A phase 1/2 study of GB1275, a novel CD11b modulator, as monotherapy and with an anti-PD-1 antibody in specified advanced solid tumors or with chemotherapy in metastatic pancreatic cancer (mPDAC)

P419

Johanna C. Bendell[1], Drew W. Rasco[2], Wungki Park[3], Lei Zhou[4], Anna Galkin[4], Debbie Slee[4], Laura L. Carter[4], David Nickle[4], Rebecca Tran[4], Jack Li[4], Beatrice Ferguson[4], Jakob Dupont[4], Vineet Gupta[5], and Eileen M. O'Reilly[3]

[1]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN; [2]The START Center for Cancer Care, San Antonio, TX, USA; [3]Memorial Sloan Kettering Cancer Center, New York, NY; [4]Gossamer Bio, San Diego, CA; [5]Rush University Medical Center, Chicago, IL

INTRODUCTION

- Tumor influx of CD11b-expressing myeloid-derived suppressor cells (MDSCs) and M2 tumor-associated macrophages (TAMs) creates an immunosuppressive tumor microenvironment associated with resistance to anti-PD-1 antibody therapy[1-3]
- GB1275 is a first-in-class, CD11b modulator that reduced MDSCs and TAMs at the tumor site, repolarized M2 immunosuppressive TAMs towards an M1 phenotype, and thus increased tumor infiltration of activated CD8+ T cells in vivo[4]
- Preclinical anti-tumor activity was seen with GB1275 as a single agent and synergistically in combination with chemotherapy and immuno-oncology therapies[4]
- GB1275 is being tested in a first-in-human Phase 1/2 study as monotherapy and in combination with an anti-PD1 antibody or standard of care (SOC) chemotherapy in tumor types that are known to be immuno-oncology resistant
- This trial is in progress (NCT04060342)

BACKGROUND

- CD11b/CD18, is a functional integrin on cell surfaces, highly expressed on leukocytes, including neutrophils, monocytes and macrophages
- GB1275 binds to an allosteric pocket in the αA-domain of CD11b, which stabilizes CD11b in a conformation required for binding to its ligands (**Figure 1**)[3]
- GB1275 enhanced CD11b-mediated adhesion and reduced transendothelial myeloid cell migration (**Figure 2)**[5]
- In preclinical models, GB1275 alleviated myeloid cell-mediated immunosuppressive effects and enhanced SOC and checkpoint blockade treatment outcomes[4]

Figure 1. GB1275 mechanism of action[3]

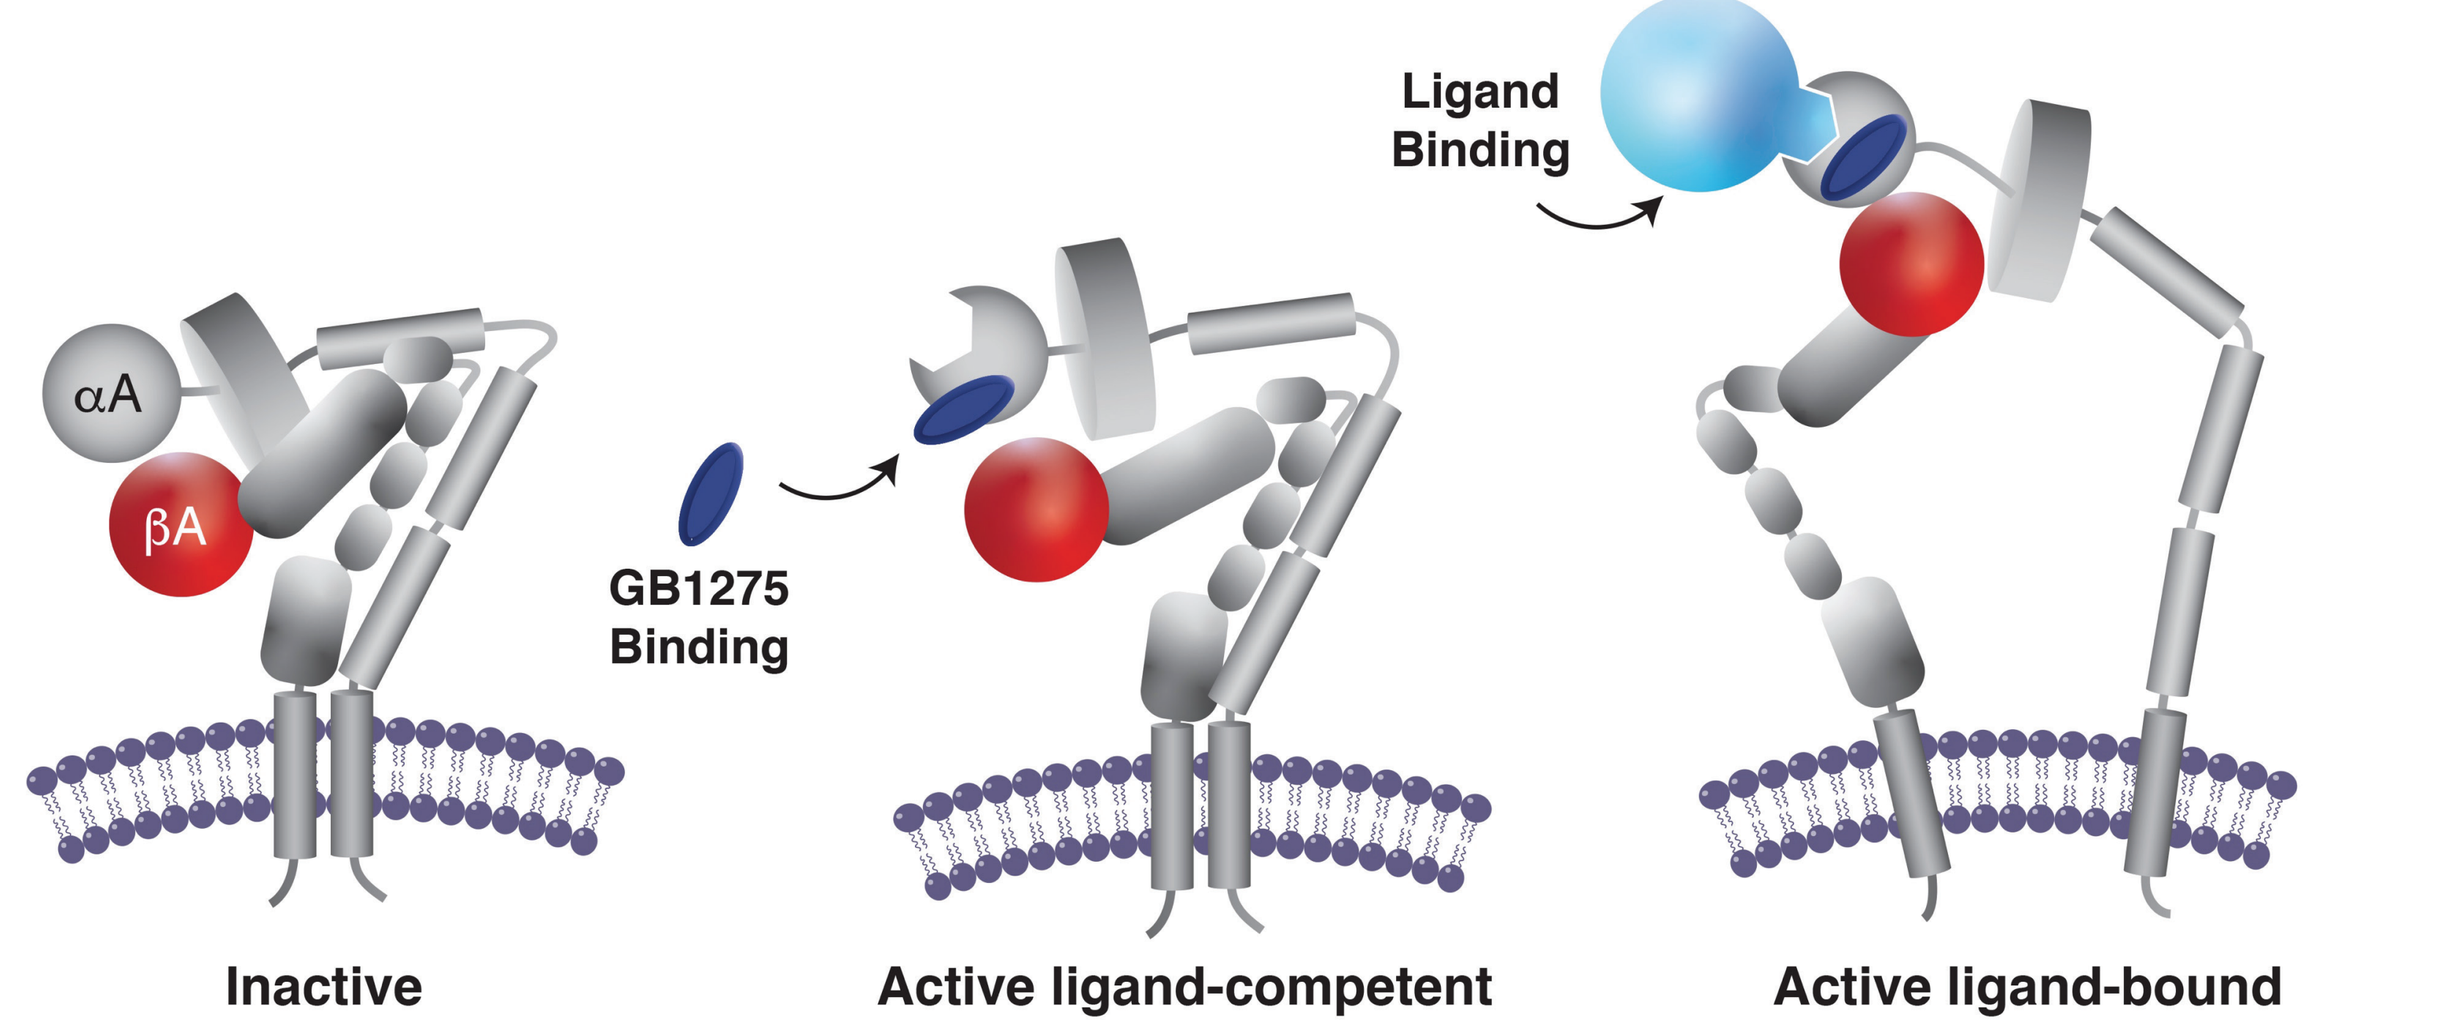
αA
βA
GB1275
Binding
Ligand
Binding
Inactive
Active ligand-competent
Active ligand-bound

