1064

THE CLINICAL DEVELOPMENT OF INHALED SERALUTINIB FOR THE TREATMENT OF PULMONARY ARTERIAL HYPERTENSION (PAH)

Benza RL[1], Zisman LS[2], Channick RN[3], Chin KM[4], Frantz RP[5], Ghofrani HA[6], Hemnes AR[7], McLaughlin VV[8], Sitbon O[9], Vachiéry JL[10], Zamanian RT[11], Escribano Subías P[12], Sahay S[13], Zolty RL[14], Aranda R[2], Bruey JM[2], Cravets M[2], Li J[2], Mottola D[2], Osterhout R[2], Parsley E[2], Roscigno RF[2], Sitapara R[2], Howard LS[15]

[1]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [2]Gossamer Bio, Inc., San Diego, CA, USA; [3]University of California Los Angeles, UCLA Medical Center, Los Angeles, CA, USA; [4]UT Southwestern Medical Center, Dallas, TX, USA; [5]Mayo Clinic, Rochester, MN, USA; [6]Justus-Liebig-University Giessen and Marburg Lung Center (UGMLC), Institute for Lung Health, Cardio-Pulmonary Institute; Member of the German Center for Lung Research (DZL), Giessen, Germany; Department of Medicine, Imperial College, London, UK; [7]Vanderbilt University, Vanderbilt University Medical Center, Nashville, TN, USA; [8]University of Michigan, Ann Arbor, MI, USA; [9]Hôpital Bicêtre (AP-HP), Université Paris-Saclay, Le Kremlin-Bicêtre, France; [10]Université Libre de Bruxelles, HUB – Hôpital Erasme, Brussels, Belgium; [11]Stanford University School of Medicine, Stanford Medicine, Stanford, CA, USA; [12]University Hospital 12 de Octubre, Complutense University, Madrid, Spain; [13]Houston Methodist Hospital/Weill Cornell Medicine, Houston, TX, USA; [14]University of Nebraska Medical Center, Omaha, NE, USA; [15]Imperial College Healthcare NHS Trust, Hammersmith Hospital, London, UK

Presented at the PHA 2024 International PH Conference
Indianapolis, IN
August 15-18, 2024

An unmet need sparks an idea

- In PAH, small vessels in the lungs ("arterioles") get thicker and stiffer due to overactive inflammation, increased cell growth ("proliferation"), and scarring ("fibrosis") – these arterioles are being "remodeled"
- As a result, the pressure in these arterioles increases, making it harder for the heart to pump blood to the lungs
- Advances have been made in the treatment of PAH, but new medicines are still needed to treat the remodeled vessels

