P406

A CNS-Penetrant BTK Inhibitor Modulates Multiple Pathogenic Pathways Implicated in Multiple Sclerosis

Isharat Yusuf, Bryan Clemons, Zachary Naiman, Susan Murphy, David Guimond, Eduardo Garcia, Kay Hou, Mark Rose, Marcos Steinberg, Kristen Taylor Meadows, Laura Carter

Gossamer Bio, Inc., San Diego, CA, USA

BACKGROUND

- Although current therapies for multiple sclerosis (MS), such as B cell depletion and S1P modulation, have been effective in relapse prevention, emerging data demonstrate continued disability progression independent of relapse activity (PIRA)[1]
- PIRA is associated with the presence of ectopic B cell follicles and slowly expanding, chronic, active lesions with microglia cells around their rim, resulting in a smoldering central nervous system (CNS)-driven inflammation, tissue damage, and disease progression[2,3]
- Bruton's tyrosine kinase (BTK), a key signaling node downstream of the B cell receptor and microglia fragment crystallizable receptors (FcR), plays a critical role in both B cell and myeloid cell activation[4,5]
- A CNS-penetrant BTK inhibitor (BTKi) has the potential to inhibit the pathogenic activity of both the adaptive B cells and innate macrophage/microglia cells (**Figure 1**)
- GB7208 is a preclinical, investigational drug product candidate designed to be a CNS-penetrant, irreversible BTKi, optimized for potential best-in-class potency, selectivity, and brain exposure

Figure 1. Targeting BTK addresses key nodes in driving autoimmunity in MS

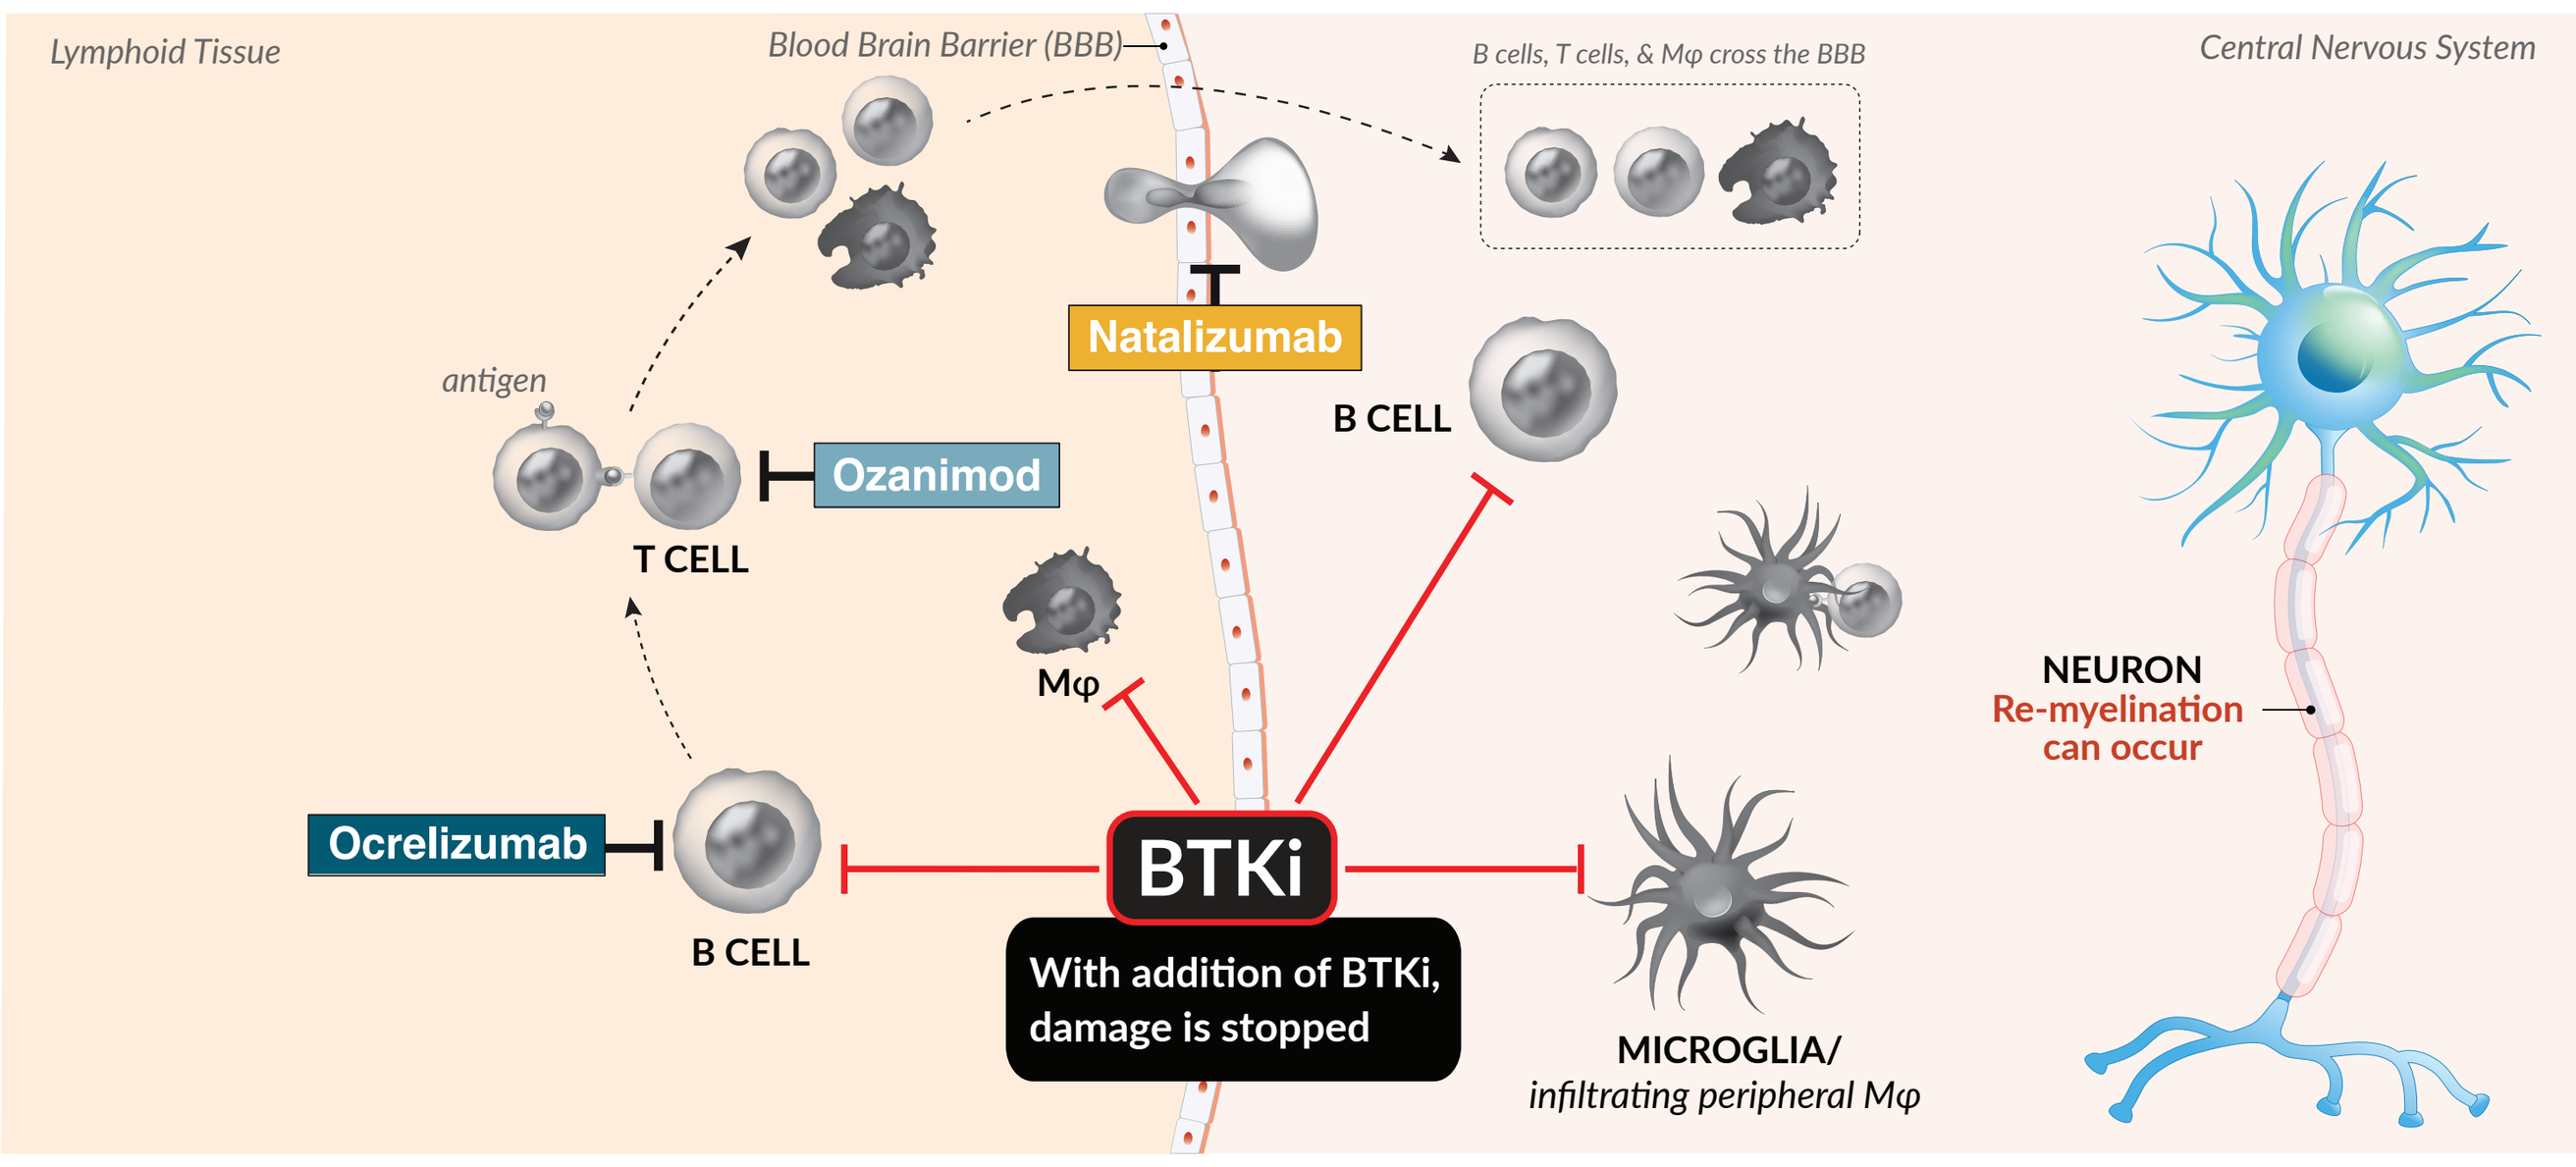
Lymphoid Tissue
Blood Brain Barrier (BBB)
B cells, T cells, & Mφ cross the BBB
Central Nervous System
Natalizumab
antigen
Ozanimod
T CELL
B CELL
Mφ
NEURON
Re-myelination can occur
Ocrelizumab
B CELL
BTKi
With addition of BTKi, damage is stopped
MICROGLIA/
infiltrating peripheral Mφ

