855

Diacylglycerol Kinase Alpha and Zeta Dual Inhibitors Enhance T Cell Responses and Promote Robust and Durable Anti-Tumor T Cell Immunity

Marcos W Steinberg, Darina Spasova, Danelle Eto, AnneMarie Pferdekamper, Phuong Nguyen, Brian Le, Zachary Naiman, Susan Murphy, Maricel Gozo, James Skipper, Taylor Ismaili, Xikui Liu, Sunil Sahdeo, Xiao Hu, Isharat Yusuf, Kristen Taylor Meadows, Laura Carter

Gossamer Bio, Inc., San Diego, CA, USA

BACKGROUND

- Diacylglycerol (DAG) is a potent intracellular secondary messenger required for efficient T cell receptor (TCR) signal transduction
- Diacylglycerol kinases (DGK) transform DAG into phosphatidic acid, limiting DAG-dependent signaling involved in T cell activation[1]
- T cells predominantly express DGKα and DGKζ isoforms[2] and deletion of either enhances T cell activity[3-4]
- DGKα and DGKζ are overexpressed in tumor-infiltrating T lymphocytes[5] and DGK activity may cause T cell dysfunction, limiting its cytotoxic potential
- Potent and selective DGKα/ζ dual inhibitors developed by Gossamer Bio have the potential to enhance T cell anti-tumor responses (**Figure 1**)

Figure 1. DGK inhibition impacts multiple mechanisms promoting enhancement of anti-tumor responses

