Fr150

GB004 Exhibits Protective Effects Directly on Epithelial Cells Using *Ex Vivo* Organoid and Monolayer Cultures

Kristen R. Taylor Meadows, Susan Murphy, Gregory J. Opiteck, Barrett G. Levesque, Laura Carter, Luisa Salter-Cid

Gossamer Bio, Inc.

INTRODUCTION

- Inflammatory bowel disease (IBD) is characterized by a breach in intestinal barrier integrity, allowing influx of luminal antigens and setting up a vicious cycle of inflammation and epithelial injury[1]
- Despite efficacy of IBD treatment with anti-tumor necrosis factor agents and anti-integrin agents, a large fraction of patients do not respond adequately to currently available therapies or biologics and do not achieve long-term remission[2,3]
- GB004 is an oral, gut-targeted, small molecule that stabilizes hypoxia inducible factor (HIF-1α), a key transcription factor involved in the adaptive and protective cellular responses at the intersection of hypoxia and inflammation[4] (**Figure 1**)
- Preclinical efficacy of GB004 has been demonstrated in mouse models of colitis and correlated with HIF-1α stabilization in colonic epithelial cells, induction of HIF-1α target genes, downmodulation of inflammatory cytokines, and improvement in histologic parameters of barrier function[5,6]
- A phase 2 study evaluating two doses of GB004 as a tablet formulation in mild to moderate UC is ongoing (NCT04556383)

Figure 1. GB004 mechanism of action

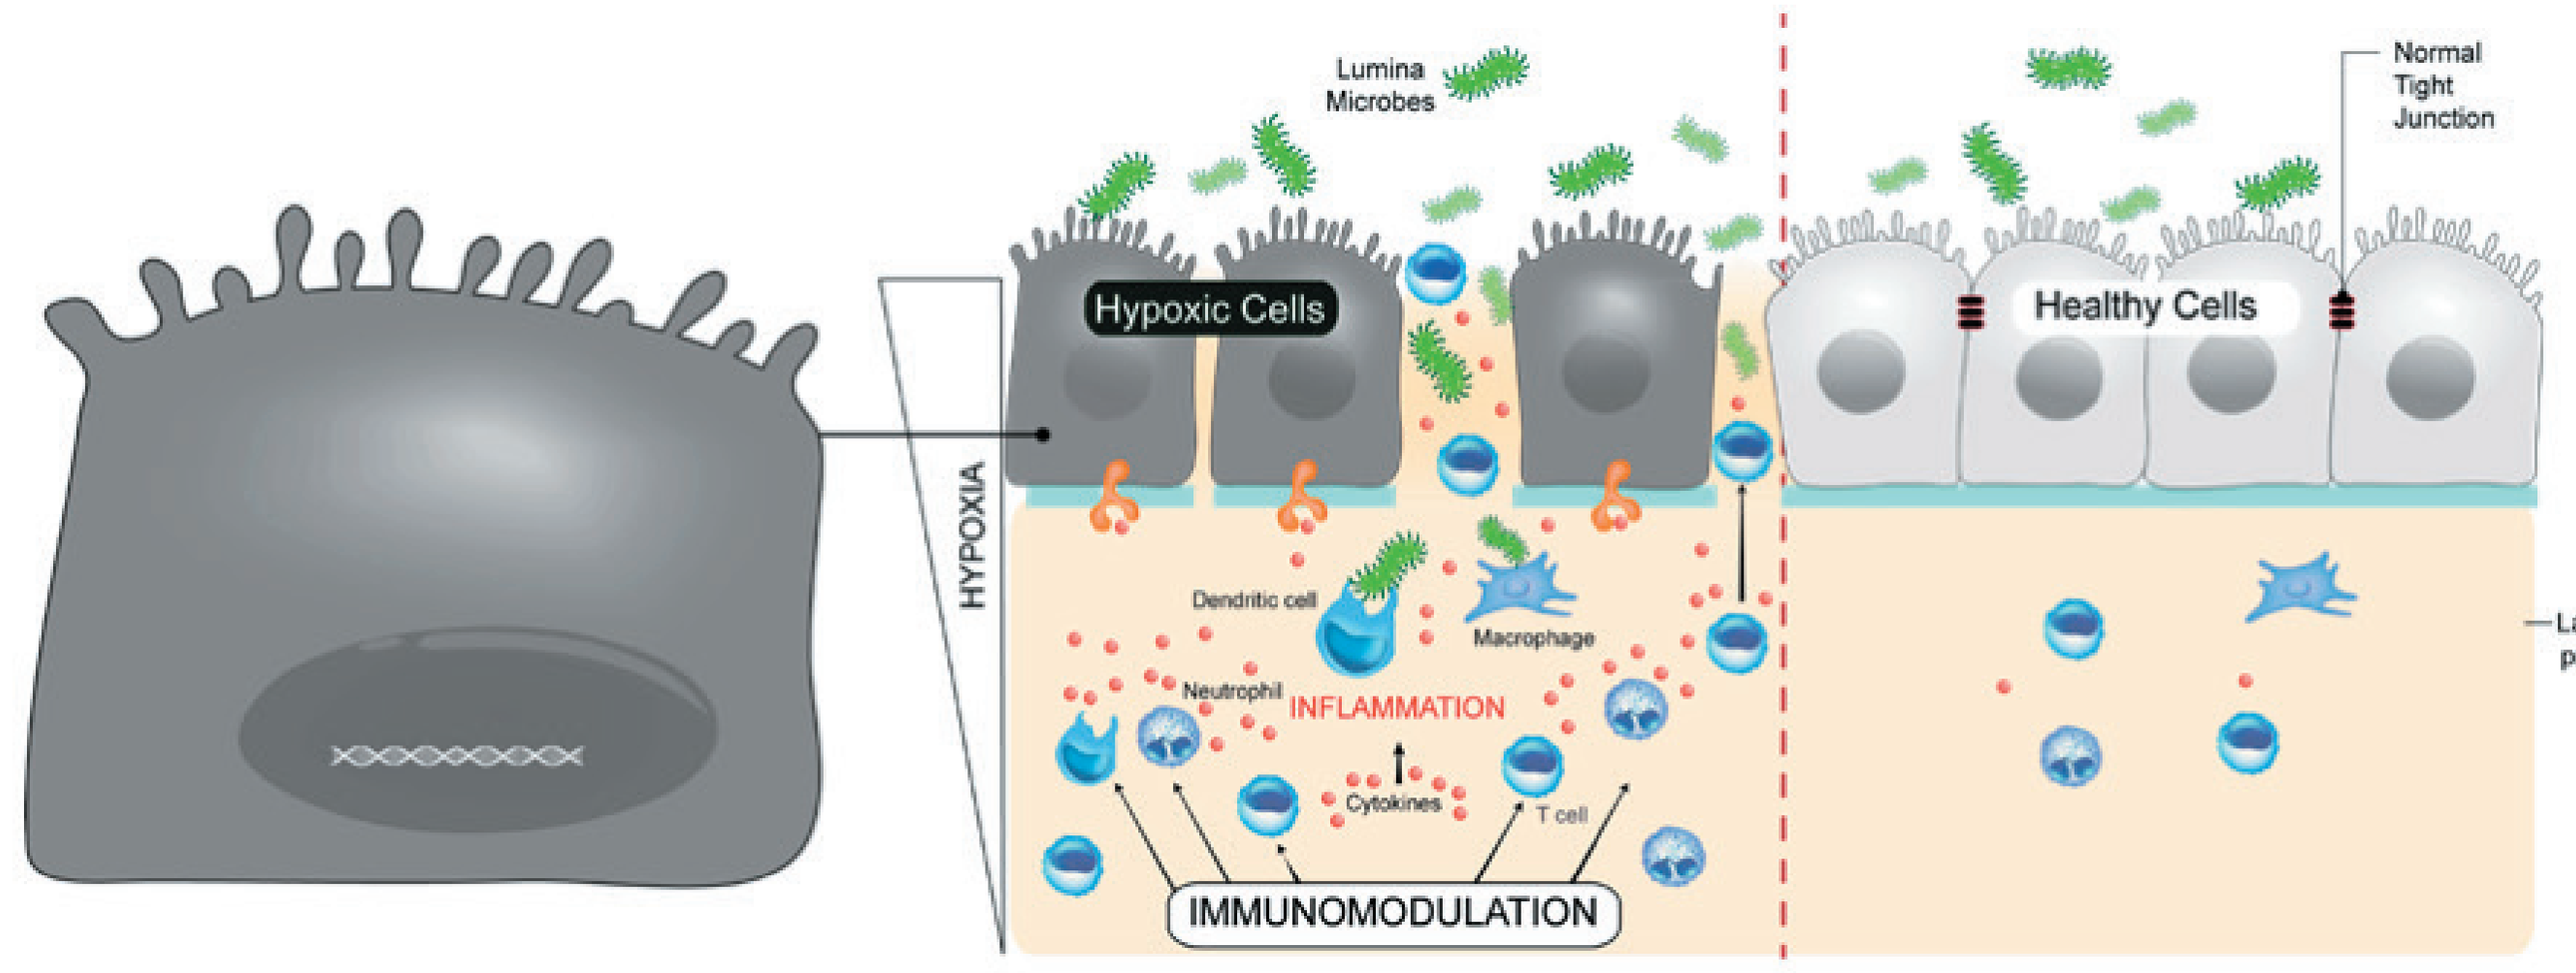

Lumina Microbes
Normal Tight Junction
Hypoxic Cells
Healthy Cells
HYPOXIA
Dendritic cell
Macrophage
Neutrophil
INFLAMMATION
Cytokines
T cell
IMMUNOMODULATION
Lamina propria

