P690

GB7208 is a Novel, Highly Potent and Selective CNS-Penetrant BTK Inhibitor for Neuroinflammatory and Neurodegenerative Diseases

Isharat Yusuf, Mark Rose, David Guimond, Zachary Naiman, Bryan Clemons, Theodore Schiff, Brian Le, Kristen Taylor Meadows, Laura Carter

Gossamer Bio, Inc., San Diego, CA, USA

BACKGROUND

- Bruton's tyrosine kinase (BTK) mediates signaling downstream of B cell receptor (BCR) and Fragment crystallizable receptors (FcR) suggesting an important role in autoimmune disease[1,2]
- BTK inhibitors (BTKi) are being explored in neuroinflammatory/neurodegenerative diseases including Multiple Sclerosis (MS) but have modest selectivity and/or limited central nervous system (CNS) penetration[3,4,5]
- GB7208 is a pre-clinical, investigational drug product candidate that has been designed to be a CNS-penetrant, irreversible small molecule BTKi optimized for potential best-in-class potency, selectivity and brain exposure

Figure 1. BTK is a key mediator of innate & adaptive immune responses implicated in neuroinflammatory and neurodegenerative diseases

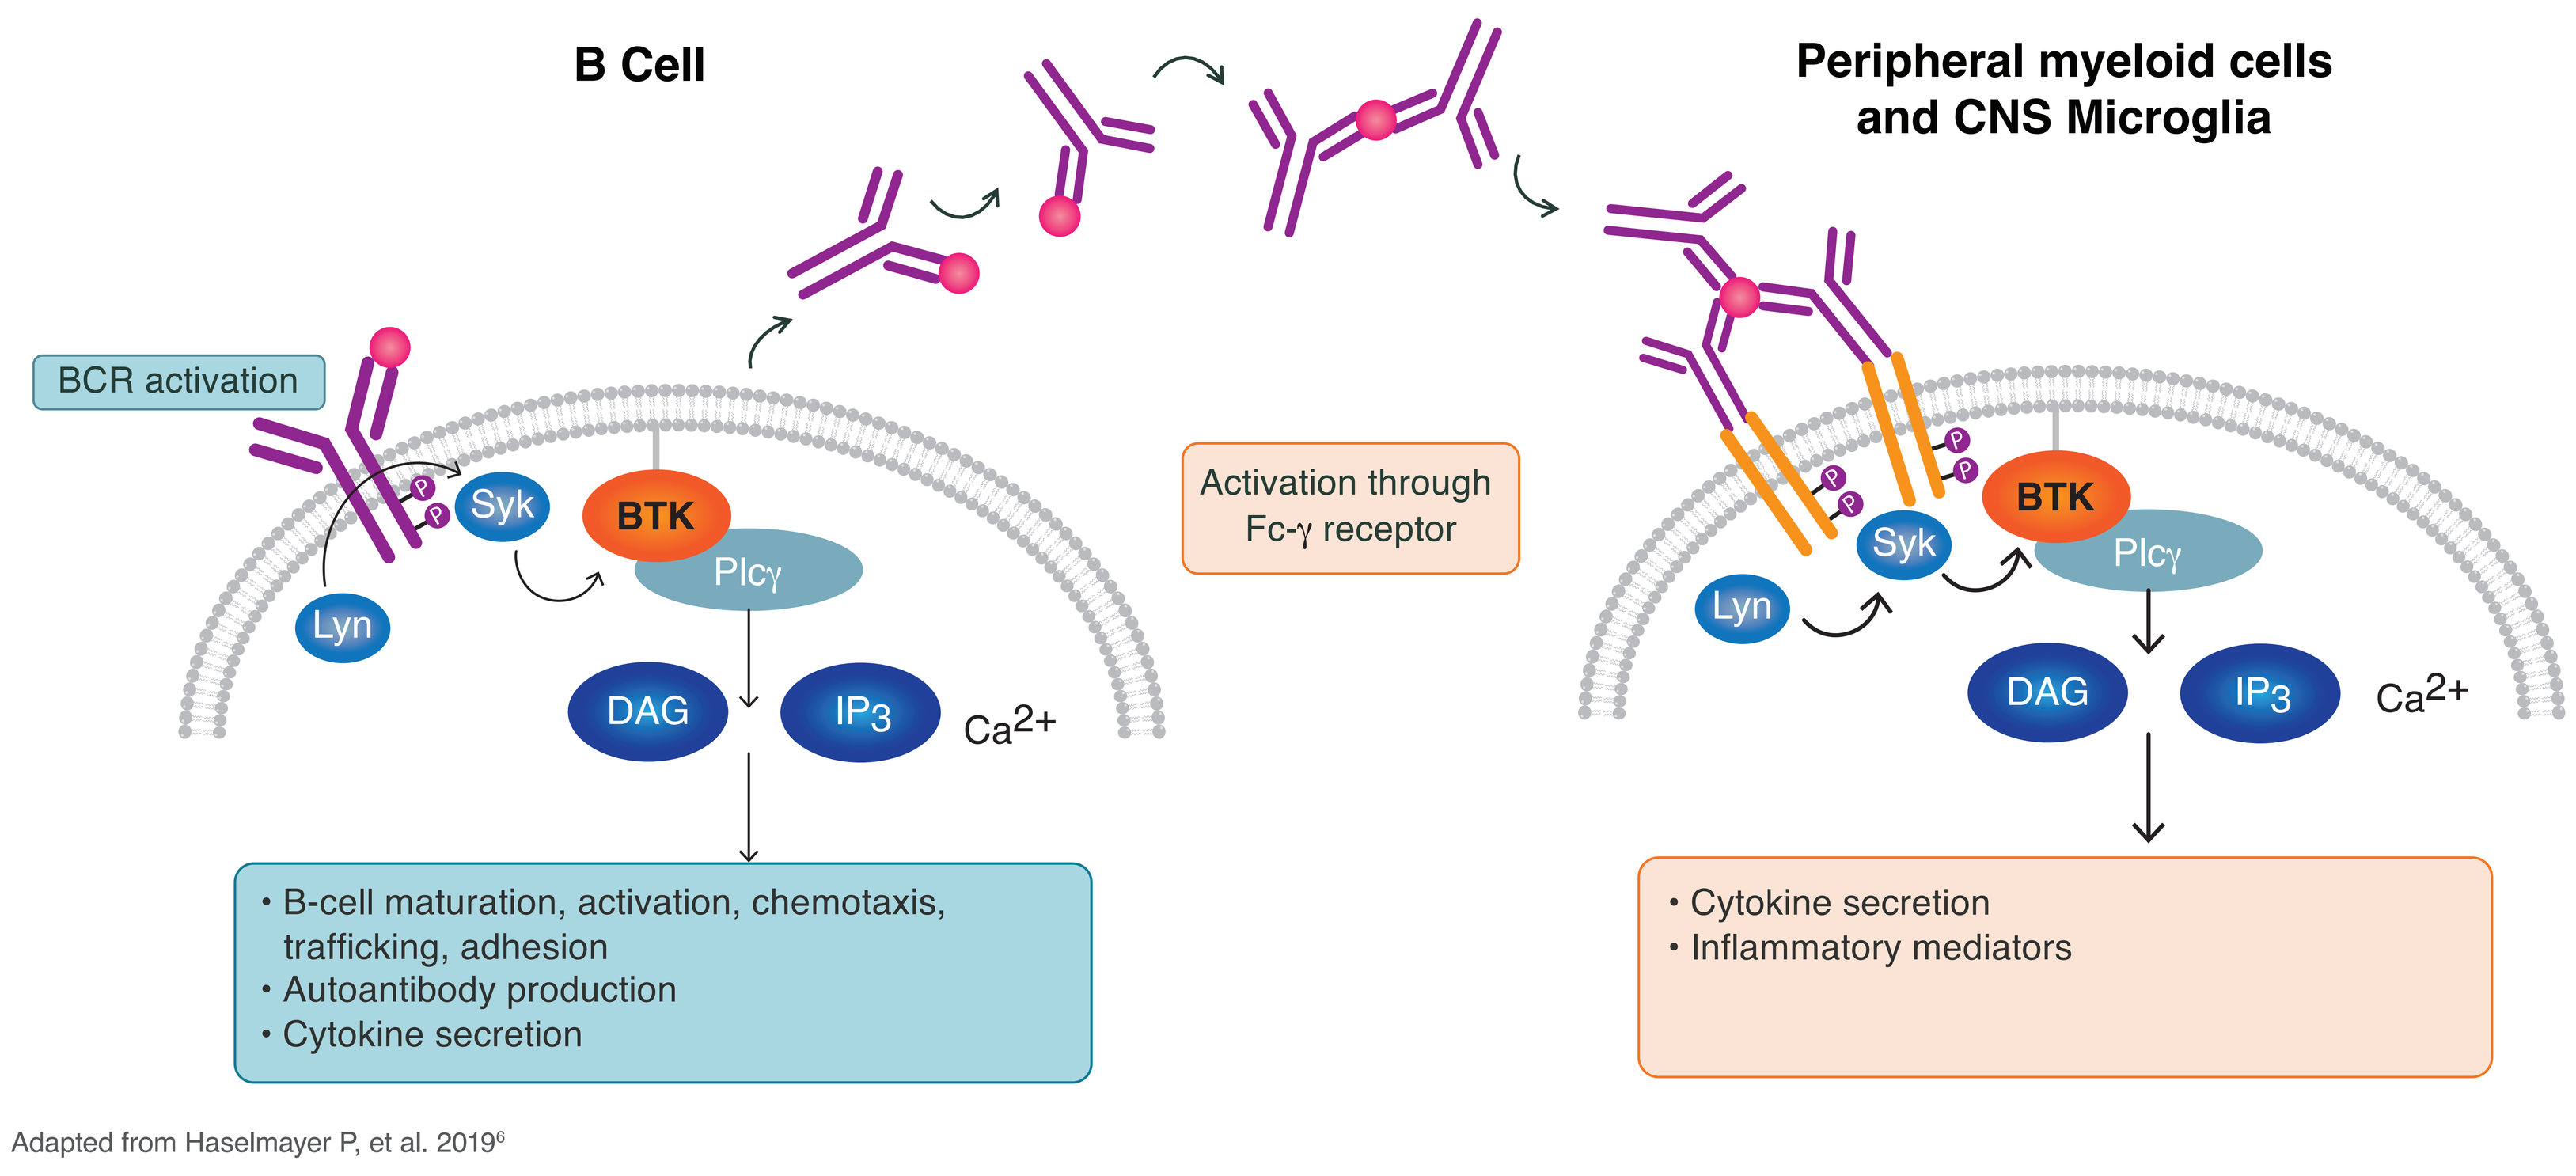
B Cell
Peripheral myeloid cells and CNS Microglia
BCR activation
Syk
BTK
Plcγ
Lyn
DAG
IP3
Ca2+
Activation through Fc-γ receptor
• B-cell maturation, activation, chemotaxis, trafficking, adhesion
• Autoantibody production
• Cytokine secretion
• Cytokine secretion
• Inflammatory mediators

