1036

INTERIM RESULTS FROM THE PHASE 1B AND PHASE 2 TORREY OPEN-LABEL EXTENSION STUDY OF SERALUTINIB IN PULMONARY ARTERIAL HYPERTENSION (PAH)

TORREY STUDY

Channick RN[1], Sahay S[2], Escribano Subías P[3], Zolty R[4], Kingrey JF[5], Penn B[6], Sobol I[7], Sood N[8], Benza RL[9], Frantz RP[10], Ghofrani HA[11], Hemnes AR[12], Howard, LS[13], McLaughlin VV[14], Sitbon O[15], Vachiéry JL[16], Zamanian RT[17], Cravets M[18], Roscigno RF[18], Mottola D[18], Parsley E[18], Aranda R[18], Zisman LS[18], Chin KM[19]

[1]UCLA Medical Center, Los Angeles, CA, USA; [2]Houston Methodist Hospital/Weill Cornell Medicine, Houston, TX, USA; [3]University Hospital 12 de Octubre, Complutense University, Madrid, Spain; [4]University of Nebraska Medical Center, Omaha, NE, USA; [5]INTEGRIS Health Pulmonary Hypertension Center of Oklahoma, Oklahoma City, OK, USA; [6]University of Utah Health, Salt Lake City, UT, USA; [7]New York Presbyterian/Weill Cornell Medical Center, New York, NY, USA; [8]University of California Davis Medical Center, Sacramento, CA, USA; [9]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [10]Mayo Clinic, Rochester, MN, USA; [11]Universities of Giessen and Marburg Lung Center (UGMLC), Institute for Lung Health (ILH); Cardio-Pulmonary Institute (CPI); Member of the German Center for Lung Research (DZL), Giessen, Germany; Department of Medicine, Imperial College, London, UK; [12]Vanderbilt University, Nashville, TN, USA; [13]Imperial College Healthcare NHS Trust, Hammersmith Hospital, London, UK; [14]University of Michigan, Ann Arbor, MI, USA; [15]Université Paris-Saclay/Hôpital Bicêtre, Le Kremlin-Bicêtre, France; [16]Université Libre de Bruxelles, HUB – Hôpital Erasme, Brussels, Belgium; [17]Stanford University School of Medicine, Stanford, CA, USA; [18]Gossamer Bio, Inc., San Diego, CA, USA; [19]UT Southwestern Medical Center, Dallas, TX, USA

Presented at the PHA 2024 International PH Conference
Indianapolis, IN
August 15-18, 2024

Background

- Seralutinib, an inhaled investigational medicine, works by blocking proteins that contribute to PAH
- Seralutinib is a fine dry powder administered by a pocket-sized inhaler device (**Figure**)
- Participants from a phase 1b study and those who completed the phase 2 TORREY study[1] had the option to enroll in an open-label extension (OLE) study,[2] which further evaluates the safety, tolerability, and efficacy of seralutinib

Learn more about seralutinib and how it works.

See poster 1064 for more information on the TORREY study.

Pocket-sized Dry-powder Inhaler.

