placebo (placebo-adjusted treatment effect: +20.0 m; nominal p=0.0207), with numerical benefit across three of four key secondary endpoints
- **Prespecified CTD-PAH subgroup:** Seralutinib showed a more pronounced improvement in 6MWD vs placebo (placebo-adjusted treatment effect: +37.0 m; nominal p=0.010)
- **Safety:** Seralutinib was generally well tolerated; the most frequently reported TEAEs were cough and hepatic enzyme elevation reversible with discontinuation of study drug

References: **1** Rothman AMK, et al. *Am J Respir Crit Care Med.* 2025;211(6):1018-1027. **2** Rothman AMK, et al. *Am J Respir Crit Care Med.* 2026;212(4):714-715. **3** Galkin A, et al. *Eur Respir J.* 2022;60(6):2102356. **4** Frantz RP, et al. *Lancet Respir Med.* 2024;12(7):523-534.

Research supported by: GB002, Inc., a subsidiary of Gossamer Bio, Inc., San Diego, CA, USA

H-AG/VVM* and J-LV/OS** are co-first and co-senior authors, respectively, with equal contribution.