3330

GB5121 is a Novel, Highly Potent and Selective CNS-Penetrant BTK Inhibitor for CNS Malignancies

Isharat Yusuf, Sunil Sahdeo, Taylor Ismaili, Zachary Naiman, David Guimond, Theodore Schiff, Bryan Clemons, Krystal Herman, Bryanna Paulson, Kay Hou, Kristen Taylor Meadows, Michael Kennedy, Mark Rose, Laura Carter

Gossamer Bio, Inc., San Diego, CA, USA

BACKGROUND

- Primary central nervous system lymphoma (PCNSL) is a rare and aggressive form of non-Hodgkin lymphoma that is restricted to the CNS
- Inhibitors of Bruton's tyrosine kinase (BTK) are approved treatments for several B cell lymphomas, but are characterized by modest selectivity and/or low CNS exposure
- GB5121 is an orally available, selective, irreversible small molecule BTK inhibitor chosen for development based on its potency, selectivity and high CNS exposure
- These features differentiate GB5121 from other BTK inhibitors and support its use in clinical trials for CNS malignancies

Figure 1. BTK inhibition targets a key survival node in PCNSL

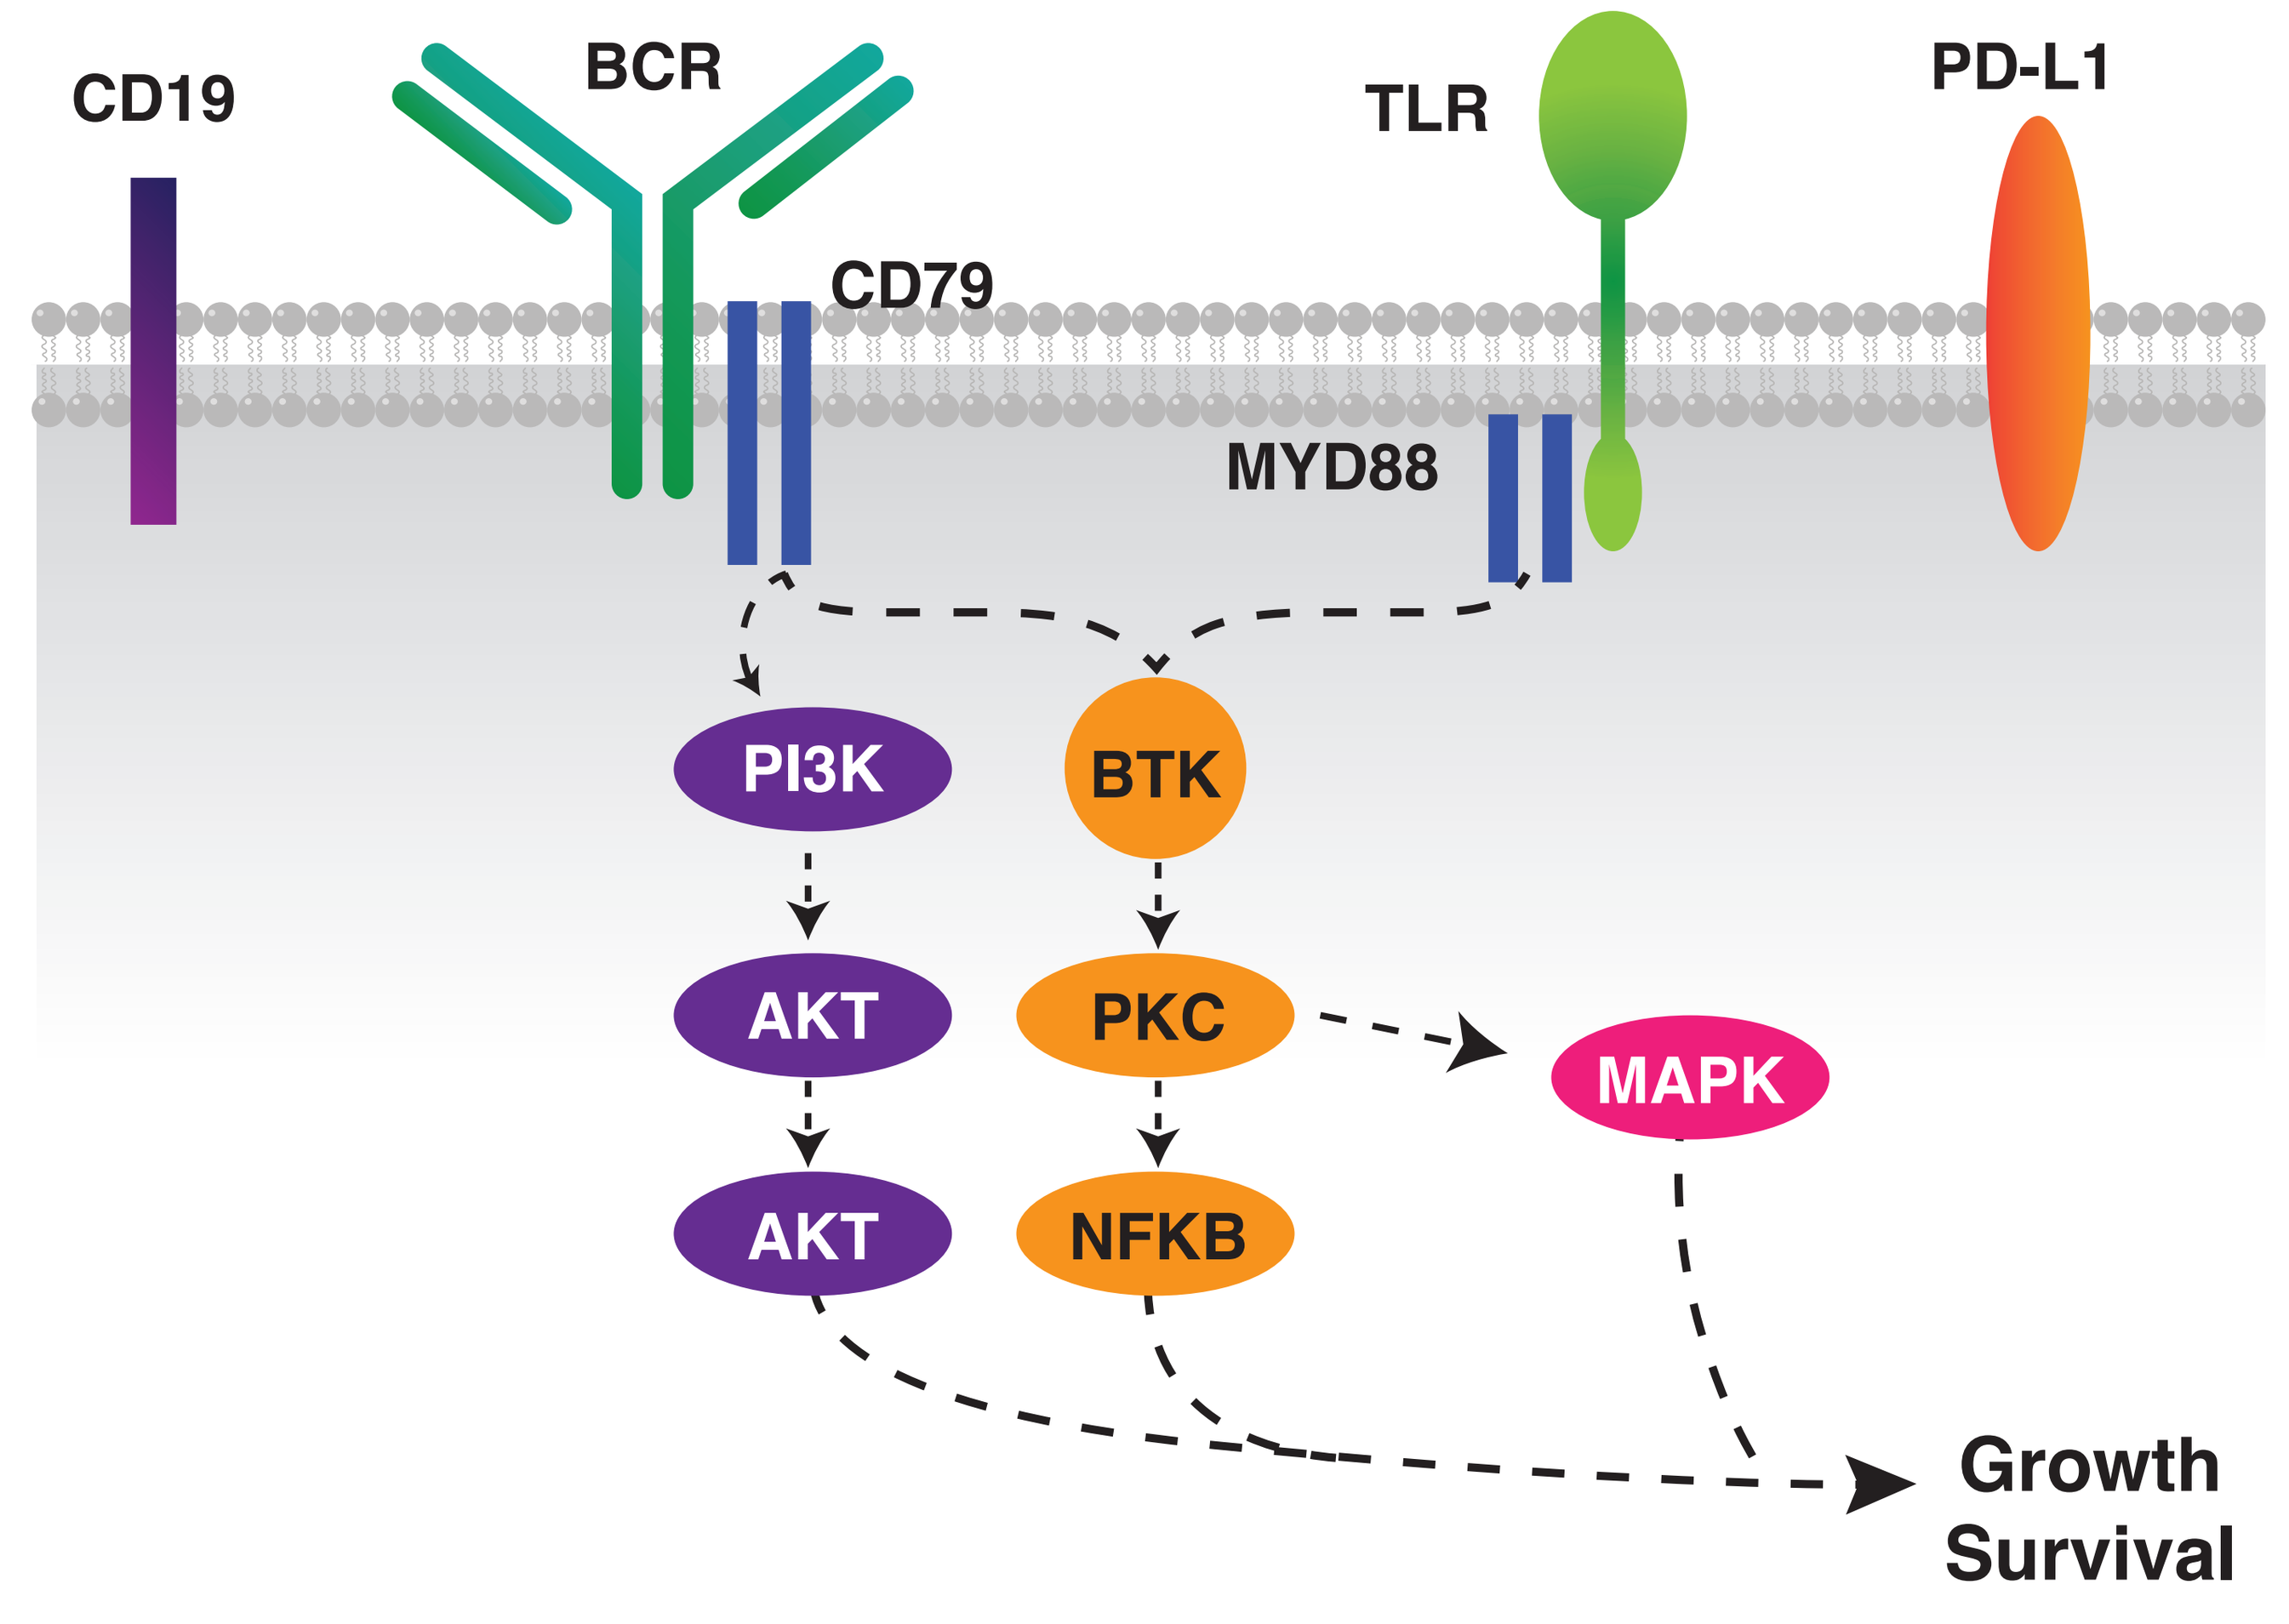
CD19
BCR
CD79
TLR
PD-L1
MYD88
PI3K
BTK
AKT
PKC
MAPK
AKT
NFKB
Growth
Survival

