401

SERALUTINIB FOR THE TREATMENT OF PULMONARY ARTERIAL HYPERTENSION (PAH): RESULTS FROM THE PHASE 2 TORREY TRIAL

Robert P. Frantz[1*], Vallerie V. McLaughlin[2*], Sandeep Sahay[3], Pilar Escribano Subías[4], Ronald L. Zolty[5], Raymond L. Benza[6], Richard N. Channick[7], Kelly M. Chin[8], Anna R. Hemnes[9], Luke S. Howard[10], Olivier Sitbon[11], Jean-Luc Vachiéry[12], Roham T. Zamanian[13], Matt Cravets[14], Robert F. Roscigno[14], David Mottola[14], Erin Elman[14], Ed Parsley[14], Richard Aranda[14], Lawrence S. Zisman[14], Hossein-Ardeschir Ghofrani[15] on behalf of the TORREY Study Investigators

[1]Mayo Clinic, Rochester, MN, USA; [2]University of Michigan, Ann Arbor, MI, USA; [3]Houston Methodist Hospital/Weill Cornell Medicine, Houston, TX, USA; [4]University Hospital 12 de Octubre, Complutense University, Madrid, Spain; [5]University of Nebraska Medical Center, Omaha, NE, USA; [6]Ohio State University, Columbus, OH, USA; [7]UCLA Medical Center, Los Angeles, CA, USA; [8]UT Southwestern Medical Center, Dallas, TX, USA; [9]Vanderbilt University, Nashville, TN, USA; [10]Imperial College Healthcare NHS Trust, London, UK; [11]Université Paris-Saclay, Le Kremlin-Bicêtre, France; [12]HUB – Hôpital Erasme, Brussels, Belgium; [13]Stanford University, Stanford, CA, USA; [14]Gossamer Bio, Inc., San Diego, CA, USA; [15]Justus Liebig University Giessen, Giessen, Germany. *Contributed equally to this work

TORREY STUDY

Presented at
The American Thoracic Society Annual Meeting, Washington, DC, May 19–24, 2023.

BACKGROUND

- PDGFR, CSF1R, and c-KIT kinase pathways play key roles in the inflammation, proliferation, and fibrosis that drive pulmonary vascular remodeling in PAH
- Vascular remodeling leads to increased pulmonary vascular resistance (PVR) and decreased pulmonary artery compliance (PAC), resulting in right heart failure
- Seralutinib is a novel tyrosine kinase inhibitor designed for dry powder inhalation that targets these dysfunctional pathways[1] and has the potential to improve PVR and PAC (**Figure 1**)

Figure 1. Seralutinib mechanism of action[1]

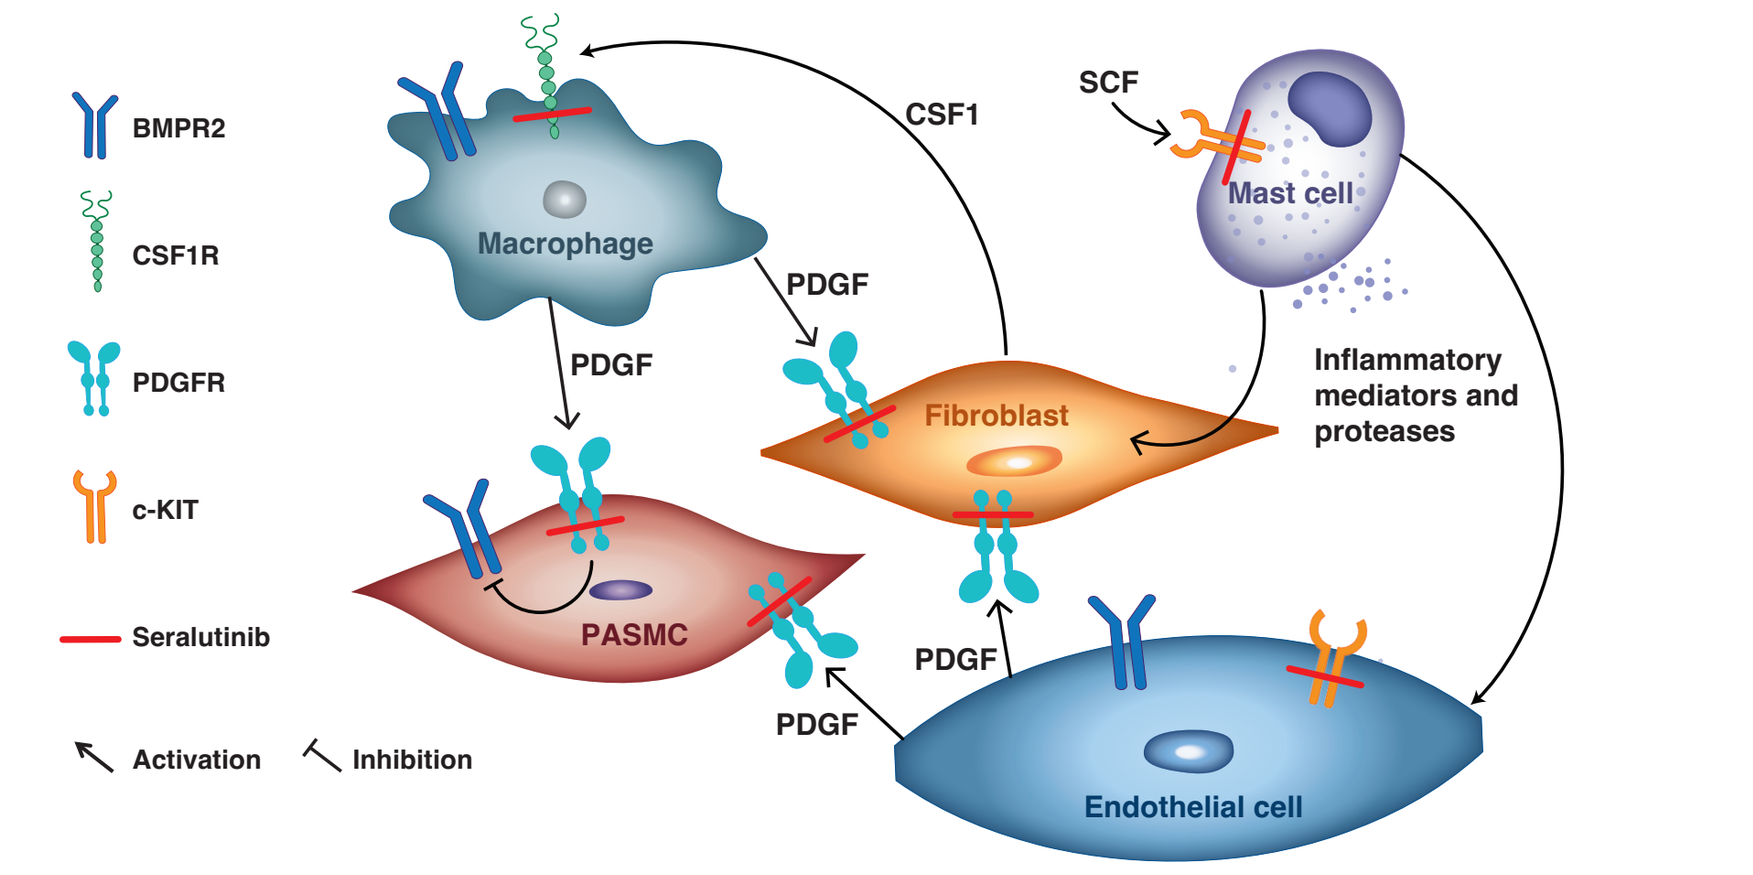

BMPR2
CSF1R
PDGFR
c-KIT
Seralutinib
Activation
Inhibition
Macrophage
CSF1
SCF
Mast cell
PDGF
Fibroblast
Inflammatory mediators and proteases
PASMC
Endothelial cell

Figure: Galkin A et al. *Eur Respir J*. 2022;60(6):2102356.