- FcR-mediated signaling by immune complex drives pathology of myeloid cells both in periphery and CNS[6,7]
- B cells, both in the periphery and CNS, contribute to pathogenesis in MS[5]
- Presence of B cell follicles in CNS correlates with poor prognosis in secondary progressive MS (SPMS)[7]
- CNS-penetrant BTKi targets pathogenic mechanisms in both the periphery and CNS
- GB7208 is differentiated from current therapies in MS, such as B cell depletion (ocrelizumab) and S1P receptor modulators (ozanimod)

Table 1. GB7208 is differentiated from other BTKi in clinical development for MS

		tolebrutinib Phase 3	evobrutinib Phase 3	GB7208 preclinical
Pharmacology	Human WB potency (CD69 IC_{50} nM)	17	119	22
	BTK rate of inactivation (× $10^{-4}nM^{-1}min^{-1}$)	4.4	0.4	3.4
	Kinome selectivity (>50% inhibition)[a]	8/349	ND	5/349
	EGFR cellular potency (µM)	0.08	>10	>10
	% CNS target occupancy (naive mouse @2.5, 5, 10 mpk)	46%, 66%, 96%	18% @3 mpk	83%, 91%, 99%
ADME	Preclinical brain:plasma (mouse, rat, monkey[b])	0.04, 0.08, 0.66	0.03, ND, ND	0.32, 0.50, 0.81
	Human dose projection (QD for MS, Kp=0.5)	60 mg QD[c]	45 mg BID[c]	<50 mg QD
Safety	DILI prediction score[d]	Medium	Medium	Low

All reported data generated by Gossamer Bio, Inc., unless otherwise noted; ND, Not determined at Gossamer Bio, Inc.
[a]1 µM compound concentration with 1 mM ATP (AssayQuant®)
[b]NHP cassette dosing (IV)
[c]clinical dose; QD, once daily; BID, twice daily; NHP, non-human primate; WB, whole blood.
[d]Drug Induced Liver Injury score based on FDA guidance of low dose, logP, and metabolites[8]

Key parameters:

✓ Excellent CNS penetration in preclinical models, including nonhuman primate

✓ High degree of selectivity, including lack of epidermal growth factor receptor (EGFR) activity

✓ Lower potential for liver toxicity and off-target effects compared to other BTKis in the MS space based on low predicted human dose, logP, metabolites, and chemical reactivity

RESULTS

Figure 2. BTK is a key mediator of innate & adaptive immune responses implicated in neuroinflammatory and neurodegenerative diseases

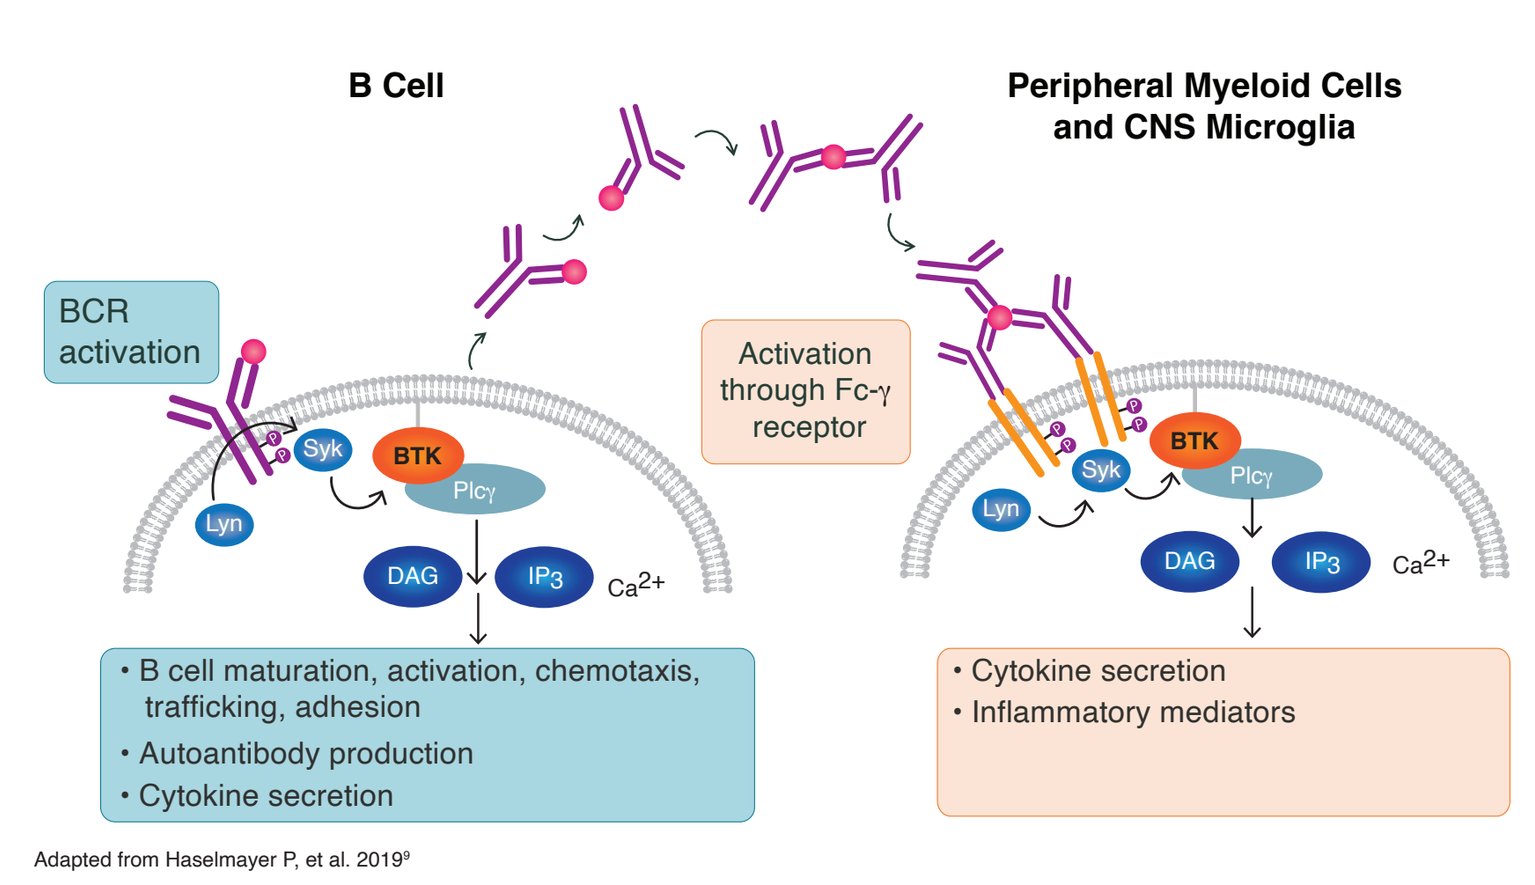
B Cell
Peripheral Myeloid Cells and CNS Microglia
BCR activation
Activation through Fc-γ receptor
Syk
BTK
Lyn
Plcγ
DAG
IP3
Ca2+
• B cell maturation, activation, chemotaxis, trafficking, adhesion
• Autoantibody production
• Cytokine secretion
• Cytokine secretion
• Inflammatory mediators
Adapted from Haselmayer P, et al. 2019[9]

