Abstract 3085

A Phase 1/2 Study of GB1275, a First-in-Class CD11b Modulator, as Monotherapy and With an Anti-PD-1 Antibody in Specified Advanced Solid Tumors or With Chemotherapy in Metastatic Pancreatic Cancer (KEYNOTE A36)

Drew W. Rasco,[1] Johanna C. Bendell,[2] Andrea Wang-Gillam,[3] Wungki Park,[4] Eileen Mary O'Reilly,[5] Lei Zhou,[6] Anna Galkin,[6] Laura L. Carter,[6] David Nickle,[6] Jack Li,[6] Beatrice Ferguson,[6] Marya F. Chaney,[7] Jakob Dupont,[6] Wells A. Messersmith[8]

[1]START, San Antonio, TX; [2]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN; [3]Washington University School of Medicine, St. Louis, MO; [4]Memorial Sloan Kettering Cancer Center, Weill Cornell Medical College, New York, NY; [5]Memorial Sloan Kettering Cancer Center, New York, NY; [6]Gossamer Bio, Inc., San Diego, CA; [7]Merck & Co., Inc, Kenilworth, NJ; [8]University of Colorado Comprehensive Cancer Center, Aurora, CO

INTRODUCTION

- Tumor influx of CD11b-expressing myeloid-derived suppressor cells (MDSCs) and M2 tumor-associated macrophages (TAMs) creates an immunosuppressive tumor microenvironment associated with resistance to anti-PD-1 antibody therapy[1-3]
- GB1275 is a first-in-class, CD11b modulator that reduced MDSCs and TAMs at the tumor site, repolarized M2 immunosuppressive TAMs towards an M1 phenotype, and thus increased tumor infiltration of activated CD8+ T cells in vivo[4]
- Preclinical anti-tumor activity was seen with GB1275 as a single agent and synergistically in combination with chemotherapy and immuno-oncology therapies[4]

Figure 1. GB1275 Mechanism of Action: CD11b Modulation is a Novel MDSC / TAM-Targeting Approach

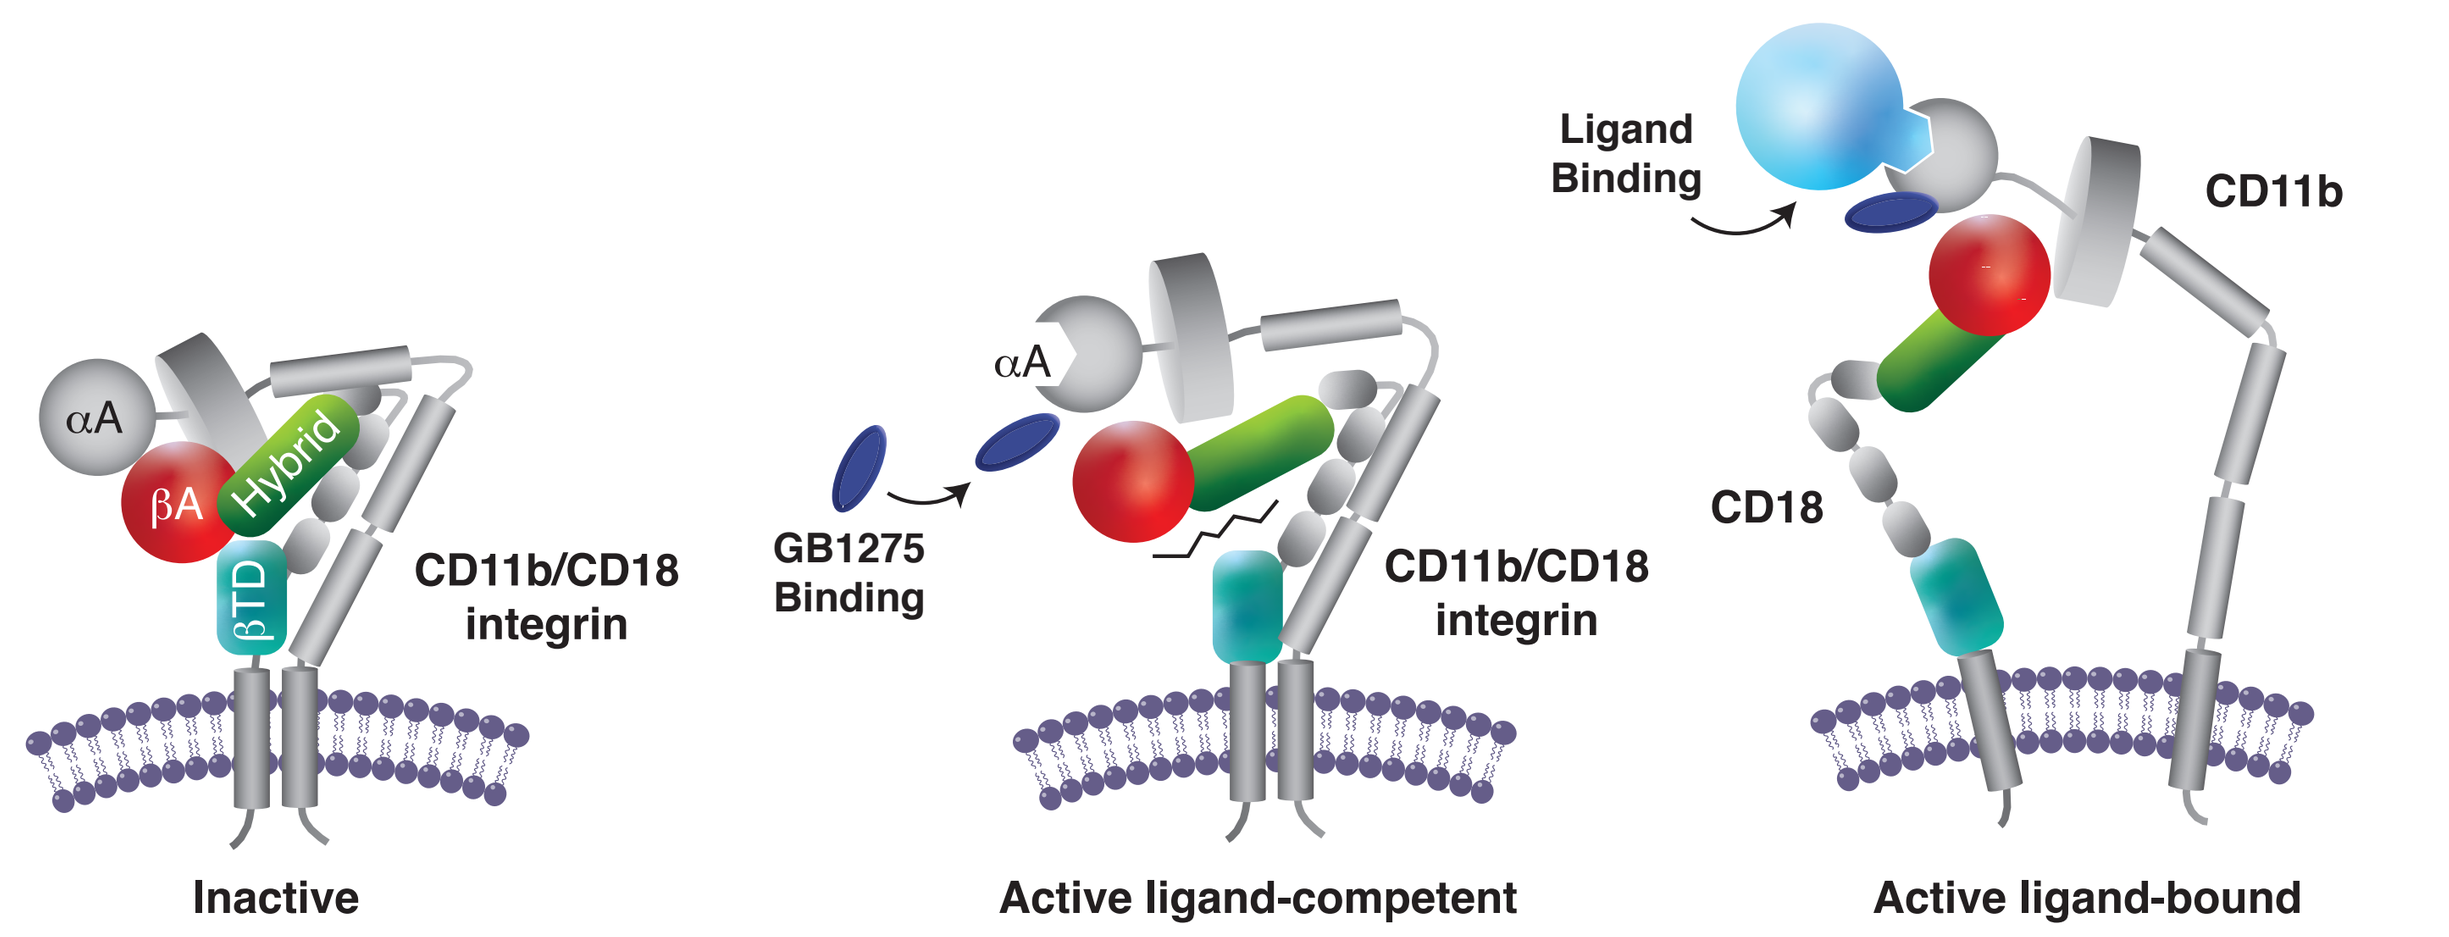
αA
βA
Hybrid
βTD
CD11b/CD18 integrin
GB1275 Binding
αA
CD11b/CD18 integrin
Ligand Binding
CD11b
CD18
Inactive
Active ligand-competent
Active ligand-bound

- GB1275 binds to an allosteric pocket in the alpha-A-domain of CD11b, which stabilizes CD11b in a conformation required for binding to its ligands (**Figure 1**)
- GB1275 is being tested in a first-in-human Phase 1/2 study as monotherapy and in combination with pembrolizumab or standard of care chemotherapy in tumor types that are known to be immuno-oncology resistant
- This trial is in progress (NCT04060342)

METHODS

- Eligible patients
 - At least 18 years of age
 - ECOG PS 0 or 1
 - Prespecified refractory tumors included in the dose escalation phase using Regimen A and B; Regimen C will be evaluated in patients with pancreatic cancer
- GB1275 is given orally at 5 dose levels, alone and with pembrolizumab 200 mg IV every 3 weeks (**Figure 2**)

