1039

Understanding Patient Experiences, Challenges, and Emotions During Diagnosis of PH-ILD: A Qualitative Study

Jennalyn Mayeux[1], Sandy L. Nguyen[2], Amy Allen[3], Sherry Danese[4], Vera Toth-Fejel[2], Julie Ulloa[4], Lori G. Reed[5]

[1]University of Utah, Salt Lake City, UT, USA; [2]Gossamer Bio, Inc., San Diego, CA, USA; [3]Nickson Healthcare Consulting, Azle, TX, USA; [4]OutcomesInsights, Calabasas, CA, USA; [5]Piedmont Healthcare Advanced Lung Disease, Atlanta, GA, USA

Presented at PHA 2026 International PH Conference and Scientific Sessions
Dallas, TX
June 11-14, 2026

BACKGROUND

- Pulmonary hypertension (PH) associated with interstitial lung disease (ILD) is a progressive condition that can greatly affect a person's daily functioning and quality of life[1,2]
- Diagnosis is often delayed because PH and ILD share many symptoms and lack standardized screening protocols[2]
- This qualitative study explored experiences, challenges, and emotions of people living with PH-ILD before and after receiving their diagnosis

METHODS

Figure 1. Qualitative research design

ONLINE BULLETIN BOARD	**18 participants** (15 patients, 3 caregivers)	From **June 9–15, 2025**
VIRTUAL IN-DEPTH INTERVIEWS	**19 patients participated**	From **July 15–30, 2025**

To participate, respondents had to:

- ☑ Live in the US
- ☑ Be age 18+
- ☑ Been diagnosed (or care for someone diagnosed) with ILD as well as PH
- ☑ Have right heart catheterization (RHC)
- ☑ Never been diagnosed with cancer
- ☑ Not have poorly controlled high blood pressure or diabetes

- The online bulletin board was a private website where participants were able to log in at their convenience to answer questions and share their thoughts
- Online bulletin board participants completed daily tasks—written responses, short videos, and ranking exercises—with a moderator probing for deeper insights
- Interview topics were informed by themes emerging from the online bulletin board
- A thematic analysis was used to identify recurring ideas and patient-reported needs
- "Patient perspectives" include input from both patients and caregivers

RESULTS

Table. Participant demographics

Characteristic	
Age, y	
40-59	20
60-69	12
70+	5
Sex, n	
Male	8
Female	29
Ethnicity, n	
White	18
Black or African American	16
Hispanic or Latino	2
Asian	1
PH-ILD–specific therapy use, n	
Yes	14
No	23
Top ILD subtypes, n	
Autoimmune / CTD ILD[a]	10
Sarcoidosis	8
IPF	7
Unspecified pulmonary fibrosis	7