OBJECTIVE

- To assess the effects of HIF stabilizer GB004 on gene expression, tight junctions, and barrier integrity using intestinal epithelial cells and organoids

METHODS

- Mouse organoids were derived from the small intestine of C57BL/6 mice and cultured in IntestiCult Organoid Growth Media (StemCell Technologies, Cambridge, MA) according to manufacturers instructions. Organoids were treated for 3 days with GB004. RNA was isolated from the organoids, and gene expression analysis was performed. Gene expression data was generated from one well of organoid cultures; data is representative of two independent experiments.

METHODS

- Human Repligut differentiated monolayers assays were performed at Altis Biosystems (Chapel Hill, NC) by proliferating and differentiating human-derived intestinal epithelial cells on a 2D monolayer platform. These monolayers were assessed with GB004 treatment under normal healthy conditions or with cytokine-stimulated conditions (25 ng/mL TNFα) to induce barrier damage. Barrier integrity was assessed through measuring Transepithelial Endothelial Electric Resistance (TEER) and a barrier integrity assay using FITC-dextran. HIF-1α target genes were assessed in cell lysates and tight junction formation and adhesion molecules were investigated by immunofluorescence staining. Three independent studies were performed to generate data presented in Figure 3, Figure 4, and Figures 5-7, respectively.
- Unless otherwise noted, data are presented as mean ± SD. Statistical analysis was carried out using GraphPad Prism and one-way ANOVA (*p < 0.05, **p < 0.01, ***p < 0.001, ***p < 0.0001).

RESULTS

Figure 2. GB004 induces HIF1α-dependent gene expression in murine organoids

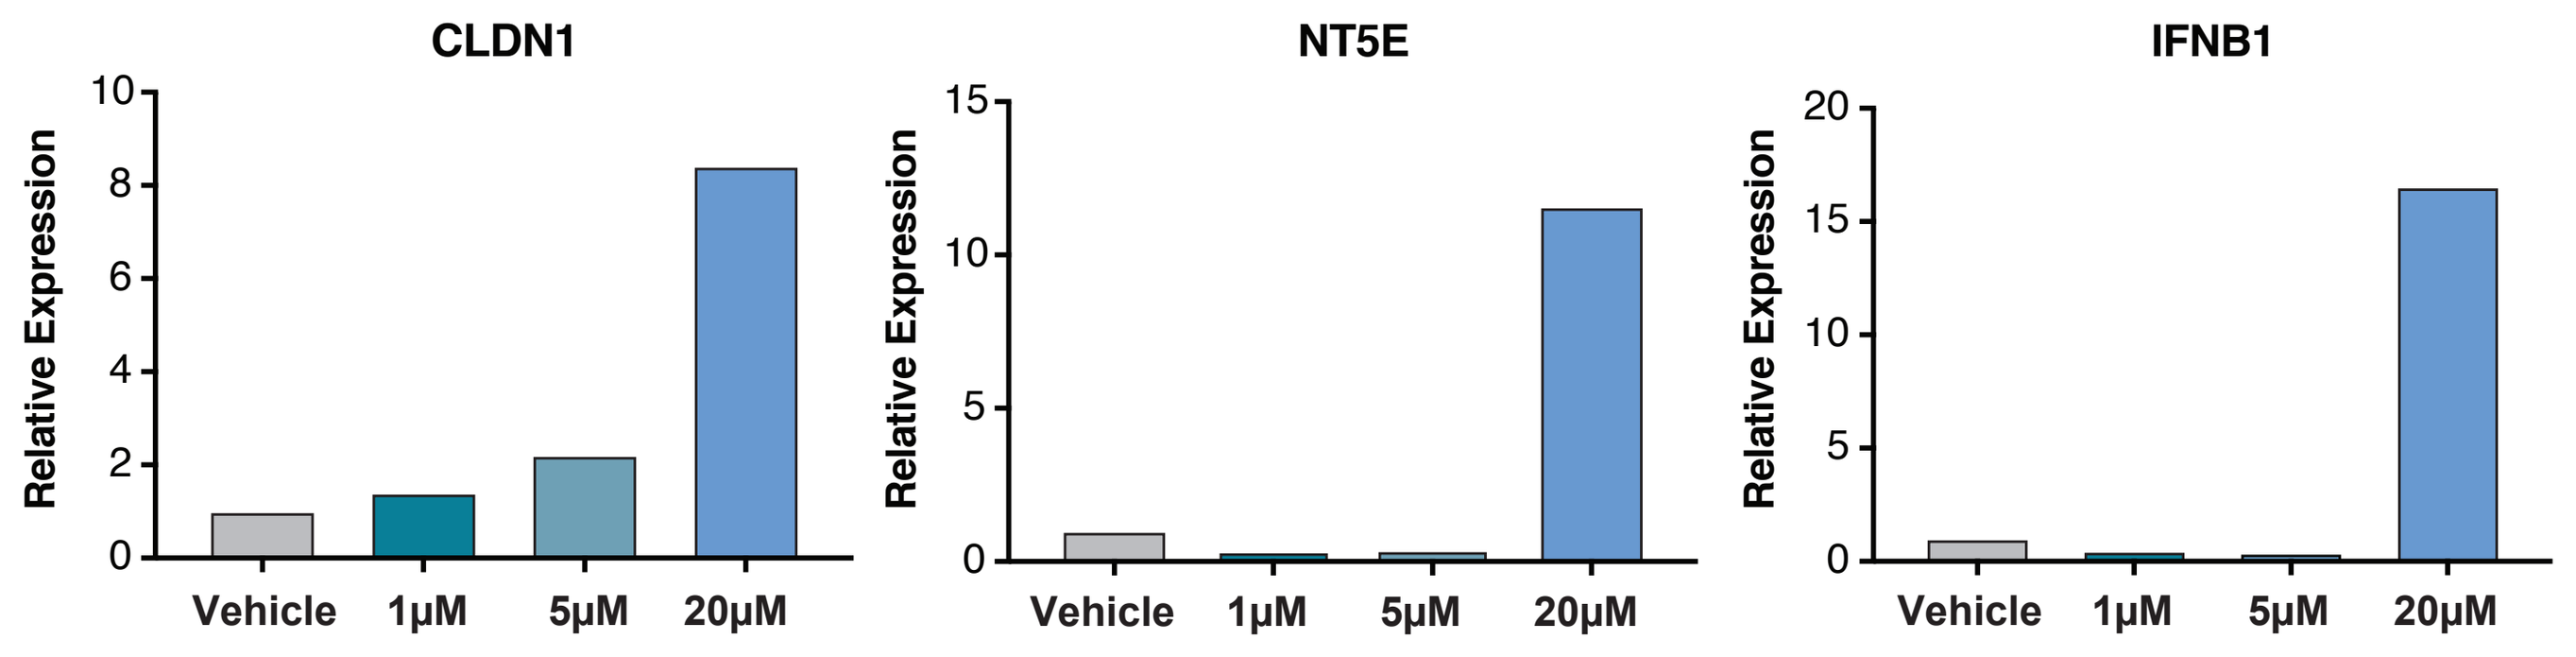

CLDN1
NT5E
IFNB1
Relative Expression
Vehicle
1μM
5μM
20μM

Figure 3. GB004 stabilizes HIF-1α in human differentiated monolayers at 2 hours (A) and 4 hours (B) after treatment depending on dose (dotted line indicates the maximum HIF-1α stabilization measurement with the positive control lysate). Normal healthy monolayers have complete barrier integrity (C) as determined by TEER.