Adapted from Haselmayer P, et al. 2019[6]

OBJECTIVE

- Profile GB7208 preclinically for pharmacologic properties, selectivity and CNS exposure across several species

METHODS

- Irreversible nature of GB7208 was evaluated using enzymatic assays
- *In vitro* potency and inactivation kinetics of GB7208 were evaluated using biochemical and cell-based assays
- Selectivity of GB7208 and tolebrutinib was profiled in kinome scans against 349 kinases at 1 μM under conditions using 1mM ATP concentration; EGFR activity was further evaluated in a cell-based assay
- Pharmacokinetic profiles of GB7208 were obtained in multiple species using oral and intravenous dosing
- Comparative BTK target occupancy of GB7208 and tolebrutinib was assessed in perfused mice with an intact blood brain barrier (BBB) using a probe-based ELISA

RESULTS

Table 1: Summary of BTK enzymatic mechanism of inhibition studies demonstrates properties of an irreversible inhibitor

Assay		GB7208
BTK Enzyme Assay Preincubation (nM IC_{50})	0 min	39.2
	5 min	8.1
	15 min	1.5
	30 min	0.8
	45 min	0.6
	60 min	0.5
BTK enzyme recovery/Residence Time		>400 min
Irreversible Inhibition		yes

Table 2: Potency and inactivation kinetics in cells and whole blood

Assay	GB7208
hWB pBTK IC_{50}	20.4 nM
Ramos calcium flux IC_{50}	7.4 nM
hWB CD69 IC_{50}	22.7 nM
B cell proliferation IC_{50}	0.5 nM
Monocyte FcR TNFα IC_{50}	0.15 nM
hWB kinact/KI	3.2 10^{-4} nM^{-1} min^{-1}
mWB kinact/KI	3.1 10^{-4} nM^{-1} min^{-1}
mB kinact/KI	1.5 10^{-4} nM^{-1} min^{-1}

hWB, human whole blood; mWB, mouse whole blood; mB, mouse brain; FcR, Fc Receptor

Figure 2. GB7208 is a highly selective BTK inhibitor

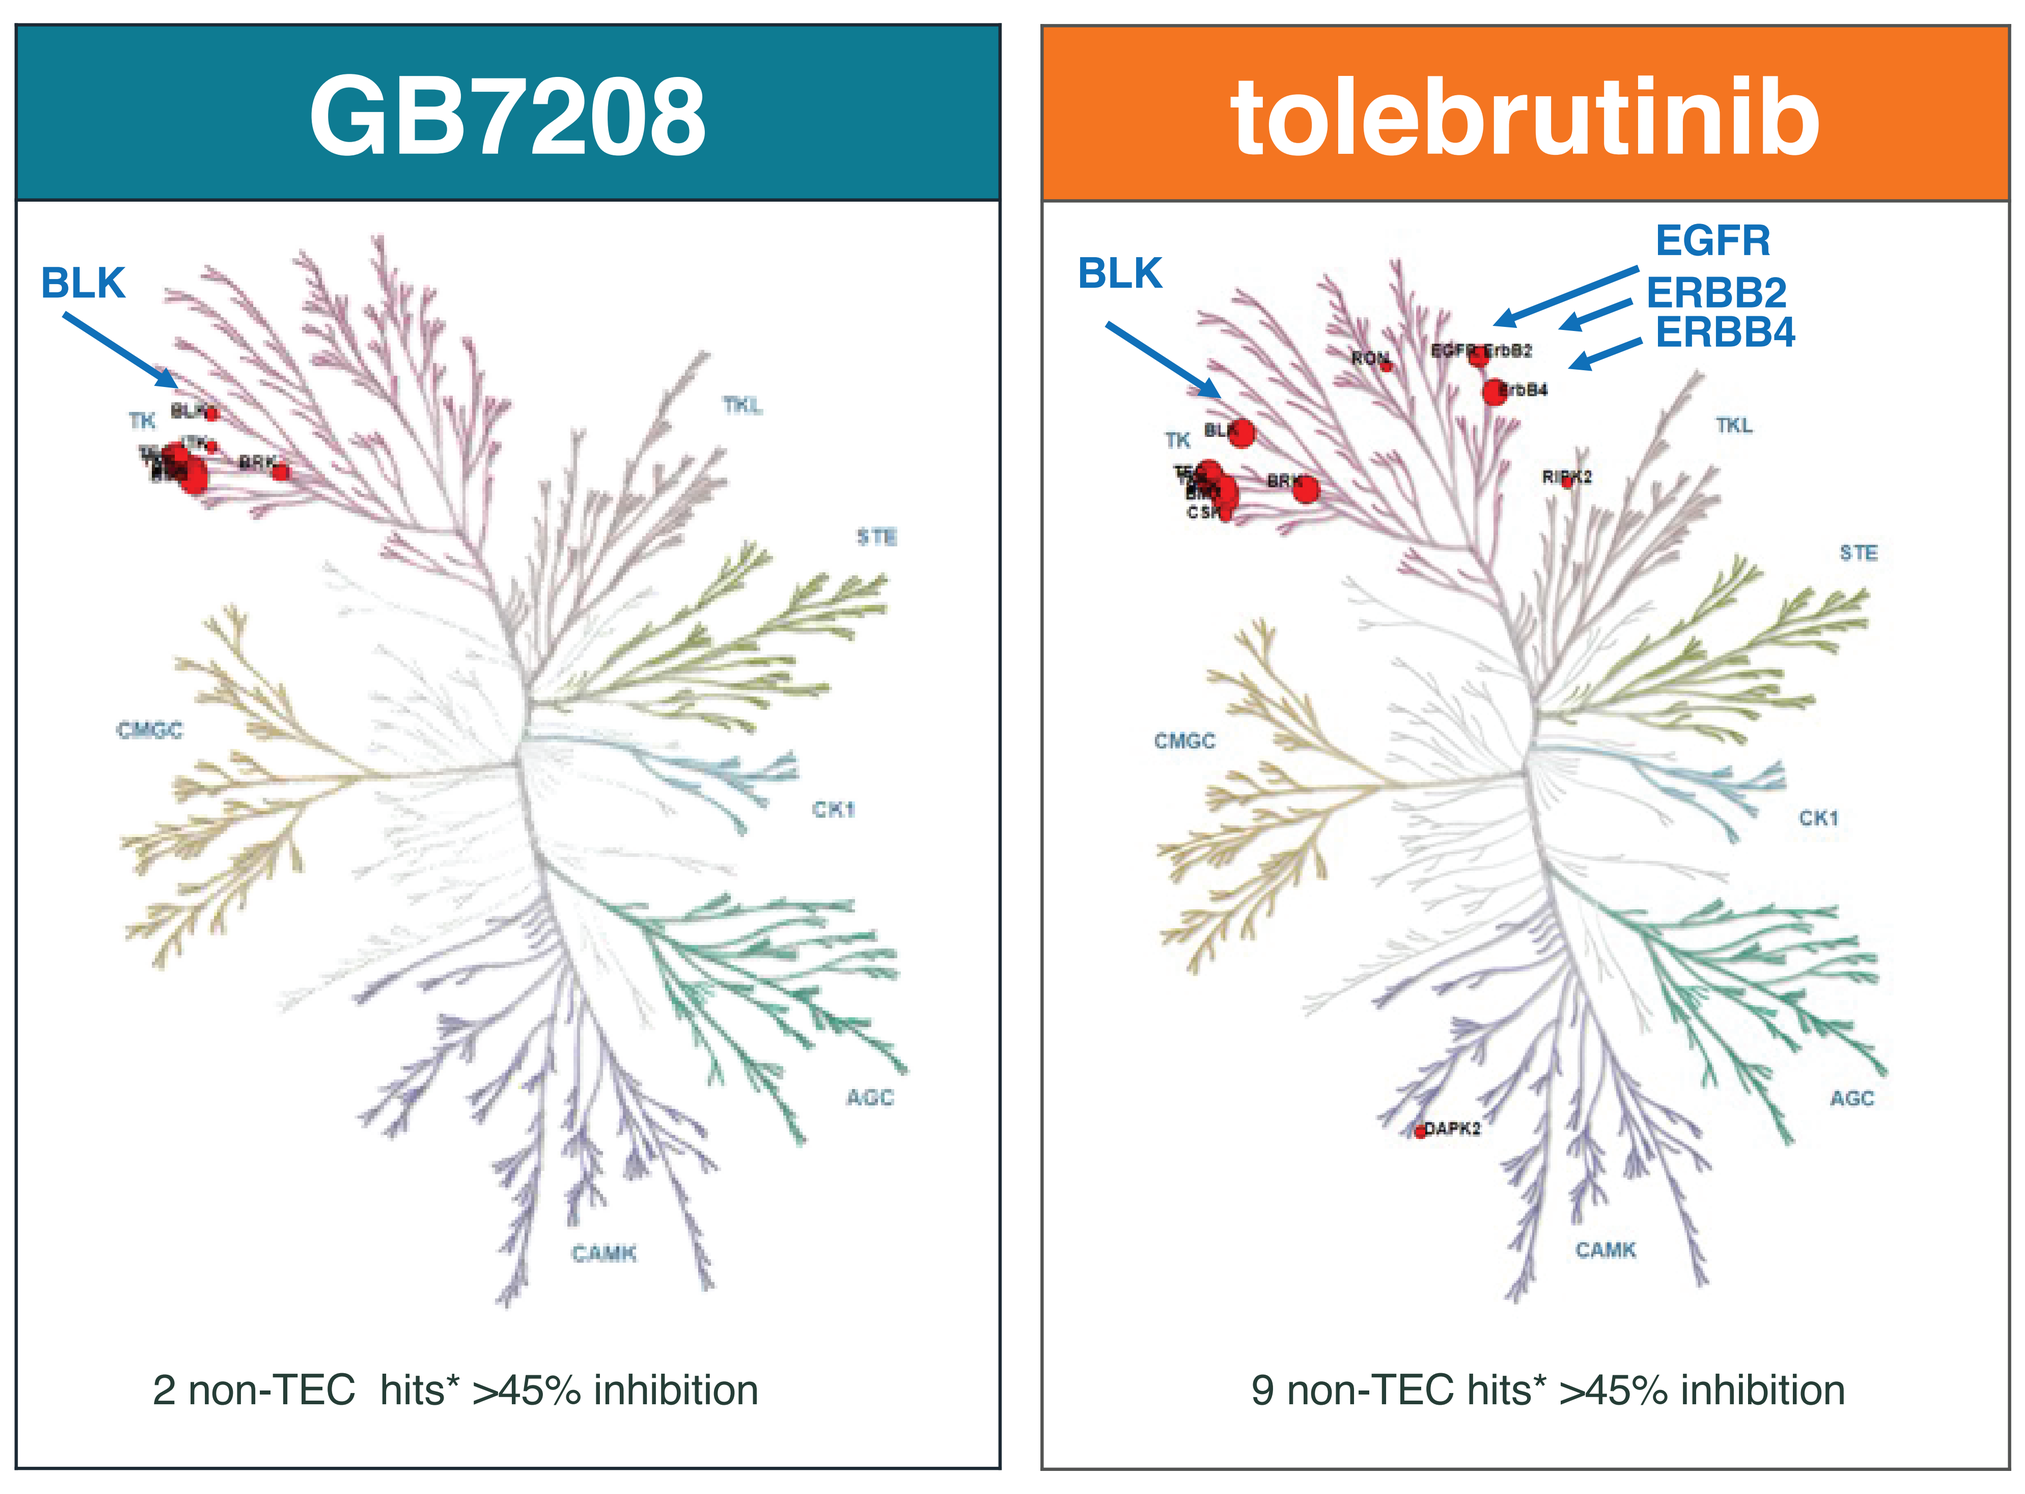
GB7208
tolebrutinib
BLK
EGFR
ERBB2
ERBB4
TK
TKL
STE
CMGC
CK1
AGC
CAMK
2 non-TEC hits* >45% inhibition
9 non-TEC hits* >45% inhibition