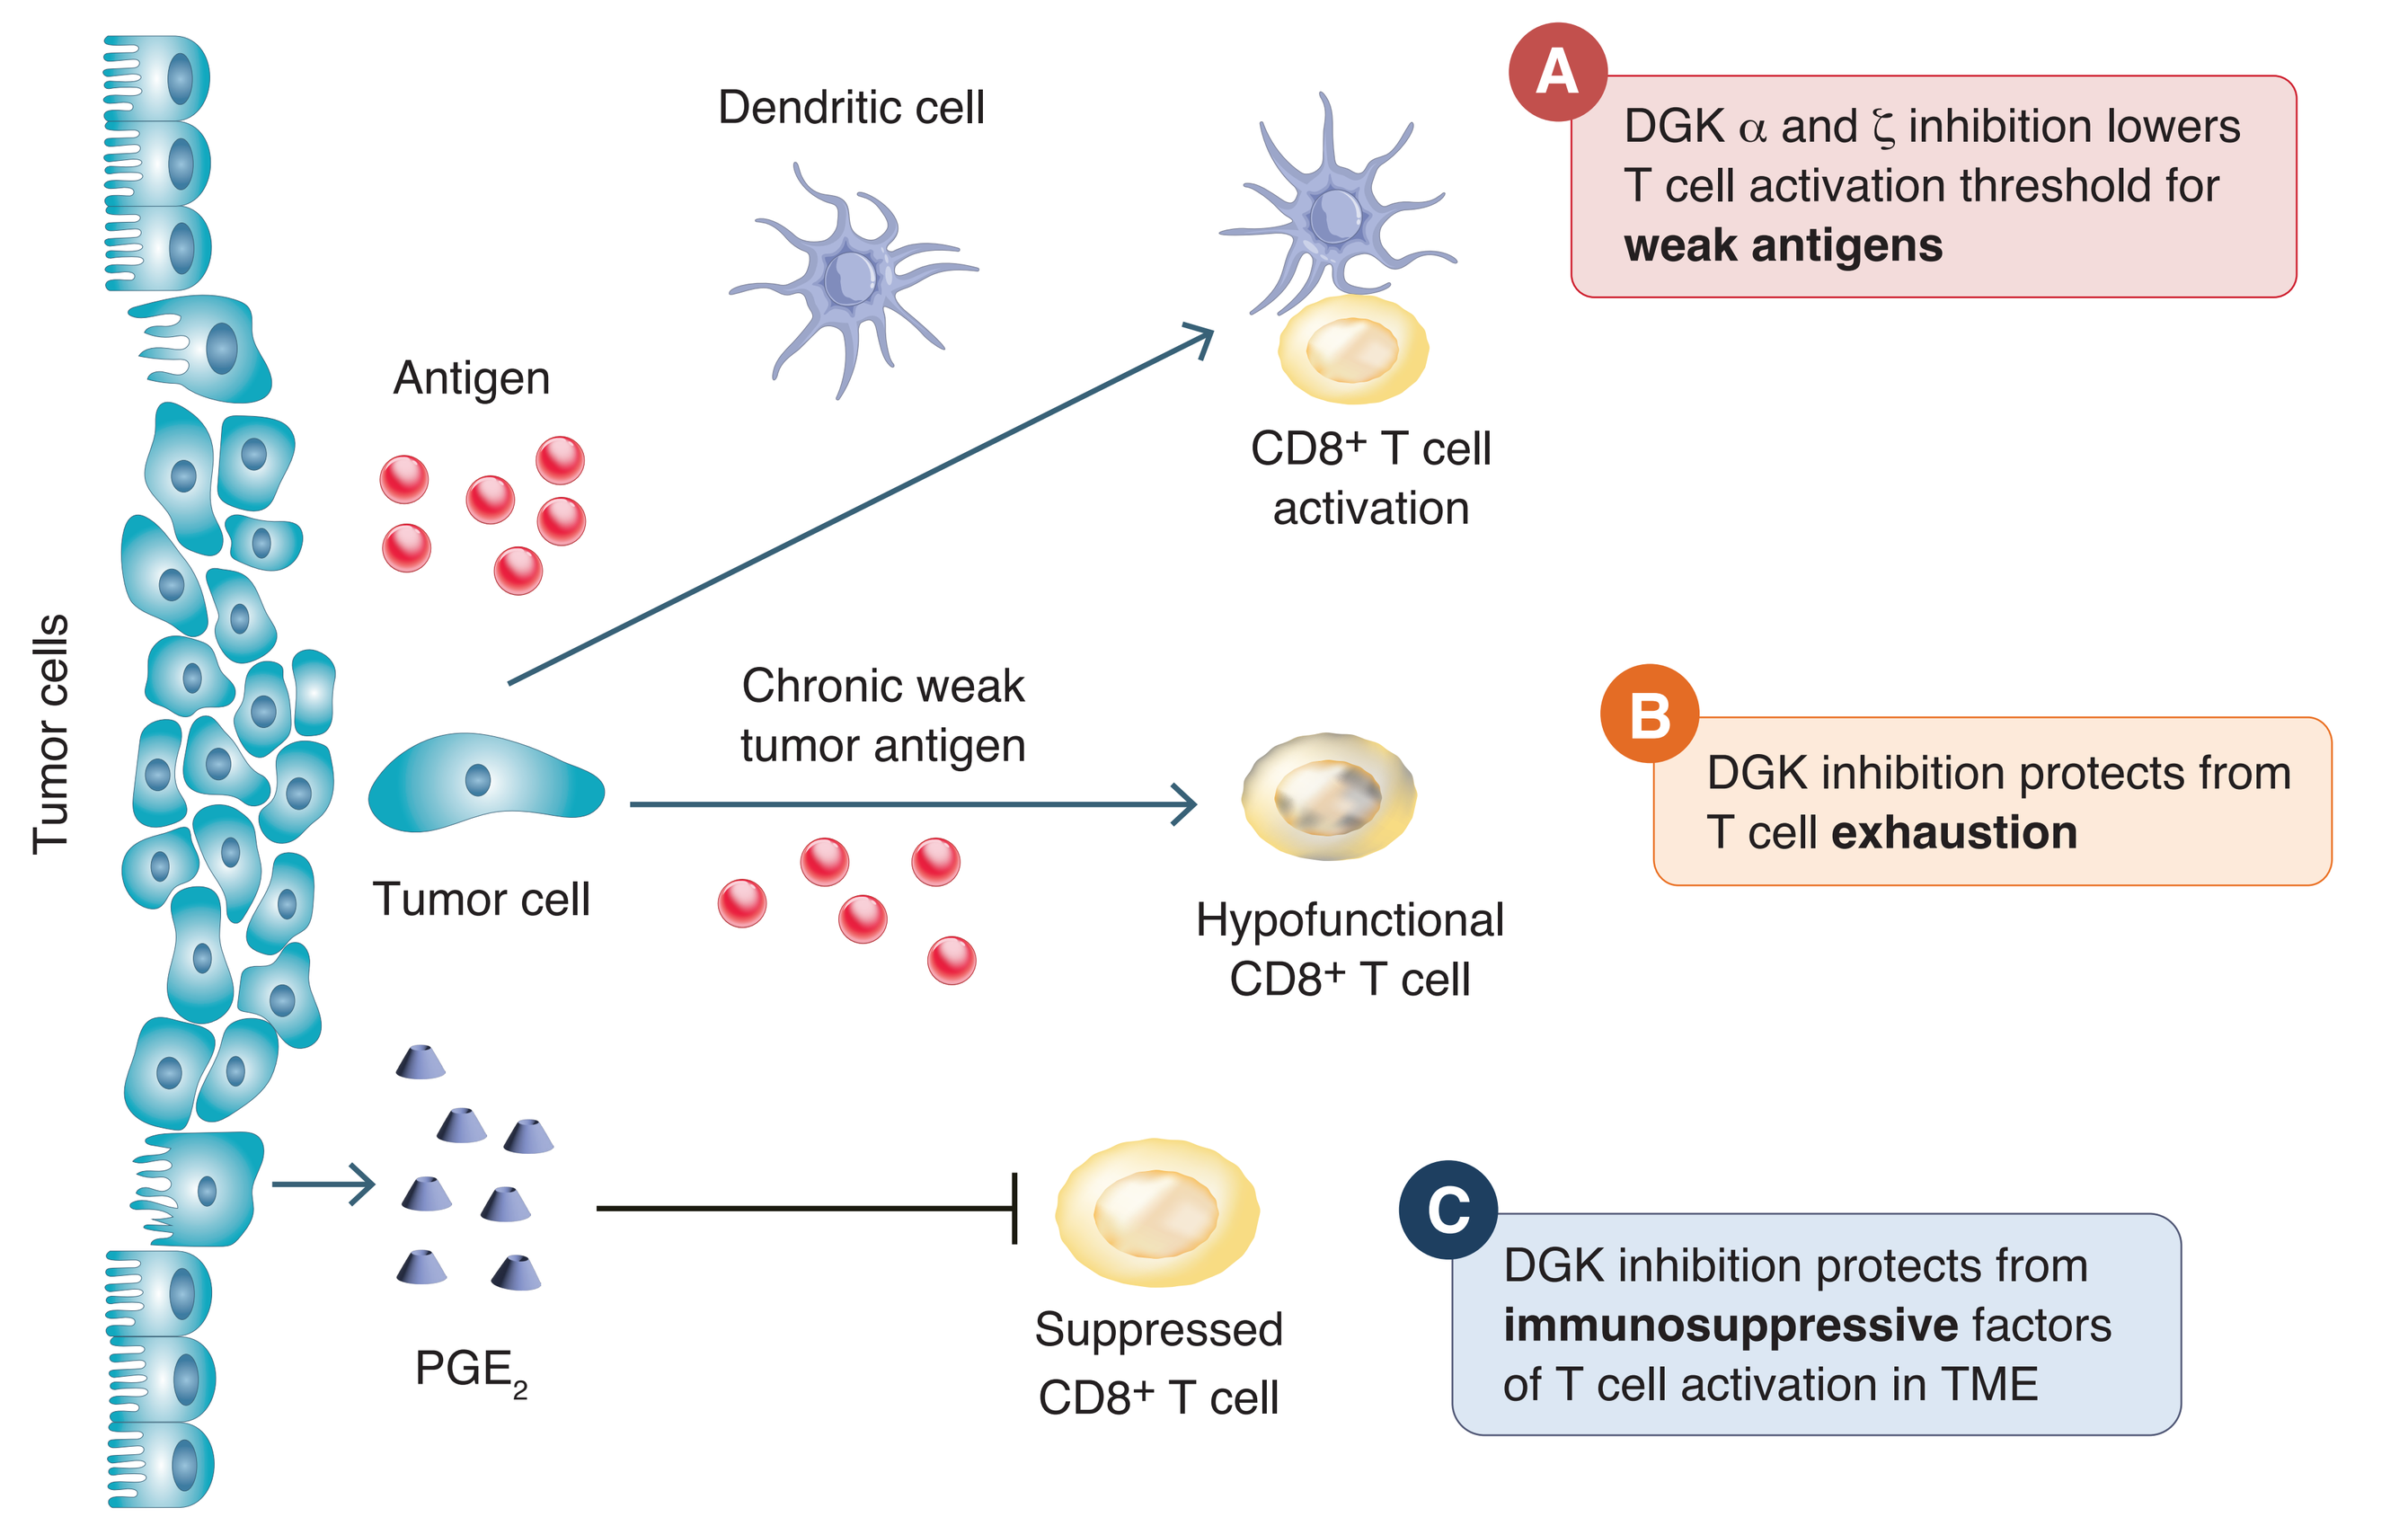
Tumor cells
Dendritic cell
Antigen
CD8+ T cell activation
A
DGK α and ζ inhibition lowers T cell activation threshold for **weak antigens**
Chronic weak tumor antigen
Tumor cell
Hypofunctional CD8+ T cell
B
DGK inhibition protects from T cell **exhaustion**
PGE2
Suppressed CD8+ T cell
C
DGK inhibition protects from **immunosuppressive** factors of T cell activation in TME