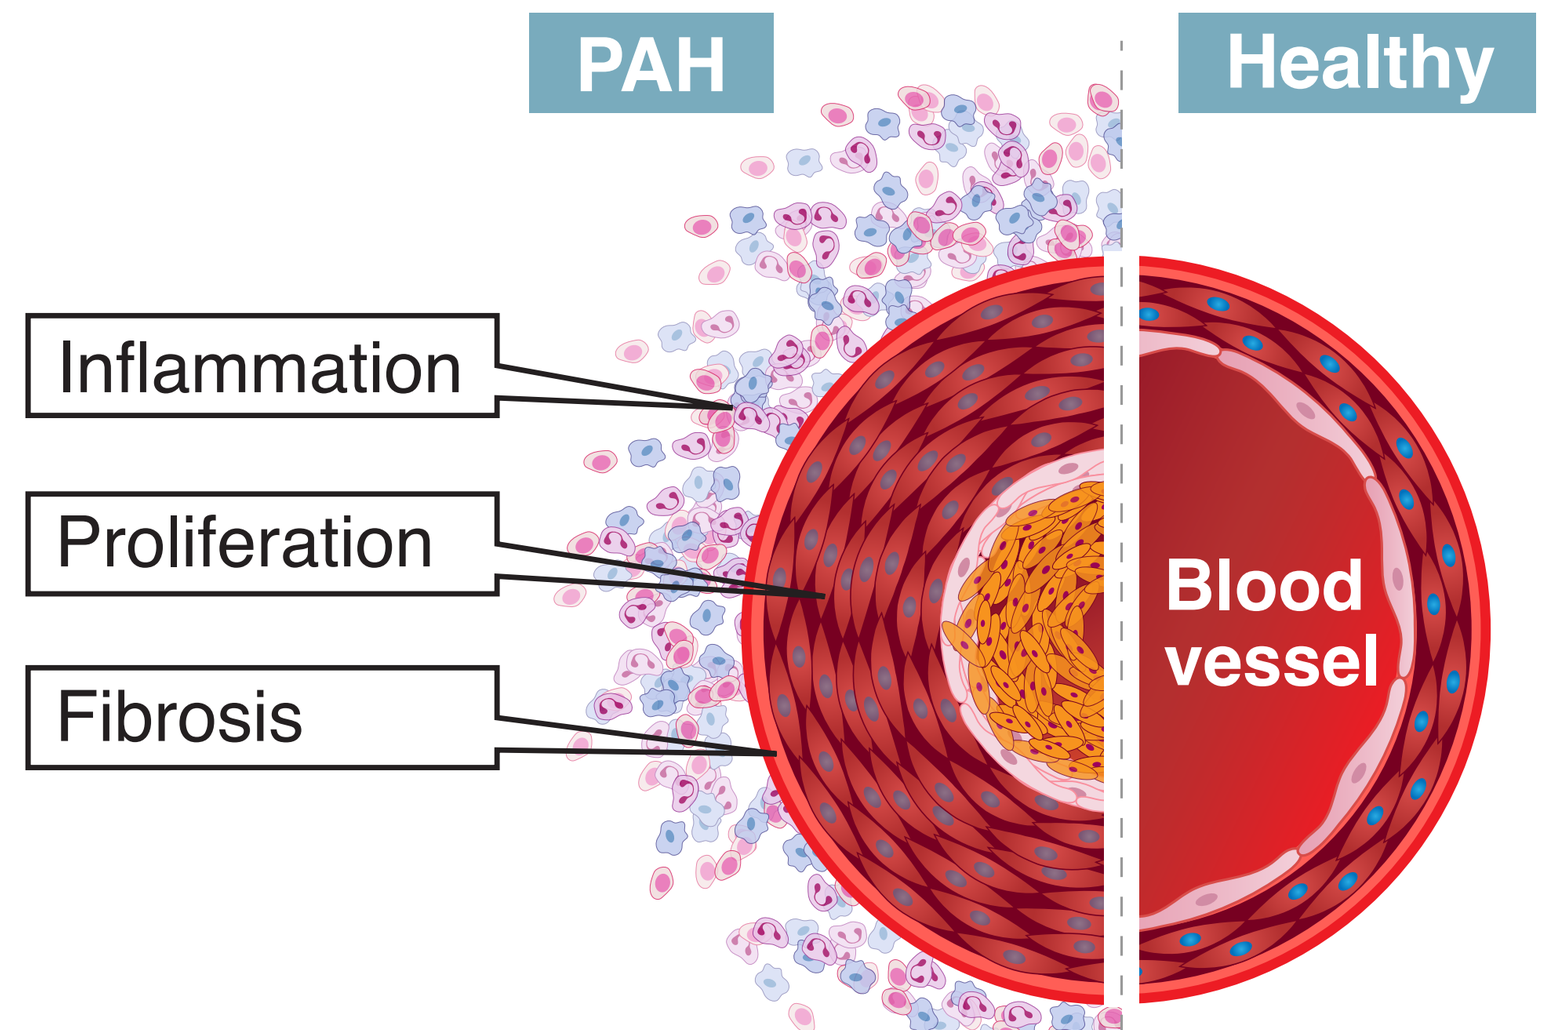
PAH
Healthy
Inflammation
Proliferation
Fibrosis
Blood vessel

