Seralutinib shows significant anti-fibrotic effects: Evidence from patient-derived models

PA1864

Ravikumar Sitapara[1], Silvia Cantoni[2], Eduardo Garcia[1], Valentina Violi[2], Zhaoqing Ding[1], Robin Osterhout[1], Lawrence S. Zisman[1], Sidra Hoffman[1], Lawrence Ho[1], Francisca Gavilanes[1], Richard Aranda[1], Daniela Miglietta[2], Tomás Pulido[3], Bruno Guedes Baldi[4], Rogério Souza[4], Jean-Marie Bruey[1], Vincent Cottin[5]

[1]Gossamer Bio, Inc., San Diego, CA, USA, [2]Chiesi Farmaceutici S.p.A., Parma, Italy, [3]Instituto Nacional de Cardiología Ignacio Chávez, México City, D. F., México, [4]Instituto do Coração, Hospital das Clínicas da Faculdade de Medicina da Universidade de São Paulo, São Paulo, Brazil, [5]Centre National de Référence des Maladies Pulmonaires rares, Service de Pneumologie, Hospices Civils de Lyon, Hôpital Louis Pradel, Lyon, France

Presented at:
ERS Congress 2025
27 September–1 October
Amsterdam, Netherlands

BACKGROUND

- Fibrosis contributes to both pulmonary vascular and parenchymal remodeling in pulmonary hypertension (PH) and interstitial lung disease (ILD)[1]
- Seralutinib is a potent, inhaled PDGFRα/ß, CSF1R, and c-KIT kinase inhibitor in clinical development for pulmonary arterial hypertension (PAH) and PH-ILD[2,3] (**Figure 1**)
- In the phase 2 TORREY study (NCT04456998) in patients with Group 1 PH, seralutinib decreased circulating fibrotic biomarkers, including collagen Iα1[4,5]
- We examined whether seralutinib inhibits established fibrosis in idiopathic pulmonary fibrosis (IPF) patient-derived precision-cut lung slices (PCLS) and human lung fibroblasts (HLF)

Figure 1. Seralutinib mechanism of action

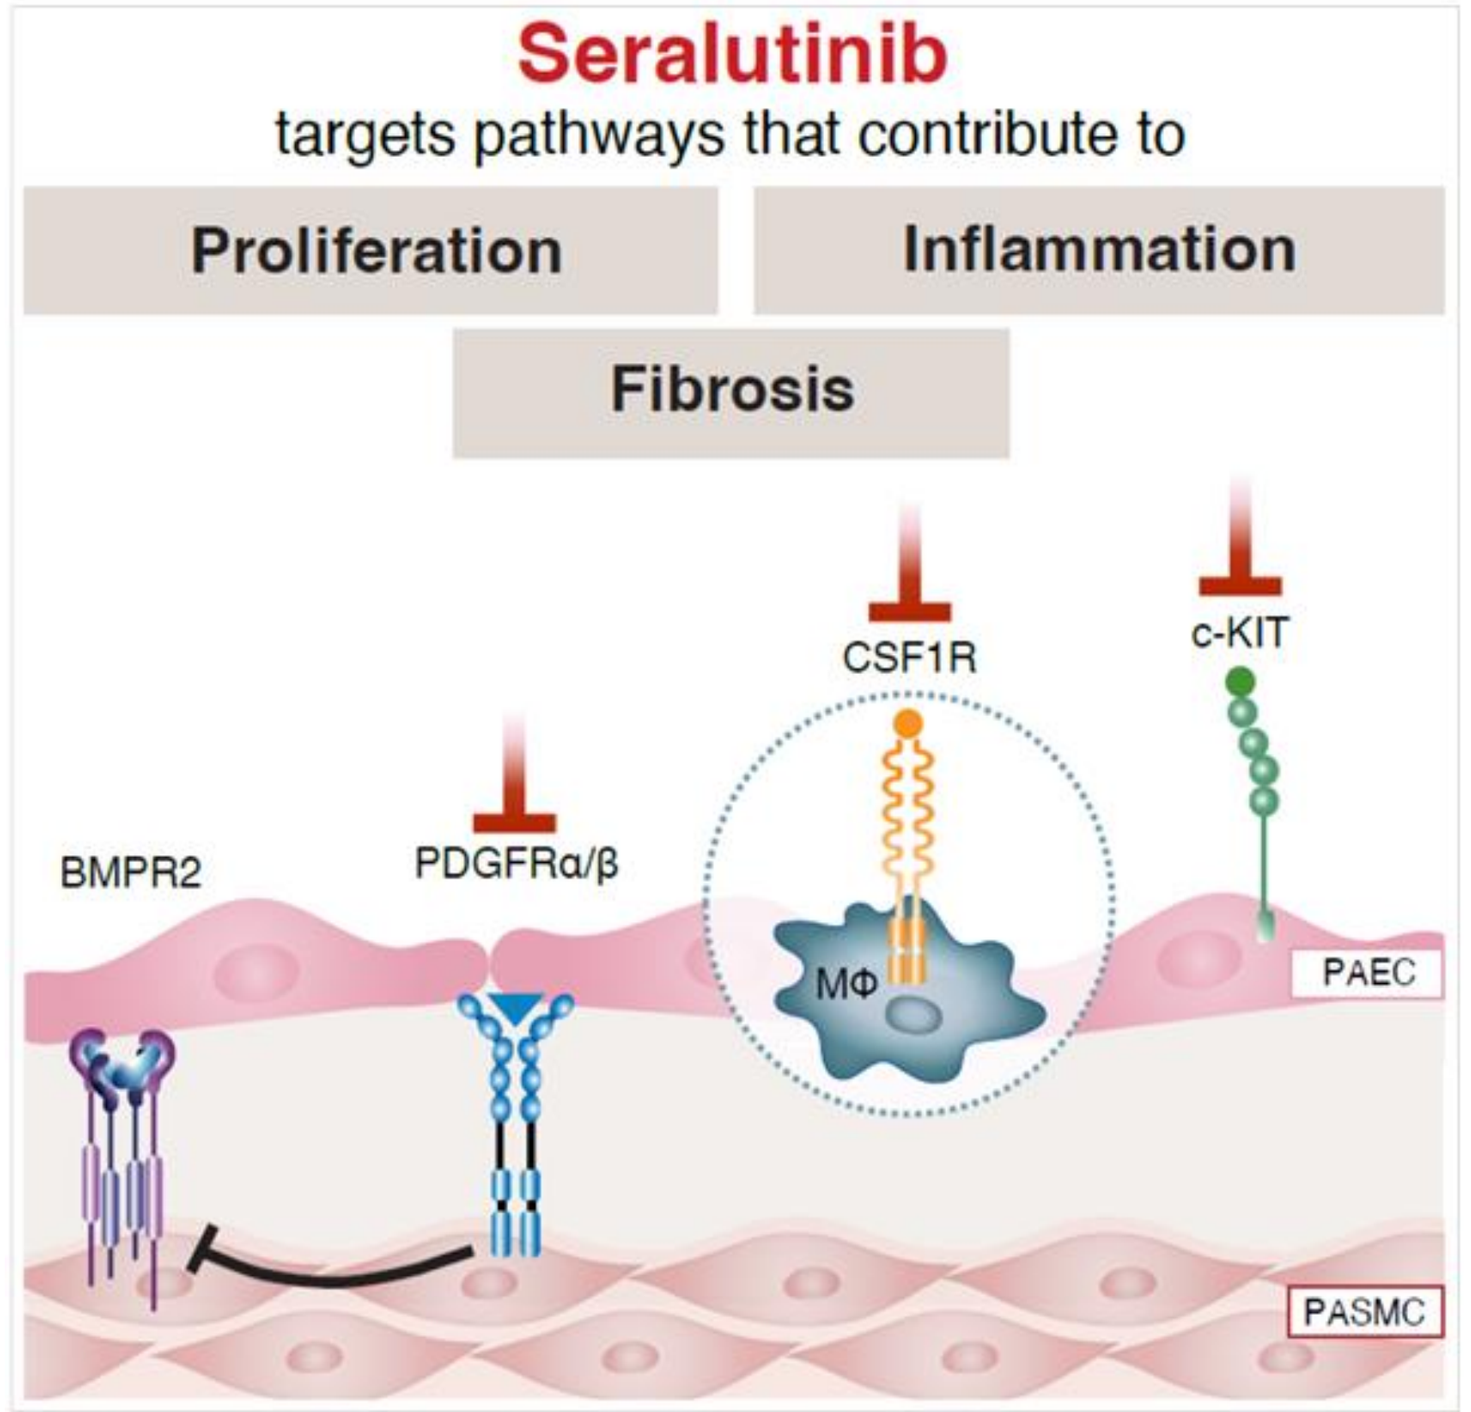
Seralutinib
targets pathways that contribute to
Proliferation
Inflammation
Fibrosis
CSF1R
c-KIT
BMPR2
PDGFRα/β
MΦ
PAEC
PASMC