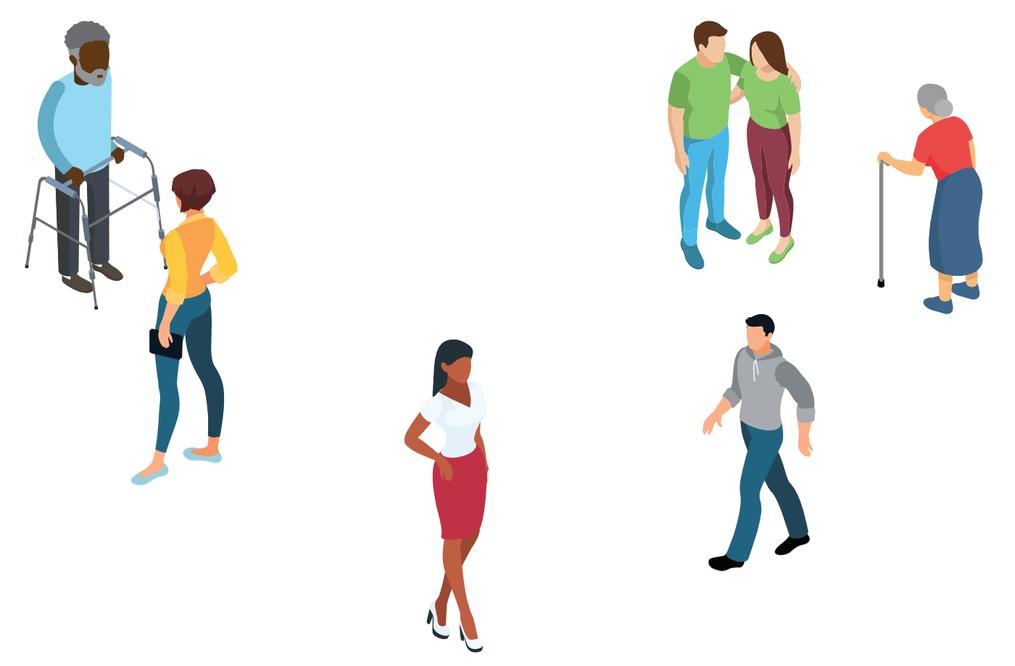

[a]Included autoimmune disease, CTD, scleroderma and polymyositis. Other ILD subtypes included bronchiectasis, hypersensitivity pneumonitis, NSIP, and unknown.

CTD, connective tissue disease; IPF, idiopathic pulmonary fibrosis; n, number; NSIP, nonspecific interstitial pneumonia; PH-ILD, pulmonary hypertension associated with interstitial lung disease; y, year.

RESULTS (continued)

Figure 2. PH-ILD patient journey: From discovery to treatment, life with PH-ILD feels like a maze that is confusing, hard to navigate, and seemingly unending

1 WHERE IT ALL BEGINS
ILD Diagnosis (1–10 Years)

ILD symptoms begin to impact daily life and independence
- Worsening shortness of breath, fatigue, and dizziness begin limiting daily activities and roles
- Symptoms are often attributed to ILD, masking emerging changes

2 SEARCHING FOR ANSWERS
PH Symptom Onset (6–18 Months)

New or worsening symptoms create confusion and uncertainty
- Overlap with ILD delays recognition of PH until more distinct signs (eg, fainting, palpitations) emerge
- Patients are referred for testing (echo, CT, RHC), often without clear understanding

3 ADDING ANOTHER CONDITION
PH Diagnosis (0–3 Months)

Patients struggle to understand what causes PH, how ILD and PH differ, and what this means for their lifespan
- Educational resources might be offered by the HCP, but are often limited
- Patients seek answers through websites, support groups, and forums to fill information gaps

4 IMPROVEMENT HOW?
PH Treatment

Treatment decisions feel unclear and overwhelming
- HCPs introduce treatment options (eg, PDE5i, inhaled treprostinil), often with limited discussion of goals, rationale, or alternatives
- Patients feel underinformed about efficacy and side effects

5 UNCERTAIN FUTURE
PH Management

Lack of clear treatment goals and limited options make PH feel out of control
- Ongoing treatment adjustments (eg, inhaled treprostinil, sildenafil add-on) offer limited perceived benefit
- Patients face worsening symptoms and increasing burden despite treatment
- Patients turn to support groups, forums, and therapy for emotional support and practical advice