Figure 2. Study Schema

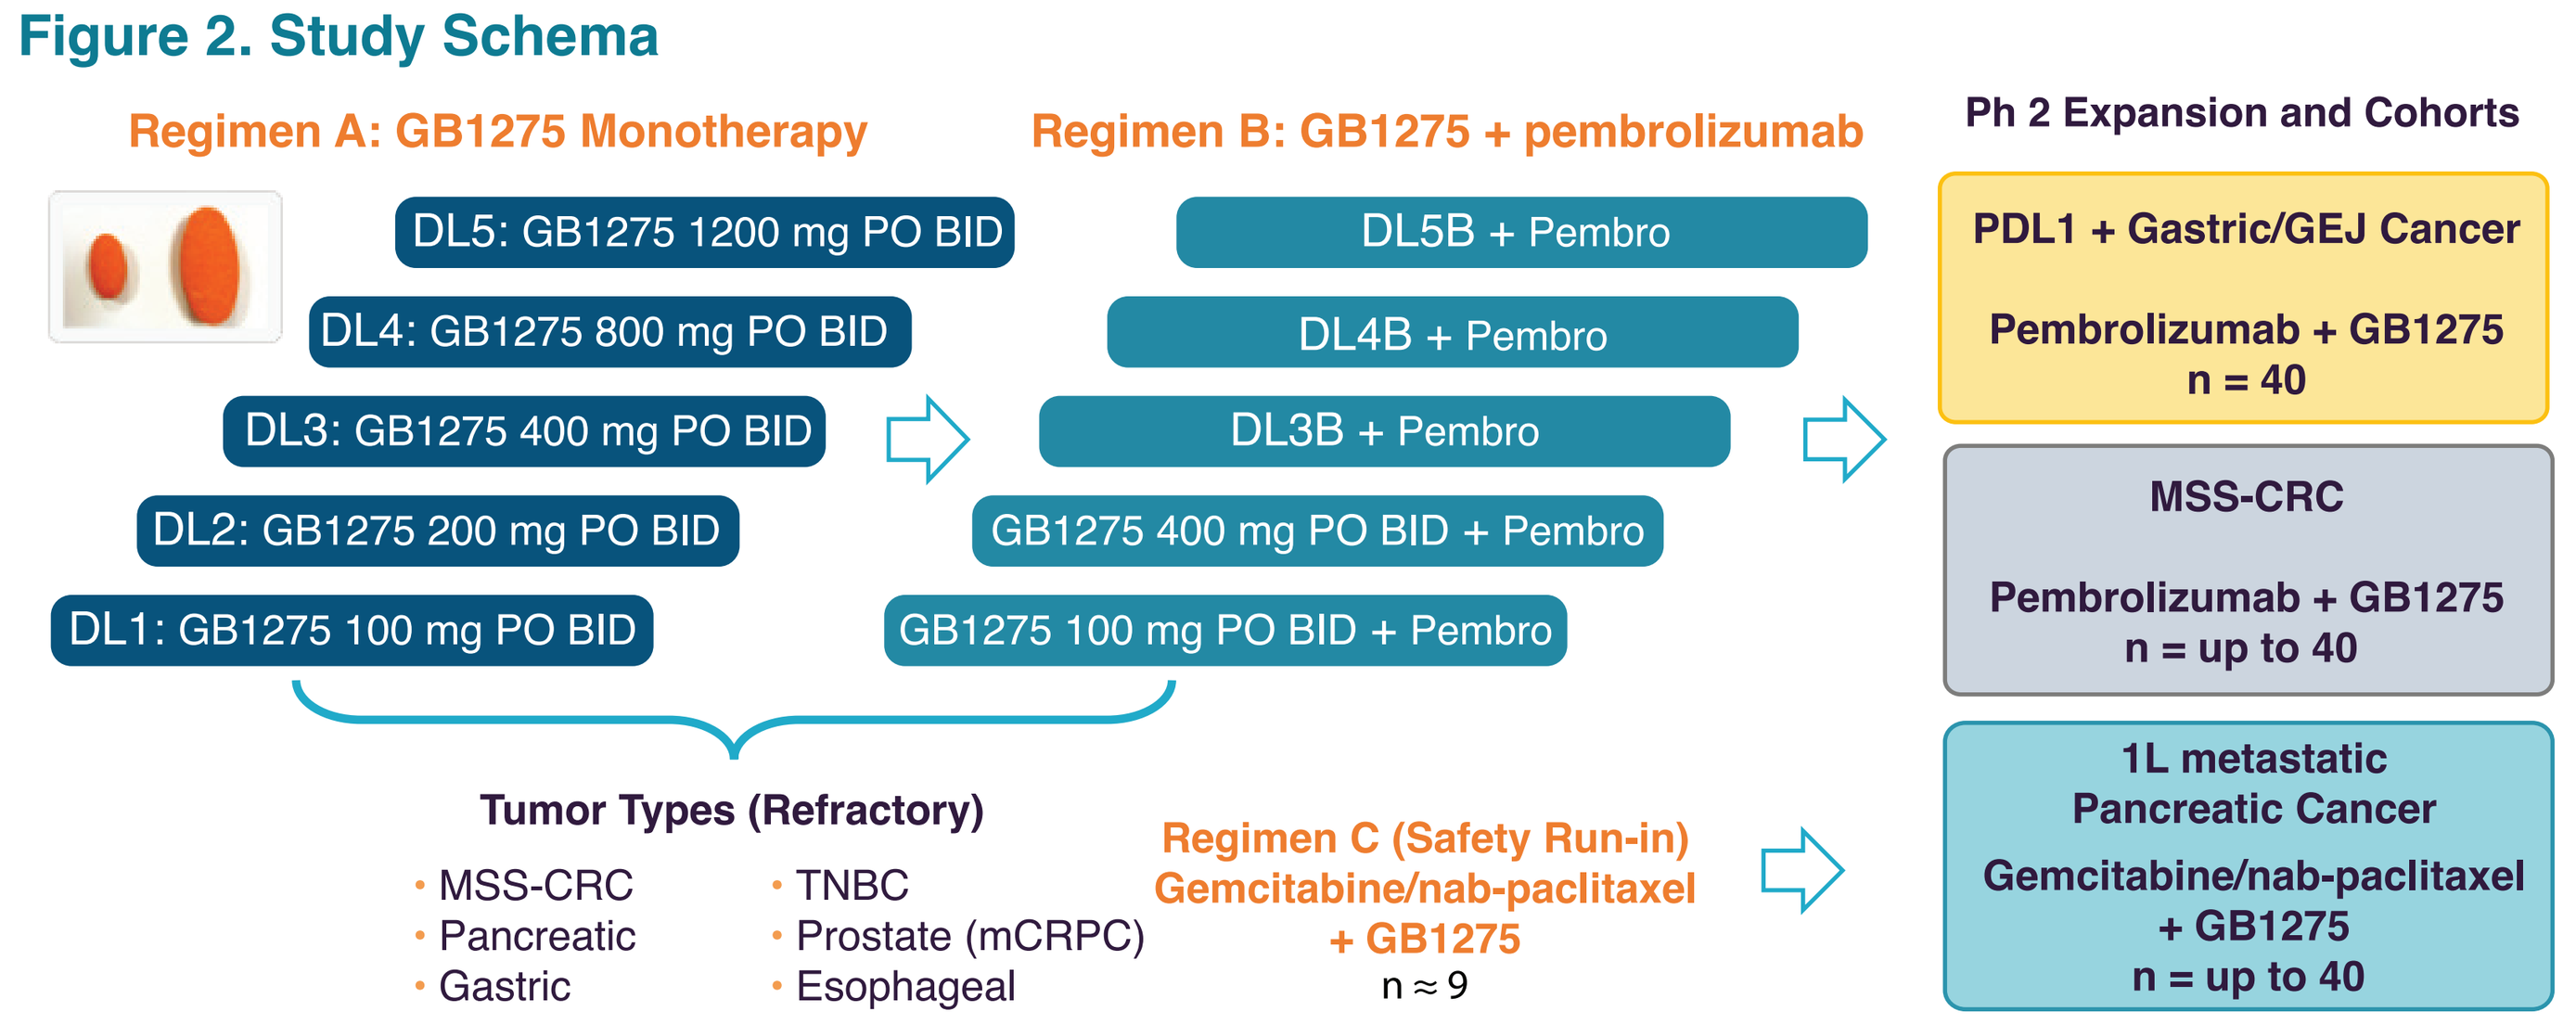
Regimen A: GB1275 Monotherapy
DL5: GB1275 1200 mg PO BID
DL4: GB1275 800 mg PO BID
DL3: GB1275 400 mg PO BID
DL2: GB1275 200 mg PO BID
DL1: GB1275 100 mg PO BID
Regimen B: GB1275 + pembrolizumab
DL5B + Pembro
DL4B + Pembro
DL3B + Pembro
GB1275 400 mg PO BID + Pembro
GB1275 100 mg PO BID + Pembro
Tumor Types (Refractory)
MSS-CRC
Pancreatic
Gastric
TNBC
Prostate (mCRPC)
Esophageal
Regimen C (Safety Run-in)
Gemcitabine/nab-paclitaxel + GB1275
n ≈ 9
Ph 2 Expansion and Cohorts
PDL1 + Gastric/GEJ Cancer
Pembrolizumab + GB1275
n = 40
MSS-CRC
Pembrolizumab + GB1275
n = up to 40
1L metastatic Pancreatic Cancer
Gemcitabine/nab-paclitaxel + GB1275
n = up to 40

DL, dose level; GEJ, gastroesophageal junction; MSS-CRC, microsatellite stable-colorectal cancer; TNBC, triple negative breast cancer

- Dose escalation was based on a standard 3 + 3 design; phase 2 basket expansion utilized a Simon's two-stage optimal design
- Study endpoints:
 - Safety: Dose-limiting toxicity, adverse events
 - Pharmacokinetic (PK) profile
 - Treatment response: Tumor assessment by RECIST 1.1 every 2 cycles (~every 42 days for Regimens A & B)
 - Pharmacodynamics: Serial blood and tumor samples were collected for PK and biomarker analyses
- Percentage of peripheral MDSCs was measured using multicolor flow cytometry (Serametrix). Whole blood transcriptome sequencing was performed at Fulgent Genetics.

RESULTS

- As of March 27, 2020, 22 patients were enrolled in the dose escalation study (**Table 1**)

Table 1. Baseline demographics and disease characteristics

	Regimen A (GB1275 monotherapy)						Regimen B (GB1275 + pembrolizumab)			
	100 mg PO BID (n = 3)	200 mg PO BID (n = 3)	400 mg PO BID (n = 3)	800 mg PO BID (n = 4)	1200 mg PO BID (n = 1)	All (n = 14)	100 mg PO BID (n = 4)	400 mg PO BID (n = 4)	All (n = 8)	Overall (N = 22)
Age, years	60 (58,78)	73 (61,74)	67 (59,73)	56.5 (37,72)	73 (73,73)	64 (37,78)	69.5 (43, 79)	69.5 (49, 75)	69.5 (43, 79)	68 (37, 79)
Male	1 (33)	1 (33)	2 (67)	3 (75)	1 (100)	8 (57)	1 (25)	1 (25)	2 (25)	10 (46)
ECOG										
0	2 (67)	2 (67)	2 (67)	–	1 (100)	7 (50)	0	0	0	7 (32)
1	1 (33)	1 (33)	1 (33)	4 (100)	–	7 (50)	4 (100)	4 (100)	8 (100)	15 (68)
Prior therapies										
< 3	1 (33)	2 (67)	1 (33)	1 (25)	1 (100)	6 (43)	1 (25)	–	1 (12)	7 (32)
≥ 3	2 (67)	1 (33)	2 (67)	3 (75)	–	8 (57)	3 (75)	4 (100)	7 (88)	15 (68)

Data as of 27 March 2020. Continuous data are presented as median (range); categorical data are presented as n (%).

