RUSH
UNIVERSITY MEDICAL CENTER

Modulation of Integrin CD11b as a Novel Therapeutic Strategy Against Lung Cancer

Terese Geraghty[1], Anugraha Rajagopalan[1], Rabail Aslam[1], and Vineet Gupta[1]

[1]Drug Discovery Center, Department of Internal Medicine, Rush University Medical Center, Chicago, IL

Abstract

A major problem facing cancer therapy is the immune suppression by the tumor and its subsequent escape from the immune response, which is mediated by the tumor microenvironment. Tumors have large numbers of myeloid derived suppressor cells (MDSCs), which suppress the adaptive immune response, increase neo-angiogenesis, and promote tumor survival. GB1275 is a novel small molecule CD11b allosteric modulator. In pancreatic cancer models, GB1275 reduced MDSC infiltration and M2 macrophage polarization leading to tumor growth inhibition. GB1275 is currently in Phase 1/2 clinical development in multiple cancer types. To investigate the impact of CD11b modulation in the context of lung cancer, GB1275 treatment effects on tumor growth and immune infiltrates were assessed in the Lewis Lung Carcinoma (LLC) syngeneic tumor model, propagated in C57BL/6 wild type (WT) mice. Additionally, we developed a transgenic CD11b knock-in (KI) mouse that renders CD11b in a partially active state. CD11b KI mice were engineered to express partially active CD11b by introducing a point mutation at position 332 causing an isoleucine to glycine switch (I332G). We studied LLC tumor growth in CD11b KI mice as an orthogonal approach to test the mechanism of action. Pharmacologic modulation of CD11b with GB1275 significantly reduced LLC tumor growth. Additionally, CD11b KI mice that showed a significant reduction in both the size and the rate of LLC tumor growth, as compared to the WT mice, mimicking treatment effects with GB1275. We found that CD11b modulation by GB2175 is T cell-dependent. Pharmacologic or genetic CD11b modulation significantly reduced CD11b⁺ myeloid cell recruitment, resulted in a phenotypic switch in innate and adaptive immune cells in lung tumors to enhance the adaptive immune response and is a novel strategy against lung cancer.

Background

- Lung cancer is the leading cause of cancer-related deaths in the United States. Of the new cancer diagnoses each year, 13% are lung cancers. With an overall 5-year survival rate at 19%, lung cancer patients are sorely in need of new therapeutic options. Non-small cell lung carcinoma (NSCLC) accounts for 85% of all lung cancers.
- Integrin heterodimer CD11b/CD18 (aka Mac-1 or αMβ2) is highly expressed on myeloid cells and plays an important role in recruitment and other biological functions.

Lewis Lung Carcinoma Model

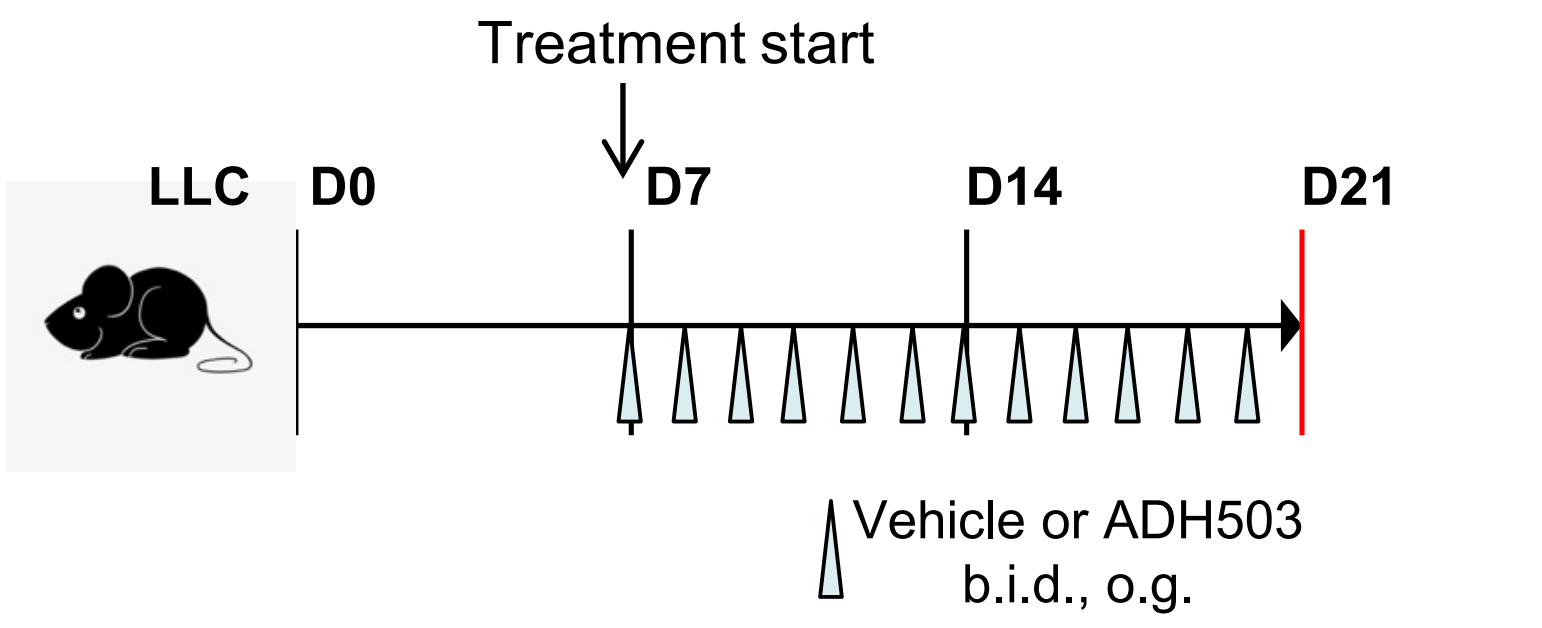
Treatment start
LLC
D0
D7
D14
D21
Vehicle or ADH503
b.i.d., o.g.