METHODS

- Phase 2, randomized, double-blind, placebo-controlled, multicenter study (NCT04456998)[2]
- Inclusion criteria: World Health Organization (WHO) Group 1 PH, Functional Class (FC) II or III, on standard background therapies, 6-minute walk distance (6MWD) ≥ 150 m and ≤ 550 m, PVR ≥ 400 dyne*s/cm[5]
- Patients randomized 1:1 to seralutinib or placebo twice daily for 24 weeks (**Figure 2**)
- Endpoints
 - Primary: Change in PVR from baseline (BL) to Week 24
 - Secondary: Change in 6MWD from BL to Week 24
 - Exploratory: NT-proBNP
 - Safety assessed during scheduled visits
- RHC analyses: PAC[a] and other cardiopulmonary hemodynamic parameters
- Statistical analyses: Change in PVR from BL to Week 24 based on analysis of covariance (ANCOVA) modelling with multiple imputation. Secondary and exploratory endpoints analyzed used mixed-effects models for repeated measures (MMRM). Analyses based on intention-to-treat (ITT) population.

[a] PAC = stroke volume/(pulmonary artery pulse pressure)

Figure 2. Study schema

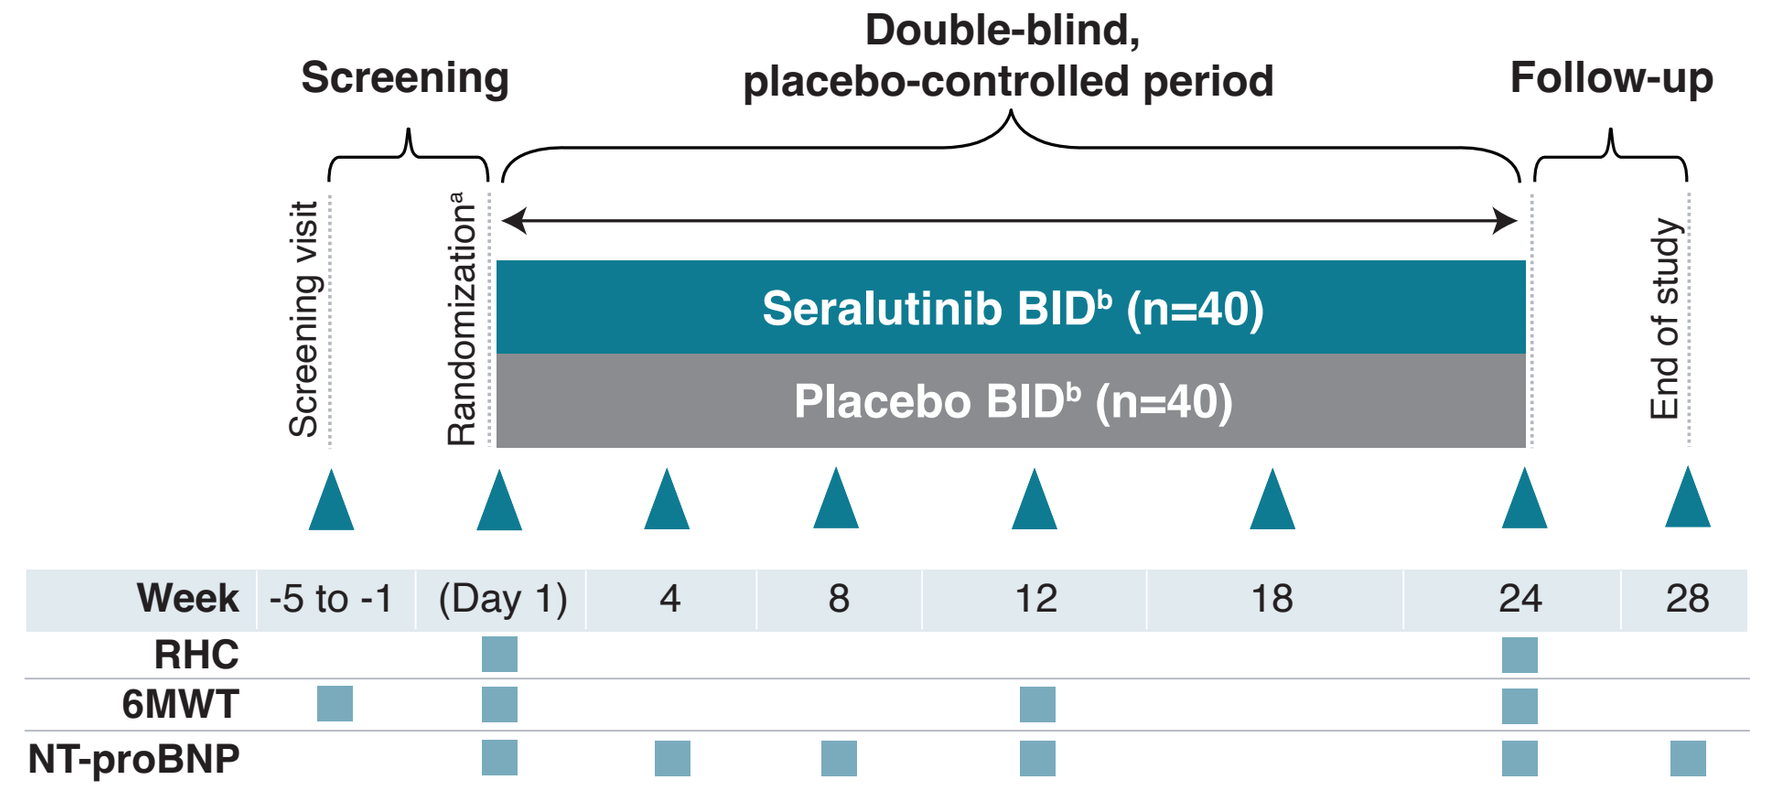

Screening
Double-blind, placebo-controlled period
Follow-up
Screening visit
Randomization[a]
Seralutinib BID[b] (n=40)
Placebo BID[b] (n=40)
End of study
Week -5 to -1 (Day 1) 4 8 12 18 24 28
RHC
6MWT
NT-proBNP

[a] Randomization stratified by PVR (< 800 dyne*s/cm[5] vs ≥ 800 dyne*s/cm[5]).
[b] Patients started on 60 mg (4 inhalations) BID and after 2 weeks escalated to 90 mg (6 inhalations) BID as tolerated.
6MWT, 6-minute walk test; BID, twice daily; RHC, right heart catheterization.