Safety

- No dose-limiting toxicities have been reported
- GB1275 as monotherapy (up to 1200 mg PO BID) and combined with pembrolizumab (up to 400 mg PO BID) was well tolerated (**Figure 3 & 4**)
- Most frequently reported treatment emergent AEs were Grade 1 or Grade 2
- Three patients discontinued study treatment due to death related to underlying disease

Figure 3. TEAEs Reported in > One Patient

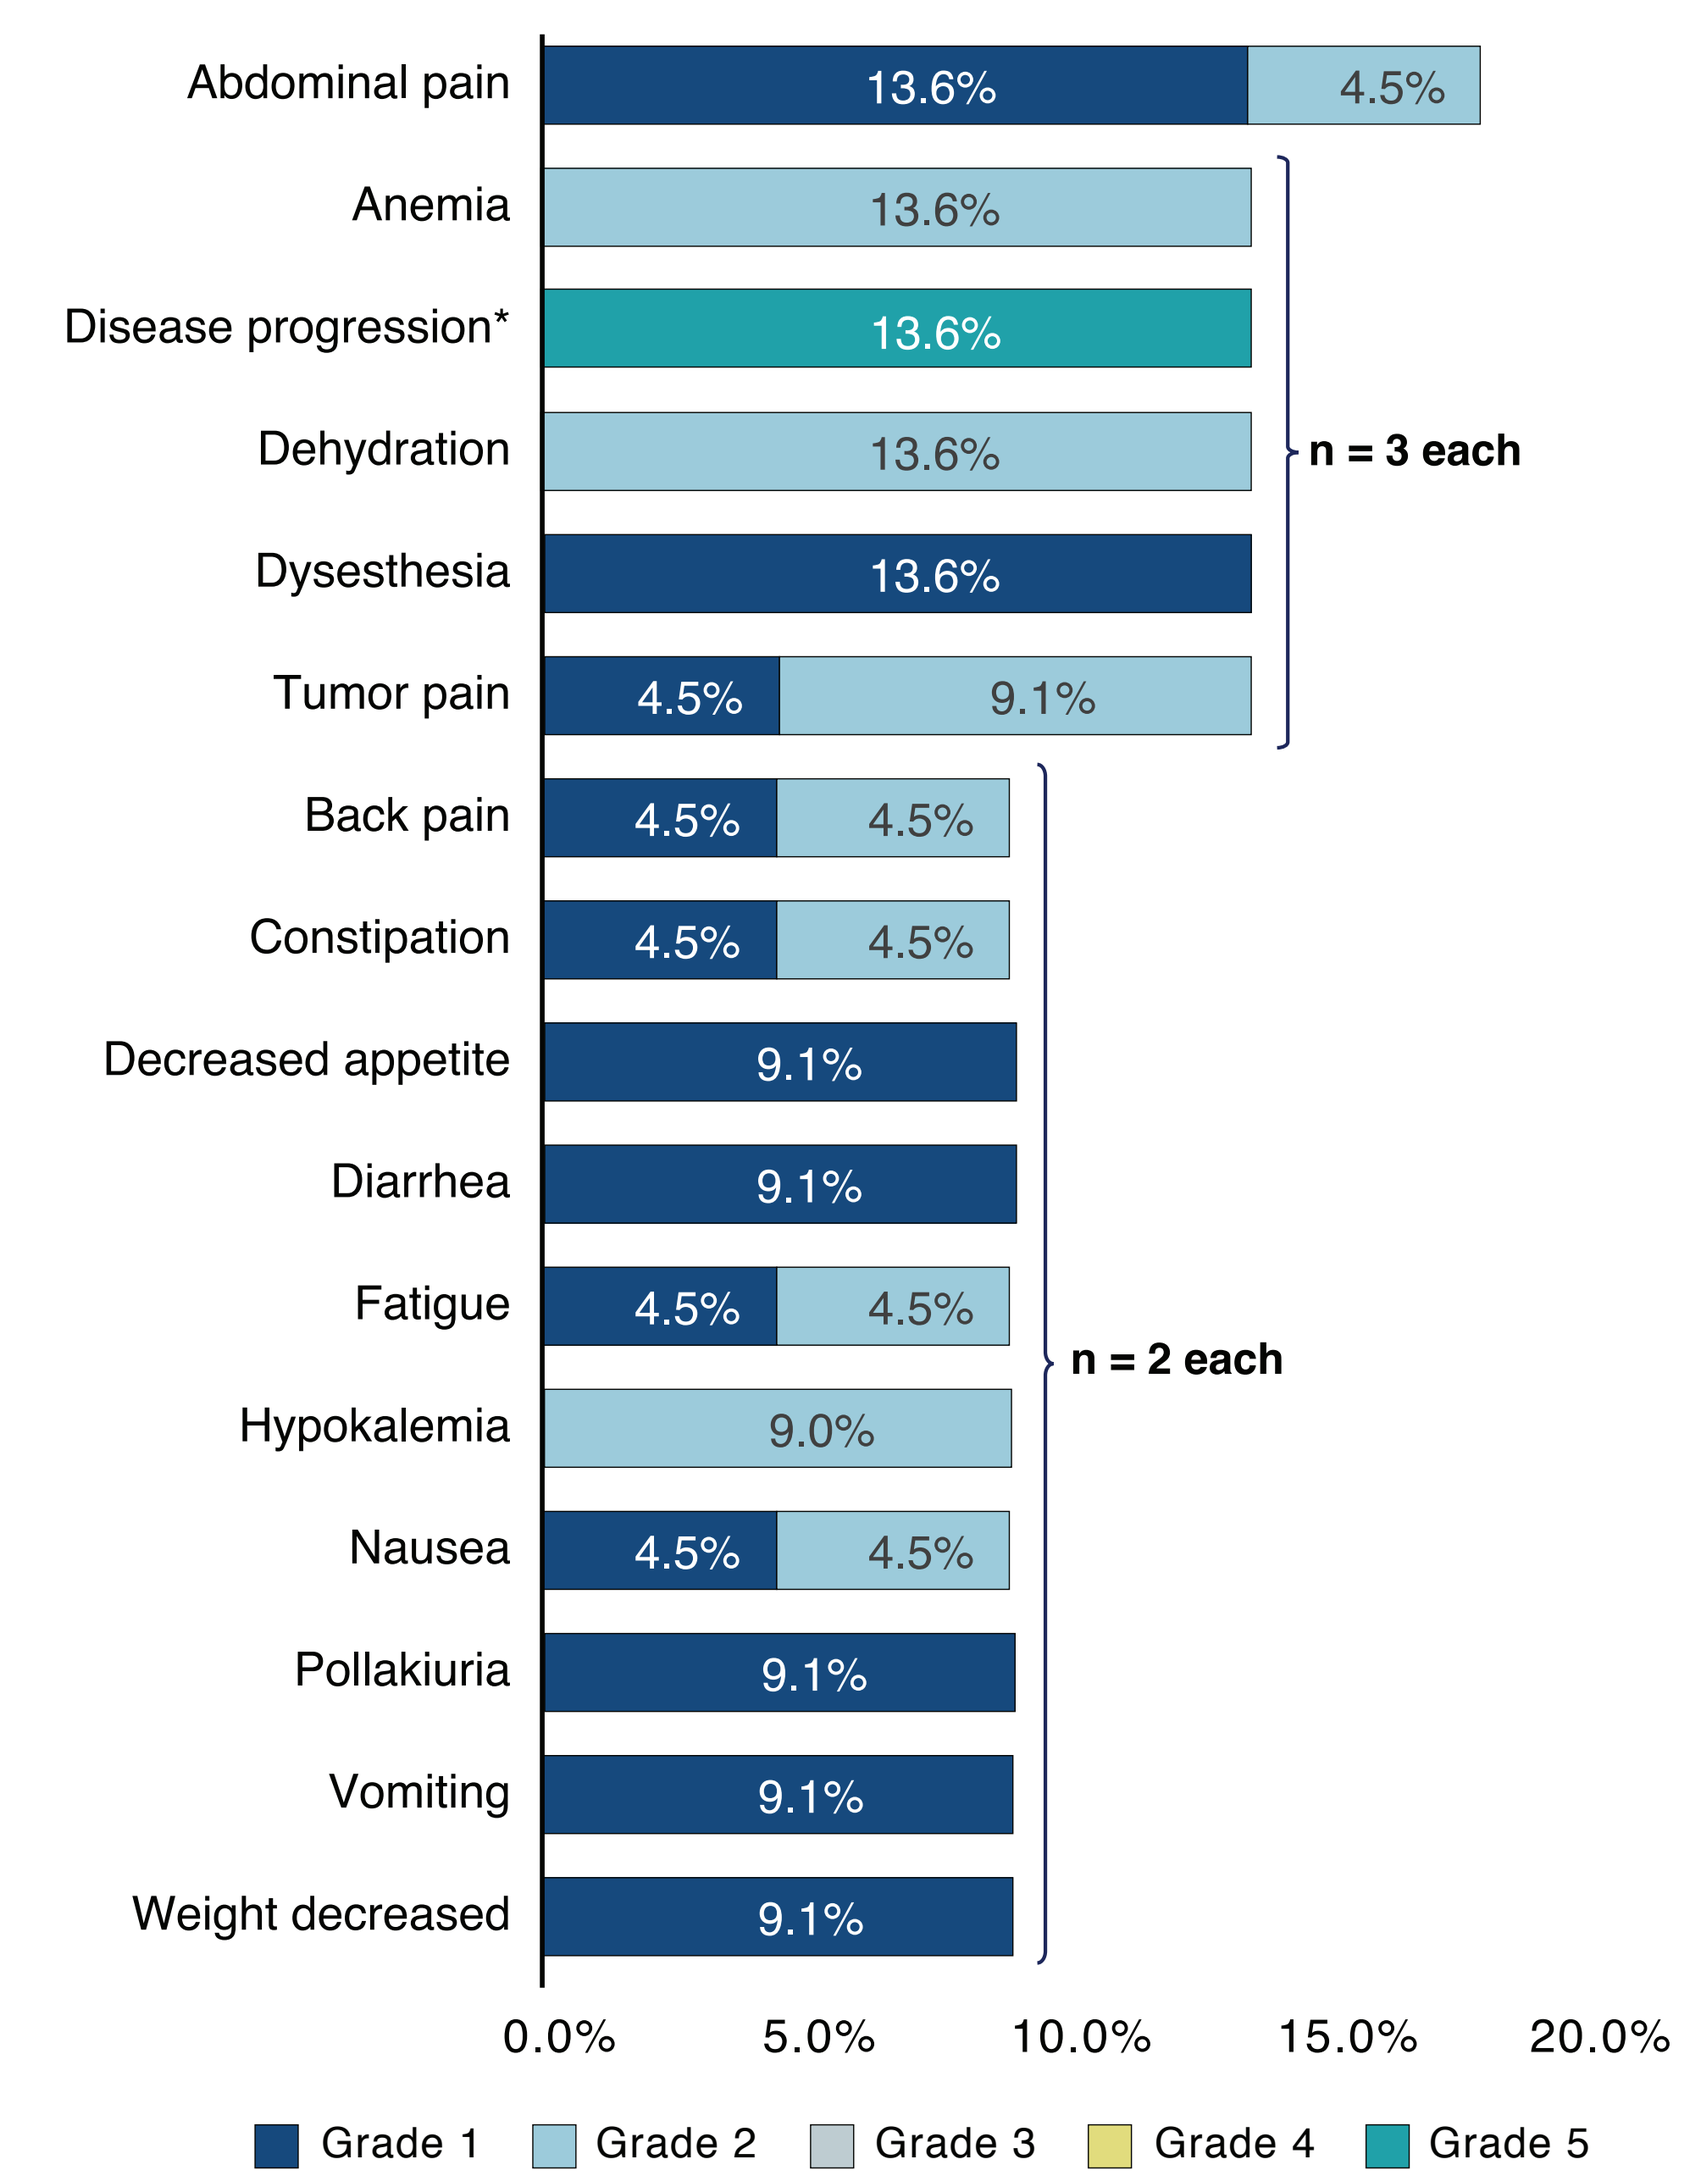
Abdominal pain 13.6% 4.5%
Anemia 13.6%
Disease progression* 13.6%
Dehydration 13.6%
Dysesthesia 13.6%
Tumor pain 4.5% 9.1%
n = 3 each
Back pain 4.5% 4.5%
Constipation 4.5% 4.5%
Decreased appetite 9.1%
Diarrhea 9.1%
Fatigue 4.5% 4.5%
Hypokalemia 9.0%
Nausea 4.5% 4.5%
Pollakiuria 9.1%
Vomiting 9.1%
Weight decreased 9.1%
n = 2 each
0.0% 5.0% 10.0% 15.0% 20.0%
Grade 1 Grade 2 Grade 3 Grade 4 Grade 5