*Non-TEC family kinases inhibited >45% at 1 μM compound concentration with 1 mM ATP (AssayQuant)

● denote >45% inhibition

→ denotes non-TEC covalent off-target interaction

** Nanobret in HEK cells

- **GB7208 has two non-TEC kinase hits >45%**
- BLK and BRK/PTK6 (De-risked in cell-based assays**)
- BLK: 1.3 μM (tolebrutinib = 0.001 μM)
- BRK/PTK6: 14 μM (tolebrutinib = 0.224 μM)
- GB7208 lacks EGFR activity

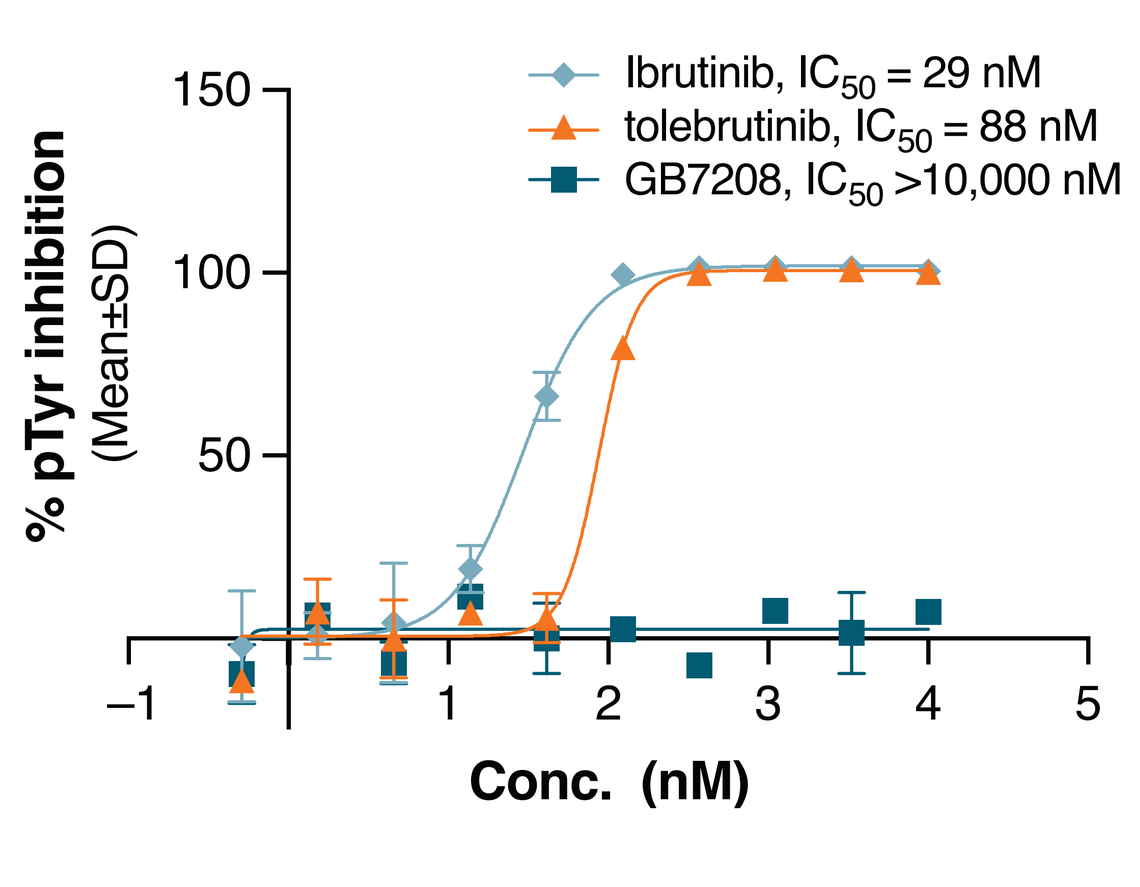
GB7208 does not inhibit EGFR signaling in a cell-based assay
Ibrutinib, IC50 = 29 nM
tolebrutinib, IC50 = 88 nM
GB7208, IC50 >10,000 nM
% pTyr inhibition (Mean±SD)
Conc. (nM)

Data are representative of 5 independent studies. Dose-response curves generated from duplicate values at each concentration of test article using a nonlinear, 4-parameter, variable slope curve-fitting function using GraphPad Prism software. SD, standard deviation, Conc., compound concentration.

Table 3. GB7208 brain to plasma ratios are high after single or multiple daily oral (PO) dosing in multiple species

Species/Strain	N	Dose (mg/kg)	Day (Time)	Plasma (ng/mL)	Brain (ng/g)	CSF (ng/mL)	Brain/ Plasma Ratio	% Brain BTK Target Occupancy
Mouse / C57Bl/6	3	10 (PO)	1 (1h)	1470 ± 329	489 ± 117	ND	0.33	99
Rat / Wistar Han	3	10 (PO)	1 (1h)	622 ± 128	317 ± 99.0	ND	0.50	ND
Dog / Beagle	4	1 (PO)	15 (6h)	184 ± 82.9	109 ± 62.4	4.16 ± 2.30	0.57	100.2 ± 0.8
	4	3 (PO)	15 (6h)	761 ± 234	332 ± 41.9	10.8 ± 3.19	0.46	101.6 ± 0.5
Monkey / Cyno	3	2 (IV)	1 (1h)	355 ± 42.4	292 ± 80.9	9.55 ± 1.67	0.81	ND

ND = not determined, ± in table is standard error of mean (SEM)

Figure 3. Simultaneous concentrations of GB7208 in plasma, brain and CSF were measured out to eight hours after an IV dose (2 mg/kg)

Nine non-naïve cisterna magna cannulated male monkeys were divided into three groups, and each given a single IV bolus dose of GB7208 (2 mg/kg). Plasma (P), CSF, and brain (B) were collected based on the schedule below. Concentrations of GB7208 were determined using LC-MS/MS.

Group	Sampling time point (hr) 0.083	0.25	0.5	1	2	4	8
1 (n=3)	P CSF	P CSF	P CSF	P CSF B	–	–	–
2 (n=3)	P CSF	P CSF	P CSF	P CSF	P CSF	P CSF B	–
3 (n=3)	P CSF	P CSF	P CSF	P CSF	P CSF	P CSF	P CSF B