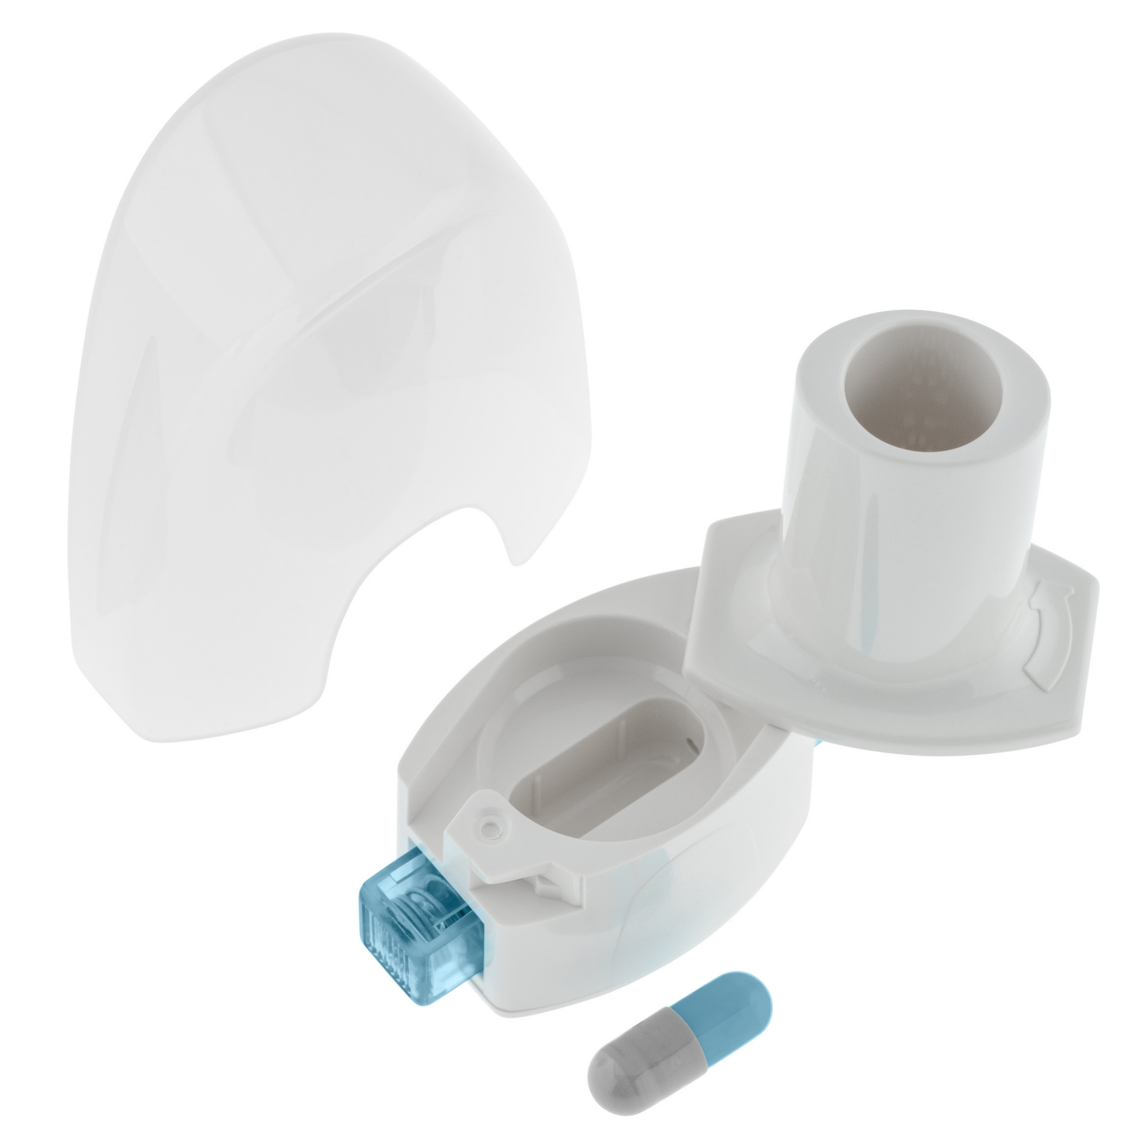

How was the study done?

Figure 1. OLE Study Design.