Data as of 27 March 2020.
*Disease progression included disease progression and worsening pancreatic cancer.
TEAE, treatment-emergent adverse event

- Treatment-related AEs were reported in 9 patients, all were Grade 1; incidence of was not dose-dependent (**Figure 4**).
- No immune-related AEs were reported with Regimen A or B
- Two patients receiving Regimen B (GB1275 100 mg PO BID) reported Grade 1 fatigue and Grade 1 constipation considered related to both GB1275 and pembrolizumab

Figure 4. GB1275 Treatment-Related Adverse Events

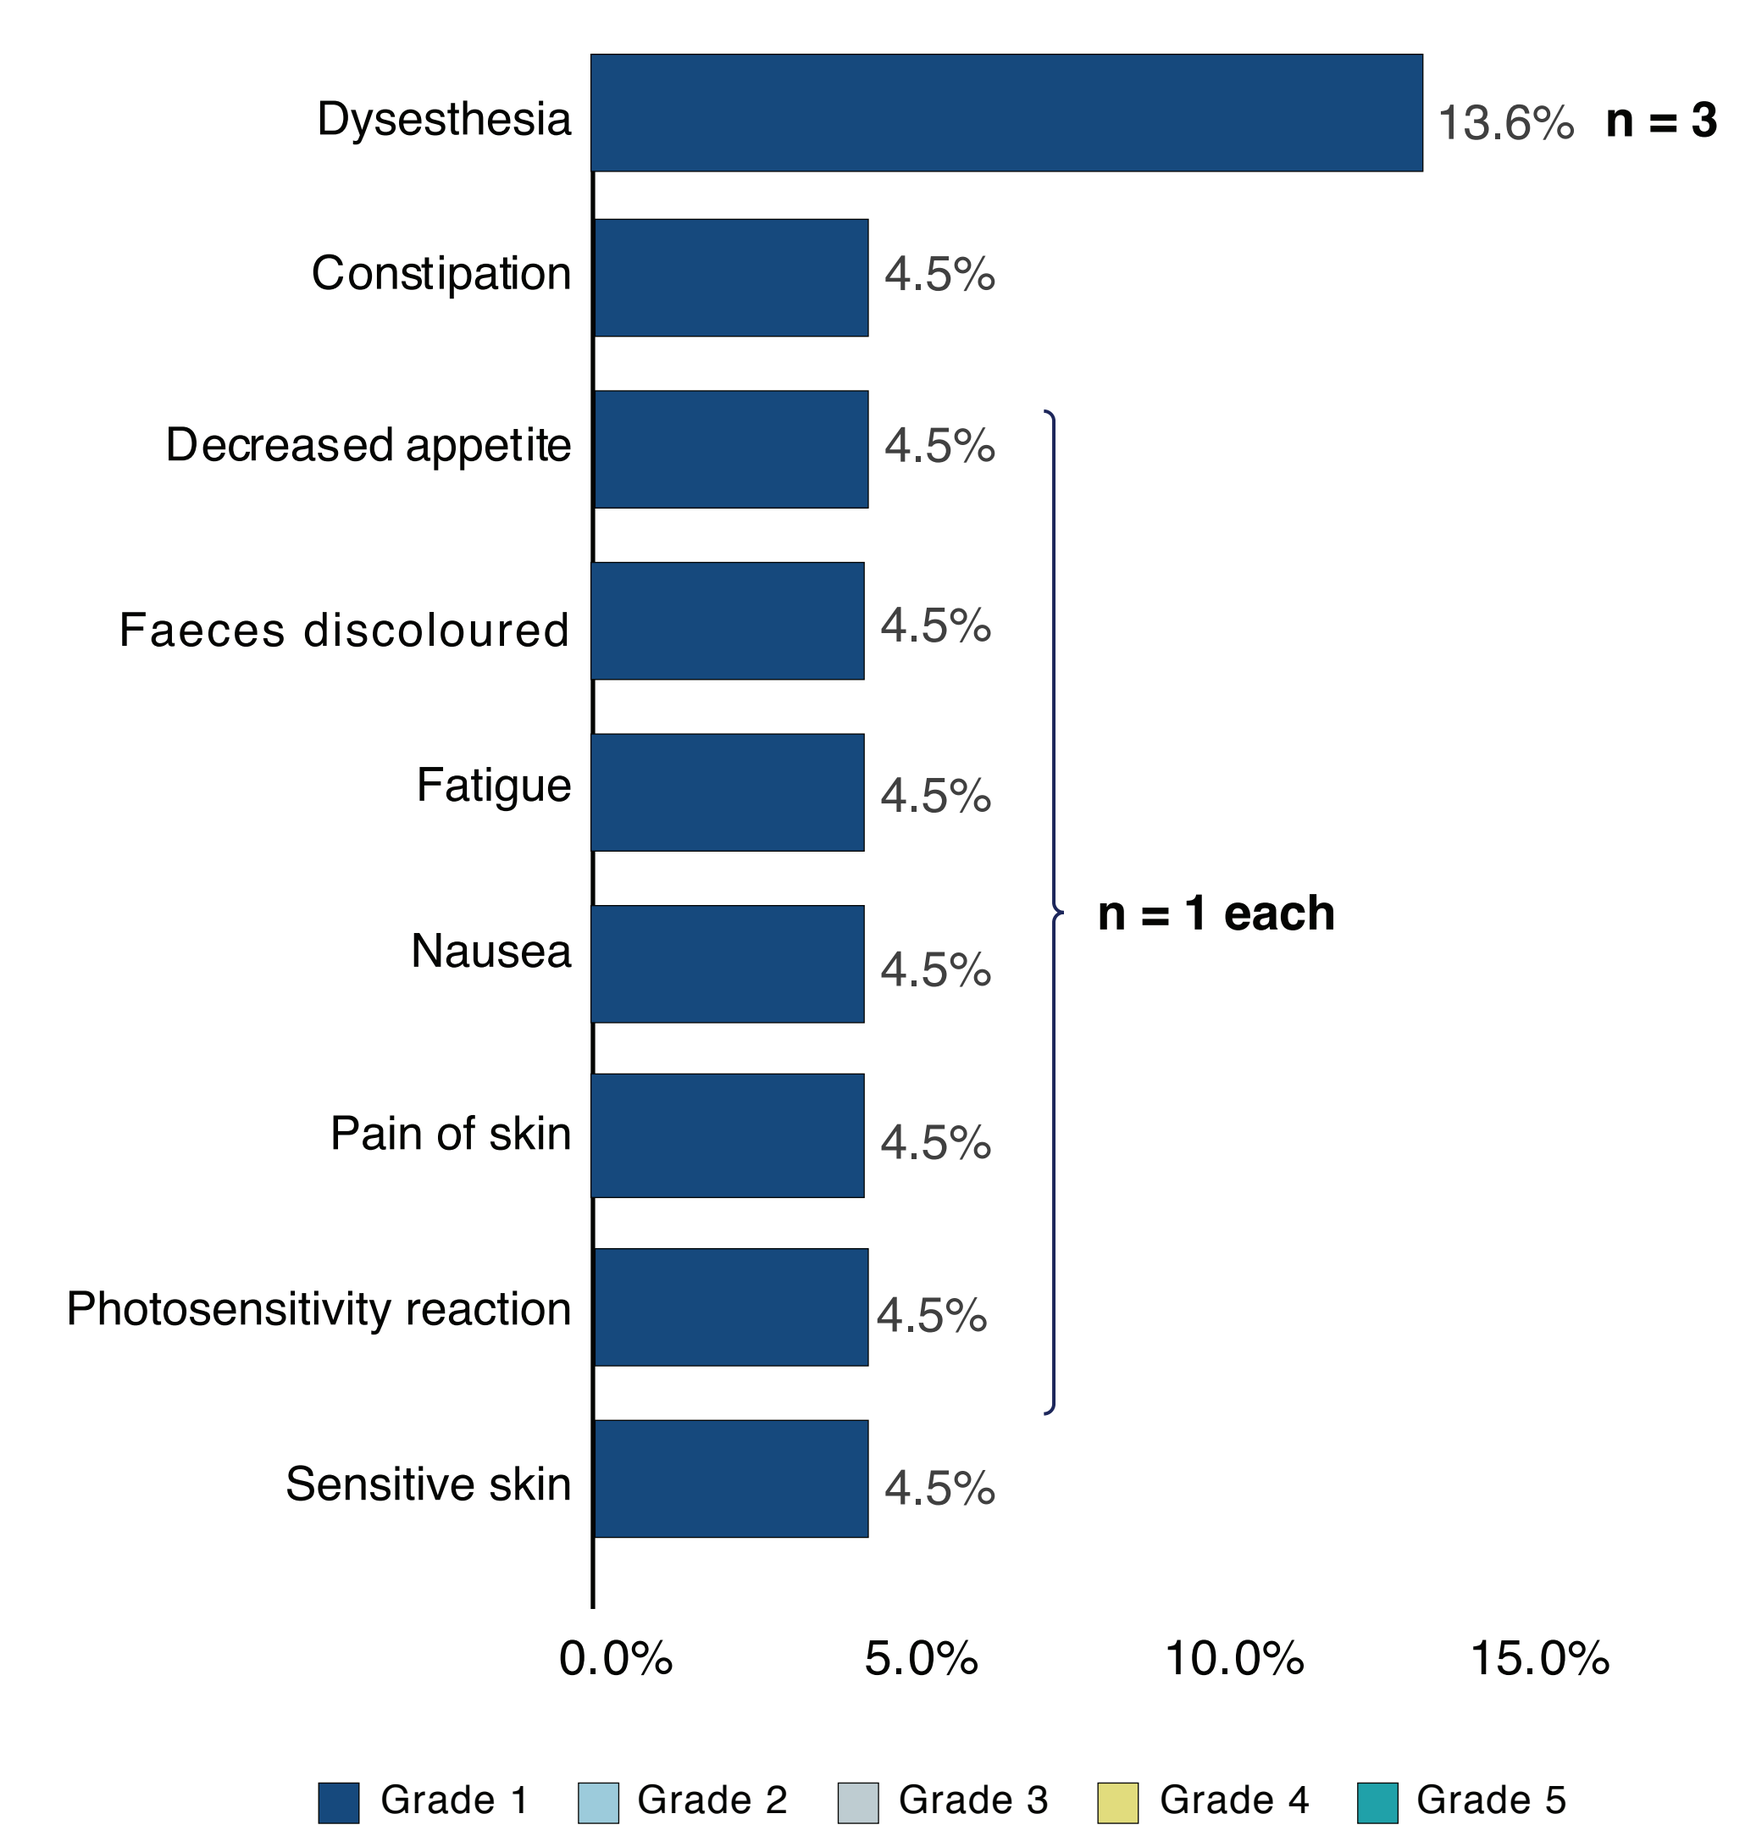
Dysesthesia 13.6% n = 3
Constipation 4.5%
Decreased appetite 4.5%
Faeces discoloured 4.5%
Fatigue 4.5%
Nausea 4.5%
Pain of skin 4.5%
Photosensitivity reaction 4.5%
Sensitive skin 4.5%
n = 1 each
0.0% 5.0% 10.0% 15.0%
Grade 1 Grade 2 Grade 3 Grade 4 Grade 5