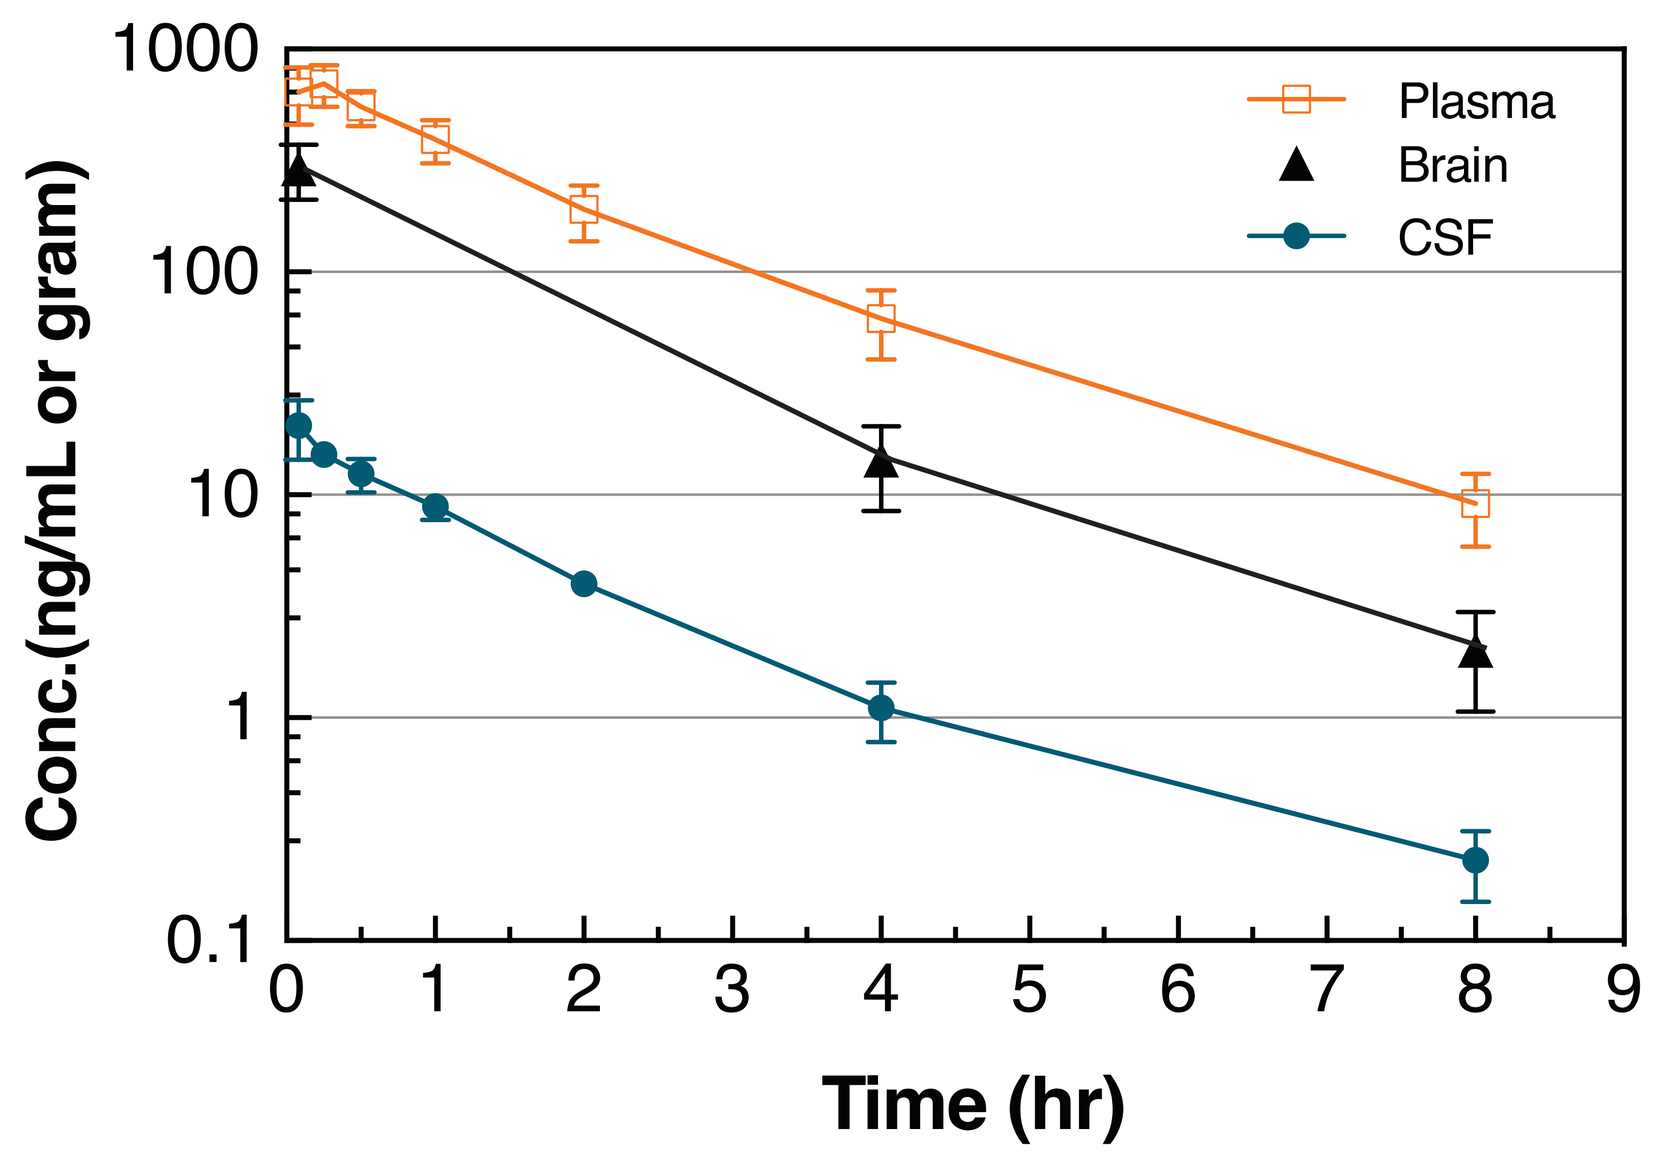
Plasma
Brain
CSF
Conc.(ng/mL or gram)
Time (hr)

Figure 4. GB7208 demonstrates high brain penetrance and target occupancy in a preclinical model