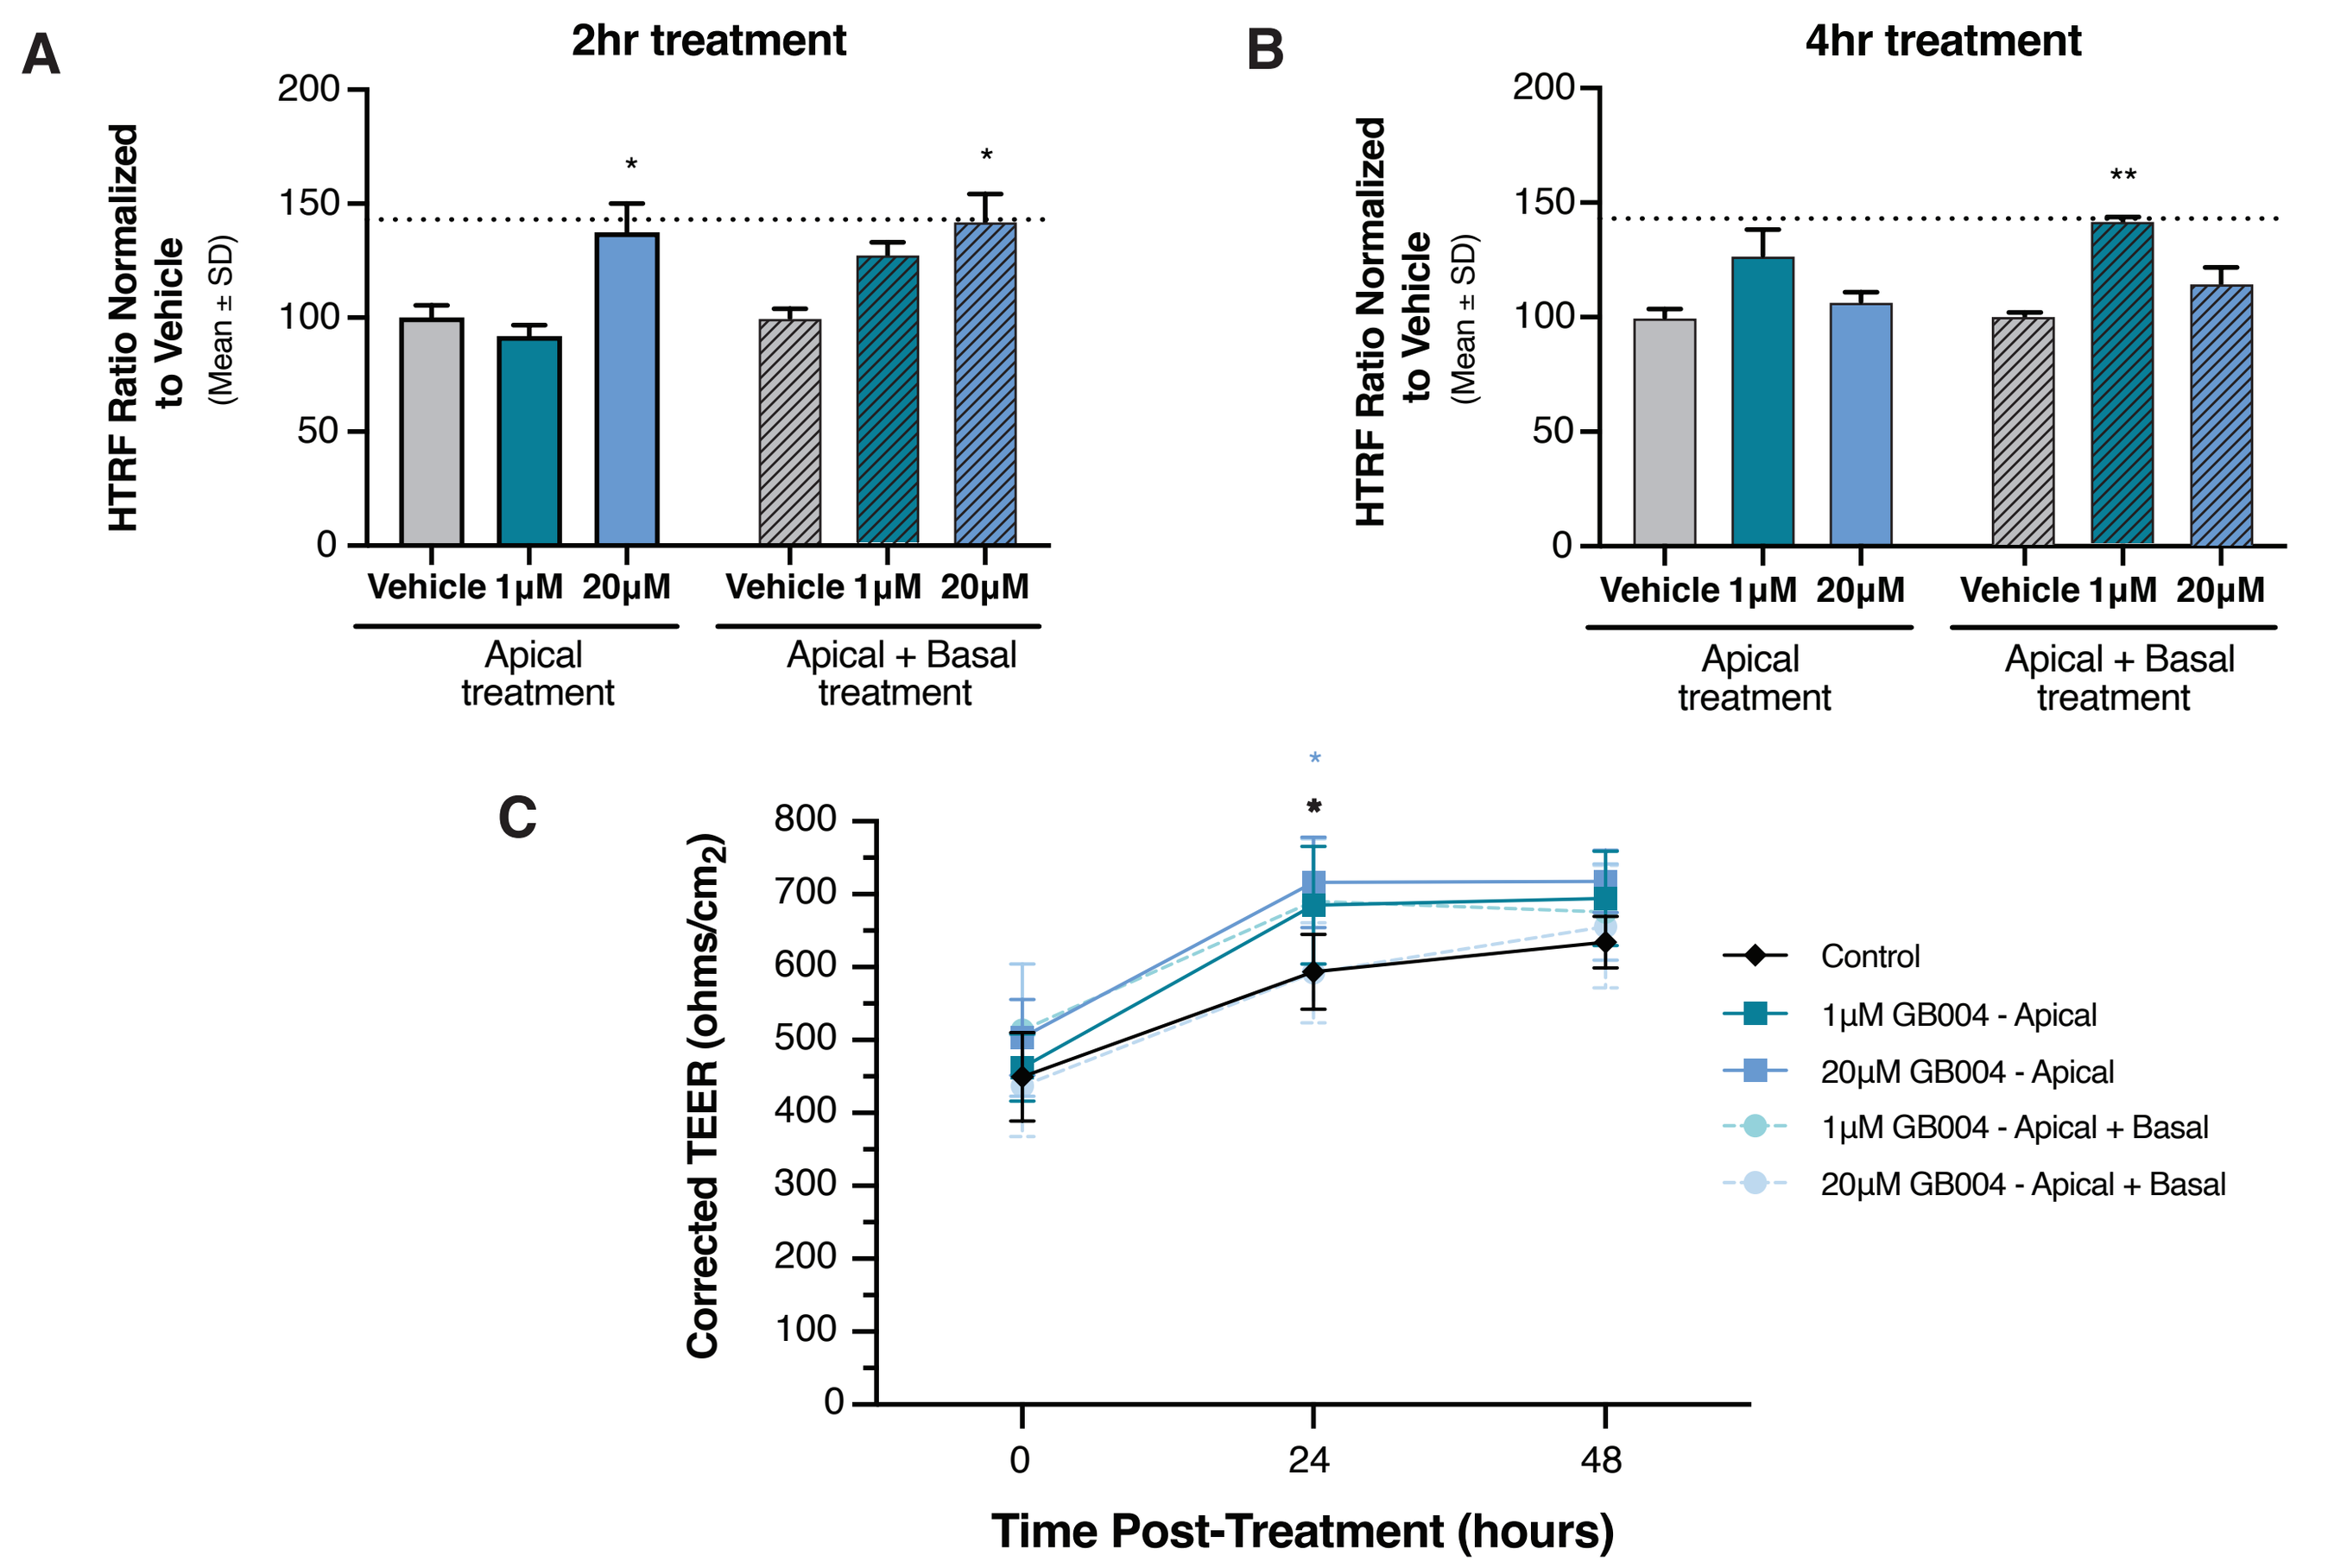

A
2hr treatment
B
4hr treatment
HTRF Ratio Normalized to Vehicle
(Mean ± SD)
Vehicle 1μM 20μM
Apical treatment
Apical + Basal treatment
C
Corrected TEER (ohms/cm2)
Time Post-Treatment (hours)
Control
1μM GB004 - Apical
20μM GB004 - Apical
1μM GB004 - Apical + Basal
20μM GB004 - Apical + Basal

RESULTS

Figure 4. TNFα or IFNγ stimulation of human differentiated monolayers disrupts barrier integrity resulting in reduced TEER measurements (A) and increased barrier permeability (B).