Data as of 27 March 2020

Pharmacokinetics

- A dose-dependent increase in GB1275 plasma concentration was observed over doses of 100 mg to 800 mg PO BID
- The elimination half-life of GB1275 was estimated at ~7 hours
- The addition of pembrolizumab (Regimen B) at the GB1275 100 mg PO BID dose level did not substantially alter the PK profile of GB1275

Anti-tumor Activity

- To date, the best response observed is stable disease (**Figure 5**)

Figure 5. Exposure & RECIST Response for Regimens A & B

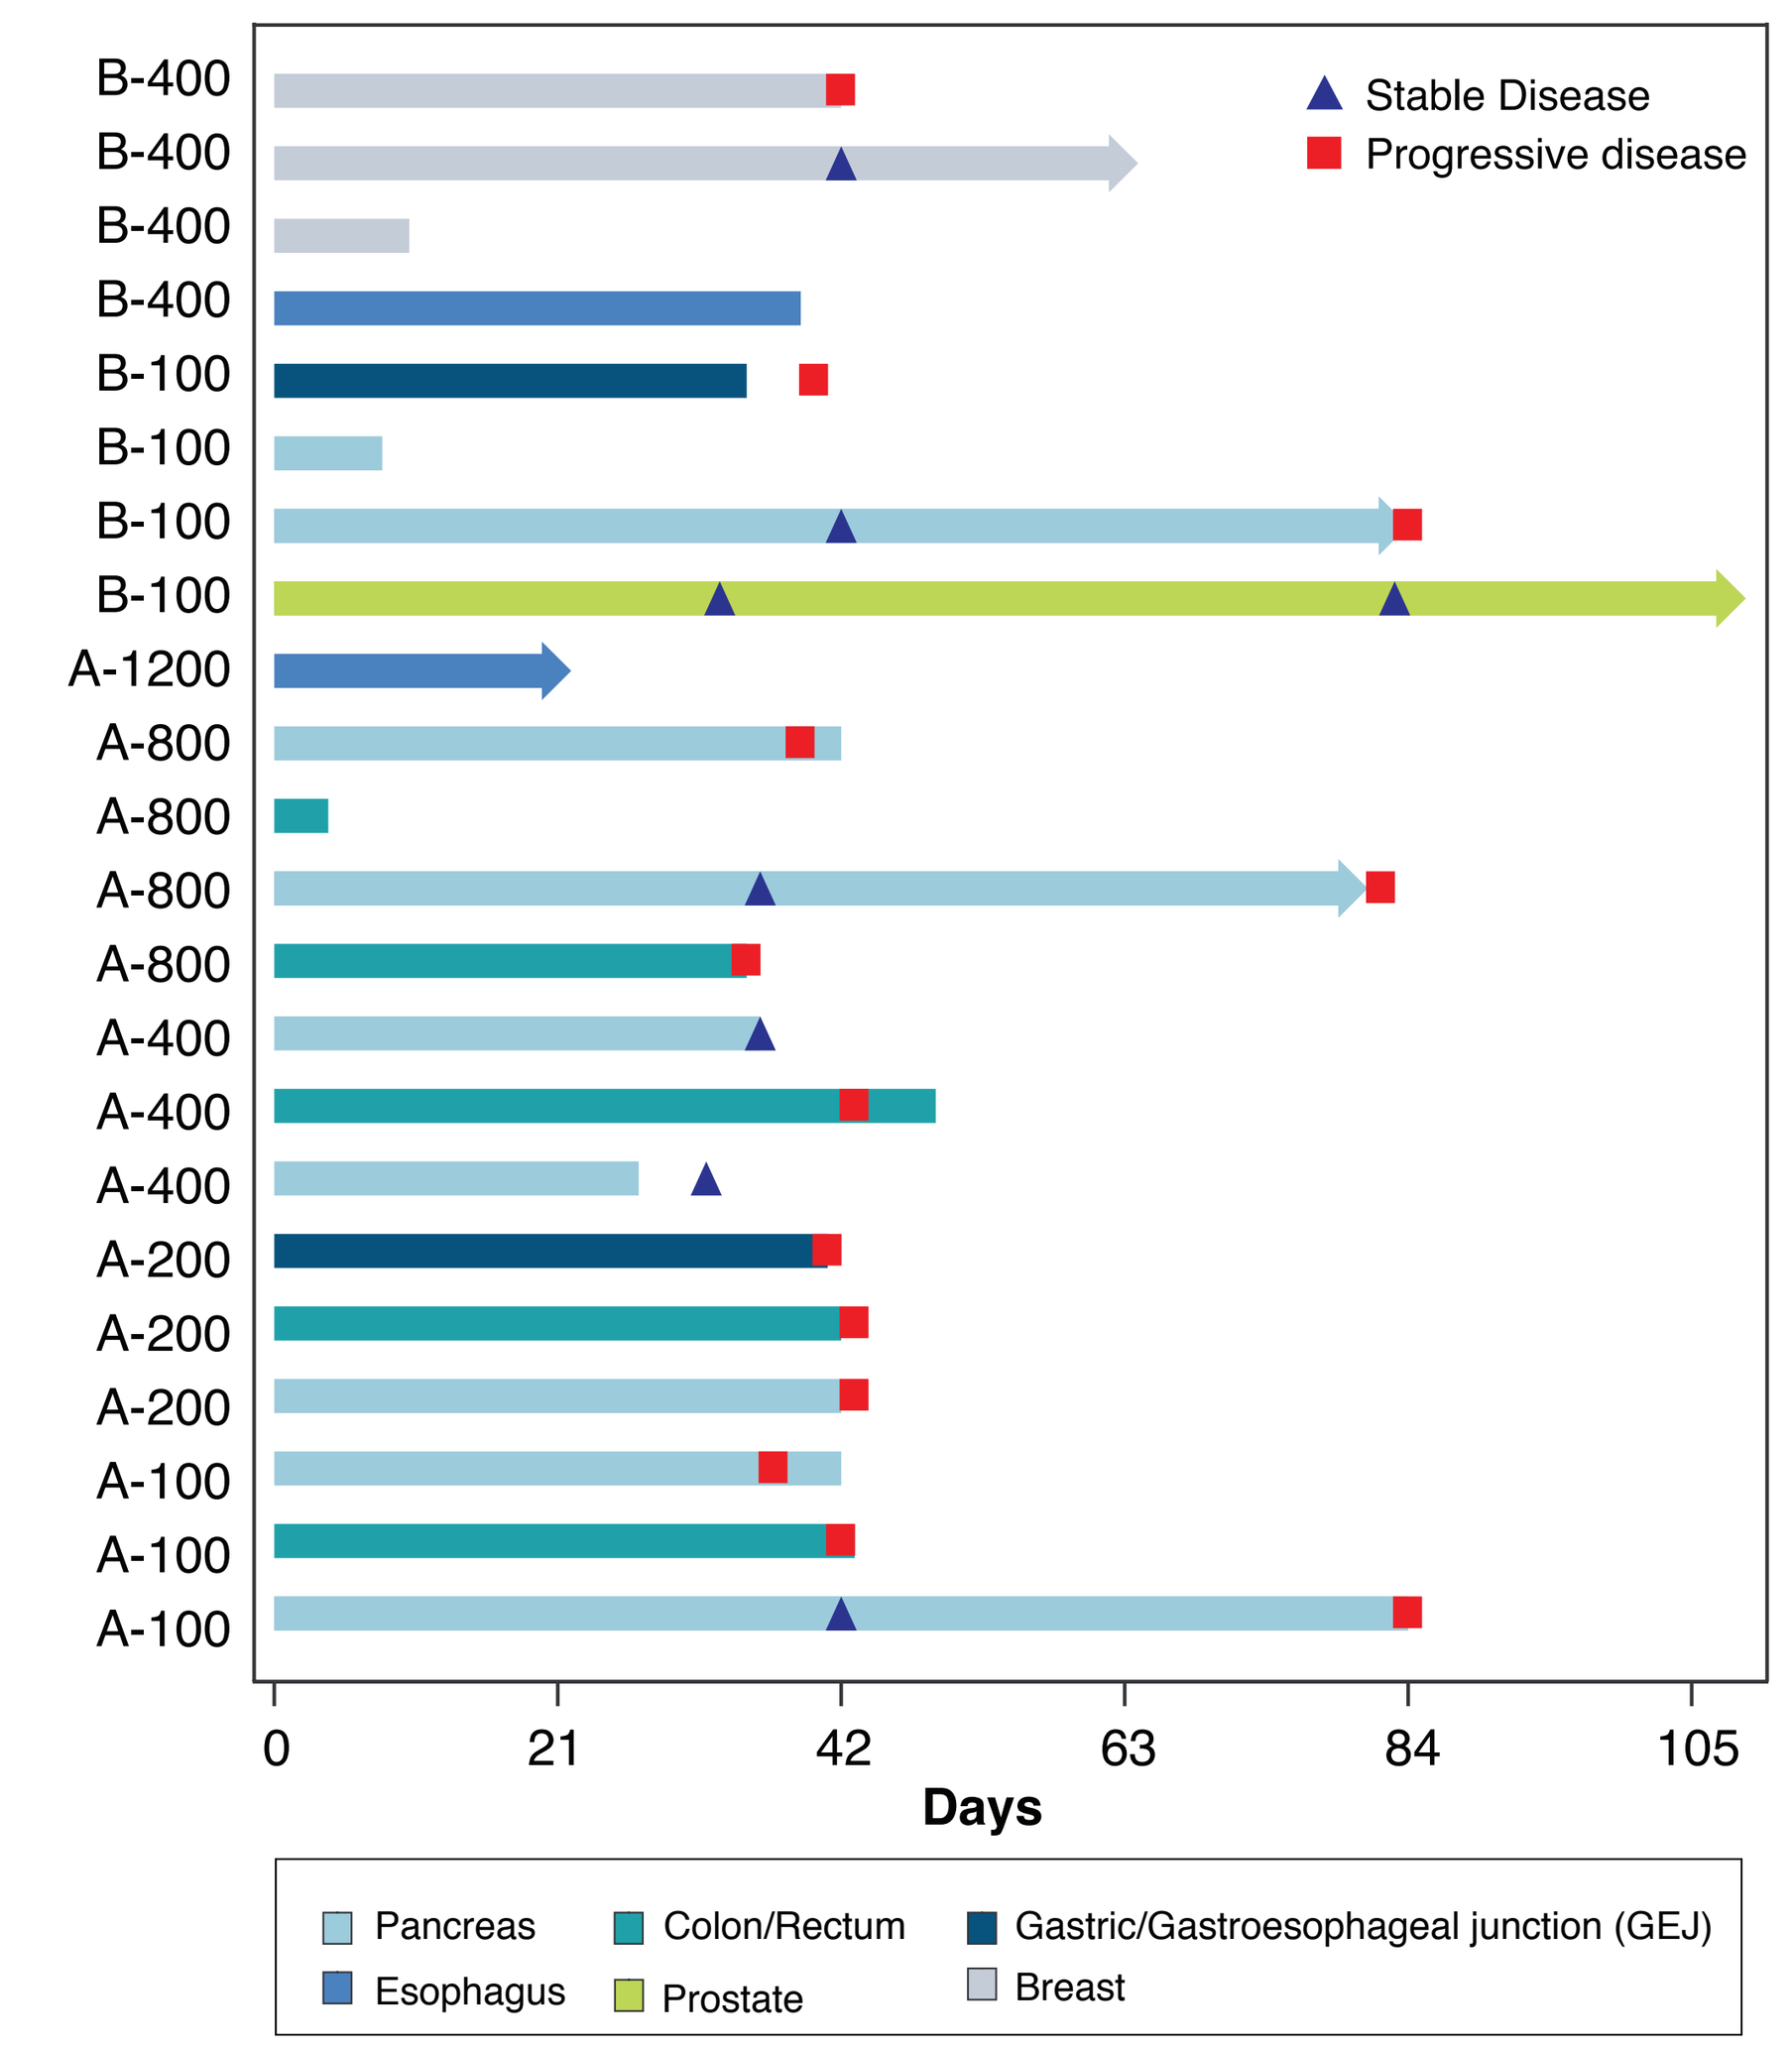
B-400
B-400
B-400
B-400
B-100
B-100
B-100
B-100
A-1200
A-800
A-800
A-800
A-800
A-400
A-400
A-400
A-200
A-200
A-200
A-100
A-100
A-100
Stable Disease
Progressive disease
0 21 42 63 84 105
Days
Pancreas
Esophagus
Colon/Rectum
Prostate
Gastric/Gastroesophageal junction (GEJ)
Breast

