1040

Understanding the Diagnostic Journey of Patients With Pulmonary Hypertension Associated With Interstitial Lung Disease (PH-ILD): A Retrospective US Claims Analysis

Kristin B. Highland[1], Sandy Nguyen[2], Samantha Rothstein[3], Nidhishant Dixit[3], Joyce Louie[3], Nicole Mayer[3], Bara' Momani[3], Yash Rathi[3], Lawrence Ho[2]

[1]Cleveland Clinic Lerner College of Medicine, Cleveland, OH, USA; [2]Gossamer Bio, Inc., San Diego, CA, USA; [3]MMIT, Norstella Consulting & Analytics, London, UK

Presented at PHA 2026 International PH Conference and Scientific Sessions
Dallas, TX
June 11-14, 2026

BACKGROUND

- PH-ILD is a severe, progressive, and life-limiting cardiopulmonary condition characterized by elevated pulmonary artery pressure in concert with irreversible interstitial fibrosis[1]
- Timely diagnosis is difficult and often delayed owing to nonspecific and frequently overlapping symptoms between ILD and PH, a low index of suspicion among non-PH specialists, and lack of standardized diagnostic algorithms for World Health Organization (WHO) Group 3 PH[1,2]
- This study characterizes the diagnostic journey of PH-ILD patients using real-world claims data, focusing on the diagnosis of PH in patients with ILD

METHODS

- A retrospective cohort study was conducted using the NorstellaLinQ fully integrated claims and electronic health record dataset from January 2019 to December 2024
- Adults with ≥1 claim for ILD and ≥1 claim for PH were included in the analysis
- PH-ILD was defined using ICD-10 codes indicative of both ILD and PH, excluding other PH groups
- Patients with chronic obstructive pulmonary disease or emphysema, cardiovascular conditions associated with WHO Group 2 PH, or heart failure in their medical record prior to the diagnosis of PH were excluded
- Time intervals were calculated from symptom presentation to PH diagnosis
- The diagnosing physician's specialty, as well as diagnostic procedures, were examined

RESULTS

- Among 18,666 PH-ILD patients identified, the majority were female, White, and ages ≥65 years (**Figure 1**)

Figure 1. Patient demographics (N=18,666)

