1038

Examining Undertreatment and Delays in Therapy for Pulmonary Arterial Hypertension: A Real-world Patient Journey Study From the US

Charles D. Burger[1], Kelly M. Chin[2], Jean M. Elwing[3], H. James Ford[4], John W. McConnell[5], John J. Ryan[6], Marc A. Simon[7], Carolynn Chang[8], Matthew Pillsbury[9], Charlotte Ward[9], Gina Nelson[8], Ioana R. Preston[10]

[1]Mayo Clinic, Jacksonville, FL, USA; [2]UT Southwestern Medical Center, Dallas, TX, USA; [3]College of Medicine, University of Cincinnati, Cincinnati, OH, USA; [4]University of North Carolina at Chapel Hill School of Medicine, Chapel Hill, NC, USA; [5]Norton Healthcare, Louisville, KY, USA; [6]University of Utah Health, Salt Lake City, UT, USA; [7]UCSF Medical Center, San Francisco, CA, USA; [8]Gossamer Bio, Inc., San Diego, CA, USA; [9]ZS, Princeton, NJ, USA; [10]Lahey Hospital & Medical Center, Burlington, MA, USA

Presented at PHA 2026 International PH Conference and Scientific Sessions
Dallas, TX
June 11-14, 2026

BACKGROUND

- Pulmonary arterial hypertension (PAH) remains a progressive and life-threatening disease despite the availability of evidence-based therapies[1]
- Treatment recommendations[1] and guidelines[2] emphasize early intensification with combination therapy across risk groups, underscoring that initial combination regimens are associated with improved outcomes versus monotherapy[1,2]
- Nevertheless, real world-evidence suggest that up to 80% of patients are initiated on monotherapy, with many patients remaining on monotherapy for prolonged periods after diagnosis[3,4]
- In addition, a substantial number of patients do not progress to treatment regimens targeting ≥3 disease pathways,[3] despite evidence supporting the benefits of comprehensive, multi-pathway therapy in appropriate patients[1,2]
- Understanding the causes and consequences of delayed treatment escalation is critical to improving outcomes
- **This mixed methods study integrated a retrospective analysis of closed claims data with qualitative interviews to explore real-world treatment patterns and stakeholder perspectives in PAH**

METHODS

Claims Analysis

- The longitudinal claims data analysis leveraged Komodo Health data and examined two cohorts: Cohort 1 (N=3,908) included PAH patients who initiated treatment between March 2020 and March 2025, with closed prescription claims enrollment from ≥1 year before treatment initiation to March 2025; Cohort 2 (N=6,600) included PAH patients who had PAH treatment between January 2023 and December 2024, with closed prescription claims enrollment from January 2023 to December 2025

INCLUSION CRITERIA

- ≥18 years of age
- ≥2 PAH diagnosis claims
- ≥1 prescription claim for advanced PAH treatment
- On PAH treatment ≥90 days

EXCLUSION CRITERIA

- On PDE5i or inhaled treprostinil not prescribed by a relevant specialty[a]
- On PDE5i or inhaled treprostinil regimens for ILD or LHD[b]
- On PDE5i monotherapy with a claim for erectile dysfunction

[a]Cardiologists, pulmonologists, rheumatologists, or NP/PA/PCP; [b]Excluded if the count of LHD diagnosis claims ≥60% of the count of PAH diagnosis claims in the 1-year look-forward period—defined as the 1-year timeframe during which patients are confirmed to have closed prescription claims enrollment prior to PAH initiation—and the count of ILD diagnosis claims was ≥2 from 120 days pre- to 60 days post-prescription. ILD, interstitial lung disease; LHD, left heart disease; NP, nurse practitioner; PA, physician's assistant; PAH, pulmonary arterial hypertension; PCP, primary care provider; PDE5i, phosphodiesterase type 5 inhibitor.

- Outcomes of interest included timing and frequency of treatment escalation (defined as addition of one or more agents to existing therapy, and excluding changes resulting in the patient receiving fewer agents than at the start of therapy), duration of monotherapy, and influence of patient characteristics on treatment patterns, including use of Chi-square tests to determine associations between comorbidities and treatment regimen

Qualitative Interviews

- Qualitative interviews were conducted to explore treatment goals, drivers of treatment decisions, perceptions of disease control, and barriers to therapy intensification
- Interview sample (N=50):

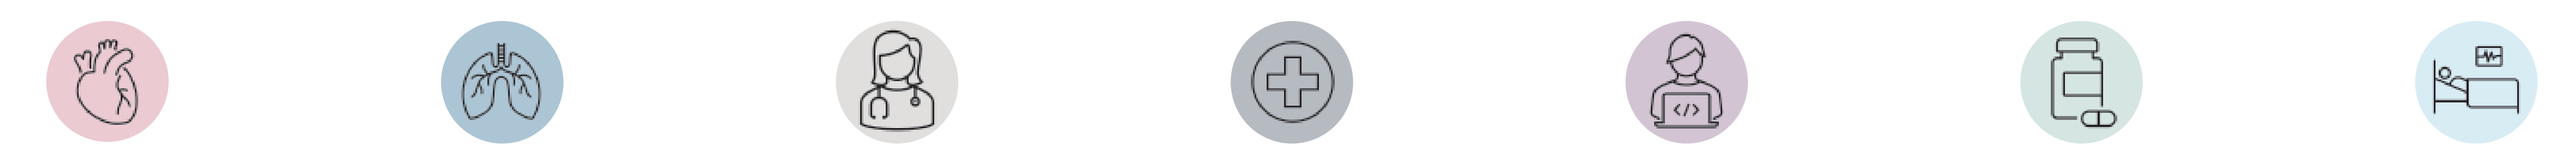

Cardiologist (n=10) | Pulmonologist (n=10) | Nurse practitioner/ Physician's assistant (n=7) | Registered nurse (n=4) | Office manager (n=2) | Specialty pharmacist (n=2) | Patient (n=15)