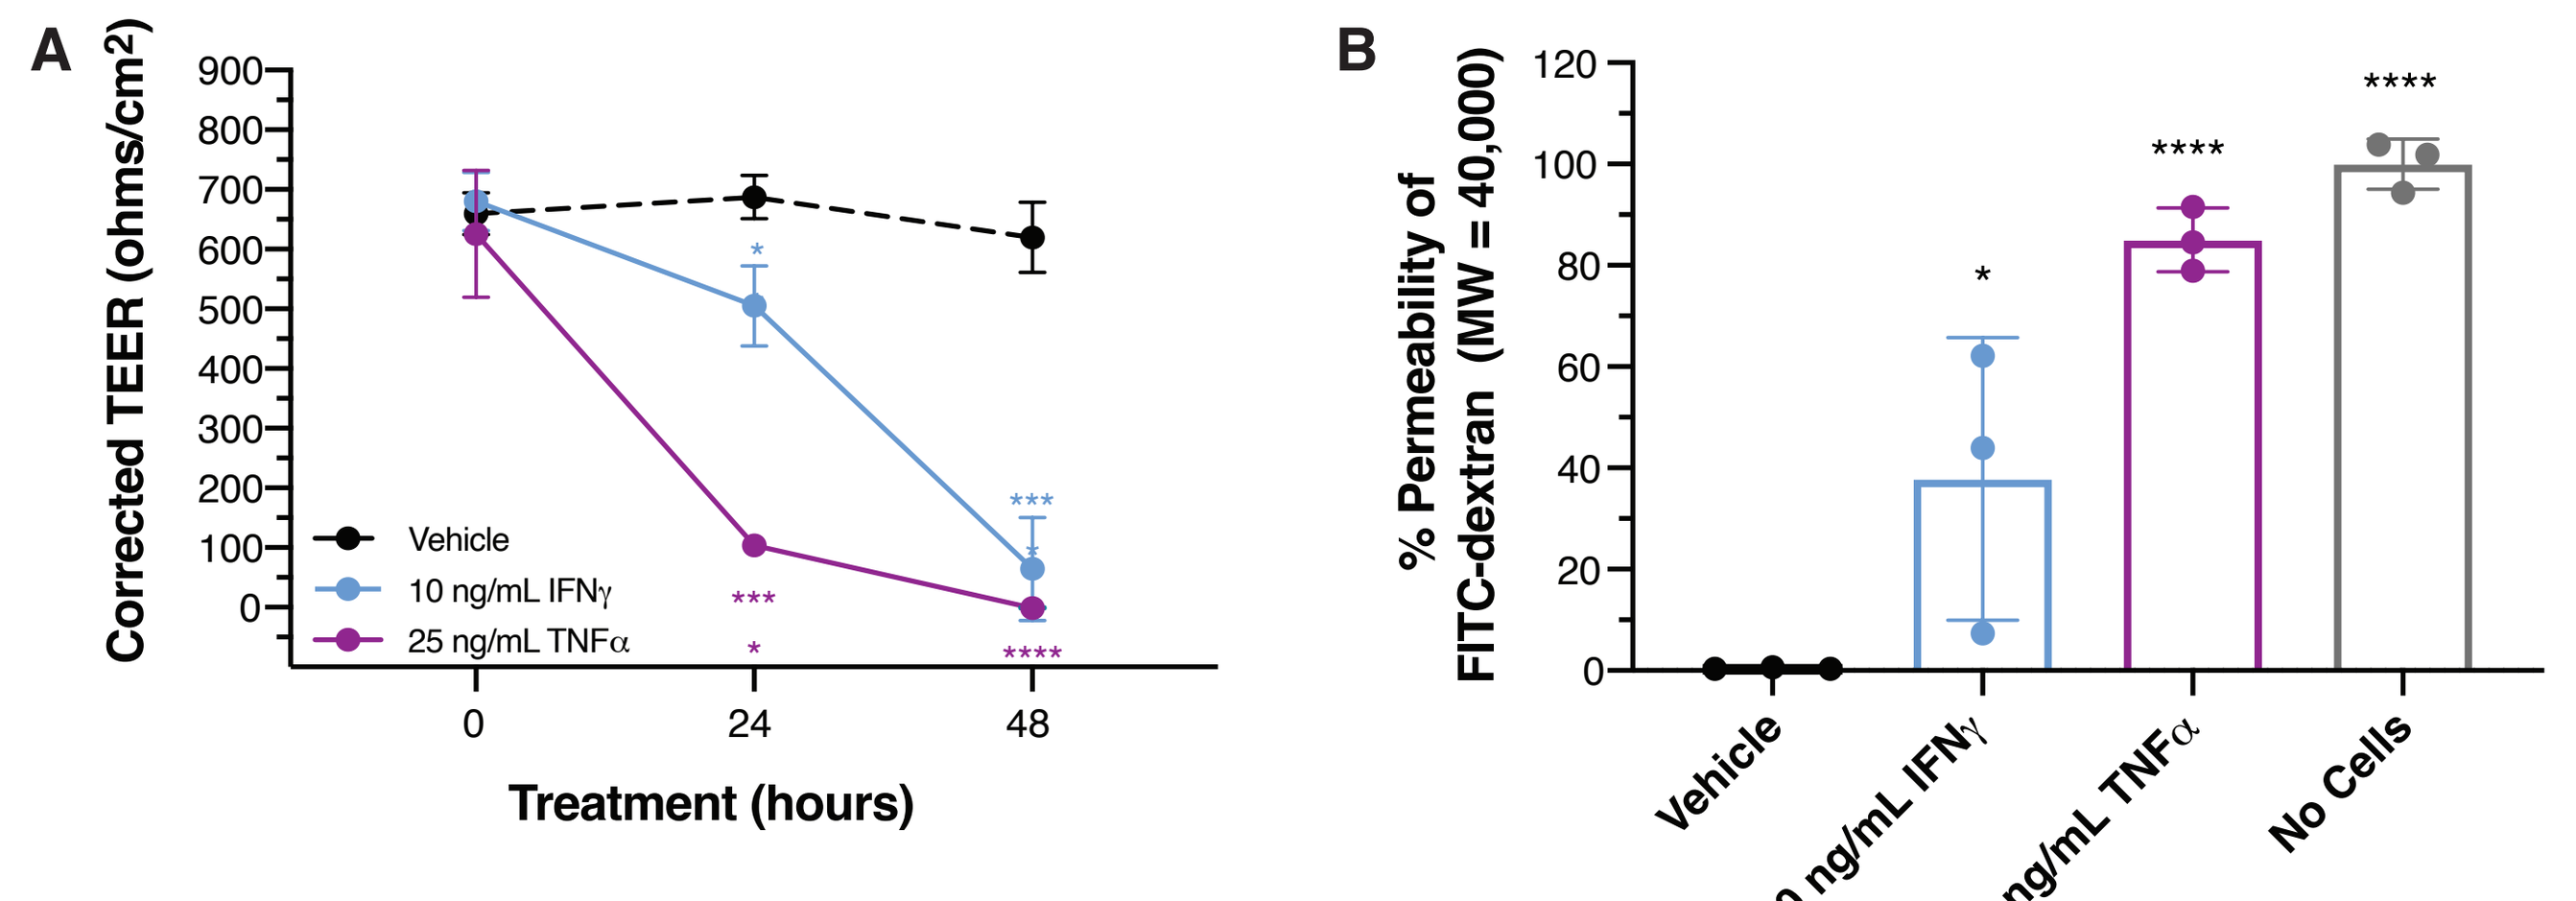

A
Corrected TEER (ohms/cm2)
Vehicle
10 ng/mL IFNγ
25 ng/mL TNFα
Treatment (hours)
B
% Permeability of FITC-dextran (MW = 40,000)
Vehicle
10 ng/mL IFNγ
25 ng/mL TNFα
No Cells

Figure 5. GB004 treatment of TNFα-stimulated of human differentiated monolayers prevents the loss in barrier integrity as measured by TEER, while tofacitinib (JAK inhibitor) had no effect on barrier integrity.

