1048

Advancing Careers and Leadership in Pulmonary Hypertension: Key Insights From PH Clinicians

Roxana Sulica[1], Jean Elwing[2], Lana Melendres-Groves[3], Karla Cruz Morel[4], Belinda Rivera-Lebron[5], Sonja Bartolome[6], K. Akaya Smith[7], Josanna Rodriguez-Lopez[8], Jill Luer[9], Natalie Sims[9], Elizabeth J. Kopras[2], Namita Sood[10]

[1]NYU Langone Health, New York, NY; [2]University of Cincinnati, Cincinnati, OH; [3]University of New Mexico, Albuquerque, NM; [4]Heart and Vascular Institute, Prisma Health, Columbia, SC; [5]University of Pittsburgh Medical Center, Pittsburgh, PA; [6]University of Texas Southwestern Medical Center, Dallas, TX; [7]University of Pennsylvania, Philadelphia, PA; [8]Massachusetts General Hospital, Boston, MA; [9]Gossamer Bio, Inc., San Diego, CA; [10]University of California Davis Medical Center, Sacramento, CA

Presented at
PHA 2026
INTERNATIONAL PH CONFERENCE AND SCIENTIFIC SESSIONS
Dallas, TX
June 11-14, 2026

BACKGROUND

- Pulmonary hypertension (PH) care depends on a highly specialized workforce to sustain excellence in clinical care, research, and advocacy
- Mentorship to prepare future PH leaders is critical to advancing the field of PH and maintaining career fulfillment and longevity
- Understanding professional experiences is essential to building a strong and enduring workforce of PH clinicians

OBJECTIVES

- Characterize the experience and contribution of providers in PH care, research, and leadership
- Identify challenges and potential gender impact on professional experience

METHODS

- A 44-question mixed-methods survey was administered electronically to PH physicians through the Pulmonary Hypertension Association (PHA) professional network listserv
- Participation was voluntary and names were not requested or recorded
- Survey domains: demographics and professional characteristics, leadership roles, clinical practice and institutional support, career satisfaction, work-life balance, and perceived barriers to advancement
- Quantitative items used multiple-choice and Likert-scale question formats. These data were analyzed using descriptive statistics and Chi-square, Fisher exact tests, and Cramér's V
- Open-ended questions captured qualitative responses describing personal experiences, perceived gaps in support, and suggestions for improving PH leadership. Responses were analyzed using conventional content analysis. Two investigators independently reviewed and coded responses to identify recurrent categories and representative quotations, with discrepancies resolved through discussion

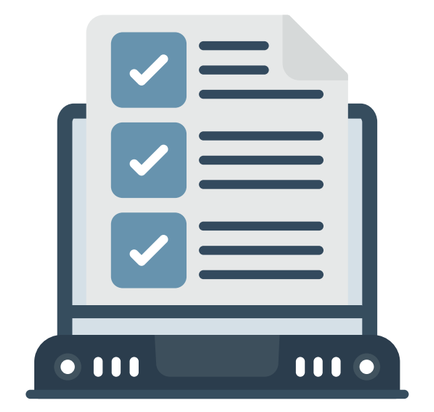

RESULTS

- 88 of 780 (11.3%) physicians in the PHA listserv completed the survey

Demographic and Professional Characteristics (Table 1)

- Respondent characteristics were similar between females and males
- Responders were 35-54 years of age (70%), worked in academic medical centers (85%), of which most were PHA-accredited, had ≥11 years of PH experience (66%), and cared for ≥26 PH patients/month (73%)

Table 1. Demographic and professional characteristics of survey respondents[a]

Characteristic	Females (n=37)	Males (n=49)	Total (N=88)
Age group, n (%)			
25–34 years	1 (2.7)	1 (2.0)	2 (2.3)
35–44 years	14 (37.8)	18 (36.7)	32 (36.4)
45–54 years	13 (35.1)	17 (34.7)	30 (34.1)
55–64 years	6 (16.2)	8 (16.3)	14 (15.9)
≥65 years	3 (8.1)	7 (14.3)	10 (11.4)
Years in PH practice,[b,c] n (%)			
<5	7 (19.4)	9 (18.8)	16 (18.4)
5–10	6 (16.7)	7 (14.6)	13 (14.9)
11–20	15 (41.7)	19 (39.6)	34 (39.1)
>20	8 (22.2)	16 (33.3)	24 (27.6)
Academic rank/role, n (%)			
Assistant Professor	9 (24.3)	11 (22.4)	20 (23.0)
Associate Professor	10 (27.0)	13 (26.5)	23 (26.4)
Professor	13 (35.1)	19 (38.8)	32 (36.8)
Community practice	5 (13.5)	6 (12.2)	11 (12.6)
Tenured	4 (10.8)	8 (16.3)	12 (13.6)
Practice setting,[d] n (%)			
Academic medical center	31 (83.8)	42 (85.7)	73 (84.9)
Private practice	4 (10.8)	4 (8.2)	8 (9.3)
Community hospital	4 (10.8)	4 (8.2)	8 (9.3)
Government hospital	1 (2.7)	1 (2.0)	2 (2.3)
PHA-accredited PH center	24 (66.7)	34 (70.8)	58 (66.7)
PH patients per month, n (%)			
1–10	3 (8.1)	3 (6.1)	6 (6.8)
11–25	8 (21.6)	10 (20.4)	18 (20.5)
26–50	13 (35.1)	19 (38.8)	32 (36.4)
>50	13 (35.1)	19 (38.8)	32 (36.4)