RESULTS

- In the claims analysis cohort with 3,908 patients (Cohort 1), 70% were female, 51% were ≥65 years of age at initial PAH treatment, 52% were covered by Medicare or Medicare Advantage, and 66% had ≥3 comorbidities
- Among the 1,800 patients in Cohort 1 who had complete information about the specialty of the diagnosing HCP, 54% of patients received their PAH diagnosis from a pulmonologist or cardiologist, most often in a community setting
- Most patients received monotherapy as their initial therapy (Tx) regimen (67%), 29% were treated with dual therapy, 4% with triple therapy, and <1% with quadruple therapy **(Figure 1)**
- Patients progressing to a second Tx regimen were on mono (41%), dual (45%), triple (13%), and quadruple (<1%) therapy

Initial Tx regimen was defined as the first advanced PAH Tx initiated after diagnosis, including all Tx added within 60 days. **Second Tx regimen** included any changes—escalations, de-escalations, switches within same MOA—occurring within a 60-day window after the initial Tx regimen. **Third Tx regimen** was defined as the next distinct regimen following that 60-day window.

Figure 1. Treatment Patterns by Therapy (Tx) Regimen (Cohort 1)

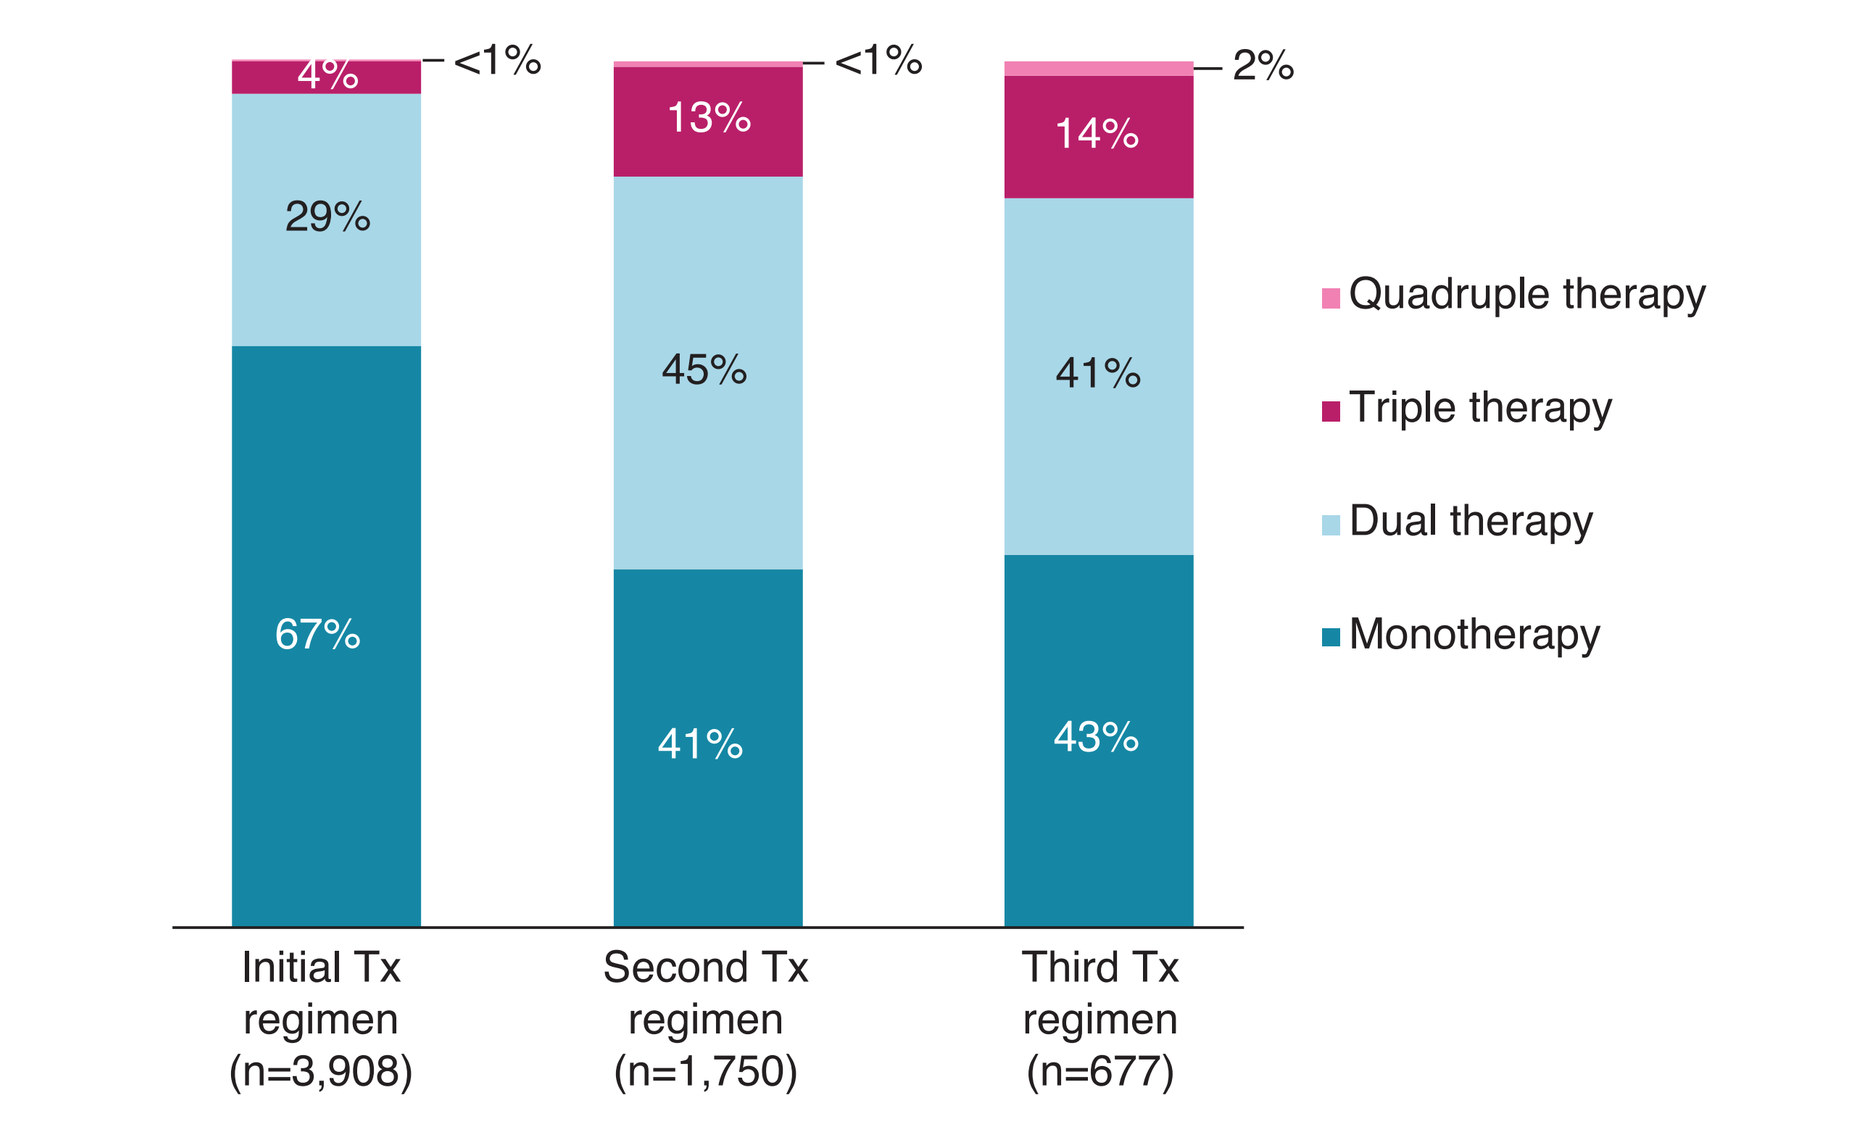
4%
<1%
29%
67%
13%
<1%
45%
41%
14%
2%
41%
43%
Quadruple therapy
Triple therapy
Dual therapy
Monotherapy
Initial Tx regimen (n=3,908)
Second Tx regimen (n=1,750)
Third Tx regimen (n=677)

3.9k patients; period March 2020—March 2025.

RESULTS (continued)

- Most patients started on phosphodiesterase type 5 inhibitor (PDE5i) monotherapy, and switched molecules or escalated to combination regimens in later therapy regimens as the disease progressed **(Figure 2)**

Figure 2. Treatment Progression: Patient Transitions Between Therapy Regimens (Cohort 1)