Results

CD11b⁺ cells are abundant in human NSCLC tissues

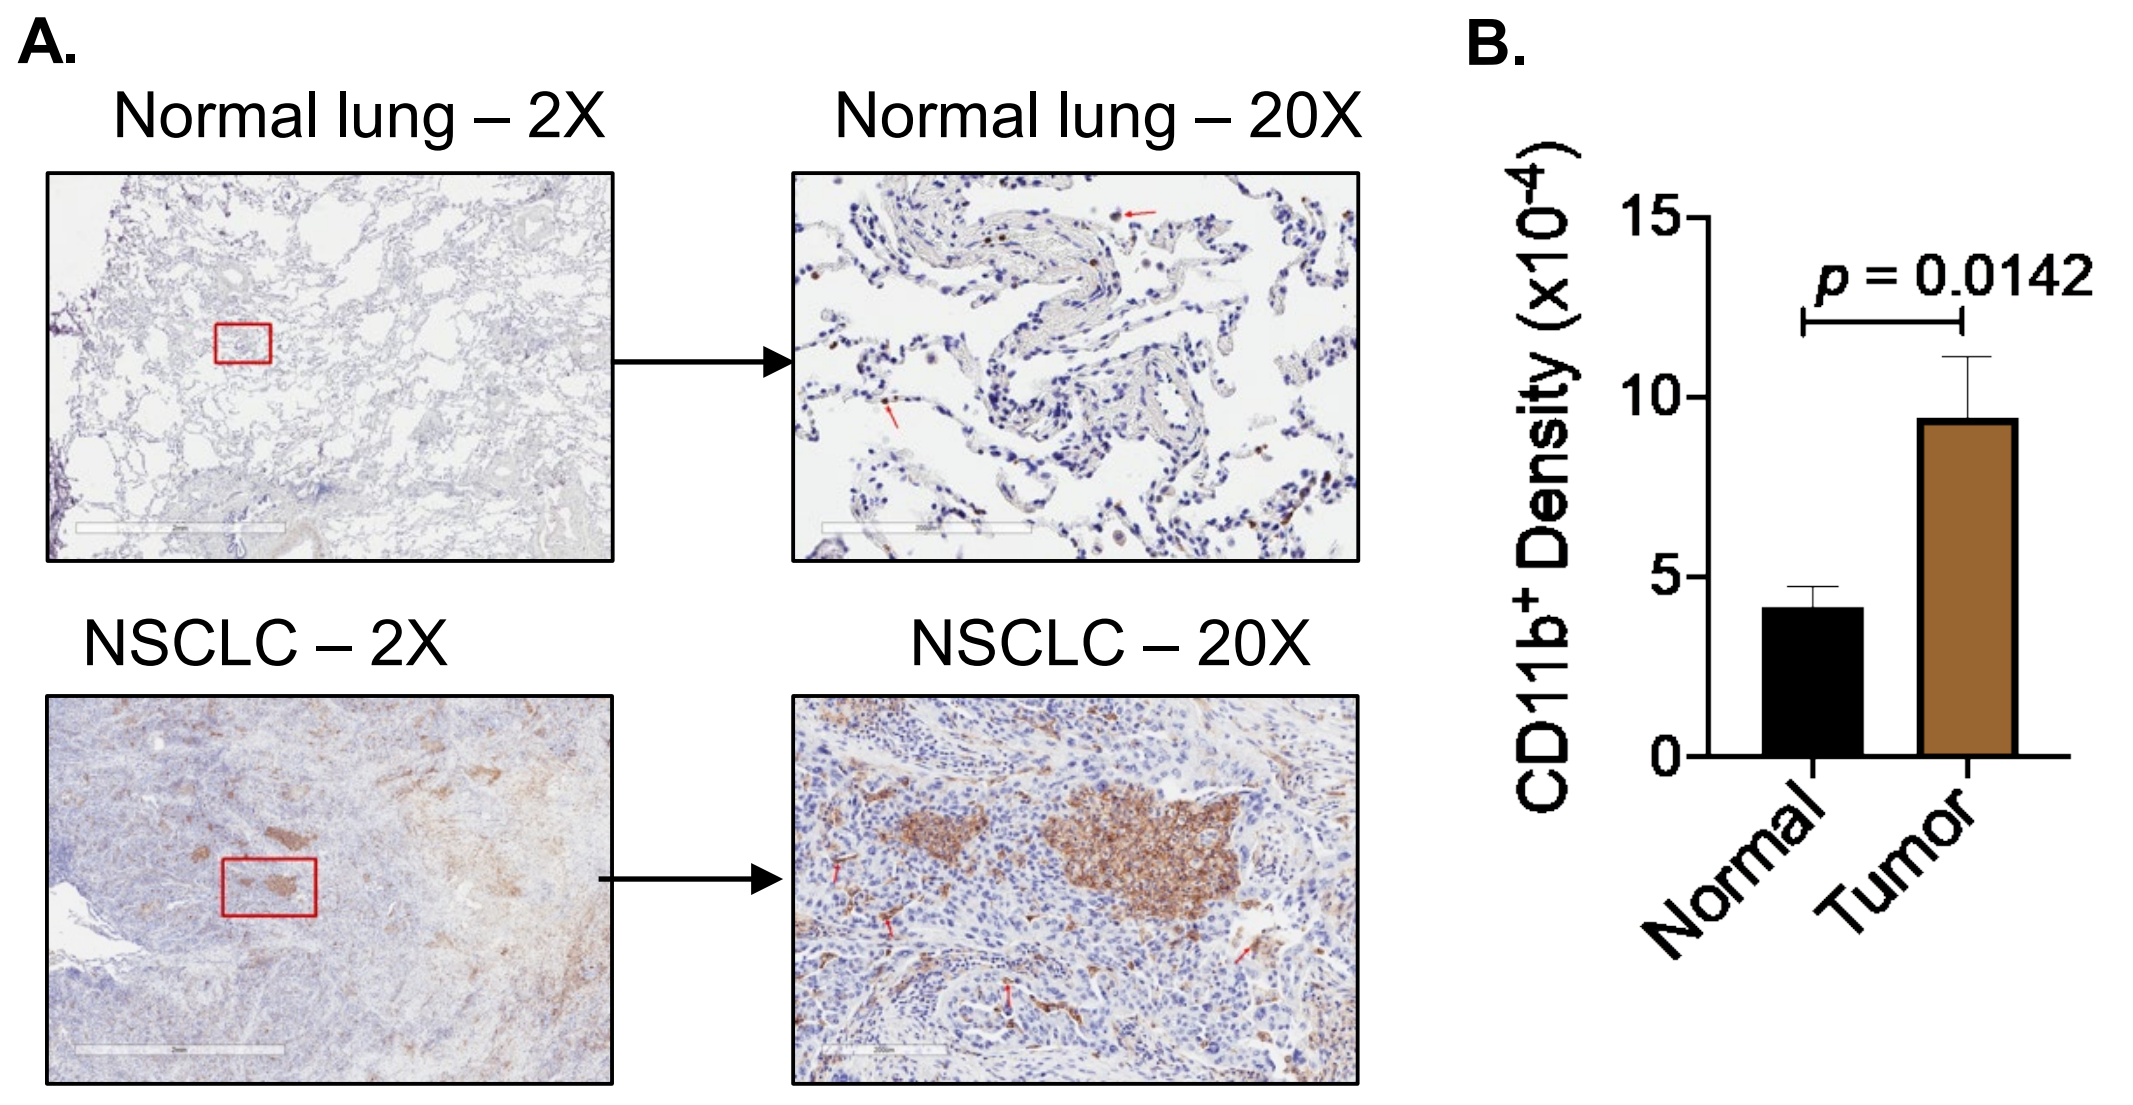
A.
Normal lung – 2X
Normal lung – 20X
NSCLC – 2X
NSCLC – 20X
B.
CD11b⁺ Density (x10⁻⁴)
p = 0.0142
Normal
Tumor

Pharmacologic CD11b modulation delays tumor growth

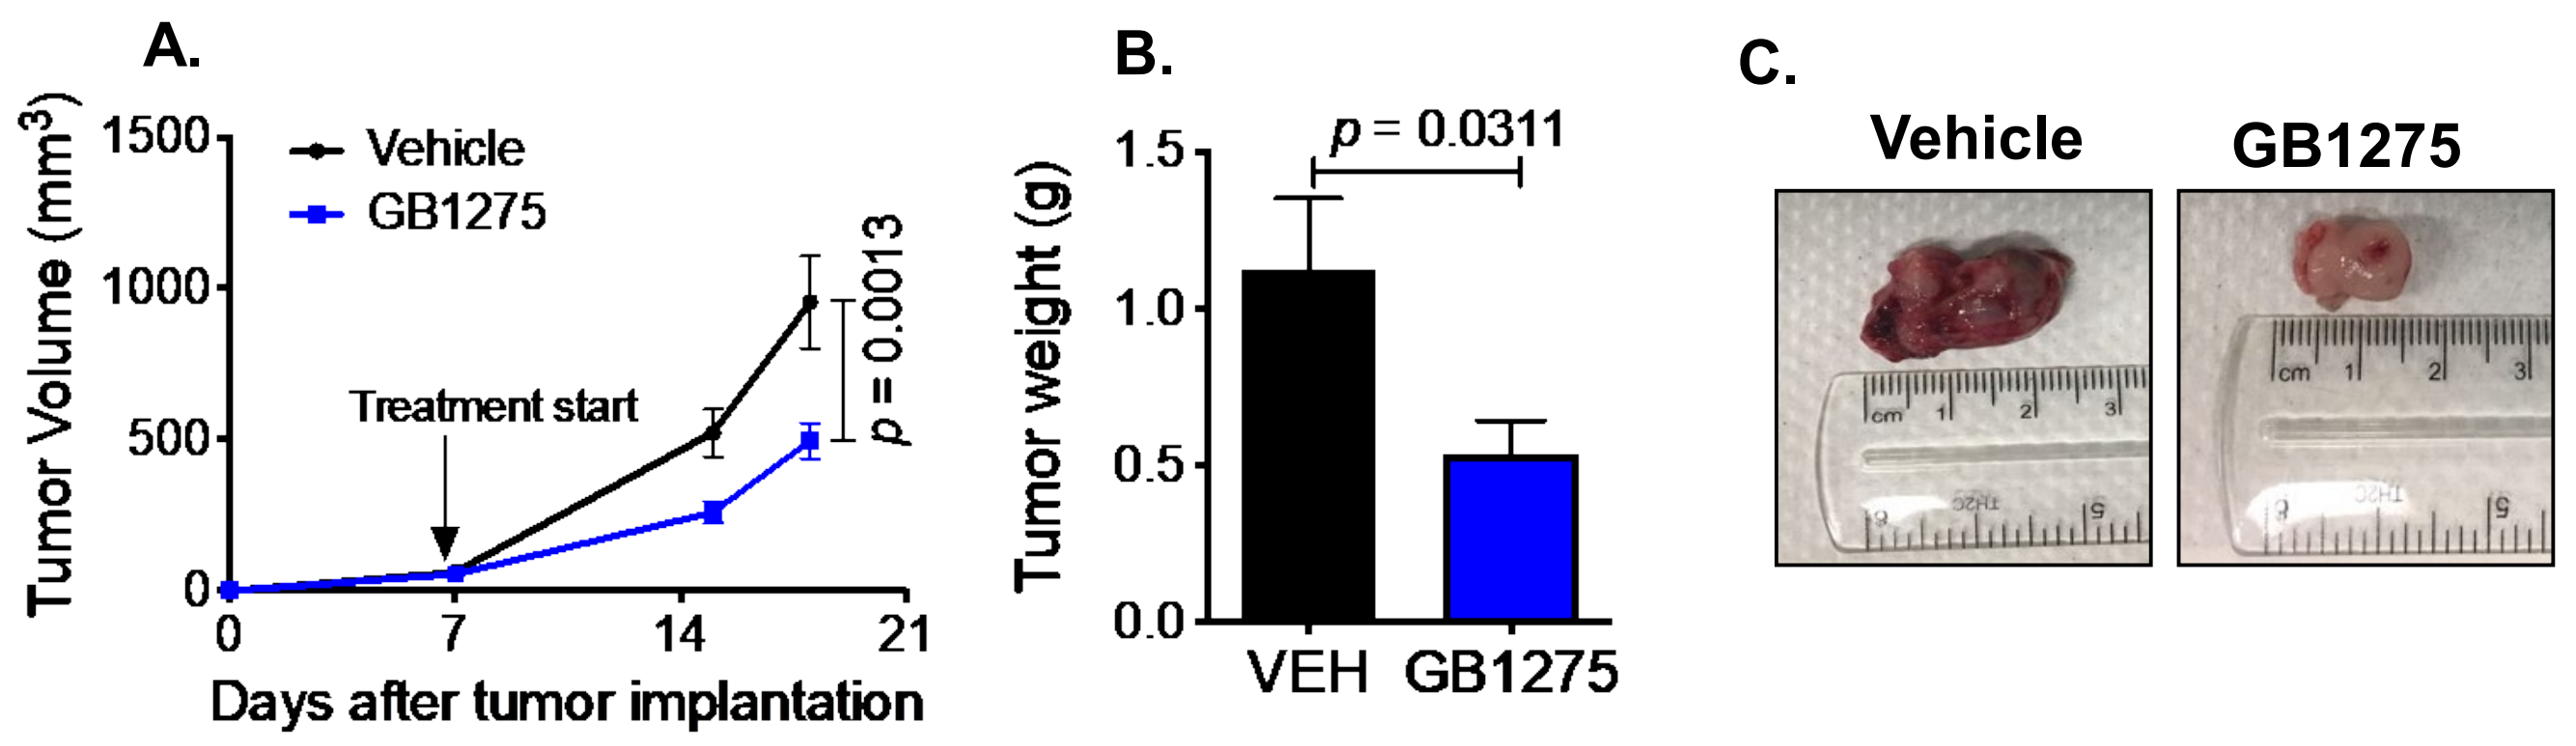
A.
Vehicle
GB1275
Tumor Volume (mm³)
Treatment start
p = 0.0013
Days after tumor implantation
B.
Tumor weight (g)
p = 0.0311
VEH
GB1275
C.
Vehicle
GB1275