Blunted arrows indicate inhibition. BMPR2, bone morphogenetic protein receptor type 2; c-KIT, mast/stem cell growth factor receptor kit; CSF1R, colony stimulating factor 1 receptor; MΦ, macrophage; PAEC, pulmonary artery endothelial cell; PASMC, pulmonary artery smooth muscle cell; PDGFR, platelet-derived growth factor receptor.

METHODS

Effect of seralutinib on release of pro-fibrotic markers from IPF human (h) PCLS cultures

- PCLS from two IPF donors were cultured and treated for 4 d with vehicle, seralutinib (0.1-10 µM), or nintedanib (1 µM)[6]
- Supernatants were collected at 96 h and pro-fibrotic markers evaluated using a custom Luminex panel. Cytotoxicity was assessed with an adenylate kinase assay

Figure 2. IPF-hPCLS experimental setup

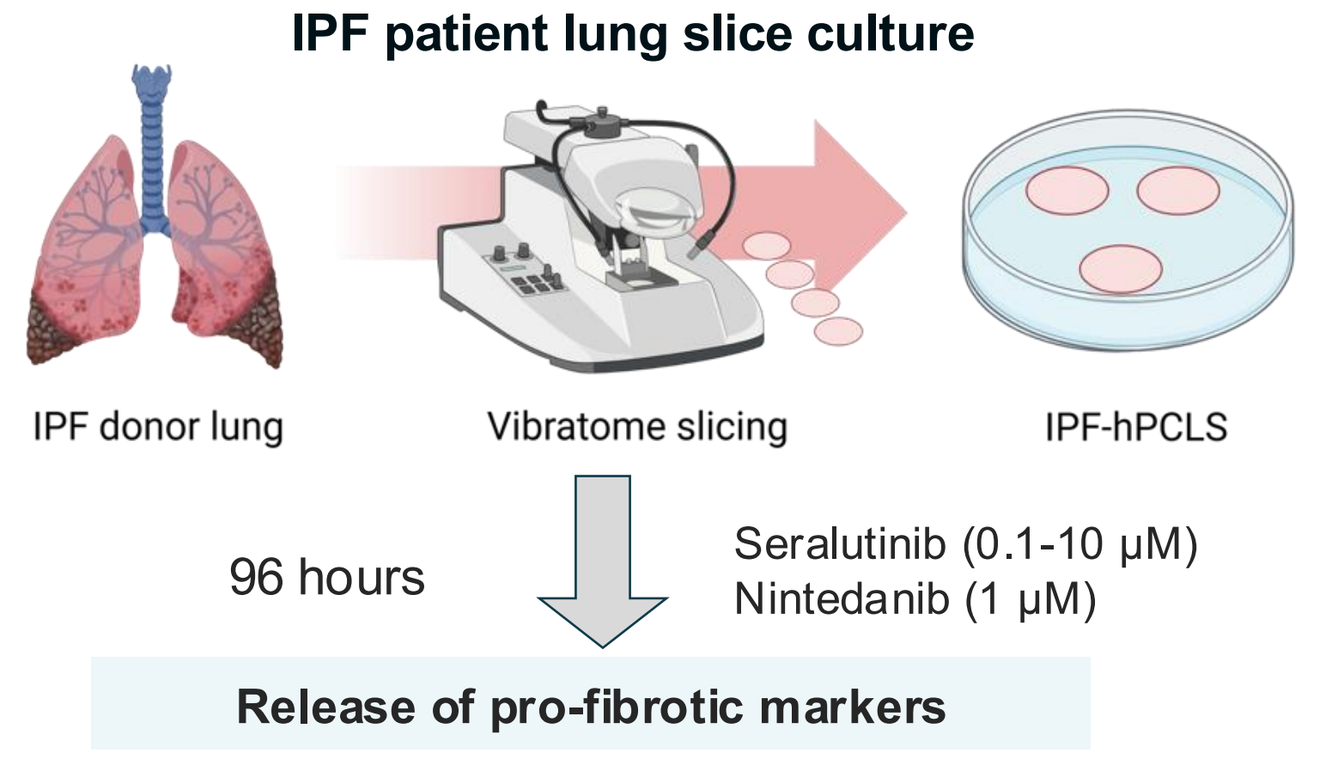
IPF patient lung slice culture
IPF donor lung
Vibratome slicing
IPF-hPCLS
96 hours
Seralutinib (0.1-10 µM)
Nintedanib (1 µM)
Release of pro-fibrotic markers

Effect of seralutinib on release of pro-fibrotic markers in IPF-HLFs

- IPF-HLF from three independent donors were treated with vehicle, seralutinib (0.1-10 µM), or nintedanib (10 µM) for 24, 48, and 72 hours
- Supernatants were collected at each timepoint and pro-fibrotic markers evaluated with a custom Luminex panel