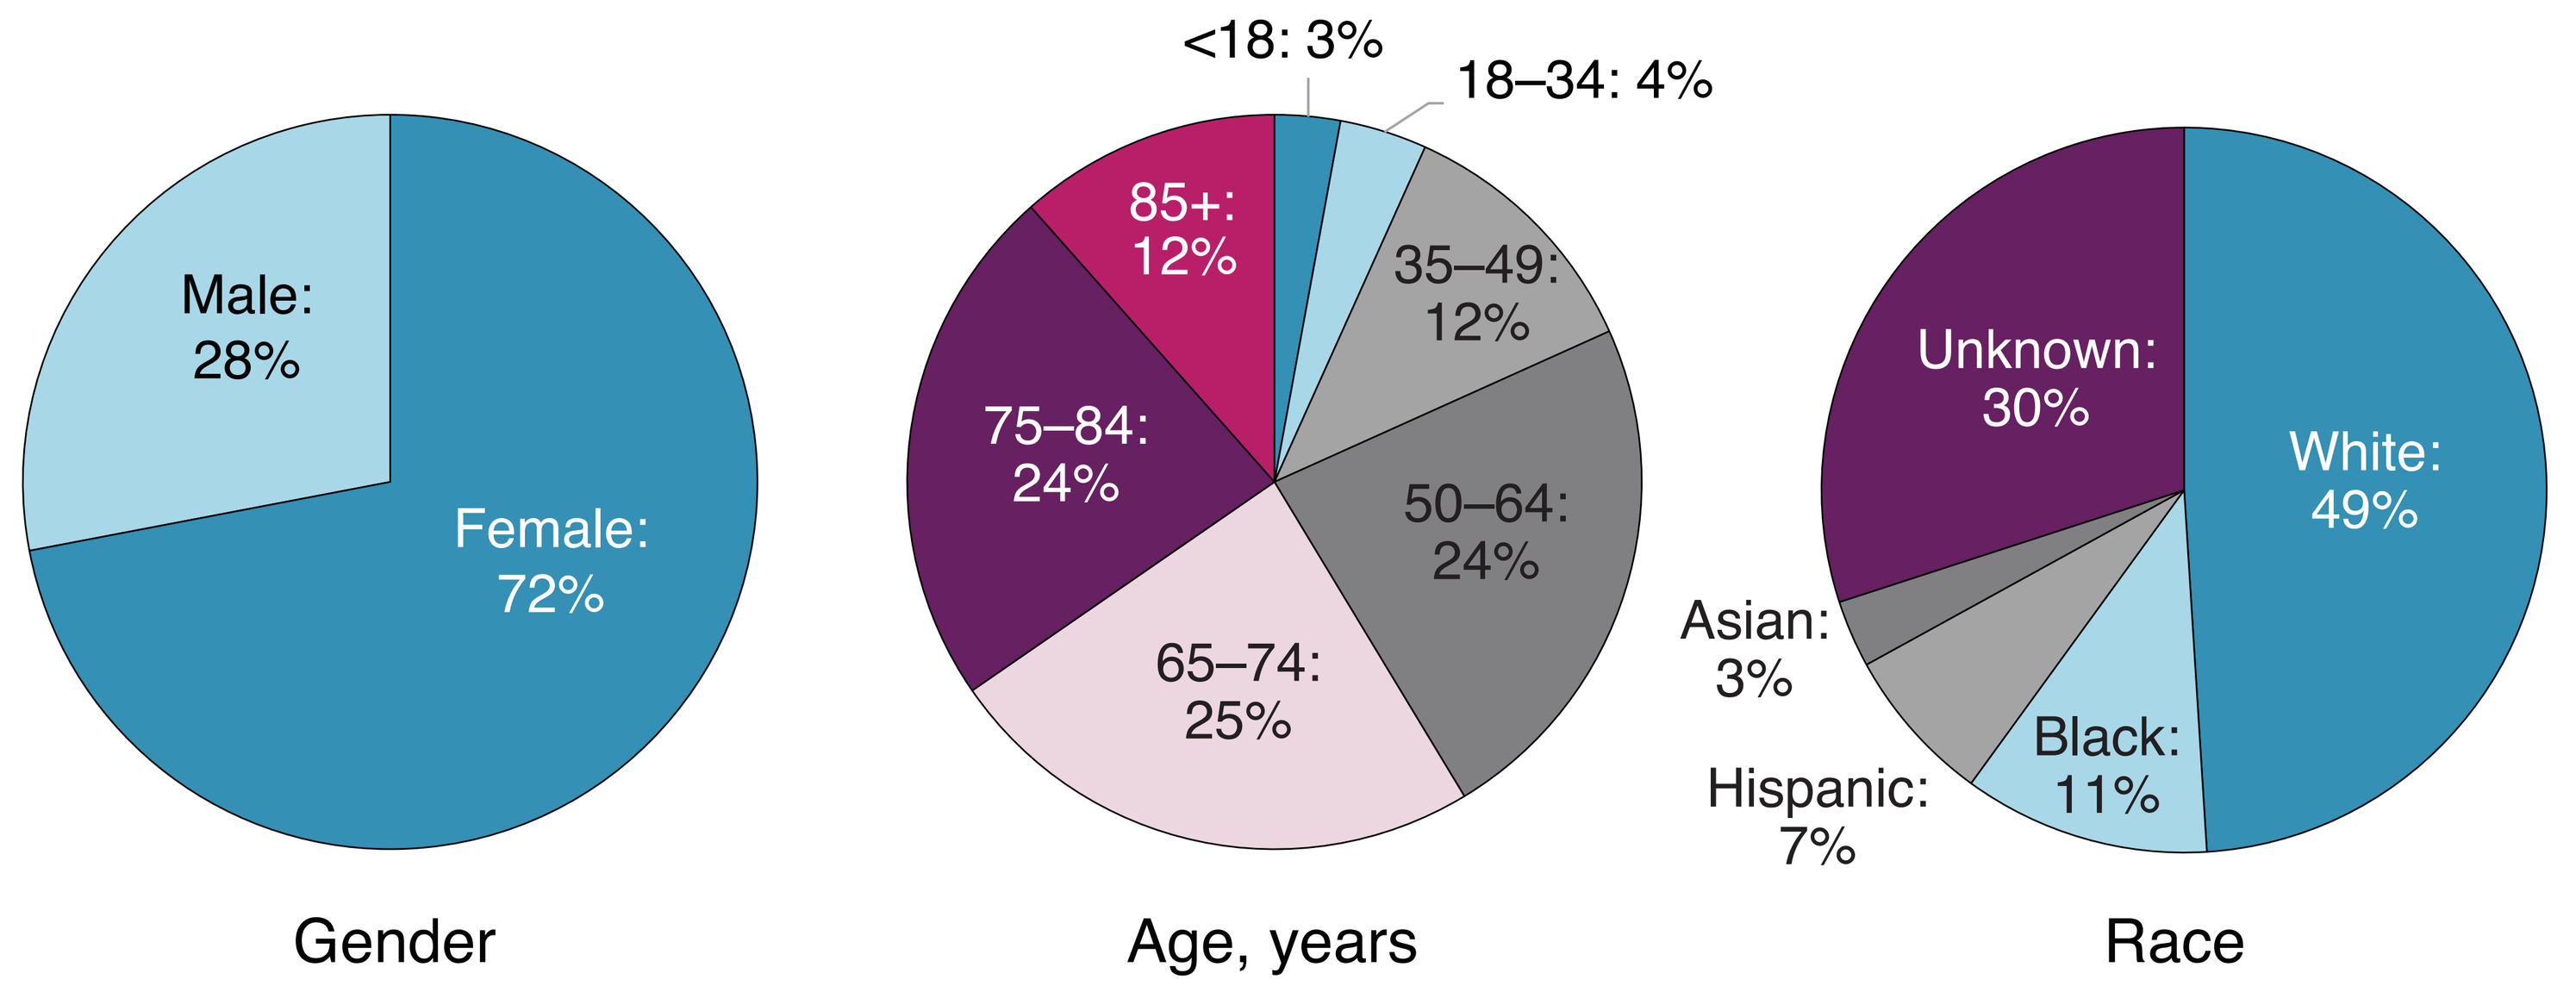
Male: 28%
Female: 72%
Gender
<18: 3%
18–34: 4%
35–49: 12%
50–64: 24%
65–74: 25%
75–84: 24%
85+: 12%
Age, years
Unknown: 30%
White: 49%
Black: 11%
Hispanic: 7%
Asian: 3%
Race