RESULTS

- 86 patients randomized to treatment at 40 sites worldwide; 80 patients completed the study
- Seralutinib and placebo groups balanced except for WHO FC (seralutinib, 68%/32% FCII/III; placebo, 48%/52% FCII/III)
- 44.2% received parenteral prostacyclin

Table 1. Baseline demographics and disease characteristics. Data presented as mean (SD) unless otherwise noted

Characteristic	Placebo (N=42)	Seralutinib (N=44)
Age, y	49.5 (11.81)	48.3 (12.70)
Female, n (%)	38 (90.5)	40 (90.9)
Race, n (%)		
White	37 (88.1)	37 (84.1)
Other	5 (11.9)	7 (15.9)
Years since PAH diagnosis	8.78 (7.218)	8.07 (7.074)
PAH classification, n (%)		
Idiopathic	22 (52.4)	20 (45.5)
Heritable	5 (11.9)	10 (22.7)
Associated with CTD	11 (26.2)	6 (13.6)
Drug or toxin-induced	4 (9.5)	5 (11.4)
Associated with congenital shunts	0	3 (6.8)
WHO FC, n (%)		
Class II	20 (47.6)	30 (68.2)
Class III	22 (52.4)	14 (31.8)

Characteristic	Placebo (N=42)	Seralutinib (N=44)
mPAP, mmHg	50.0 (9.58)	51.9 (11.71)
PCWP or LVEDP, mmHg	10.8 (2.87)	11.0 (2.82)
Cardiac output, L/min	4.865 (0.985)	5.033 (1.037)
PVR, dyne*s/cm[5]	661.3 (164.91)	675.8 (240.35)
6MWD, m	407.1 (107.02)	408.6 (75.11)
NT-proBNP, ng/L	645.6 (1158.75)	611.0 (714.58)
Number of background therapies, n (%)		
< 3	18 (42.9)	19 (43.2)
3	24 (57.1)	25 (56.8)
Prostacyclin/Prostacyclin receptor agonist use, n (%)		
Parenteral	19 (45.2)	19 (43.2)
Oral	10 (23.8)	10 (22.7)

6MWD, 6-minute walk distance; CTD, connective tissue disease; FC, functional class; LVEDP, left ventricular end-diastolic pressure; mPAP, mean pulmonary arterial pressure; PAH, pulmonary arterial hypertension; PCWP, pulmonary capillary wedge pressure; PVR, pulmonary vascular resistance; WHO, World Health Organization.

RESULTS

Figure 3. A. Seralutinib significantly reduced PVR at Week 24 vs placebo (14.3%, p=0.0310). **B.** Reduction of PVR in FC III patients (20.8%, p=0.0427). **C.** Reduction of PVR in patients with a baseline REVEAL 2.0 risk score of ≥ 6 (22.7%, p=0.0134)