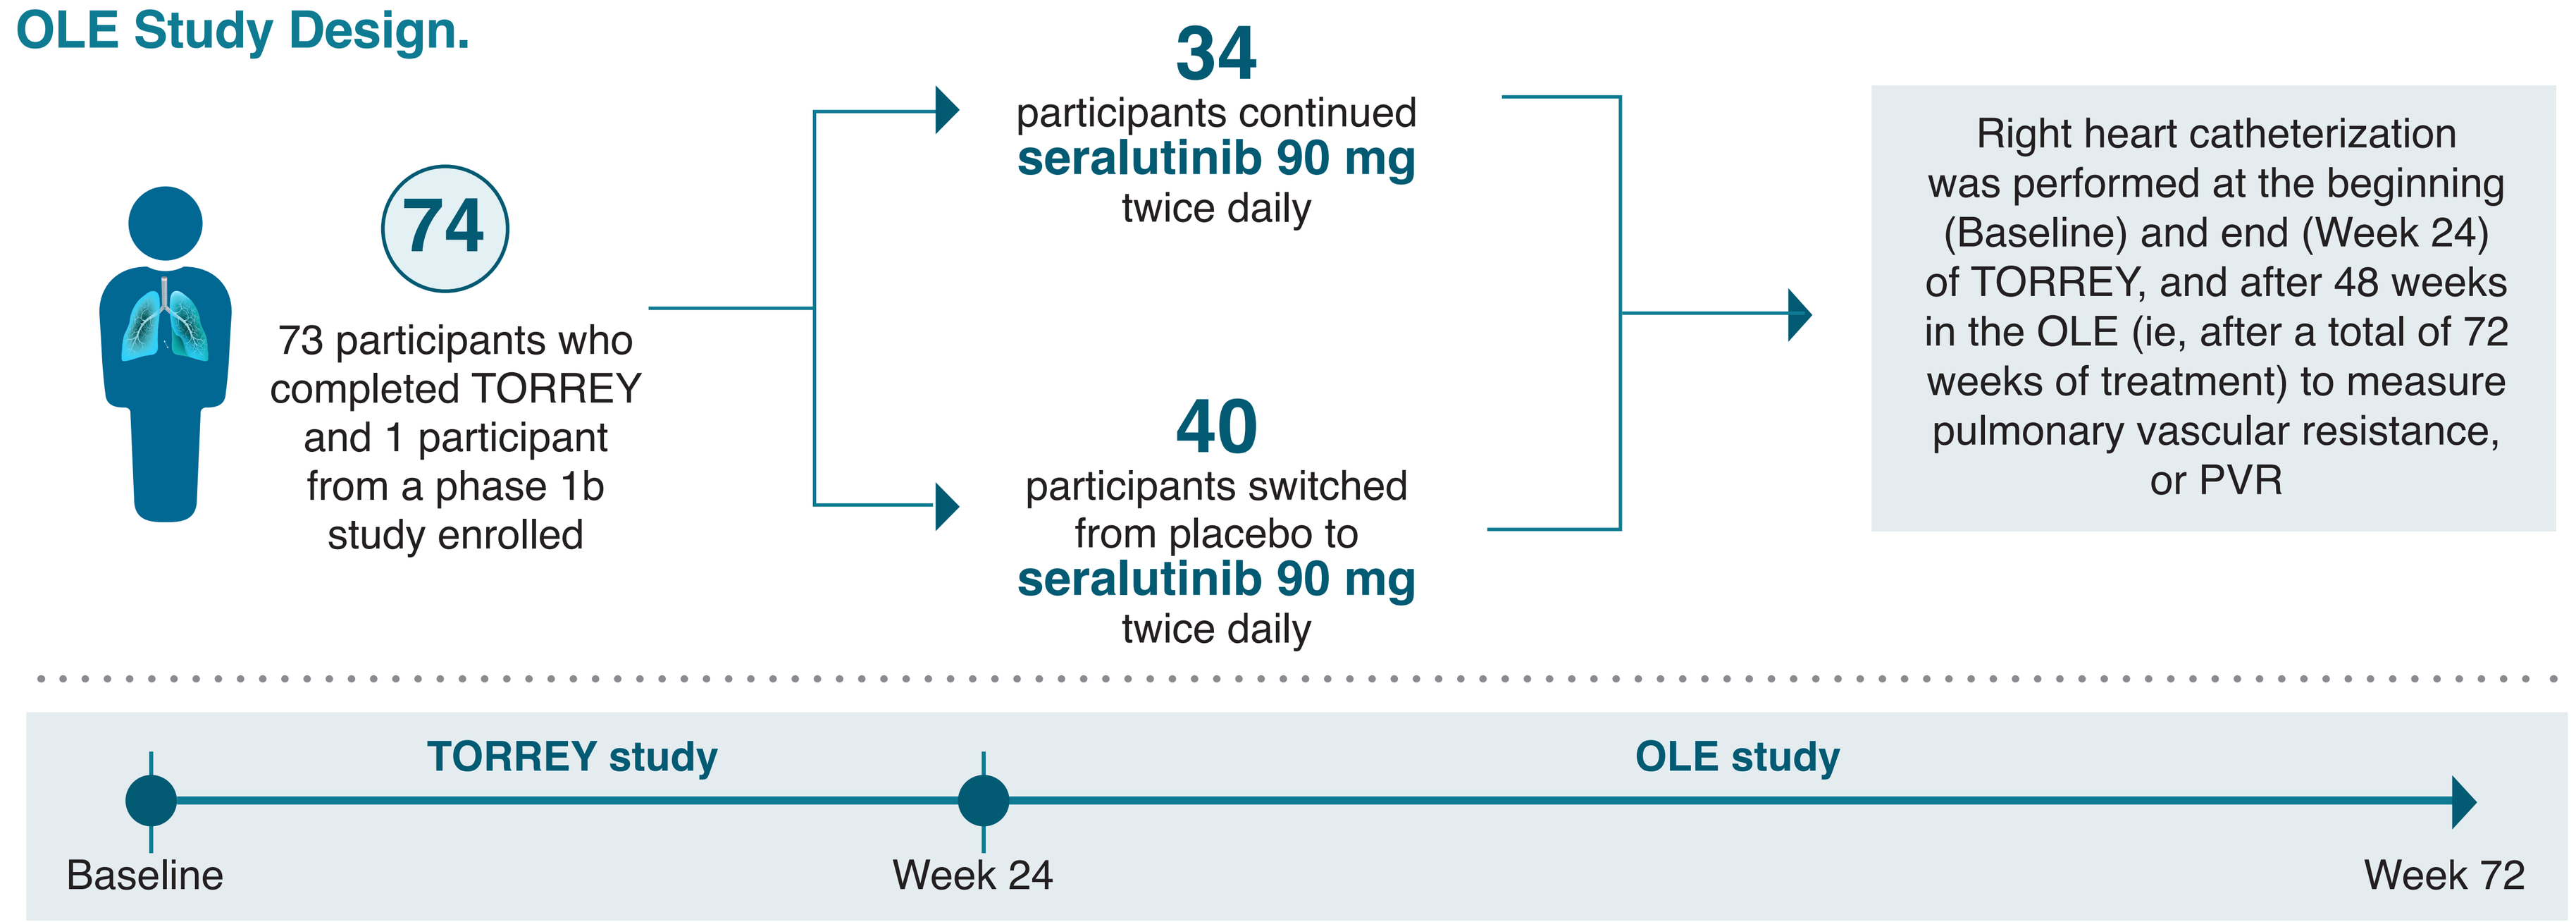
74
73 participants who completed TORREY and 1 participant from a phase 1b study enrolled
34
participants continued
seralutinib 90 mg
twice daily
40
participants switched from placebo to
seralutinib 90 mg
twice daily
Right heart catheterization was performed at the beginning (Baseline) and end (Week 24) of TORREY, and after 48 weeks in the OLE (ie, after a total of 72 weeks of treatment) to measure pulmonary vascular resistance, or PVR
TORREY study
OLE study
Baseline
Week 24
Week 72