Genetic CD11b modulation in CD11b knock-in mice showed reduced tumor growth

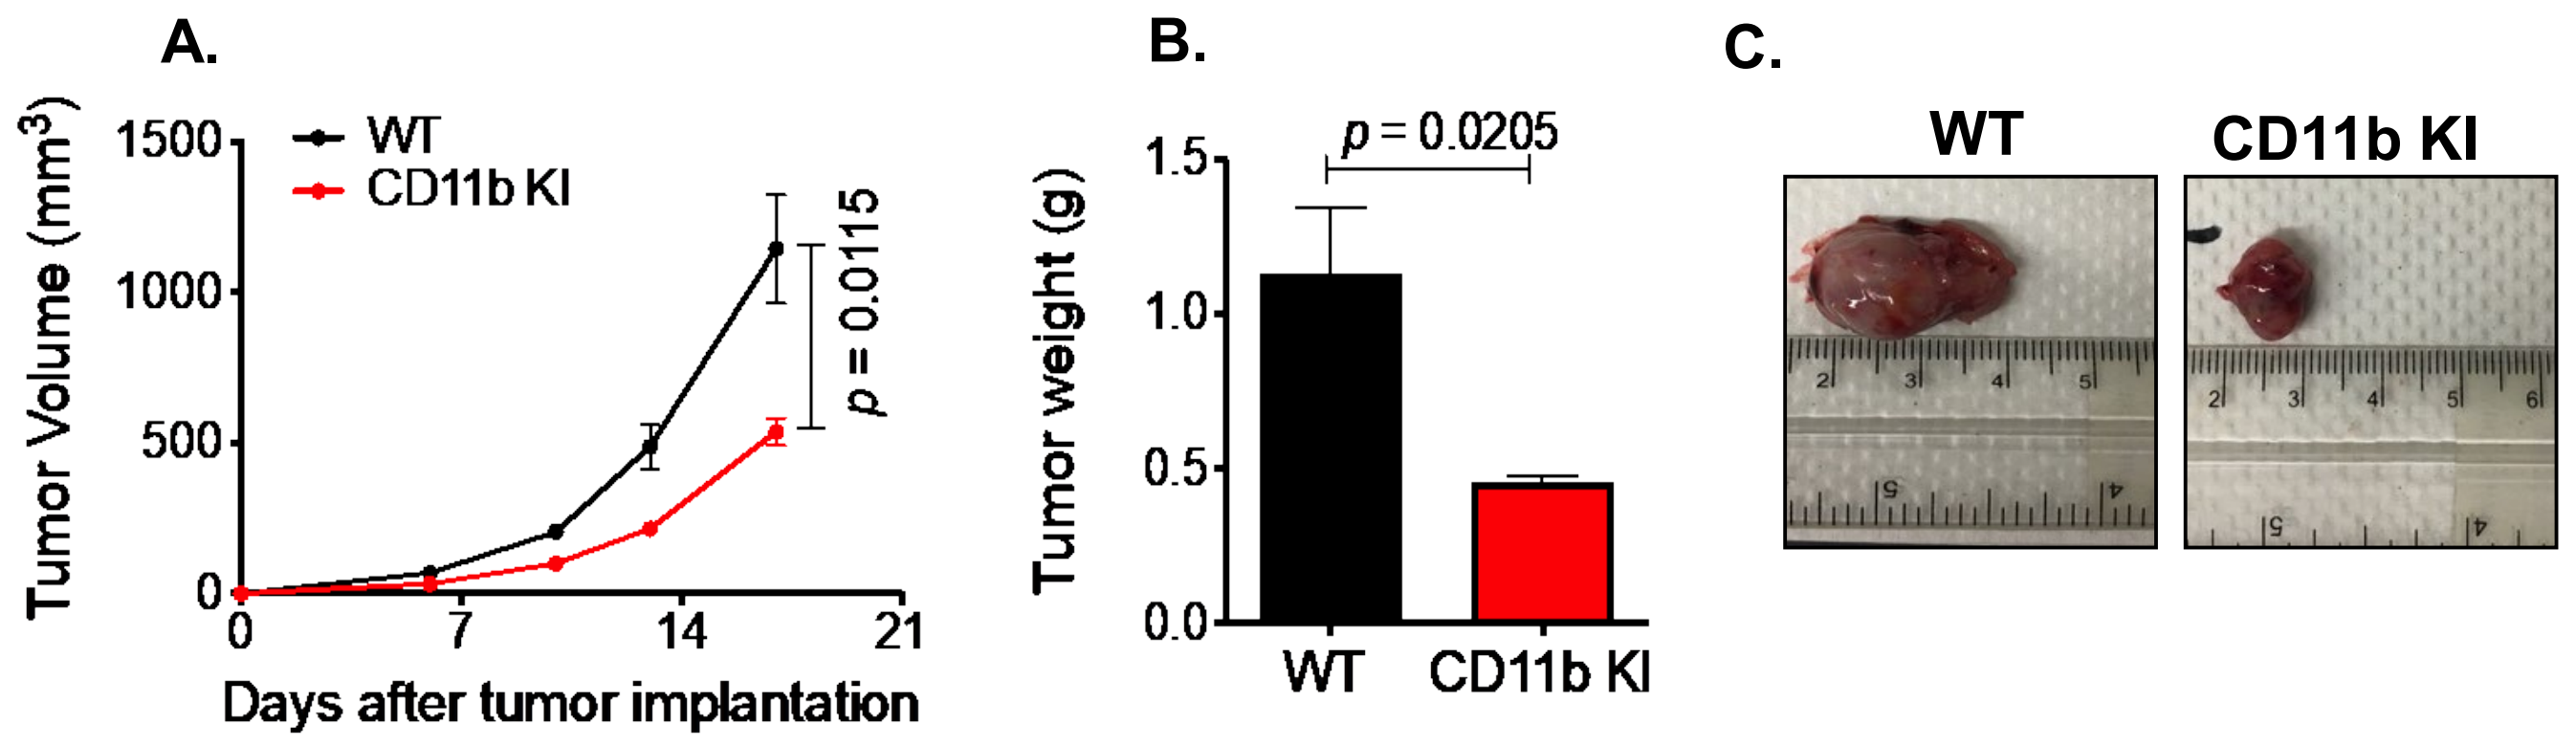
A.
WT
CD11b KI
Tumor Volume (mm³)
p = 0.0115
Days after tumor implantation
B.
Tumor weight (g)
p = 0.0205
WT
CD11b KI
C.
WT
CD11b KI

