P556

GB7208 is a CNS-Penetrant BTK Inhibitor Demonstrating Potent Activity on Pathogenic Pathways Implicated in Multiple Sclerosis

Isharat Yusuf, Bryan Clemons, Zachary Naiman, Susan Murphy, David Guimond, Theodore Schiff, Eduardo Garcia, Mark Rose, Marcos Steinberg, Kristen Taylor Meadows, Laura Carter

Gossamer Bio, Inc., San Diego, CA, USA

BACKGROUND

- Bruton's tyrosine Kinase (BTK) plays an important role in B cell and myeloid cell activation and is a critical signaling node downstream of both the B cell receptor (BCR) and Fragment crystallizable receptors (FcR)[1, 2]
- Aberrant BTK signaling in B cells and central nervous system (CNS) resident microglia cells has been associated with Multiple Sclerosis (MS)[3-5]
- GB7208, a selective, CNS-penetrant small molecule BTK inhibitor (BTKi) has the potential to modulate multiple pathogenic mechanisms in MS, including CNS resident microglia

Figure 1. Targeting BTK could address multiple nodes driving autoimmunity in Multiple Sclerosis

BTK plays a role in both peripheral and CNS-resident cells contributing to MS pathogenesis | **BTKi both in the periphery and CNS targets multiple pathogenic mechanisms**

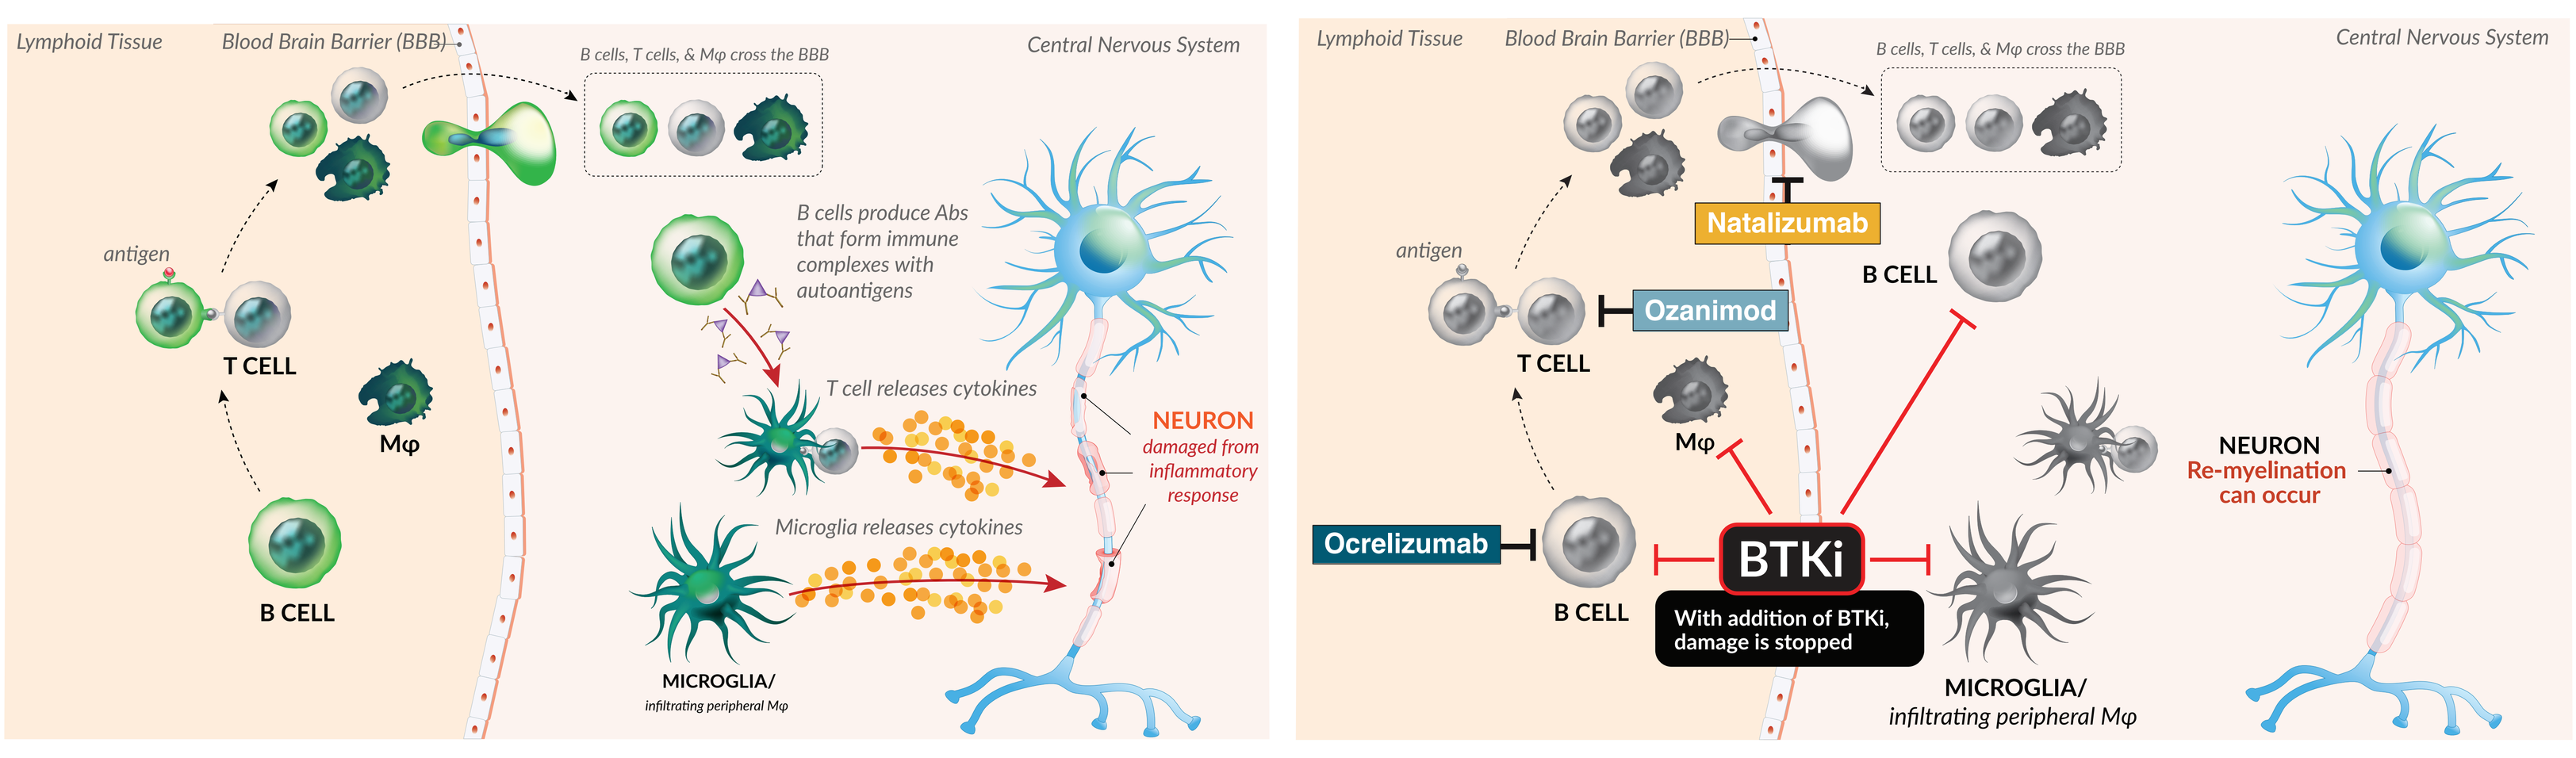
Lymphoid Tissue
Blood Brain Barrier (BBB)
B cells, T cells, & Mφ cross the BBB
Central Nervous System
antigen
T CELL
Mφ
B CELL
B cells produce Abs that form immune complexes with autoantigens
T cell releases cytokines
NEURON damaged from inflammatory response
Microglia releases cytokines
MICROGLIA/ infiltrating peripheral Mφ
Natalizumab
Ozanimod
Ocrelizumab
BTKi
With addition of BTKi, damage is stopped
NEURON Re-myelination can occur