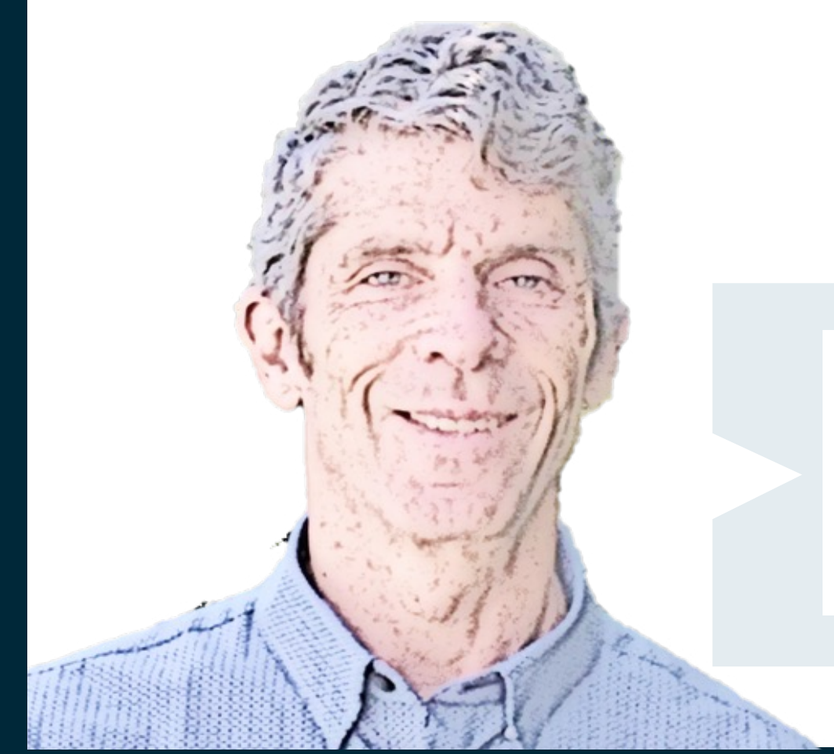

Dr. Larry S. Zisman talks about his motivation to support the PAH community

- **Dr. Lawrence S. Zisman**, a cardiologist and researcher who treated patients with PAH, wanted to find a way to develop a medicine to treat this remodeling, which is believed to be the underlying root cause of PAH

The journey continues

A novel approach is developed

- In the years to follow, Dr. Zisman and his team developed **seralutinib**, an investigational medicine they designed intentionally to treat PAH
- Seralutinib was created as a fine dry powder to be delivered deep into the lungs, where PAH occurs
- Inhaled administration concentrates seralutinib in the lung arterioles and thus minimizes side effects in the rest of the body

Dr. Larry S. Zisman elaborates on how seralutinib was designed to treat PAH

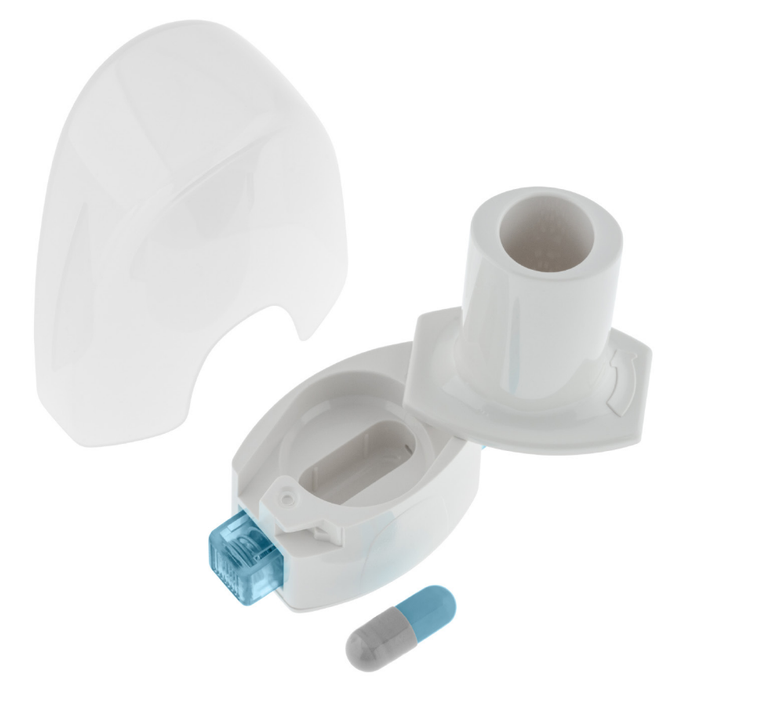

Seralutinib capsules are inserted into a pocket-sized dry-powder inhaler.

How does seralutinib work?

- Seralutinib is described by scientists as a *tyrosine kinase inhibitor* that works by blocking proteins (PDGFR, CSF1R, c-KIT) that contribute to PAH[1,2]
- Blocking these proteins reduces inflammation, proliferation, and fibrosis, potentially slowing or reversing damage caused by vascular remodeling[1,2]