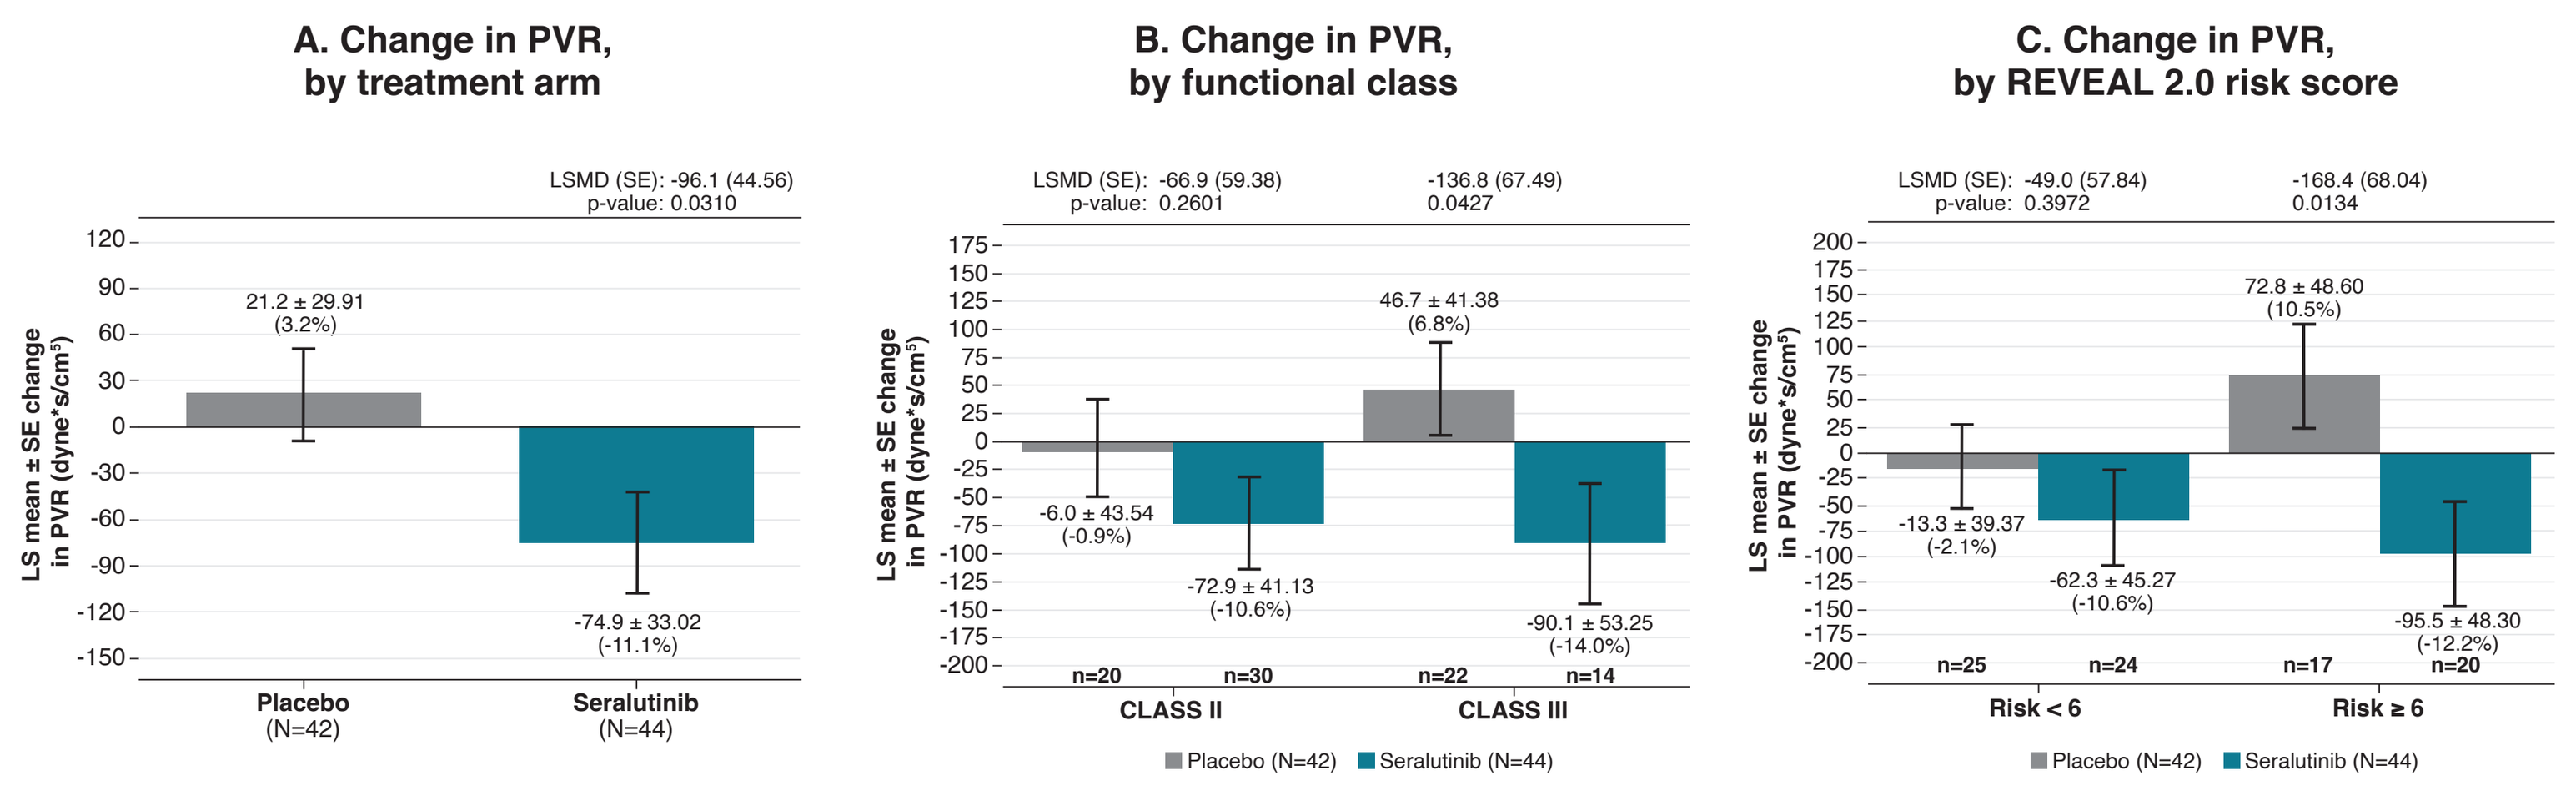

A. Change in PVR, by treatment arm
LSMD (SE): -96.1 (44.56)
p-value: 0.0310
21.2 ± 29.91 (3.2%)
-74.9 ± 33.02 (-11.1%)
Placebo (N=42)
Seralutinib (N=44)
LS mean ± SE change in PVR (dyne*s/cm5)
B. Change in PVR, by functional class
LSMD (SE): -66.9 (59.38) p-value: 0.2601
-136.8 (67.49) 0.0427
-6.0 ± 43.54 (-0.9%)
-72.9 ± 41.13 (-10.6%)
46.7 ± 41.38 (6.8%)
-90.1 ± 53.25 (-14.0%)
n=20 n=30 n=22 n=14
CLASS II
CLASS III
C. Change in PVR, by REVEAL 2.0 risk score
LSMD (SE): -49.0 (57.84) p-value: 0.3972
-168.4 (68.04) 0.0134
-13.3 ± 39.37 (-2.1%)
-62.3 ± 45.27 (-10.6%)
72.8 ± 48.60 (10.5%)
-95.5 ± 48.30 (-12.2%)
n=25 n=24 n=17 n=20
Risk < 6
Risk ≥ 6
Placebo (N=42) Seralutinib (N=44)

Figure 4. Consistent reduction of PVR across pre-specified subgroups with seralutinib treatment

Subgroup	Placebo N	Seralutinib N	LSMD (95% CI)[a]	p-value[a]
Overall	42	44	-96.1 (-183.5, -8.8)	0.0310
Sex				
Female	38	40	-97.5 (-190.2, -4.9)	0.0390
PAH background medications				
< 3	18	19	-105.8 (-235.4, 23.9)	0.1099
≥ 3	24	25	-89.1 (-205.8, 27.5)	0.1343
Prostacyclins at baseline				
Yes	29	29	-93.9 (-201.9, 14.1)	0.0885
No	13	15	-105.4 (-252.8, 41.9)	0.1609
Age groups				
< Median (47 y)	19	24	-143.6 (-268.1, -19.0)	0.0239
≥ Median (47 y)	23	20	-49.1 (-171.1, 72.9)	0.4303
< 65 y	37	36	-84.0 (-178.1, 10.1)	0.0801
≥ 65 y	5	8	-192.2 (-423.9, 39.5)	0.1040
Race				
White	37	37	-100.3 (-192.1, -8.4)	0.0324
Non-White	5	7	-44.8 (-290.7, 201.1)	0.7210
Region				
North America	30	29	-65.9 (-172.6, 40.8)	0.2262
Rest of world	12	15	-164.7 (-316.8, -12.7)	0.0337
PVR strata				
< 800 dyne*s/cm[5]	34	34	-91.0 (-186.1, 4.1)	0.0606
≥ 800 dyne*s/cm[5]	8	10	-119.6 (-332.9, 93.6)	0.2712
WHO functional class				
II	20	30	-66.9 (-183.3, 49.5)	0.2601
III	22	14	-136.8 (-269.1, -4.5)	0.0427
Connective tissue disease				
Yes	11	6	-55.8 (-294.4, 182.7)	0.6459
No	31	38	-95.5 (-190.3, -0.8)	0.0482
REVEAL 2.0 risk score				
< 6	25	24	-49.0 (-162.3, 64.4)	0.3972
≥ 6	17	20	-168.4 (-301.7, -35.0)	0.0134