- FcR-mediated signaling by immune complex drives pathology of myeloid cells both in periphery and CNS.[6,7]
- B cells, both in the periphery and CNS, contribute to pathogenesis in MS[6]
- B cell follicles in CNS correlates with poor prognosis in SPMS[7]
- CNS-penetrant BTKi targets pathogenic mechanisms in both the periphery and CNS
- GB7208 is differentiated from current therapies in MS, such as B cell depletion (ocrelizumab) and S1P receptor modulators (ozanimod)

OBJECTIVE

- Profile the potency and efficacy of GB7208 in disease-relevant mechanisms and models

METHODS

- Potency of GB7208 was measured in human B cells, downstream of BCR activation with receptor crosslinking antibodies and human CD14+ monocytes after FcR crosslinking with plate bound IgG
- Effects of GB7208 were evaluated in an experimental autoimmune encephalomyelitis (EAE) mouse model that shares many features of human MS
- A mouse model of anti-myelin oligodendrocyte glycoprotein (MOG) antibody driven microglia proliferation in the brain of wild-type C57BL/6 mice was used to assess the *in vivo* activity of GB7208 on microglia
- GB7208 activity was evaluated in induced pluripotent stem cells (iPSC) derived microglia stimulated with FcR crosslinking by heat aggregated immune complexes

RESULTS

Figure 2. GB7208 inhibits activation of both innate and adaptive immune responses

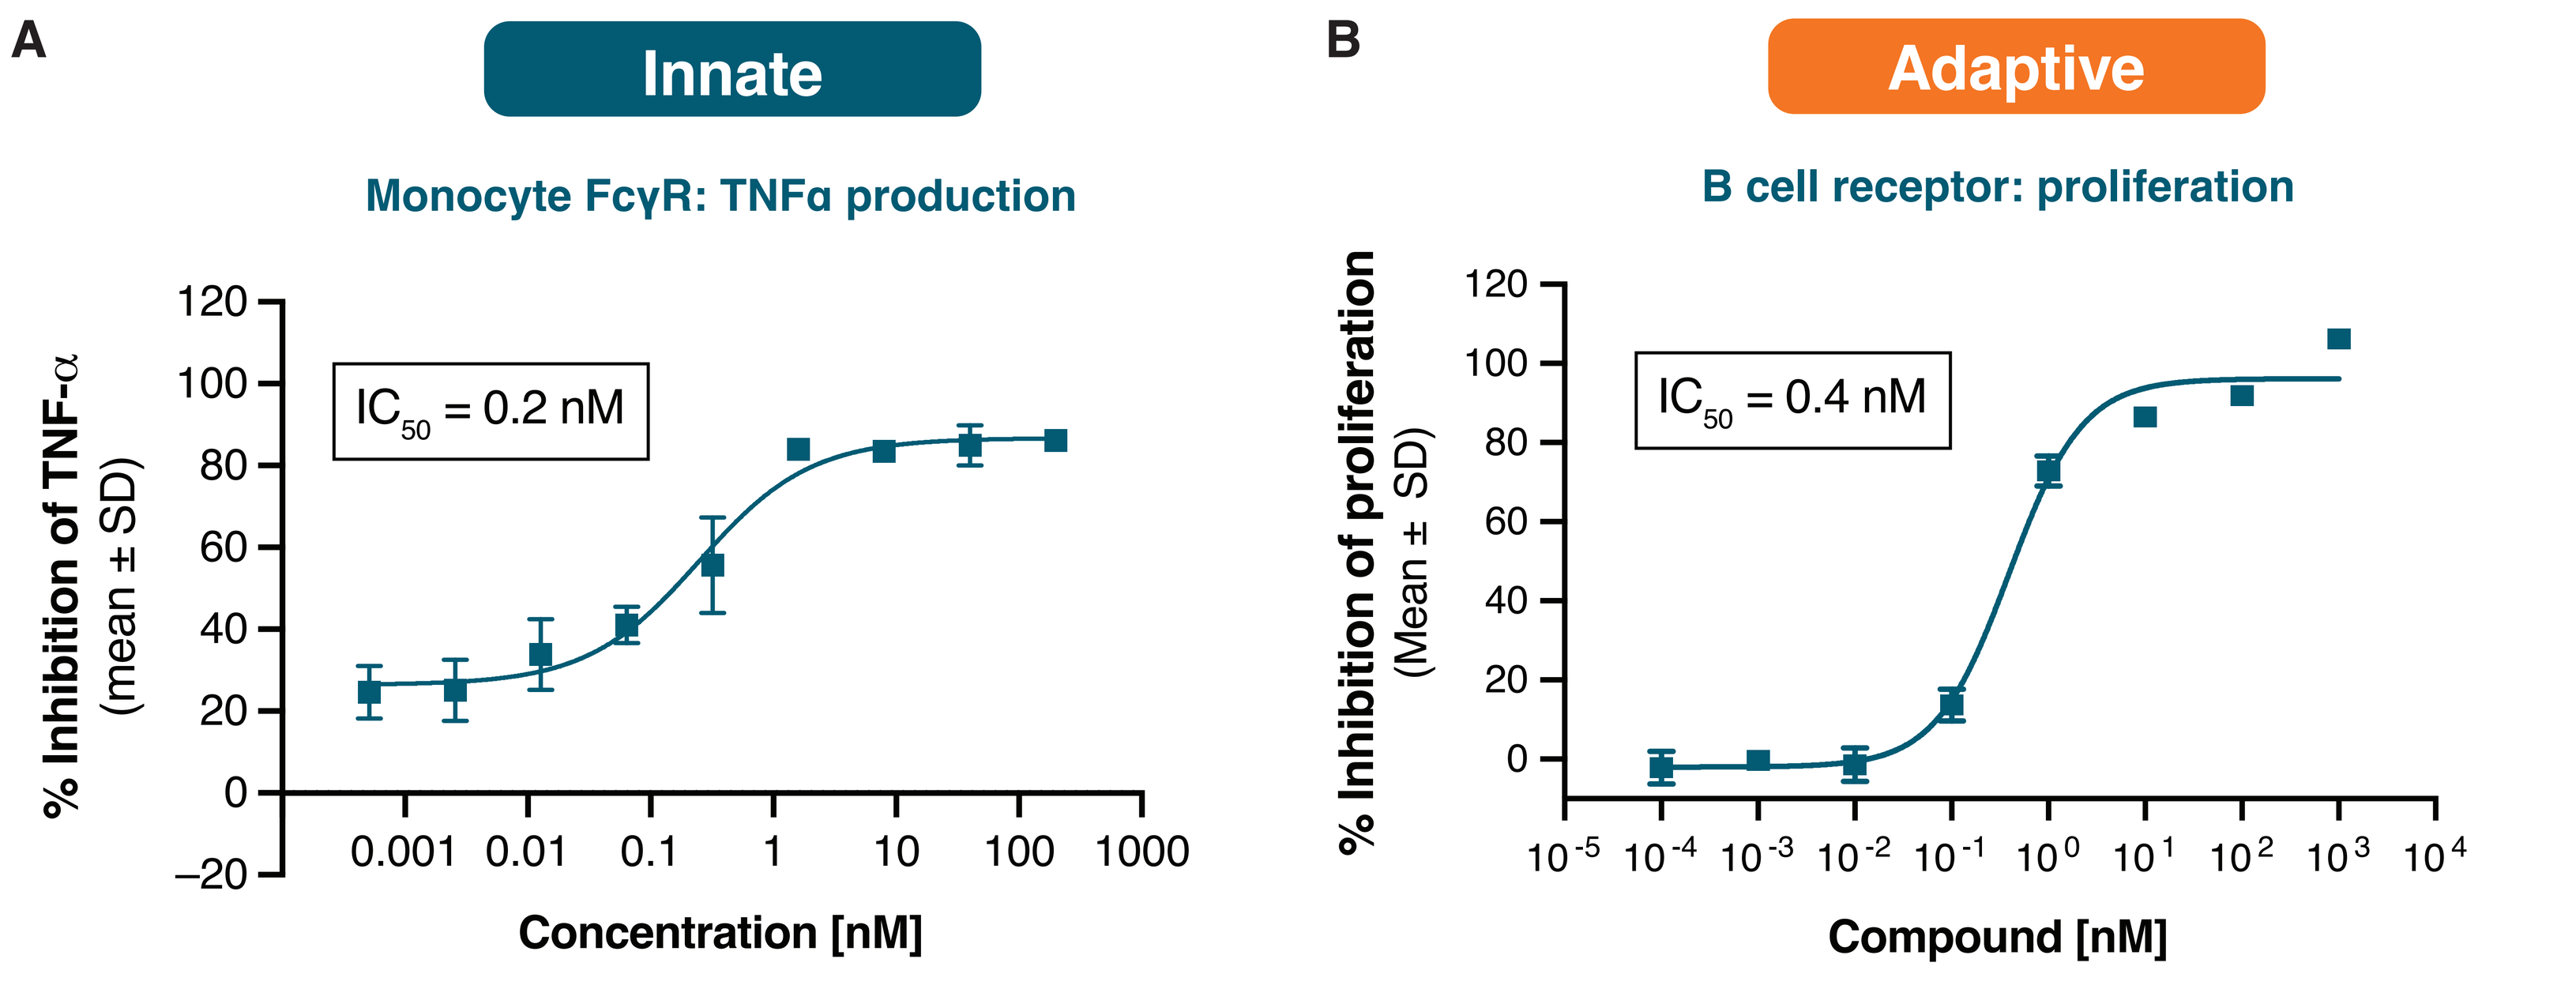
A
Innate
Monocyte FcγR: TNFα production
IC50 = 0.2 nM
% Inhibition of TNF-α (mean ± SD)
Concentration [nM]
B
Adaptive
B cell receptor: proliferation
IC50 = 0.4 nM
% Inhibition of proliferation (Mean ± SD)
Compound [nM]