- GB1275 stabilizes CD11b in ligand competent state
- Upon exposure to ligand, GB1275 promotes cell adhesion and reduces cell migration

Figure 2. LA-1 reduces CD11b+ THP1 cell transendothelial migration in vitro[5]

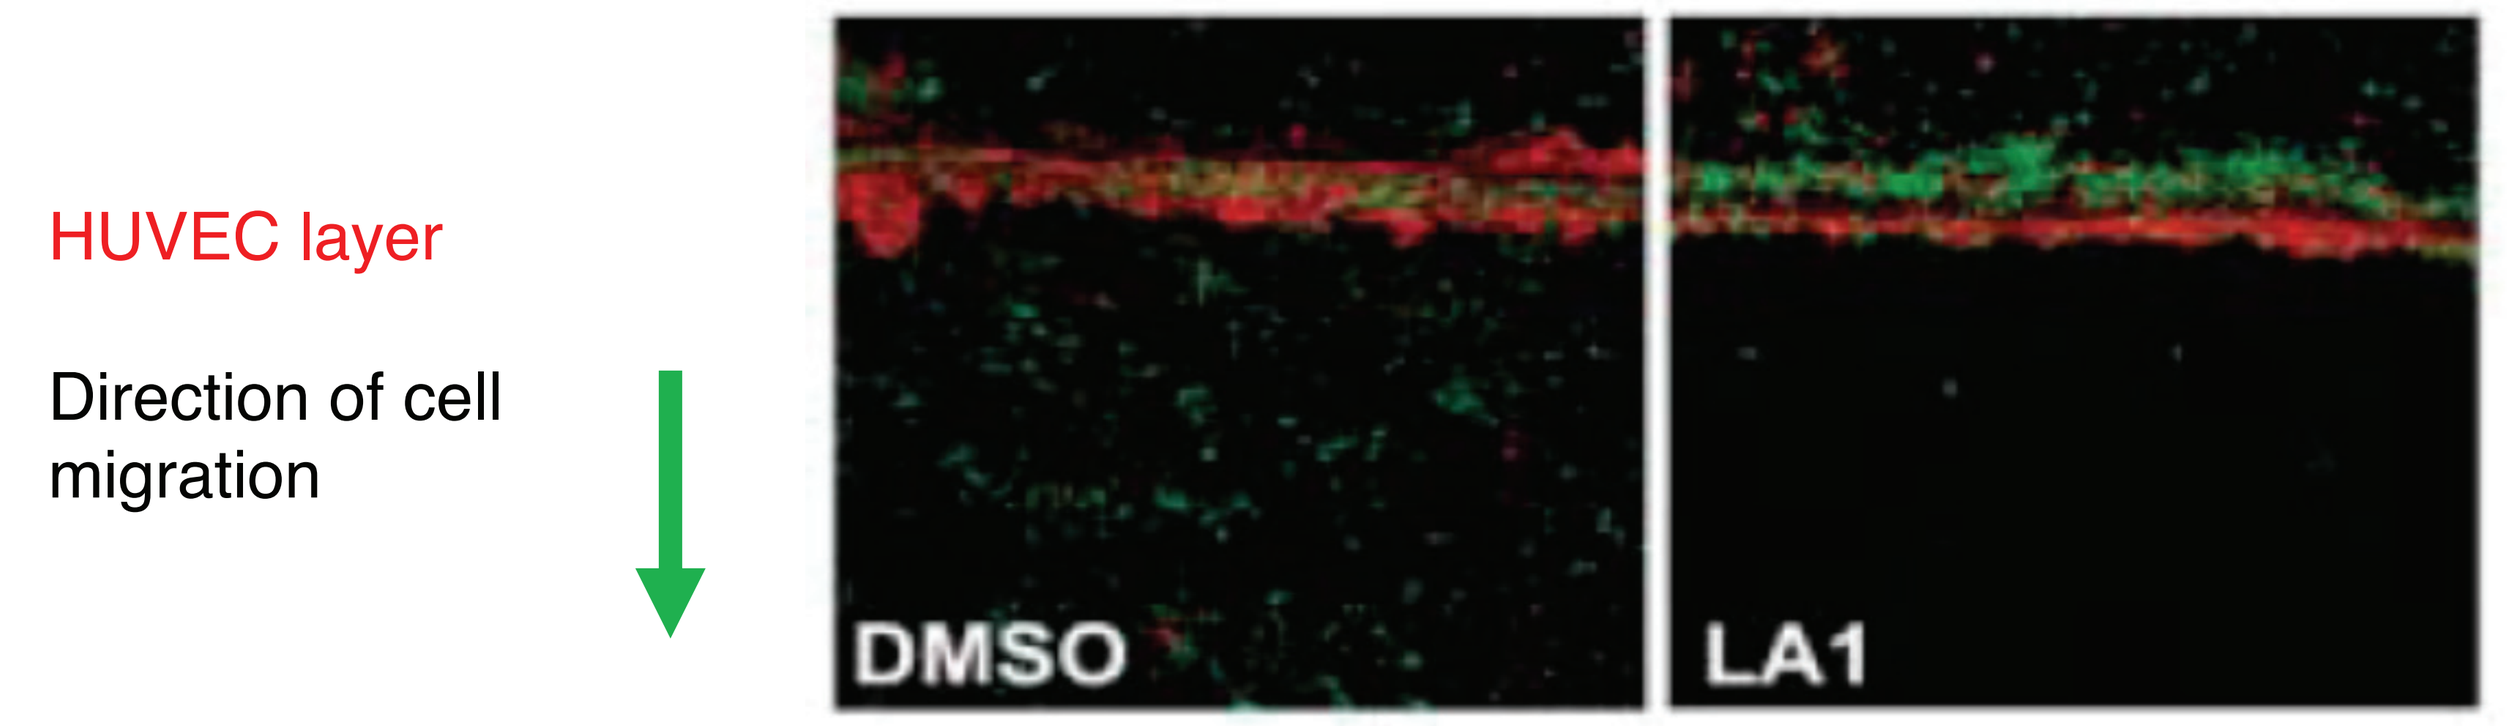
HUVEC layer
Direction of cell migration
DMSO
LA1