GB7208 is a potent inhibitor of B cell and myeloid cell function

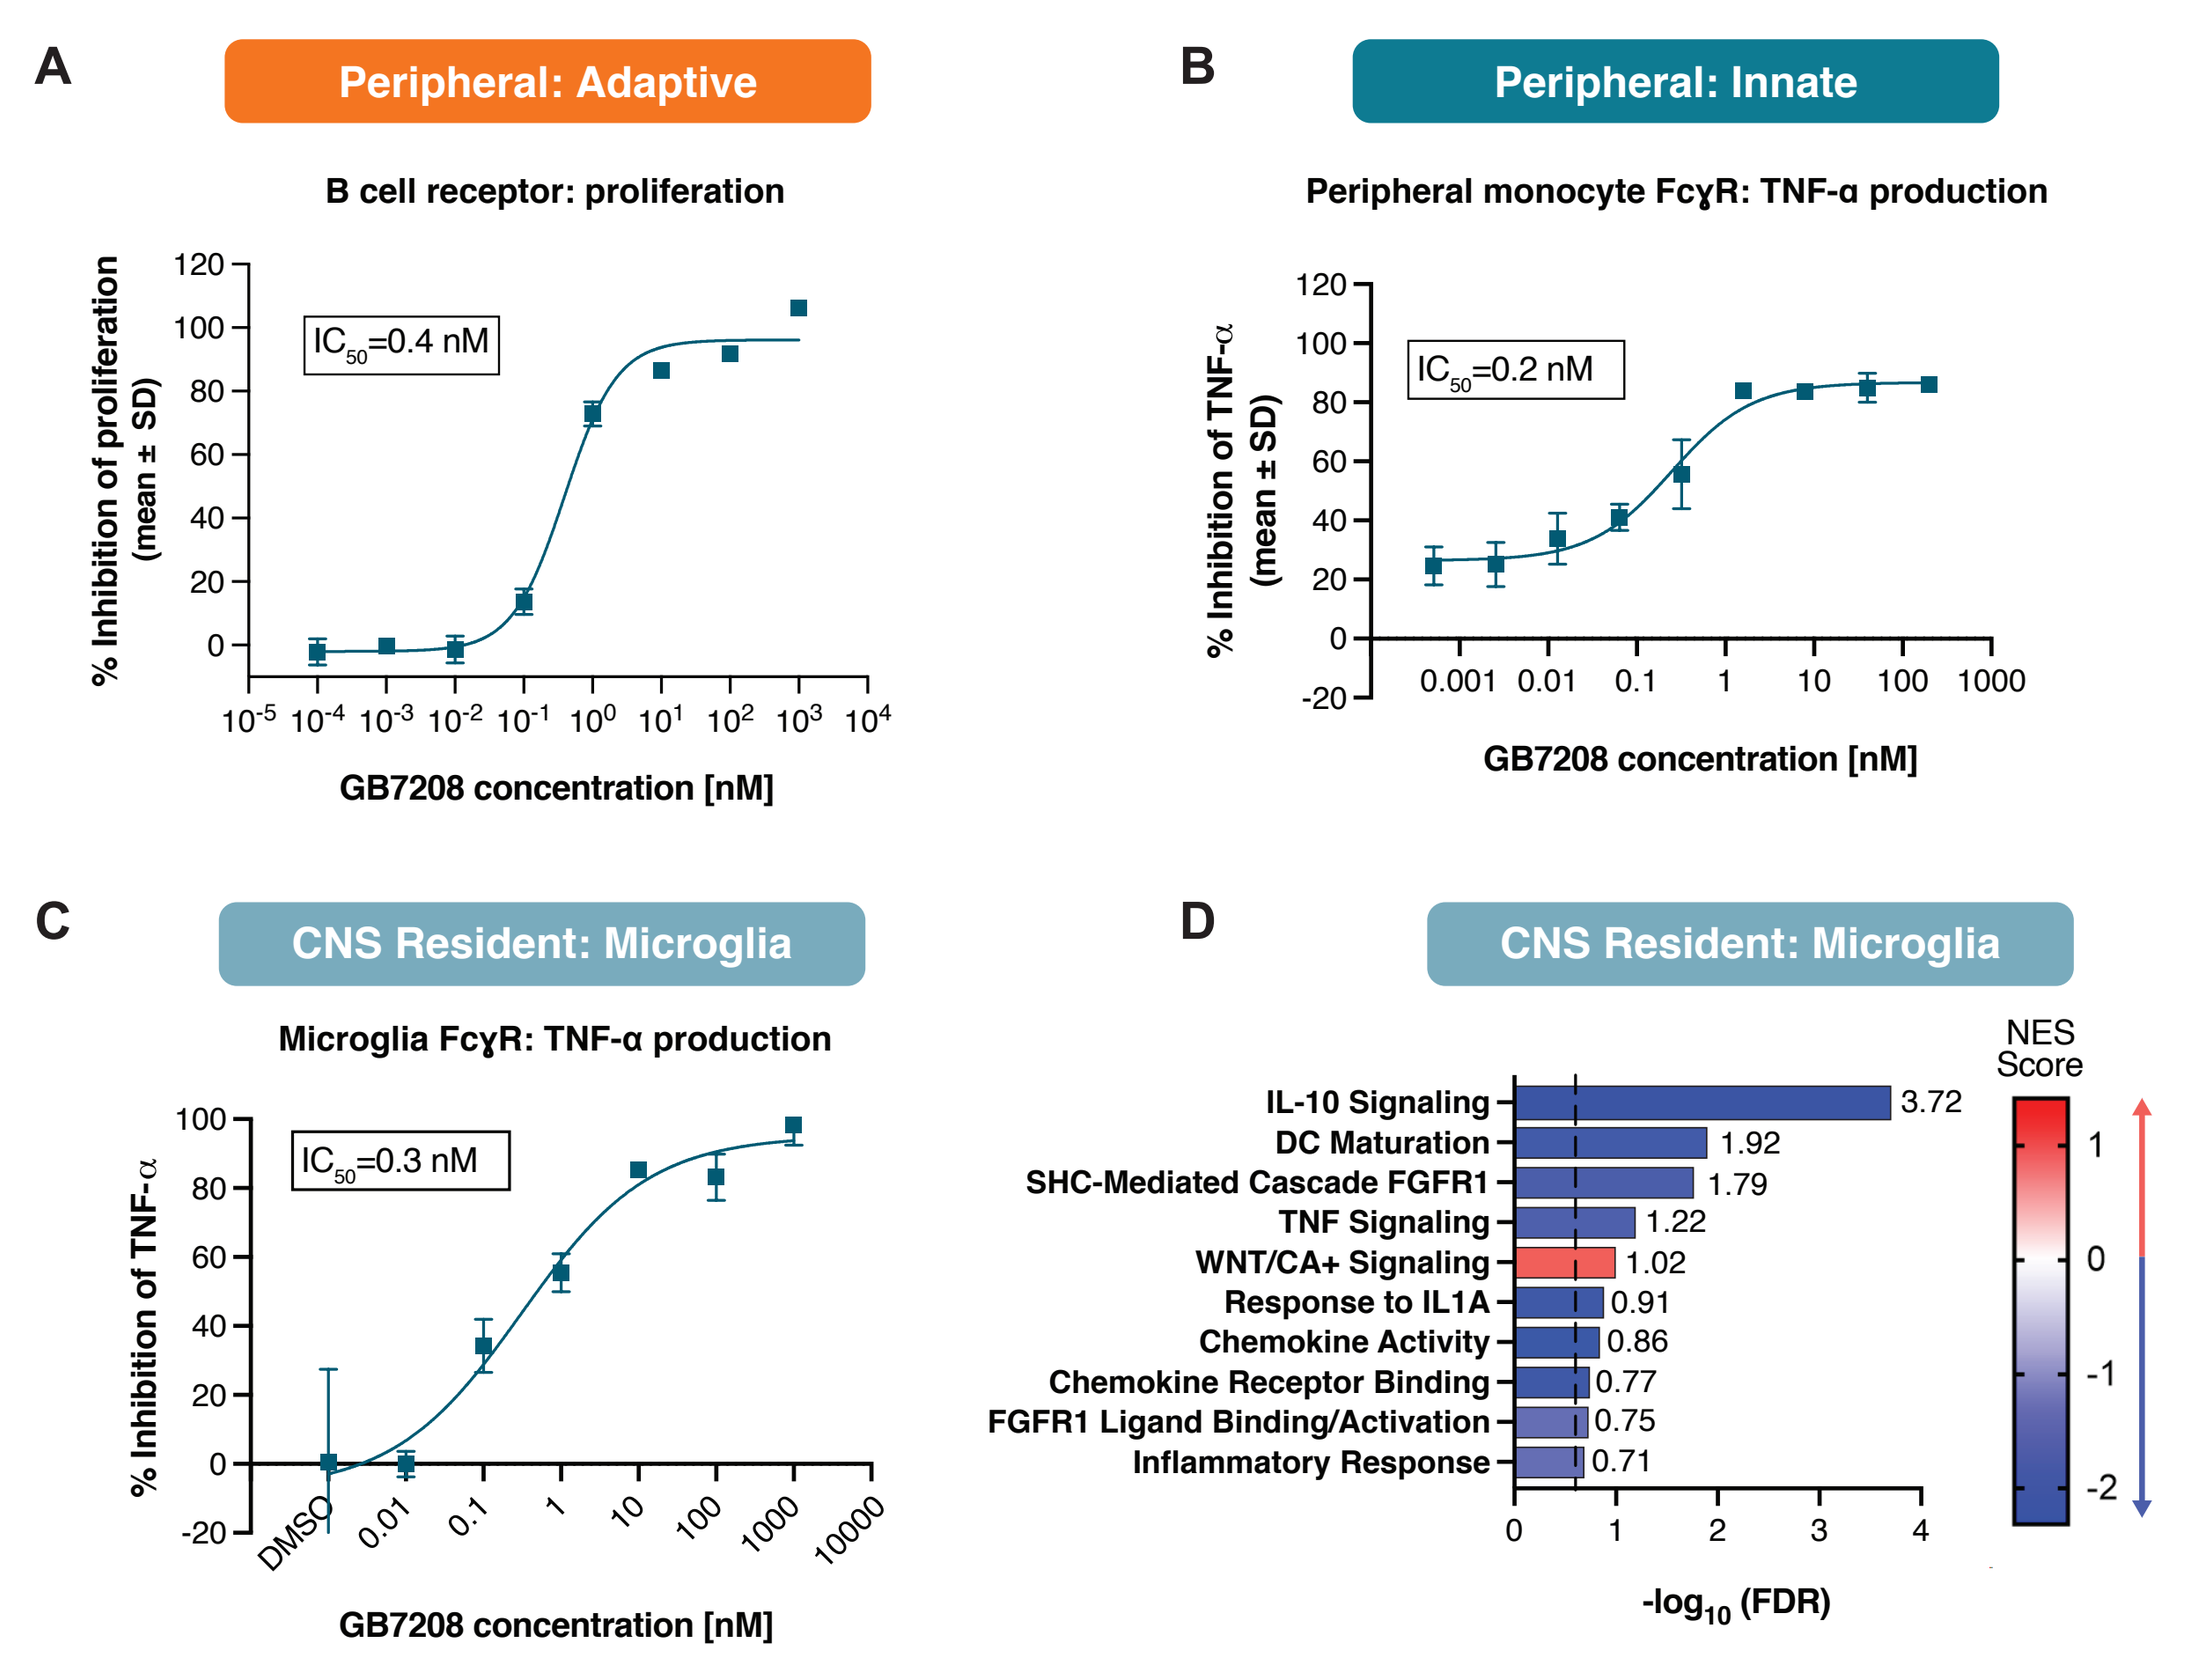
A
Peripheral: Adaptive
B cell receptor: proliferation
IC50=0.4 nM
% Inhibition of proliferation (mean ± SD)
GB7208 concentration [nM]
B
Peripheral: Innate
Peripheral monocyte FcγR: TNF-α production
IC50=0.2 nM
% Inhibition of TNF-α (mean ± SD)
GB7208 concentration [nM]
C
CNS Resident: Microglia
Microglia FcγR: TNF-α production
IC50=0.3 nM
% Inhibition of TNF-α
GB7208 concentration [nM]
D
CNS Resident: Microglia
NES Score
IL-10 Signaling 3.72
DC Maturation 1.92
SHC-Mediated Cascade FGFR1 1.79
TNF Signaling 1.22
WNT/CA+ Signaling 1.02
Response to IL1A 0.91
Chemokine Activity 0.86
Chemokine Receptor Binding 0.77
FGFR1 Ligand Binding/Activation 0.75
Inflammatory Response 0.71
-log10 (FDR)