Tumor growth inhibition mediated by CD11b modulation is T cell-dependent

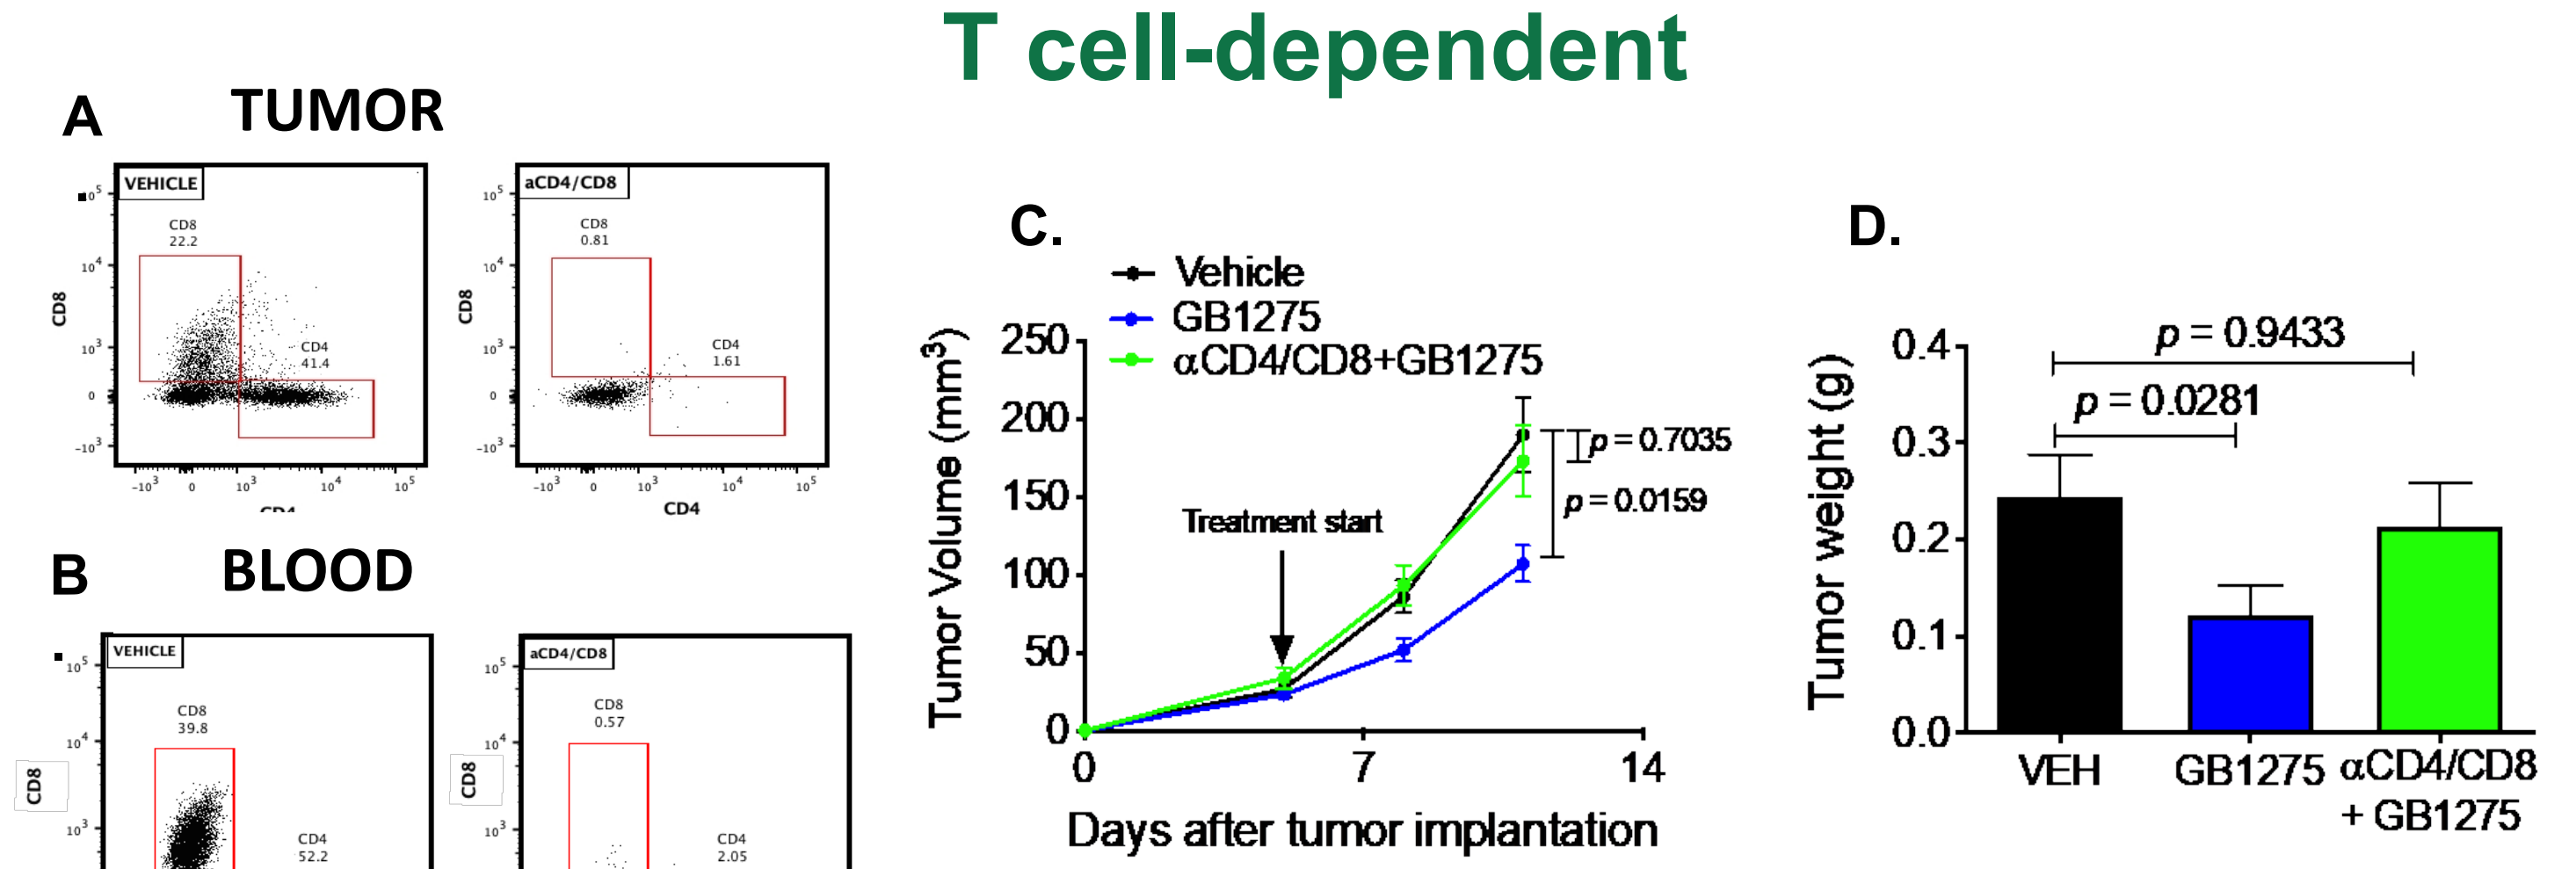
A
TUMOR
B
BLOOD
VEHICLE
aCD4/CD8
CD8
CD4
C.
Vehicle
GB1275
αCD4/CD8+GB1275
Tumor Volume (mm³)
Treatment start
p = 0.7035
p = 0.0159
Days after tumor implantation
D.
Tumor weight (g)
p = 0.9433
p = 0.0281
VEH
GB1275
αCD4/CD8 + GB1275