Adapted from Graham MS et al. 2018

OBJECTIVE

- Profile GB5121 for selectivity and CNS exposure in comparison to ibrutinib

METHODS

- Selectivity of GB5121 and ibrutinib was profiled in kinome scans against 349 kinases at 1μM under conditions using 1mM ATP concentration; EGFR activity was further evaluated in a cell-based assay
- Irreversible and covalent nature of GB5121 was evaluated using enzymatic assays and no warhead control compounds
- *In vitro* potency and inactivation kinetics of GB5121 were evaluated using biochemical and cell-based assays
- Comparative BTK target occupancy of GB5121 and ibrutinib was assessed in perfused mice with an intact blood brain barrier (BBB) using a probe-based ELISA
- Pharmacokinetic profiles of GB5121 were obtained in multiple species using intravenous and oral dosing

RESULTS

Figure 2. GB5121 is a highly selective BTK inhibitor

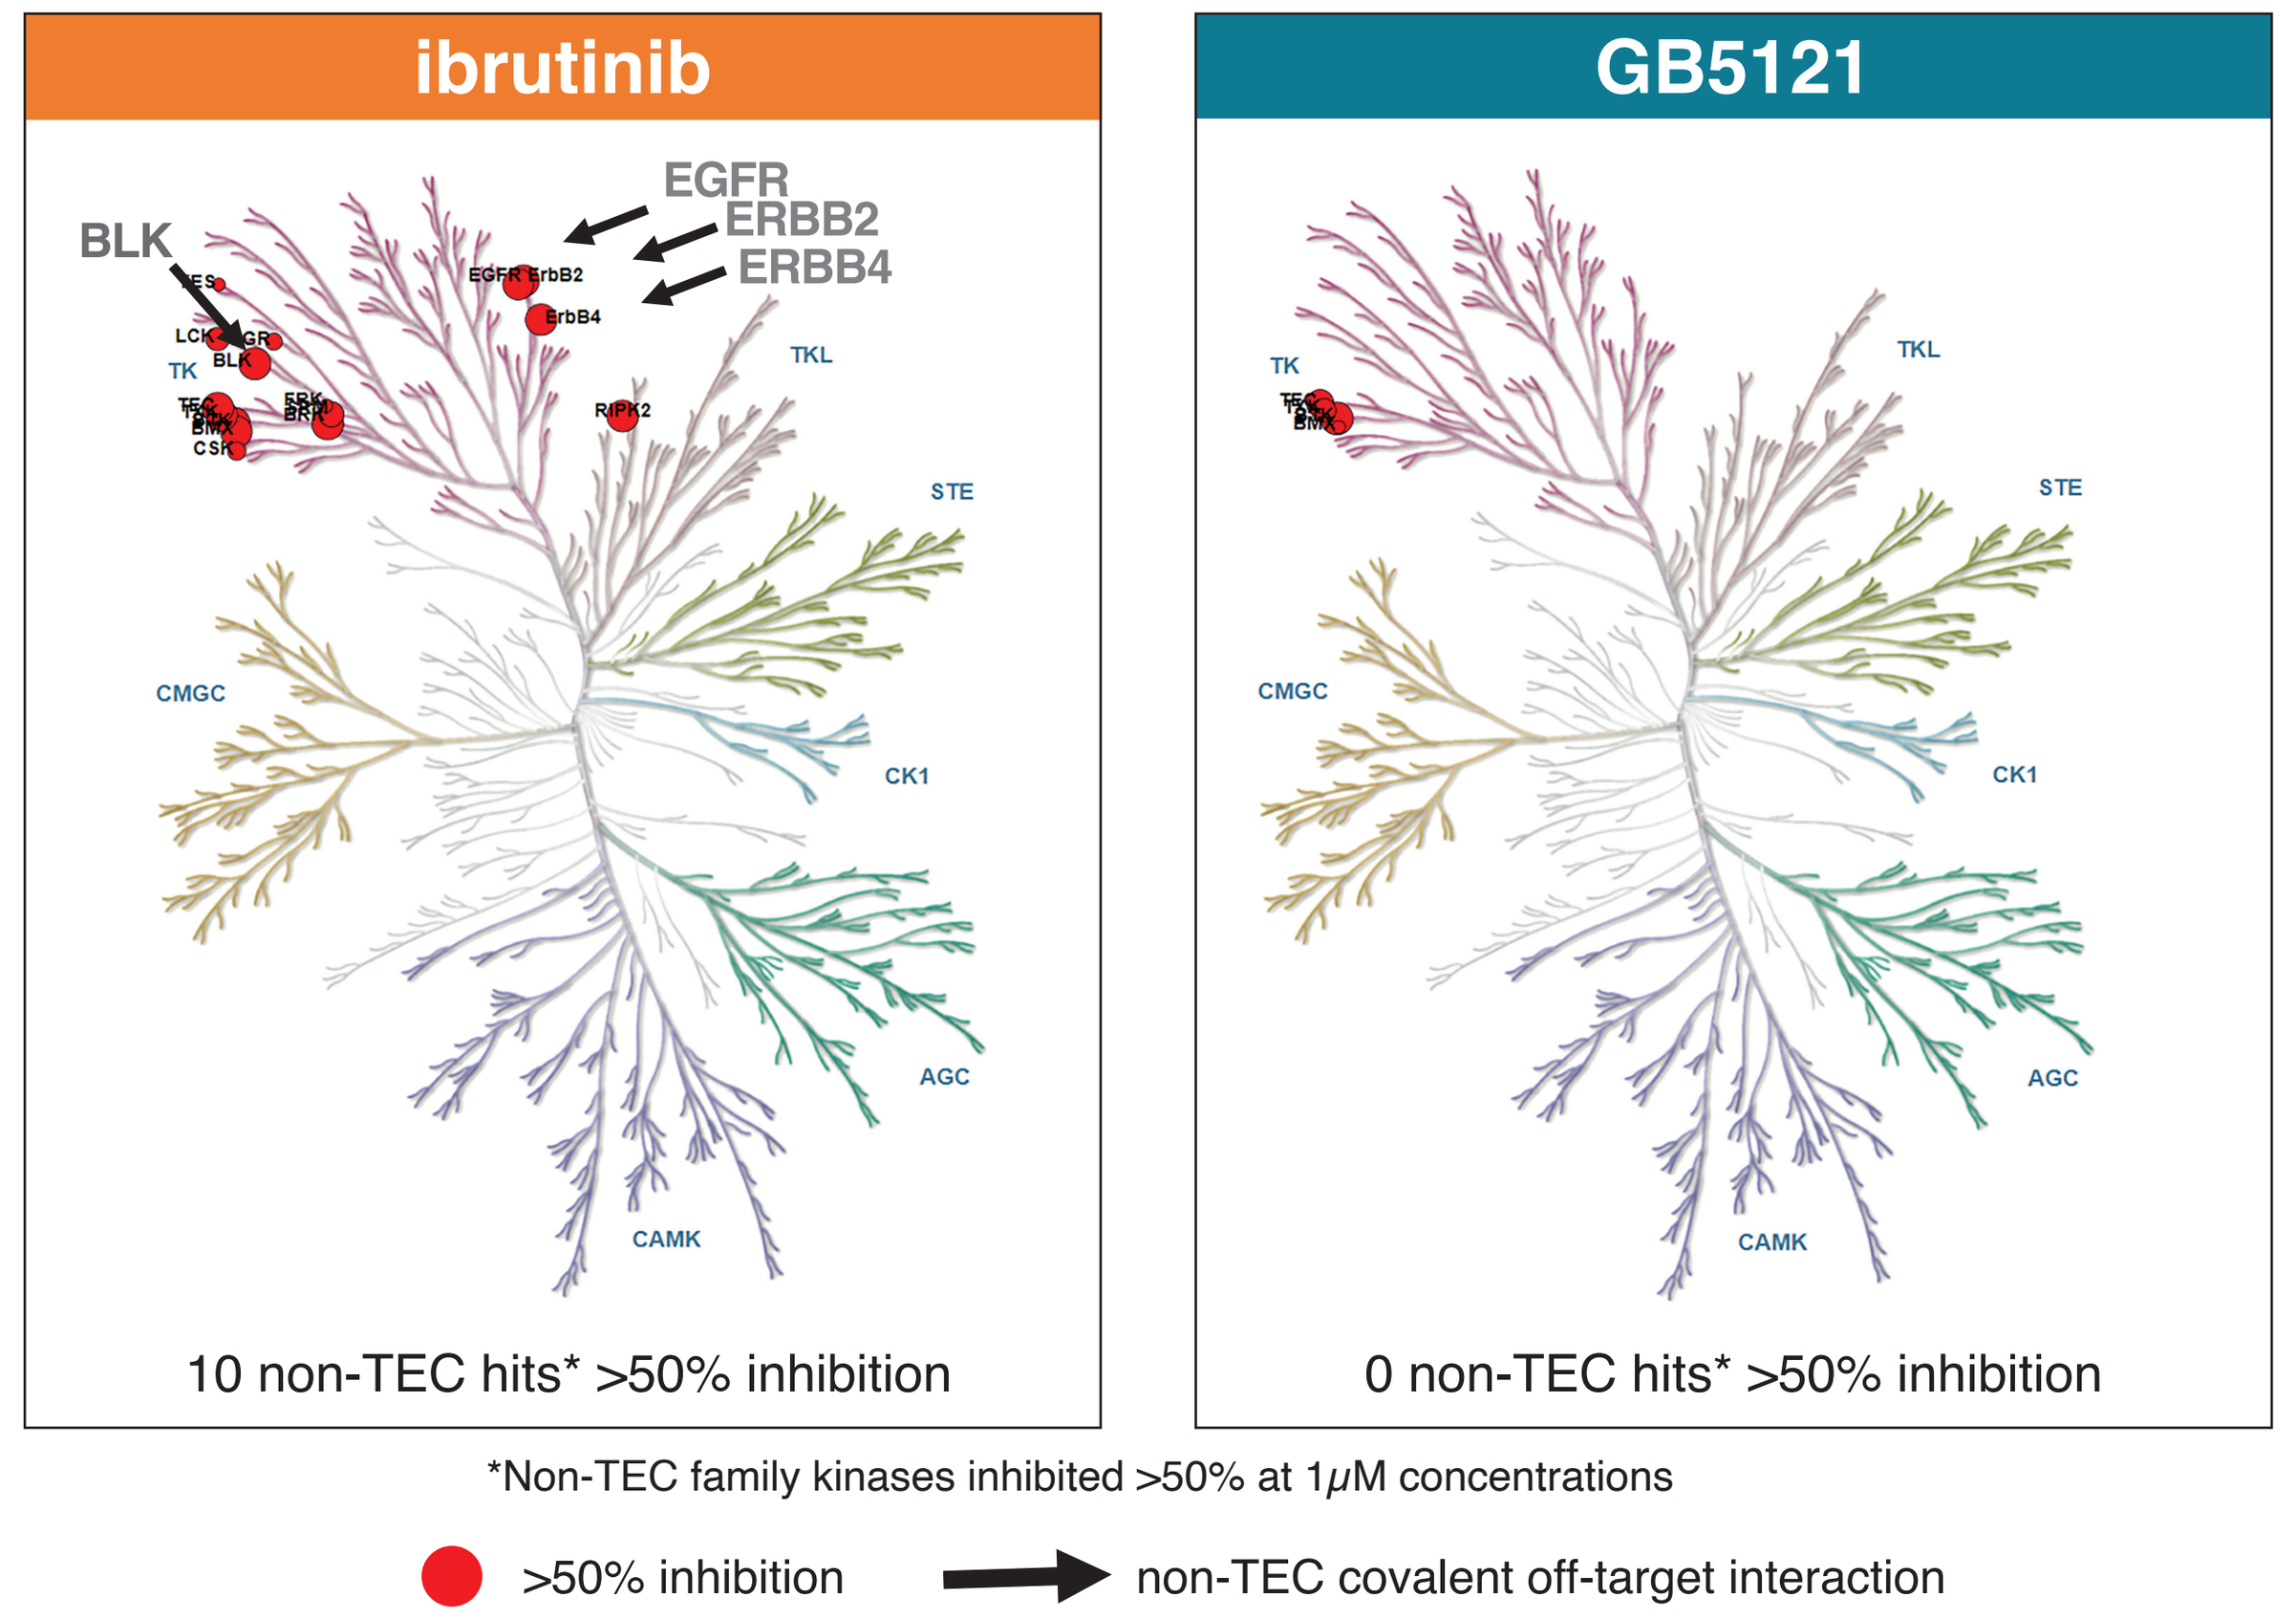
ibrutinib
GB5121
BLK
EGFR
ERBB2
ERBB4
TK
TKL
STE
CMGC
CK1
AGC
CAMK
10 non-TEC hits* >50% inhibition
0 non-TEC hits* >50% inhibition
>50% inhibition
non-TEC covalent off-target interaction