Who participated in this study?

Figure 2. Participant Characteristics at the Start of the OLE (N=74). Numbers shown are averages ("means") or %.

AGE AT DIAGNOSIS
42 years

TIME SINCE PAH DIAGNOSIS
8.5 years

BACKGROUND PAH TREATMENT
Double/triple therapy:
39%/57%
Parenteral prostacyclins/PRA:
46%

WHO FUNCTIONAL CLASS
I: 7% II: 57% III: 31% IV: 4%

PVR
647 dynes*s/cm⁵

6-MINUTE WALK DISTANCE
415 meters

PAH, pulmonary arterial hypertension; PRA, prostacyclin receptor agonist; PVR, pulmonary vascular resistance; WHO, World Health Organization.

What did the study show?

- Seralutinib was generally well tolerated during the OLE
- Increased levels of liver enzymes can be a sign of stress on the liver. A small number of participants (6.8%) taking seralutinib in the TORREY and OLE studies experienced an increase in levels above average
- No new side effects were observed during the OLE, including the serious side effects seen with other tyrosine kinase inhibitors such as oral imatinib

Table 1. Most Common Side Effects Seen in the OLE, by Number and Percentage of Participants in Both Groups With Each Listed Side Effect.
"Most common" means side effect in 10% or more of participants.