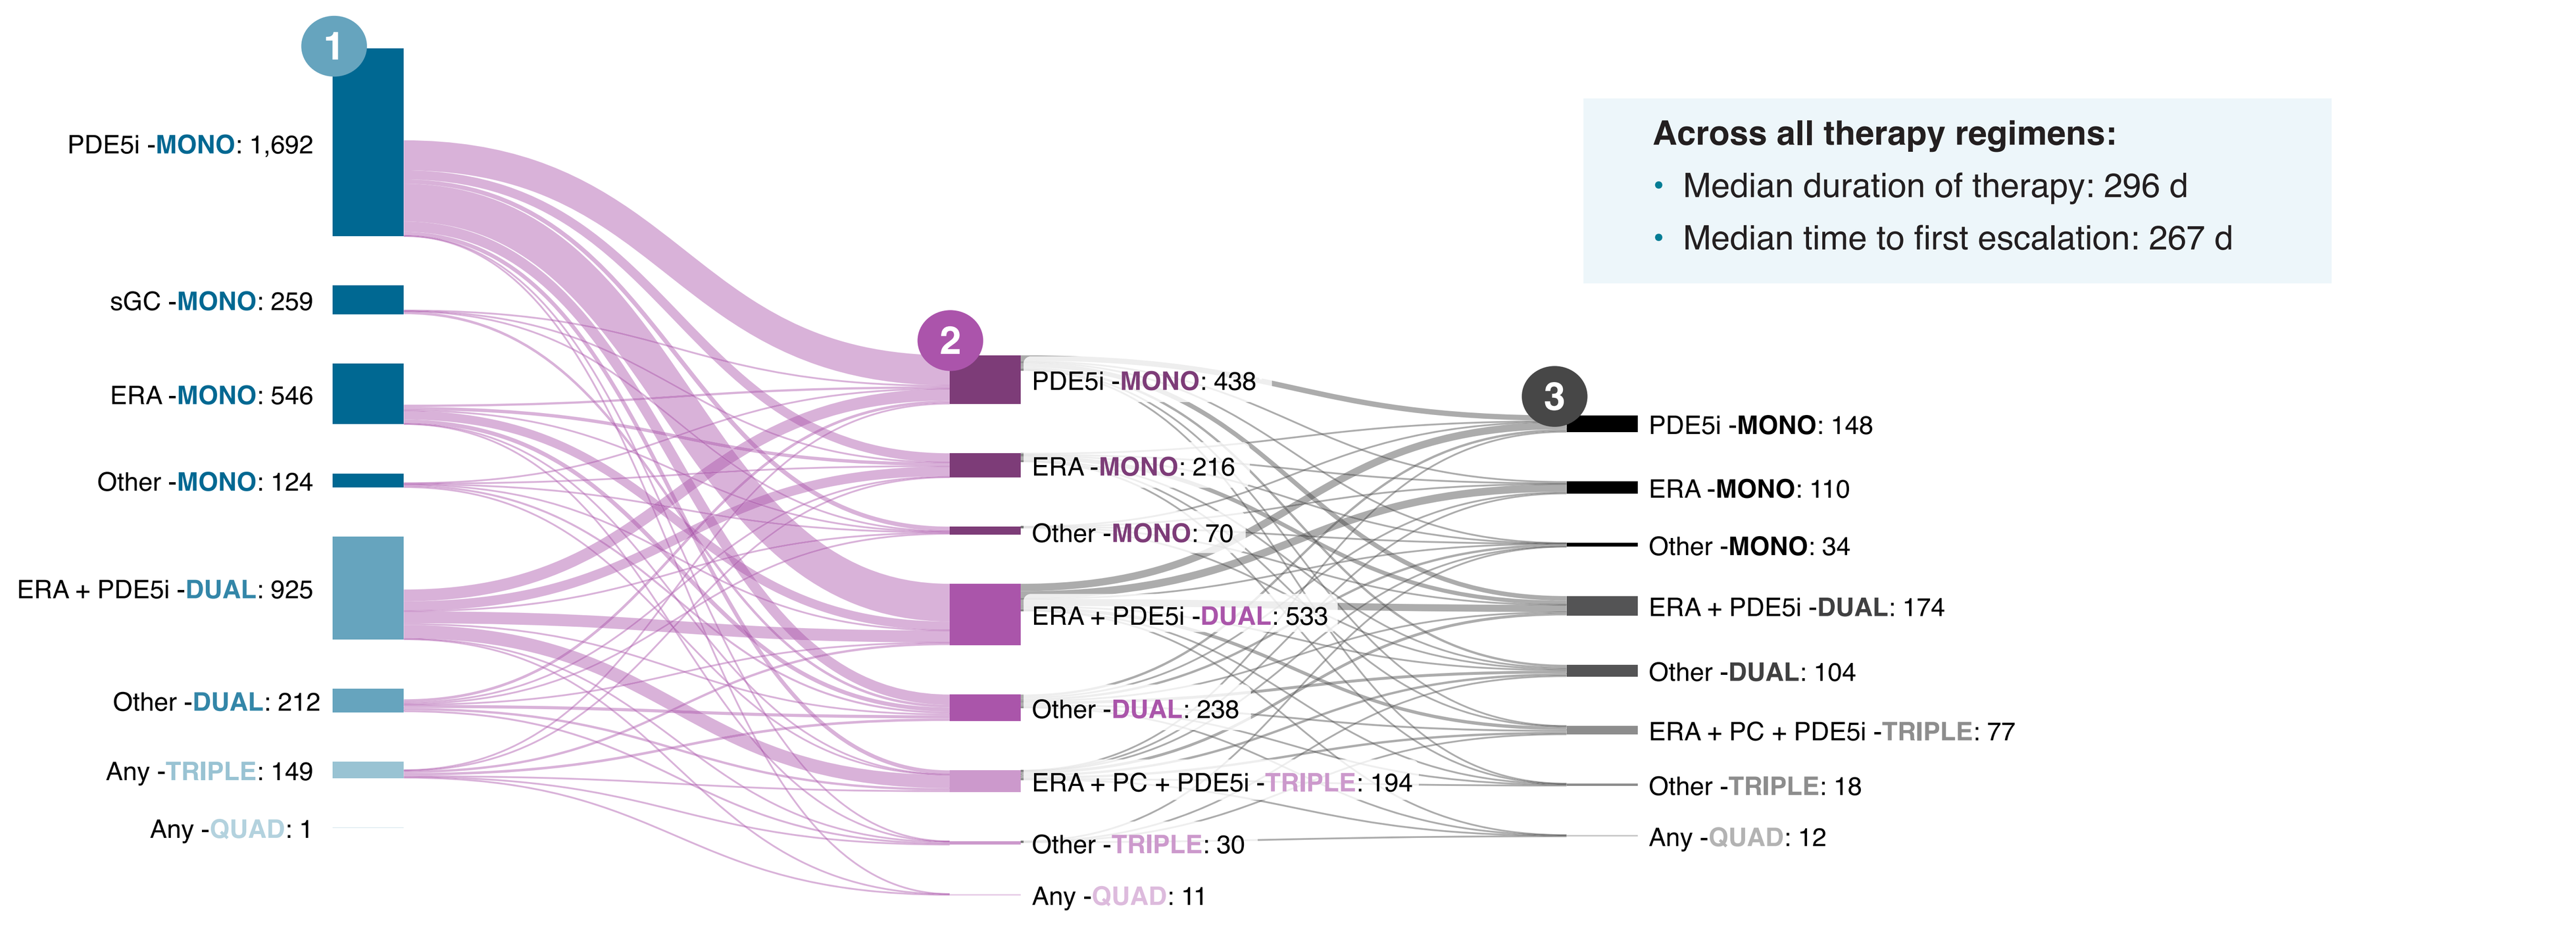
1
PDE5i -MONO: 1,692
sGC -MONO: 259
ERA -MONO: 546
Other -MONO: 124
ERA + PDE5i -DUAL: 925
Other -DUAL: 212
Any -TRIPLE: 149
Any -QUAD: 1
2
PDE5i -MONO: 438
ERA -MONO: 216
Other -MONO: 70
ERA + PDE5i -DUAL: 533
Other -DUAL: 238
ERA + PC + PDE5i -TRIPLE: 194
Other -TRIPLE: 30
Any -QUAD: 11
3
PDE5i -MONO: 148
ERA -MONO: 110
Other -MONO: 34
ERA + PDE5i -DUAL: 174
Other -DUAL: 104
ERA + PC + PDE5i -TRIPLE: 77
Other -TRIPLE: 18
Any -QUAD: 12
Across all therapy regimens:
Median duration of therapy: 296 d
Median time to first escalation: 267 d