LA1, free acid form of GB1275

- GB1275 (formerly ADH503) is the salt form of Leukadherin-1 (LA-1)

Figure 3. Proposed Impact of GB1275 on the Tumor Microenvironment[6]

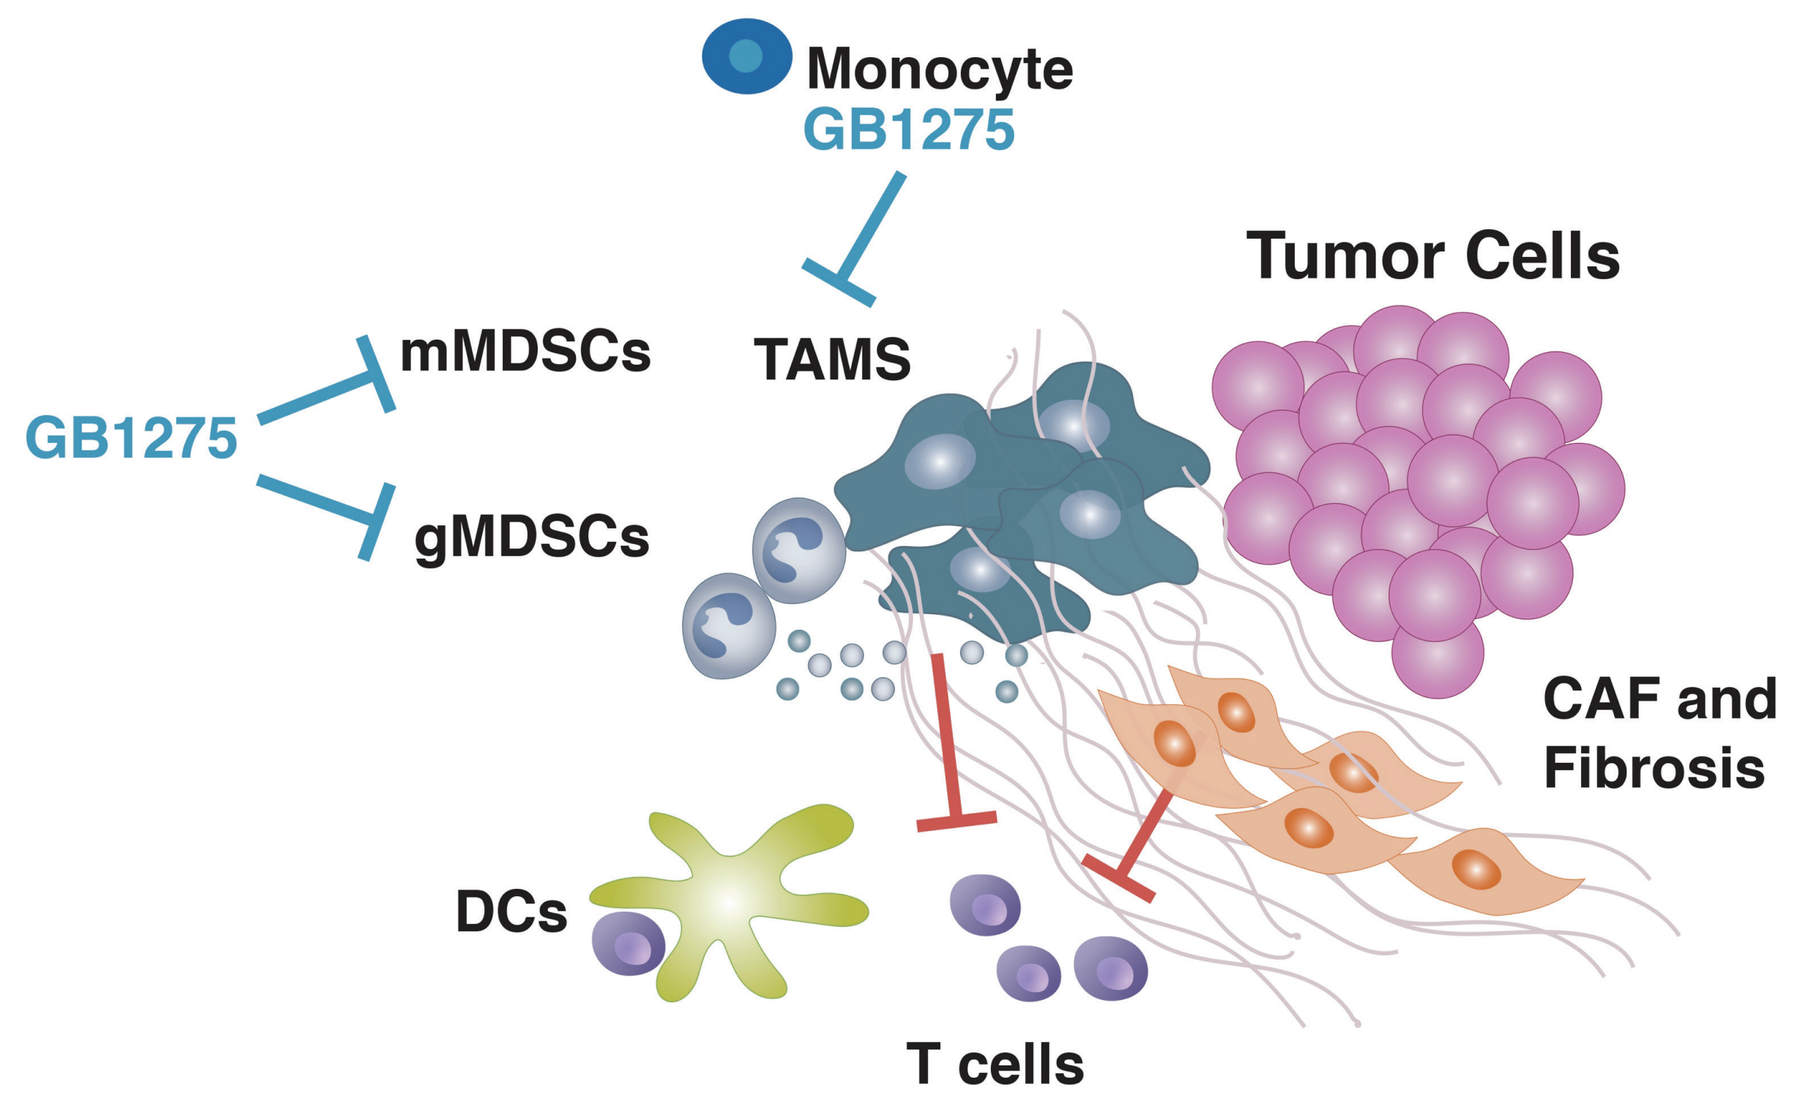
Monocyte
GB1275
TAMS
Tumor Cells
GB1275
mMDSCs
gMDSCs
CAF and Fibrosis
DCs
T cells

Figure 4. Increased CD11b expression and myeloid cell infiltration observed in primary PDAC tumors compared to adjacent normal tissues[4]