A. CD14+ human monocytes were stimulated with plate-bound IgG for 6 hours. TNFα was measured in supernatant by Mesoscale Discovery kit. Data are representative of 2 independent experiments. **B.** Purified human B cells were stimulated with anti-IgD for 3 days. Proliferation was measured using Cell Titer Glo. Data are are representative of 3 independent experiments representing unique donors. Dose-response curves generated from triplicate values at each concentration of test article using a nonlinear, 4-parameter, variable slope curve-fitting function using GraphPad Prism software. SD, standard deviation.

Figure 3. GB7208 demonstrates dose-dependent efficacy in autoimmune encephalomyelitis (EAE) model of MS

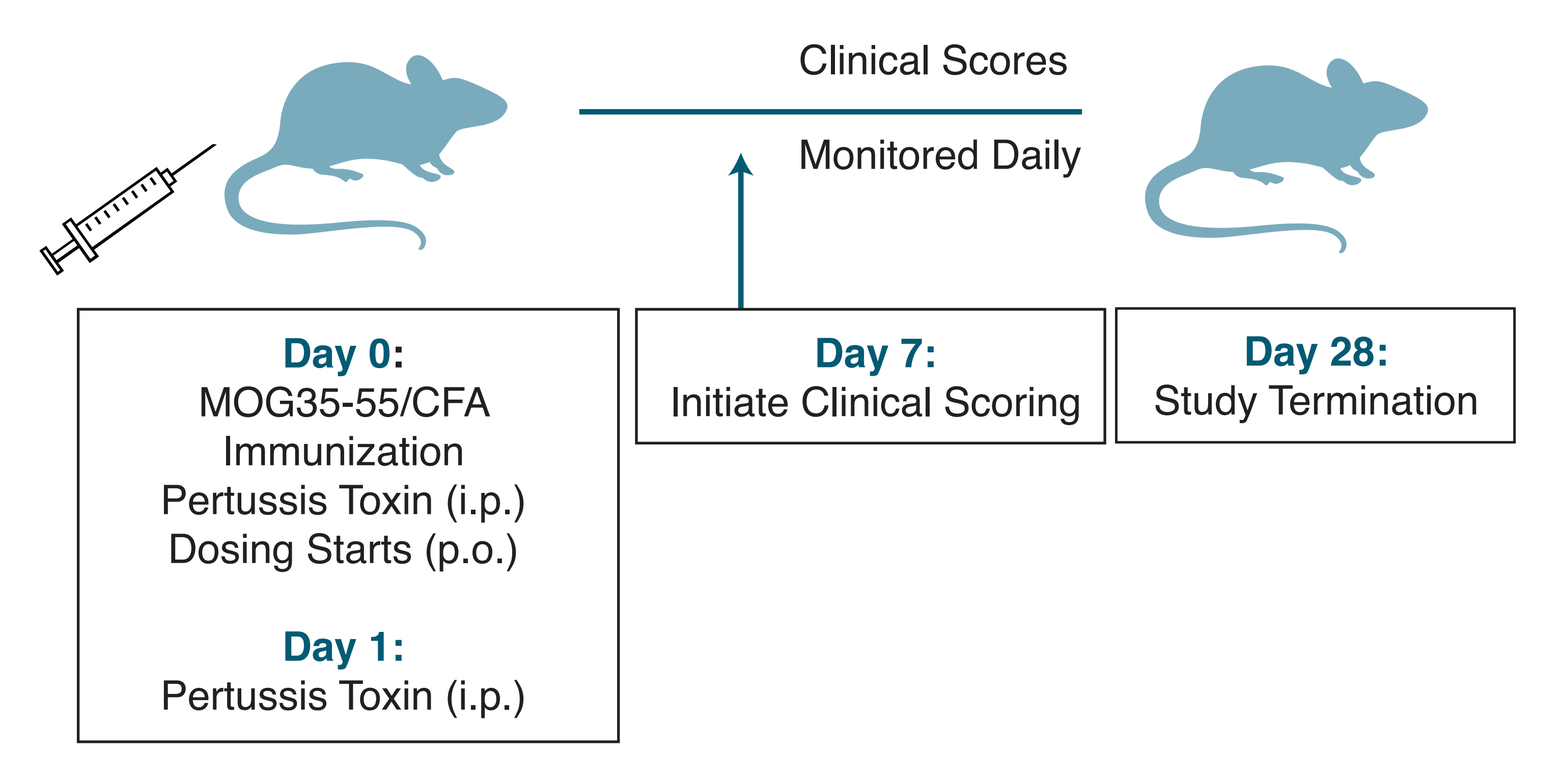
A
Clinical Scores
Monitored Daily
Day 0:
MOG35-55/CFA Immunization
Pertussis Toxin (i.p.)
Dosing Starts (p.o.)
Day 1:
Pertussis Toxin (i.p.)
Day 7:
Initiate Clinical Scoring
Day 28:
Study Termination