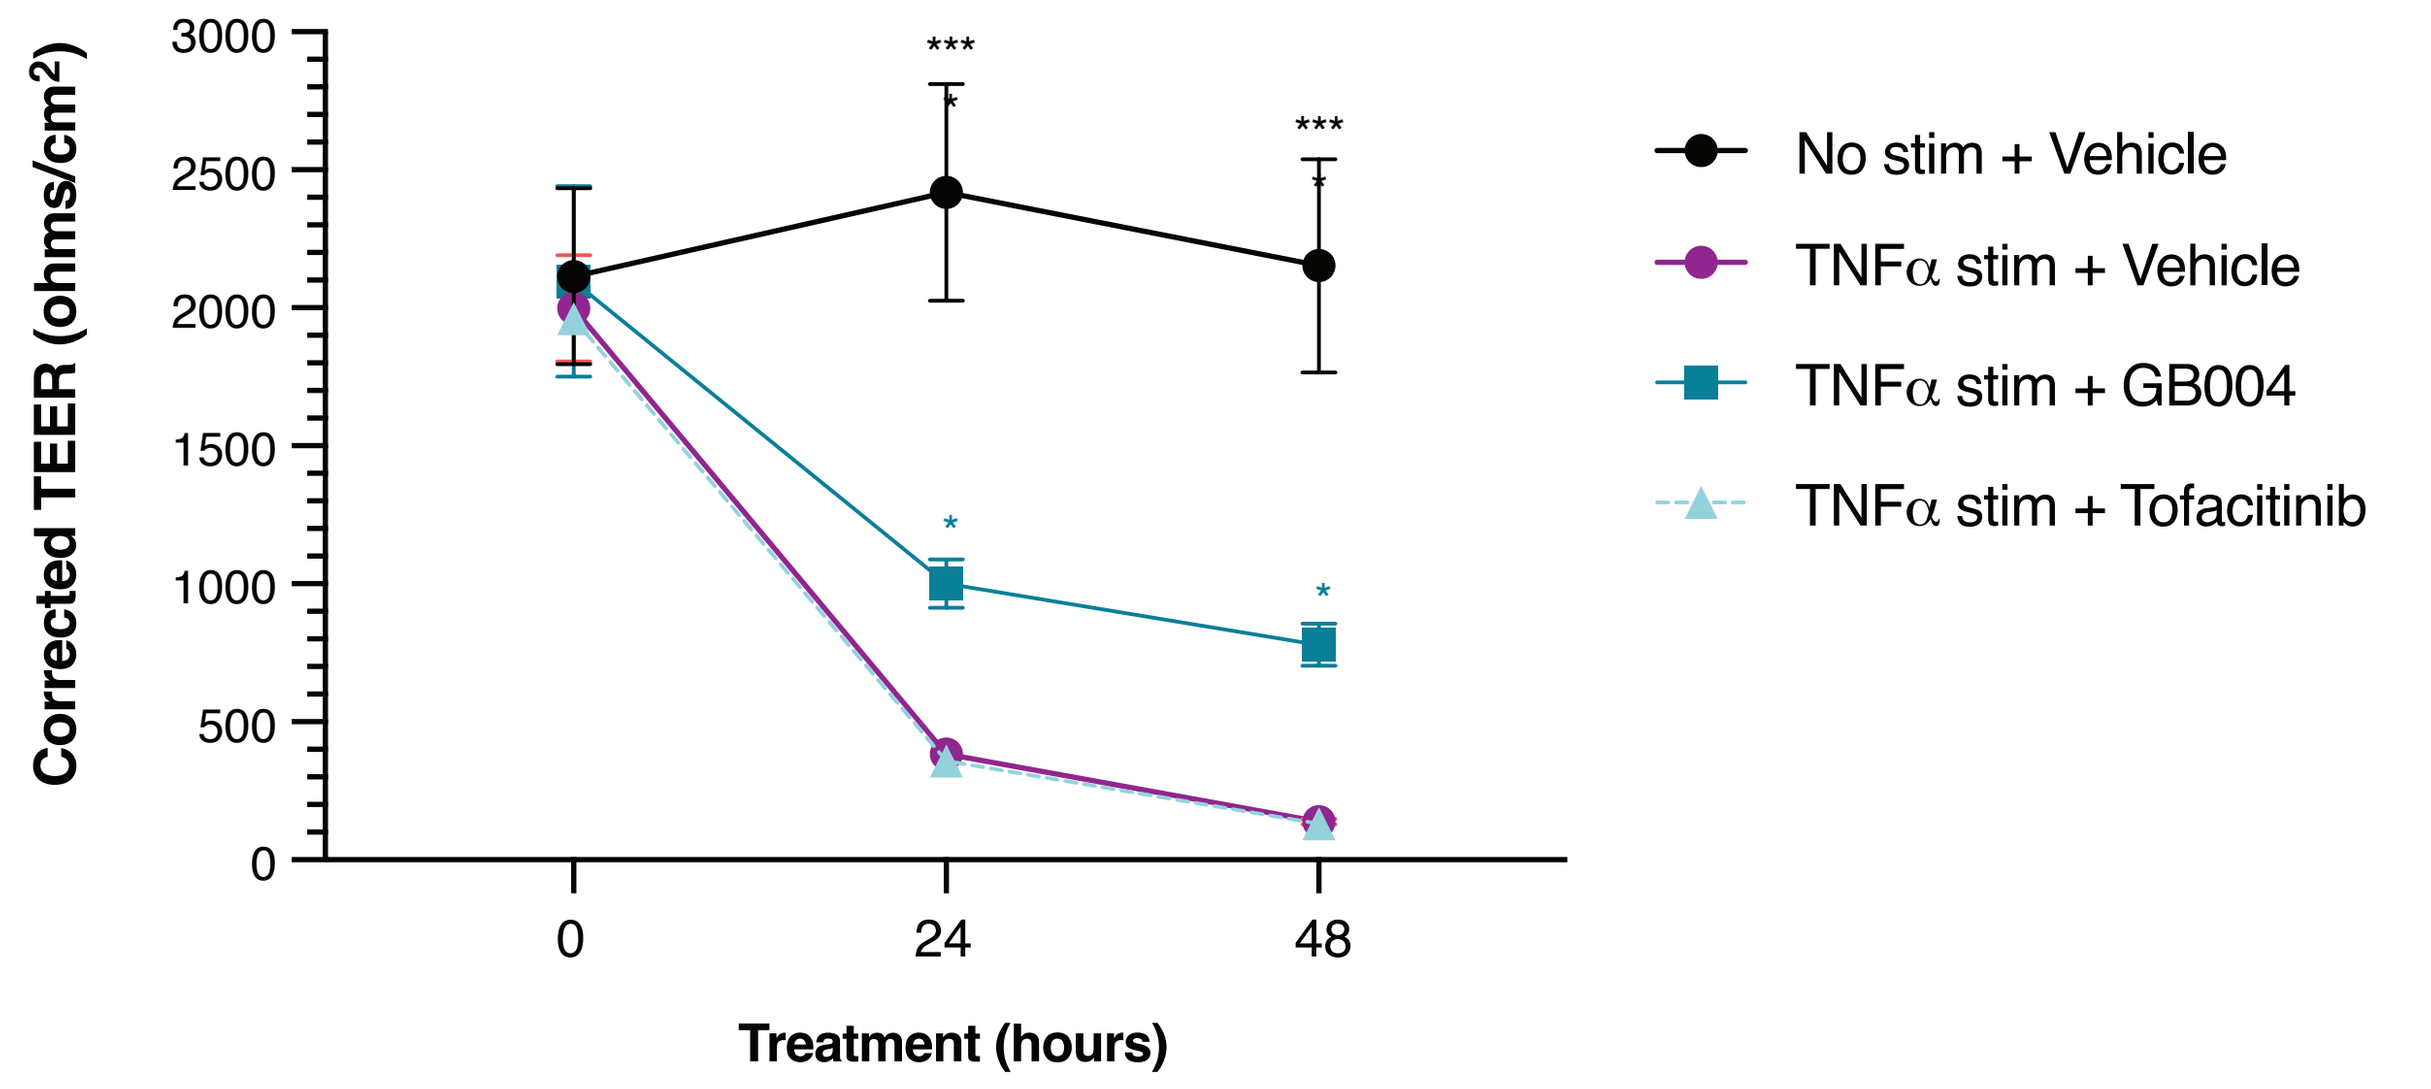

Corrected TEER (ohms/cm2)
Treatment (hours)
No stim + Vehicle
TNFα stim + Vehicle
TNFα stim + GB004
TNFα stim + Tofacitinib

Figure 6. TNFα treatment of human differentiated monolayer results in epithelial cell death at 48 hours. GB004 treatment prevents TNFα-induced cell death, whereas tofacitinib (Tofa) had no effect.