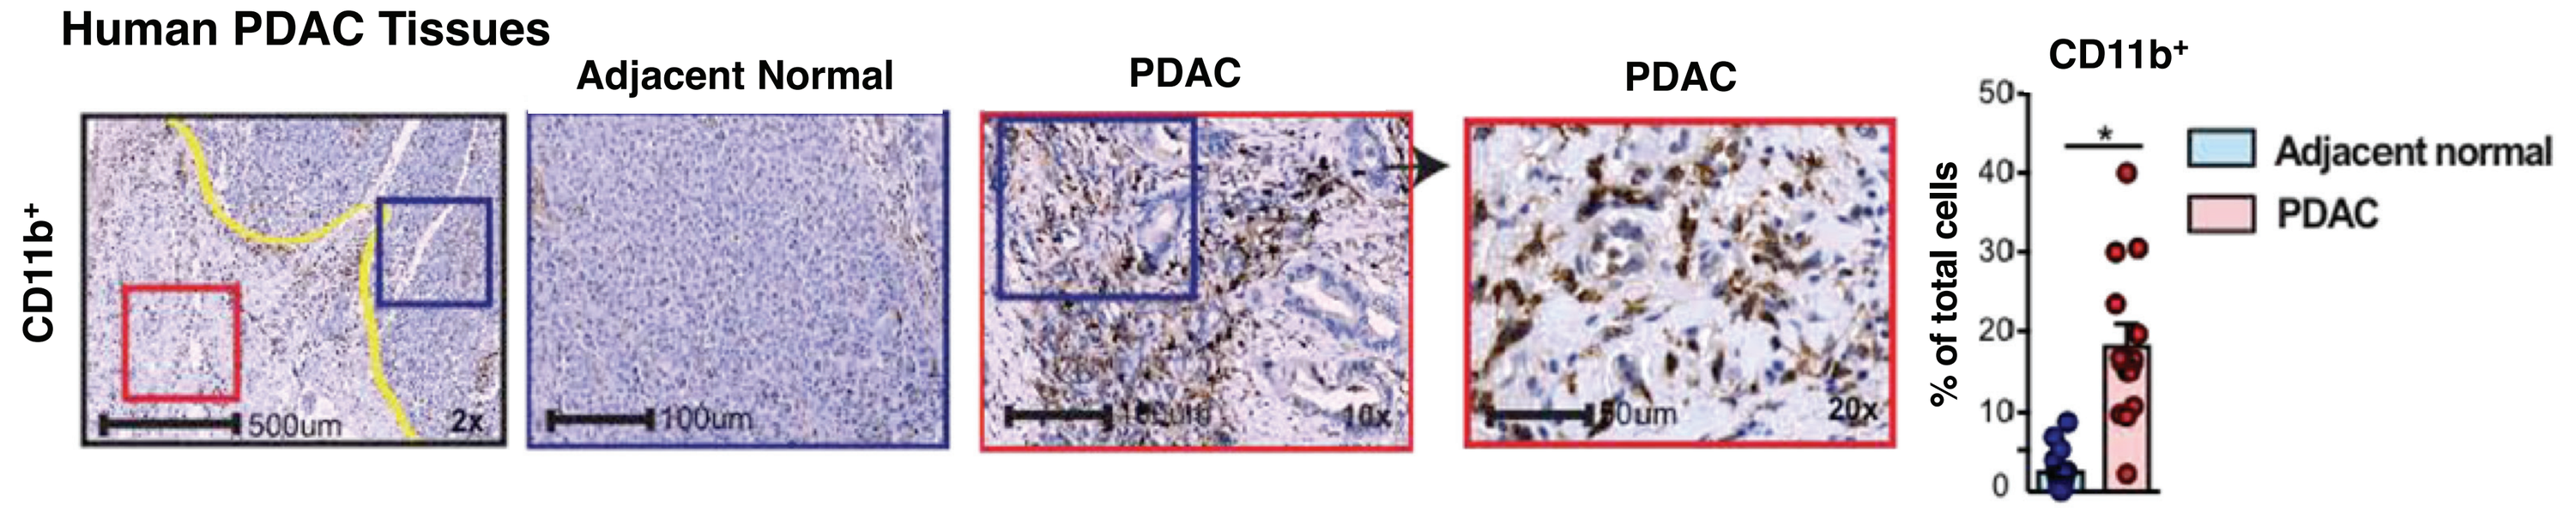
Human PDAC Tissues
Adjacent Normal
PDAC
PDAC
CD11b+
500um
2x
100um
10x
50um
20x
CD11b+
% of total cells
Adjacent normal
PDAC

Figure 5. GB1275 reduces tumor Infiltration of CD11b+ myeloid cells and increases influx of activated CD8+ T cells in the PDAC model[4]

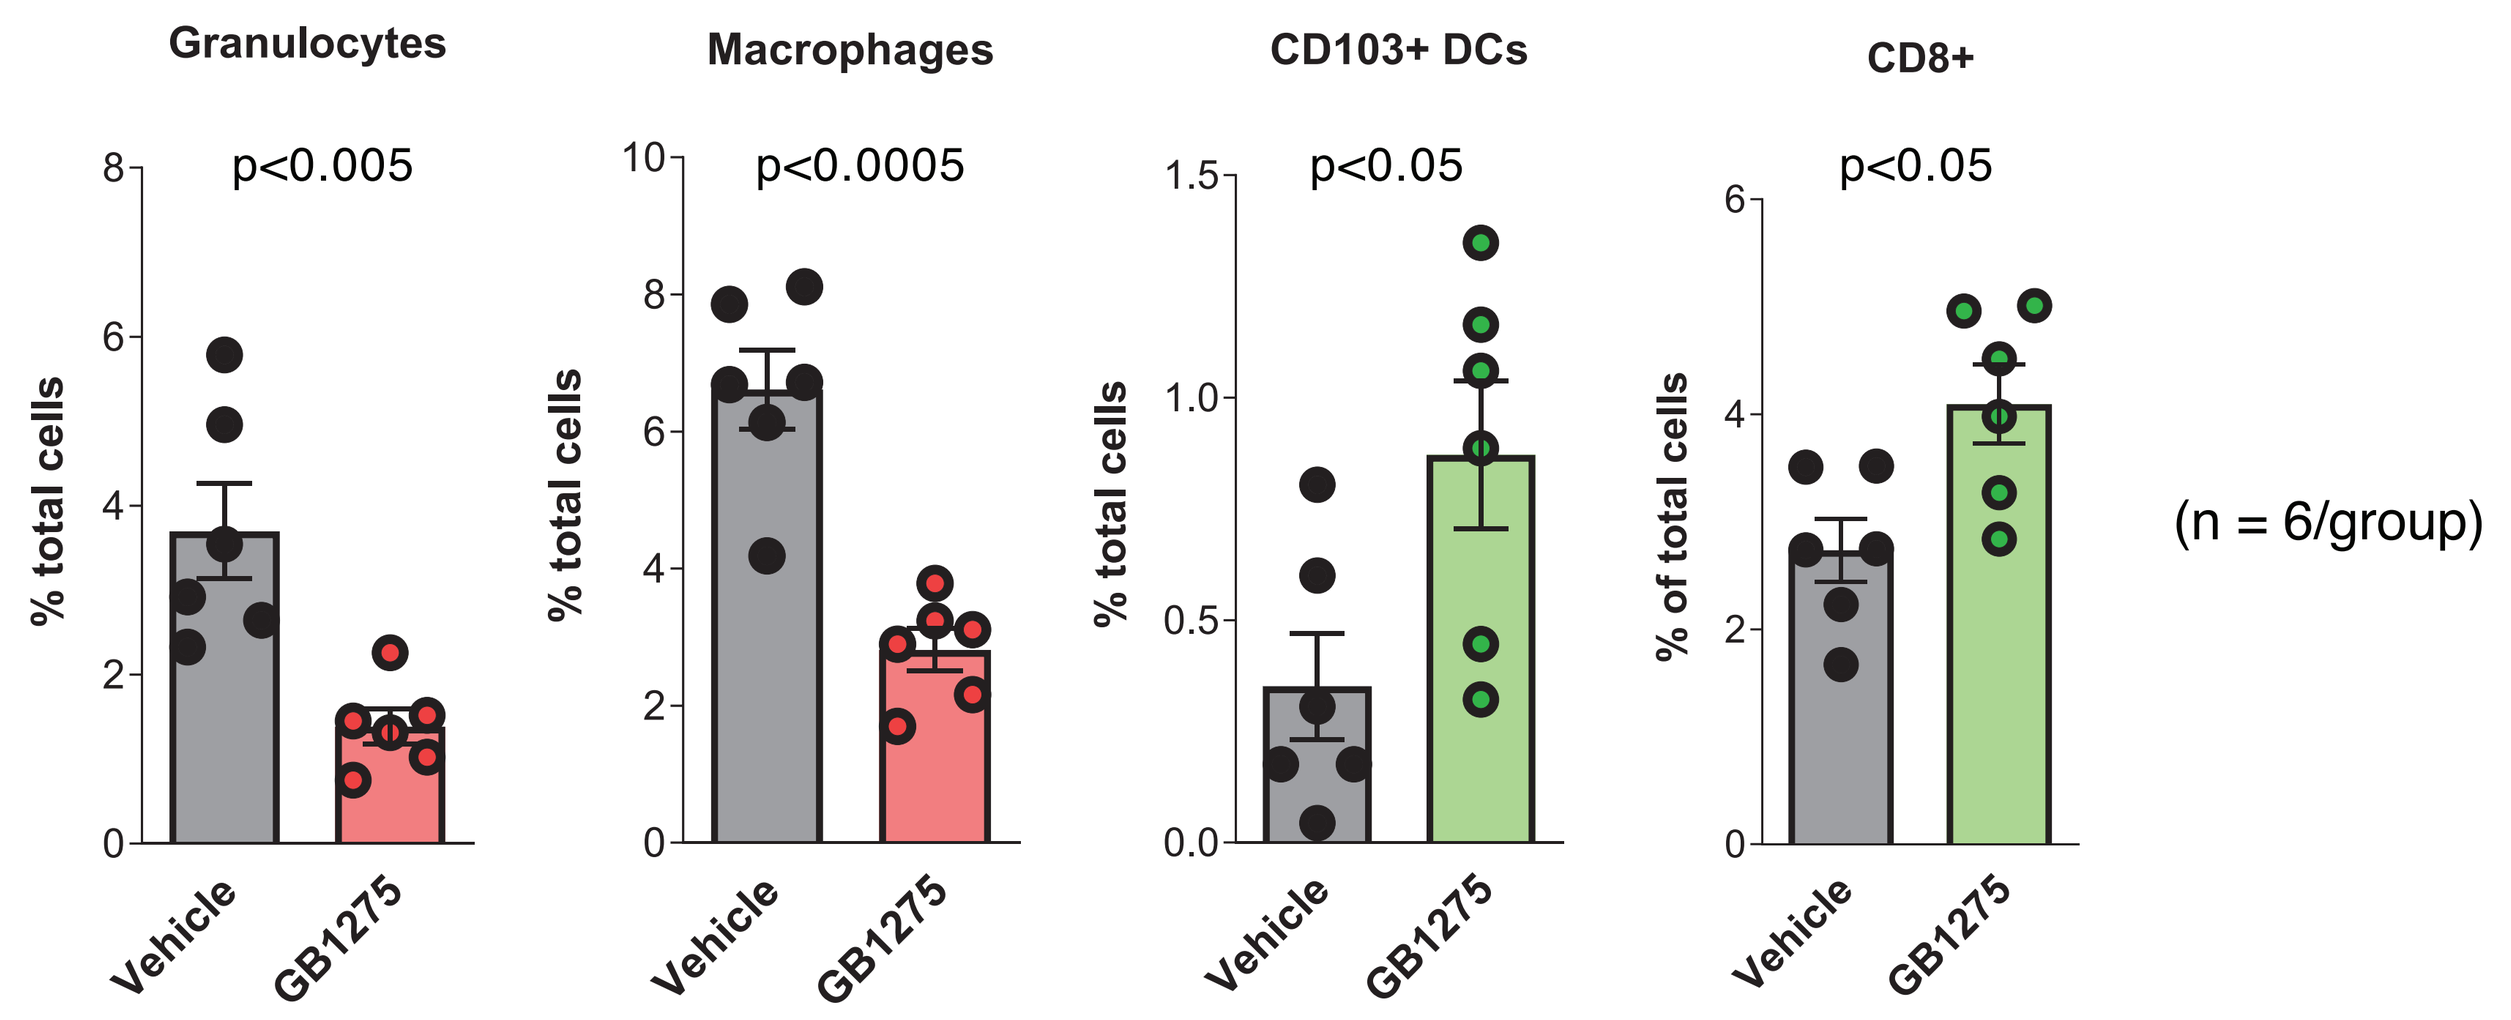
Granulocytes
p<0.005
Macrophages
p<0.0005
CD103+ DCs
p<0.05
CD8+
p<0.05
% total cells
% of total cells
Vehicle
GB1275
(n = 6/group)