Clinical Score Scale:
1 – limp tail
2 – partial hind leg paralysis
3 – complete hind leg paralysis
4 – complete hind and partial front leg paralysis
5 – moribund

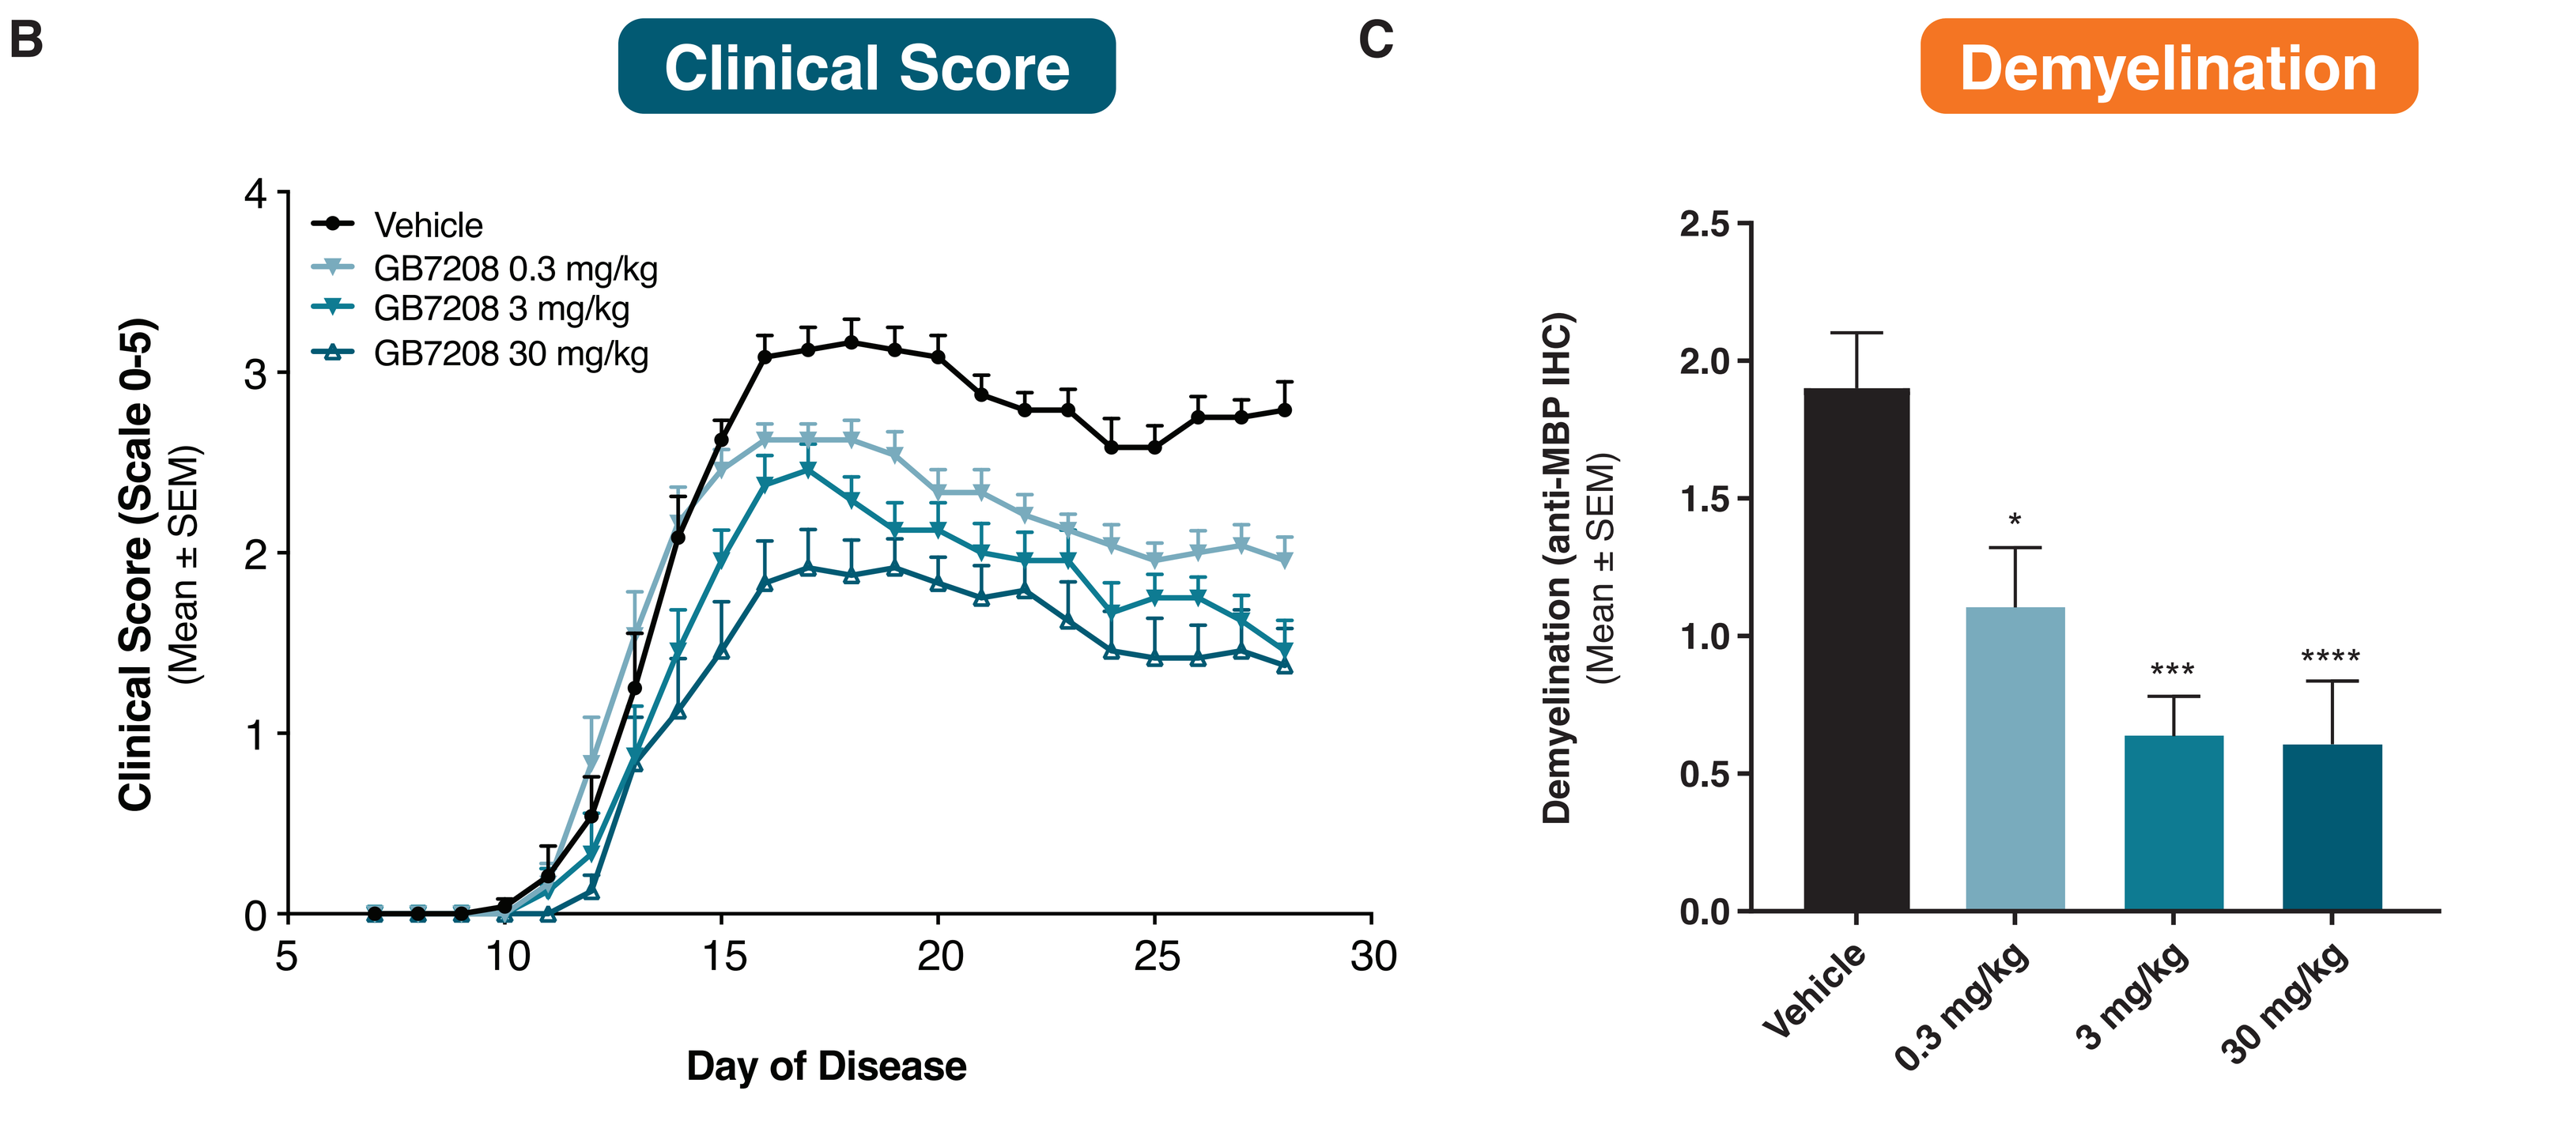
B
Clinical Score
Vehicle
GB7208 0.3 mg/kg
GB7208 3 mg/kg
GB7208 30 mg/kg
Clinical Score (Scale 0-5) (Mean ± SEM)
Day of Disease
C
Demyelination
Demyelination (anti-MBP IHC) (Mean ± SEM)
Vehicle
0.3 mg/kg
3 mg/kg
30 mg/kg