*Non-TEC family kinases inhibited >50% at 1μM concentrations

- GB5121 demonstrates >50% inhibition of only two kinases besides BTK (TEC and TXK)
- Compared to ibrutinib, GB5121 lacks activity against EGFR, ERBB2 (HER2) and ERBB4 (HER4)

Figure 3. GB5121 does not inhibit phosphorylation of EGFR in A431 cell line

Data are representative of 10 independent studies. Dose response curves were generated from duplicate values at each concentration of test article using a nonlinear, 4-parameter, variable slope curve-fitting function using GraphPad Prism software

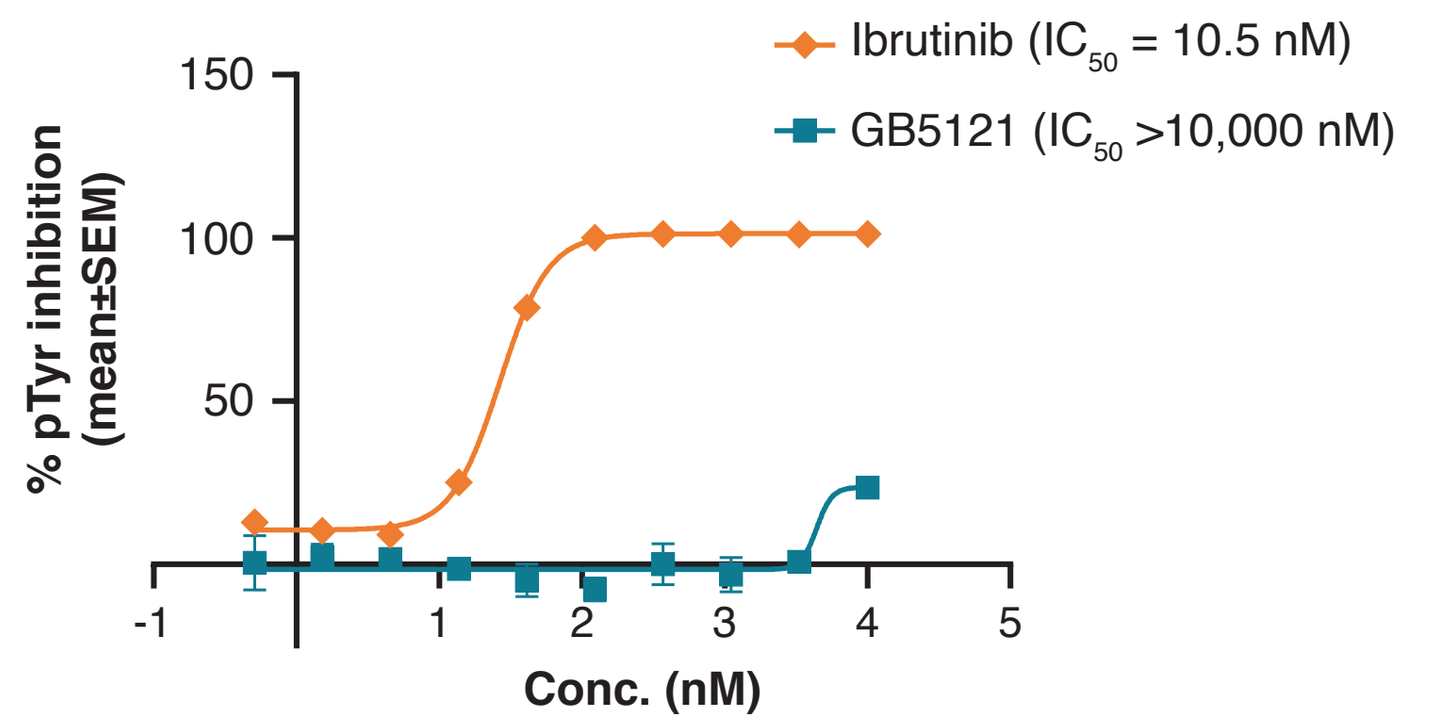
Ibrutinib (IC_{50} = 10.5 nM)
GB5121 (IC_{50} >10,000 nM)
% pTyr inhibition (mean±SEM)
150
100
50
-1
1
2
3
4
5
Conc. (nM)

Table 1. BTK enzymatic mechanism of inhibition studies demonstrate properties of an irreversible covalent inhibitor