CD11b modulation reduced neo-angiogenesis in LLC tumors

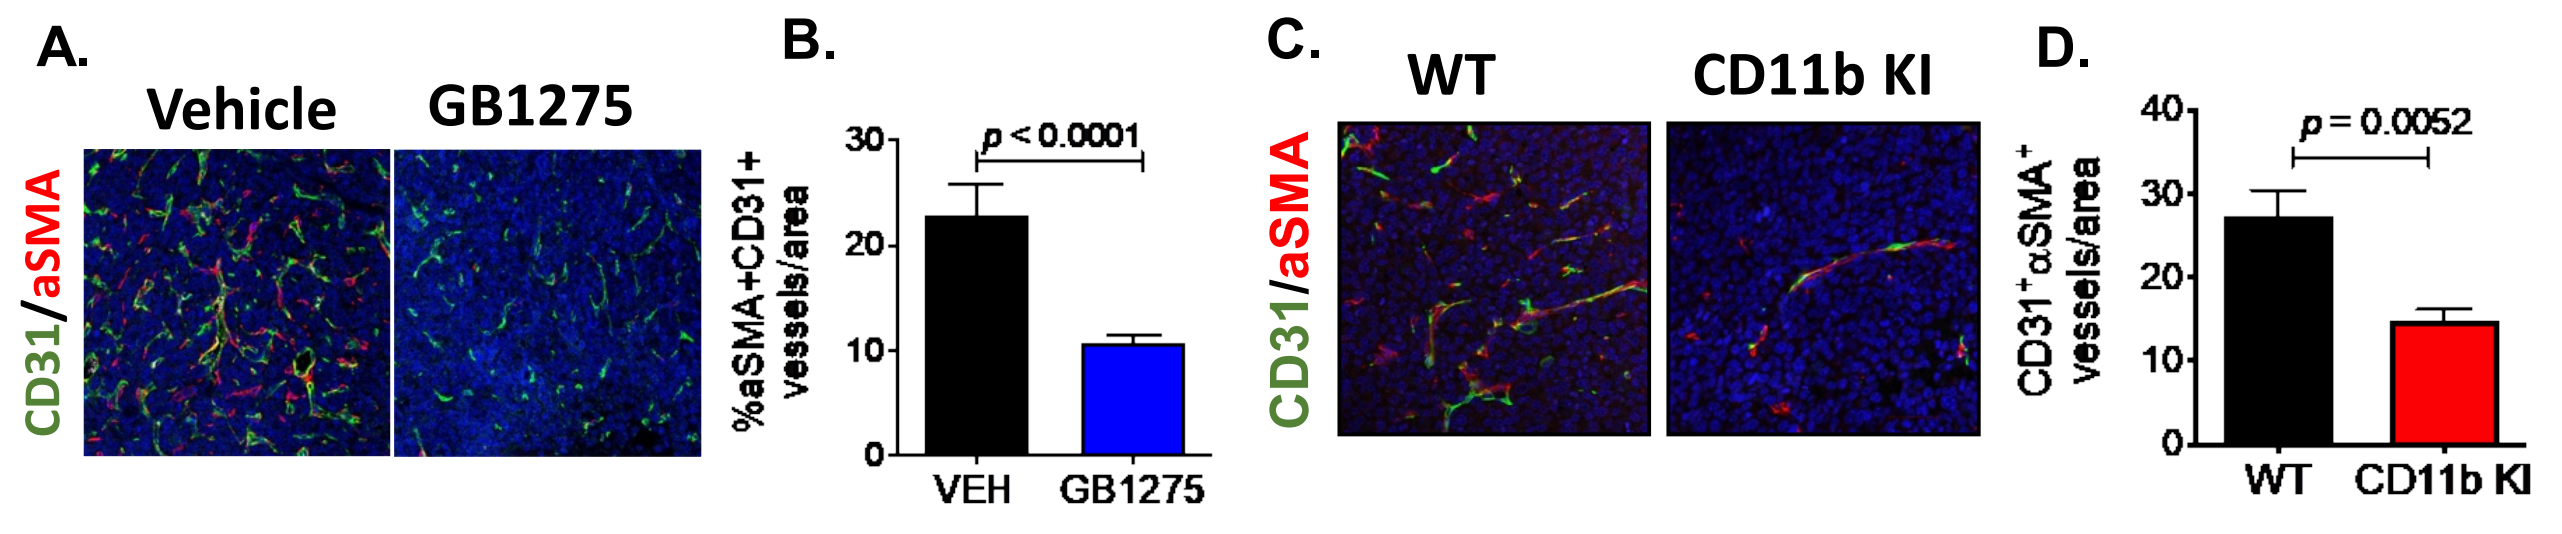
A.
Vehicle
GB1275
CD31/aSMA
B.
%aSMA+CD31+ vessels/area
p < 0.0001
VEH
GB1275
C.
WT
CD11b KI
CD31/aSMA
D.
CD31⁺αSMA⁺ vessels/area
p = 0.0052
WT
CD11b KI

CD11b modulation reduces myeloid cell recruitment

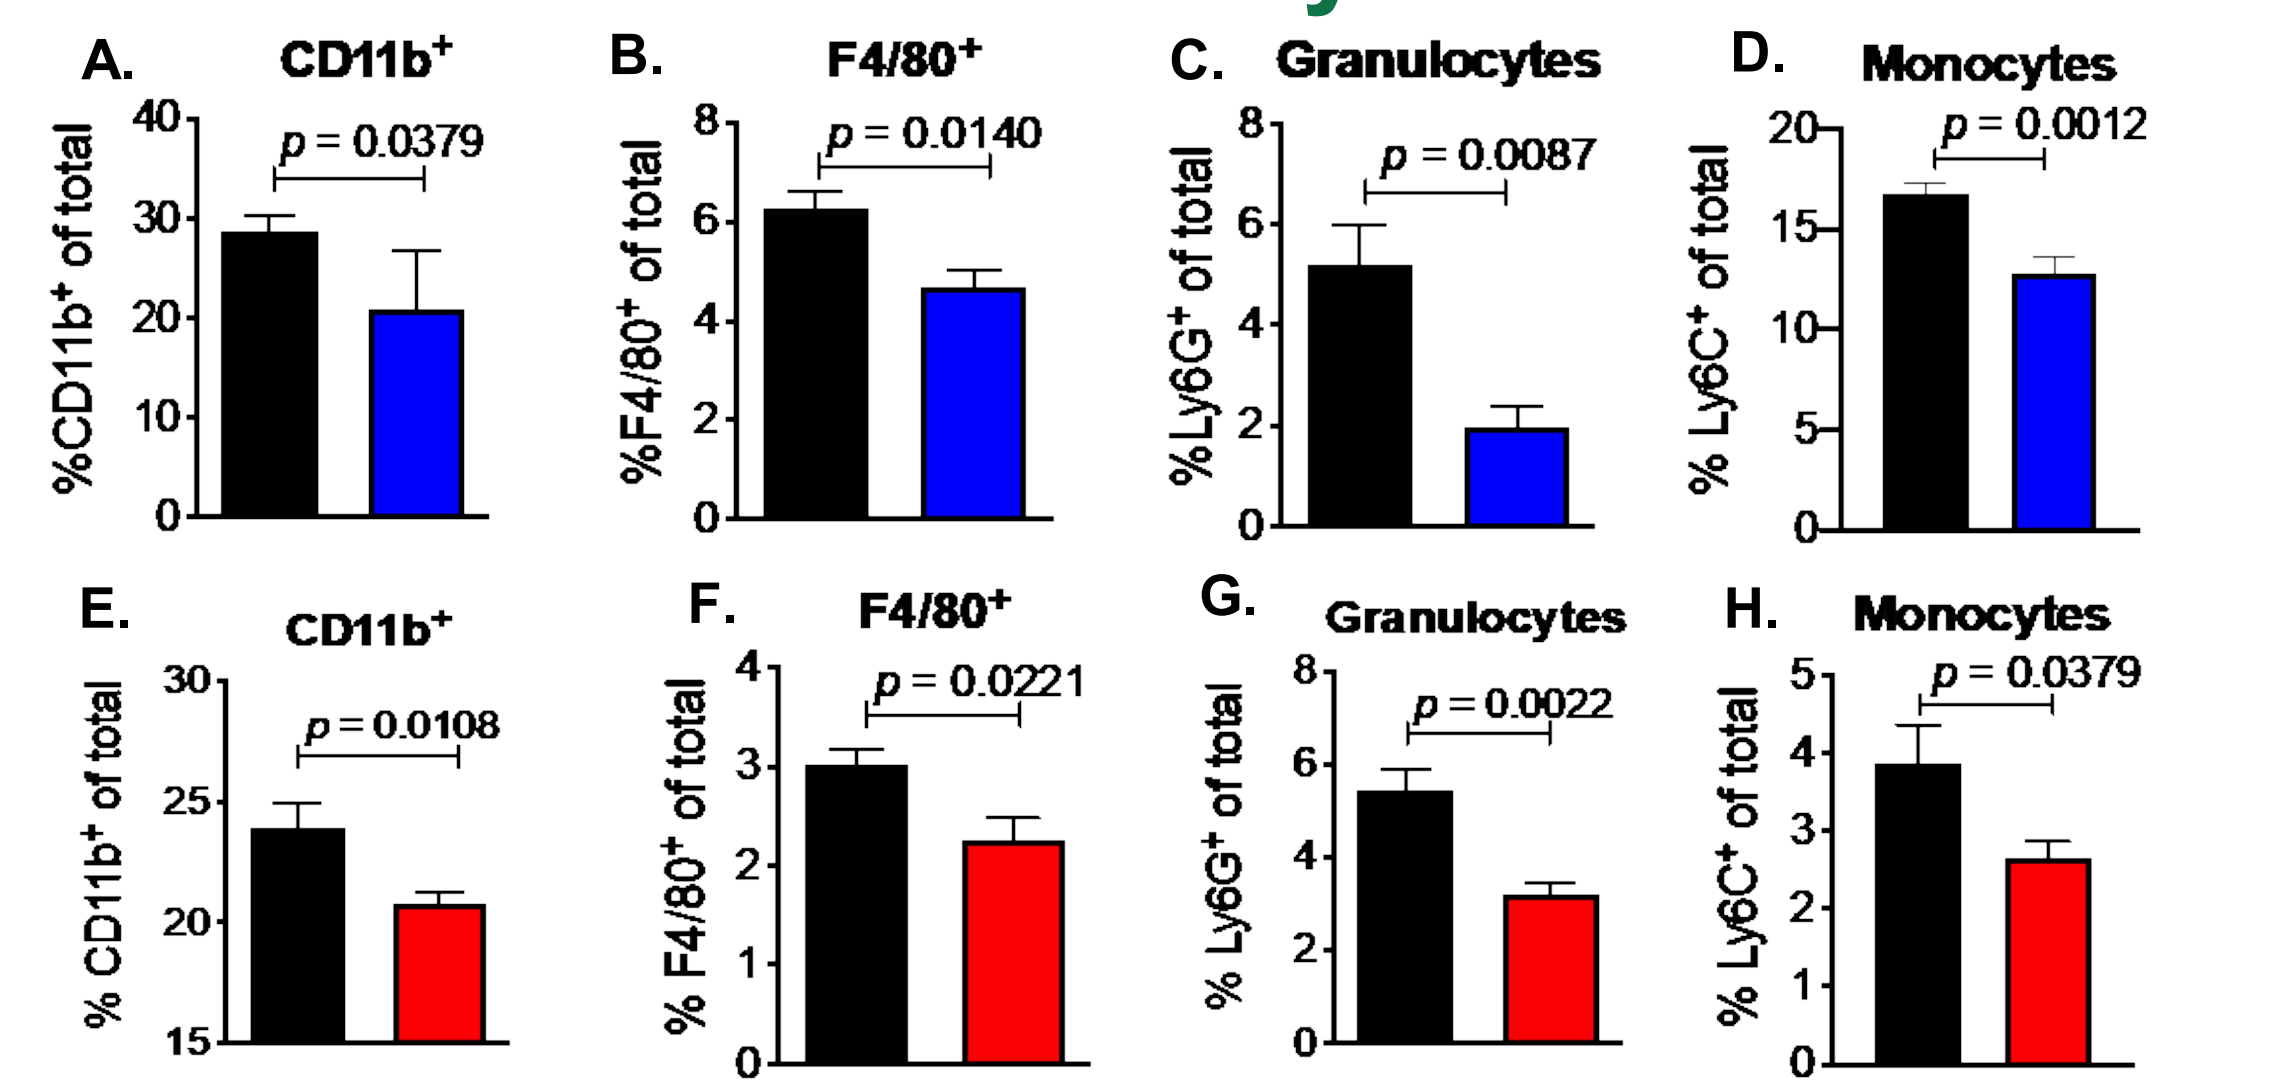
A. CD11b⁺
%CD11b⁺ of total
p = 0.0379
B. F4/80⁺
%F4/80⁺ of total
p = 0.0140
C. Granulocytes
%Ly6G⁺ of total
p = 0.0087
D. Monocytes
% Ly6C⁺ of total
p = 0.0012
E. CD11b⁺
% CD11b⁺ of total
p = 0.0108
F. F4/80⁺
% F4/80⁺ of total
p = 0.0221
G. Granulocytes
% Ly6G⁺ of total
p = 0.0022
H. Monocytes
% Ly6C⁺ of total
p = 0.0379