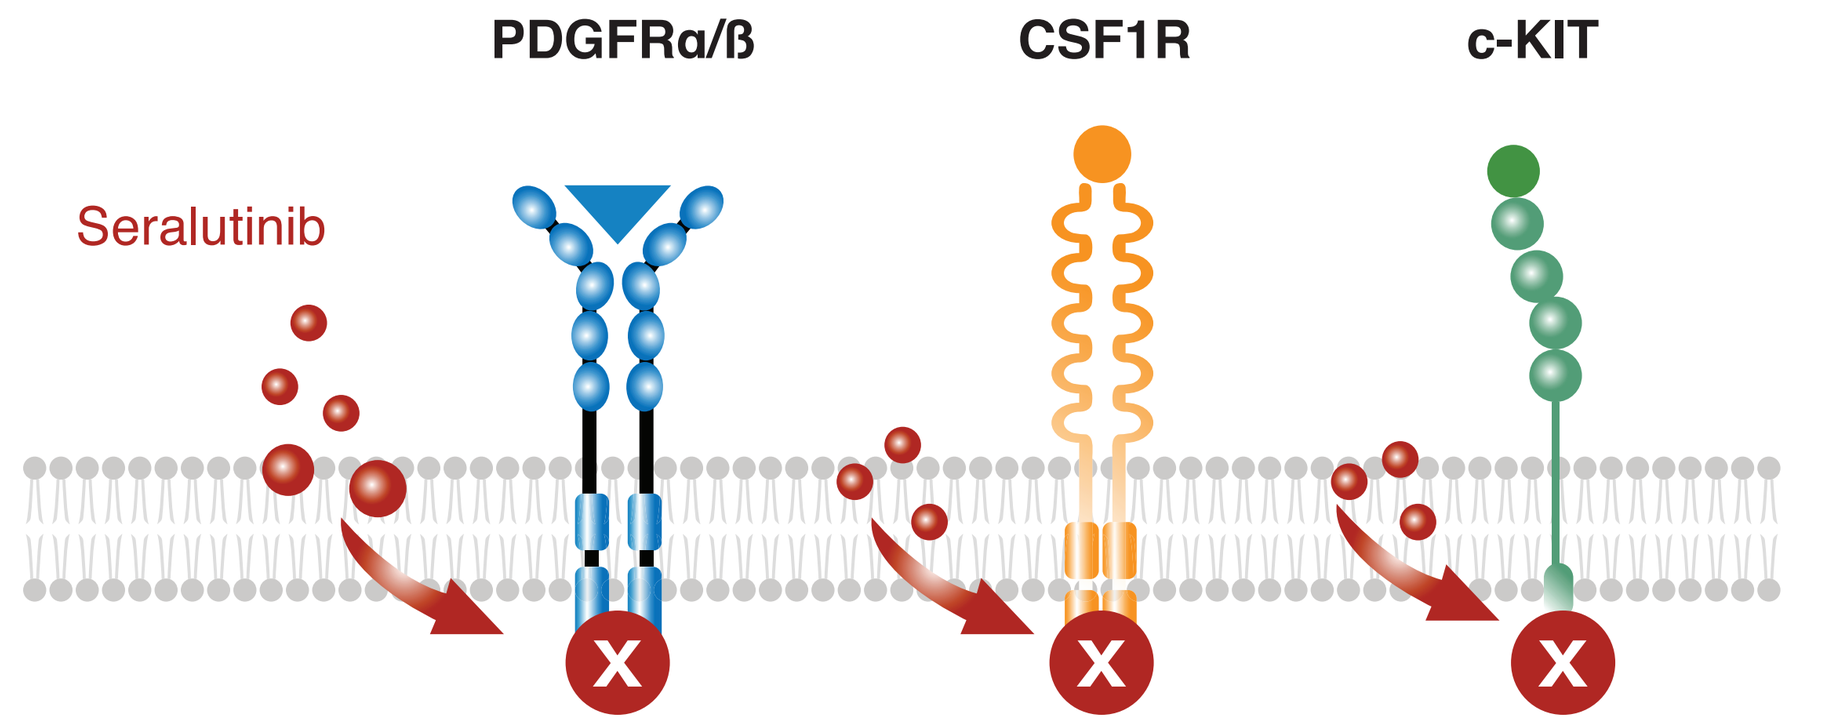
PDGFRα/ß
CSF1R
c-KIT
Seralutinib
X
X
X

c-KIT, mast/stem cell growth factor receptor kit; CSF1R, colony stimulating factor 1 receptor; PDGFR, platelet-derived growth factor receptor.

Prof. Luke S. Howard (Imperial College, London, UK) explains how seralutinib works

Promising early evidence

- In experimental models of PAH, researchers found that inhaled seralutinib was mostly present in the lungs, meaning that it was delivered to the site of the disease[1]
- Further preclinical investigation showed that seralutinib reversed structural changes in lung arterioles[1]