RESULTS (continued)

- Most patients (65%) were diagnosed with PH at the time of or after the diagnosis of ILD (**Figure 2**)

Figure 2. PH diagnosis occurrence relative to ILD diagnosis

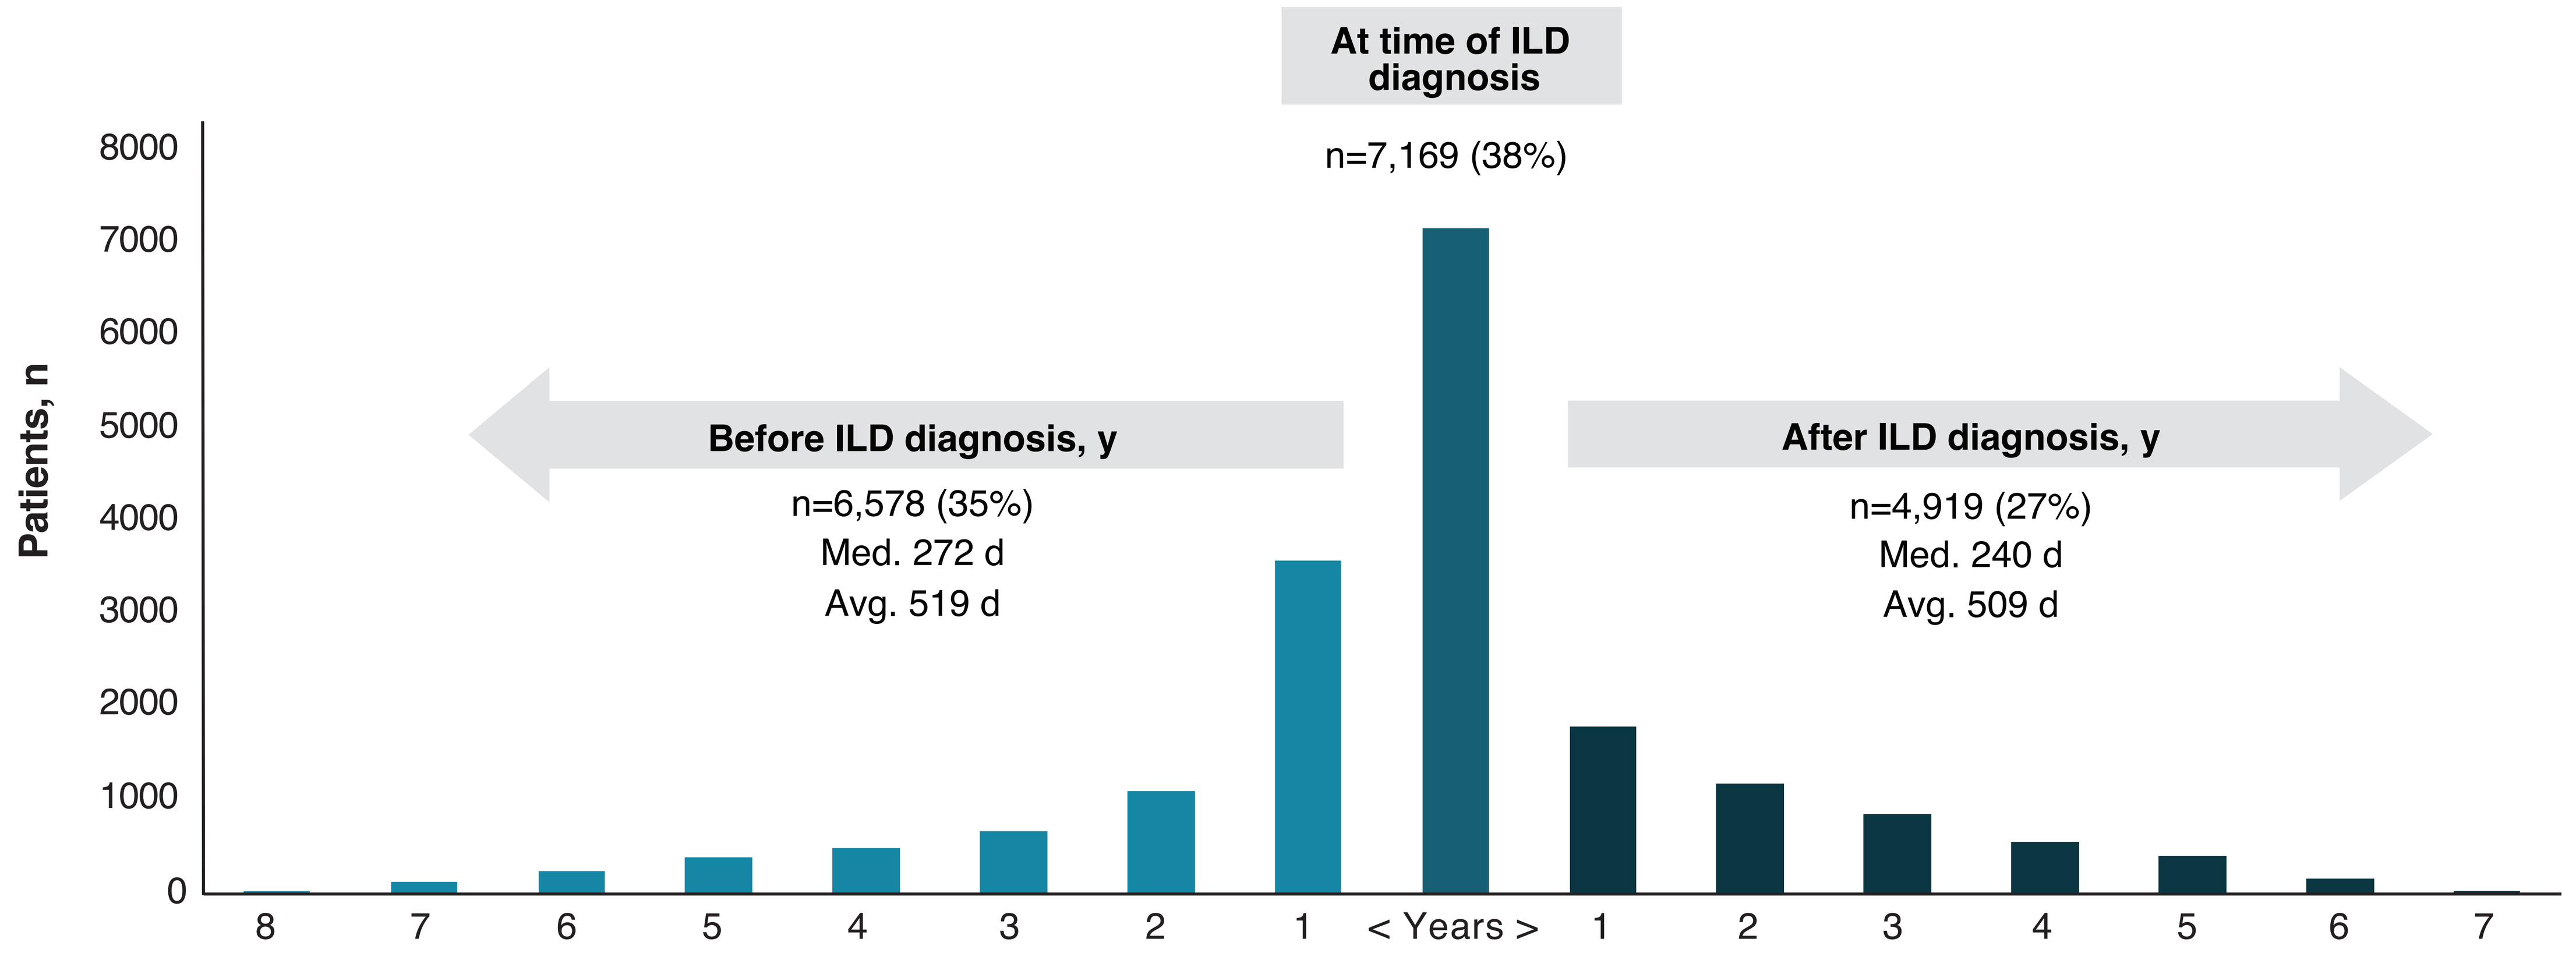
At time of ILD diagnosis
n=7,169 (38%)
Before ILD diagnosis, y
n=6,578 (35%)
Med. 272 d
Avg. 519 d
After ILD diagnosis, y
n=4,919 (27%)
Med. 240 d
Avg. 509 d
Patients, n
8000
7000
6000
5000
4000
3000
2000
1000
0
8 7 6 5 4 3 2 1 < Years > 1 2 3 4 5 6 7