[a]Two respondents reported gender as "None of the above"; associated data were not included to avoid identification. [b]Denominators vary slightly due to missing responses. [c]Responses were collected by predefined ranges. [d]Multiple selections were allowed. PH, pulmonary hypertension; PHA, Pulmonary Hypertension Association.

Quantitative Analysis

- No statistically significant gender differences were observed in perceptions of leadership roles held, tenure status, research involvement, or authorship (**Figure 1**)
- However, perceptions of gender-related barriers to career success were reported by 51.4% of female physicians vs 2% of males (p<0.001)
 - No significant association was observed between gender and tenure status; the effect size was small to moderate (Cramér's V=0.22)
 - No significant difference between gender and leadership roles was observed, but no female physician reported identification as a division chief or department chair
- Most respondents (86.9%) conducted PH research; however, few (28.6%) have applied for federal funding

Figure 1. Reported experiences and contributions to PH care

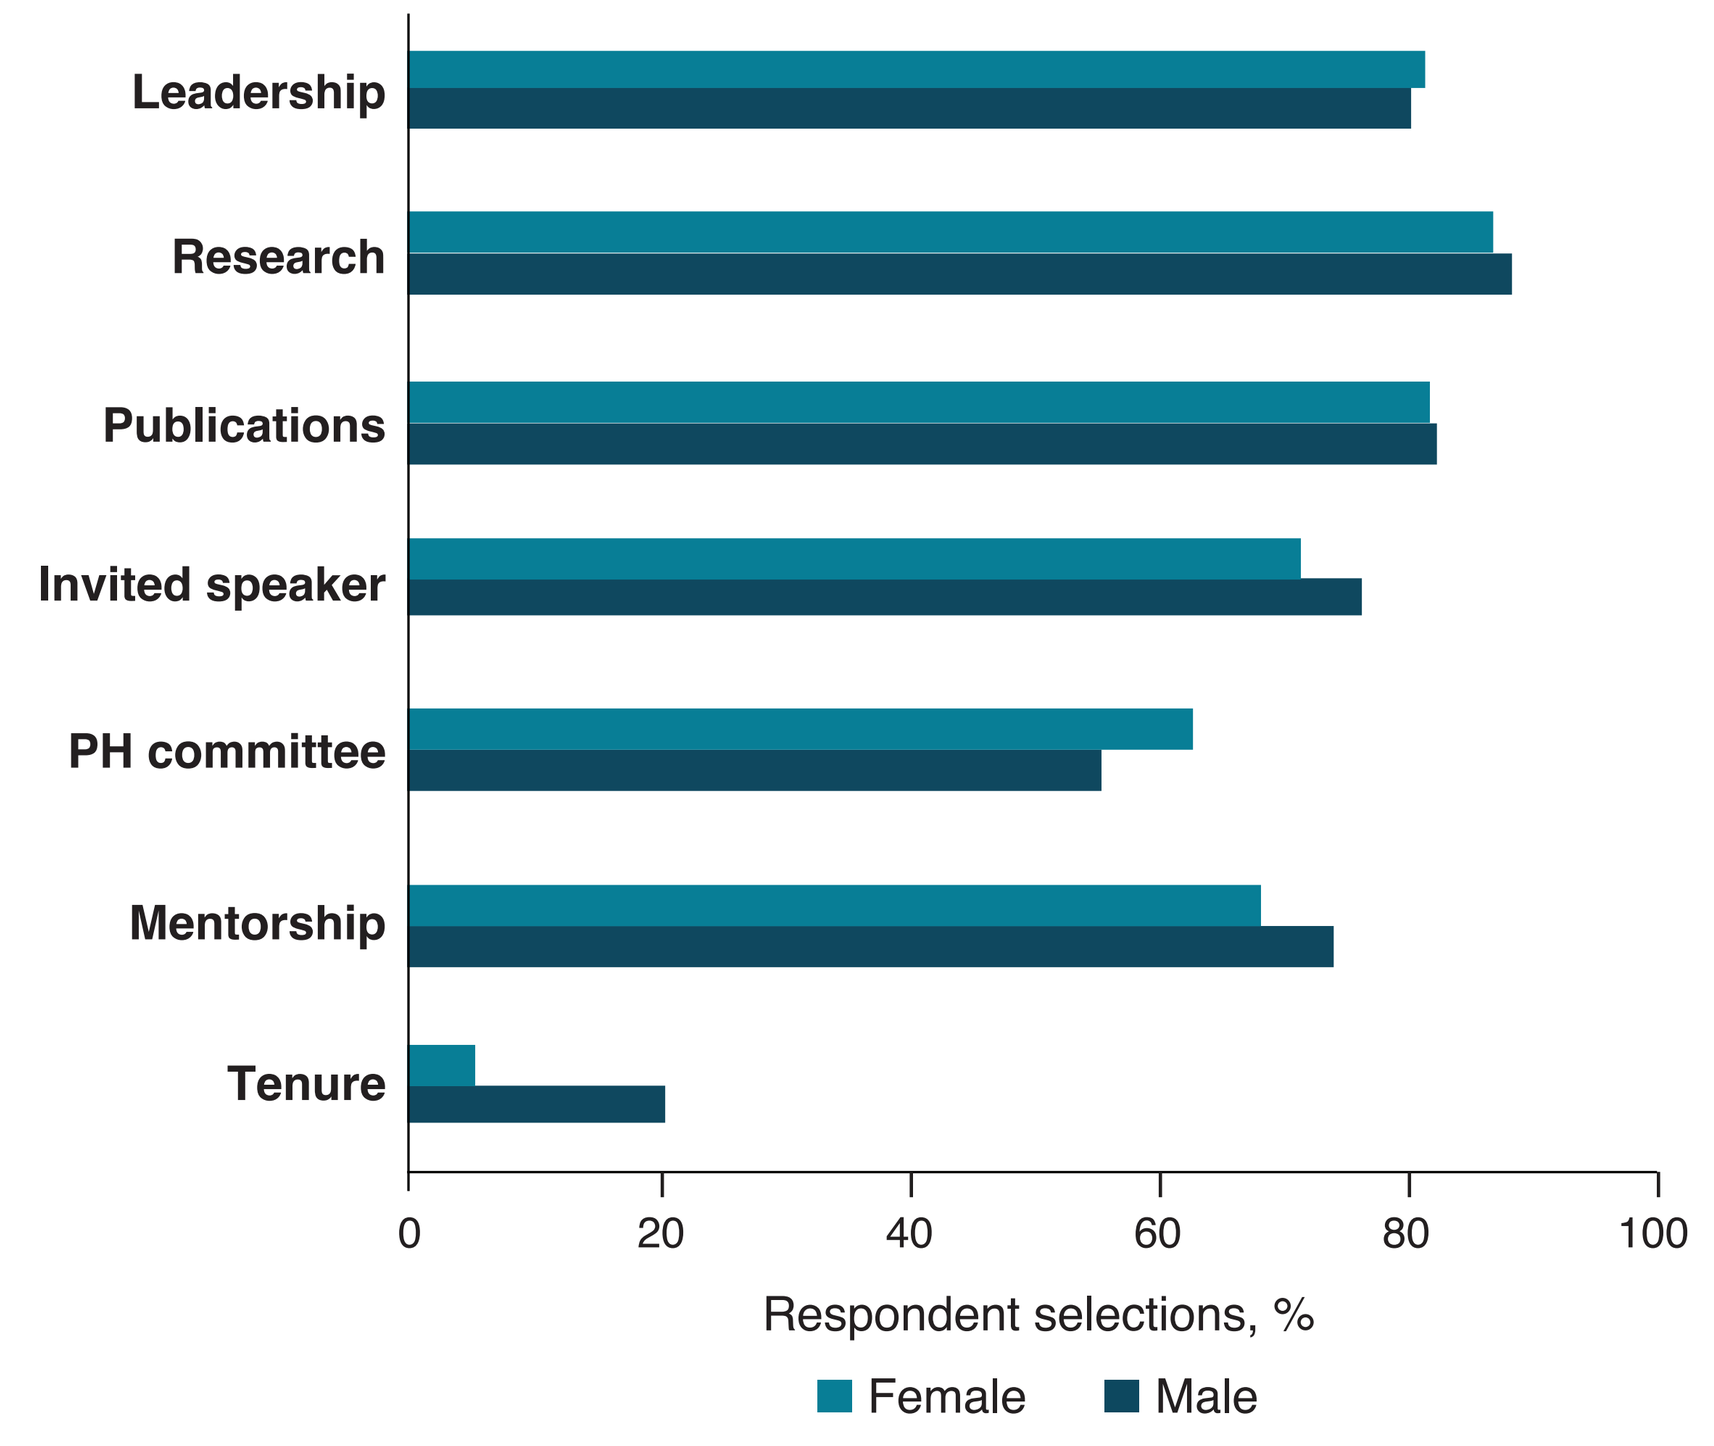
Leadership
Research
Publications
Invited speaker
PH committee
Mentorship
Tenure
0
20
40
60
80
100
Respondent selections, %
Female
Male

Two respondents reported gender as "None of the above"; data for these respondents were not included to avoid identification. PH, pulmonary hypertension.