- GB1275 immunomodulatory effects were tumor-localized
- GB1275 did not alter immune cell phenotypes in the periphery, spleen or bone marrow

Figure 6. GB1275 displays single agent activity and sensitizes PDAC model to anti-PD-1 Ab checkpoint blockade[4]

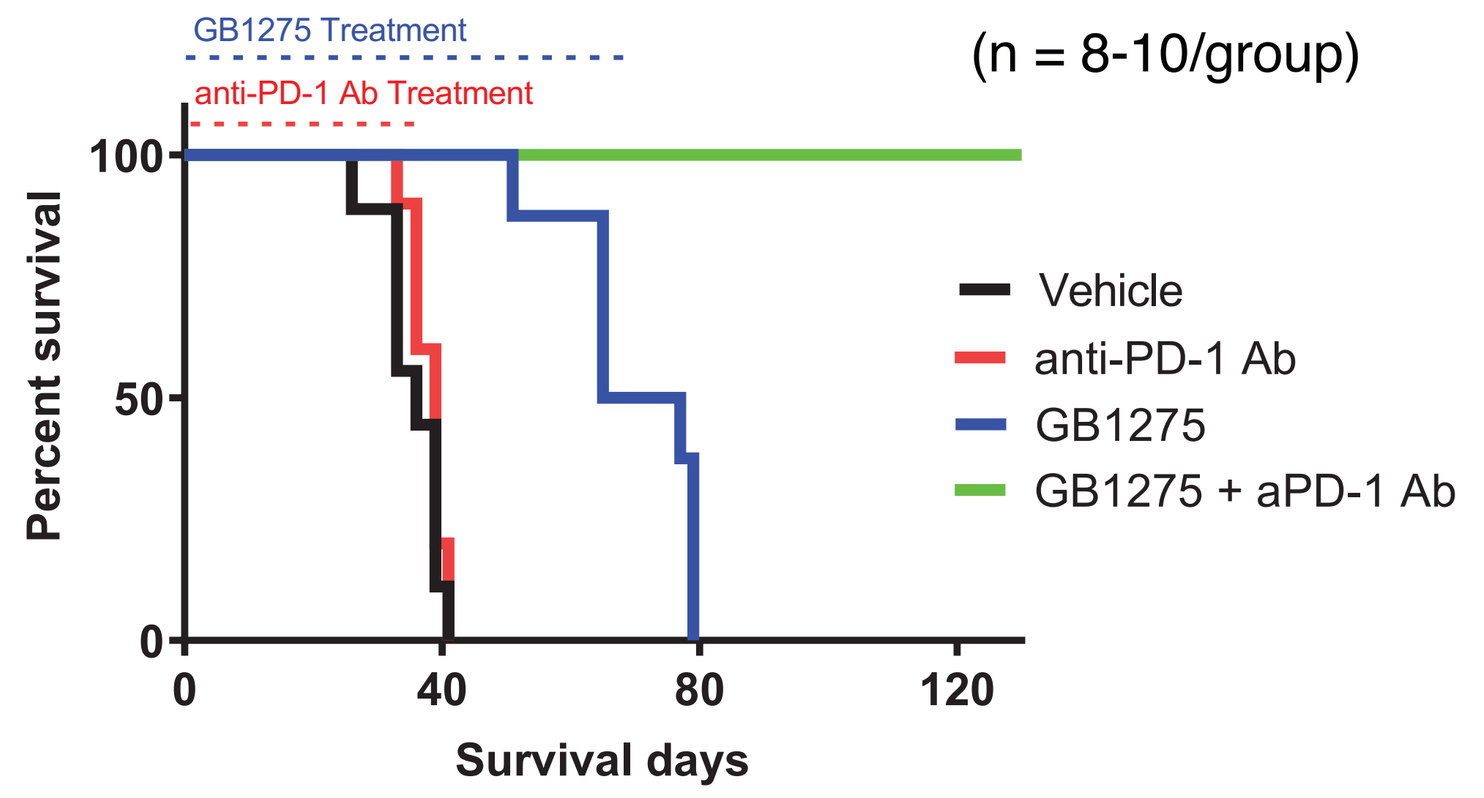
GB1275 Treatment
anti-PD-1 Ab Treatment
(n = 8-10/group)
Percent survival
Survival days
Vehicle
anti-PD-1 Ab
GB1275
GB1275 + aPD-1 Ab

Figure 7. GB1275 in combination with chemotherapy extends survival in a PDAC model[4]