Assay		GB5121	GBD-07476 (GB5121 with unreactive warhead)	GBD-07547 (GB5121 with no warhead)
BTK Enzyme Assay Preincubation (nM IC_{50})	0 min	69	9010	3830
	30 min	14	8610	3780
	60 min	6	7210	4900
BTK enzyme recovery/ Residence Time		>300 min	<5 min	<5 min
Irreversible Inhibition		yes	no	no

Table 2. Potency and inactivation kinetics in cells and whole blood

Assay	GB5121
hWB pBTK IC_{50}	63.5 nM
Ramos calcium flux IC_{50}	23.8 nM
hWB CD69 IC_{50}	181.3 nM
B cell proliferation IC_{50}	4.7 nM
hWB kinact/KI	4.7 10^{-4} nM^{-1} min^{-1}
mWB kinact/KI	7.2 10^{-4} nM^{-1} min^{-1}
mB kinact/KI	3.8 10^{-4} nM^{-1} min^{-1}
Pfeiffer DLBCL viability IC_{50}	14.0 nM
TMD8 DLBCL viability IC_{50}	23.5 nM

hWB = human whole blood, mWB = mouse whole blood, mB = mouse brain lysate

Figure 4. GB5121 shows potent activity in vitro in diffuse large B cell lymphoma (DLBCL) cell lines regardless of phenotype and mutational profile

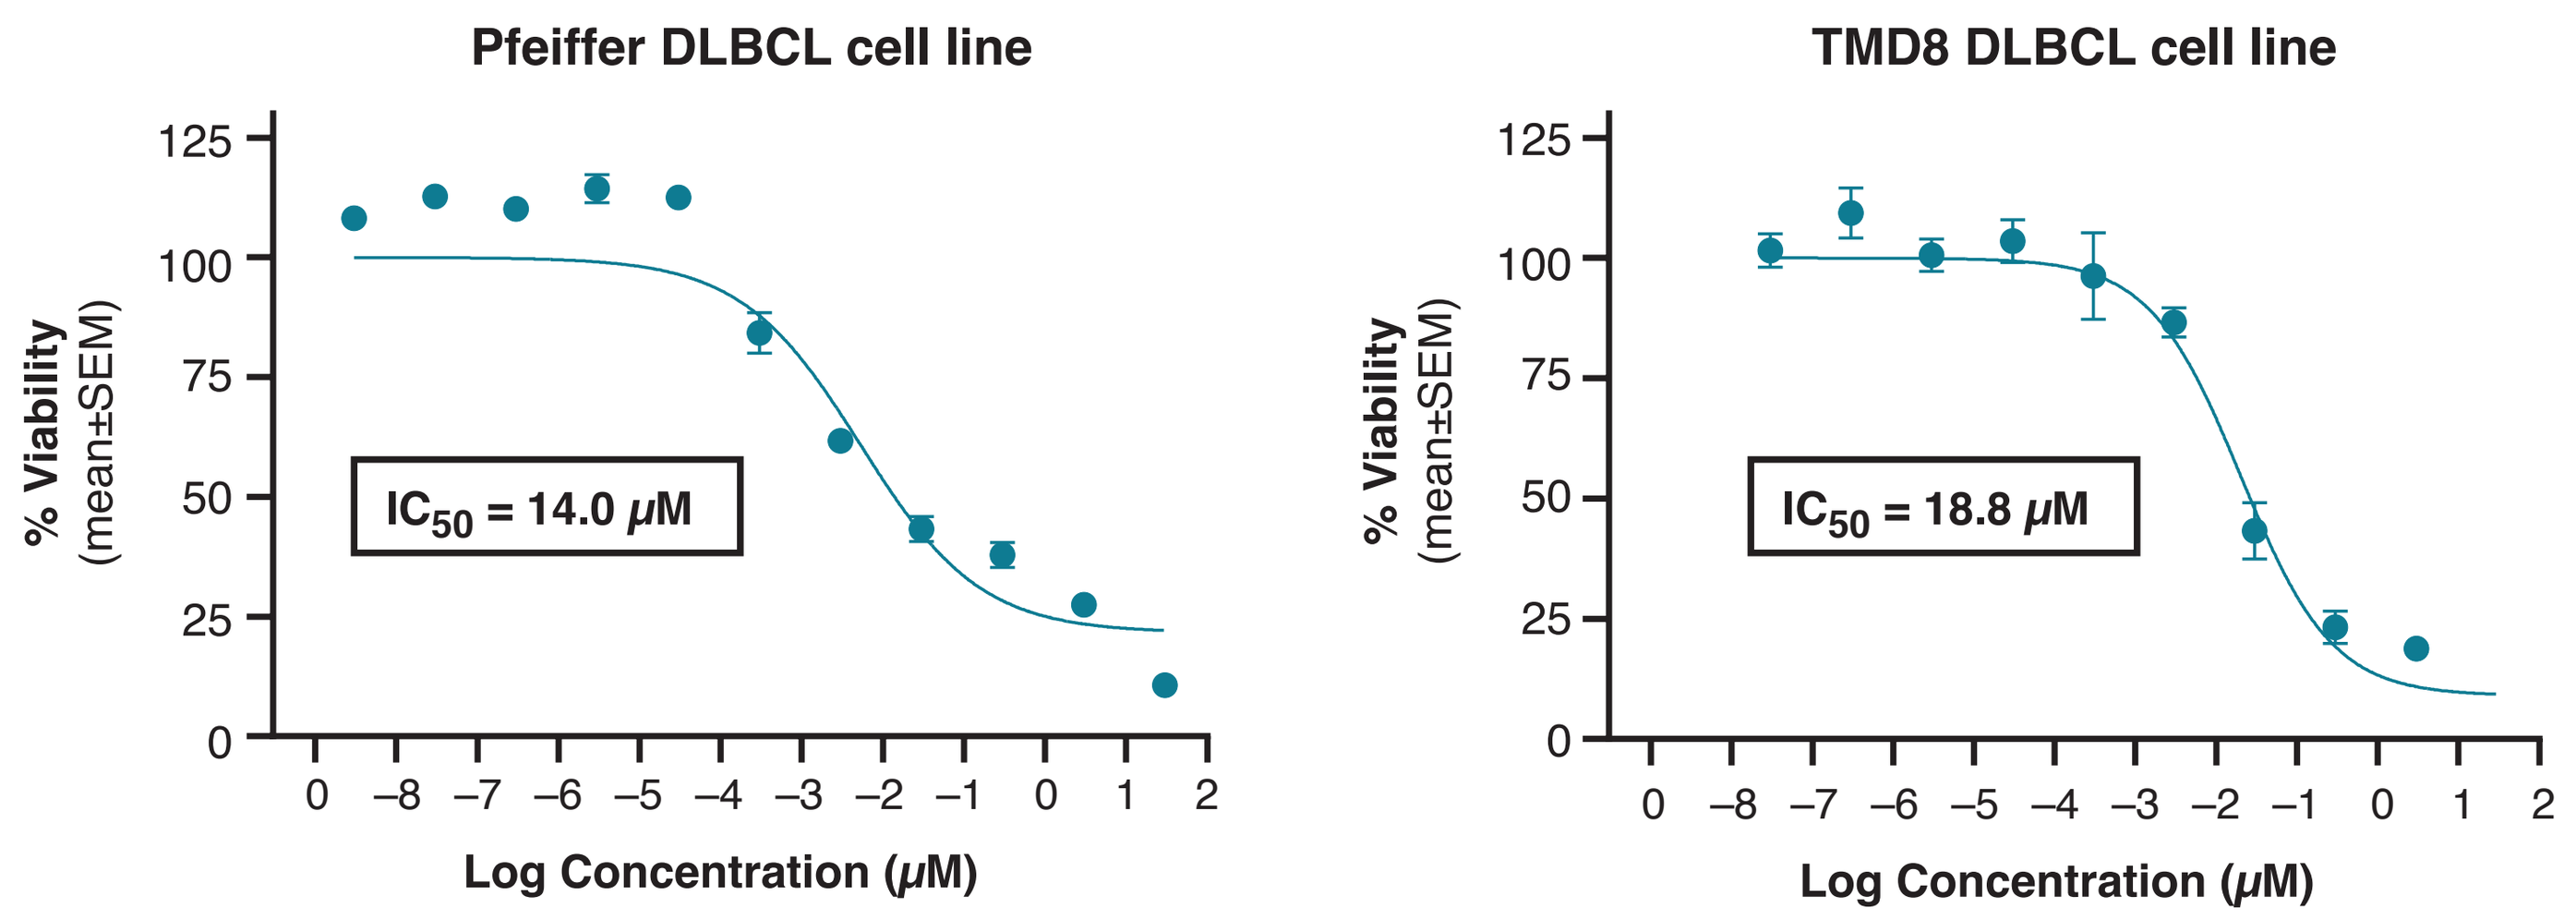
Pfeiffer DLBCL cell line
TMD8 DLBCL cell line
% Viability (mean±SEM)
IC_{50} = 14.0 μM
IC_{50} = 18.8 μM
Log Concentration (μM)