- Mentorship and career guidance was most frequently selected as an important driver of career success, followed by institutional advocacy and protected time for academic/leadership work (**Figure 2**). No significant differences were observed between genders in driver selection frequency

Figure 2. Reported important drivers of career success

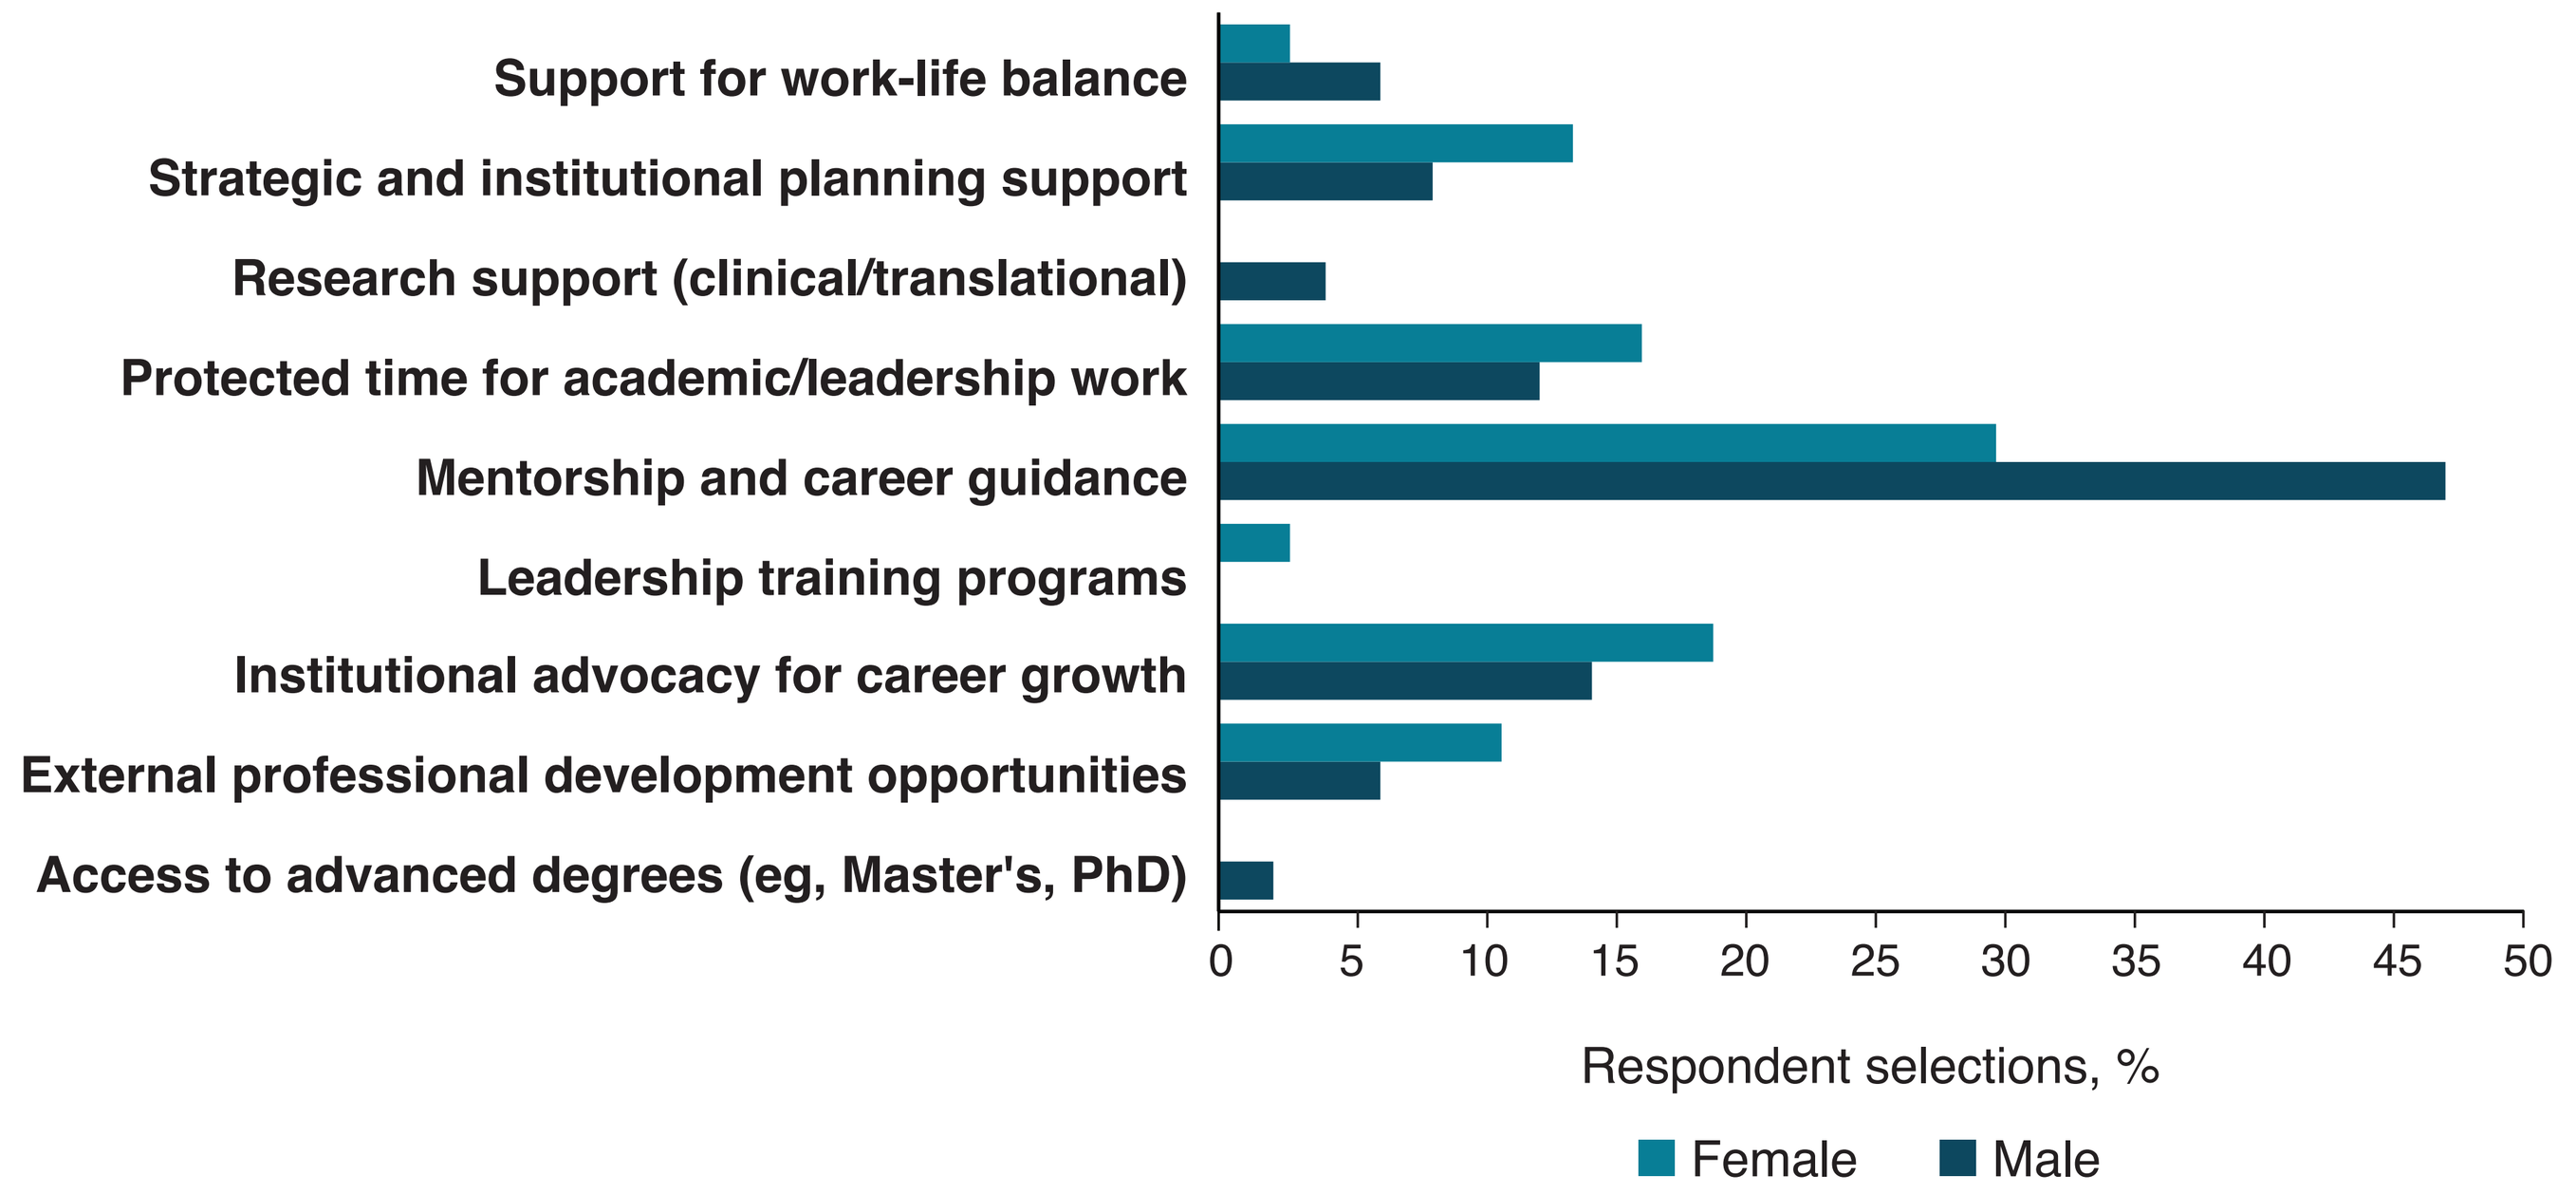
Support for work-life balance
Strategic and institutional planning support
Research support (clinical/translational)
Protected time for academic/leadership work
Mentorship and career guidance
Leadership training programs
Institutional advocacy for career growth
External professional development opportunities
Access to advanced degrees (eg, Master's, PhD)
0
5
10
15
20
25
30
35
40
45
50
Respondent selections, %
Female
Male