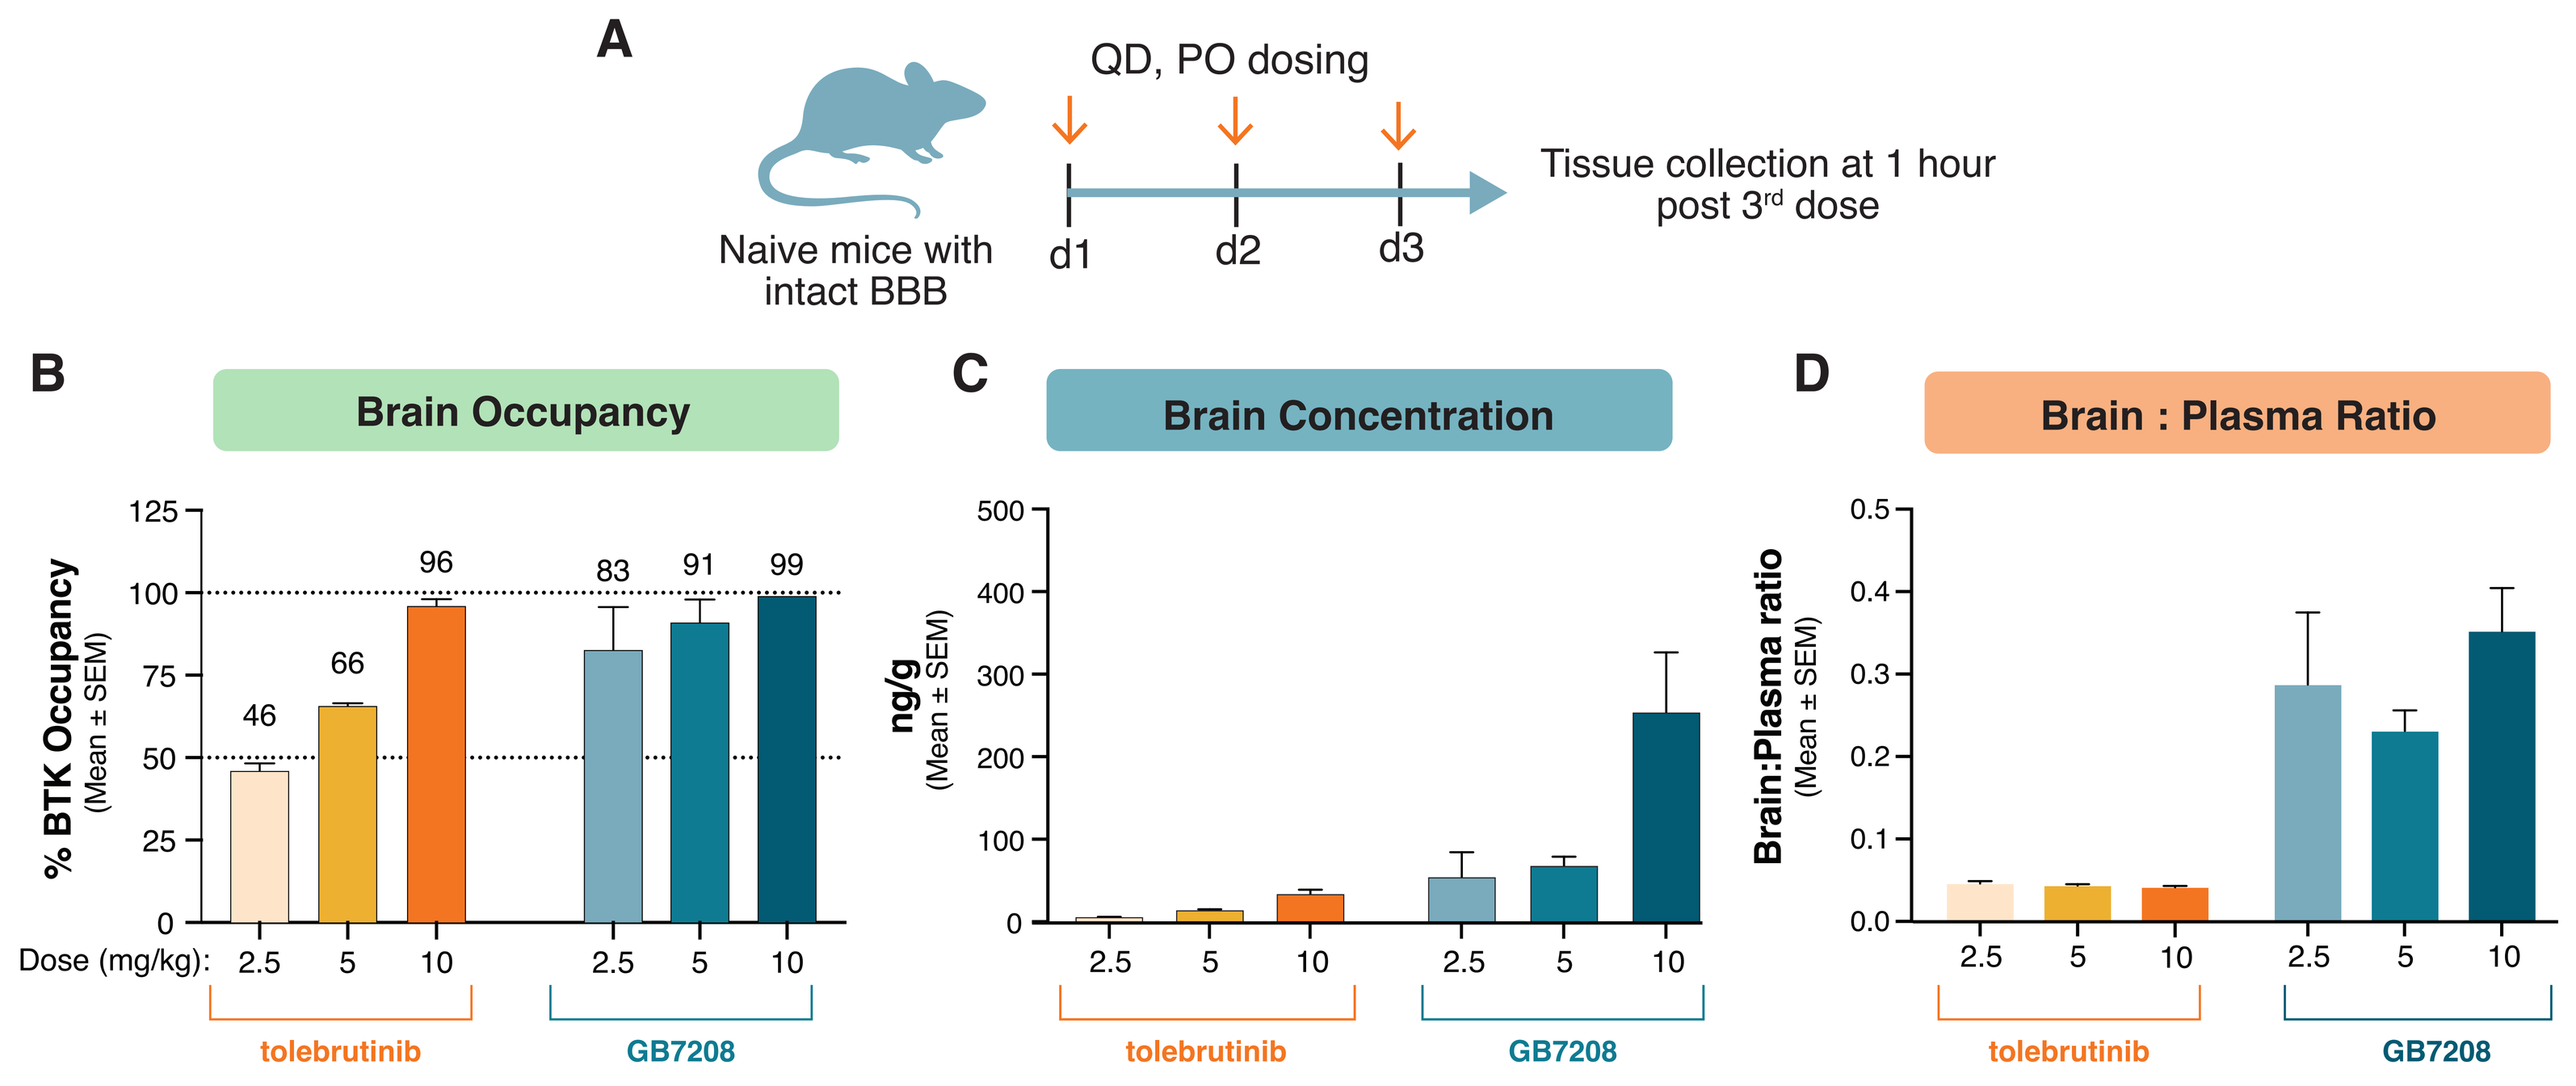
A
QD, PO dosing
Naive mice with intact BBB
d1
d2
d3
Tissue collection at 1 hour post 3rd dose
B
Brain Occupancy
% BTK Occupancy (Mean ± SEM)
46
66
96
83
91
99
Dose (mg/kg):
C
Brain Concentration
ng/g (Mean ± SEM)
D
Brain : Plasma Ratio
Brain:Plasma ratio (Mean ± SEM)
tolebrutinib
GB7208