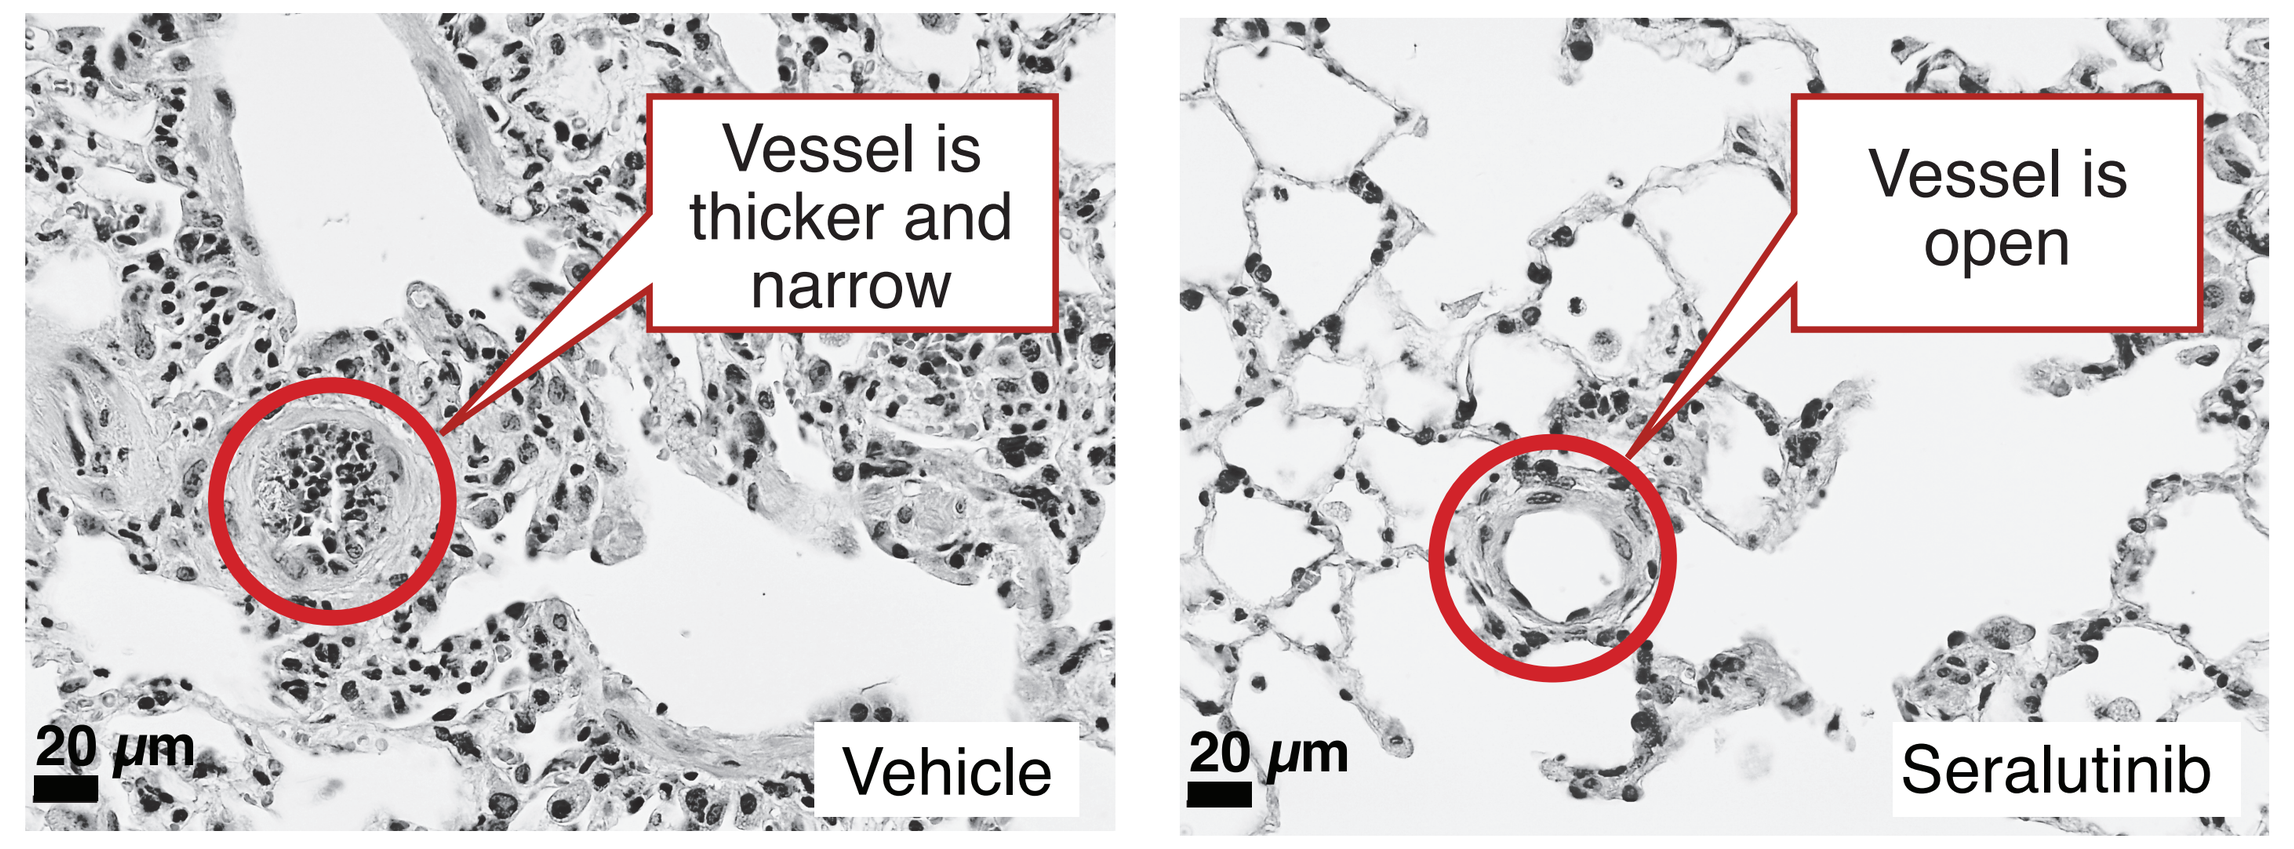
Vessel is thicker and narrow
20 μm
Vehicle
Vessel is open
20 μm
Seralutinib