1 Of 3,908 patients on initial Tx regimen:
- **45%** continued to a second Tx regimen
- **21%** added ≥1 agents in their second Tx regimen
- **8%** started a PC in their second Tx regimen

2 Of 1,750 patients on a second Tx regimen:
- **39%** continued to a third Tx regimen
- **5%** added ≥1 agents in their third Tx regimen
- **2%** started a PC in their third Tx regimen

3 Of 677 patients on a third Tx regimen:
- **37%** continued to a fourth Tx regimen
- **15%** added ≥1 agents in their fourth Tx regimen
- **3%** started a PC in their fourth Tx regimen

3.9k patients; period March 2020—March 2025.
dual, dual therapy; ERA, endothelin receptor antagonist; mono, monotherapy; PC, prostacyclin; PDE5i, phosphodiesterase type 5 inhibitor; quad, quadruple therapy; sGC, soluble guanylyl cyclase; triple, triple therapy; Tx, therapy.

- Qualitative interviews identified key factors influencing treatment choice, including clinical classification, age, and comorbidities, which guided subsequent claims-based analyses
- Given the absence of clinical classification in claims data, we focused on evaluating the impact of age and comorbidities on treatment decisions

- Younger patients tended to be treated with more aggressive treatment than did older patients **(Figure 3)**

Figure 3. Treatment Patterns by Age (Percentage of Patients) (Cohort 2)[a]