Figure 3. IPF-HLF experimental setup

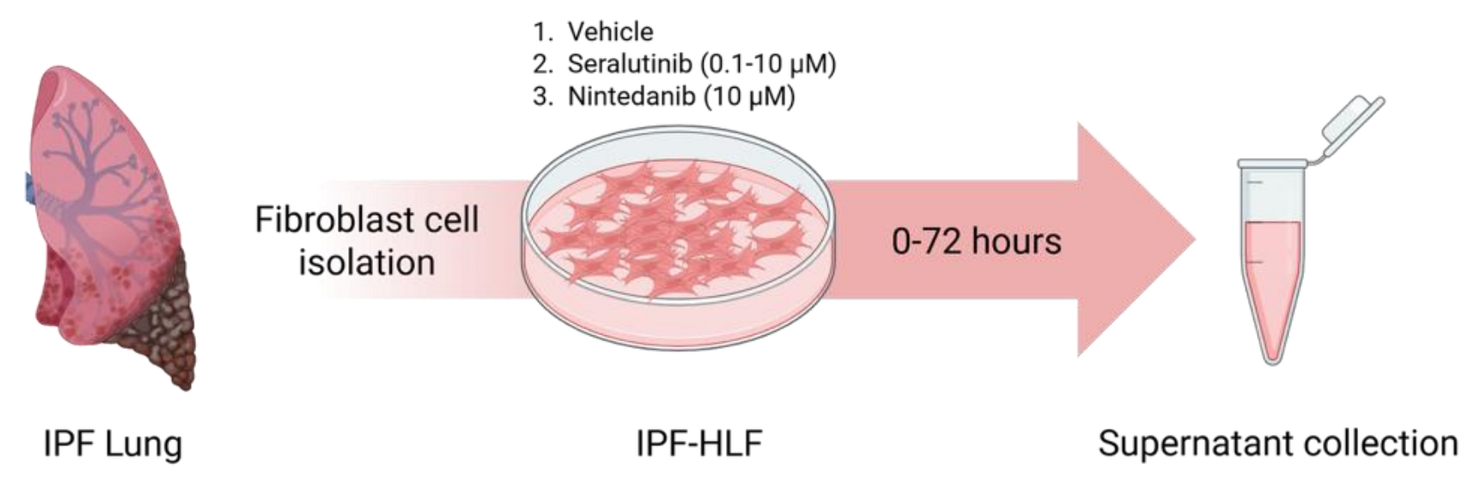
1. Vehicle
2. Seralutinib (0.1-10 µM)
3. Nintedanib (10 µM)
Fibroblast cell isolation
0-72 hours
IPF Lung
IPF-HLF
Supernatant collection

Effect of seralutinib on fibroblast-to-myofibroblast transition in IPF-HLFs

- IPF-HLF were pre-treated with seralutinib (0.1-10 µM) or nintedanib (3 µM) and stimulated with 10 ng/mL TGF-β for 72 hours
- Alpha-smooth muscle actin (α-SMA) gene expression levels were determined by quantitative PCR and protein expression was assessed by immunofluorescent staining

PCR, polymerase chain reaction. Figures created with BioRender.