Data as of 27 March 2020.

- 79 year-old male with mCRPC, Gleason score 5 at diagnosis, tumor mutation burden (TMB) 4, blood BRCA1-m, prostate specific antigen (PSA) 3730 ng/mL at study entry, >10 lines of treatment, progressive disease to atezolizumab. Initiated Regimen B: GB1275 100 mg PO BID + pembrolizumab.
- After 8 weeks of treatment: stable disease, serum PSA 1790 ng/mL (a 52% decrease), maximum decrease in neutrophil-to-lymphocyte ratio (NLR): 52%, sustained for 4 cycles up to data cut. Subject continues on study treatment. **Figure 6** shows a potential difference in gene expression in this patient.

Figure 6. Heatmap of Per Patient Gene Expression Changes in Serial Blood Biopsies and Correlation With Tumor Type. Note the unique gene signature for the 79-year-old patient with mCRPC.

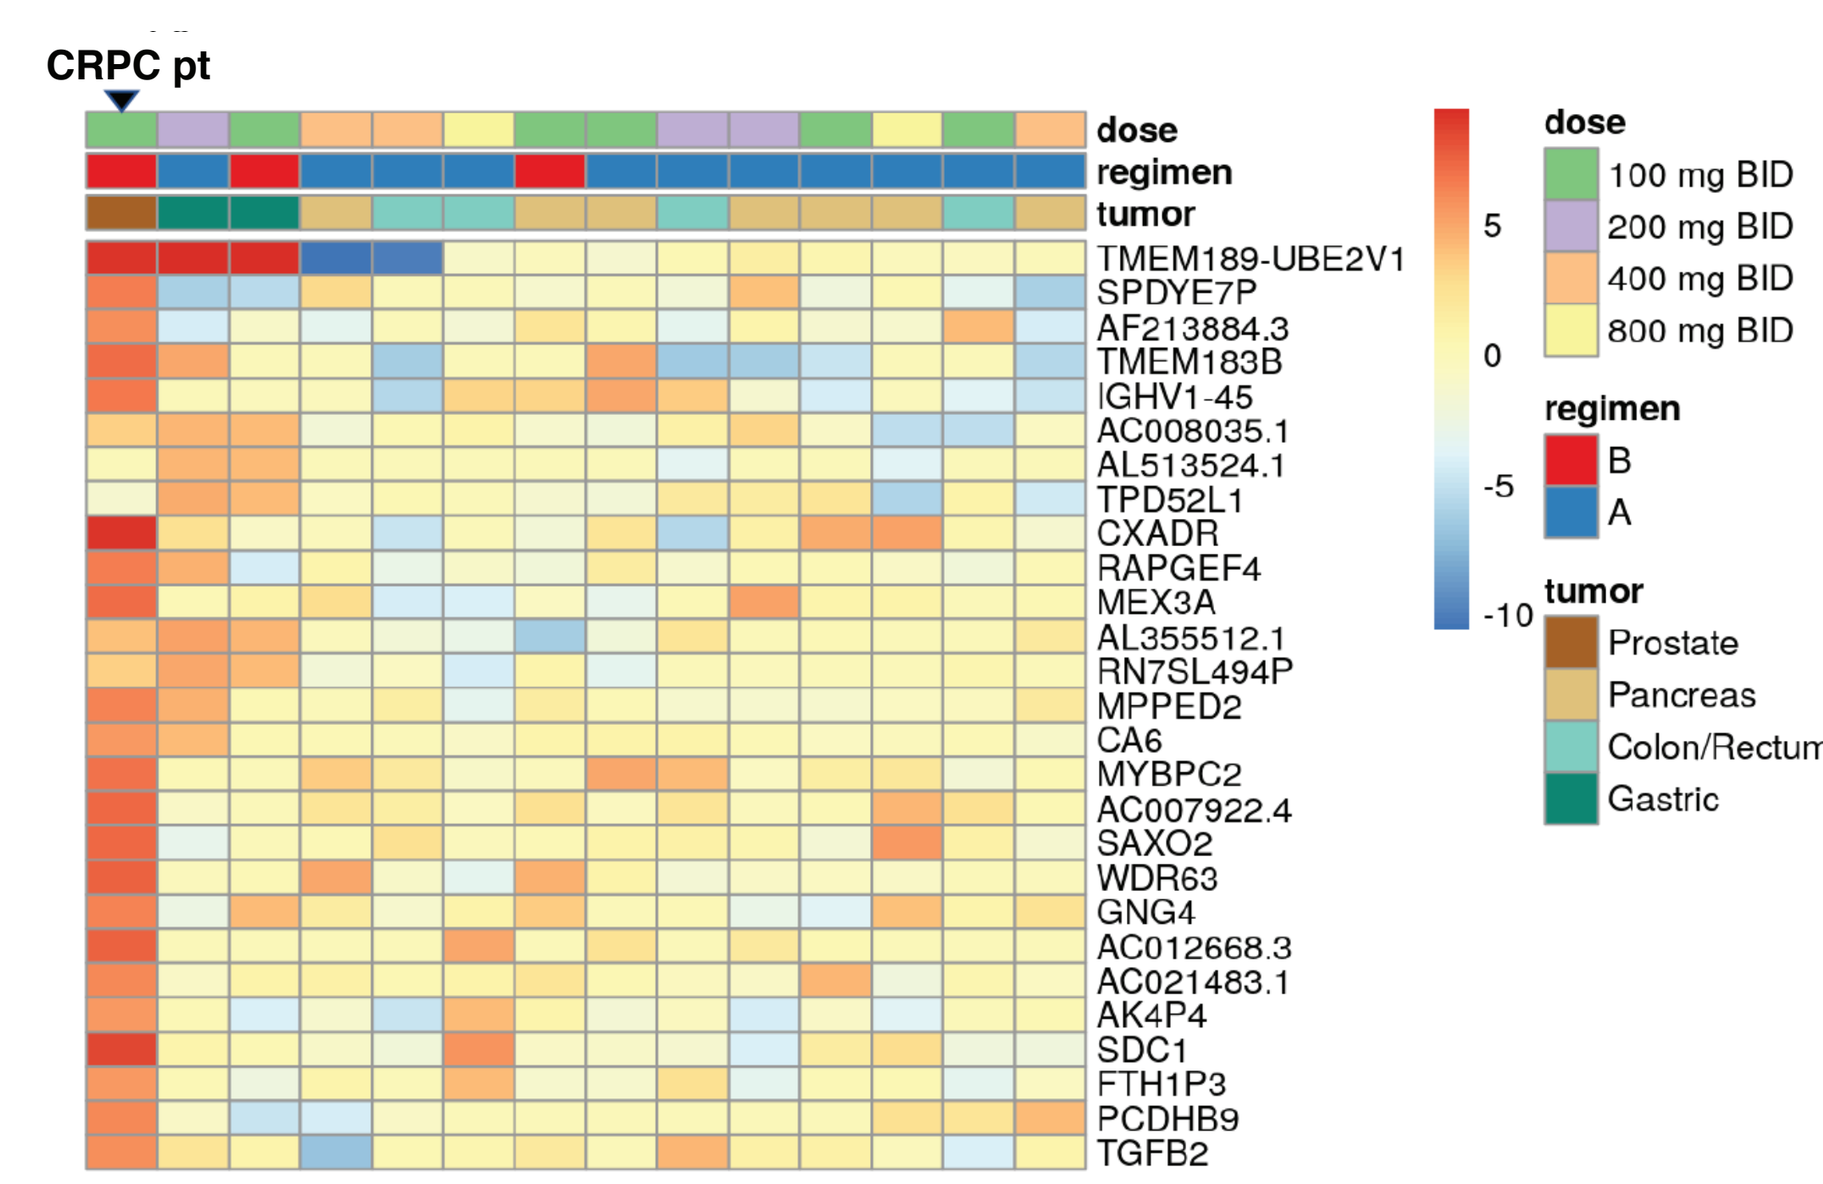
CRPC pt
dose
regimen
tumor
TMEM189-UBE2V1
SPDYE7P
AF213884.3
TMEM183B
IGHV1-45
AC008035.1
AL513524.1
TPD52L1
CXADR
RAPGEF4
MEX3A
AL355512.1
RN7SL494P
MPPED2
CA6
MYBPC2
AC007922.4
SAXO2
WDR63
GNG4
AC012668.3
AC021483.1
AK4P4
SDC1
FTH1P3
PCDHB9
TGFB2
5
0
-5
-10
dose
100 mg BID
200 mg BID
400 mg BID
800 mg BID
regimen
B
A
tumor
Prostate
Pancreas
Colon/Rectum
Gastric