A. Purified human B cells were stimulated with anti-IgD for 3 days. Proliferation measured with CellTiter-Glo®. **B.** CD14+ human monocytes stimulated with plate-bound IgG for 6 hours. TNF-α was measured in supernatant. **C.** iPSC-derived microglia were stimulated with heat-aggregated IgG for 24 hours. TNF-α was measured in supernatant. **D.** Pathway analysis (GSEA) of RNAseq data from human iPSC-derived microglia stimulated with HA-IgG+IFNγ. Pathways shown had significant changes (FDR <0.25) with GB7208 treatment compared to the vehicle-treated cells. Color mappings for each pathway show normalized enrichment score (NES), indicating either an activation (red) or inhibition (blue) of each pathway.

Table 2. GB7208 has a favorable *in vitro* ADME profile

Assay	GB7208	Conclusions
LM stability (mouse, rat, dog, monkey, human)	15.6, 7.05, 49.2, 4.62, 73.2 ($t_{1/2}$, min)	Low metabolic turnover in human
In vitro met ID	No major (>10%) human metabolites	Minimal DDI and tox risk from metabolites
CYP inhibition IC_{50} 1A2, 2B6, 2C8, 2C9, 2D6, 3A4 (M)[a]	25.0, 35.5, 5.00, 1.89, 23.6, 8.24 µM	Minimal DDI risk
Efflux transporter inhibition IC_{50} (BSEP, MRP2, MRP3, and MRP4)	42.1, >100, >100, 38.0 µM	Minimal DILI risk
GSH trapping	Relative % of GSH conjugates: 1.13%	Minimal DILI risk
CYP450 phenotyping	Multiple metabolic pathways	Minimal DDI risk

[a]IC_{50} was determined using midazolam as a substrate.

Figure 3. GB7208 demonstrated potent activity in collagen antibody-induced arthritis (CAIA) model

A CAIA models FcR-driven inflammation and disease[10,11]

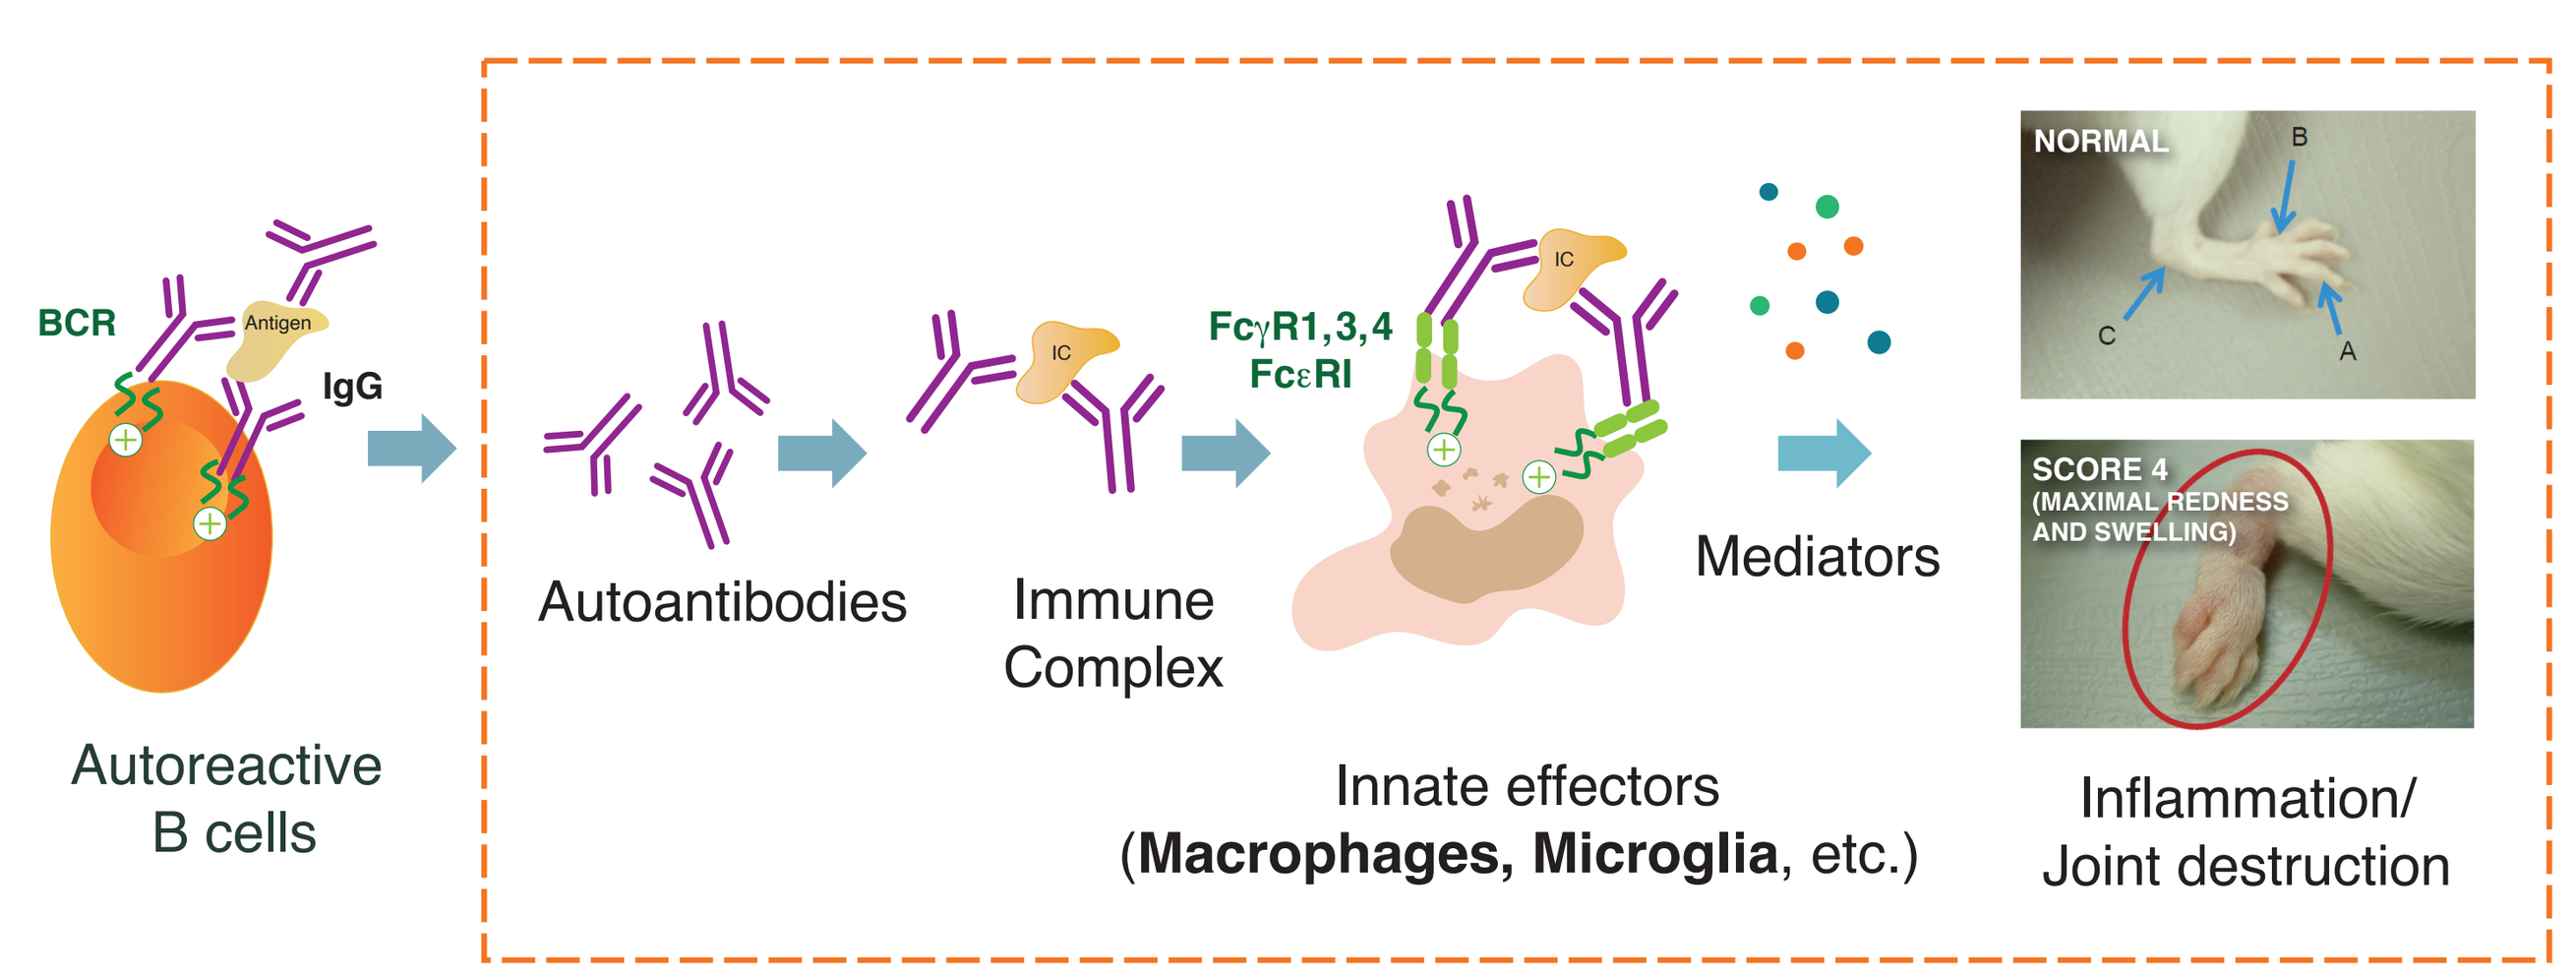
BCR
Antigen
IgG
Autoreactive B cells
Autoantibodies
IC
Immune Complex
FcγR1,3,4
FcεRI
Innate effectors (Macrophages, Microglia, etc.)
Mediators
NORMAL
SCORE 4 (MAXIMAL REDNESS AND SWELLING)
Inflammation/ Joint destruction