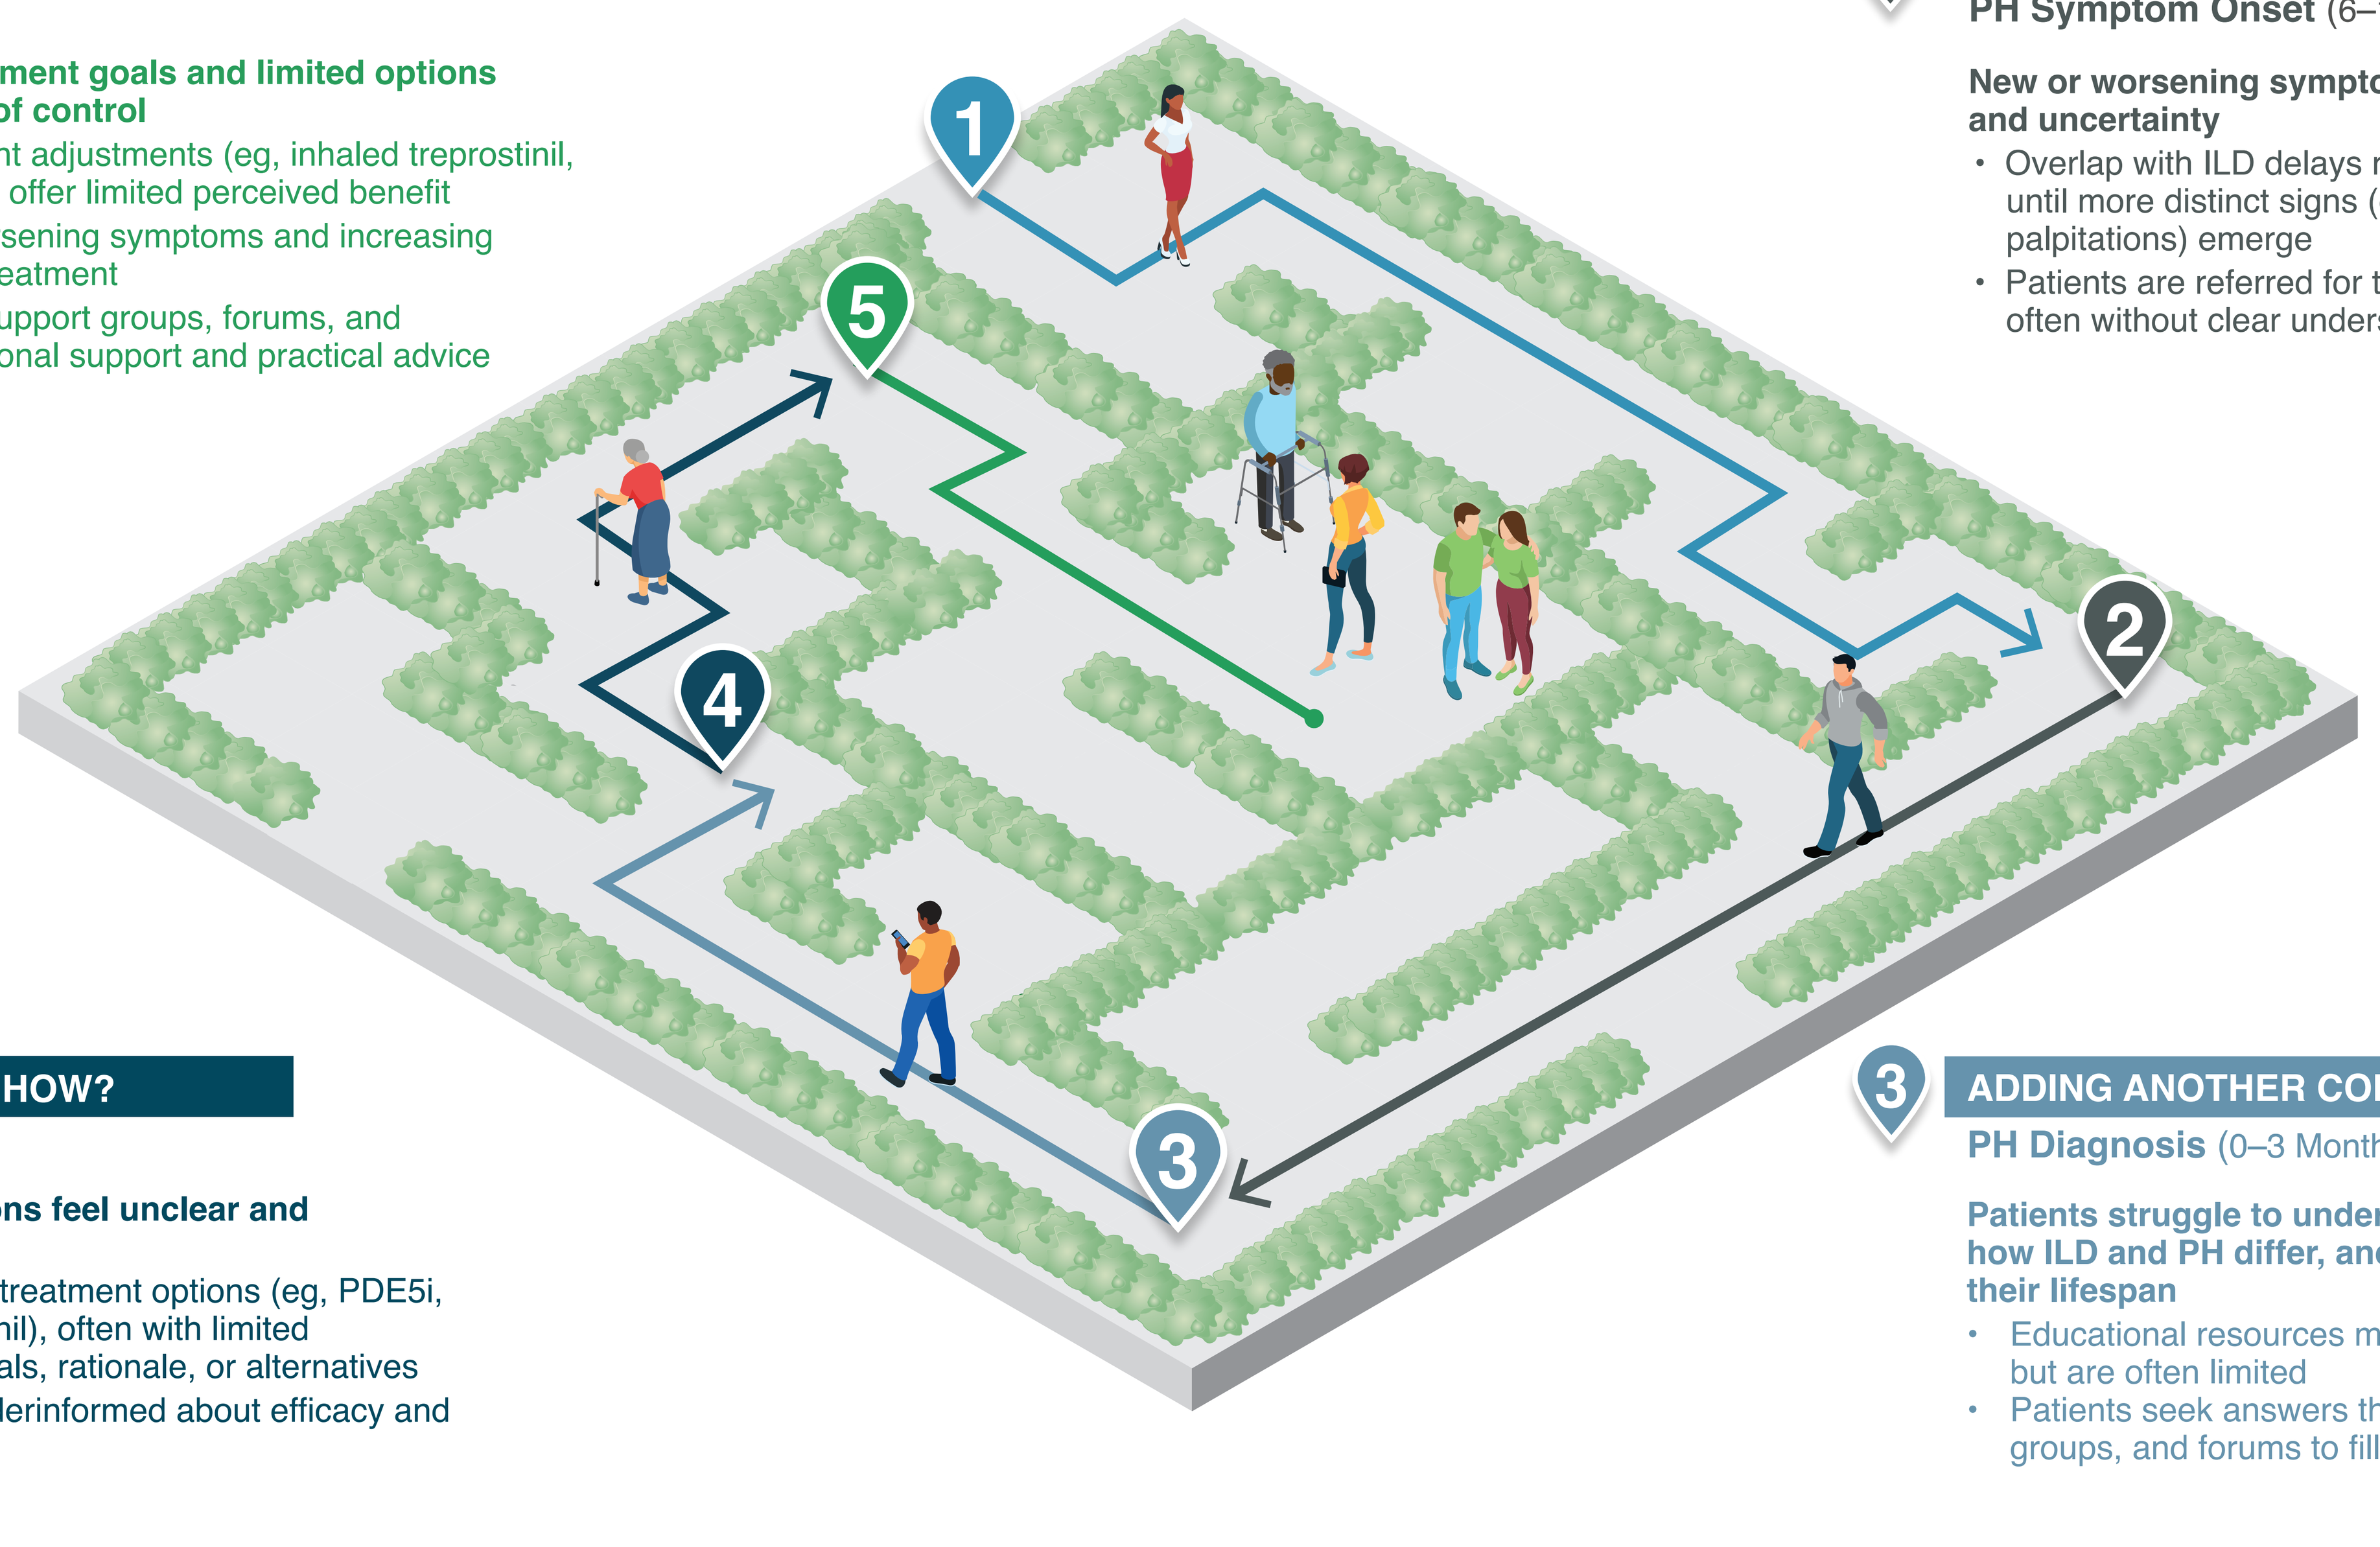
1
2
3
4
5

CT, computed tomography; echo, echocardiography; HCP, healthcare provider; ILD, interstitial lung disease; PDE5i, phosphodiesterase type 5 inhibitor; PH, pulmonary hypertension; PH-ILD, pulmonary hypertension associated with interstitial lung disease; RHC, right heart catheterization.

Participants described several challenges related to diagnosis and understanding their disease:

Among those diagnosed with ILD before PH (n=25):

56% said they were **unaware that PH could develop**

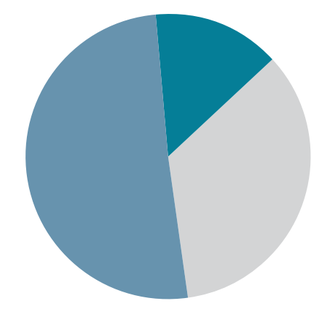

and only 16% recalled **healthcare providers (HCP) discussing this risk**

- Most patients who did not know about the risk of PH **(79%) wished they had been told earlier**

Among all participants (n=37):

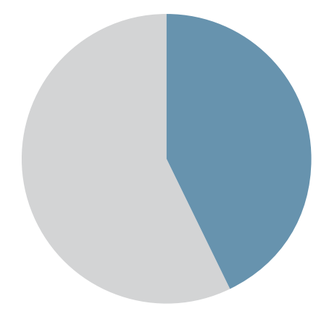

43% reported **emotional distress intensified by uncertainty** about worsening and severity of symptoms prior to diagnosis of PH-ILD

- After hearing their provider's explanation of PH-ILD, many reported **feeling confused (35%) and/or overwhelmed (22%)**
- Reasons for confusion included **explanations that were too general (41%)** or relied on **unfamiliar medical terminology (19%)**