Pfeiffer cell line: Germinal Center (GC) phenotype with wild type MyD88. **TMD8 cell line:** Activated B Cell (ABC) phenotype carrying MyD88 driver mutation. Dose response curves were generated from triplicate (Pferiffer) or duplicate (TMD8) values at each concentration of test article using a nonlinear, 4-parameter, variable slope curve-fitting function using GraphPad Prism software

Figure 5. GB5121 shows superior CNS exposure with lower plasma exposures compared to ibrutinib in naïve mice with intact BBB

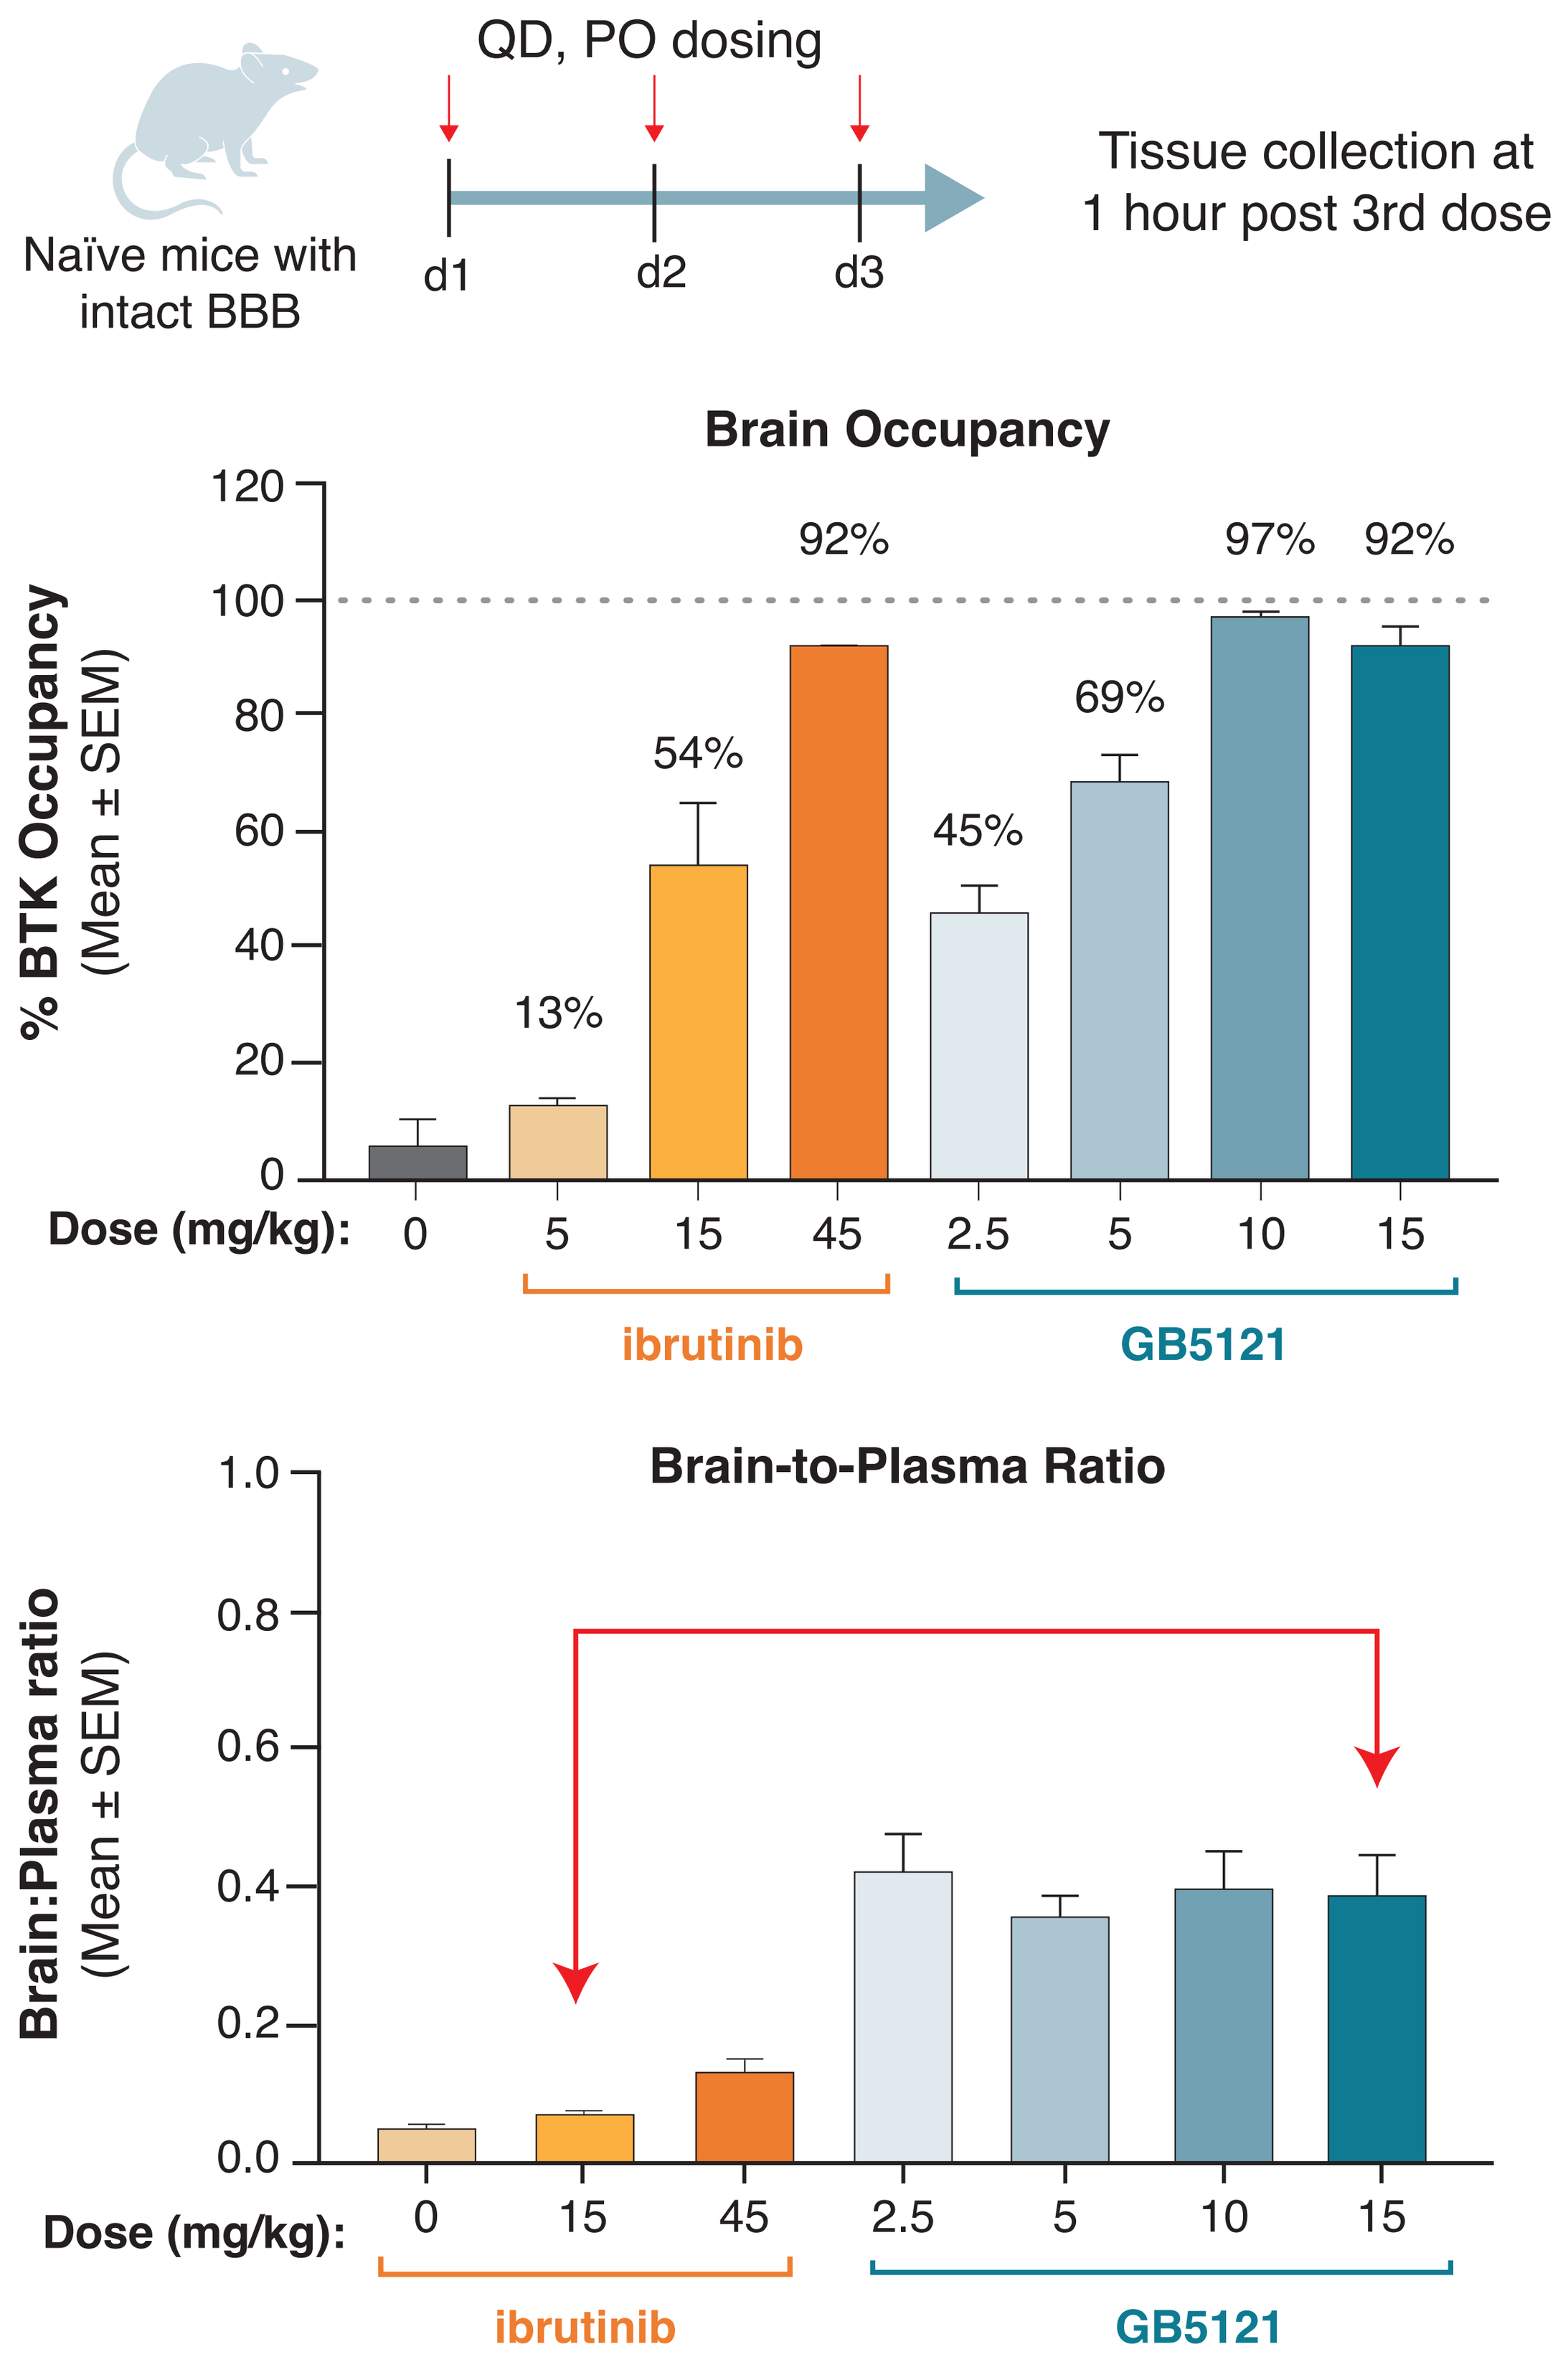
QD, PO dosing
Naïve mice with intact BBB
d1
d2
d3
Tissue collection at 1 hour post 3rd dose
Brain Occupancy
% BTK Occupancy (Mean ± SEM)
13%
54%
92%
45%
69%
97%
92%
Dose (mg/kg): 0 5 15 45 2.5 5 10 15
ibrutinib
GB5121
Brain-to-Plasma Ratio
Brain:Plasma ratio (Mean ± SEM)
Dose (mg/kg): 0 15 45 2.5 5 10 15
ibrutinib
GB5121

15 mg/kg = Mouse comparable dose of ibrutinib 560 mg QD clinical dose based on plasma AUC.
Data are representative of 2 independent experiments. Graphs were generated in GraphPad Prism software. Study included 3 mice per group.