RESULTS

Figure 4. Seralutinib inhibited release of pro-fibrotic markers in IPF-hPCLS

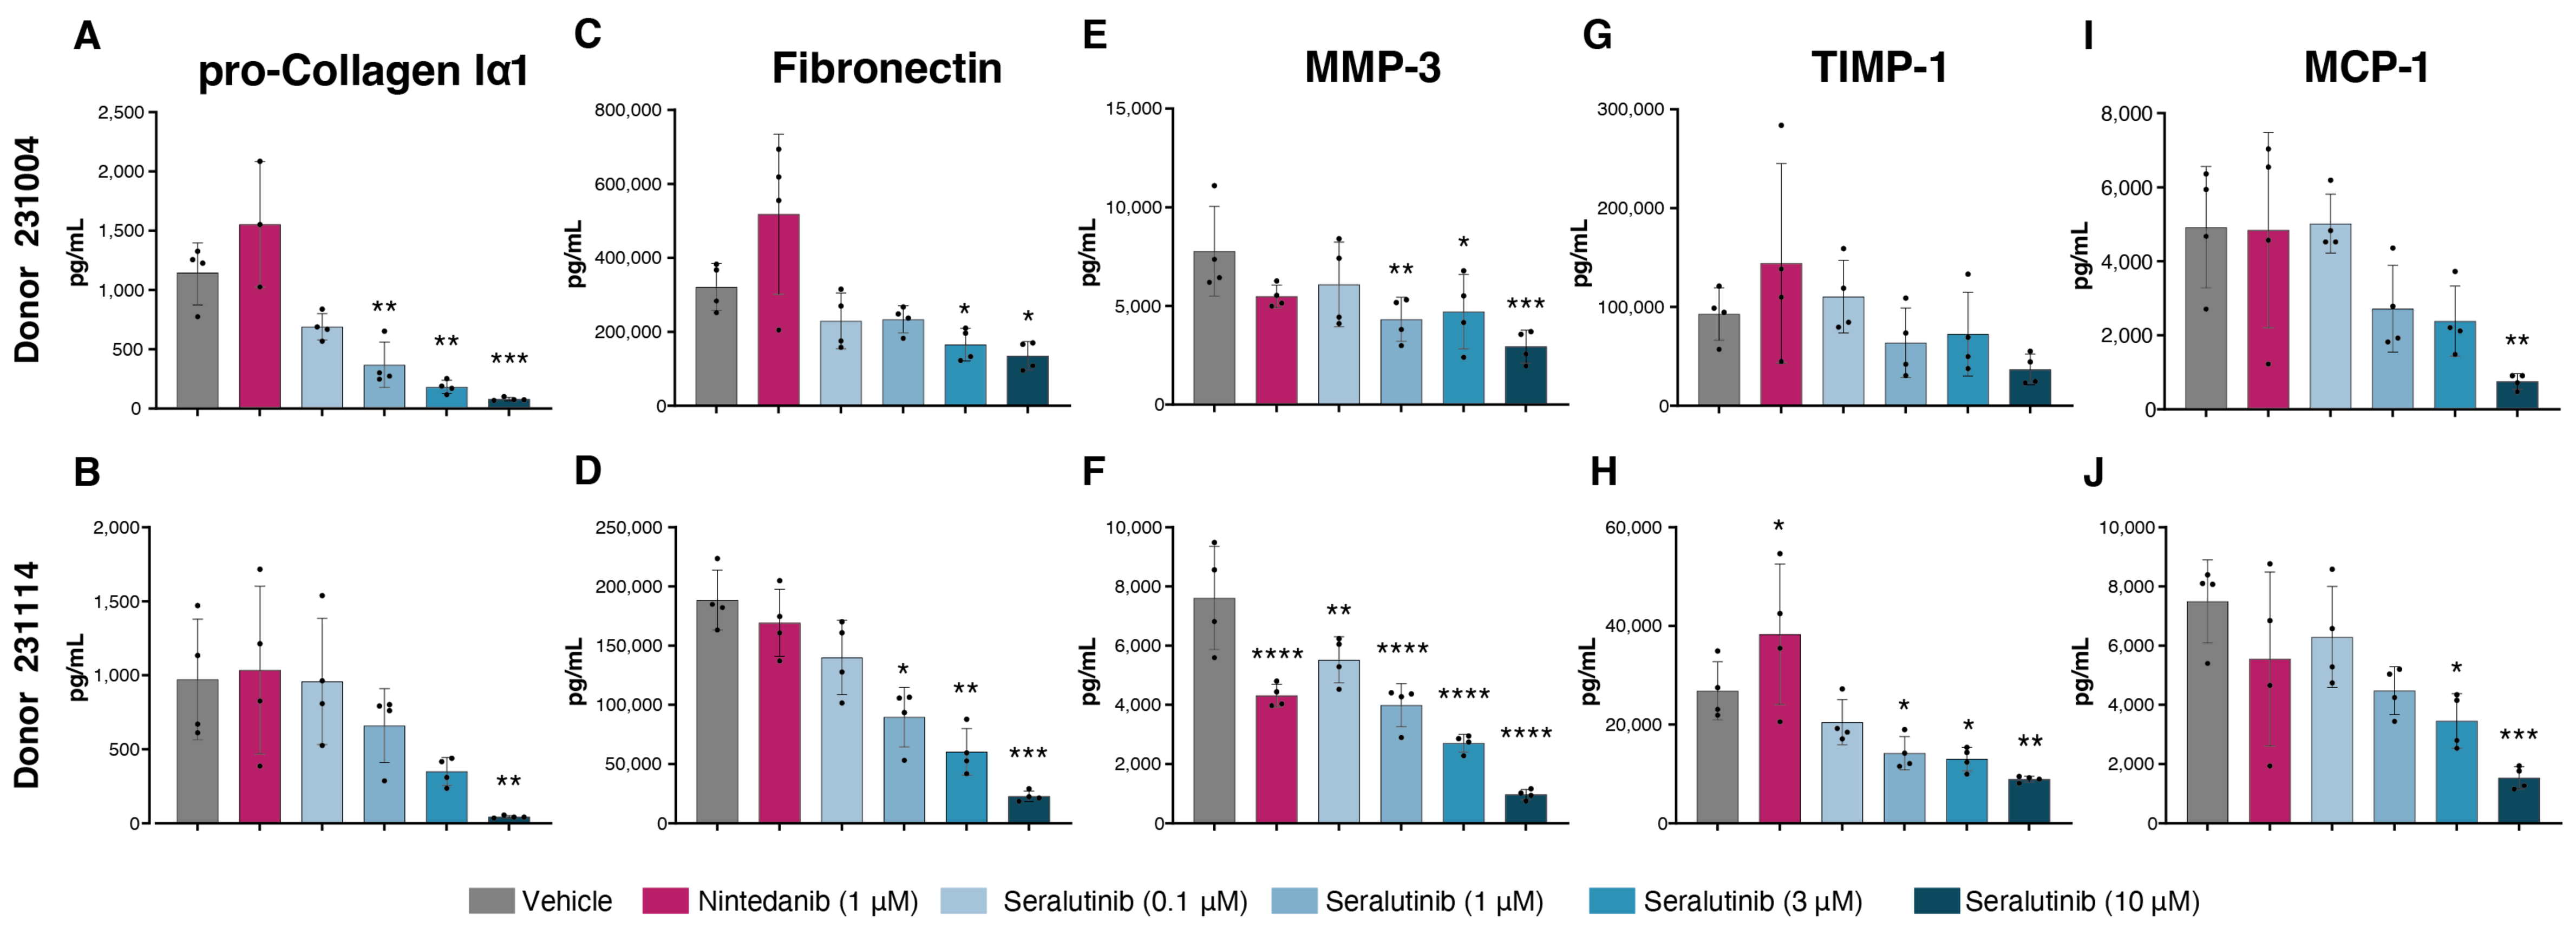
A pro-Collagen Iα1
C Fibronectin
E MMP-3
G TIMP-1
I MCP-1
B
D
F
H
J
Donor 231004
Donor 231114
pg/mL
Vehicle
Nintedanib (1 µM)
Seralutinib (0.1 µM)
Seralutinib (1 µM)
Seralutinib (3 µM)
Seralutinib (10 µM)

Graphs depict effect of vehicle control, seralutinib, and nintedanib on the release of pro-fibrotic markers pro-collagen Iα1 **(A,B)**, fibronectin **(C,D)**, MMP-3 **(E,F)**, TIMP-1 **(G,H)**, and MCP-1 **(I,J)** secreted by IPF-hPCLS at 96 h for both donors tested. Data are expressed as mean ± SEM (n=4). Statistical analysis was performed using one-way ANOVA with Fisher's LSD Test. *p<0.05, **p<0.01, ***p<0.001, and ****p<0.0001 versus vehicle.

Figure 5. Seralutinib did not induce cytotoxicity at clinically relevant concentrations in IPF-hPCLS