CD11b modulation stimulates M1 polarization

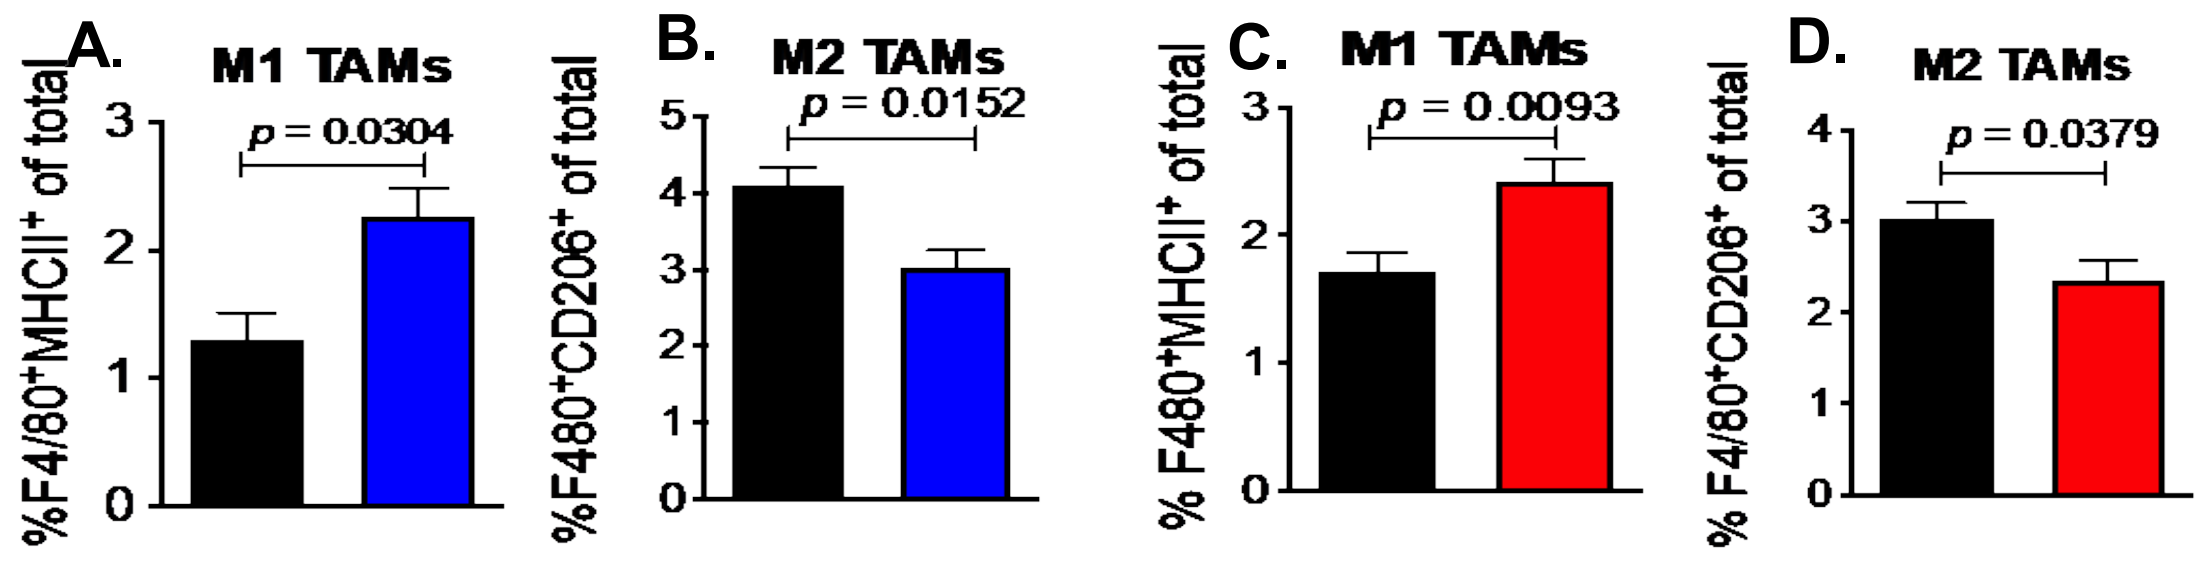
A. M1 TAMs
%F4/80⁺MHCII⁺ of total
p = 0.0304
B. M2 TAMs
%F480⁺CD206⁺ of total
p = 0.0152
C. M1 TAMs
% F480⁺MHCII⁺ of total
p = 0.0093
D. M2 TAMs
% F4/80⁺CD206⁺ of total
p = 0.0379