Table 3. GB5121 brain to plasma ratios after single or multiple daily oral (PO) dosing in multiple species

Species Strain	N	PO Dose (mg/kg)	Day (Time)	Brain/Plasma Ratio (Mean ± SD)
Mouse C57BL/6	15	10	3 (1h)	0.187 ± 0.022
Rat Wistar Han	3	10	1 (1h)	0.285 ± 0.044
NHP Cyno	4	30	15 (4h)	1.60 ± 0.42

Figure 6. GB5121 demonstrates a 1:1 brain to plasma ratio in non-human primates (n=3) after an IV dose

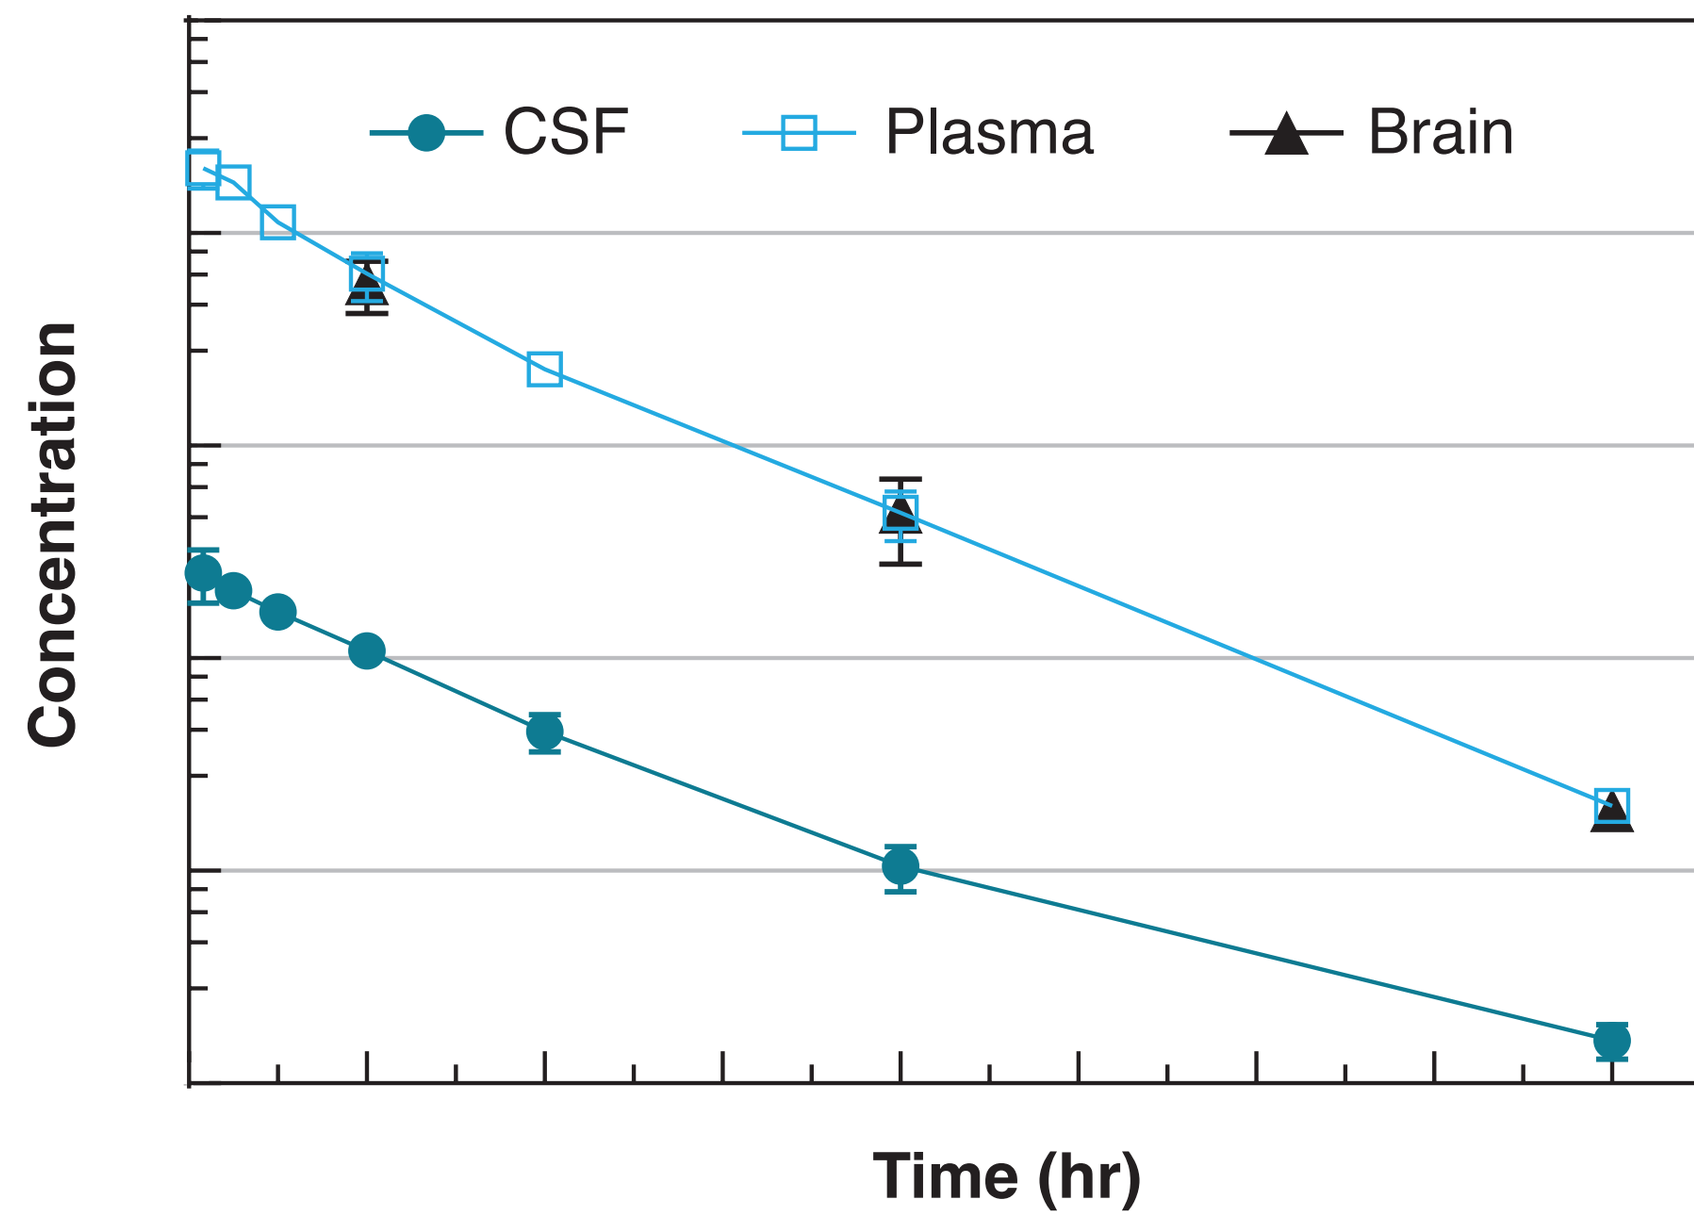
CSF
Plasma
Brain
Concentration
Time (hr)

SUMMARY AND CONCLUSIONS

- GB5121 is an orally-available covalent BTK inhibitor that is currently being investigated in a phase 1b/2 trial in patients with relapsed/refractory primary/secondary CNS lymphoma and primary vitreoretinal lymphoma (NCT05242146)
- Preclinical studies demonstrated GB5121 to exhibit several characteristics differentiating it from approved BTK inhibitors and those currently under clinical investigation, including
 - rapid equilibrium into the brain
 - superior BTK inactivation rate and brain target occupancy
 - high selectivity over other kinases
- Excellent brain exposure and selectivity combined with activity against DLBCL cell lines support the use of GB5121 as a novel molecule to treat human BTK-driven malignancies including CNS lymphoma

REFERENCES

1. Graham MS, DeAngelis LM. *Best Pract Res Clin Haematol.* 2018; 31: 262-269.

ACKNOWLEDGEMENTS

Funded by GB005, Inc, a wholly owned subsidiary of Gossamer Bio,Inc. The authors gratefully acknowledge AssayQuant and Pharmaron for support of *in vitro* studies.

Presented at the AACR Annual Meeting, April 8 – 13, 2022