Image from Chondrex, Inc. Mouse arthritis scoring system. www.chondrex.com

B GB7208 significantly reduced disease (day 10)

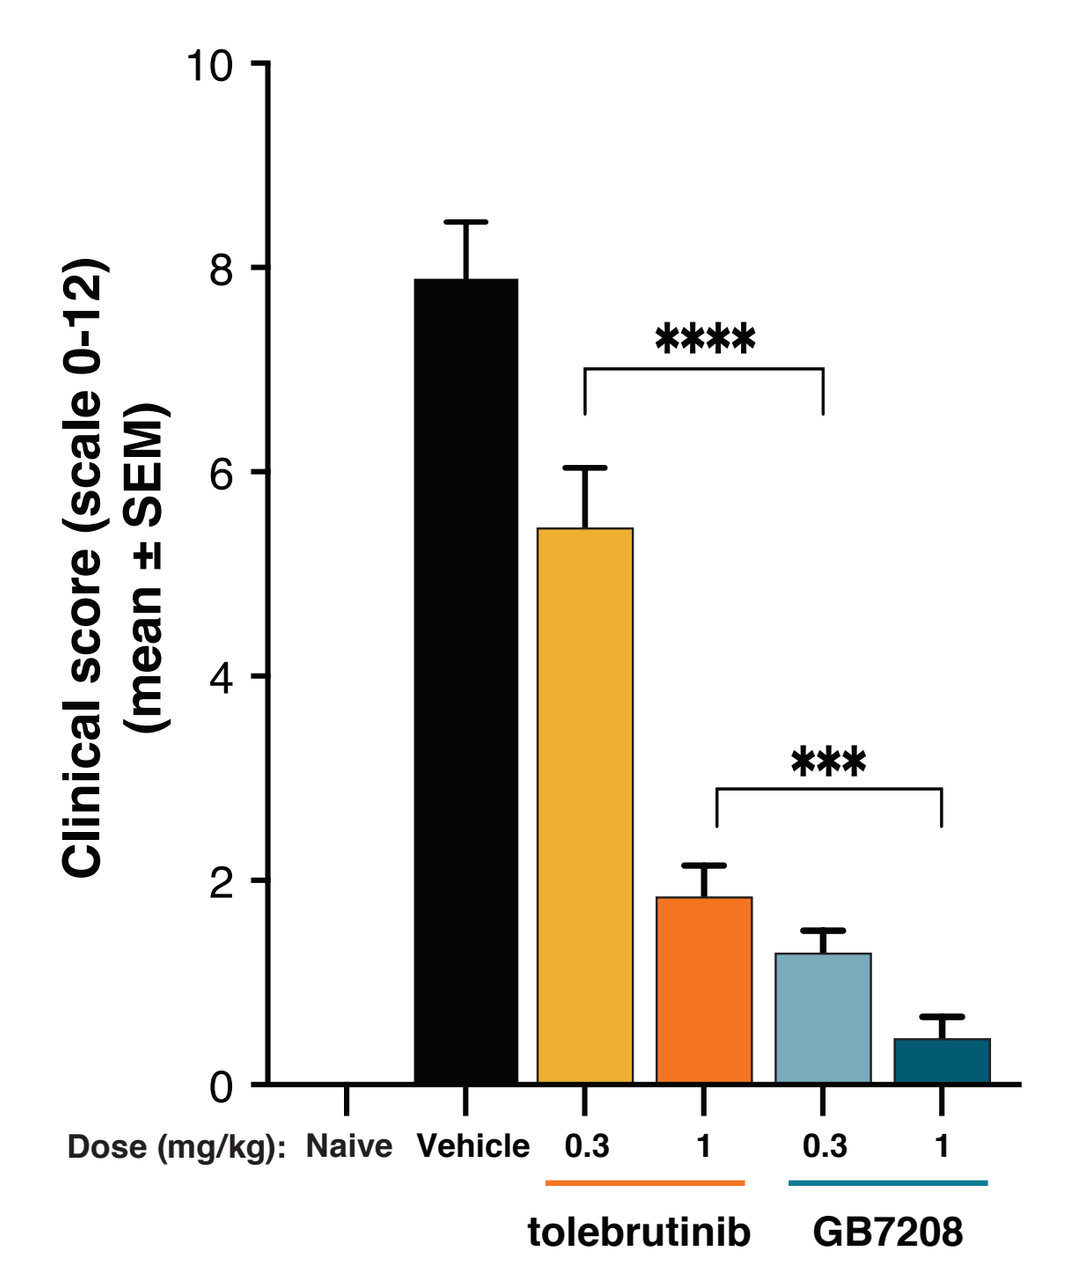
Clinical score (scale 0-12) (mean ± SEM)

Dose (mg/kg): Naive Vehicle 0.3 1 0.3 1
tolebrutinib GB7208

Mice were injected IV with anti-collagen antibody cocktail on day 0, followed by an LPS injection (IP) on day 3. Mice were subsequently randomized and compound dosing was initiated (PO, QD). Mice were dosed for nine days with either GB7208 or tolebrutinib. The severity of arthritis was graded on three separate joints on each paw on a 0-4 scale on days 3, 7 (not shown), and 10 (**panel B**). Clinical score representative of the sum of scores of all paws. Statistics determined by Student's *t*-test. ***P<0.001, ****P<0.0001.