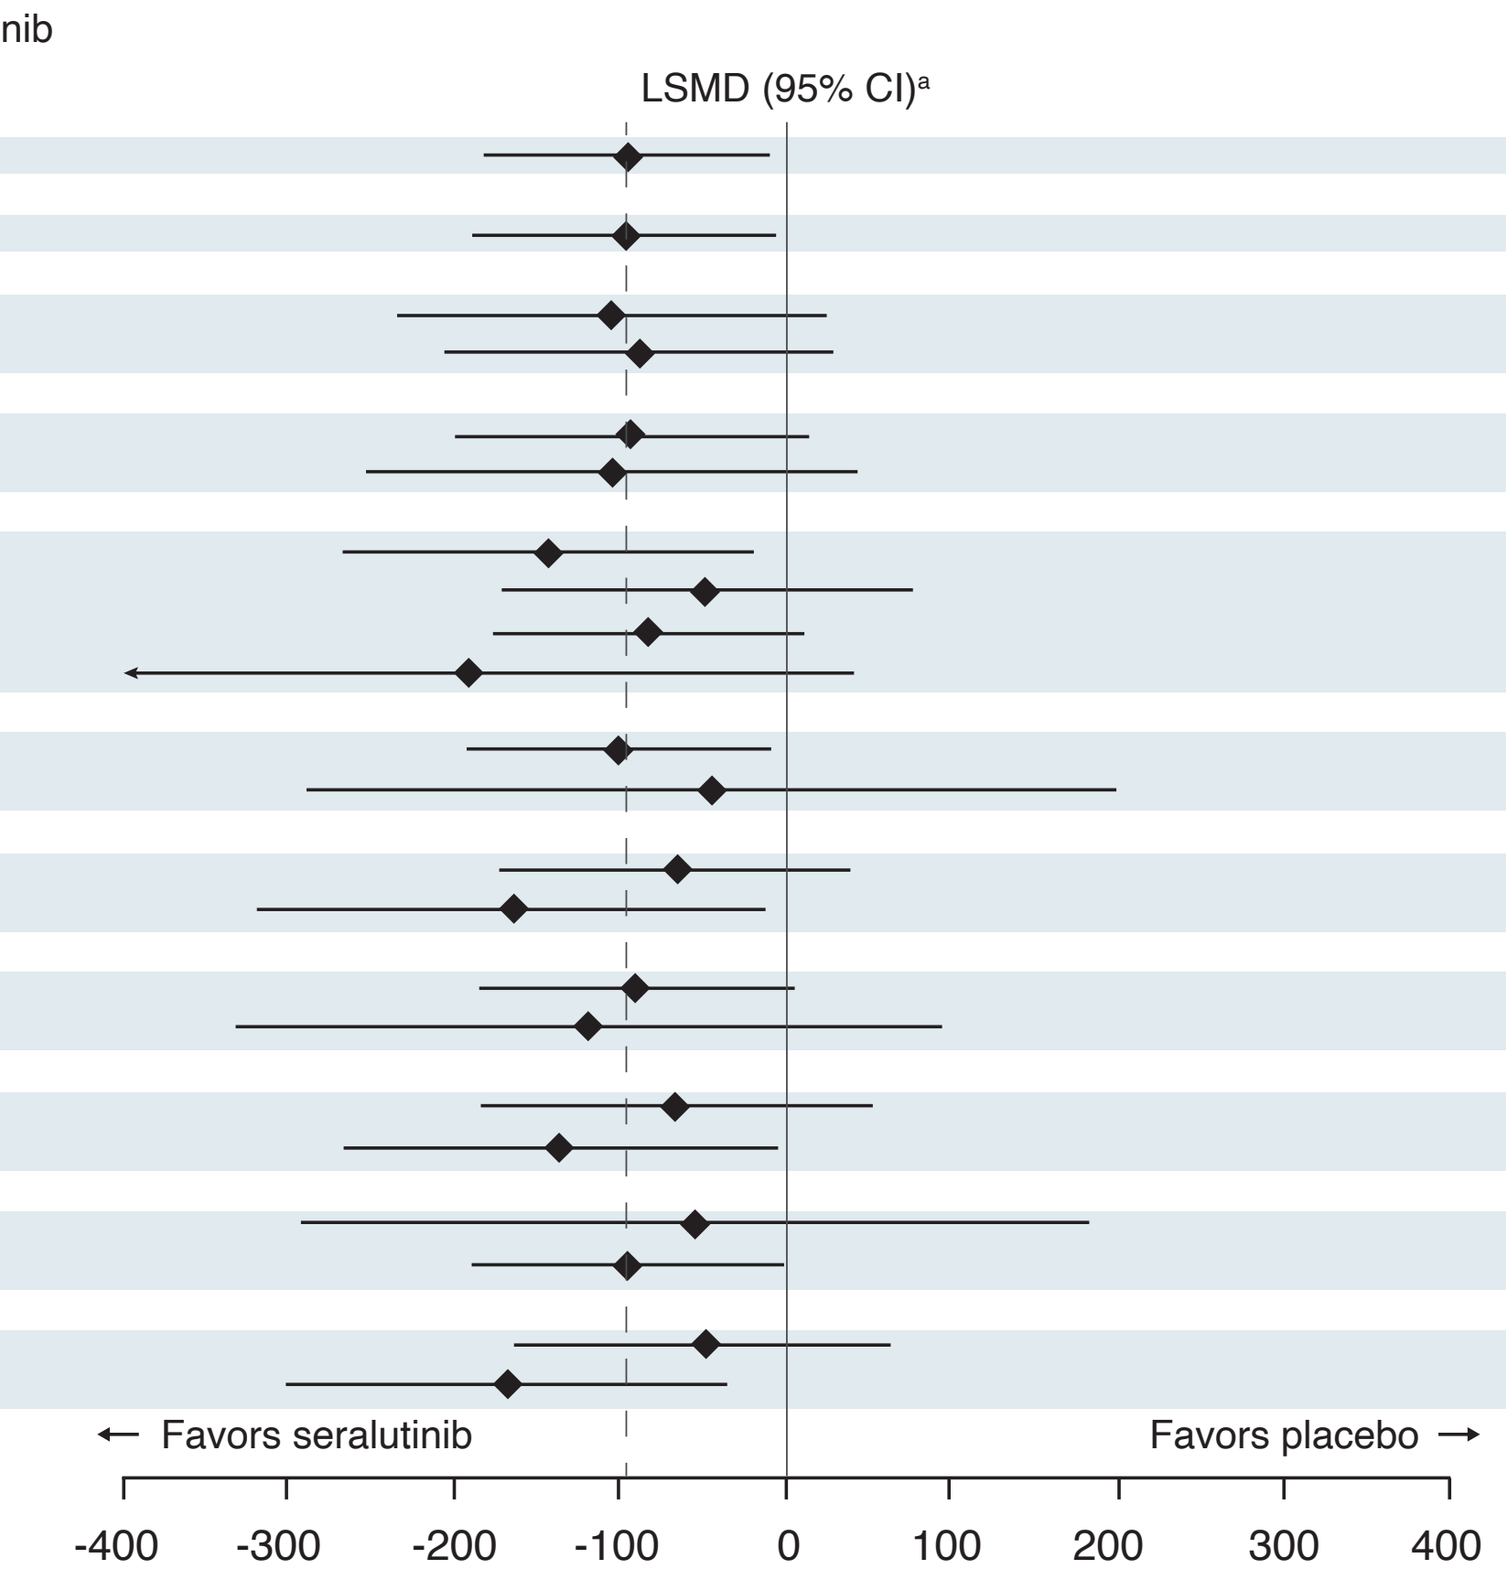

LSMD (95% CI)[a]
← Favors seralutinib
Favors placebo →
-400 -300 -200 -100 0 100 200 300 400

[a] Based on an ANCOVA model with multiple imputation.

Table 2. Change from baseline to Week 24 in pulmonary hemodynamic parameters. A significant reduction in mPAP (p=0.0094) was the main driver of the observed reduction in PVR. Treatment with seralutinib was associated with a significant improvement in PAC (p=0.0410)

Parameter	Placebo (N=42)	Seralutinib (N=38)	
	LS mean change ± SE	LS mean change ± SE	LS mean difference (95% CI)
mRAP, mmHg	0.85 ± 0.532	-0.14 ± 0.576	-0.99 (-2.350, 0.367)
PASP, mmHg	1.74 ± 2.321	-5.24 ± 2.469	-6.98 (-12.77, -1.19)*
PADP, mmHg	1.95 ± 1.127	-1.47 ± 1.197	-3.43 (-6.21, -0.64)*
mPAP, mmHg	2.12 ± 1.415	-2.58 ± 1.508	-4.70 (-8.203, -1.188)*
Cardiac output, L/min	-0.15 ± 0.165	0.06 ± 0.173	0.20 (-0.204, 0.605)
Cardiac index, L/min/m²	-0.02 ± 0.092	0.11 ± 0.097	0.13 (-0.100, 0.359)
PCWP or LVEDP, mmHg	1.04 ± 0.574	0.54 ± 0.608	-0.50 (-1.963, 0.963)
PVR, dyne*s/cm[5]	21.2 ± 29.91	-74.9 ± 33.02	-96.1 (-183.5, -8.8)*
PA compliance, mL/mmHg[a]	-0.02 ± 0.085	0.19 ± 0.089	0.22 (0.009, 0.423)*
Stroke volume, mL	-4.57 ± 2.206	-0.78 ± 2.313	3.79 (-1.606, 9.190)
Stroke volume index, mL/m²	-1.81 ± 1.263	0.38 ± 1.313	2.19 (-0.917, 5.299)

* p<0.05. [a] PA compliance = stroke volume/PA pulse pressure; PA pulse pressure = PASP-PADP. Observed cases, except for PVR, which used multiple imputation.
LS, least squares; LVEDP, left ventricular end-diastolic pressure; mPAP, mean pulmonary arterial pressure; mRAP, mean right atrial pressure; PA, pulmonary artery; PAC, pulmonary artery compliance; PASP/PADP, pulmonary artery systolic/diastolic pressure; PCWP, pulmonary capillary wedge pressure; PVR, pulmonary vascular resistance; SE, standard error.