A. Study design. Study included n=12 C57BL/6 mice per group. **B.** Clinical score assigned for each mouse based on criteria outlined in Figure 3A. **C.** Demyelination score on mouse spines isolated at conclusion of study. The spine of each mouse was fixed, paraffin-embedded, sectioned, and stained with anti-myelin basic protein antibody. An area on each section was scored on a scale from 0–5 (0: no demyelination; 5: >50% demyelination). Graph represents the average scores generated from scores given to the cervical, thoracic and lumbar sections of the spine. Graphs generated using GraphPad Prism software. SEM, standard error of the mean. Statistics determined by one-way ANOVA. *p<0.05, ***p<0.001, ****p<0.0001.

RESULTS

Figure 4. GB7208 inhibits proliferation in a mouse model of microglia activation induced by anti-MOG autoantibodies

- MOG autoantibodies drive activation and proliferation of microglia in mice with an intact BBB
- Activation is driven by BTK-dependent FcR signaling

A. Model overview. Study included n=6 C57BL/6 mice per group. On study day 0, mice were given a single intraperitoneal injection of 30 mg/kg anti-MOG IgG and compound dosing (p.o. QD) was initiated. Mice were dosed for a total of four days with either GB7208 or tolebrutinib. Brain and spinal cord samples were collected 1 hour after the final dose. Animals were perfused with PBS before tissue collection. **B.** Ki-67 was measured by flow cytometry and **C.** BTK target occupancy was measured using a probe-based ELISA. Statistics determined by one-way ANOVA. *p<0.05, **p<0.01, ***p<0.001, ****p<0.0001. Tolebrutinib clinical dose of 60mg QD = 10 mg/kg based on allometric scaling or ~0.07 mg/kg based on plasma AUC. Abs, antibodies; MOG, myelin oligodendrocyte glycoprotein; FcR, Fc receptor; BBB, blood brain barrier.