Dr. Larry S. Zisman explains early research on seralutinib

The phase 2 TORREY study is successful

- Encouraged by data from early studies, a clinical study involving adults with PAH was conducted – the **TORREY** study (NCT04456998) (see **figure, right**)

TORREY STUDY

86 participants with PAH

- **44** participants took **seralutinib 90 mg** twice daily
- **42** participants took **placebo** twice daily

Treatment lasted **24 weeks**

Hear **Dr. Robert P. Frantz** (Mayo Clinic, Rochester, MN, USA) discuss highlights of the TORREY study

What did the TORREY study find?[3]

- Inhaled seralutinib reduced the pressure in pulmonary blood vessels (also called pulmonary vascular resistance, or PVR) after 24 weeks compared to placebo. Reducing PVR makes it easier for blood to flow from the heart to the lungs
- Seralutinib reduced stress on the heart, as measured by blood levels of NT-proBNP
- Participants with more severe symptoms (FC III or REVEAL 2.0 risk score ≥6) experienced greater improvements in PVR and exercise capacity (measured by 6-minute walk distance, or 6MWD)
- Seralutinib was well tolerated. The most common side effect was cough (seralutinib group: 43%; placebo group: 38%)

PVR			6MWD			NT-proBNP		
Pulmonary Vascular Resistance measures how hard it is for blood to flow from the heart to the lungs			*6-Minute Walk Distance* reflects how well one can perform daily activities			A protein in the blood that measures stress on the heart		
All participants	FC III	REVEAL ≥6	All participants	FC III	REVEAL ≥6	All participants	FC III	REVEAL ≥6
−14%*	−21%*	−23%*	+7 meters	+37 meters*	+22 meters	−408 ng/L*	−527 ng/L*	−732 ng/L*

*p-value ≤0.05.

Imaging the lung vessels[4]

- An imaging substudy examined the effect of seralutinib on improving small blood vessels, which are typically narrowed in PAH
- The study found that seralutinib increased the blood distribution to the small vessels, which suggests that seralutinib is having a favorable outcome on underlying vascular remodeling (**figure**)