Figure 5. Top challenges in long-term PH-ILD management

Emotional and Psychological Impact
- Fears about disease progression and anxiety over potential health crises grow
- Patients become more depressed over further loss of independence, often turning to caregiver and loved ones for emotional support
- Progressively worsening physical limitations lead to social isolation, increased depression and guilt over loss of purpose and role in family and life
- Caregivers struggle with the changing nature of relationship with their loved one

Logistical and Financial Burden
- Increasing polypharmacy, testing, insurance hassles, transportation costs and time off work increase financial burden
- Coordinating lengthy appointments and insurance paperwork adds complexity
- Managing multiple medications with varied administration needs is overwhelming
- Patients increasingly rely on caregivers to manage household chores, transportation, medication reminders, and basic shopping, increasing burden on caregivers

Physical Limitations and Symptom Management
- Physical symptoms remain a significant challenge, particularly breathing difficulties, fatigue, and need for limiting therapies like oxygen
- Many need increasing periods of rest, and activities can be unexpectedly draining, derailing their day
- Frustration with treatment efficacy grows, dealing with new or worsening side effects and limited efficacy with few alternatives
- As physical symptoms worsen, caregivers take on greater responsibility, sometimes losing their careers, income, and time for themselves

Figure 4. Patients appreciate when providers combine expertise with genuine compassion

PATIENTS FEEL AT EASE AND RESPECTED BY THEIR CARE TEAM WHEN THEY:

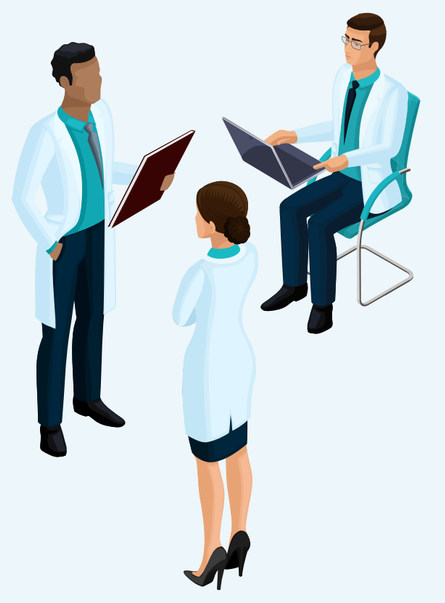

- **Genuinely listen** to patient needs and concerns
- **Explain concepts** clearly and don't rush
- **Are honest** about PH-ILD prognosis
- **Include patients** in treatment decisions
- **Show genuine concern** for patient well-being
- **Ask about or include caregivers** in discussions

"Clearly explaining things, shared decision-making, offering hopeful suggestions and concrete plans."
– Patient

Figure 5. Ideal resources: Patients desire more two-way communication with HCPs and expanded ways to obtain targeted and practical peer-to-peer advice

MOST MENTIONS

- **HCP conversations** across specialists and nurses, with outreach options as questions arise
- **Centralized resource hubs** with articles, support links, trusted websites
- **Informational videos and infographics** with easy-to-digest content that simplifies complexities
- **Smaller support groups** that are specific (eg, PH or sarcoidosis) versus generic
- **Expert-led webinars and conferences** about PH-ILD, potentially with guest specialist Q&A
- **Topic-specific educational sessions** on practical issues such as managing flare-ups, side effects, and other real-life challenges
- **Peer mentorship/buddy programs** for newly diagnosed patients and their caregivers

CONCLUSIONS

- This study highlights challenges and unmet needs for people living with PH-ILD throughout their diagnostic journey
- Interpretation is limited by self-reported data and potential selection bias
- Opportunities to improve the patient experience include using consistent terminology, clearly communicating PH risk in ILD, acknowledging uncertainty, equipping patients with clear guidance on when and how to seek care, and making patient-friendly information and support resources accessible
- Additional research is needed to evaluate strategies that enhance communication, education, and support for people living with PH-ILD

References:
1 Heresi G, et al. *BMC Pulm Med.* 2024;24(1):335.
2 Nathan SD, et al. *J Manag Care Spec Pharm.* 2025;31(1-a Suppl):S2-S17.

Acknowledgements: We would like to sincerely thank the patients and caregivers for sharing their perspectives and stories.
Research supported by: Gossamer Bio, Inc.