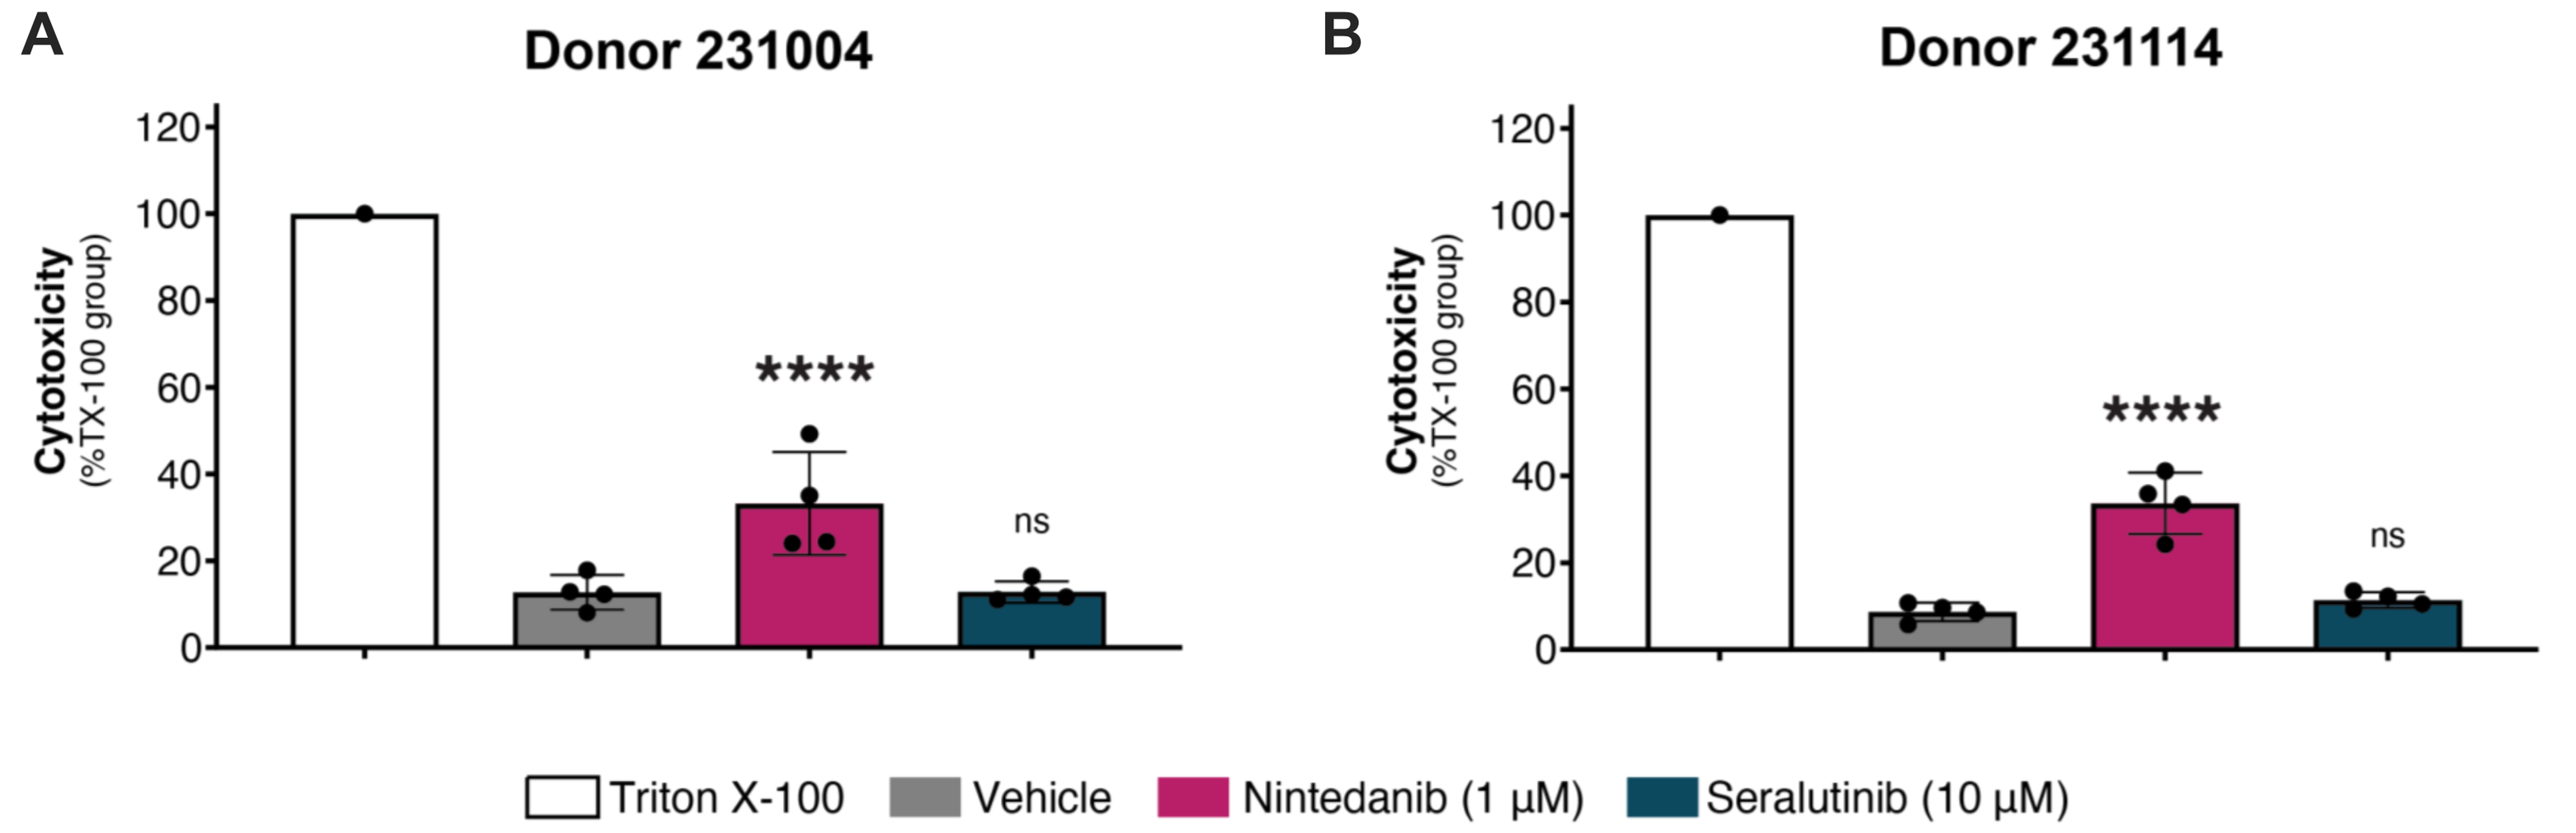
A Donor 231004
B Donor 231114
Cytotoxicity (%TX-100 group)

ns
Triton X-100
Vehicle
Nintedanib (1 µM)
Seralutinib (10 µM)

Change in adenylate kinase levels, a measurement of cytotoxicity, at 96 h for both donors **(A,B)**. Data are expressed as mean ± SEM (n=4). Triton X-100 was used as a positive control for cytotoxicity in this assay. Statistical analysis was performed using one-way ANOVA with Fisher's LSD Test. *p<0.05, **p<0.01, ***p<0.001, and ****p<0.0001 versus vehicle.

Figure 6. Seralutinib dose-dependently decreased release of fibrotic markers from IPF-HLFs