Figure 5. A. Seralutinib treatment resulted in significant reduction in NT-proBNP vs placebo at Week 12 (-309.6 ng/L, p=0.0116) and at Week 24 (-408.3 ng/L, p=0.0012). **B.** Directional improvement in PAC-PVR relationship in seralutinib-treated patients vs placebo from BL to Week 24

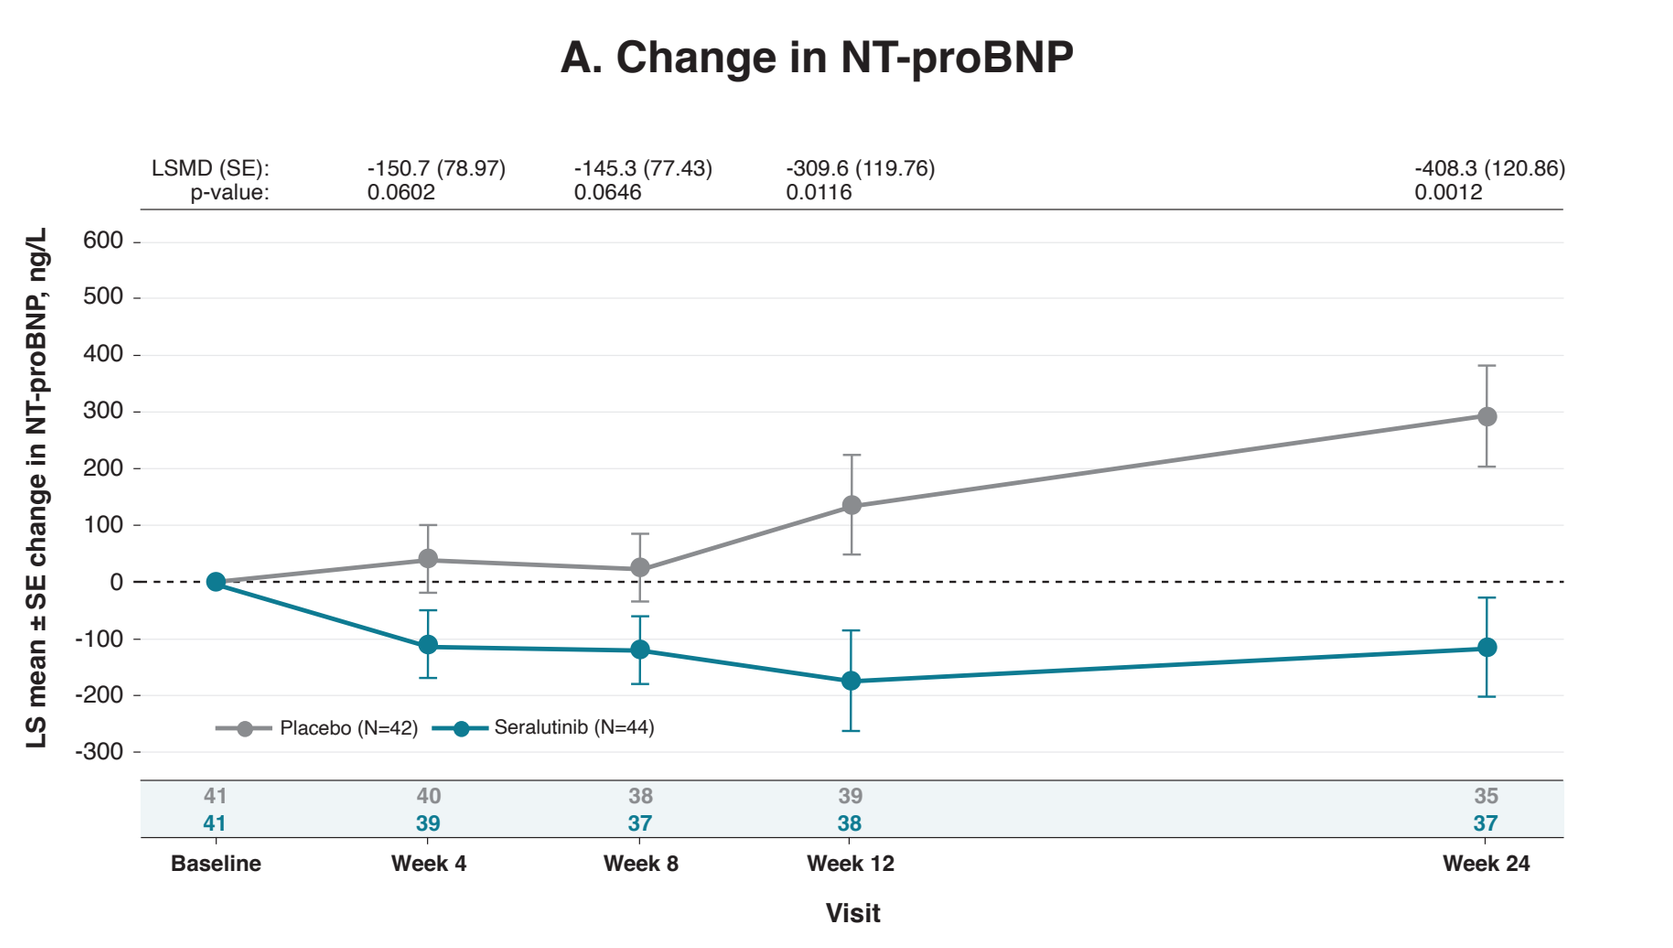

A. Change in NT-proBNP
LSMD (SE): -150.7 (78.97) -145.3 (77.43) -309.6 (119.76) -408.3 (120.86)
p-value: 0.0602 0.0646 0.0116 0.0012
LS mean ± SE change in NT-proBNP, ng/L
Placebo (N=42) Seralutinib (N=44)
41 40 38 39 35
41 39 37 38 37
Baseline Week 4 Week 8 Week 12 Week 24
Visit