The median of a set of numbers is the "middle" value, the one that separates the higher half from the lower half of a set of numbers. Avg, average; d, day; ILD, interstitial lung disease; med, median; n, number; PH, pulmonary hypertension; y, year.

- The most common symptoms preceding PH diagnosis were dyspnea (20%), dizziness (14%), edema (14%), and palpitations (12%) (**Figure 3**)

Figure 3. Symptom occurrence in the years prior to and after PH diagnosis

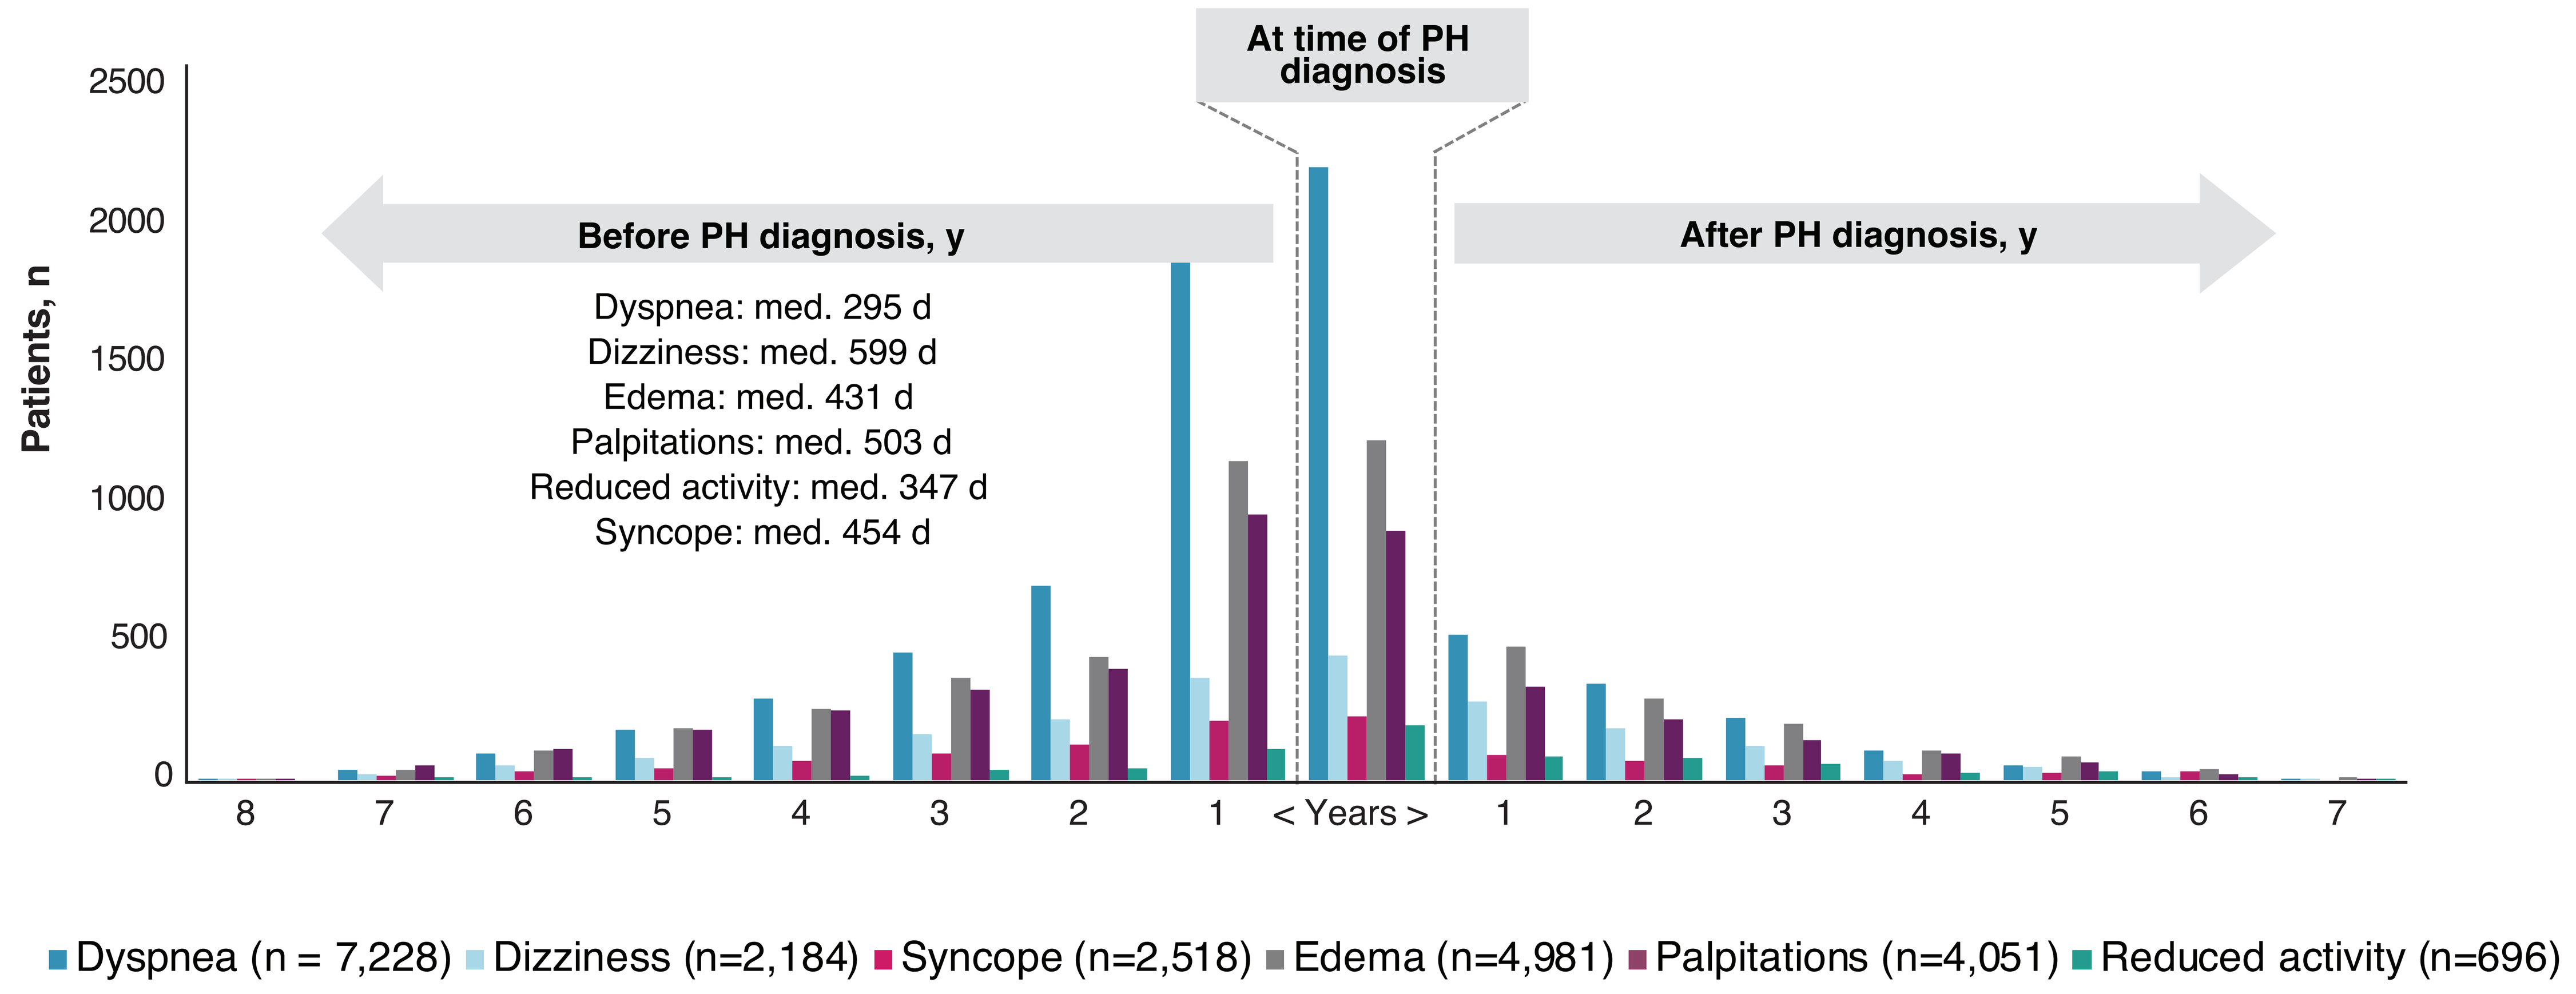
At time of PH diagnosis
Before PH diagnosis, y
After PH diagnosis, y
Dyspnea: med. 295 d
Dizziness: med. 599 d
Edema: med. 431 d
Palpitations: med. 503 d
Reduced activity: med. 347 d
Syncope: med. 454 d
Patients, n
2500
2000
1500
1000
500
0
8 7 6 5 4 3 2 1 < Years > 1 2 3 4 5 6 7
Dyspnea (n = 7,228)
Dizziness (n=2,184)
Syncope (n=2,518)
Edema (n=4,981)
Palpitations (n=4,051)
Reduced activity (n=696)