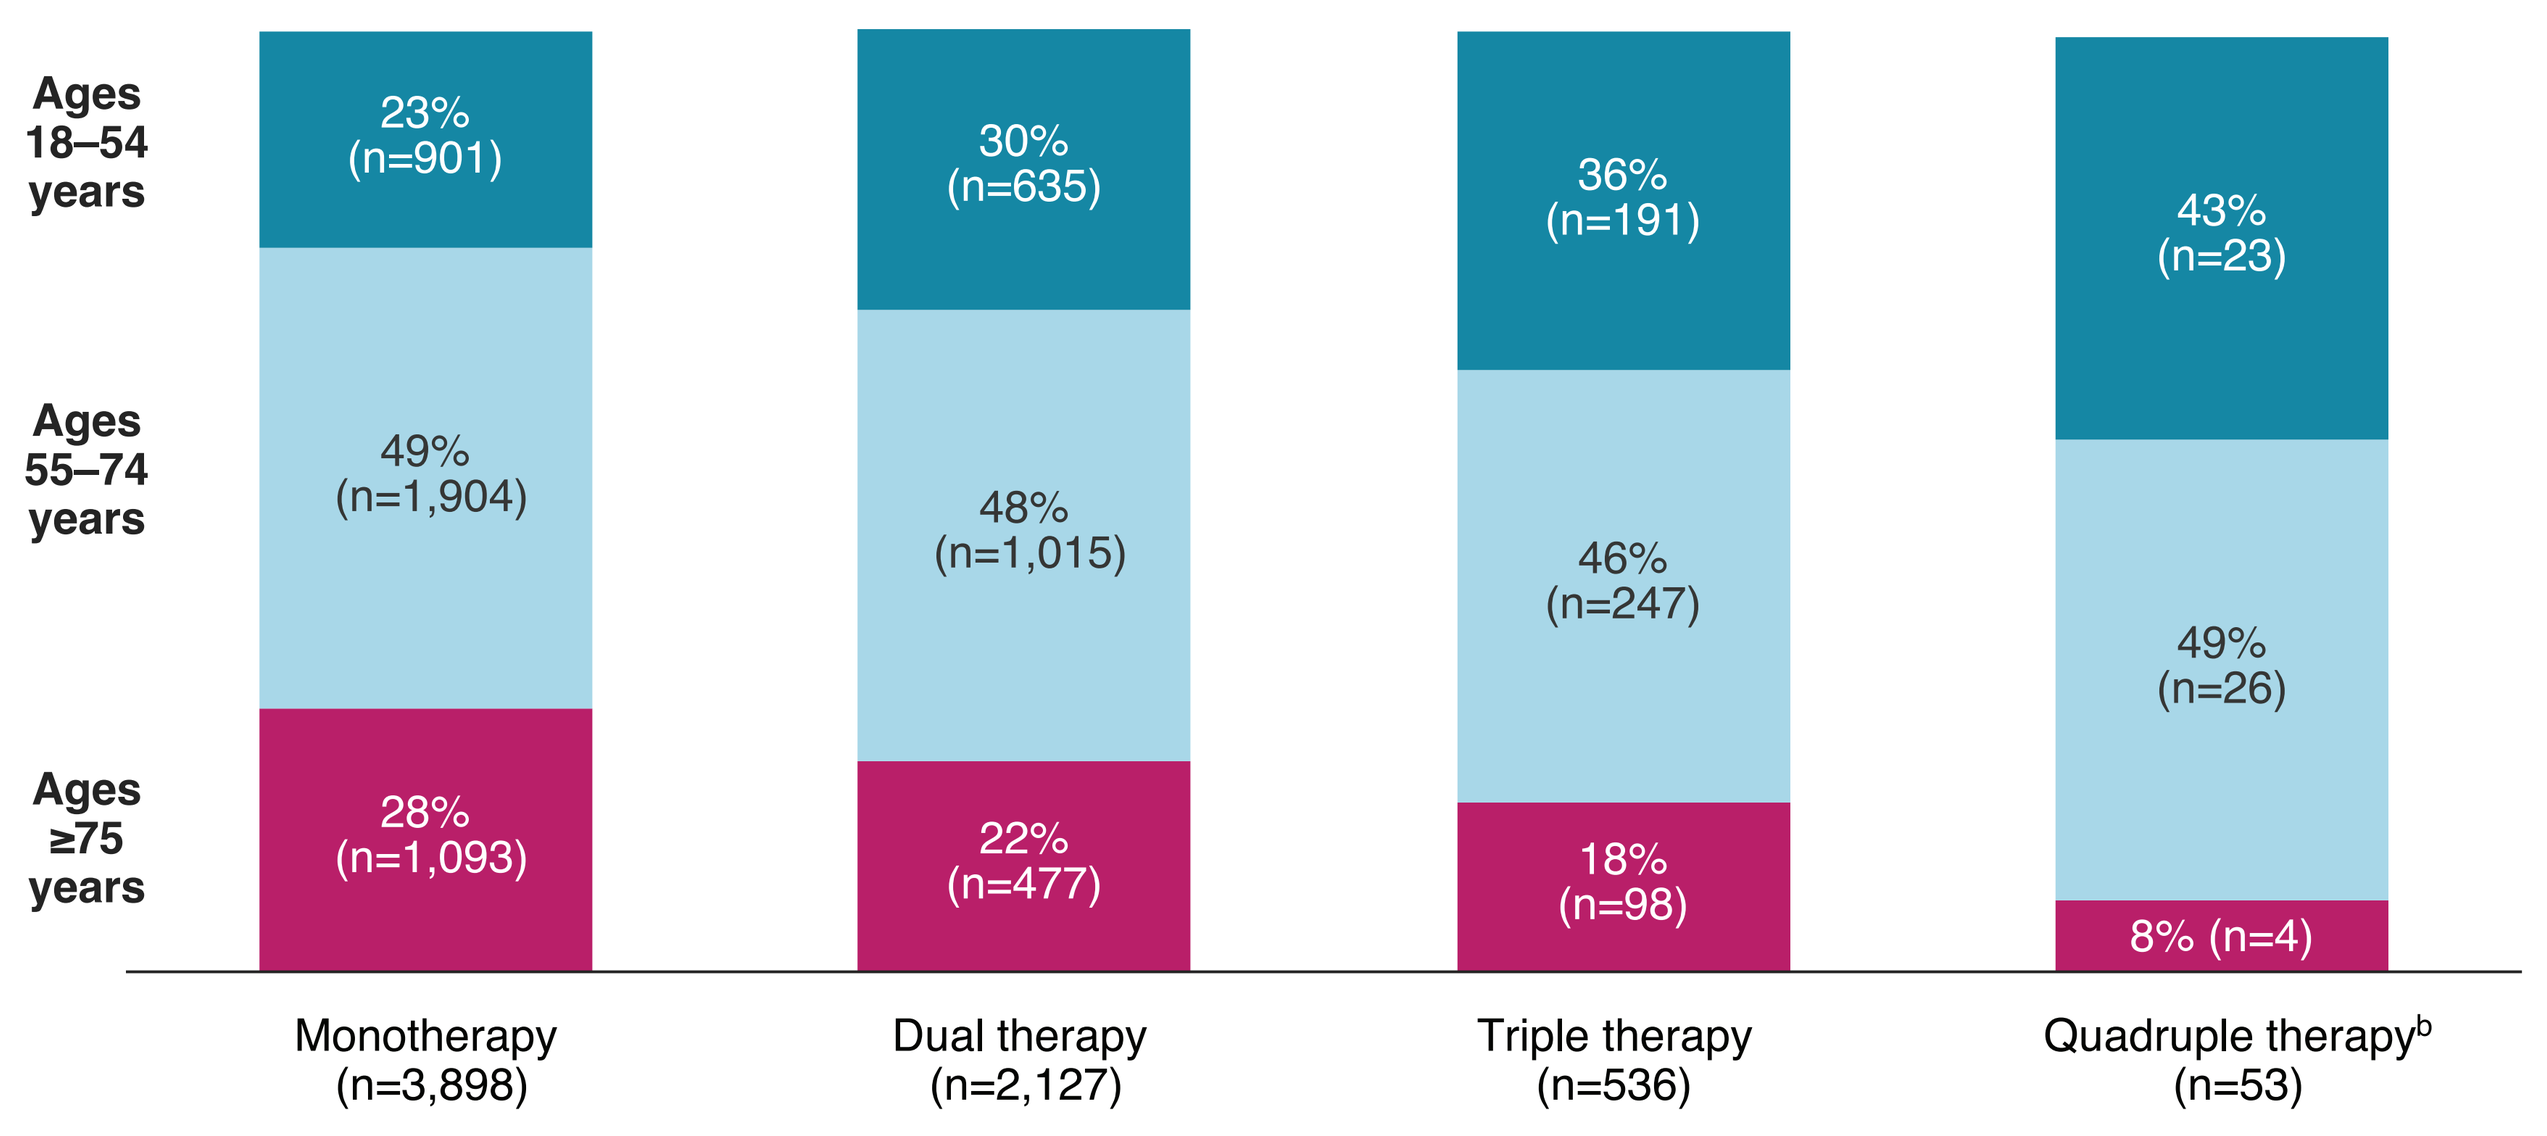
Ages 18–54 years
Ages 55–74 years
Ages ≥75 years
23% (n=901)
49% (n=1,904)
28% (n=1,093)
30% (n=635)
48% (n=1,015)
22% (n=477)
36% (n=191)
46% (n=247)
18% (n=98)
43% (n=23)
49% (n=26)
8% (n=4)
Monotherapy (n=3,898)
Dual therapy (n=2,127)
Triple therapy (n=536)
Quadruple therapy[b] (n=53)