Data as of 27 March 2020.

Biomarker Assessment

- Biomarker profiles differ with the addition of pembolizumab and with 800 mg dose (**Figure 7**)

Figure 7. Blood Transcriptomics: A. Regimen A (GB1275 Alone) vs Regimen B (GB1275 + Pembrolizumab) Cluster and B. Dose-dependent Cluster for Regimens A + B

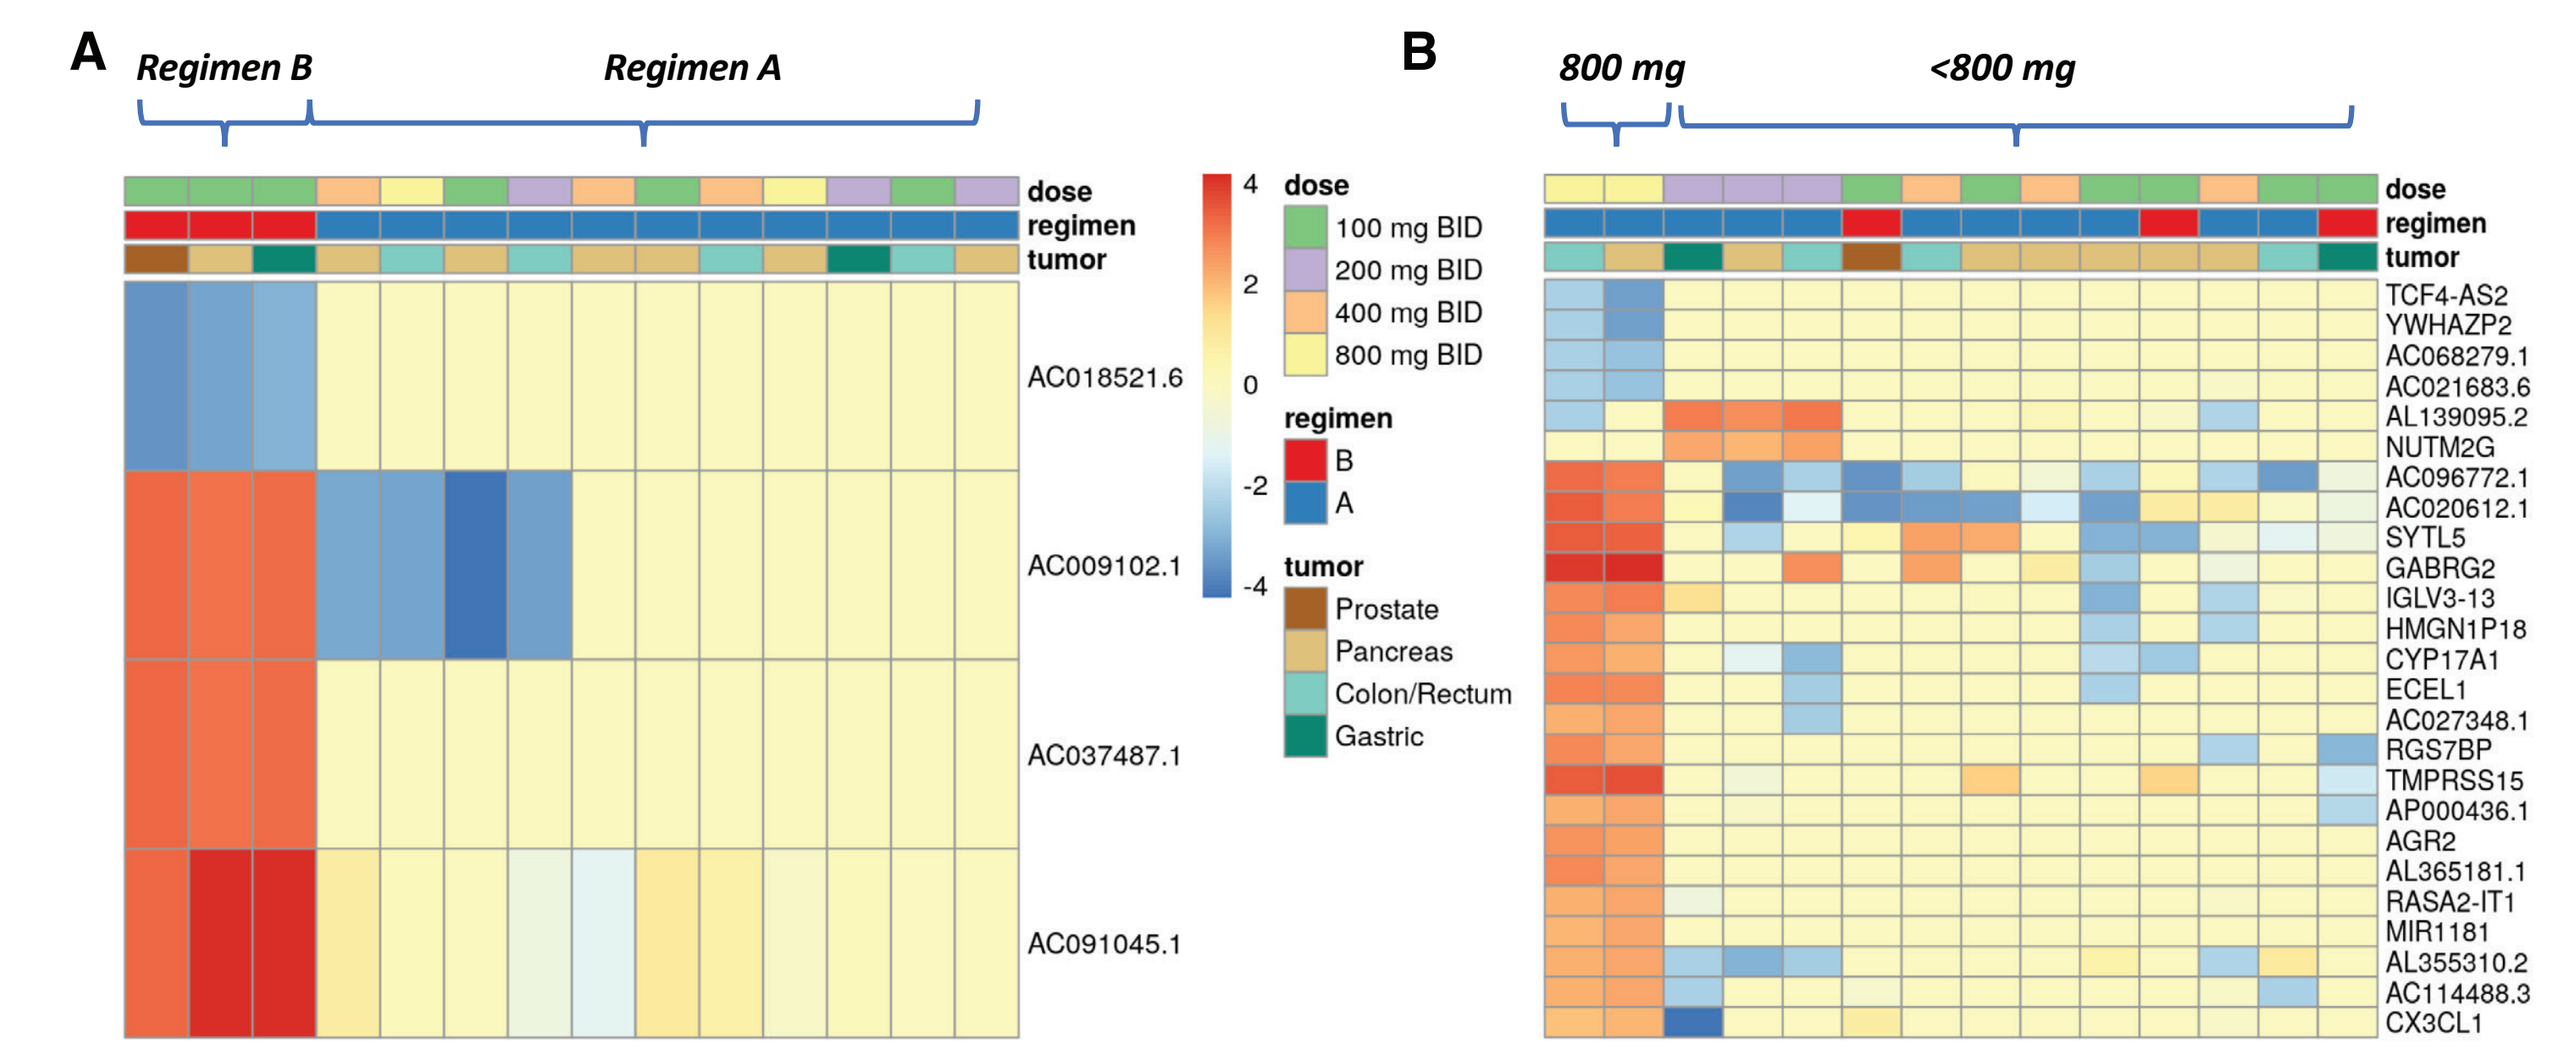
A
Regimen B
Regimen A
dose
regimen
tumor
AC018521.6
AC009102.1
AC037487.1
AC091045.1
4
2
0
-2
-4
dose
100 mg BID
200 mg BID
400 mg BID
800 mg BID
regimen
B
A
tumor
Prostate
Pancreas
Colon/Rectum
Gastric
B
800 mg
<800 mg
dose
regimen
tumor
TCF4-AS2
YWHAZP2
AC068279.1
AC021683.6
AL139095.2
NUTM2G
AC096772.1
AC020612.1
SYTL5
GABRG2
IGLV3-13
HMGN1P18
CYP17A1
ECEL1
AC027348.1
RGS7BP
TMPRSS15
AP000436.1
AGR2
AL365181.1
RASA2-IT1
MIR1181
AL355310.2
AC114488.3
CX3CL1