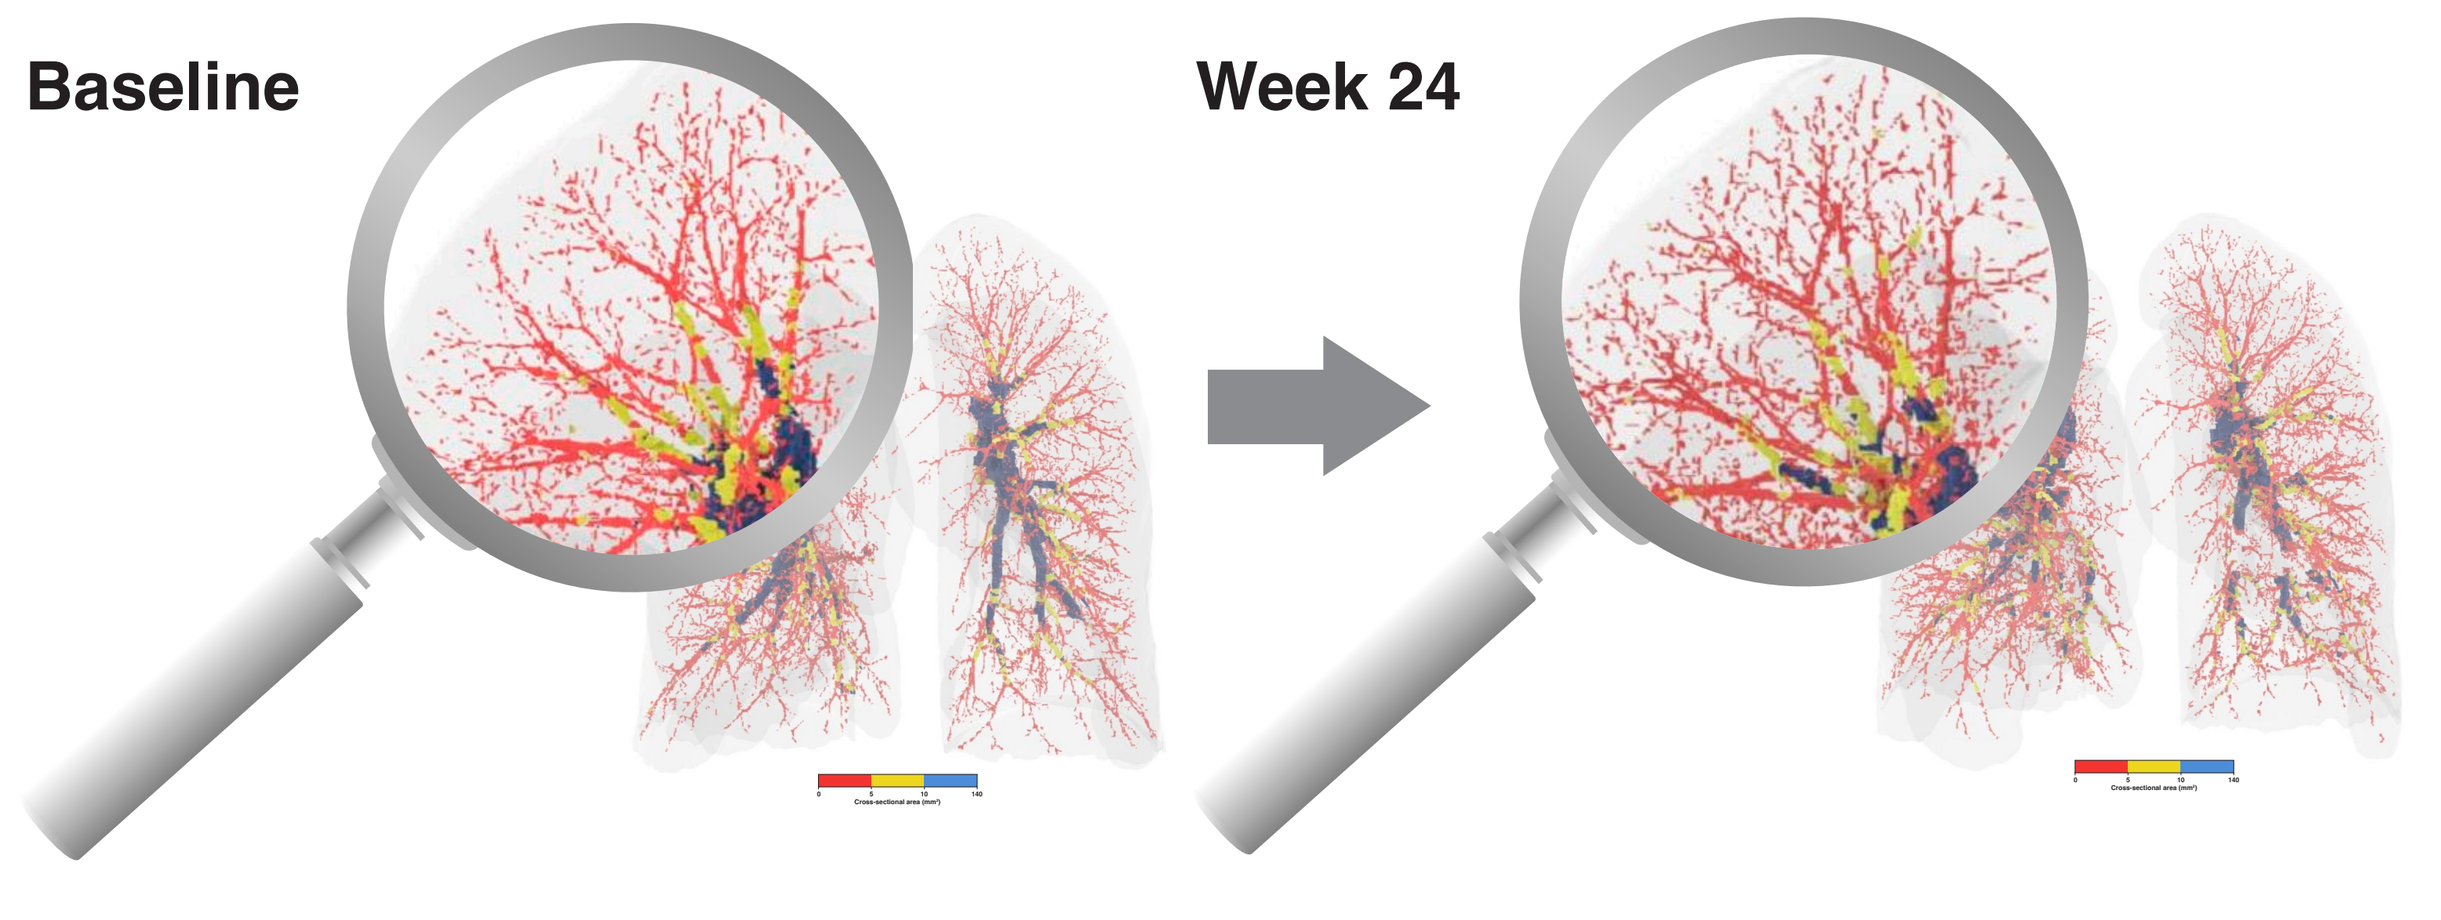
Baseline
Week 24

The representative example shows changes in the pulmonary vessels of a seralutinib-treated participant from the beginning of the study to 24 weeks.

6MWD, 6-minute walk distance; FC, functional class; NT-proBNP, N-terminal pro-brain natriuretic peptide; PVR, pulmonary vascular resistance; REVEAL, Registry to Evaluate Early and Long-term PAH Disease Management.

Continued improvement in the OLE study

- After completing TORREY, participants could enroll in an **open-label extension** (OLE; NCT04816604), during which all participants received seralutinib to study long-term safety and efficacy

Dr. Richard N. Channick (UCLA Medical Center, Los Angeles, CA, USA) discusses the OLE study

74: 73 participants who completed TORREY and 1 participant from a phase 1b study enrolled

- **34** participants continued **seralutinib 90 mg** twice daily
- **40** participants switched from placebo to **seralutinib 90 mg** twice daily

Right heart catheterization was performed at the beginning (Baseline) and end (Week 24) of TORREY, and after 48 weeks in the OLE (ie, after a total of 72 weeks of treatment) to measure PVR

TORREY study: Baseline – Week 24; OLE study: Week 24 – Week 72

What were the results of the OLE study?[5]

Median **drop in PVR** from the beginning of TORREY to Week 72 (dyne*s/cm^5)

- Continued seralutinib: -146
- Placebo crossed: -56

Mean **improvement in 6MWD** from the beginning of TORREY to Week 72

- Continued seralutinib: +36 meters
- Placebo crossed: +12 meters

- In some participants, PVR and 6MWD improved during TORREY, and continued to improve even more during the OLE