[a]6.6k patients; period January 2023–December 2024. [b]53 patients were on quadruple therapy, leading to higher variance in percentage.

- Connective tissue disease and other forms of pulmonary hypertension appeared to be associated with more aggressive treatment regimens, while PAH patients with cardiovascular comorbidities appeared more likely to be on PDE5i monotherapy **(Figure 4)**

Figure 4. Treatment Patterns by Comorbidities

A. Percentage of patients with most frequent comorbidities (Cohort 1)

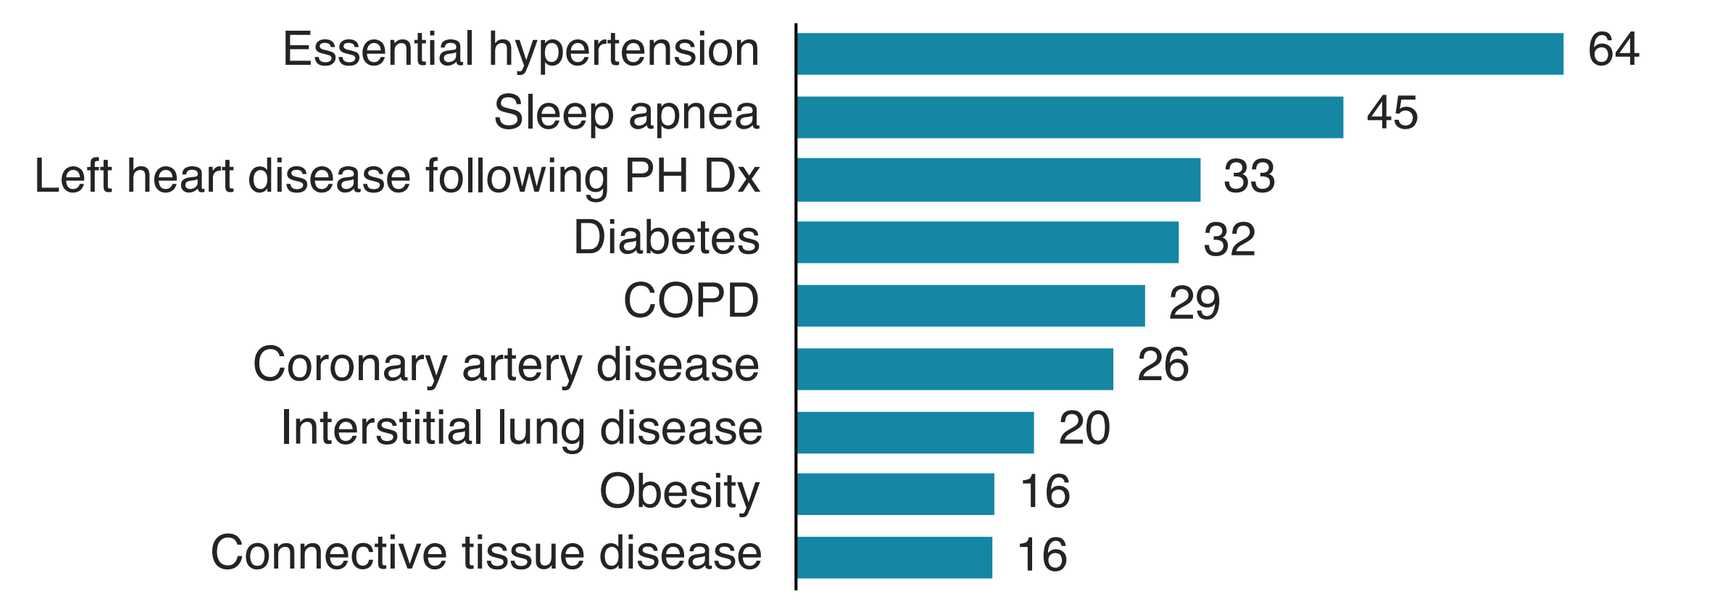
Essential hypertension 64
Sleep apnea 45
Left heart disease following PH Dx 33
Diabetes 32
COPD 29
Coronary artery disease 26
Interstitial lung disease 20
Obesity 16
Connective tissue disease 16