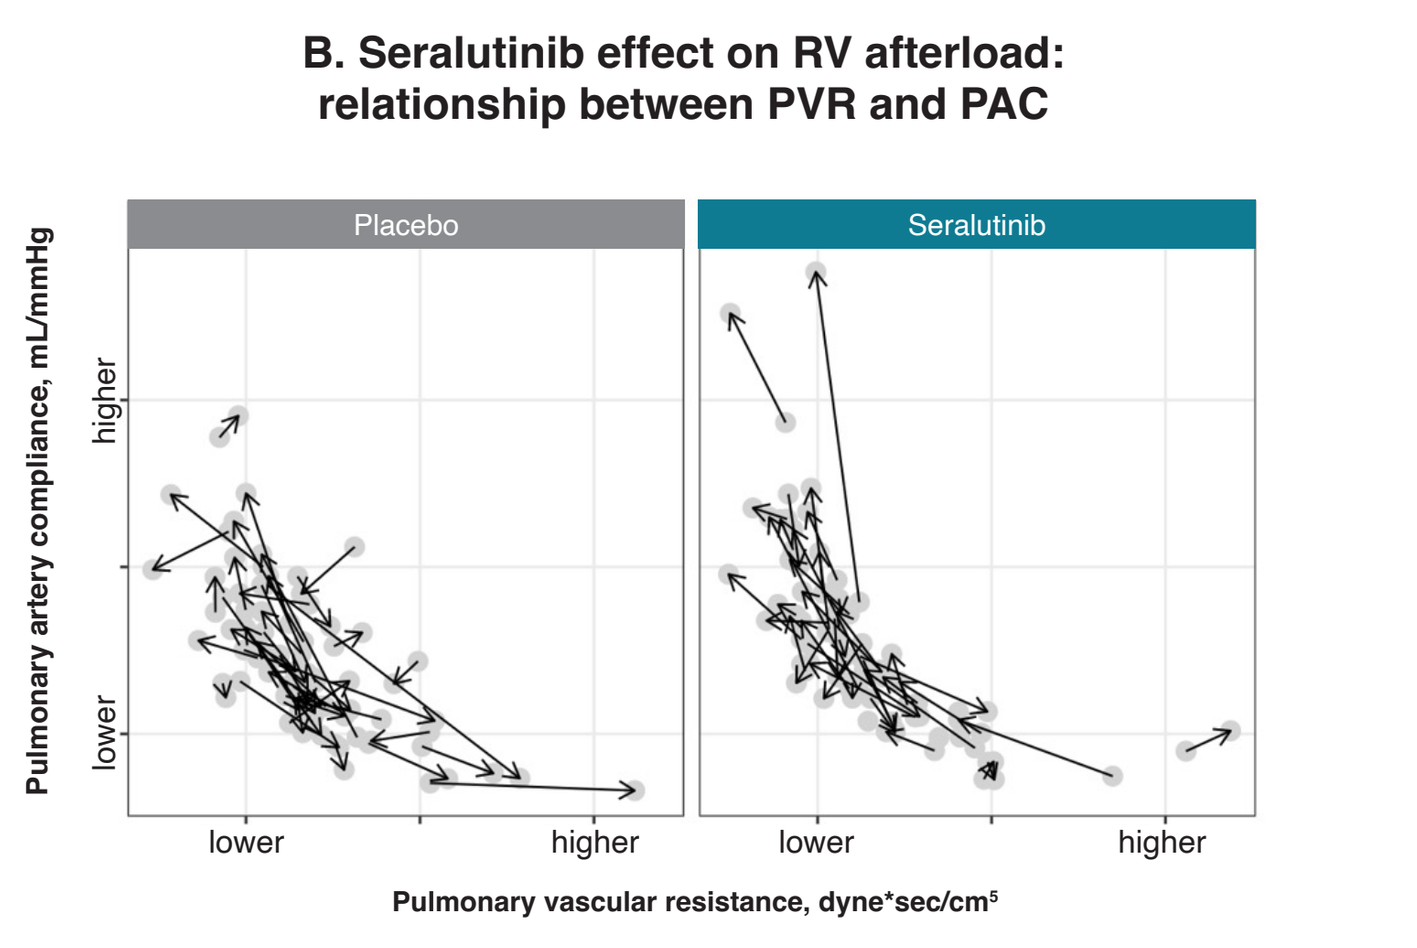

B. Seralutinib effect on RV afterload: relationship between PVR and PAC
Placebo
Seralutinib
Pulmonary artery compliance, mL/mmHg
higher
lower
Pulmonary vascular resistance, dyne*sec/cm5

Scale is the same for both placebo and seralutinib graphs.

Figure 6. A. At Week 24, the least-square mean difference in 6MWD between seralutinib and placebo groups was 6.5 m (p=NS). **B.** Significant improvement in 6MWD in FC III patients (+37.3 m, p=0.0476). **C.** Numerical trend favoring seralutinib in patients with a REVEAL 2.0 risk score of ≥ 6 (+21.9m, p=NS)