Phenotypic switch in dendritic cells after CD11b modulation

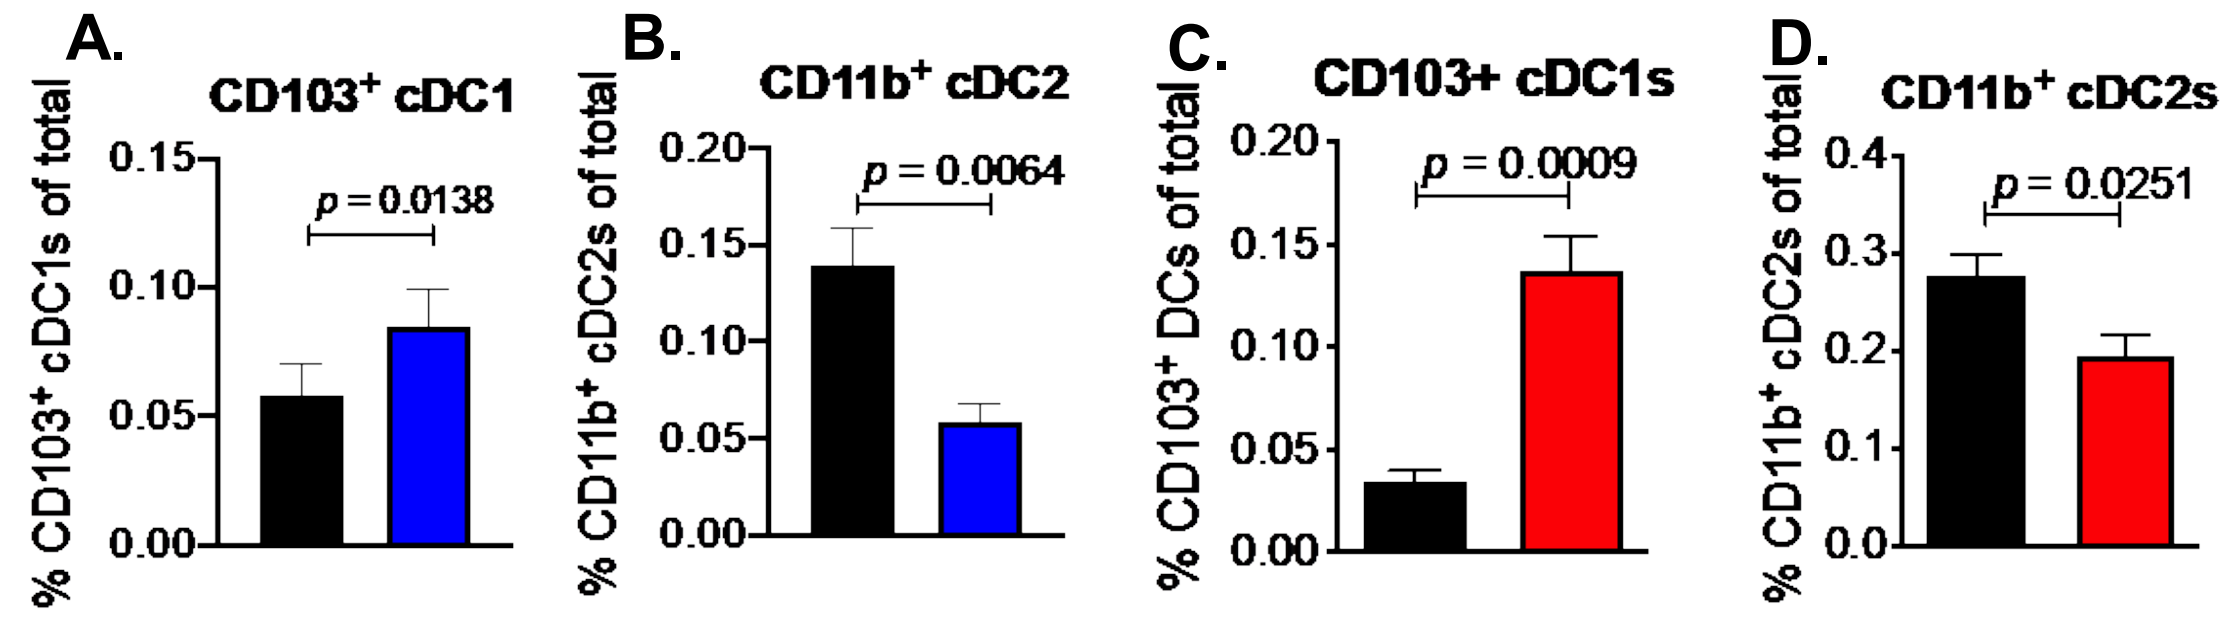
A. CD103⁺ cDC1
% CD103⁺ cDC1s of total
p = 0.0138
B. CD11b⁺ cDC2
% CD11b⁺ cDC2s of total
p = 0.0064
C. CD103+ cDC1s
% CD103⁺ DCs of total
p = 0.0009
D. CD11b⁺ cDC2s
% CD11b⁺ cDC2s of total
p = 0.0251

CD11b modulation reduces T cell immune suppression

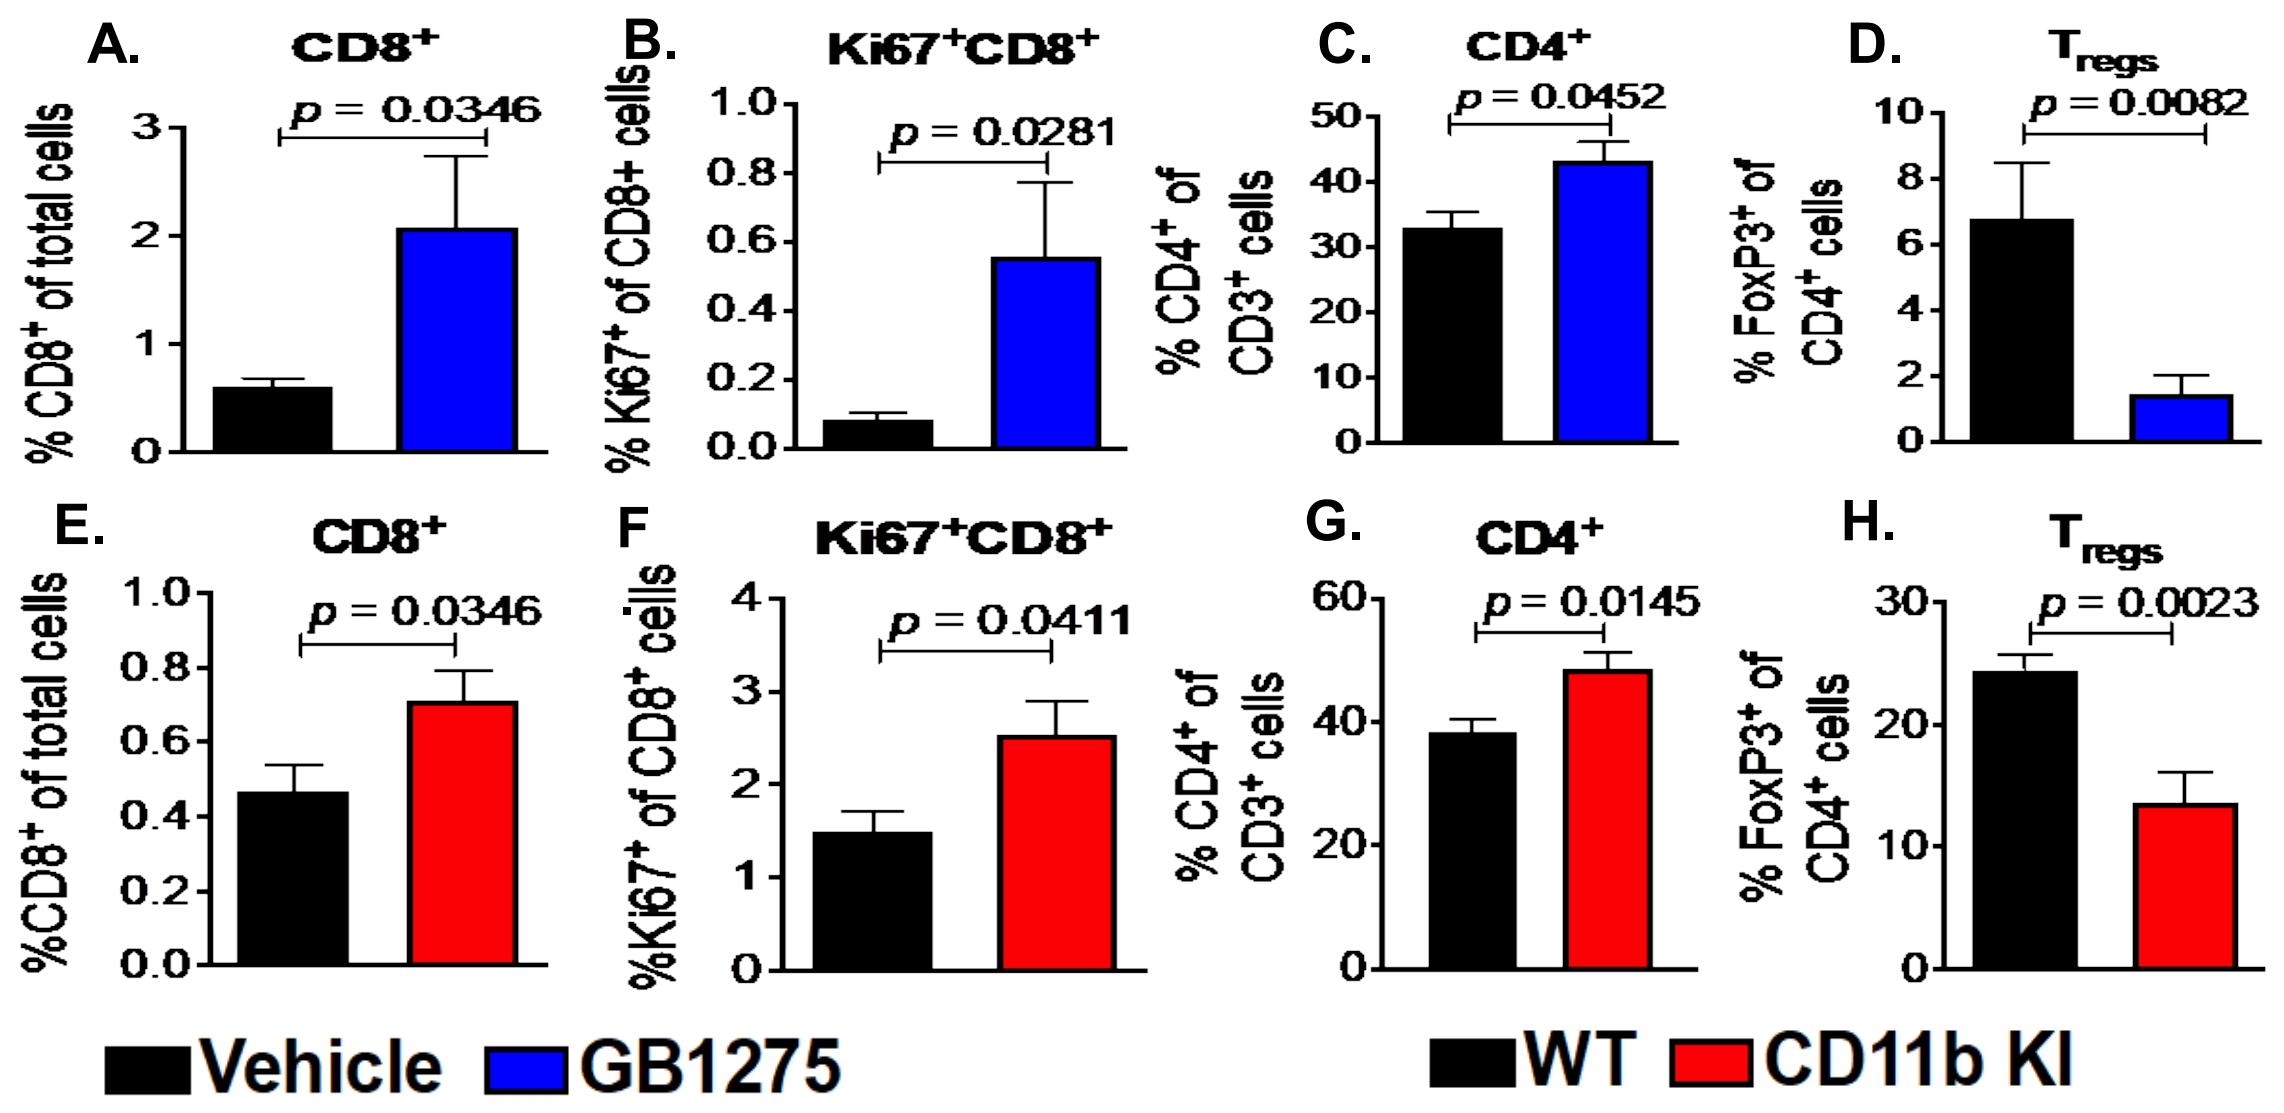
A. CD8⁺
% CD8⁺ of total cells
p = 0.0346
B. Ki67⁺CD8⁺
% Ki67⁺ of CD8+ cells
p = 0.0281
C. CD4⁺
% CD4⁺ of CD3⁺ cells
p = 0.0452
D. Tregs
% FoxP3⁺ of CD4⁺ cells
p = 0.0082
E. CD8⁺
%CD8⁺ of total cells
p = 0.0346
F Ki67⁺CD8⁺
%Ki67⁺ of CD8⁺ cells
p = 0.0411
G. CD4⁺
% CD4⁺ of CD3⁺ cells
p = 0.0145
H. Tregs
% FoxP3⁺ of CD4⁺ cells
p = 0.0023
Vehicle
GB1275
WT
CD11b KI