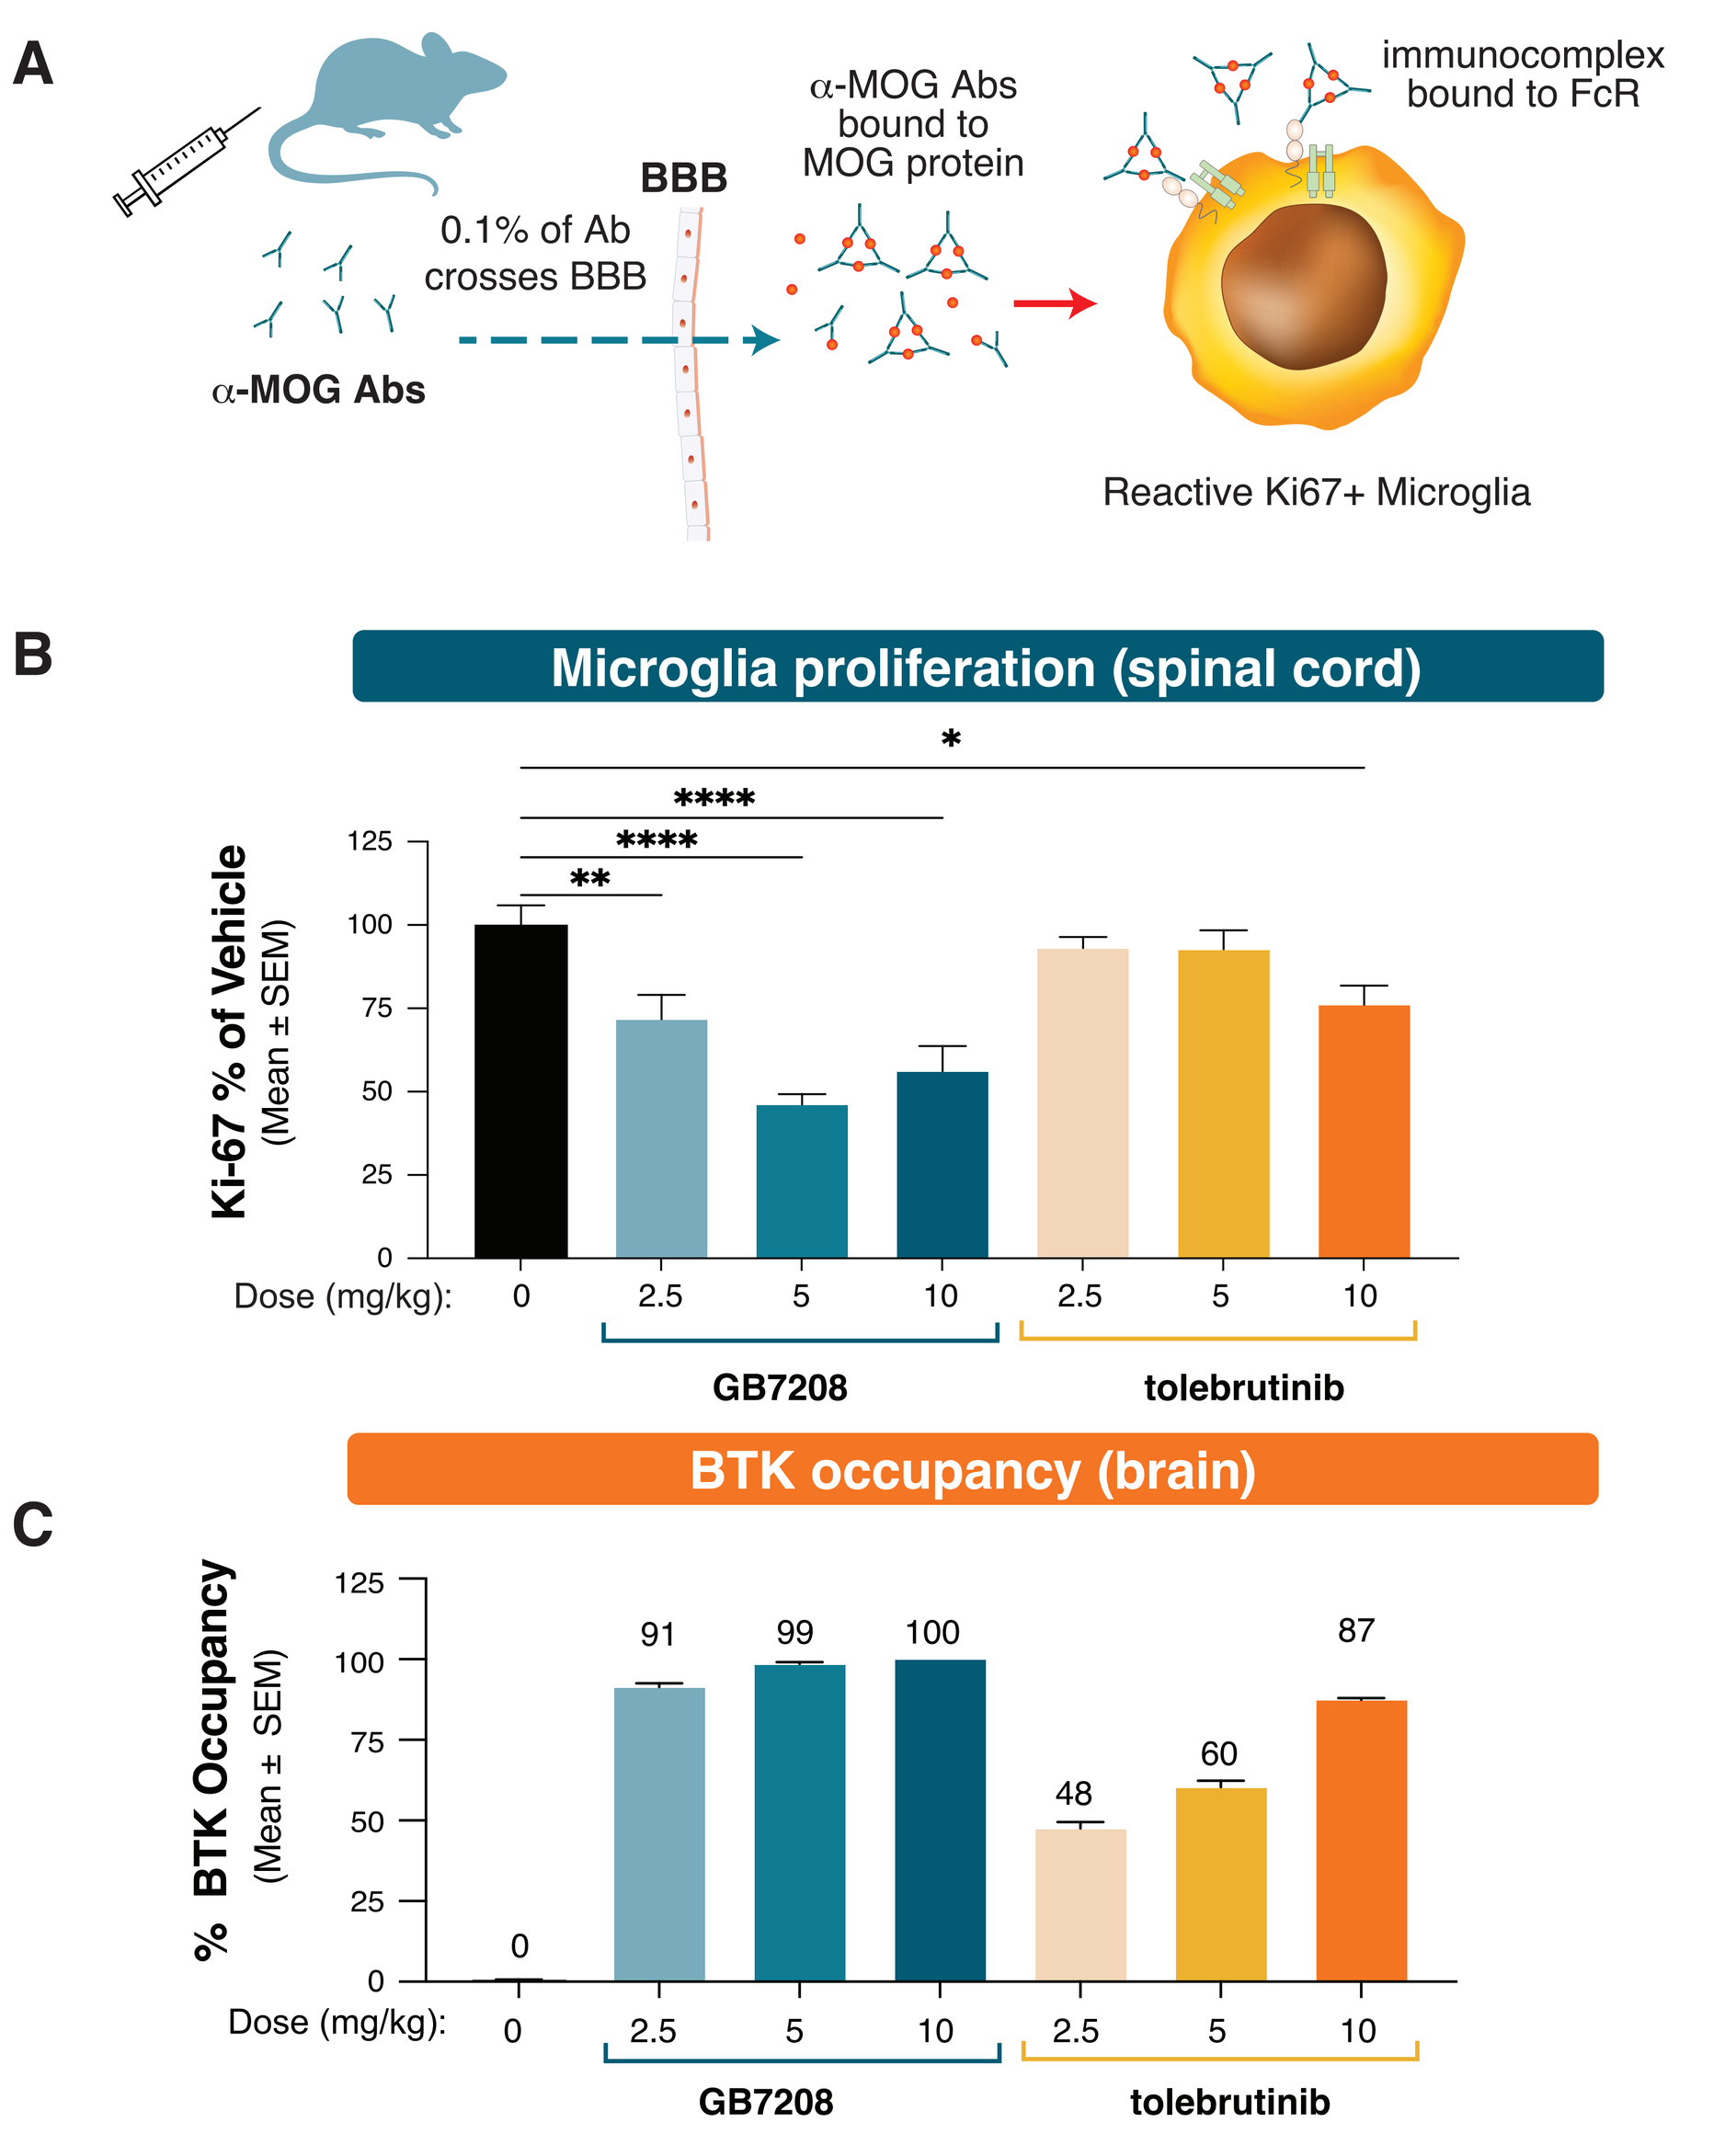
A
α-MOG Abs
0.1% of Ab crosses BBB
BBB
α-MOG Abs bound to MOG protein
immunocomplex bound to FcR
Reactive Ki67+ Microglia
B
Microglia proliferation (spinal cord)
Ki-67 % of Vehicle (Mean ± SEM)
Dose (mg/kg): 0 2.5 5 10 2.5 5 10
GB7208
tolebrutinib
C
BTK occupancy (brain)
% BTK Occupancy (Mean ± SEM)
0
91
99
100
48
60
87
Dose (mg/kg): 0 2.5 5 10 2.5 5 10
GB7208
tolebrutinib