Respondents were asked to select one category. Two respondents reported gender as "None of the above"; data for these respondents were not included to avoid identification.

- 50% or more of respondents chose five leadership skills or resources as key for physician success (**Figure 3**)
- Although no significant differences were observed between genders in selection frequency:
 - Males most frequently selected mentorship and trainee support; females most frequently selected strategic and program planning
 - Females selected research development more often than males (55% vs 30%, respectively)
 - Males selected patient and community advocacy as a key leadership skill more frequently than females (28% vs 8%, respectively)
- No female physicians chose health policy/system-based thinking as a leadership skill/resource needed for leadership success

Figure 3. Reported top leadership skills/resources for PH physician success

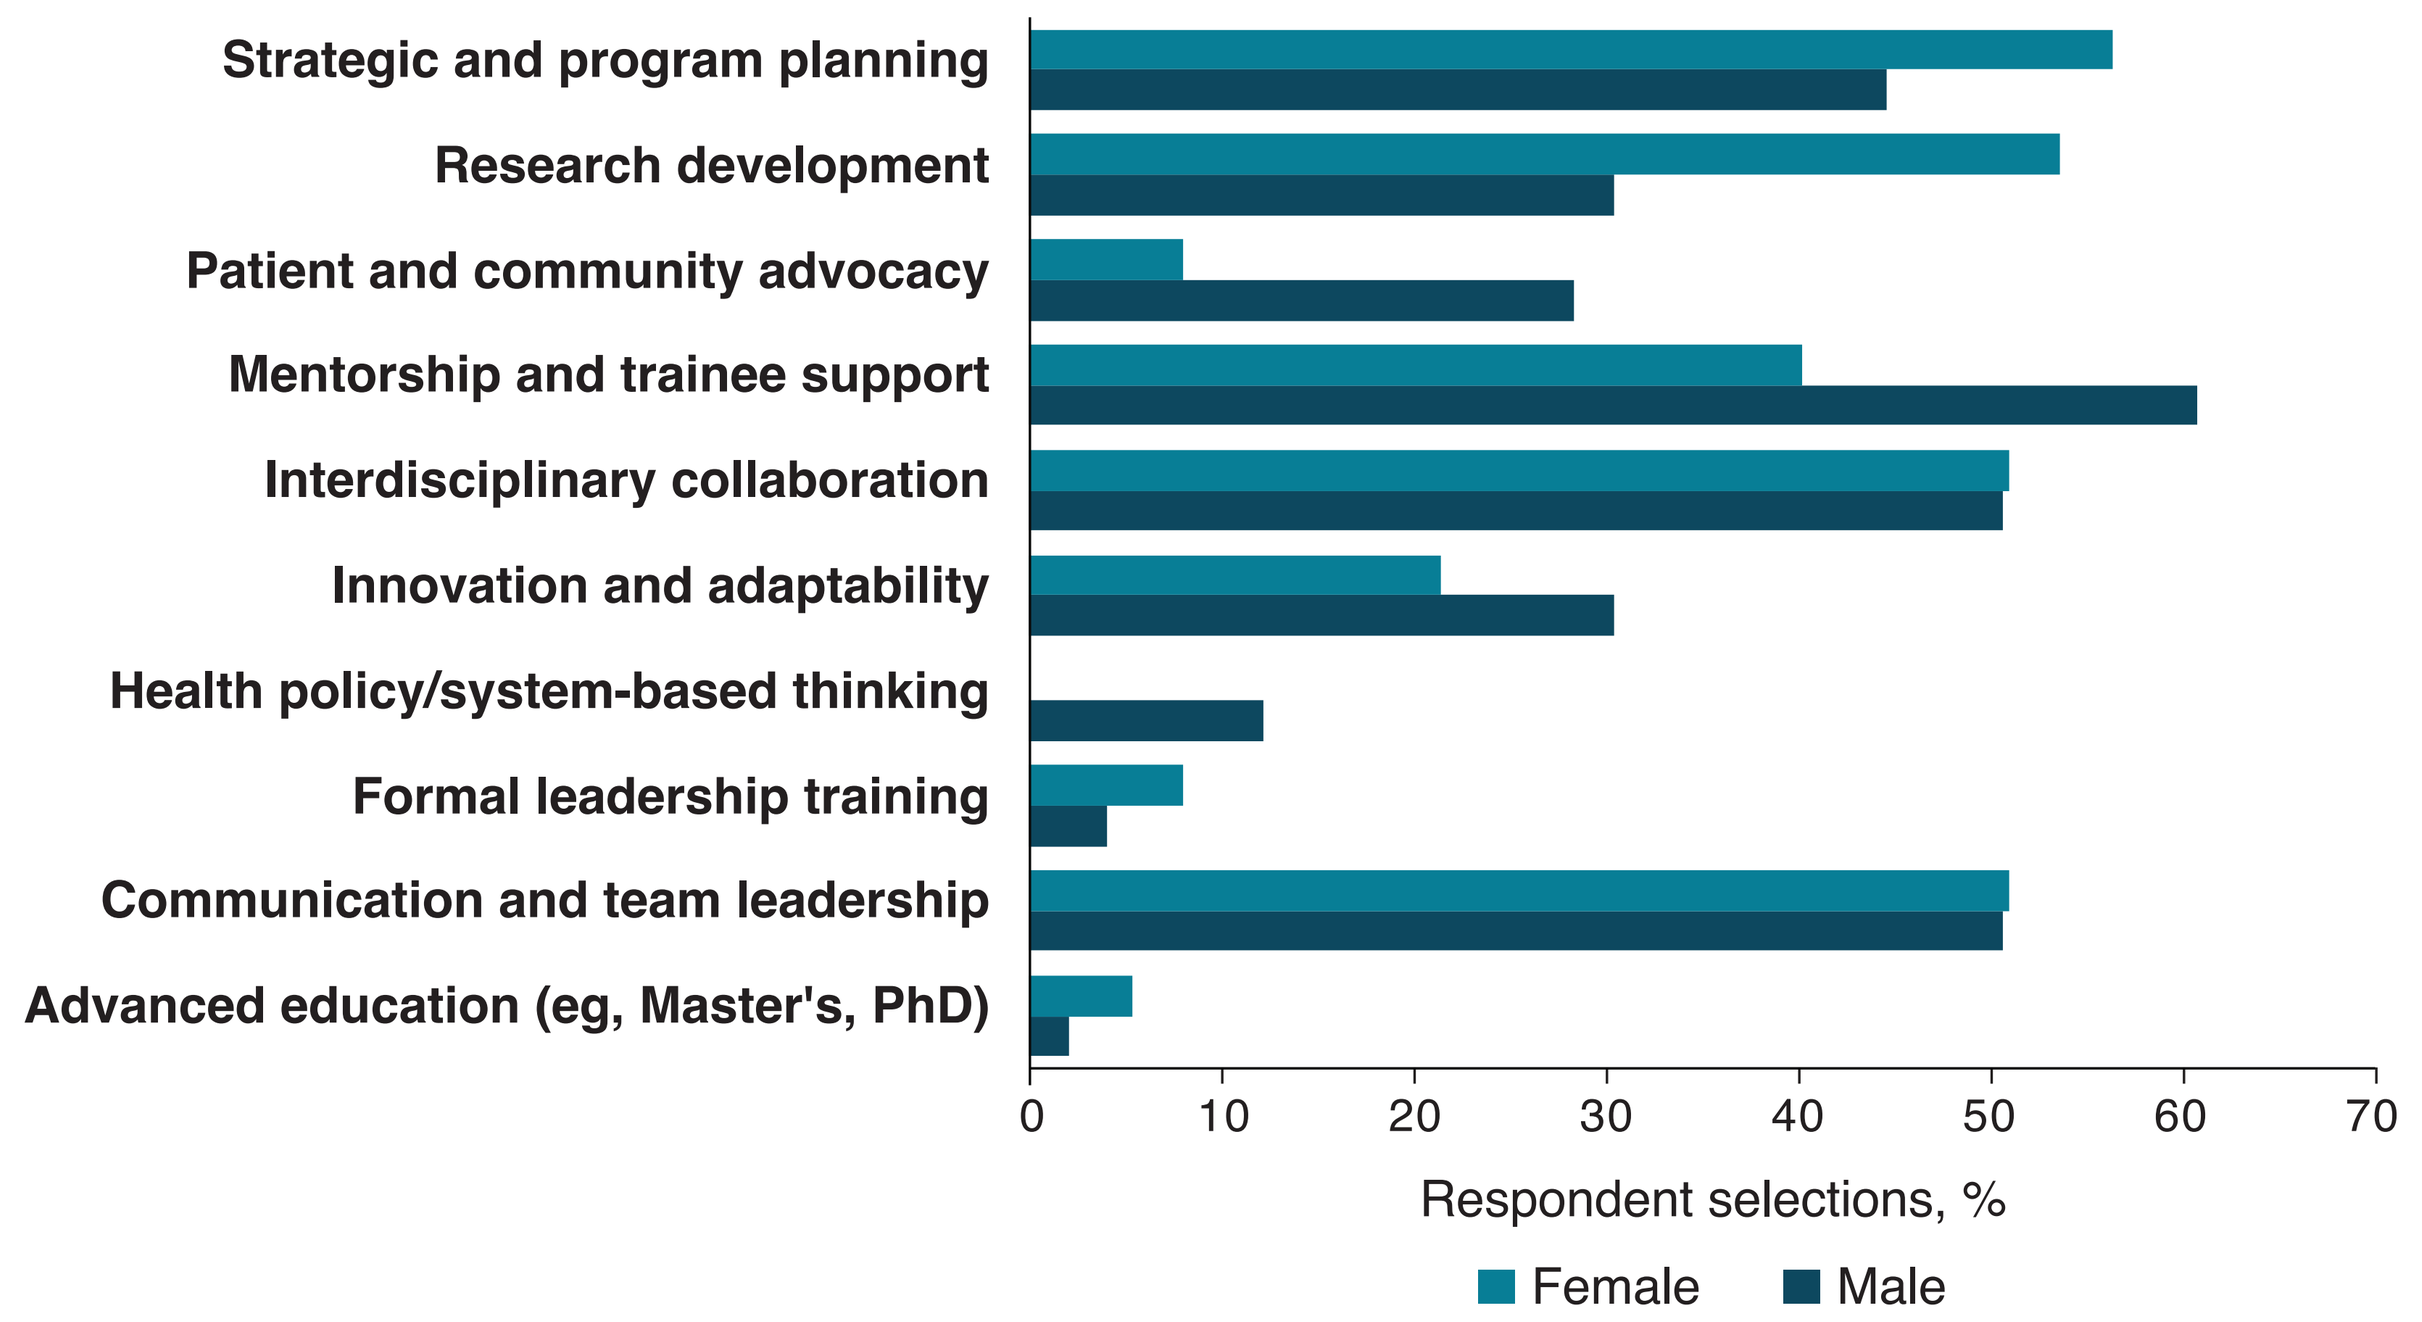
Strategic and program planning
Research development
Patient and community advocacy
Mentorship and trainee support
Interdisciplinary collaboration
Innovation and adaptability
Health policy/system-based thinking
Formal leadership training
Communication and team leadership
Advanced education (eg, Master's, PhD)
0
10
20
30
40
50
60
70
Respondent selections, %
Female
Male