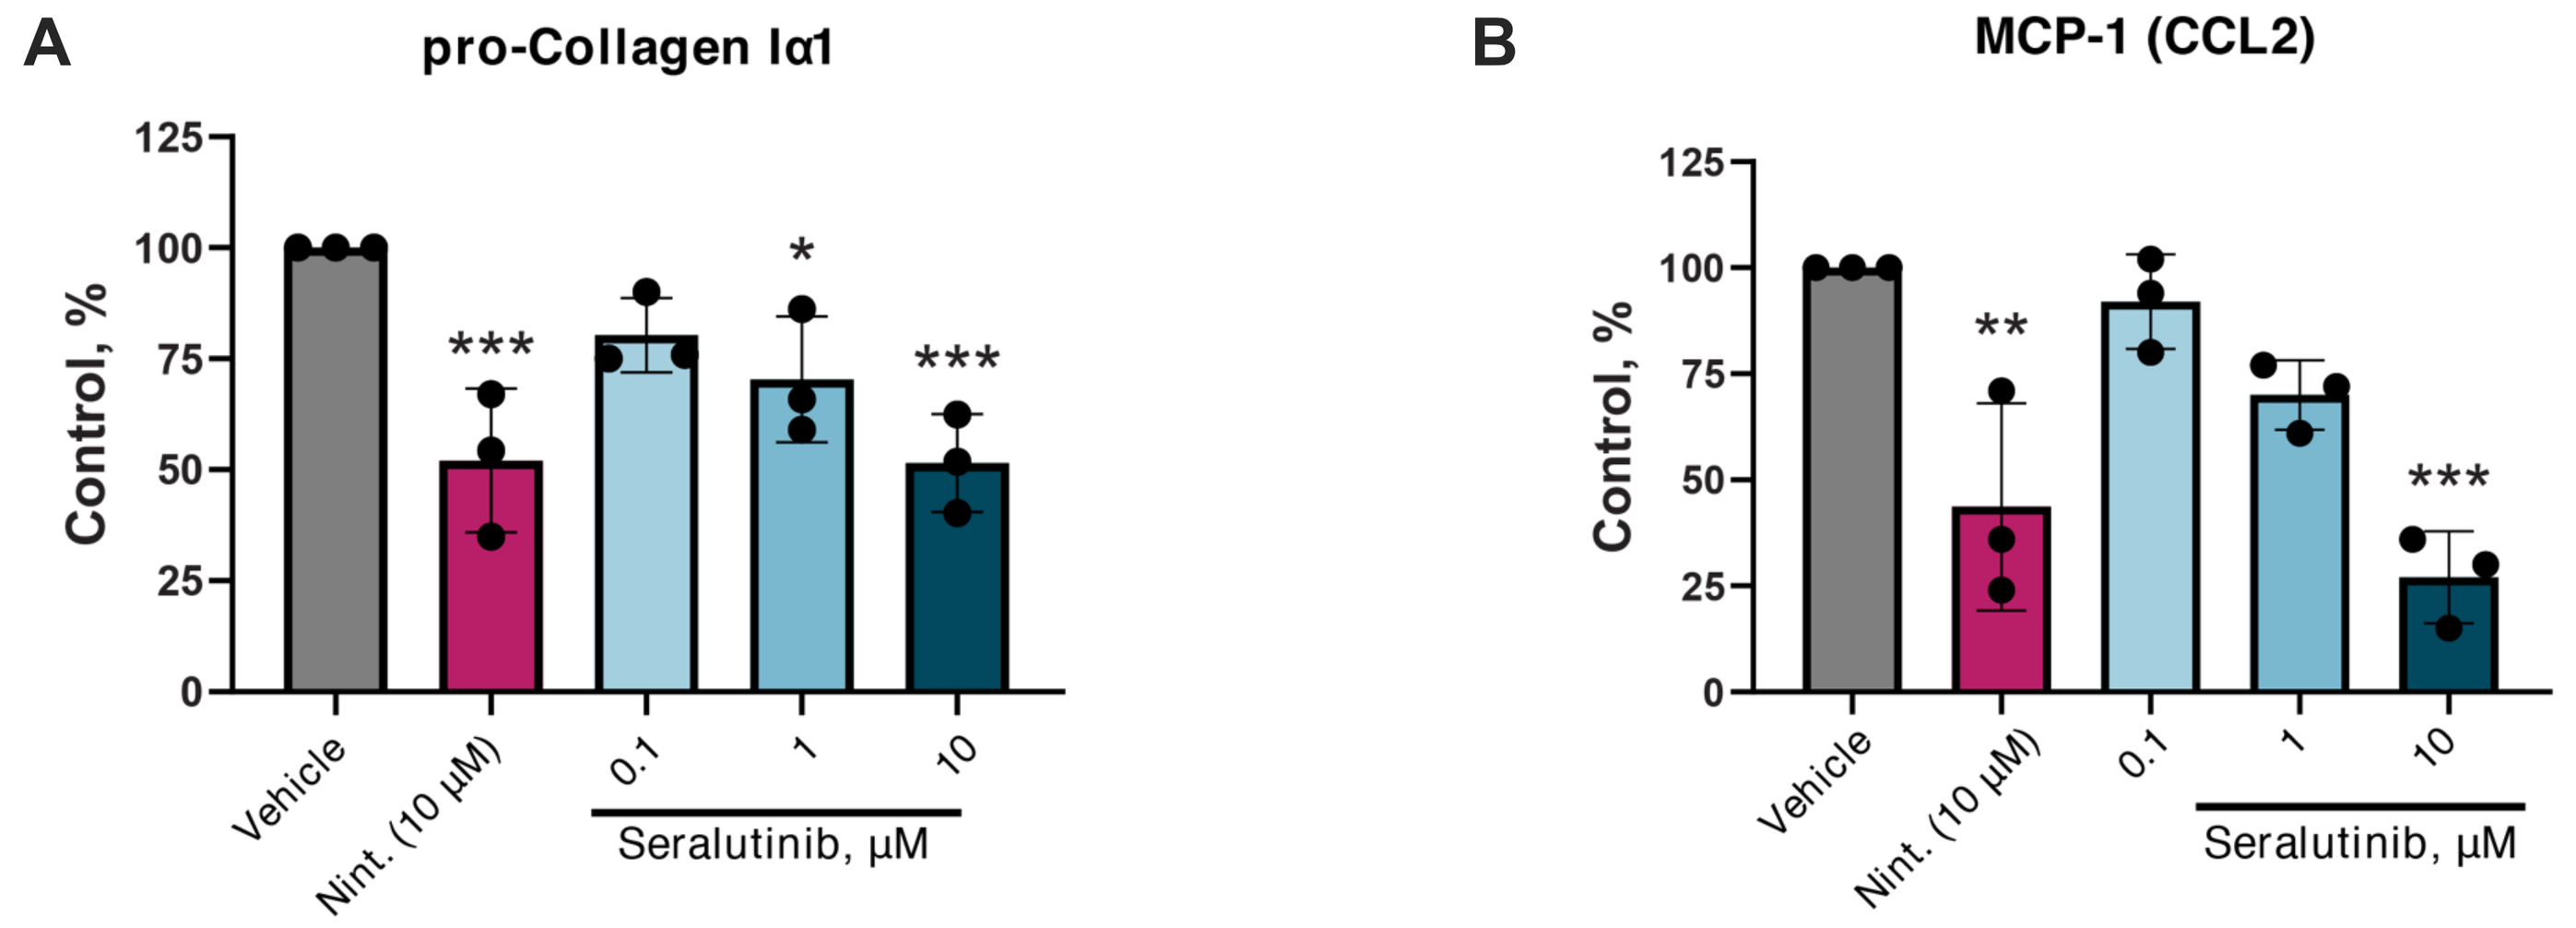
A pro-Collagen Iα1
B MCP-1 (CCL2)
Control, %
Vehicle
Nint. (10 µM)
0.1
1
10
Seralutinib, µM

Measured release of **(A)** pro-collagen Iα1 and **(B)** MCP-1 in supernatant of IPF-HLF cells averaged and normalized. Data are expressed as mean ± SEM (n=3). Statistical analysis was performed using one-way ANOVA followed by Dunnett's multiple comparisons test. *p<0.05, **p<0.01, ***p<0.001, and ****p<0.0001 versus vehicle at 72 h. Nint., Nintedanib.