Data as of 27 March 2020.

- Modulation of peripheral MDSCs was observed in patients with serial samples (**Figure 8**)

Figure 8. Percent gMDSCs and mMDSCs in the Regimen B 400 mg BID Cohort

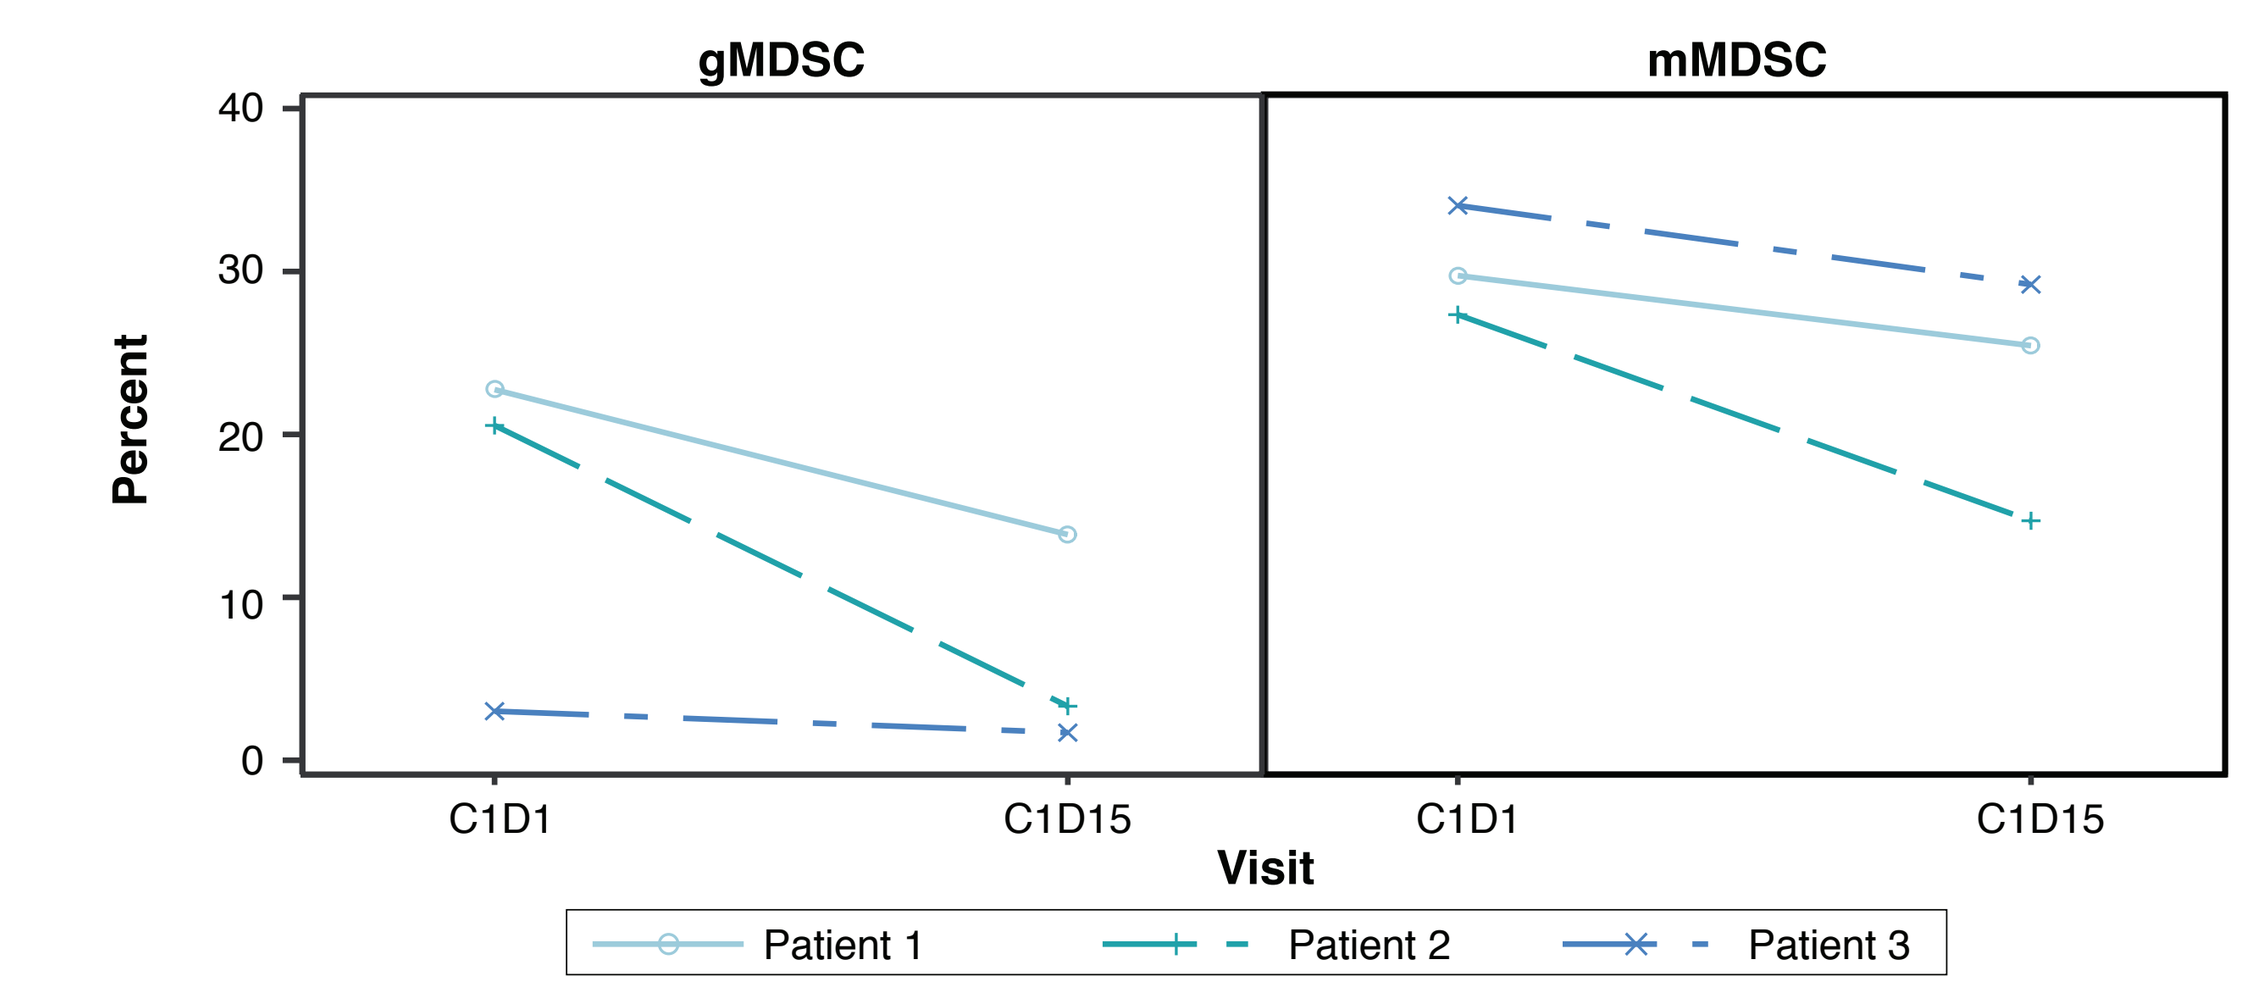
gMDSC
mMDSC
Percent
40
30
20
10
0
C1D1
C1D15
C1D1
C1D15
Visit
Patient 1
Patient 2
Patient 3

Data as of 27 March 2020.
mMDSC, monocytic myeloid-derived suppressor cells; gMDSC, granulocytic myeloid-derived suppressor cells

CONCLUSIONS

- Clinical safety data to date suggest that GB1275 monotherapy (up to 1200 mg PO BID) and in combination with pembrolizumab (up to 400 mg PO BID) is well tolerated
- The maximum tolerated dose of GB1275 has not been reached
- GB1275's elimination half-life of approximately 7 hours supports BID dosing
- Modulation of peripheral MDSCs was observed in patients treated with Regimens A and B. Additional biomarker analysis in blood and tumor biopsies is ongoing.
- Preliminary anti-tumor activity was observed in one checkpoint inhibitor-resistant patient with mCRPC, supported by PSA and NLR data

REFERENCES

1. Fleming V, et al. *Front Immunol.* 2018; 9:398.
2. Kumar V, et al. *Trends Immunol.* 2016; 37(3):208-220.
3. Mantovani A, et al. *Trends Immunol.* 2002; 23(11):549-555.
4. Panni R, et al. *Sci Transl Med.* 2019; 11: eaau9240.

ACKNOWLEDGMENTS

This study is funded by GB006, Inc, a wholly owned subsidiary of Gossamer, Bio, Inc.

The authors gratefully acknowledge the support of Jean Marie Bruey in the review of this poster.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Prepared for ASCO20 Virtual Scientific Program May 29 – June 2, 2020.

For questions about this poster, contact Drew Rasco at Drew.Rasco@startsa.com