A. Study design. Study included n=3 C57BL/6 mice per group. Mice were dosed for a total of three days (p.o. QD) with either GB7208 or tolebrutinib and then 1 hour after the final dose. Animals were perfused with PBS before tissue collection. **B**. BTK target occupancy was measured using a probe-based ELISA. **C**. Brain concentration, **D**. Brain to plasma ratio. Graphs were generated in GraphPad Prism software. SEM = Standard error of the mean. Tolebrutinib clinical dose of 60 mg QD = 10 mg/kg based on allometric scaling or ~0.07 mg/kg based on plasma AUC.

Table 4. GB7208 is a differentiated BTK Inhibitor designed to be potent, selective, and highly CNS penetrant with low human dose predictions

		Tolebrutinib (Phase 3)	Evobrutinib (Phase 3)	GB7208 (preclinical)
Pharmacology	Human WB potency (CD69 IC_{50} nM)	17	119	22
	BTK rate of inactivation (x $10^{-4}nM^{-1}min^{-1}$)	4.4	0.4	3.4
	Kinome selectivity (>50% inhibition)*	8/349	ND[6]	5/349
	EGFR cellular potency (μM)	0.08	>10	>10
	% CNS target occupancy (naïve mice; 2.5, 5, 10 mg/kg dose)	46%, 66%, 96%	18% (3 mg/kg dose)	83%, 91%, 99%
Safety	DILI prediction score**	Medium	Medium	Low
ADME	Preclinical Brain/Plasma ratio (mouse, rat, monkey***)	0.04, 0.08, 0.66	0.03, ND, ND	0.32, 0.50, 0.81
	Human Dose Projection (Kp=0.5)	60 mg QD#	45 mg BID#	<50 mg QD

All reported data generated by Gossamer Bio unless otherwise noted, ND = Not determined at Gossamer Bio
*1 μM compound concentration with 1 mM ATP (AssayQuant)
**Drug Induced Liver Injury score based on FDA guidance of low dose, logP, and metabolites[7]
***NHP cassette dosing (IV)
#clinical dose
QD, once daily; BID, twice daily; NHP, non-human primate; WB, whole blood

PLEASE SEE RELATED POSTER
"GB7208 is a CNS-penetrant BTK inhibitor demonstrating potent activity on pathogenic pathways implicated in multiple sclerosis"

SUMMARY AND CONCLUSIONS

- **GB7208 is an irreversible, selective and potent BTK inhibitor being developed for neuroinflammatory and neurodegenerative diseases**
- **GB7208 has excellent CNS penetration across several preclinical species including nonhuman primates**
- **In naïve mice with an intact BBB, GB7208 demonstrates a high brain to plasma ratio and brain target occupancy**
- **Preclinical assessment of GB7208 demonstrates a differentiated profile suitable for development for neuroinflammatory and neurodegenerative diseases such as MS**

REFERENCES

1. Corneth OBJ, et al. *Curr Top Microbiol Immunol*. 2016 (393) 67-105.
2. Weber ANR, et al. *Front Immunol* 2017; 8(1454).
3. Montalban X, et al. *N Engl J Med*. 2019;380(25):2406-2417.
4. Reich DS, et al. *Lancet Neurol* 2021;20(9):729-738.
5. Hauser S et al. *Mult Scler* 2020; 26(3_suppl): P0211
6. Haselmayer P, et al. *J Immunology* 2019; 10: 2888-2906.
7. Chen M., et al. *Hepatology*, 2016, 64(3): 931-940.

ACKNOWLEDGEMENTS

The authors gratefully acknowledge Daniel Buzzard, Vijay Kumar, Nidhi Arora, Martin Rowbottom and Craig Coburn for medicinal chemistry efforts in the development and identification of GB7208. Authors also acknowledge Byranna Paulson and Kay Hou for DMPK support.

DISCLOSURES

All authors are employees and stockholders of Gossamer Bio, Inc. This research is being funded by GB005, Inc., an affiliate of Gossamer Bio, Inc.

Presented at the 38th Congress of the European Committee for Treatment and Research in Multiple Sclerosis, Amsterdam, The Netherlands, October 26-28, 2022