A. Selective inhibition of DGKα and DGKζ enhances T cell activation and cytokine production in response to weak tumor antigens

B. DGKα/ζ inhibitors revert the phenotype of "exhausted", non-responsive CD8+ T cells induced by chronic exposure to weak tumor antigens

C. DGKα/ζ inhibitors rescue cytokine release and T cell activity attenuated by immunosuppressive factors found in the tumor microenvironment (TME)

OBJECTIVES

- Evaluate the relative contribution of DGKα vs. DGKζ to T cell activation and function
- Characterize novel small molecule DGKα/ζ dual inhibitors developed by Gossamer Bio

RESULTS

Figure 2. Dual inhibition of DGKα and DGKζ is required to fully enhance T cell activity

- In Jurkat cells, DGKα and DGKζ negatively regulate NFκB activation, and DGKα/ζ deficiency increases IL-2 production upon T cell receptor-stimulation (**Figure 2A**)
- Dual DGKα/ζ inhibition more potently enhances primary human T cell activation than selective inhibition of DGKα or DGKζ isoforms (**Figure 2B**)

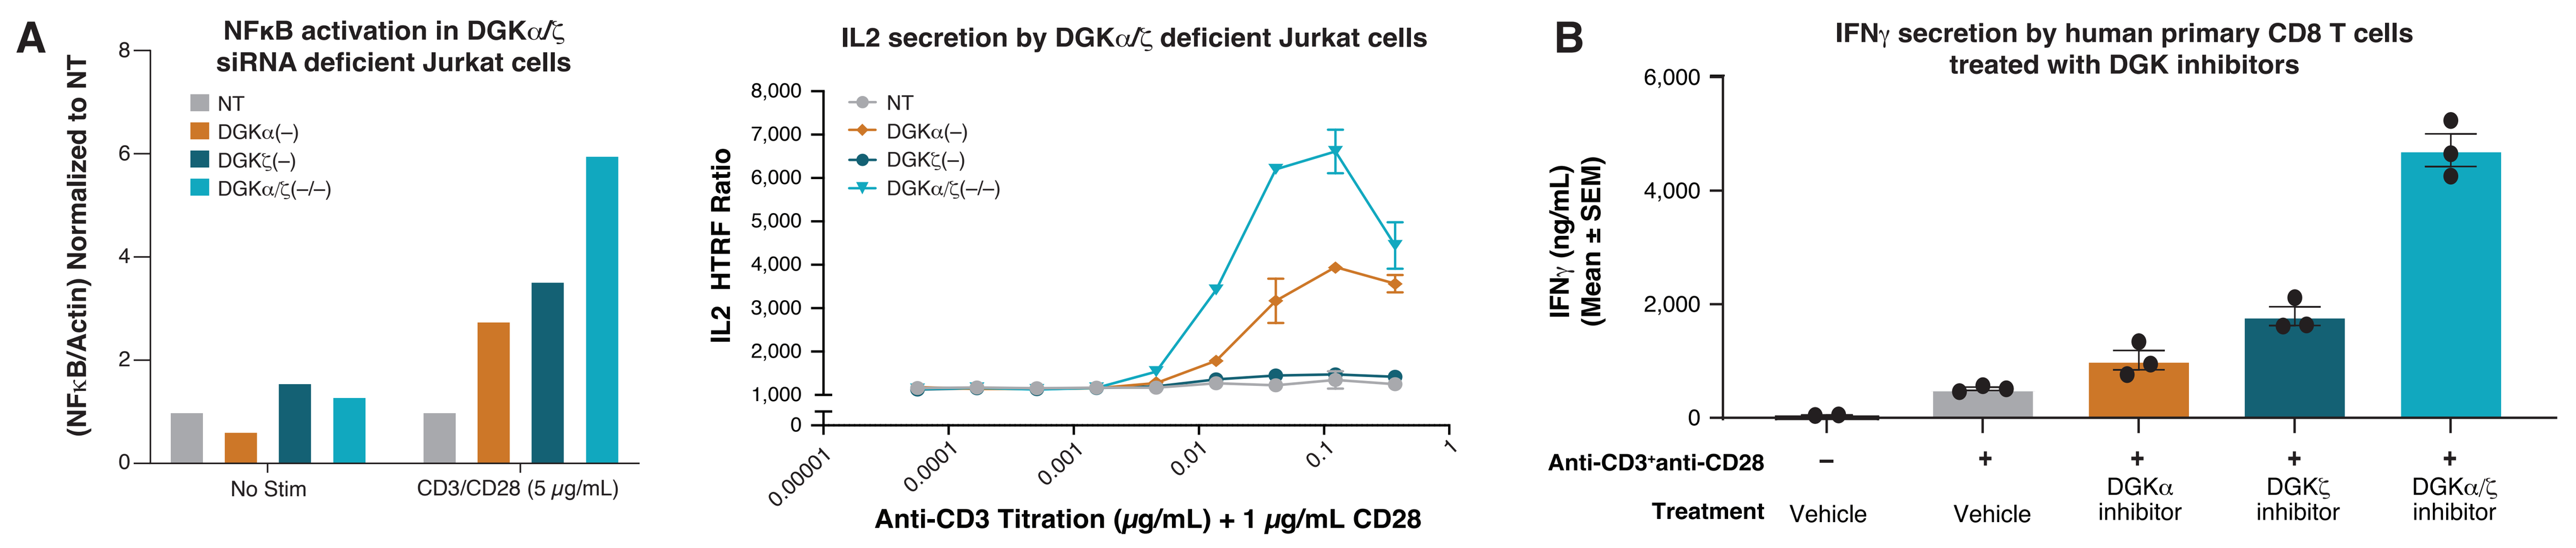
A
NFκB activation in DGKα/ζ siRNA deficient Jurkat cells
(NFκB/Actin) Normalized to NT
NT
DGKα(–)
DGKζ(–)
DGKα/ζ(–/–)
No Stim
CD3/CD28 (5 μg/mL)
IL2 secretion by DGKα/ζ deficient Jurkat cells
IL2 HTRF Ratio
Anti-CD3 Titration (μg/mL) + 1 μg/mL CD28
B
IFNγ secretion by human primary CD8 T cells treated with DGK inhibitors
IFNγ (ng/mL) (Mean ± SEM)
Anti-CD3+anti-CD28
Treatment
Vehicle
Vehicle
DGKα inhibitor
DGKζ inhibitor
DGKα/ζ inhibitor

A. DGKα and DGKζ deficient Jurkat cells were generated using siRNA or CRISPR. NFκB activation and IL2 secretion in control or DGK-deficient Jurkat cells were measured 24 hours after stimulation of cells with the indicated concentration of anti-CD3 and anti-CD28 mAbs. **B.** Purified human CD8+ T cells were stimulated with 2.5 μg/mL αCD3/αCD28 mAbs in the presence of DGKα-, DGKζ- or DGKα/ζ-selective inhibitors. Secretion of IFNγ in the supernatants was measured 24 hours after cell stimulation.