Figure 4. GB7208 inhibited proliferation in a mouse model of microglia activation induced by anti-MOG autoantibodies

- MOG autoantibodies drive activation and proliferation of microglia in mice with an intact BBB
- Activation is driven by BTK-dependent FcR signaling

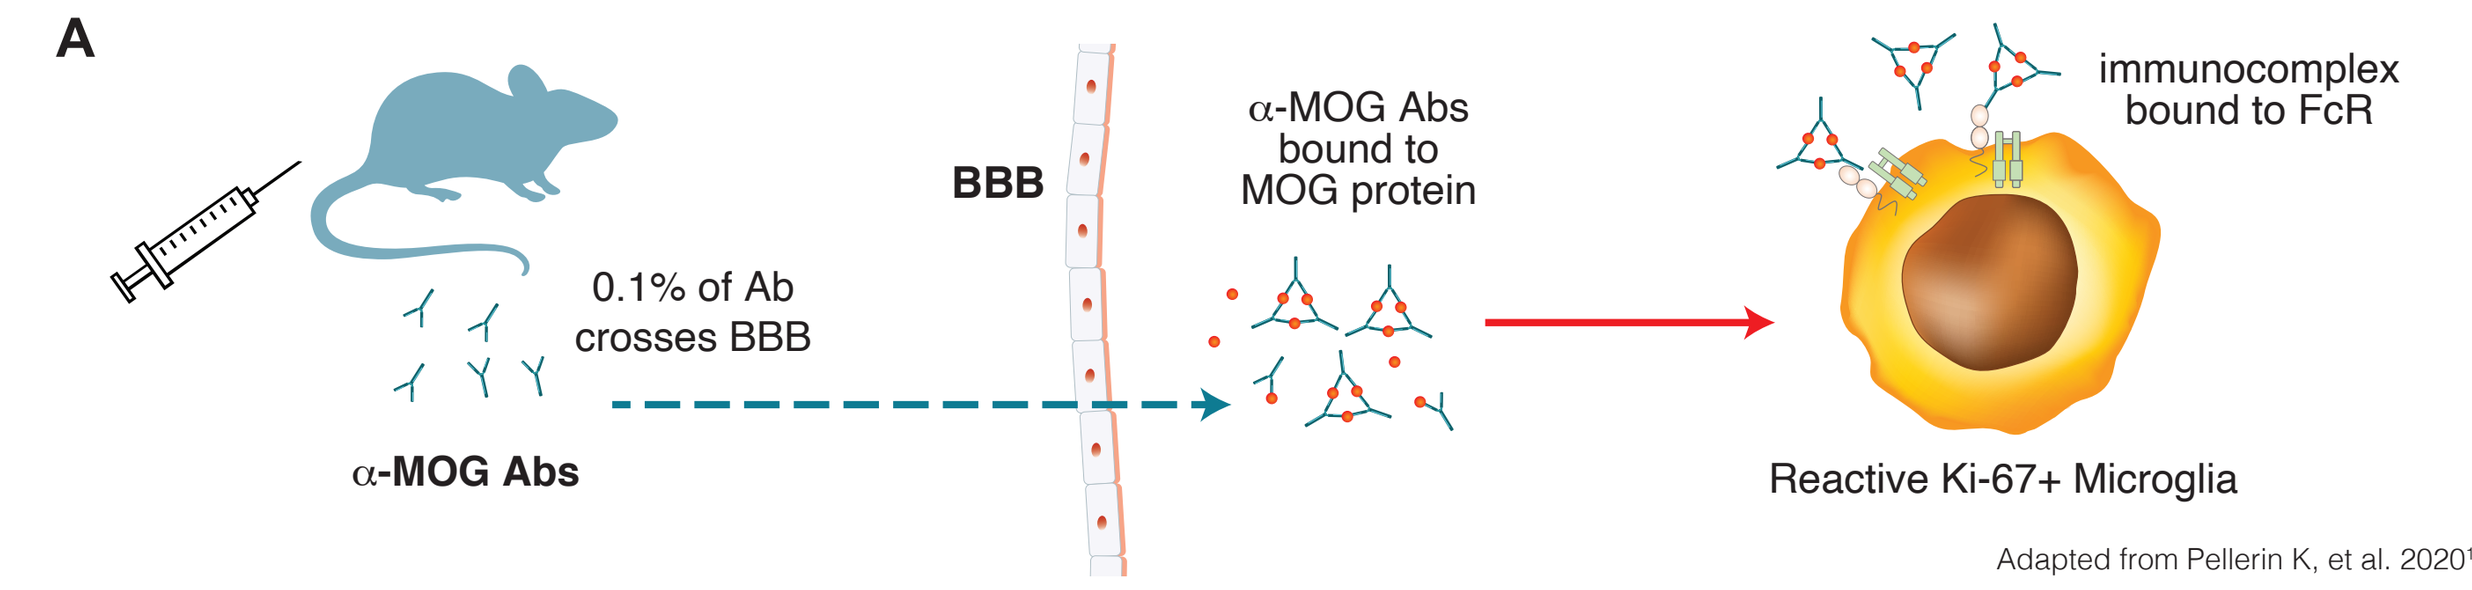
A
0.1% of Ab crosses BBB
α-MOG Abs
BBB
α-MOG Abs bound to MOG protein
immunocomplex bound to FcR
Reactive Ki-67+ Microglia
Adapted from Pellerin K, et al. 2020[12]

A. On day 0, mice were injected IP with 30 mg/kg anti-MOG IgG and compound dosing (PO, QD) was initiated. Mice were dosed for four days with either GB7208 or tolebrutinib; 1 hour after the final dose, brain and spinal cord samples were collected. **B.** Ki-67 was measured by flow cytometry and **C.** BTK target occupancy was measured using a probe-based ELISA. Statistics determined by one-way ANOVA. *P<0.05, **P<0.01, ***P<0.001, ****P<0.0001.

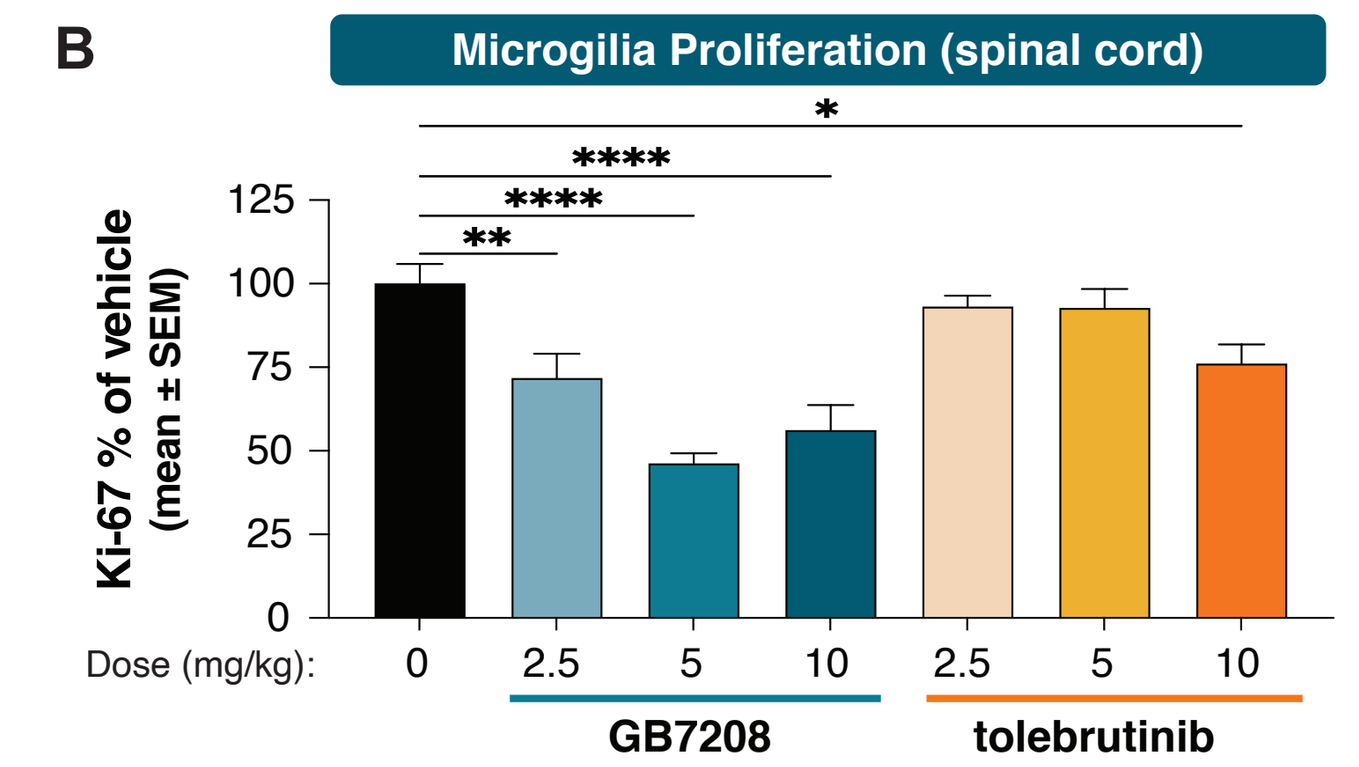
B
Microglia Proliferation (spinal cord)
Ki-67 % of vehicle (mean ± SEM)
Dose (mg/kg): 0 2.5 5 10 2.5 5 10
GB7208 tolebrutinib