Figure 5. GB7208 inhibits FcR-dependent TNFα production by human microglia

- GB7208 dose-dependently inhibits the production of TNFα in human microglia stimulated with IgG-immunocomplexes and IFNγ
- TNFα is elevated in PPMS and causes excitotoxic neurodegeneration[9]
- GB7208 shows superior potency to tolebrutinib and orelabrutinib

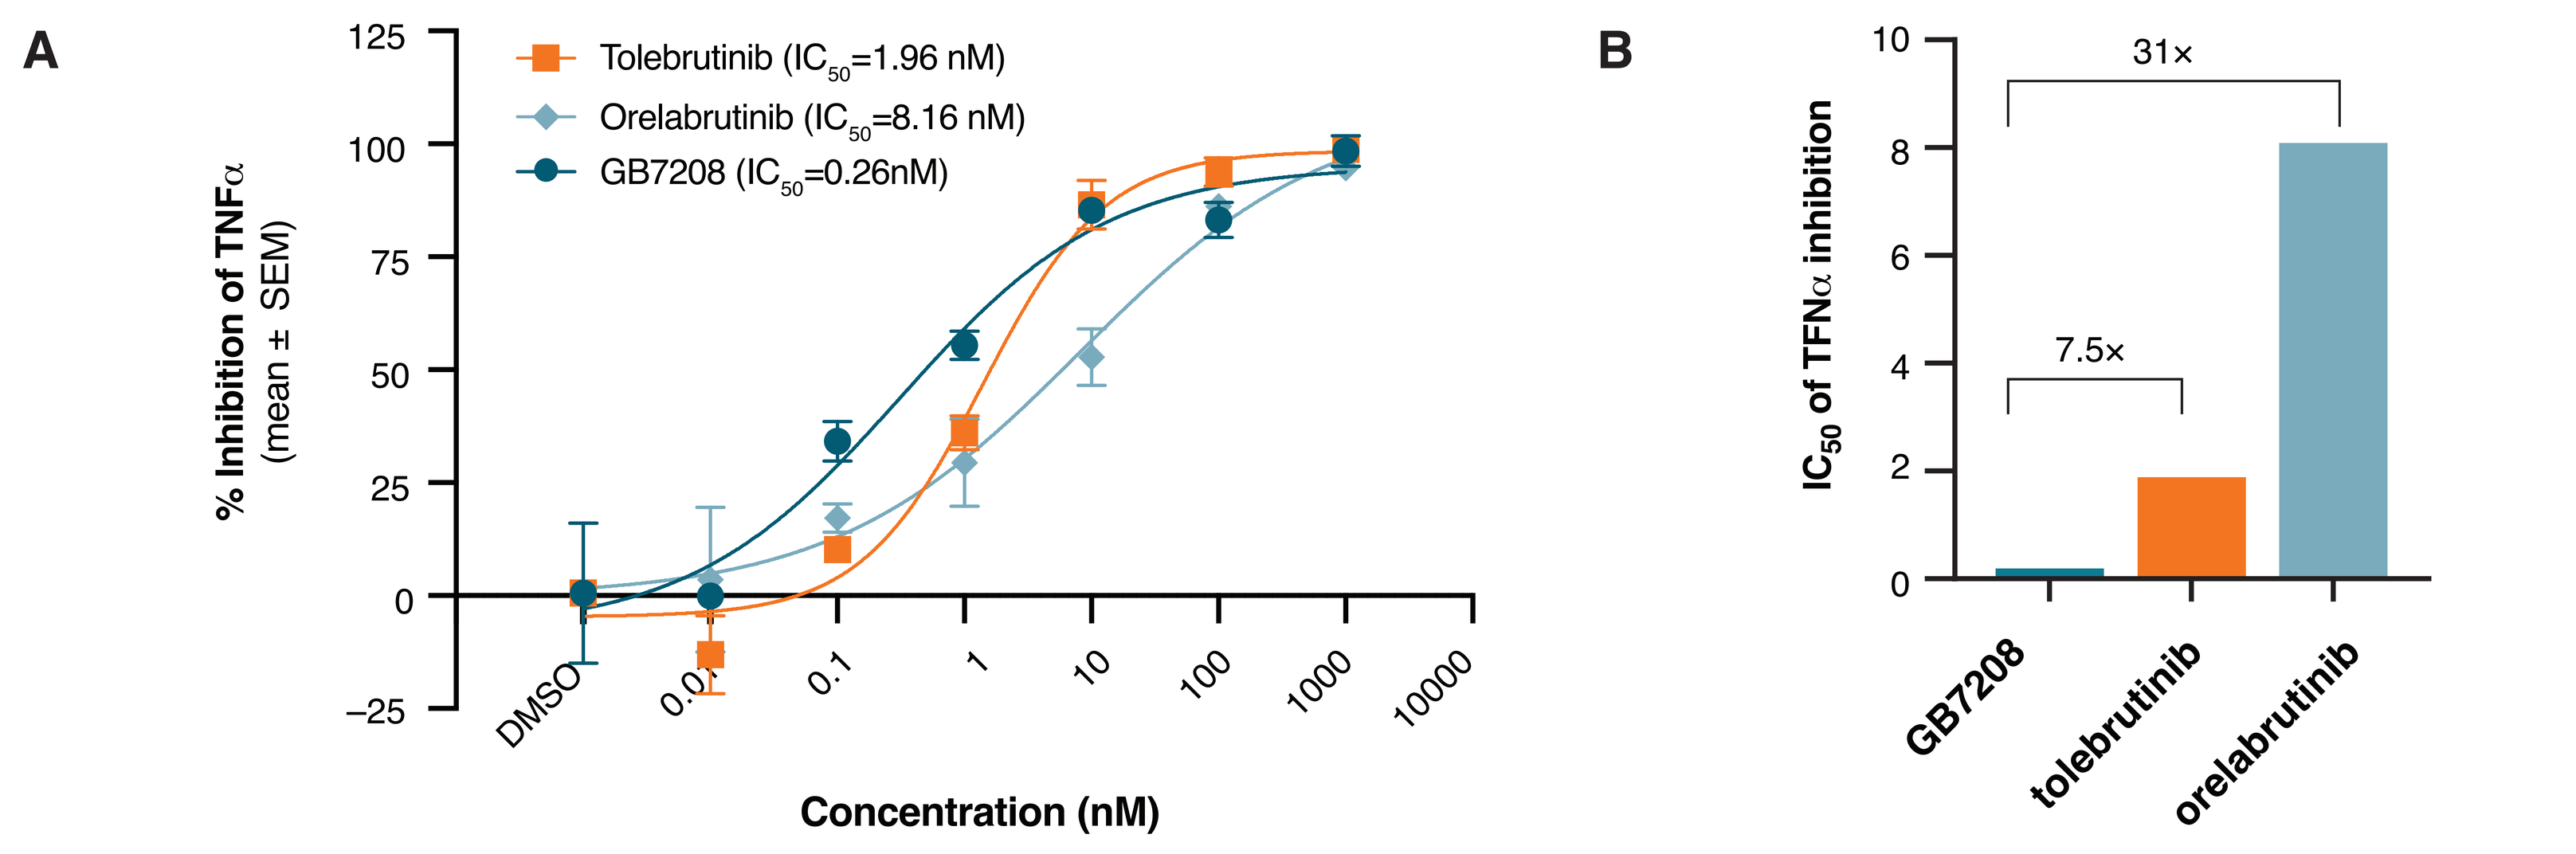
A
Tolebrutinib (IC50=1.96 nM)
Orelabrutinib (IC50=8.16 nM)
GB7208 (IC50=0.26nM)
% Inhibition of TNFα (mean ± SEM)
Concentration (nM)
B
31×
7.5×
IC50 of TFNα inhibition
GB7208
tolebrutinib
orelabrutinib