Respondents could select up to 3 leadership skills or resources from the list provided. Two respondents reported gender as "None of the above"; data for these respondents were not included to avoid identification.

- Established mentorship programs, formal leadership training, and flexible work schedules were considered important opportunities for facilitating career advancement by both male and female PH physicians
- Overall, most respondents indicated being happy and content or generally satisfied working in PH, with nearly 60% describing their work as fulfilling
 - However, a trend towards gender differences was reported in happiness/contentment: females, 70.3%; males, 87.7%; Cramér's V=0.30
- A substantial proportion of respondents reported challenges with work-life balance: females, 75.0%; males, 58.3%; p=0.2282

Thematic Content Analysis

- Although physicians are deeply fulfilled by their PH work, demands and expectations are high in the context of workforce scarcity and geographic isolation, with potential for high burnout
- PH clinicians experience inadequate institutional support due to poor understanding by administrators of the complexities and demands of their field. Career success depends heavily on investment in support staff and protected time for research and other leadership opportunities
- Administrative burden is particularly challenging given the increased demands of PH care. Clinicians feel pulled away from the very activities defining career success in PH – patient care, mentoring, research, program development, and advocacy
- Although survey results suggest similar access to roles for female and male physicians, feedback from females reveals that recognition, sponsorship, flexibility, and support may not be experienced equally

CONCLUSIONS

- PH care is intense and immersive. While most physicians surveyed are happy with their career choice, institutional support to enhance career growth and work-life balance is needed
- Mentorship is key for the development of future PH specialists and advancement of PH care to ensure improved outcomes for patients with PH
- Specific challenges identified suggest potential gender disparities in tenure status and top leadership roles, and limited time for grant development (for both genders), which may warrant further investigation
- These survey results provide a foundation for targeted leadership development, mentorship, and institutional initiatives aimed at strengthening and sustaining the PH workforce

Acknowledgements: We sincerely thank survey participants for sharing their valuable perspectives and time. We also thank the Pulmonary Hypertension Association for its support of this endeavor.

Research supported by: Gossamer Bio, Inc.