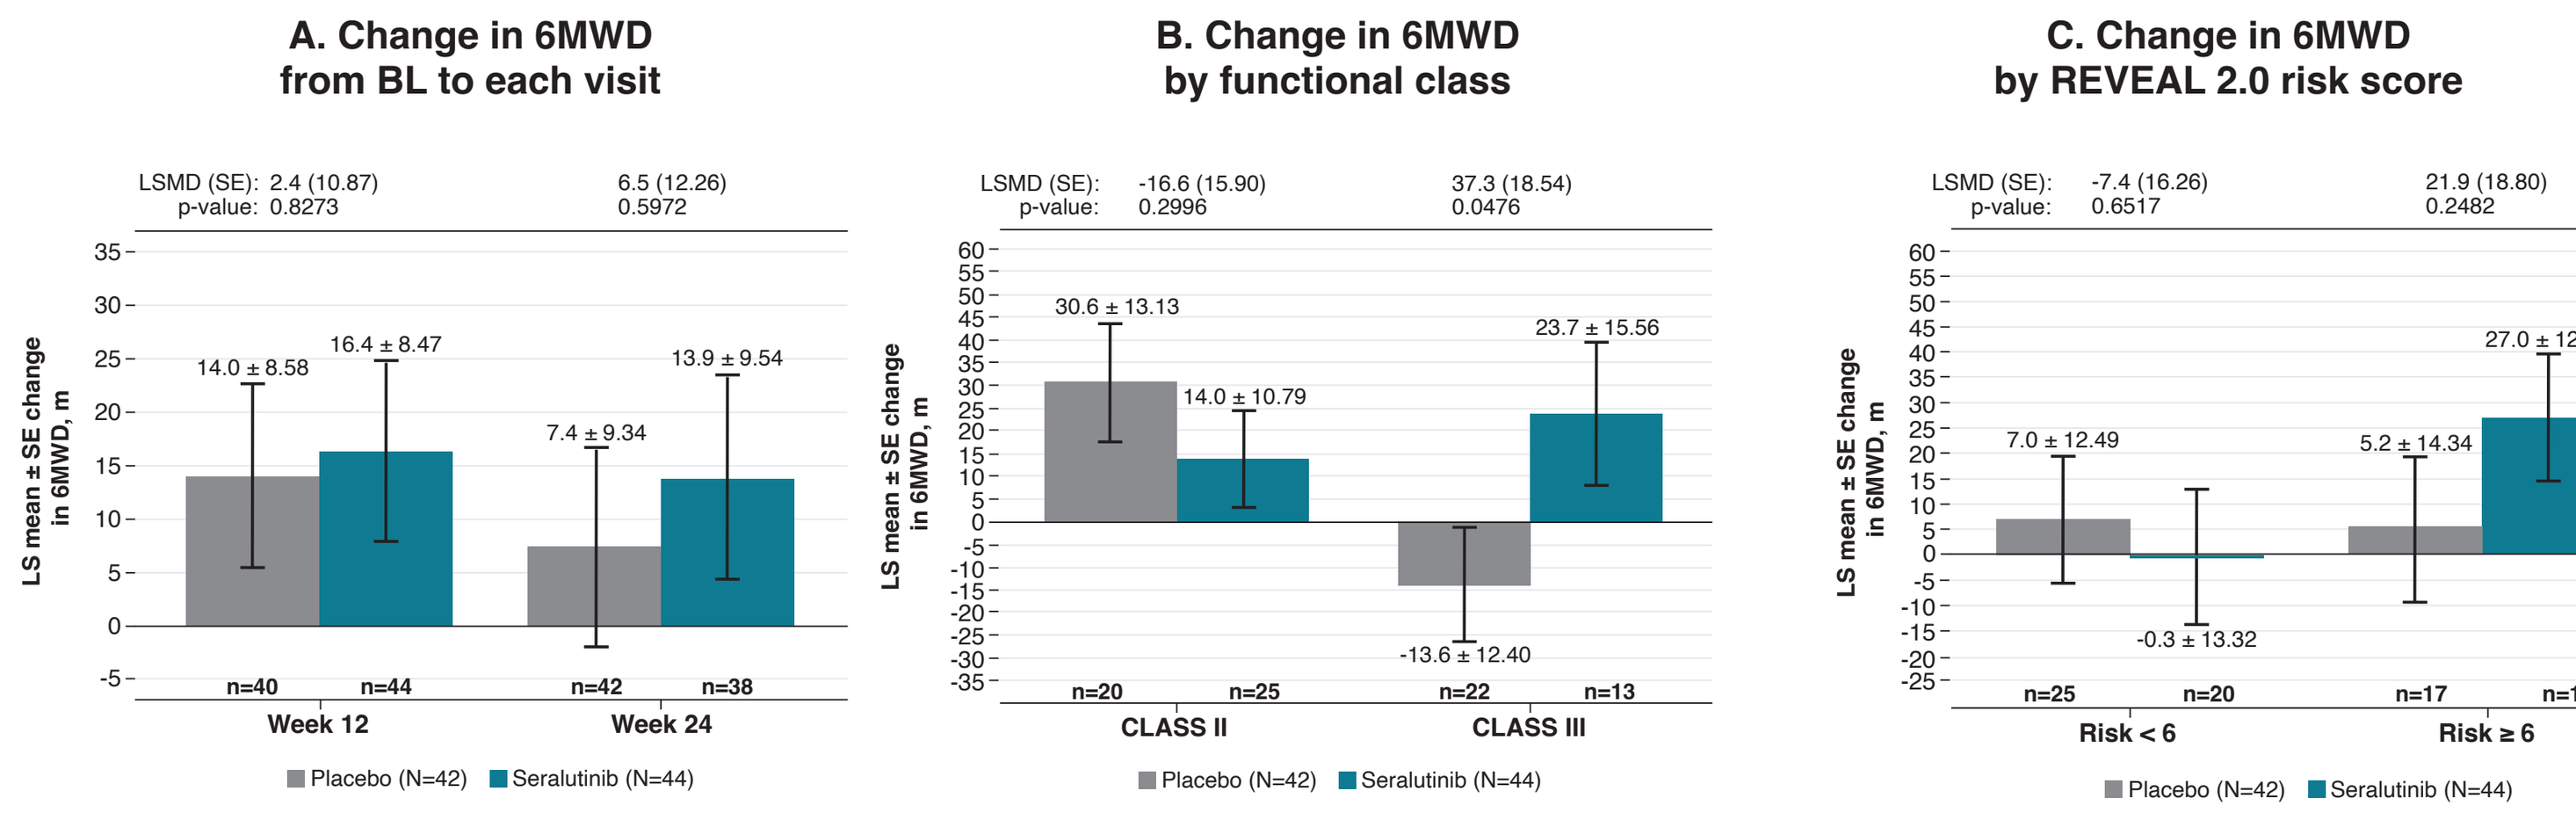

A. Change in 6MWD from BL to each visit
LSMD (SE): 2.4 (10.87) p-value: 0.8273
6.5 (12.26) 0.5972
14.0 ± 8.58
16.4 ± 8.47
7.4 ± 9.34
13.9 ± 9.54
n=40 n=44 n=42 n=38
Week 12 Week 24
LS mean ± SE change in 6MWD, m
B. Change in 6MWD by functional class
LSMD (SE): -16.6 (15.90) p-value: 0.2996
37.3 (18.54) 0.0476
30.6 ± 13.13
14.0 ± 10.79
-13.6 ± 12.40
23.7 ± 15.56
n=20 n=25 n=22 n=13
CLASS II CLASS III
C. Change in 6MWD by REVEAL 2.0 risk score
LSMD (SE): -7.4 (16.26) p-value: 0.6517
21.9 (18.80) 0.2482
7.0 ± 12.49
-0.3 ± 13.32
5.2 ± 14.34
27.0 ± 12.81
n=25 n=20 n=17 n=18
Risk < 6 Risk ≥ 6
Placebo (N=42) Seralutinib (N=44)

- Types of adverse events (AEs) observed were consistent with an inhalation therapy, i.e., mild-to-moderate cough
- Most treatment-emergent AEs reported were mild-to-moderate in severity
- No fatal AE was reported
- No adverse effect on pulmonary function or hematologic parameters
- Liver enzyme elevations > 3x upper limit of normal (3 seralutinib patients, 2 placebo patients)

Table 3. Overall summary of adverse events

	Placebo (N=42)	Seralutinib (N=44)
Frequency of adverse events		
Number of patients with at least one (%):		
TEAE	36 (85.7)	41 (93.2)
Severe TEAE	2 (4.8)	6 (13.6)
Related TEAE	22 (52.4)	28 (63.6)
TEAE leading to discontinuation of study drug	1 (2.4)	6 (13.6)
TEAE leading to withdrawal from study	0	4 (9.1)
SAE	6 (14.3)	10 (22.7)
Incidence of TEAEs by preferred term[a]: ≥ 5% higher in seralutinib group, n (%)		
Cough	16 (38.1)	19 (43.2)
Diarrhea	3 (7.1)	6 (13.6)
Dizziness	2 (4.8)	5 (11.4)
Nightmare	1 (2.4)	4 (9.1)
Abdominal pain lower	0	3 (6.8)
Nasopharyngitis	0	3 (6.8)
Throat irritation	0	3 (6.8)

Safety Population.
[a] Coded using MedDRA v 24.0.
MedDRA, Medical Dictionary for Regulatory Activities; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

CONCLUSIONS

- Seralutinib, an inhaled PDGFR, CSF1R, and Kit tyrosine kinase inhibitor, demonstrated clinical activity and safety in the Phase 2 TORREY trial
- TORREY met the primary endpoint of reduction in PVR in a heavily treated, prevalent study population on standard of care background medications
- Prespecified subgroup analyses showed greater benefit in FC III and patients with REVEAL 2.0 risk score ≥ 6
- The reduction in PVR and increase in PAC in conjunction with a reduction of NT-proBNP indicates that seralutinib is reducing RV afterload and having a beneficial effect on the right heart
- Proof of concept has been demonstrated and a global registrational Phase 3 program in PAH is planned

References: 1 Galkin A et al. *Eur Respir J.* 2022;60(6):2102356. **2** Frantz RP et al. *Pulm Circ.* 2021;11(4):20458940211057071.

Acknowledgements: We thank all patients, their families, and all the TORREY study investigators and study coordinators who participated in TORREY.

Research supported by: Gossamer Bio, Inc.

Authors' relevant interests: RPF: Gossamer Bio, Inc., Janssen, Liquidia, Merck, ShouTi, Tenax Therapeutics, Insmed (Advisory Committee); Janssen, Liquidia (Consultant); Aerovate Therapeutics (DSMB). **HAG:** Gossamer Bio, Inc., Aerovate, Altavant, Bayer AG, Attgeno, Janssen/Actelion, MSD/Acceleron, Pfizer (Consultant, Advisory Committee); Bayer AG, Janssen/Actelion, MSD/Acceleron (Speaker); Insmed (DSMB).

Download your copy of the **poster**

Download the **plain language summary**