Side effect	Total (N=74)		
Participants with a side effect, n (%)	71 (95.9%)		
Headache	19 (25.7%)	**Nausea**	13 (17.6%)
Cough	18 (24.3%)	**Nasopharyngitis**	10 (13.5%)
COVID-19	17 (23.0%)	**Fatigue**	8 (10.8%)
Diarrhea	15 (20.3%)	**Pyrexia**	8 (10.8%)
Dyspnea	13 (17.6%)	**Rash**	8 (10.8%)

Dyspnea, shortness of breath; nasopharyngitis, runny nose, sneezing, and coughing; pyrexia, fever.

Figure 3. Pulmonary Vascular Resistance (PVR). During the OLE, PVR continued to decrease in participants who received seralutinib to Week 72 (the continued-seralutinib group) and decreased in participants who received placebo to Week 24, then switched to seralutinib to Week 72 (the placebo-crossed group).

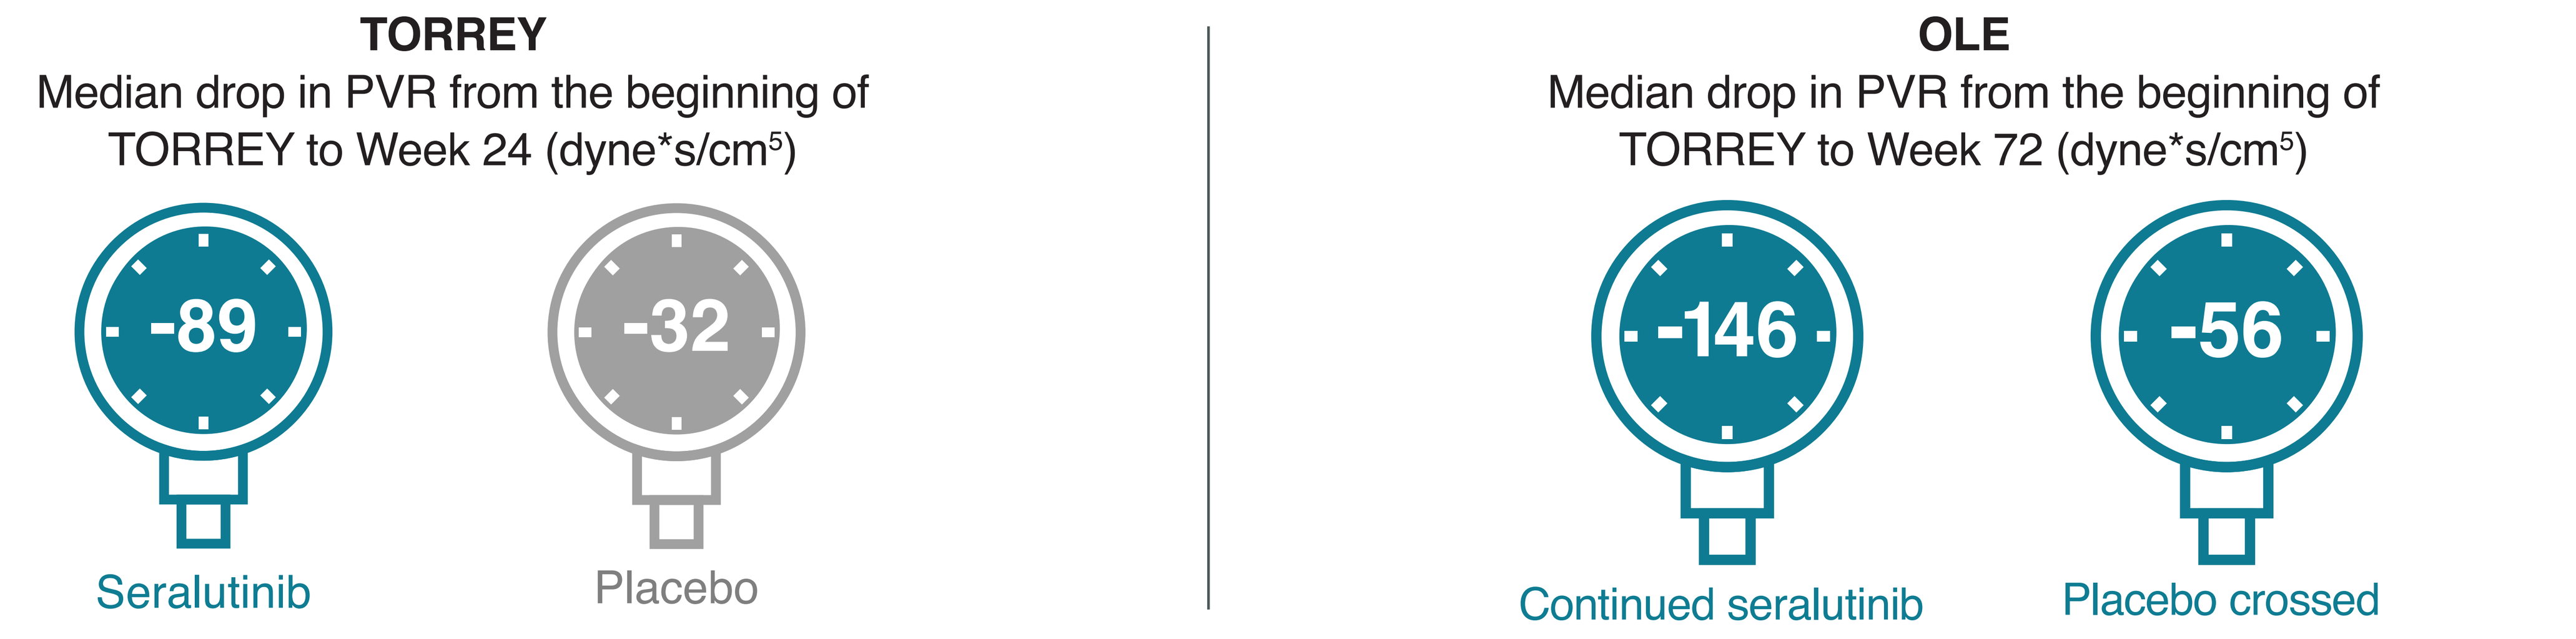
TORREY
Median drop in PVR from the beginning of TORREY to Week 24 (dyne*s/cm⁵)
-89
Seralutinib
-32
Placebo
OLE
Median drop in PVR from the beginning of TORREY to Week 72 (dyne*s/cm⁵)
-146
Continued seralutinib
-56
Placebo crossed