- Treatment with seralutinib for up to 72 weeks was generally well tolerated. Cough decreased for both groups (continued seralutinib: 21%; placebo crossed: 23%) at Week 72

The phase 3 PROSERA study is ongoing

- The positive results of TORREY and Gossamer Bio's strong commitment to the PAH community led to an even bigger study, which is required to confirm these results
- **PROSERA** (NCT05934526) is a global, randomized, double-blind study, enrolling participants with PAH, 18–75 years of age and on stable PAH-specific medication (other criteria apply)
- Participants who complete the placebo-controlled portion of the study may enroll in an extension study, in which all participants receive seralutinib

PROSERA STUDY

Screening (up to 4 weeks) → **Randomization**

- **175** participants will take **seralutinib 90 mg** twice daily
- **175** participants will take **placebo** twice daily

Treatment will last between **24 and 48 weeks**

Where can I learn more?

- Please visit **www.proserastudy.com** or scan the QR code

Listen to **Dr. Raymond L. Benza** (Icahn School of Medicine at Mount Sinai, New York, NY, USA) talk about PROSERA

References: 1 Galkin A, et al. *Eur Respir J.* 2022;60(6):2102356. **2** Pullamsetti SS, et al. *Int J Mol Sci.* 2023;24(16):12653. **3** Frantz RP, et al. *Lancet Respir Med.* 2024;12(7):523-534. **4** Zamanian RT, et al. *Eur Respir J.* 2023;62(suppl 67):OA742. **5** Sitbon O, et al. *Am J Respir Crit Care Med.* 2024;209:A1011.

The clinical development of seralutinib is supported by **Gossamer Bio, Inc.**