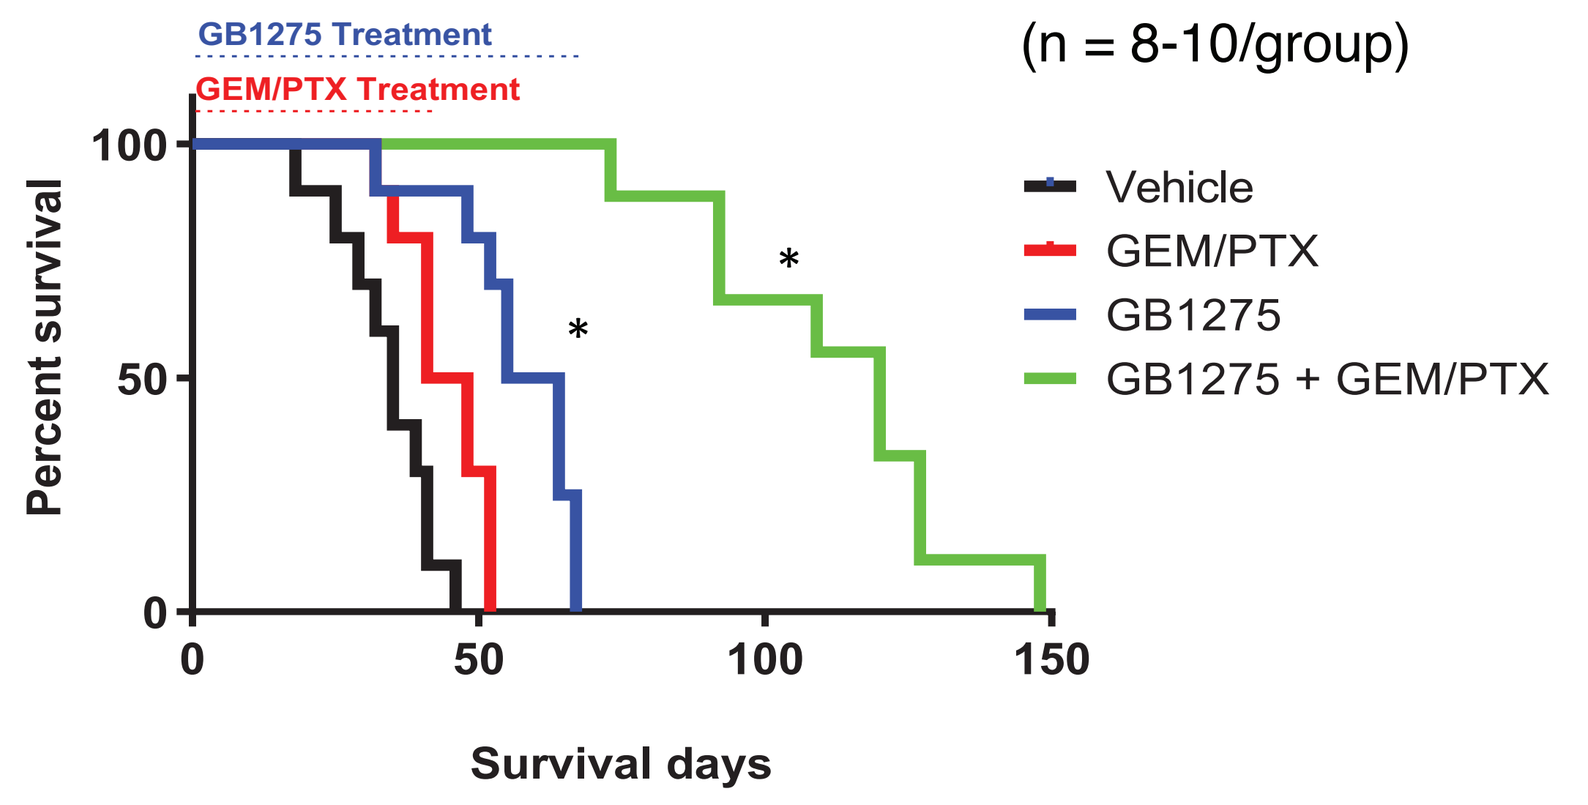
GB1275 Treatment
GEM/PTX Treatment
(n = 8-10/group)
Percent survival
Survival days
Vehicle
GEM/PTX
GB1275
GB1275 + GEM/PTX

GEM/PTX: Gemcitabine / Paclitaxel; *p<0.05

Figure 8. LA-1 displays single agent activity in the CL66-Luc orthotopic breast cancer model[7]

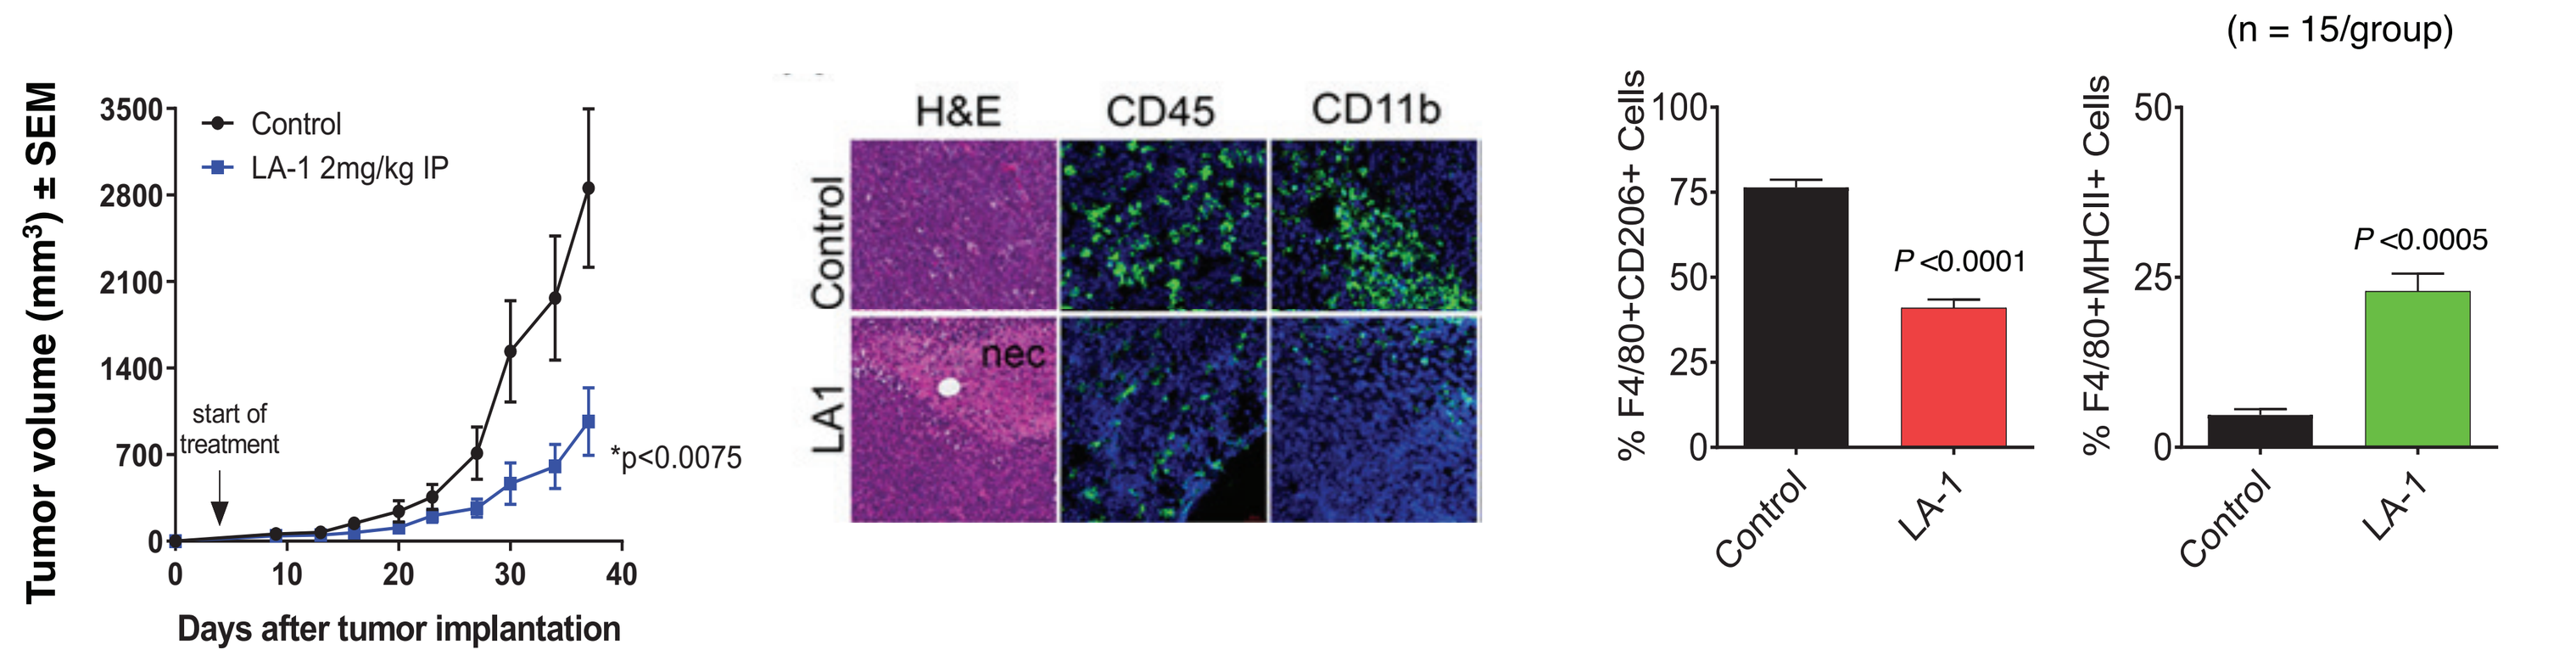
(n = 15/group)
Tumor volume (mm³) ± SEM
Control
LA-1 2mg/kg IP
start of treatment
*p<0.0075
Days after tumor implantation
H&E
CD45
CD11b
Control
LA1
nec
% F4/80+CD206+ Cells
P<0.0001
% F4/80+MHCII+ Cells
P<0.0005
Control
LA-1