Figure 7. Seralutinib inhibited TGF-β–induced α-SMA in IPF-HLFs

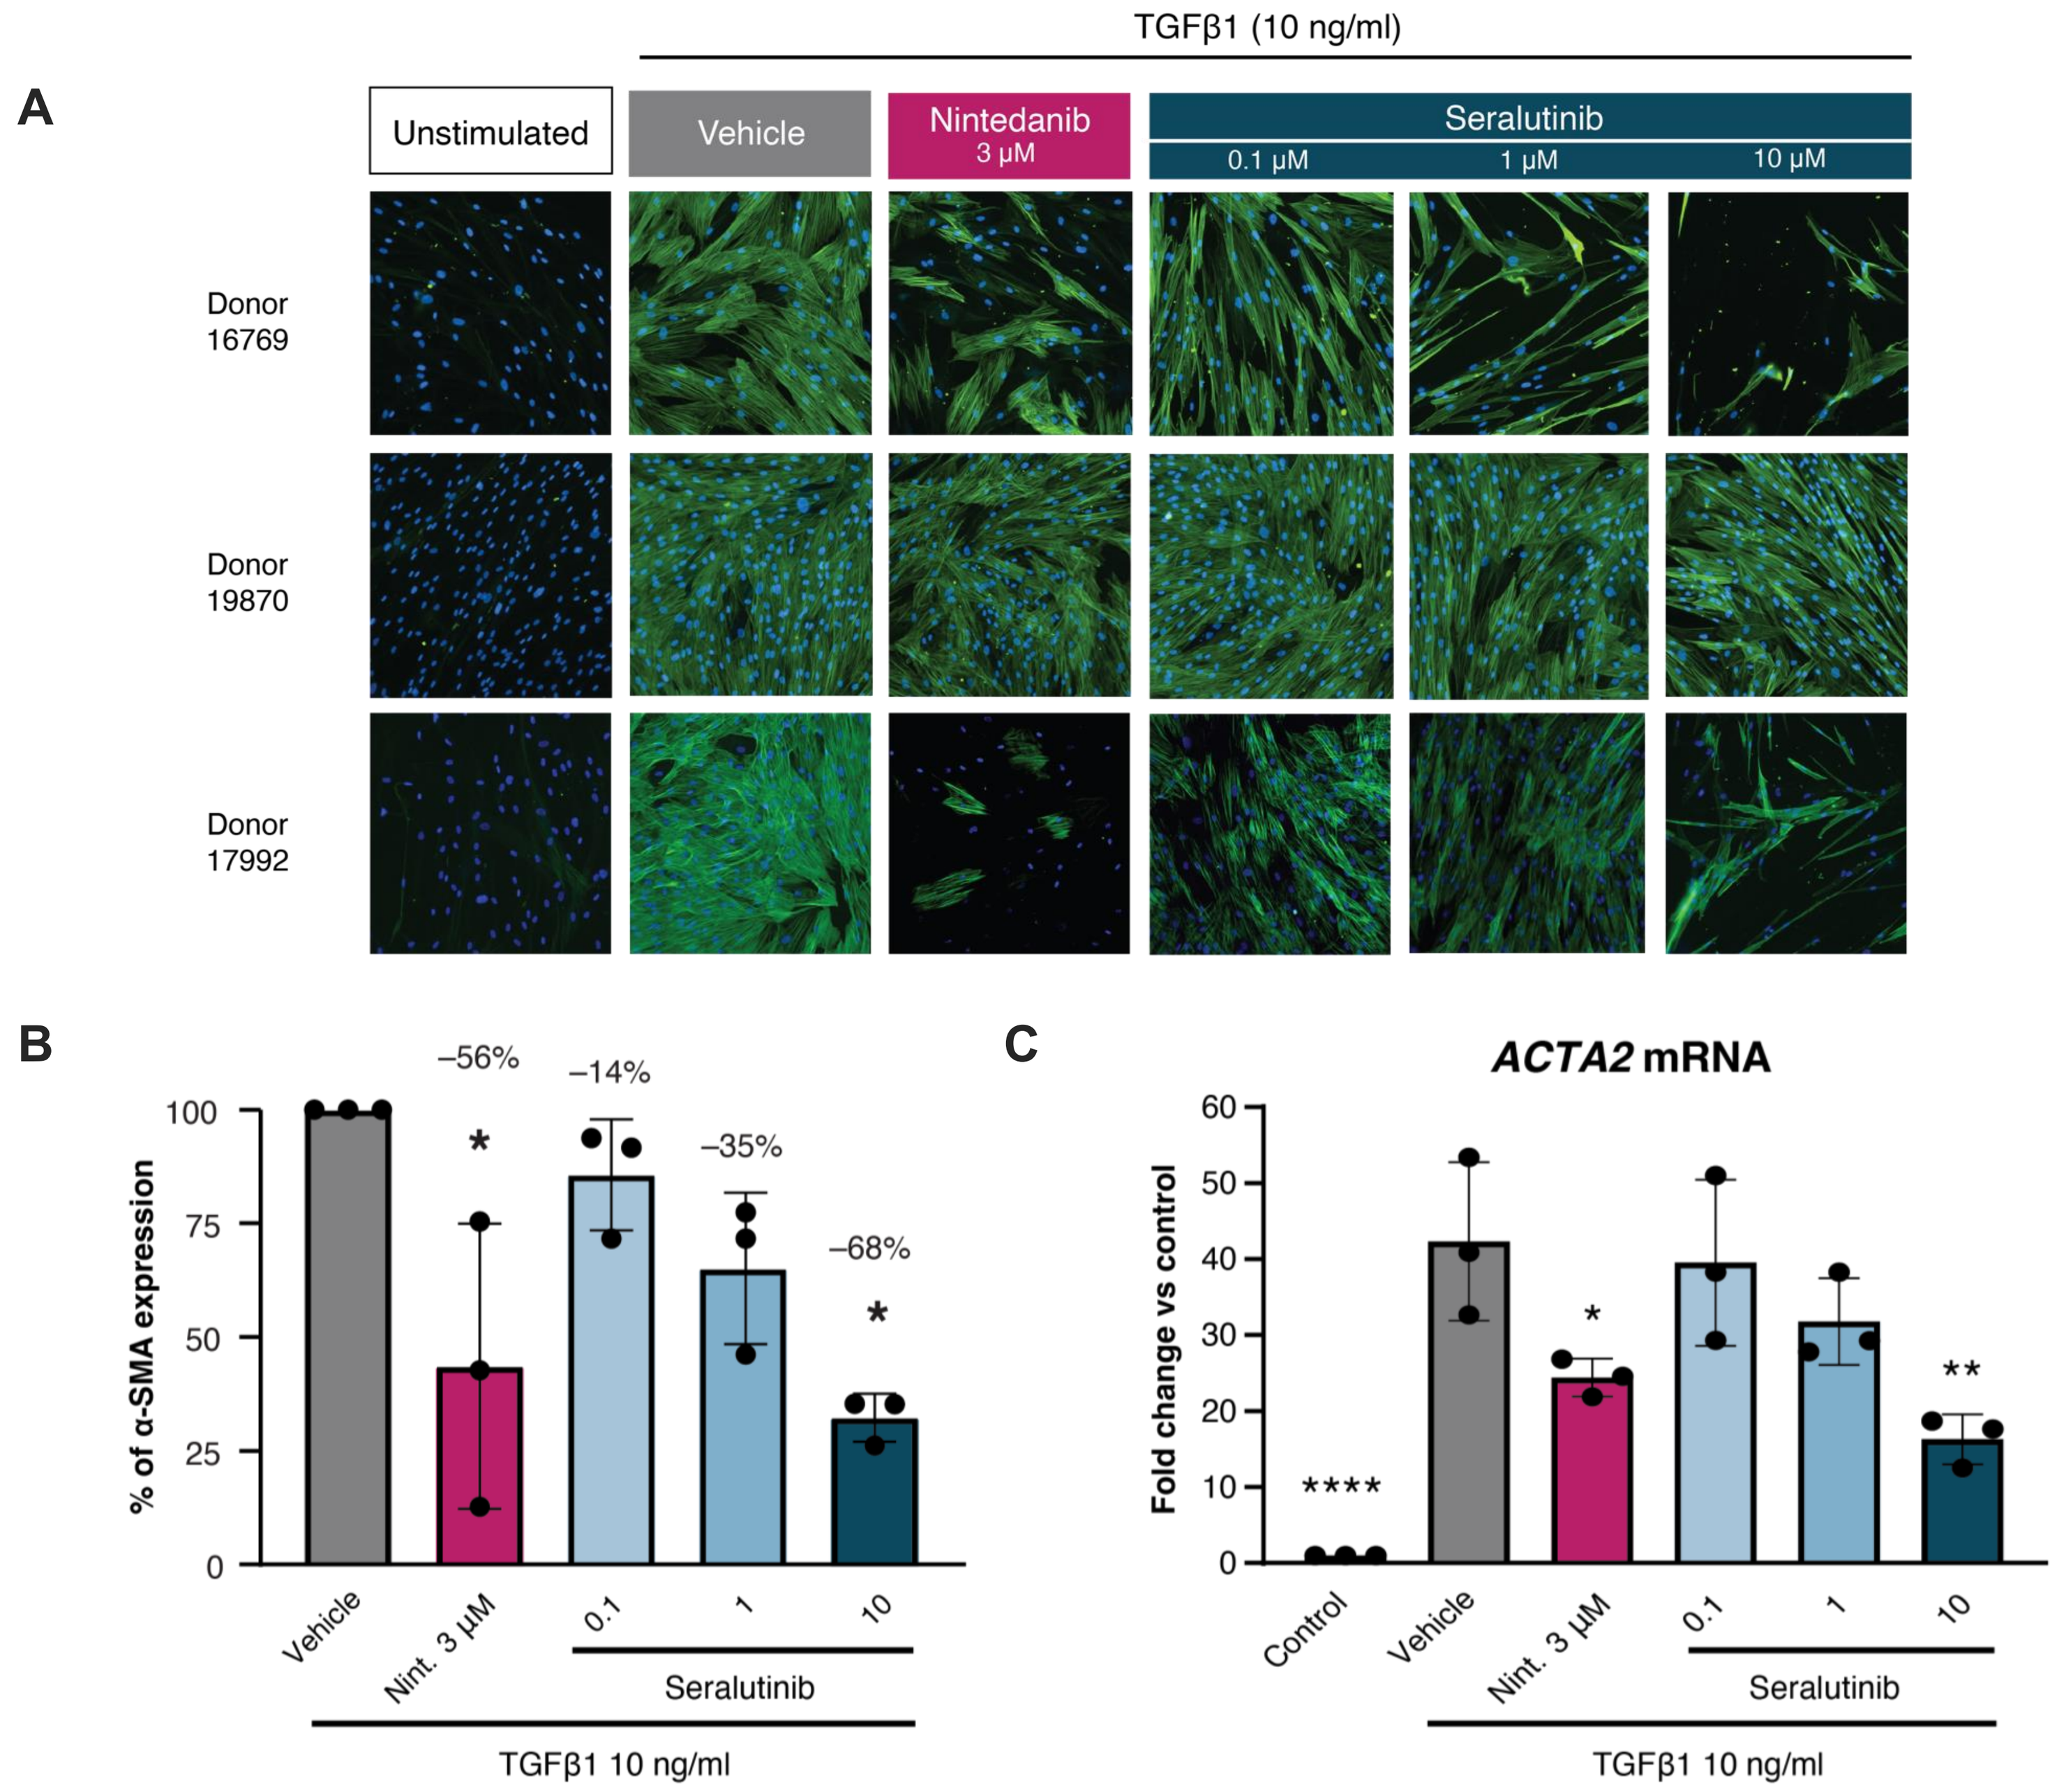
TGFβ1 (10 ng/ml)
A
Unstimulated
Vehicle
Nintedanib 3 µM
Seralutinib
0.1 µM
1 µM
10 µM
Donor 16769
Donor 19870
Donor 17992
B
% of α-SMA expression
–56%
–14%
–35%
–68%
Vehicle
Nint. 3 µM
Seralutinib
TGFβ1 10 ng/ml
C
ACTA2 mRNA
Fold change vs control
Control