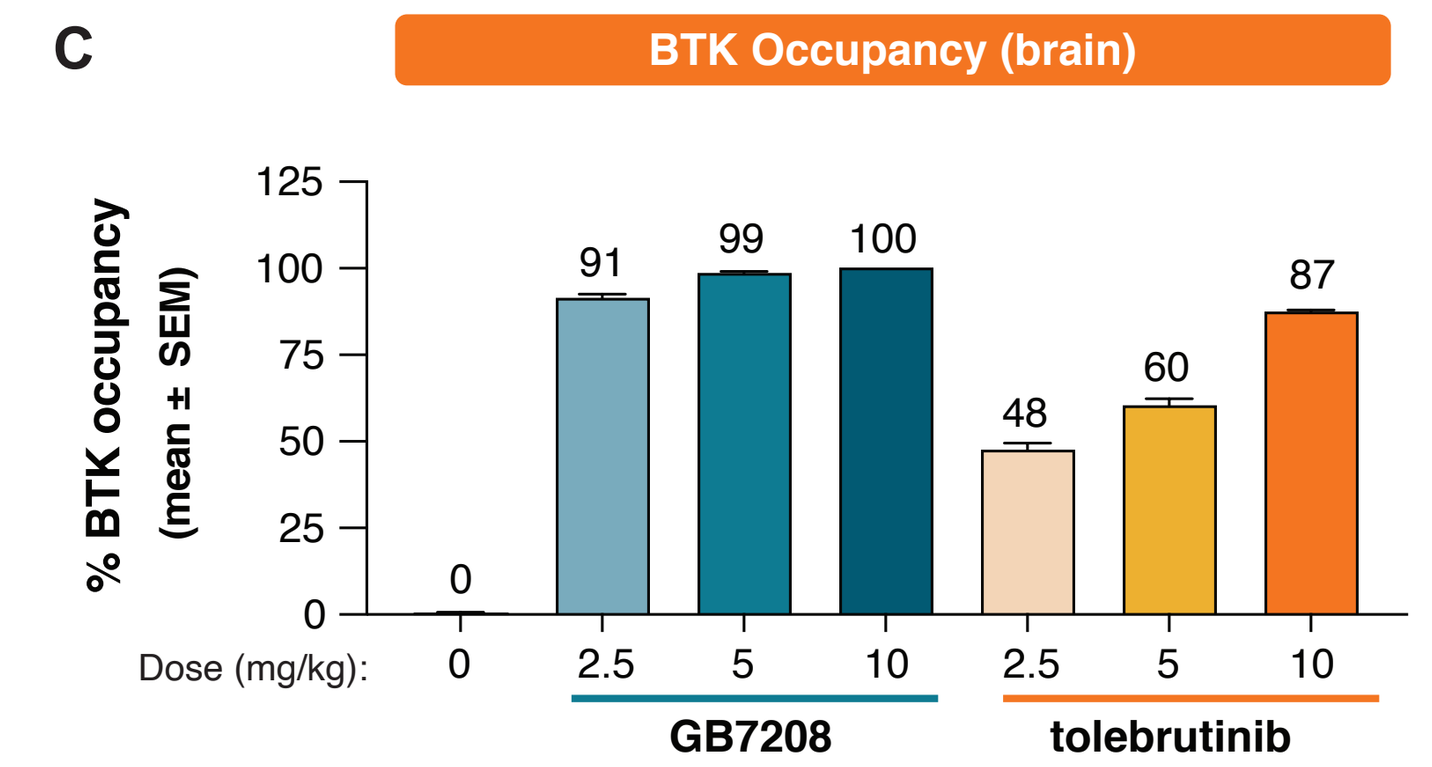
C
BTK Occupancy (brain)
% BTK occupancy (mean ± SEM)
0 91 99 100 48 60 87
Dose (mg/kg): 0 2.5 5 10 2.5 5 10
GB7208 tolebrutinib

Figure 5. GB7208 demonstrated efficacy in multiple preclinical models of MS

Study included n=12 C57BL/6 mice (top) and n=15 NOD/ShiLtJ mice per group (bottom). For the C57BL/6 model, mice were immunized with MOG_{35-55}/CFA on day 0, administered pertussis toxin on days 0 and 1. Dosing initiated day 0 (PO, QD) and study terminated day 28. For the NOD/ShiLtJ model, mice were immunized with MOG_{35-55}/CFA on day 0 and administered pertussis toxin on days 0 and 1. A second immunization with MOG_{35-55}/CFA occurred on day 21, with pertussis toxin administered on days 21 and 22. Dosing was initiated on day 34 (PO, QD) and continued out to day 60, when the study was terminated. **A.** Clinical score.
B. Demyelination score on mouse spines isolated at conclusion of study. The spine was fixed, paraffin-embedded, sectioned, and stained with anti-myelin basic protein antibody. An area on each section was scored on a scale from 0–5 (0: no demyelination; 5: >50% demyelination). Graphs represent the average scores generated from scores given to the cervical, thoracic, and lumbar sections of the spine. Graphs generated using GraphPad Prism software. SEM, standard error of the mean. Statistics determined by one-way ANOVA. *P<0.05, ***P<0.001, ****P<0.0001.

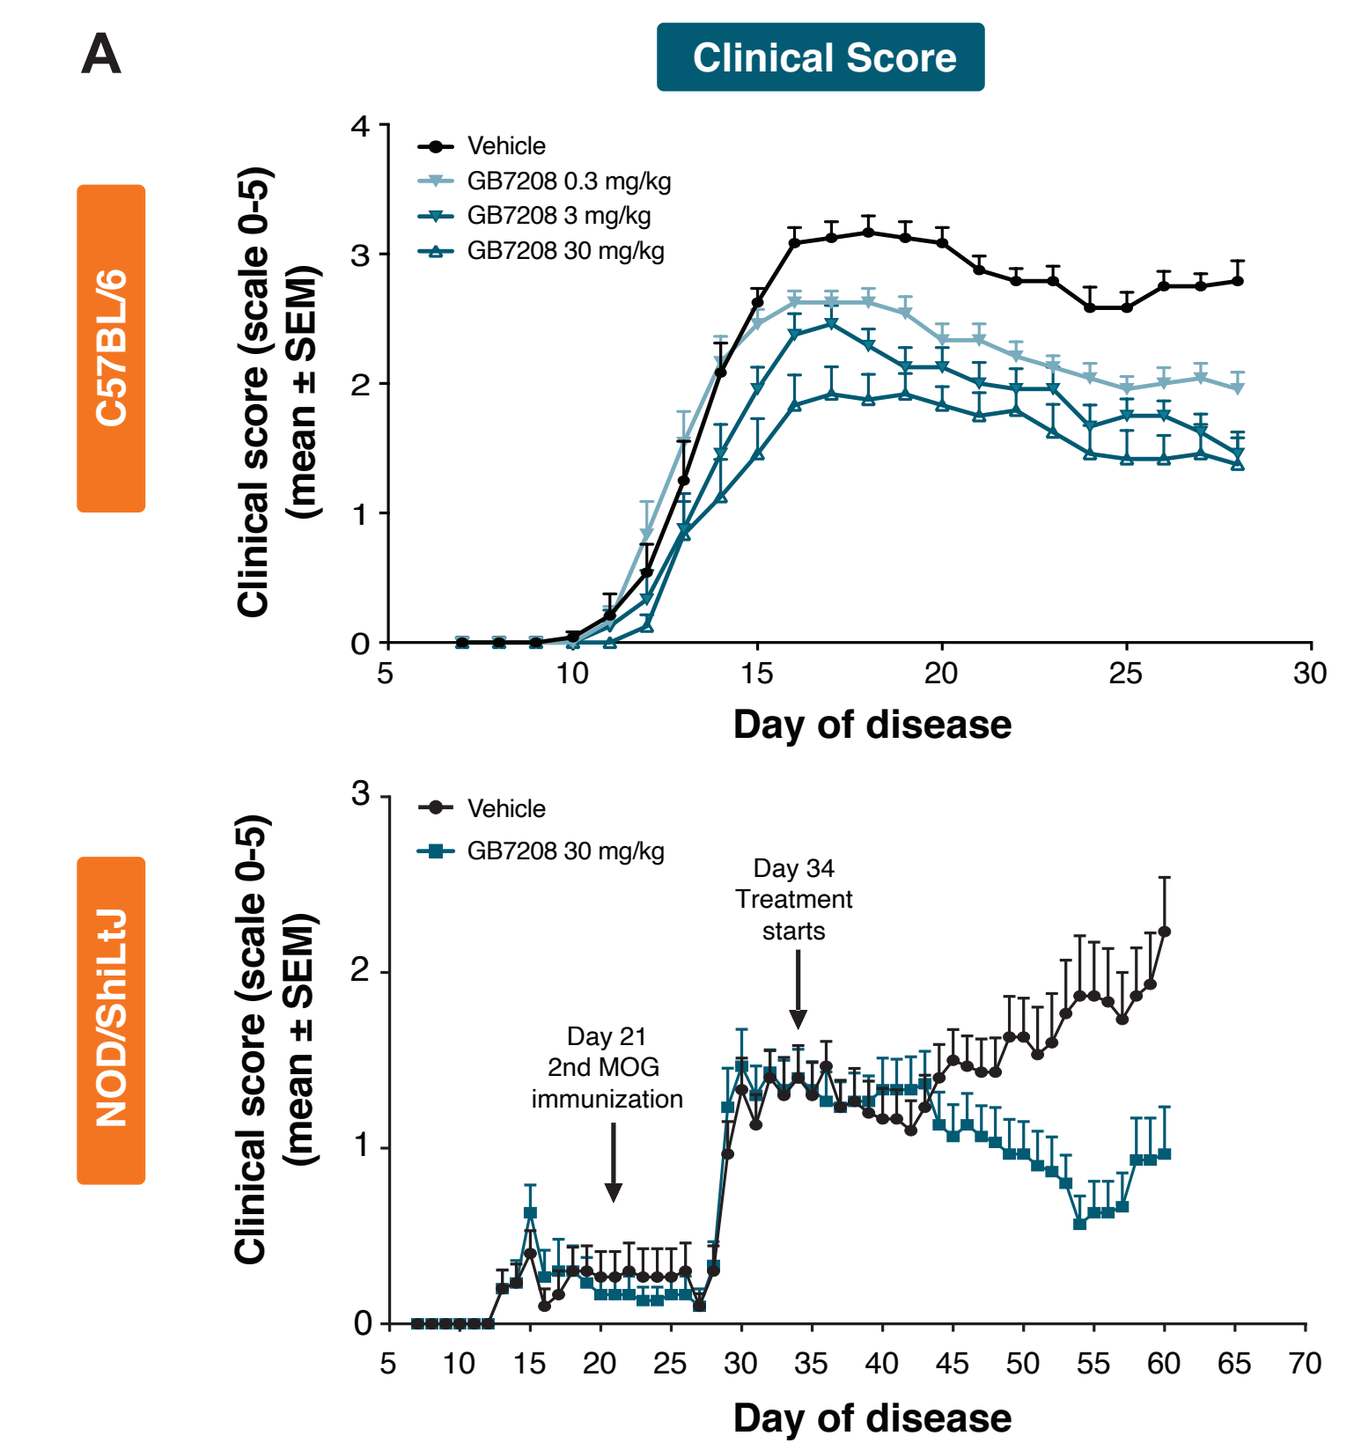
A
Clinical Score
C57BL/6
Vehicle
GB7208 0.3 mg/kg
GB7208 3 mg/kg
GB7208 30 mg/kg
Clinical score (scale 0-5) (mean ± SEM)
Day of disease
NOD/ShiLtJ
Vehicle
GB7208 30 mg/kg
Day 21 2nd MOG immunization
Day 34 Treatment starts
Day of disease