Left heart disease only included patients who had their first left heart disease diagnosis after initiating PAH treatment.

B. Comorbidity clusters (Cohort 2)[a]

Respiratory[b]	15%
Connective tissue disease associated[c]	26%
Other forms of PH	22%
Cardiovascular/cardiometabolic[d]	24%
None[e]	14%

C. Statistically significant correlations between comorbidity and treatment regimen (Cohort 2)[f]

Cluster	Comorbidity	Monotherapy	Dual	Triple	Quadruple
Respiratory	Acute pulmonary embolism	Y	N	N	N
Connective tissue disease associated	Connective tissue disease (excl. CREST)	N	Y	N	N
	Limited cutaneous systemic sclerosis/CREST	N	Y	Y	N
	Interstitial lung disease	N	Y	Y	N
Other forms of PH	Left heart disease following PH Dx	N	Y	Y	N
	Other PH unspecified	N	Y	Y	Y
Cardiovascular/ cardiometabolic	Essential hypertension	Y	N	N	N
	Diabetes	Y	N	N	N
	Coronary artery disease	Y	N	N	N

Left heart disease only included patients who had their first left heart disease diagnosis after initiating PAH treatment.

Panel A: 3.9k patients; period March 2020—March 2025; Panels B and C: 6.6k patients; period January 2023–December 2024.
[a]Since many PAH patients have multiple comorbidities, comorbidities were made mutually exclusive to enable segmentation of patients into clusters. Comorbidities were prioritized in the following order: 1. Respiratory; 2. Associated with CTD; 3. Other PH; 4. Cardiovascular/cardiometabolic; 5. None. As an example, if a patient had CTD and any other comorbidity outside of a respiratory comorbidity, they were assigned to the CTD cohort. [b]Included acute pulmonary embolism; [c]Included CTD (excluding CREST), limited cutaneous systemic sclerosis/CREST, and interstitial lung disease; [d]Included essential hypertension, diabetes, coronary artery disease; [e]This group may have had comorbidities not associated with the clusters, but these did not significantly influence treatment decisions; [f]Of the 23 comorbidities present in the cohort, only the 9 listed here had a statistically significant association ($P<0.05$ with Benjamini–Hochberg correction) with any treatment regimen; Y, indicates a significant association.

COPD, chronic obstructive pulmonary disease; CTD, connective tissue disease; Dx, diagnosis; excl, excluding; N, no; P(A)H, pulmonary (arterial) hypertension; Y, yes.

LIMITATIONS

- Despite criteria to exclude patients with other forms of PH beyond PAH, the claims data analysis cohort may have included some of these patients, which could influence the findings
- Claims data did not include clinical findings (such as risk status, functional class or right heart catheterization and echocardiography parameters) that could have provided information on why treatment changes were made

CONCLUSIONS

- This mixed methods analysis in a large, contemporary US PAH cohort identified that despite current treatment guidelines and recommendations, a large number of patients started on and remained on monotherapy
- Qualitative interviews indicated that risk classification and comorbidities are driving treatment choice; consistent with this, claims data showed that comorbidities and age were associated with treatment patterns
- These findings may inform targeted strategies to better align real-world care with guideline-directed therapy, reduce delays in escalation, and ultimately improve both clinical and healthcare utilization outcomes

References:

1 Chin KM, et al. *Eur Respir J*. 2024;64(4):2401325.
2 Humbert M, et al. *Eur Respir J*. 2023;61(1):2200879.
3 Leary PJ, et al. *Am J Respir Crit Care Med*. 2025;211(4):619-627.
4 Stubbe B, et al. *BMC Pulm Med*. 2021;21(1)130.

Research supported by: Chiesi Farmaceutici S.p.A. and Gossamer Bio, Inc.