DAPI = blue staining, ZO-1 = green staining

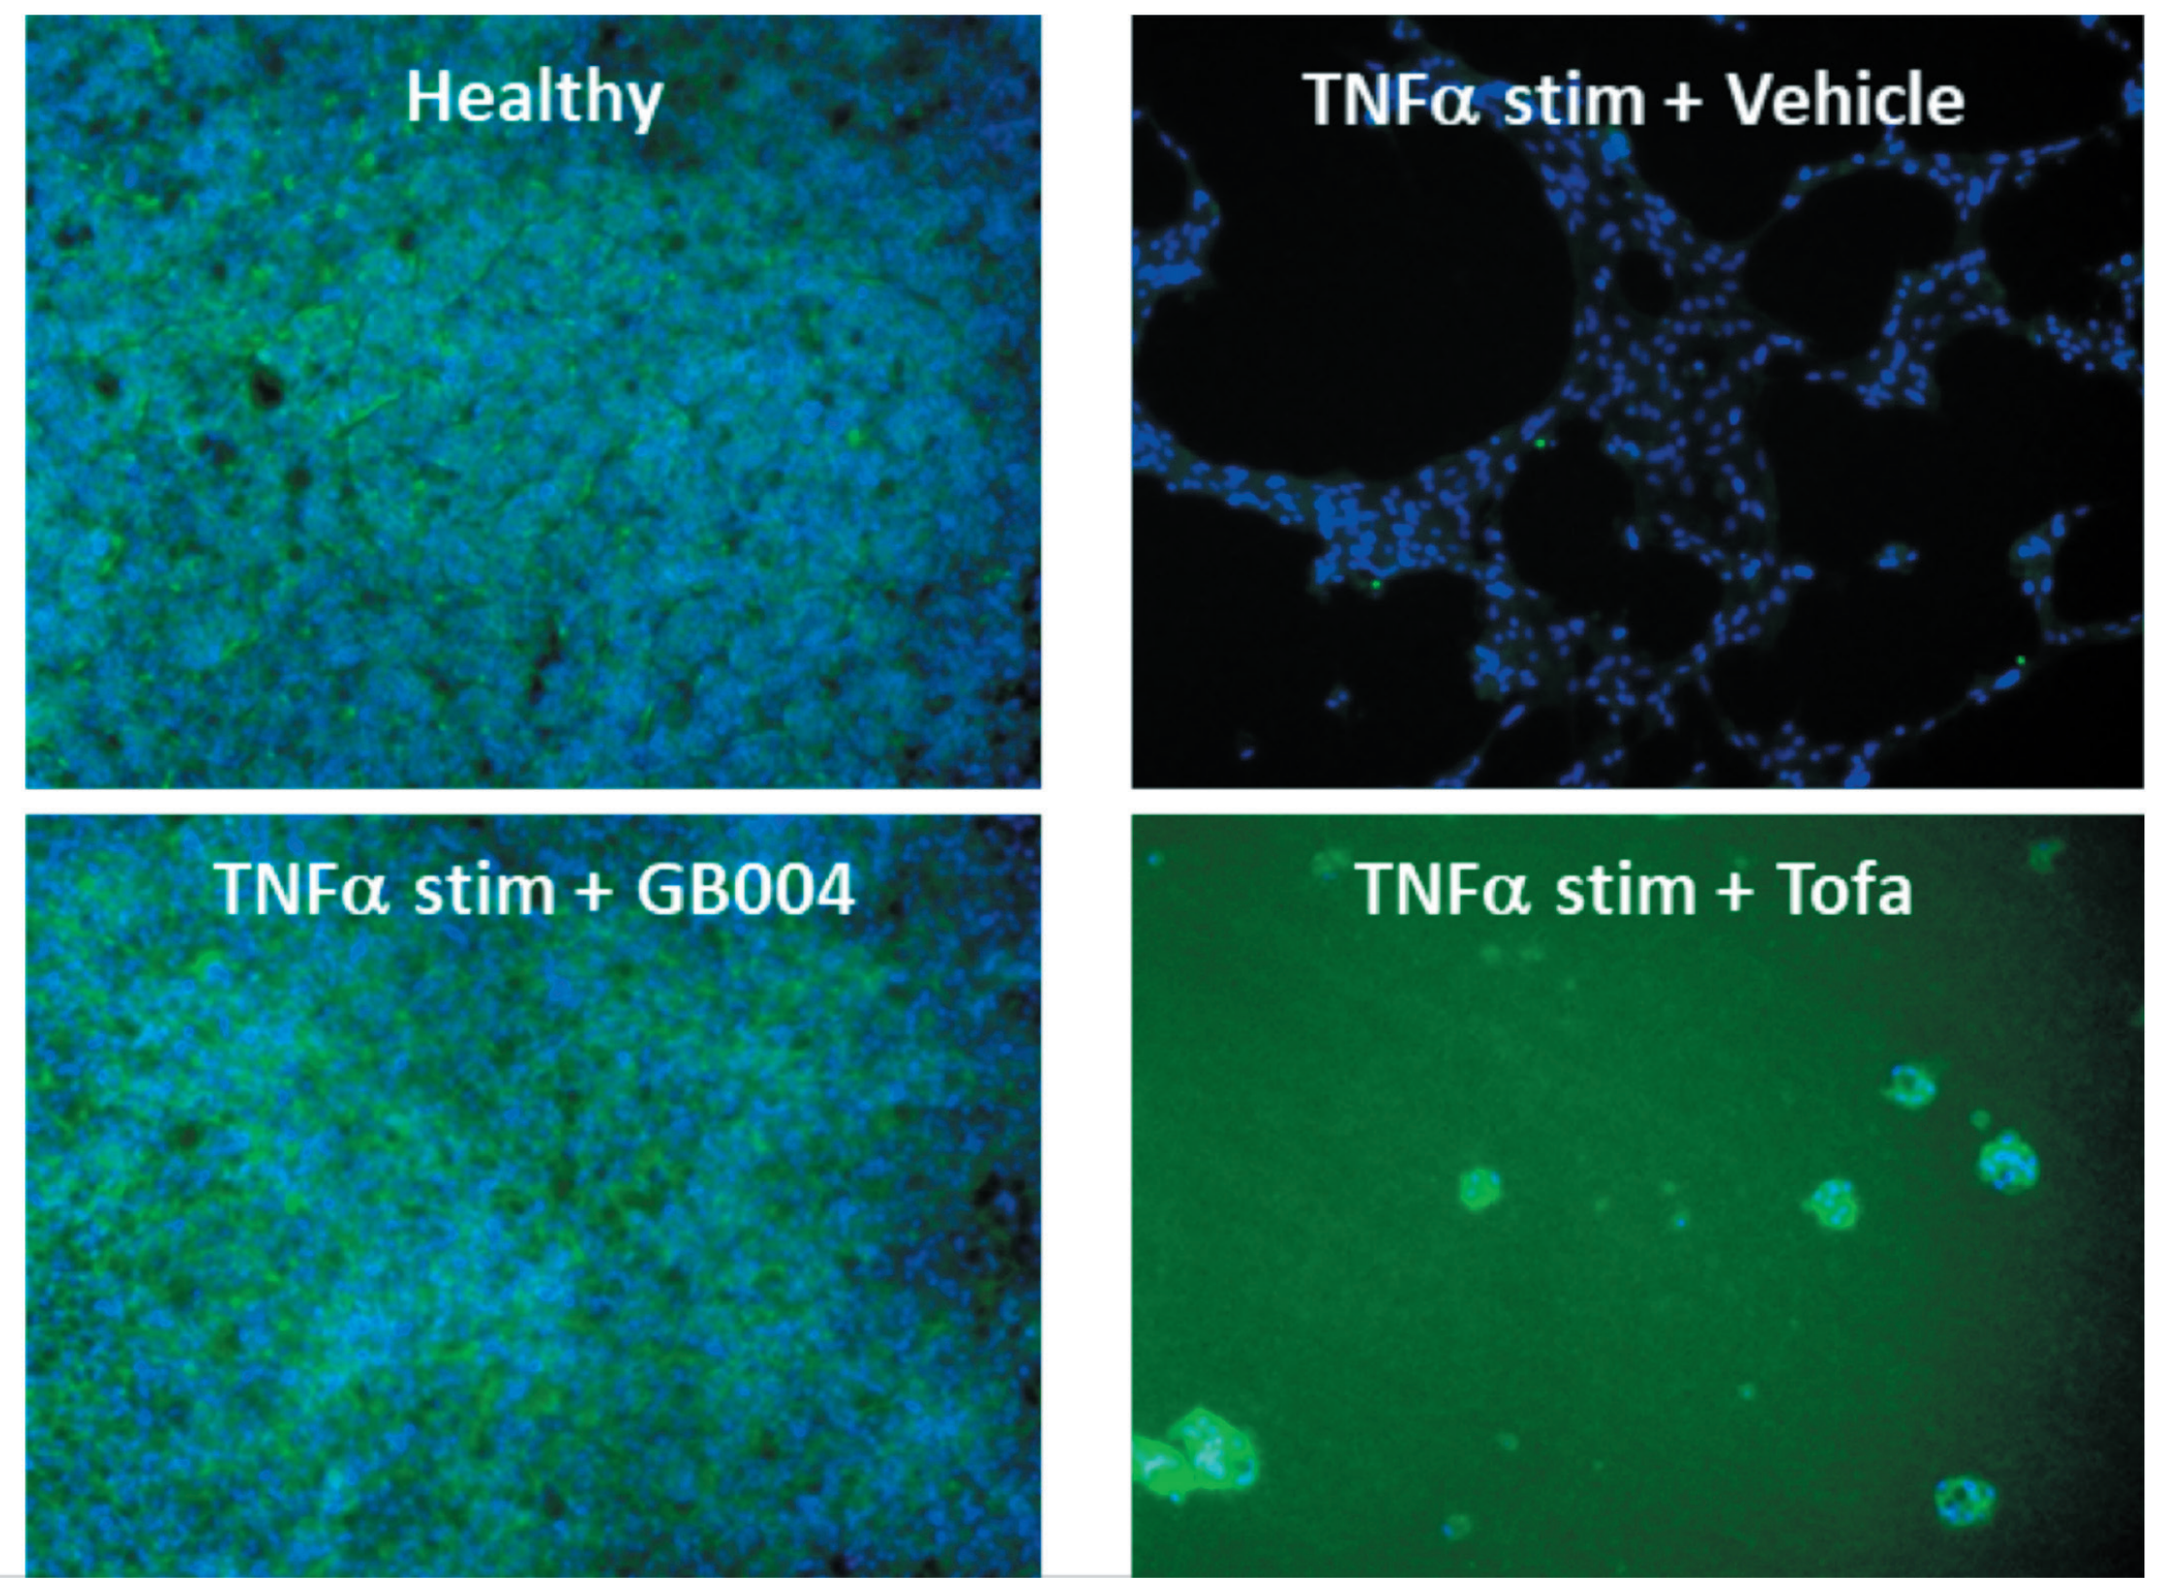

Healthy
TNFα stim + Vehicle
TNFα stim + GB004
TNFα stim + Tofa

Figure 7. GB004 modulates a total of 266 genes in human differentiated monolayers stimulated with TNFα that are related to the HIF1A, PHD and hypoxia pathways as determined by RNAseq. Genes are grouped into tightly correlated modules and key genes are highlighted with green boxes.