iPSC-derived microglia were stimulated with heat-aggregated IgG immune complexes for 24 hours. TNFα was measured in supernatant by Mesoscale Discovery kit. Data are representative of 1-3 independent experiments. **A.** Dose response curves generated from triplicate values at each concentration of test article using a nonlinear, 4-parameter, variable slope curve-fitting function using GraphPad Prism software. SD, standard deviation. **B.** Fold difference in IC_{50} potency

Table 1. GB7208 is a differentiated BTKi primed to make transition to the clinic

		Tolebrutinib (Phase 3)	Evobrutinib (Phase 3)	GB7208 (preclinical)
Pharmacology	Human WB potency (CD69 IC_{50} nM)	17	119	22
	BTK rate of inactivation (x $10^{-4}nM^{-1}min^{-1}$)	4.4	0.4	3.4
	Kinome selectivity (>50% inhibition)*	8/349	ND[10]	5/349
	EGFR cellular potency (μM)	0.08	>10	>10
	% CNS target occupancy (naïve mice; 2.5, 5, 10 mg/kg dose)	46%, 66%, 96%	18% (3 mg/kg dose)	83%, 91%, 99%
ADME	Preclinical Brain/Plasma ratio (mouse, rat, monkey**)	0.04, 0.08, 0.66	0.03, ND, ND	0.32, 0.50, 0.81
	Human Dose Projection (Kp=0.5)	60 mg QD#	45 mg BID#	<50 mg QD

*1 µM compound concentration with 1 mM ATP (AssayQuant)
**monkey cassette dosing (IV)
#clinical dose
ND, not determined by Gossamer Bio

PLEASE SEE RELATED POSTER
"GB7208 is a novel, highly potent and selective CNS-penetrant BTK inhibitor for neuroinflammatory and neurodegenerative diseases"

SUMMARY

- **GB7208 potently inhibits BTK-dependent innate and adaptive immune response**
- **GB7208 demonstrates dose-dependent efficacy in the MOG_{35-55} EAE preclinical model of MS**
- **In a mouse model of microglia activation driven by anti-MOG antibodies, GB7208 leads to significant inhibition of proliferation which is consistent with high CNS target occupancy**
- **GB7208 potently inhibits TNFα production by human iPSC derived microglia cells stimulated with immune complexes**
- **GB7208 is differentiated CNS-penetrant BTKi that modulates several pathogenic mechanisms in MS**

REFERENCES

1. Corneth OBJ, et al. *Curr Top Microbiol Immuno*l. 2016;(393)67-105.
2. Weber ANR, et al. *Front Immunol* 2017; 8(1454).
3. Myhr KM, et al. *Expert Opin Biol Ther*. 2019;19(3):261-271.
4. Mitsdoerffer M, et al. *Front Immunol.* 2016;7:451.
5. Li Q, et al. *Nat Rev Immunol.* 2018;18 (4):225-242.
6. von Büdingen HC, et al. *J Clin Invest* 2012;122(12):4533-4543.
7. Serafini B, et al. *Brain Pathol.* 2004;14(2):164-174.
8. Pellerin K. et al. *Mult Scler* 2020; 26(3_suppl): P0941.
9. Rossi S, et al. *Mult Scler* 2014; 20(3):304-312.
10. Haselmayer P, et al. *J Immunology*. 2019; 10: 2888-2906.

ACKNOWLEDGEMENTS

The authors gratefully acknowledge Daniel Buzzard, Vijay Kumar, Nidhi Arora, Martin Rowbottom and Craig Coburn for medicinal chemistry efforts in the development and identification of GB7208.

DISCLOSURES

All authors are employees and stockholders of Gossamer Bio, Inc. This research is being funded by GB005, Inc., an affiliate of Gossamer Bio, Inc.

Presented at the 38th Congress of the European Committee for Treatment and Research in Multiple Sclerosis, Amsterdam, The Netherlands, October 26-28, 2022