Conclusions

- CD11b modulation works to slow LLC tumor growth by reducing myeloid cell recruitment, decreasing neo-angiogenesis, and by significantly changing tumor immune profiles.
- Our findings suggest a novel therapeutic strategy against lung cancer via pharmacologic CD11b modulation

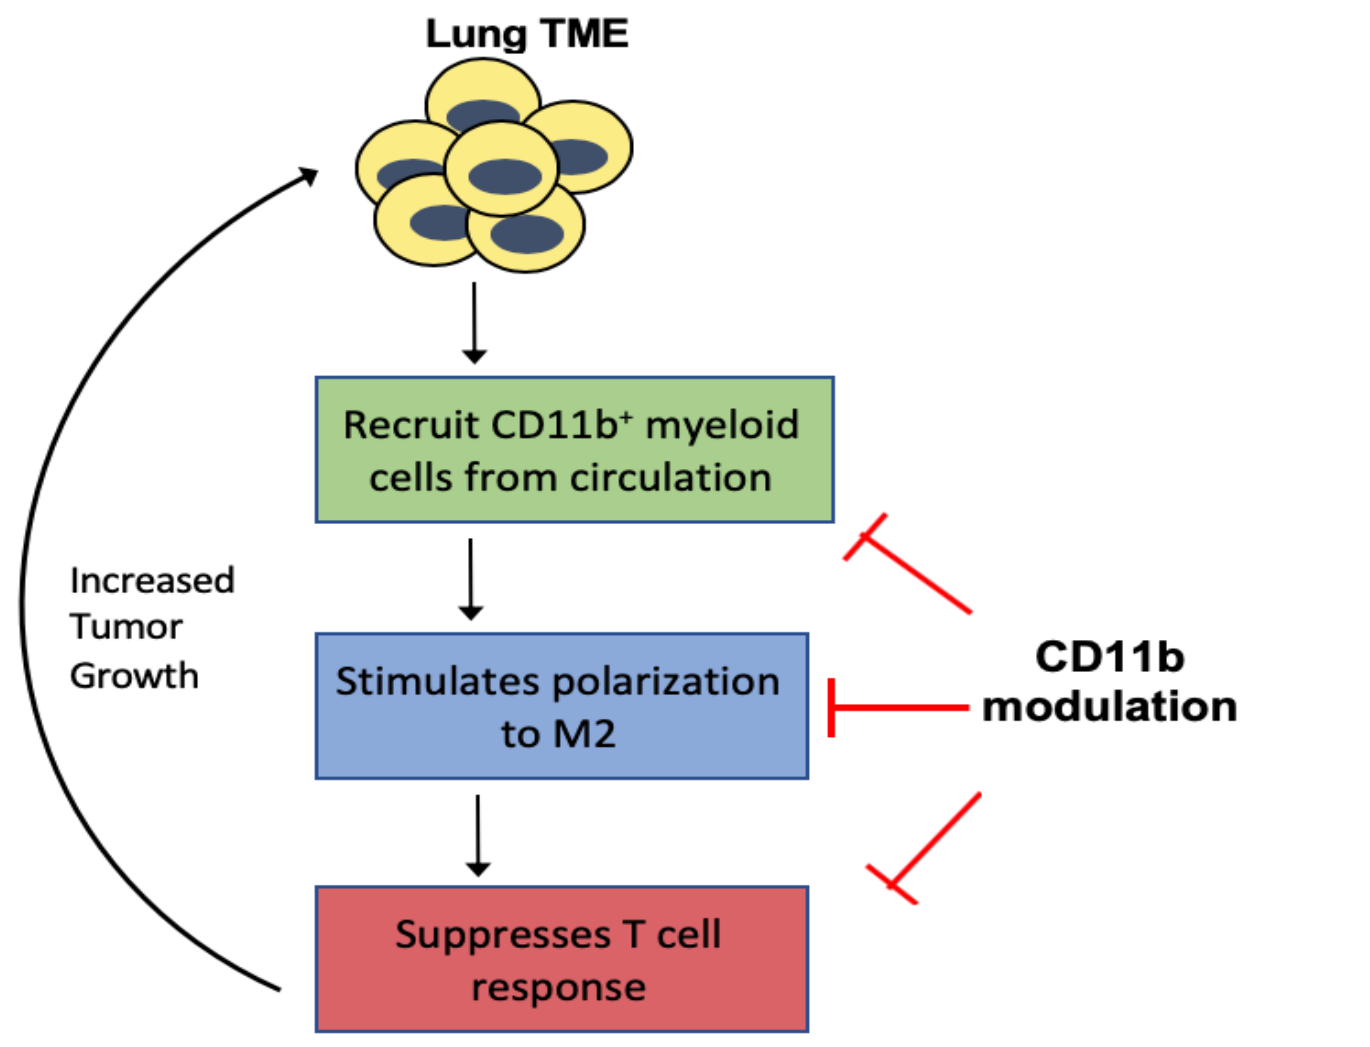
Lung TME
Recruit CD11b⁺ myeloid cells from circulation
Stimulates polarization to M2
Suppresses T cell response
Increased Tumor Growth
CD11b modulation