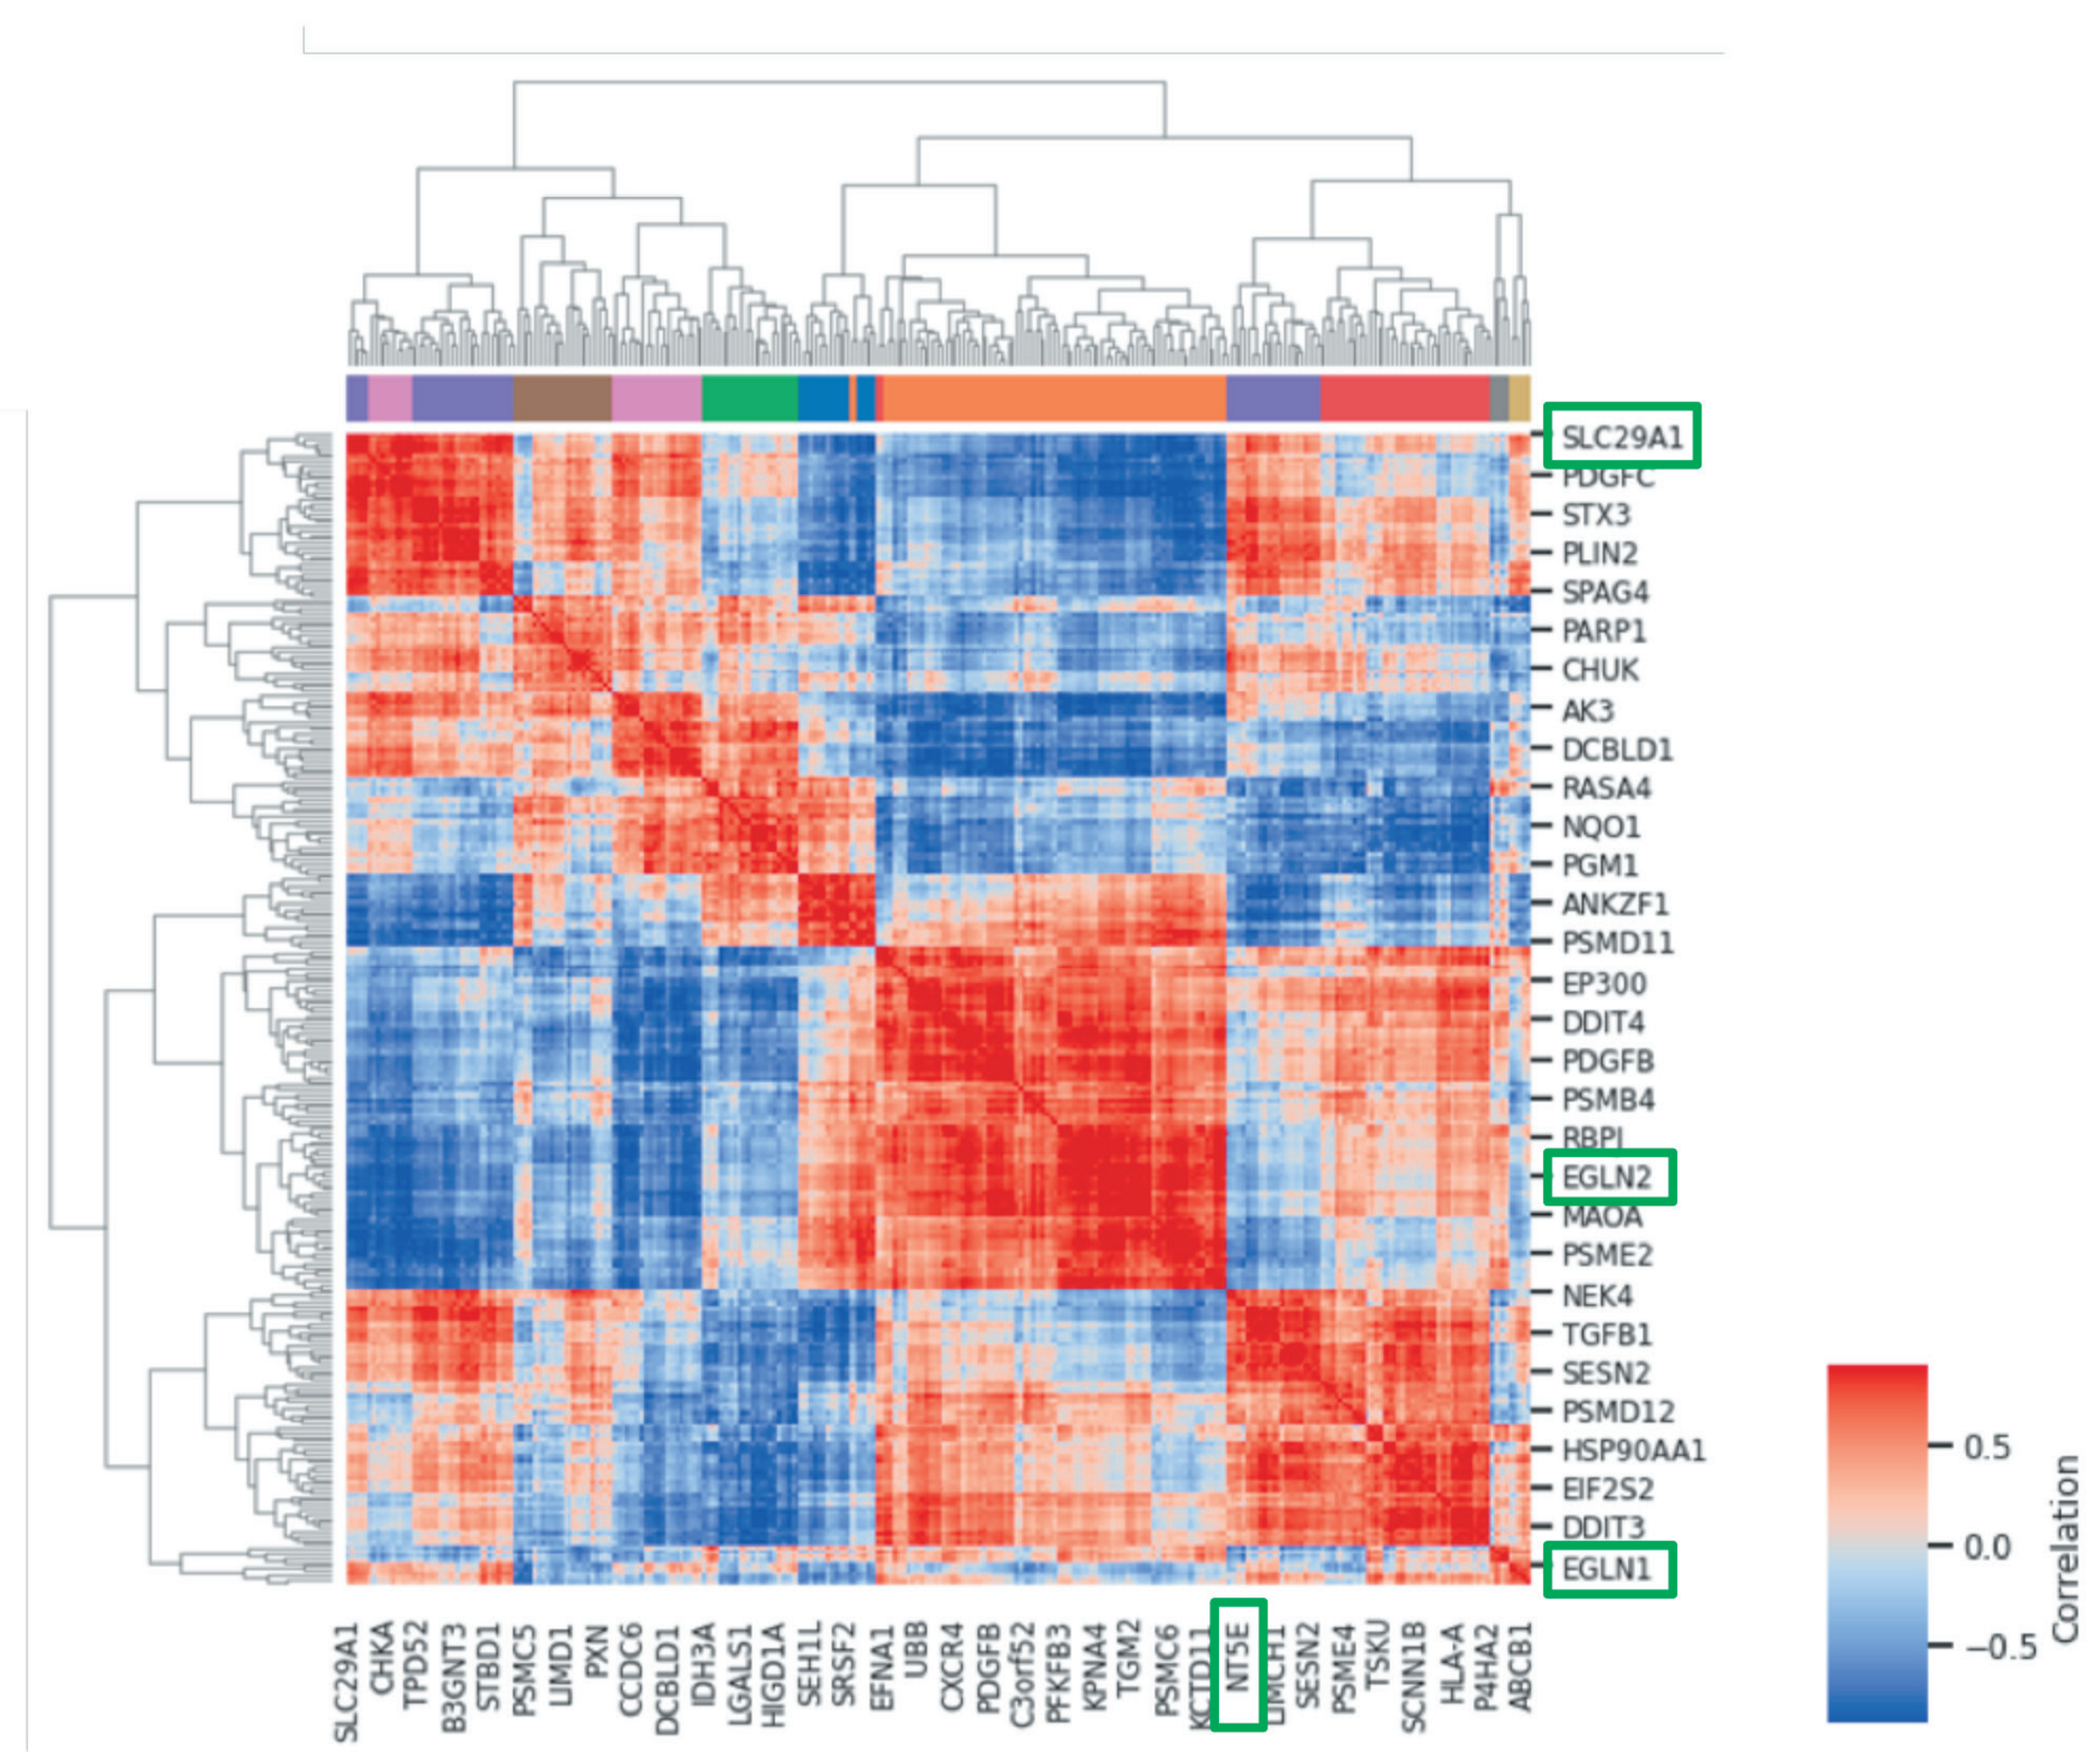

SLC29A1
EGLN2
EGLN1
NT5E
Correlation

CONCLUSIONS

- GB004 demonstrates direct protective effects on mouse organoid and human-derived differentiated monolayer epithelial cultures
- GB004 induces HIF-dependent genes, specifically genes that drive barrier integrity, which are critical to mucosal repair in IBD
- GB004 preserves barrier integrity in human monolayer cultures that have been stimulated with cytokines
- GB004 prevents TNFα-induced cell death and preserves epithelial cell survival
- This data complements data generated in mouse models of colitis demonstrating induction of HIF-1α dependent genes, reduced barrier dysfunction and beneficial efficacy

REFERENCES

1. Danese S, Fiocchi C. *N Eng J Med.* 2011;365:1713-1725.
2. Nielsen OH., Ainsworth MA. *N Eng J Med.* 2013;369:754-762.
3. Petkau JM, Eksteen B. *Biologics.* 2016;10:33-52.
4. Robinson S, Keeley A, Karhausen J, et al. *Gastroenterology.* 2008;134:145-155.
5. Keeley S, Campbell EL, Baird, AW, et al. *Mucosal Immunology.* 2014;7: 114-123.
6. Marks E, Goggins BJ, Cardona C, et al. *Inflamm Bowel Dis.* 2015;21:267-275.

DISCLOSURES

KRTM, SM, GJO, BGL, LC, and LSC are employed by Gossamer Bio, Inc.

Prepared for presentation at Digestive Disease Week (DDW) virtual meeting, May 21-23, 2021.