A drop, expressed by a negative number, indicates a reduction of pressure in pulmonary blood vessels, which means that it became easier for the blood to flow from the heart to the lungs. Values are from 27 participants per group who had a RHC procedure done at the beginning of TORREY, after 24 weeks, and after 72 weeks.

Figure 4. 6-minute Walk Distance (6MWD), a Measure of Participants' Exercise Capacity, Increased in Both the Group That Continued on Seralutinib and the Group That Crossed Over to Seralutinib From Placebo in the OLE.

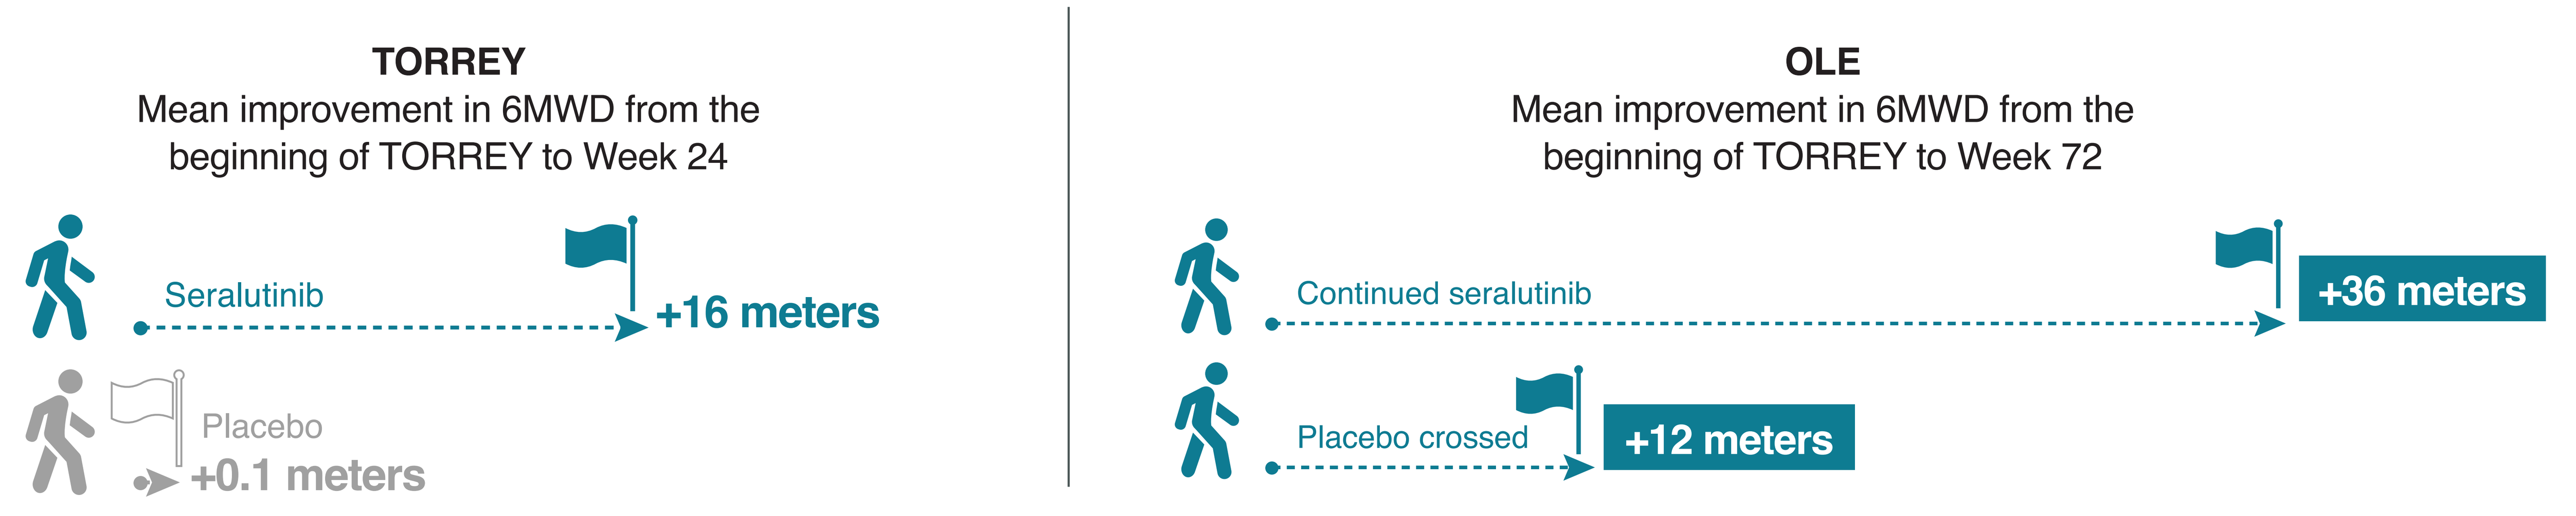
TORREY
Mean improvement in 6MWD from the beginning of TORREY to Week 24
Seralutinib
+16 meters
Placebo
+0.1 meters
OLE
Mean improvement in 6MWD from the beginning of TORREY to Week 72
Continued seralutinib
+36 meters
Placebo crossed
+12 meters

WHAT DO THE RESULTS OF THIS STUDY MEAN?

- **Seralutinib was well tolerated** over a period of up to 72 weeks
- **PVR and exercise capacity continued to improve** in some participants who received inhaled seralutinib in TORREY and the OLE
- These positive results encourage **further development of seralutinib** as a potential new treatment for PAH
- The **phase 3 PROSERA study** of inhaled seralutinib in adults with PAH is now enrolling

Learn more at proserastudy.com or scan the QR code

References: 1 Frantz RP, et al. *Lancet Respir Med.* 2024;12(7):523-534. **2** Sitbon O, et al. *Am J Respir Crit Care Med.* 2024;209:A1011.

Acknowledgements: We thank all participants, their families, and all the study investigators and study coordinators who participated in TORREY and the OLE.

Research supported by: Gossamer Bio, Inc.

This poster presents results from the OLE as of March 4, 2024.

Download your copy of this poster here.