- LA-1 reduced tumor influx of CD11b+ myeloid cells
- LA-1 repolarizes F4/80+ macrophages from M2 (CD206+) to M1 (MHCII+) subtype

OBJECTIVES

Phase 1 Dose Escalation

- Evaluate safety and tolerability to determine the maximum tolerated dose (MTD) or recommended Phase 2 dose (RP2D)
- Describe the pharmacokinetic-pharmacodynamic (PK-PD) profile of GB1275 as monotherapy and in combination

Phase 2 Basket Expansion

- Assess clinical efficacy of GB1275 in combination with pembrolizumab or SOC chemotherapy
- Evaluate safety and tolerability of GB1275 in combination with pembrolizumab or SOC chemotherapy

Exploratory objectives for Phase 1/2

- Evaluate changes in biomarkers to assess target engagement (gMDSCs and mMDSCs in blood and tumor, immune cell distribution by flow cytometry, CD11b+ cells at the tumor site and CD8+ T cells, etc.)

INCLUSION CRITERIA

- Age of ≥ 18 years
- ECOG performance status of 0 or 1
- Adequate organ function within 14 days of first dose of study drug

Phase 1

- **Regimen A and B:** one of the following relapsed/refractory tumor types
 - ✓pancreatic adenocarcinoma
 - ✓gastric/gastroesophageal junction (GEJ) adenocarcinoma
 - ✓esophageal adenocarcinoma or squamous cell carcinoma
 - ✓MSS colorectal
 - ✓metastatic castrate-resistant prostate
 - ✓Triple negative breast cancer (TNBC)
- **Regimen C:** mPDAC

Phase 2

- Newly diagnosed mPDAC
- MSS colorectal
 - ✓Refractory or intolerant to at least 1 fluoropyrimidine-containing regimen
 - ✓No prior pembrolizumab or other immune-modulating antitumor therapies
- Gastric/GEJ
 - ✓PD-L1 expression with combined positive score ≥1
 - ✓Refractory or intolerant to standard of care therapy after receiving at least 2 lines chemotherapy that include fluoropyrimidine- and platinum-based regimens
 - ✓No prior pembrolizumab or other immune-modulating antitumor therapies

KEY EXCLUSION CRITERIA: All subjects

- History of another malignancy within 2 years prior to enrollment
- Pregnant or nursing
- Known history of testing positive for human immunodeficiency virus (HIV)
- Positive test for Hepatitis B or Hep C infection
- Inability to take oral medication

STUDY SCHEMA

Phase 1: Dose Escalation of GB1275 monotherapy and in combination with pembrolizumab in subjects with locally advanced or metastatic pancreatic, or esophageal, or gastric/GEJ or MSS colorectal adenocarcinoma; or esophageal squamous cell carcinoma, or prostate cancer, or TNBC; or in combination with nab-paclitaxel and gemcitabine (Nab-P+Gem) in m PDAC