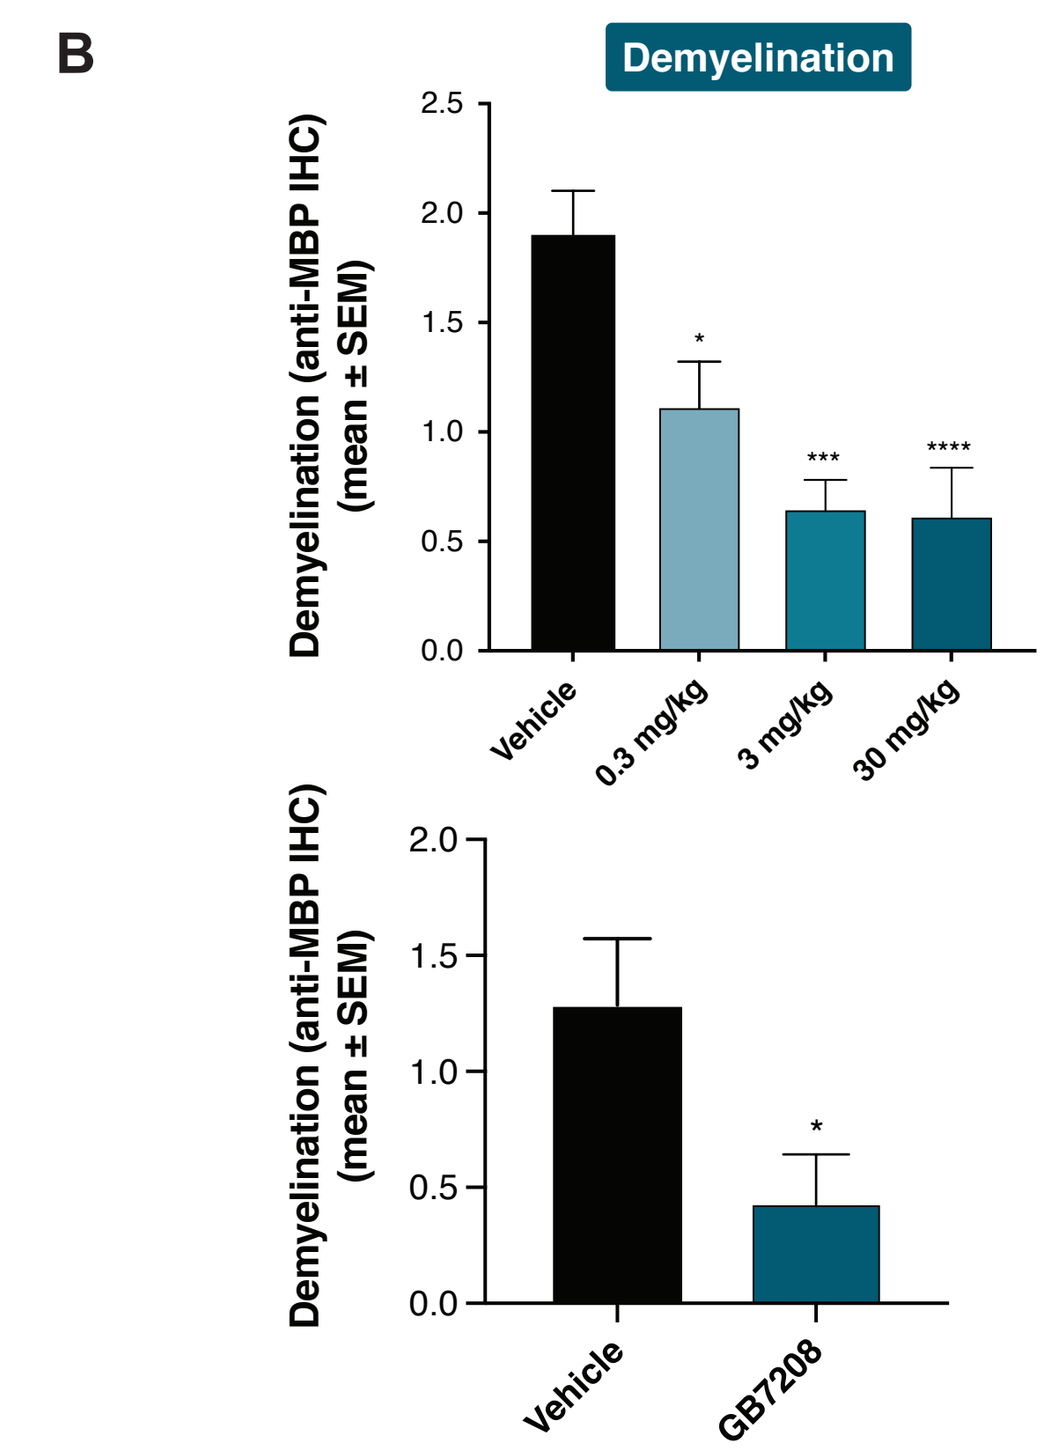
B
Demyelination
Demyelination (anti-MBP IHC) (mean ± SEM)
Vehicle
0.3 mg/kg
3 mg/kg
30 mg/kg
Vehicle
GB7208

SUMMARY

- GB7208 is a differentiated BTK inhibitor designed to be potent, selective, and highly CNS penetrant, with low human dose predictions
- GB7208 potently inhibits the BTK-dependent innate and adaptive immune responses relevant to MS pathogenesis
- The GB7208 preclinical profile predicts a lower risk of liver toxicity
- GB7208 demonstrated high CNS penetrance across multiple species, including nonhuman primates
- In both peripheral and CNS mouse models of FcR-driven activation of myeloid cells, GB7208 led to significant reduction in disease activity and inhibition of microglia proliferation, which is consistent with high CNS target occupancy

REFERENCES

1. Portaccio E, et al. *Brain.* 2022;145(8):2796-2805. **2.** Kee R, et al. *Biomedicines.* 2022;10(10):2604. **3.** Giovannoni G, et al. *Ther Adv Neurol Disord.* 2022;15:17562864211066751. **4.** Corneth OBJ, et al. *Curr Top Microbiol Immunol.* 2016;393:67-105. **5.** Weber ANR, et al. *Front Immunol.* 2017;8:1454. **6.** von Büdingen HC, et al. *J Clin Invest.* 2012;122(12):4533-4543. **7.** Serafini B, et al. *Brain Pathol.* 2004;14(2):164-174. **8.** Chen M, et al. *Hepatology.* 2016;64(3):931-940. **9.** Haselmayer P, et al. *J Immunol.* 2019;202(10):2888-2906. **10.** Gillooly KM, et al. *PLoS ONE.* 2017;12(7):e0181782. **11.** Kagari T, et al. *J Immunol.* 2003;170(8):4318–4324. **12.** Pellerin K, et al. *Mult Scler.* 2020;26(3 suppl):P0941.

ACKNOWLEDGEMENTS

The authors gratefully acknowledge Warren Plaisted and Kevin Lu. The authors also acknowledge Daniel Buzzard, Vijay Kumar, Nidhi Arora, Martin Rowbottom, and Craig Coburn for medicinal chemistry efforts in the development and identification of GB7208.

DISCLOSURES

All authors are employees and stockholders of Gossamer Bio, Inc. This research was funded by GB005, Inc., a wholly owned subsidiary of Gossamer Bio, Inc.

Presented at the ACTRIMS Forum 2023, San Diego, California, February 23-25, 2023