The median of a set of numbers is the "middle" value, the one that separates the higher half from the lower half of a set of numbers. d, day; med; median; n, number; PH, pulmonary hypertension; y, year.

- PH diagnoses were performed by cardiologists (29%), pulmonologists (19%), and non-PH focused specialties (45%) (**Table 1**)

Table 1. Patients with PH diagnosed by HCPs with PH-focused specialties

HCPs diagnosing PH-ILD	Patients, n (%)
Cardiology	5,371 (29)
Pulmonology	3,610 (19)
Rheumatology	1,070 (6)
Other	53 (<1)
Total diagnosed by PH-focused specialties	**10,189 (55)**

HCP, healthcare professional; ILD, interstitial lung disease; n, number; PH, pulmonary hypertension.

- Diagnostic modalities used for PH evaluation before or in the same year as PH diagnosis were echocardiography (52%), CT scan (29%), PFT/spirometry (25%; includes DLCO), 6MWT (20%), and RHC (12%) (**Figure 4**)
- 22% of patients used supplemental oxygen before or in the same year as PH diagnosis

Figure 4. Most common diagnostic tests performed before or in the same year as PH diagnosis

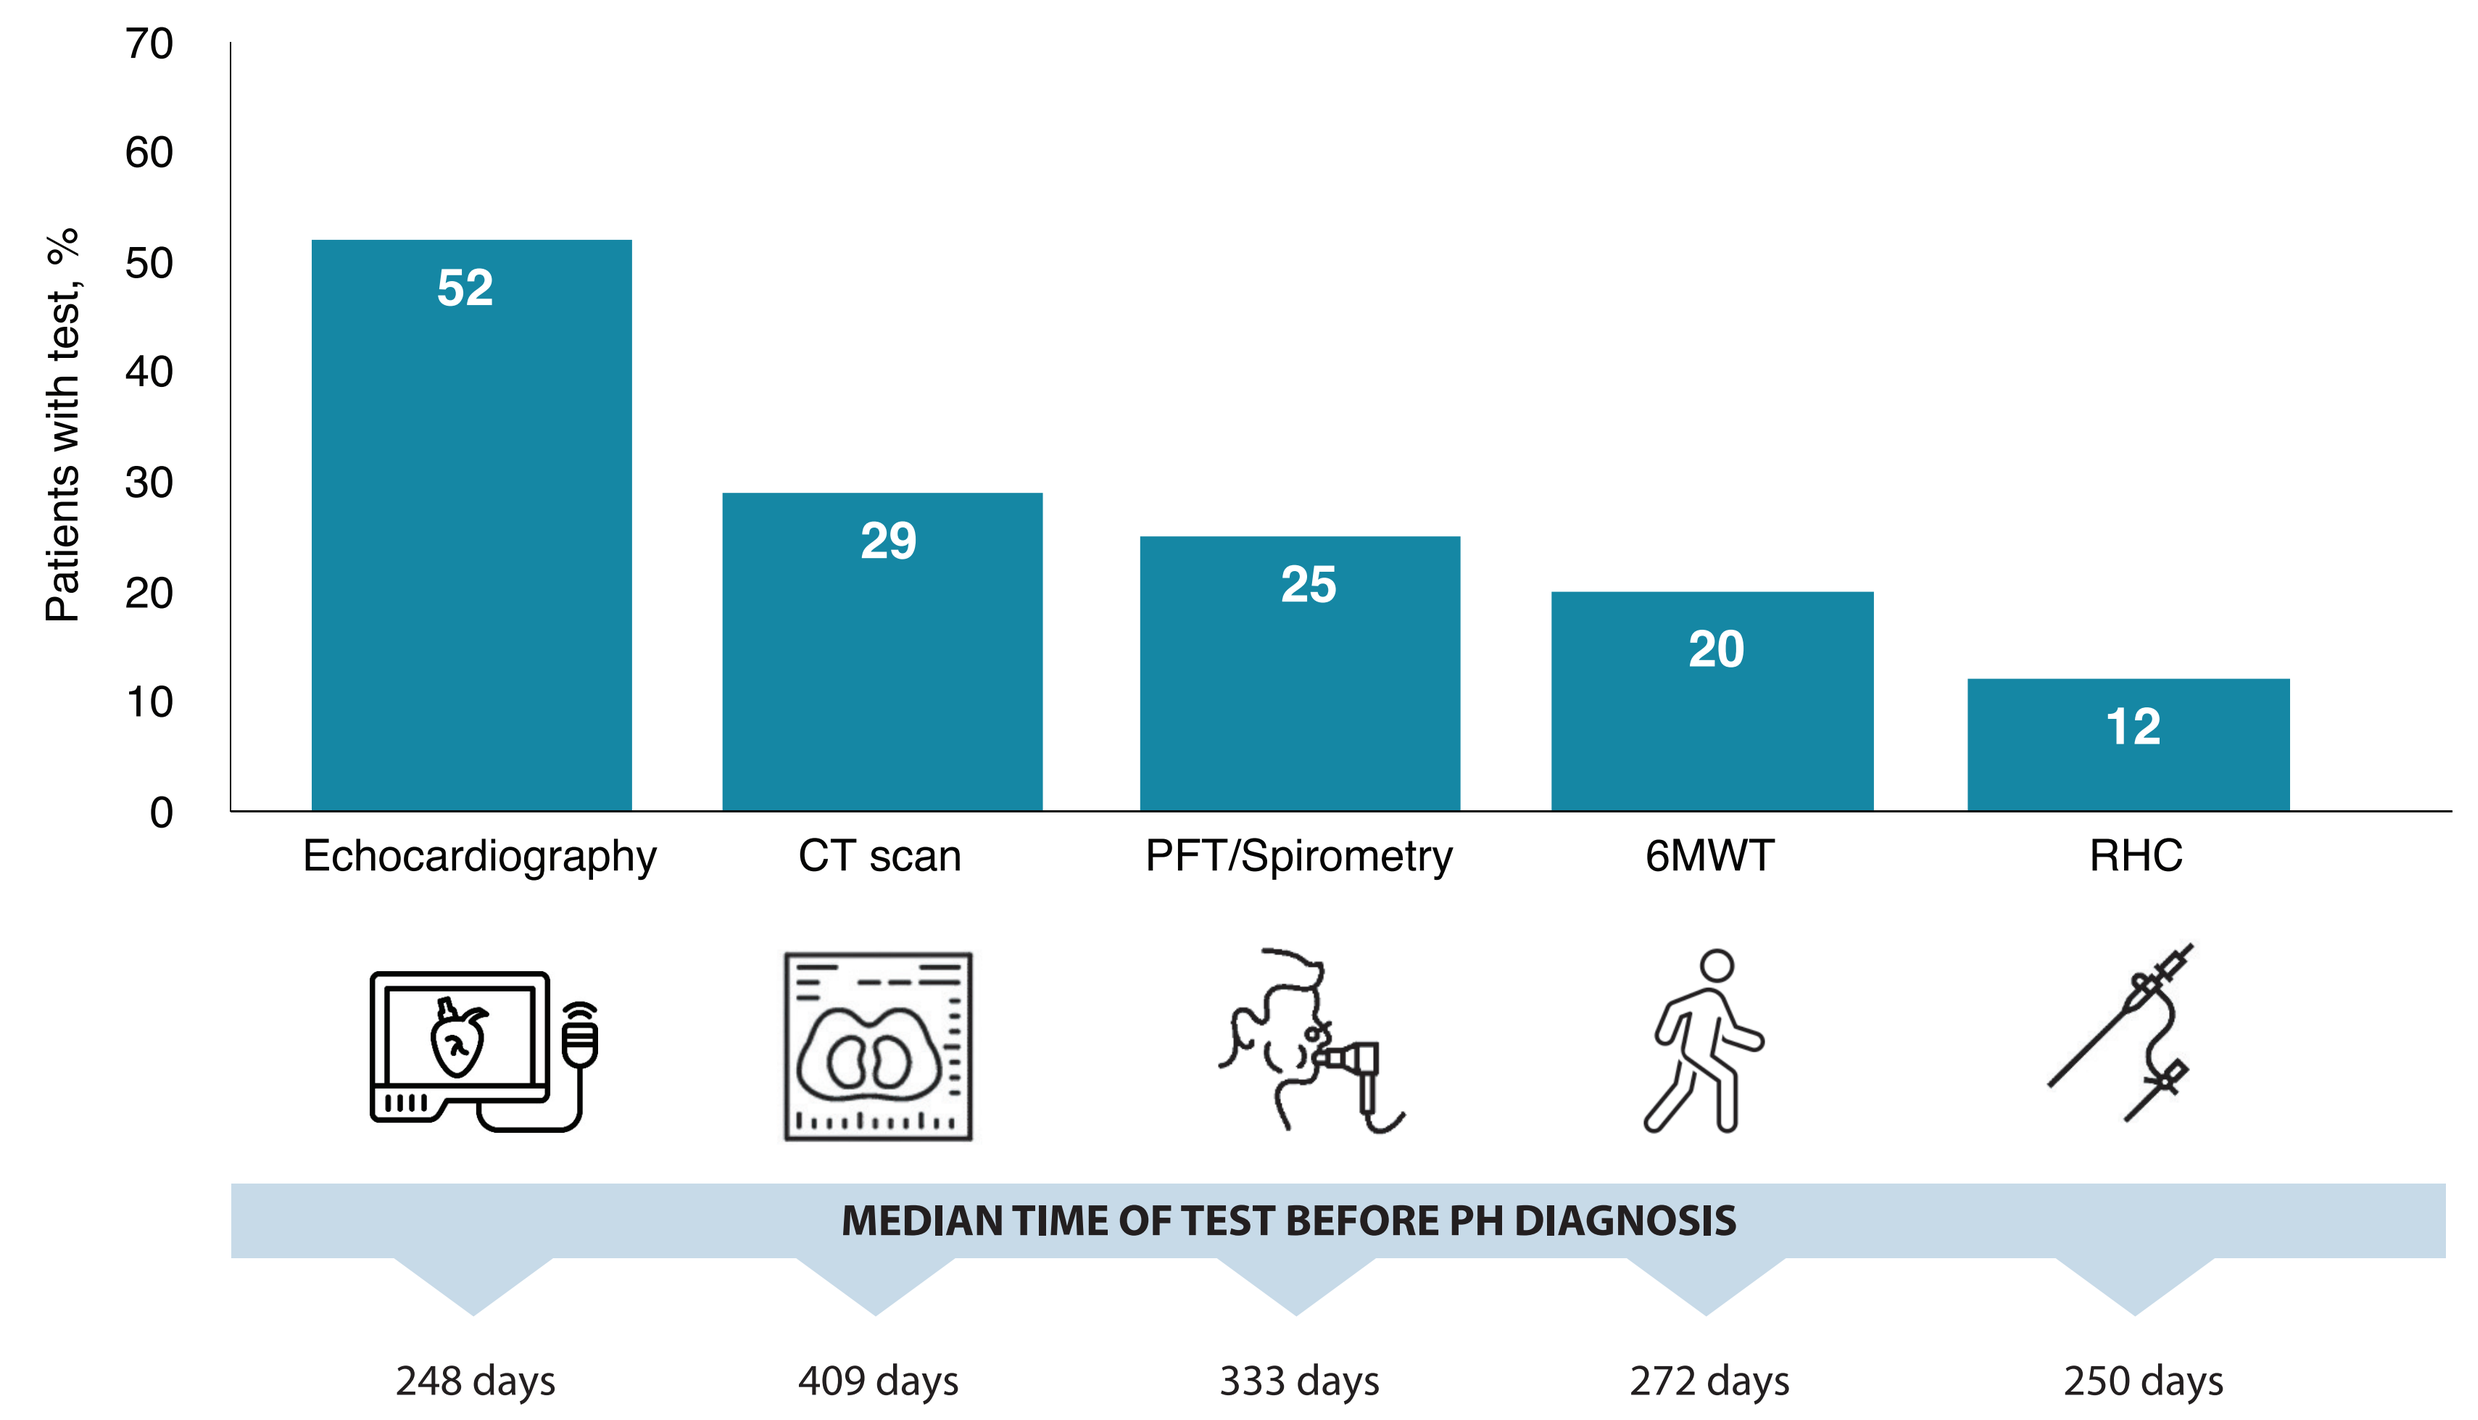
Patients with test, %
70
60
50
40
30
20
10
0
52
29
25
20
12
Echocardiography
CT scan
PFT/Spirometry
6MWT
RHC
MEDIAN TIME OF TEST BEFORE PH DIAGNOSIS
248 days
409 days
333 days
272 days
250 days

6MWT, six-minute walk test; CT, computed tomography; DLCO, diffusing capacity of the lungs for carbon monoxide; PFT, pulmonary function test; PH, pulmonary hypertension; RHC, right heart catheterization.

CONCLUSIONS

- This analysis offers insights into symptom presentation, physician specialties, and modalities used for PH-ILD diagnosis
- There was a delay of up to 1.6 years between initial symptom presentation and PH diagnosis, likely attributed to the overlapping symptomatology of PH and ILD
- A high percentage of diagnoses was made by non-PH specialists in acute and primary care settings, indicating potential additional delays in referral to appropriate specialty care centers
- Key diagnostic modalities—including RHC, PFTs, and 6MWT—remain underutilized and may represent missed opportunities for comprehensive evaluation and timely intervention in this high-risk population
- Opportunities exist to accelerate disease recognition and referral to reduce diagnostic delays and optimize outcomes for this high-risk population

References:
1 Nathan SD, et al. *J Manag Care Spec Pharm*. 2025;31(1-a suppl):S1-S18.
2 de Belen E, et al. *J Am Heart Assoc*. 2023;12(2):e026413.

Research supported by: Chiesi Farmaceutici S.p.A. and Gossamer Bio, Inc.