(A) Immunofluorescence imaging and **(B)** quantification of α-SMA levels. Seralutinib reduced amount of TGF-β–induced α-SMA expressed at clinically relevant doses in IPF-HLF cells. **(C)** Change in gene expression levels of *ACTA2* (smooth muscle actin gene) in IPF-HLF cells. Gene upregulated due to TGF-β stimulation and dose-dependent inhibition by seralutinib.

Data are expressed as mean ± SEM (n=3 donors). Statistical analysis was performed using one-way ANOVA Dunnett's multiple comparisons test. *p<0.05, **p<0.01, ****p<0.0001 versus vehicle.

CONCLUSIONS

- Seralutinib dose-dependently decreased pro-collagen Iα1, fibronectin, MMP-3, TIMP-1, and MCP-1 in IPF-hPCLS, with no cytotoxicity observed
- In IPF-HLFs, seralutinib significantly inhibited the release of pro-collagen Iα1, MCP-1, and TGF-β–induced α-SMA expression in a dose-dependent manner
- These data support investigation of seralutinib as a potential inhaled treatment option for diseases characterized by underlying pulmonary vascular and interstitial fibrosis

References: **1** Dhont S, et al. *ERJ Open Res*. 2022;8(4):00272-2022. **2** Galkin A, et al. *Eur Respir J*. 2022;60(6):2102356. **3** Pullamsetti SS, et al. *Int J Mol Sci*. 2023;24(16):12653. **4** Ghofrani HA, et al. *Am J Respir Crit Care Med*. 2024;209:A7383. **5** Osterhout R, et al. *Eur Respir J*. 2024;64:OA1872. **6** Marimoutou M, et al. *Toxics*. 2024;12(9):637.

Disclosures: RS is an employee of, and owns stocks or stock options from, Gossamer Bio, Inc.

Research supported by: Chiesi Farmaceutici S.p.A. and Gossamer Bio, Inc.

The ERS is not responsible for and does not endorse the data and information presented on external sites.