Phase 2 Expansion: up to 40 patients per disease cohort

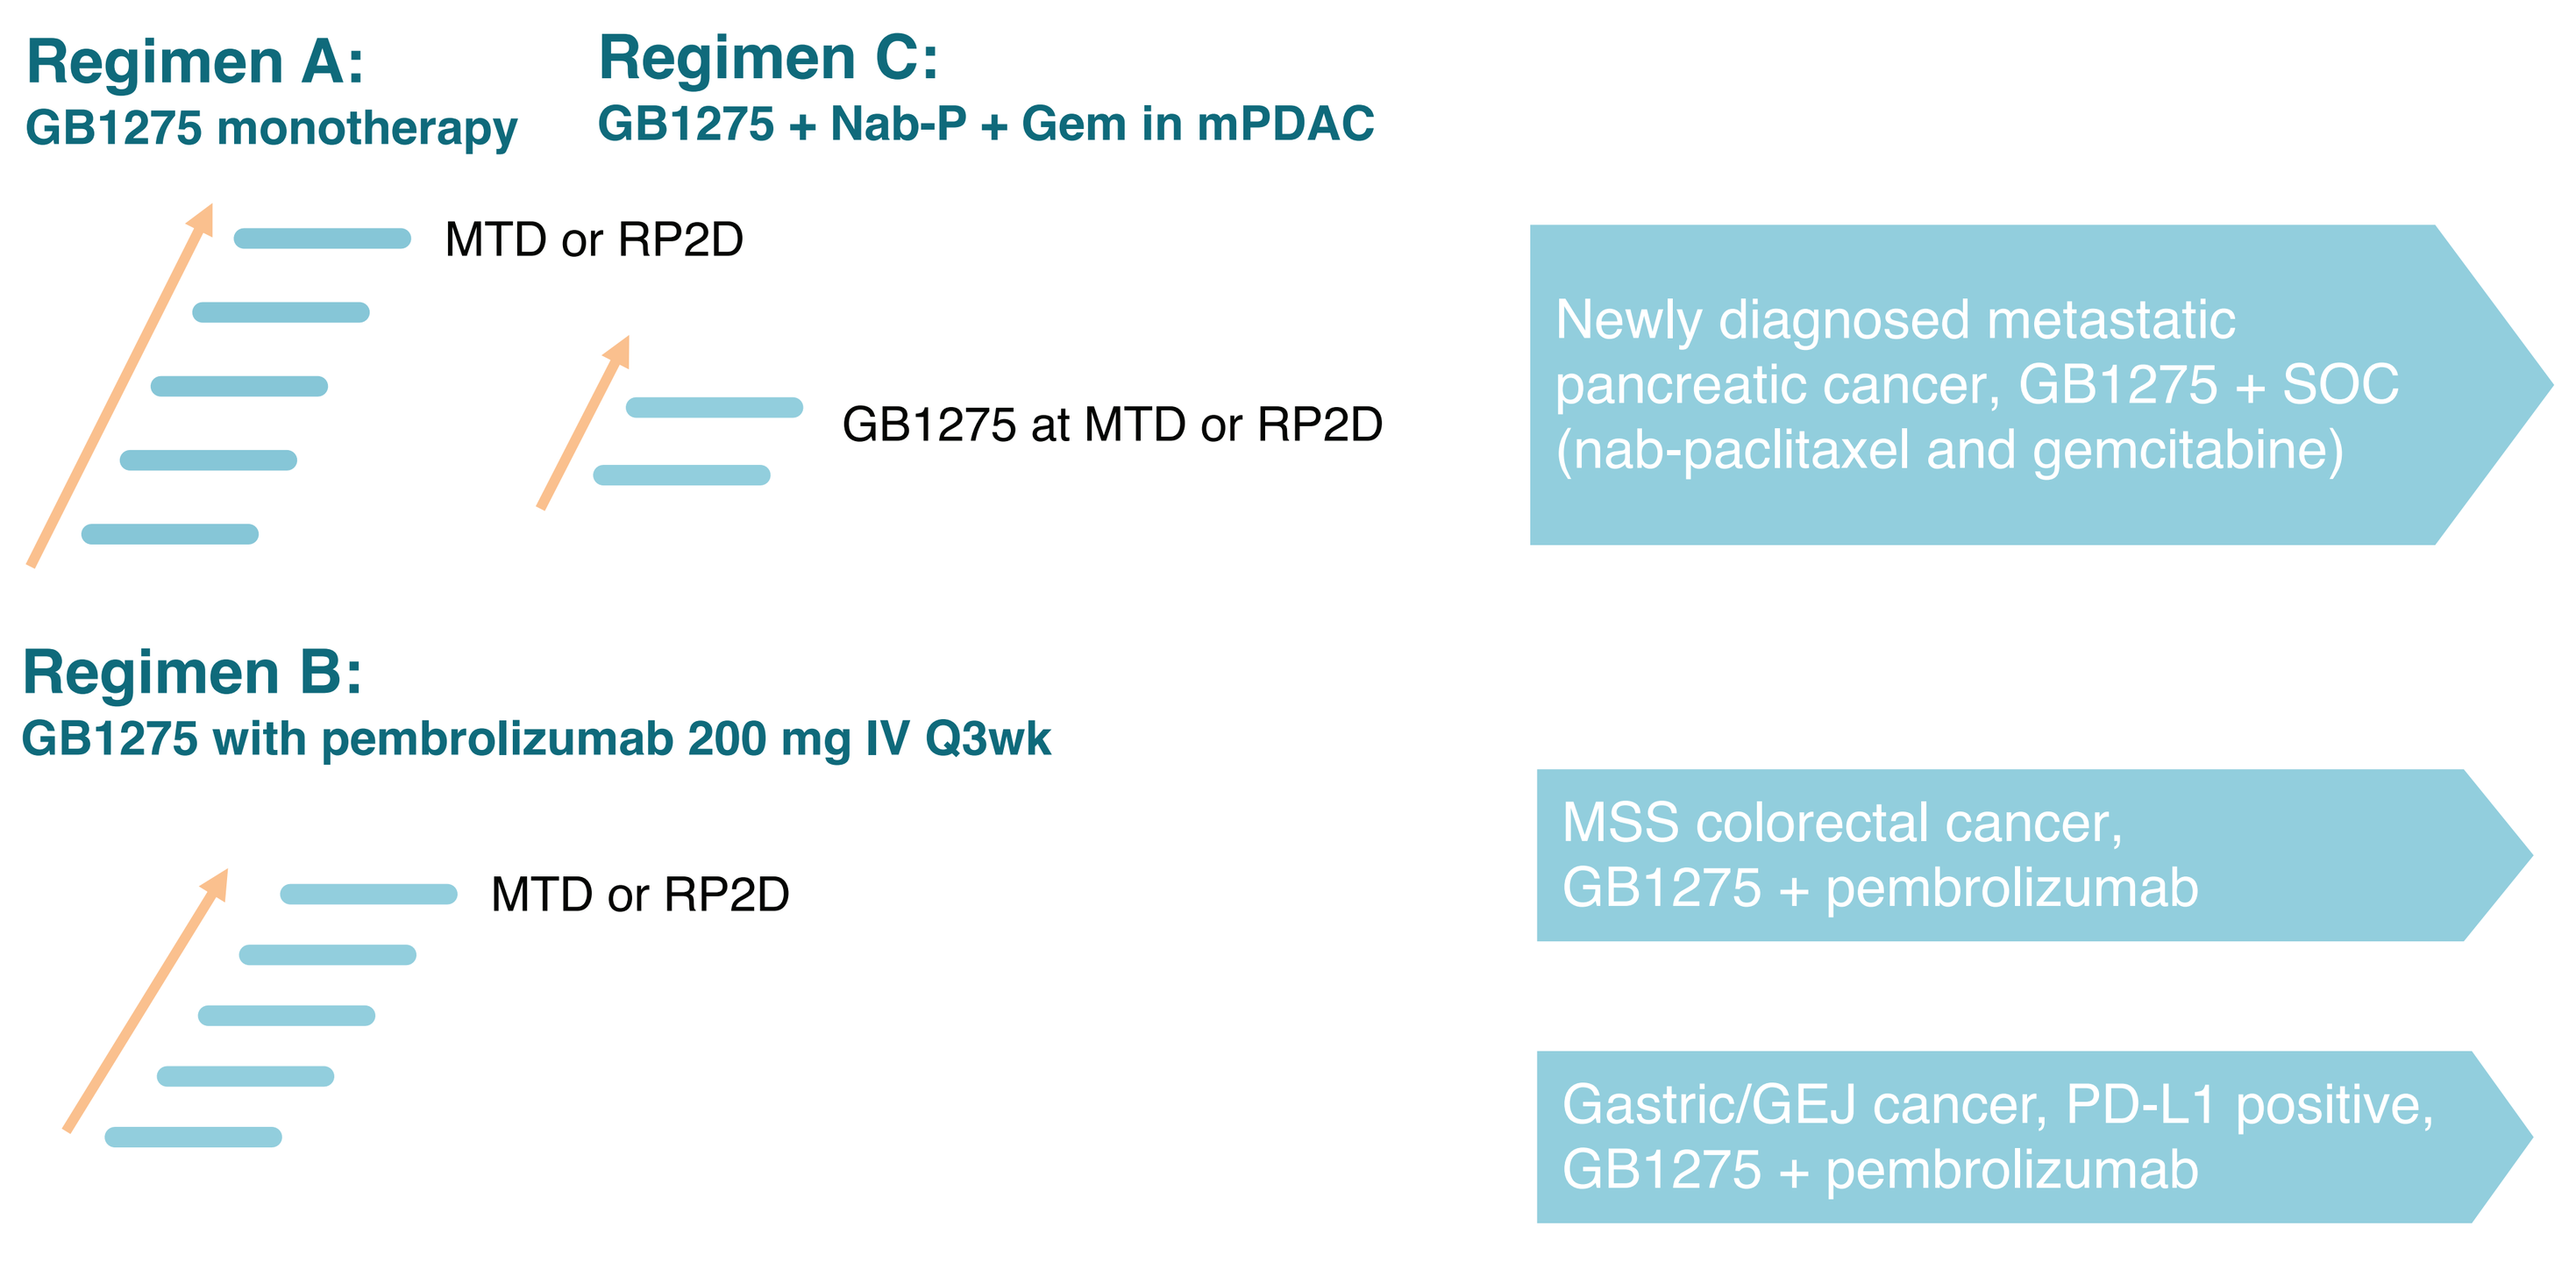
Regimen A:
GB1275 monotherapy
Regimen C:
GB1275 + Nab-P + Gem in mPDAC
MTD or RP2D
GB1275 at MTD or RP2D
Newly diagnosed metastatic pancreatic cancer, GB1275 + SOC (nab-paclitaxel and gemcitabine)
Regimen B:
GB1275 with pembrolizumab 200 mg IV Q3wk
MTD or RP2D
MSS colorectal cancer, GB1275 + pembrolizumab
Gastric/GEJ cancer, PD-L1 positive, GB1275 + pembrolizumab

TRIAL STATUS

- Four investigator sites have been activated:
 - ✓ Sarah Cannon Research Institute, Nashville, TN
 - ✓ South Texas Accelerated Research Therapeutics, San Antonio, TX
 - ✓ UC Health at University of Colorado, Denver, CO
 - ✓ Washington University Hospital, St. Louis, MO
- Enrollment on this trial has commenced
- Additional sites will be initiated and recruited
- Phase 2 portion of the study may be initiated in 2020
- Interim data presentations for monotherapy and combination cohorts are planned

References

1. Fleming V, Hu X, Weber, R, *et al. Front Immunol.* 2018; 9: 398
2. Kumar V, Patel S, Tcyganov E, et al. *Trends Immunol.* 2016; 37(3): 208-220.
3. Mantovani A, Sozzani S, Locati, M, et al. *Trends Immunol.* 2002; 23(11): 549-555.
5. Maiguel D, Faridi MH, Wei C, et al. *Sci Signal* 2011;4(189):ra57
6. Zhu Y, Li M, Bo C, et al. *Cancer Immunol Immunother.* 2017;66(3):343-354.
7. Schmid MC, Khan SQ, Kaneda MM, et al. *Nat Commun* 2018;9:5379

Acknowledgements

We would like to thank our patients, their families, and investigator site staff for their participation in this trial. We gratefully acknowledge Dr. David DeNardo and his laboratory at Washington University for generating preclinical data to support the mechanism of action and preclinical efficacy model to enable clinical development of GB1275.

Presented at the SITC 2019, National Harbor, Maryland USA, November 6–10, 2019