RESULTS

Figure 3. DGKα/ζ inhibitors enhance T cell proliferation and cytokine release in response to low affinity antigens

- DGKα/ζ inhibitors enhance IFNγ production by OT-I cells stimulated with low affinity peptides (**Figure 3A**)
- In human melanoma-specific T cells, DGKα/ζ inhibitors enhance IFNγ production after stimulation with the weak affinity MART1 antigen (**Figure 3B**)

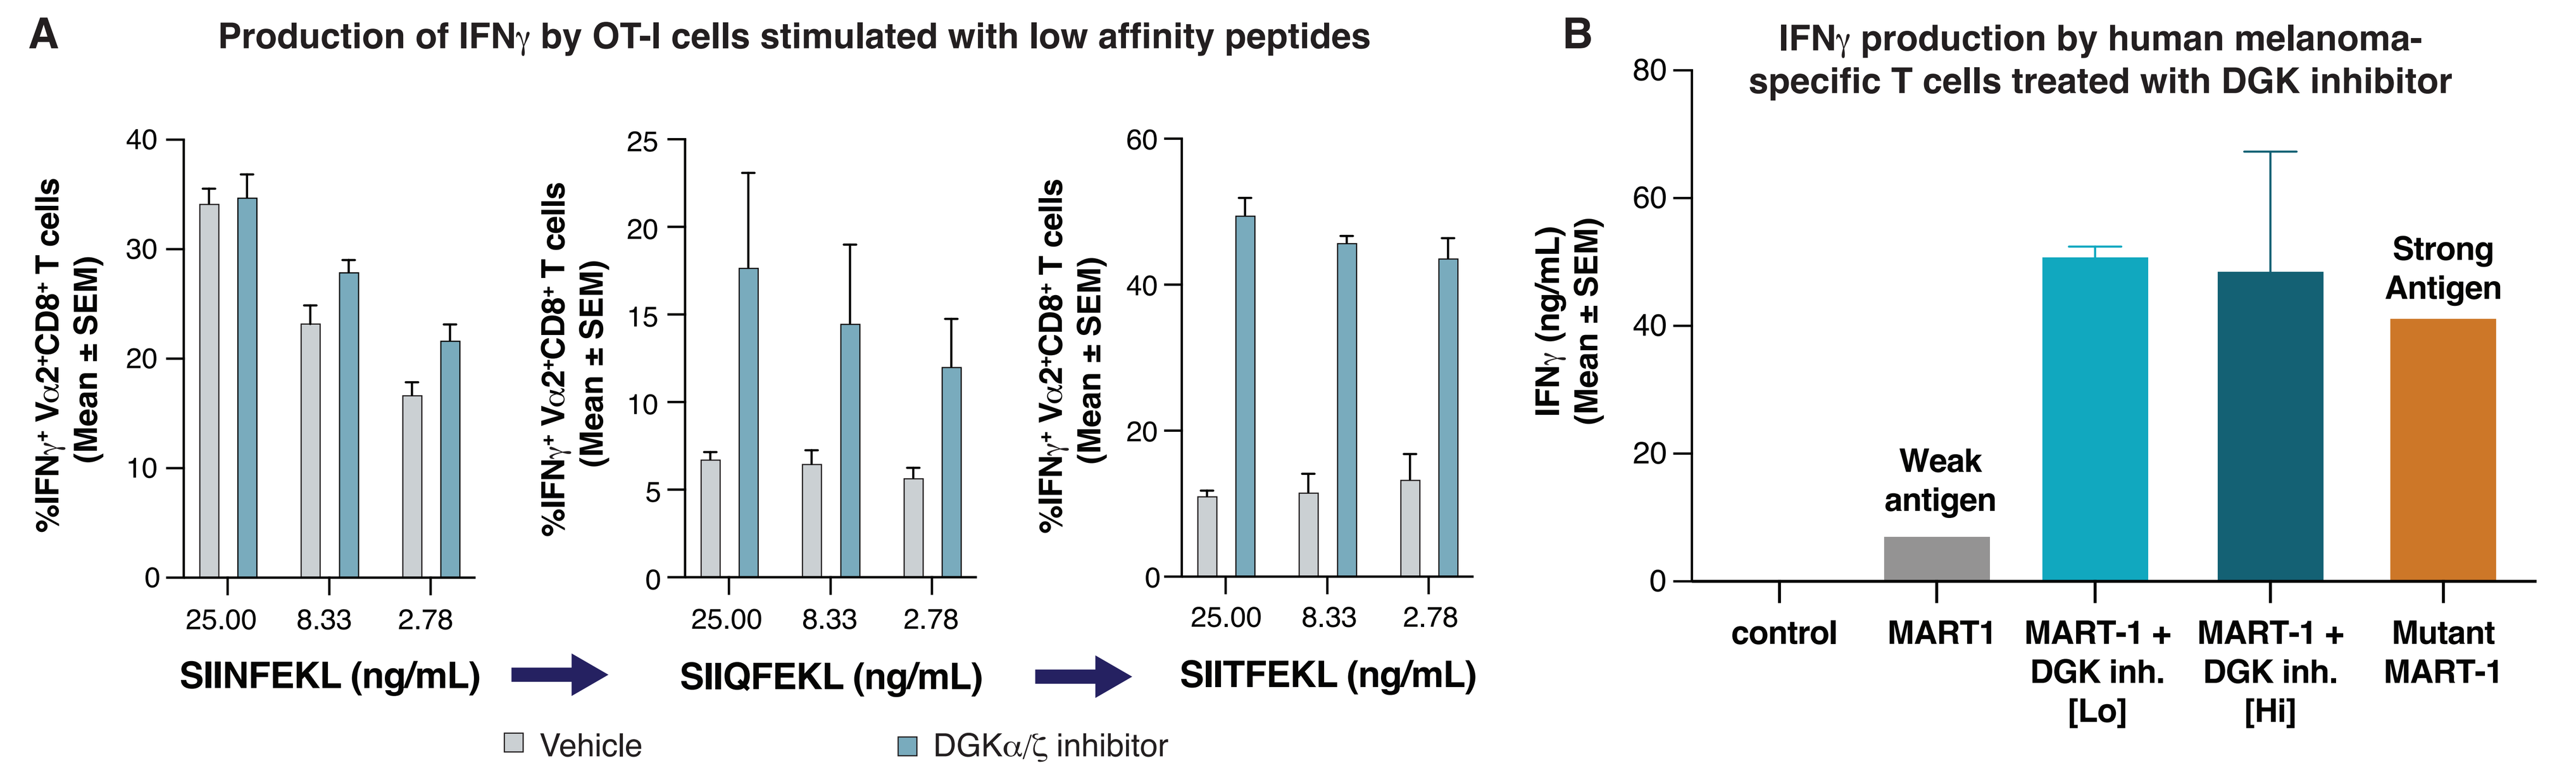
A
Production of IFNγ by OT-I cells stimulated with low affinity peptides
%IFNγ+ Vα2+CD8+ T cells (Mean ± SEM)
SIINFEKL (ng/mL)
SIIQFEKL (ng/mL)
SIITFEKL (ng/mL)
Vehicle
DGKα/ζ inhibitor
B
IFNγ production by human melanoma-specific T cells treated with DGK inhibitor
IFNγ (ng/mL) (Mean ± SEM)
Weak antigen
Strong Antigen
control
MART1
MART-1 + DGK inh. [Lo]
MART-1 + DGK inh. [Hi]
Mutant MART-1

A. Production of IFNγ by OT-I cells treated with different concentrations of the high (SIINFEKL), medium (SIIQFEKL), or low (SIITFEKL) affinity OVA-peptides, with or without DGKα/ζ inhibitor, was assessed by flow cytometry measuring the percentage of IFNγ+Vα2+CD8+T cells at day 3. **B.** Human melanoma-specific CD8 T cells were stimulated with 1 μg/mL of MART-1 WT peptide (AAGIGILTV) in the presence of vehicle (labeled as week antigen), or DGKα/ζ inhibitor at low [Lo] or high [Hi] concentration. The high affinity MART-1 mutant peptide ELAGIGILTV was used as a positive control. IFNγ production in the culture supernatants at day 3 was measured by MSD.

Figure 4. DGKα/ζ inhibition reverts CD8 T Cell exhaustion and protect cells from immune suppression

- DGKα/ζ inhibitors rescue exhausted T cell function in the LCMV *ex vivo* model. DGK inhibition enhances IFNγ and granzyme B production by LCMV-exhausted CD8 T cells (**Figure 4A**)
- DGKα/ζ inhibition protects T cells from immunosuppression and rescues cytokine release from human T cells treated with adenosine, TGFβ or PGE_2 (**Figure 4B**)

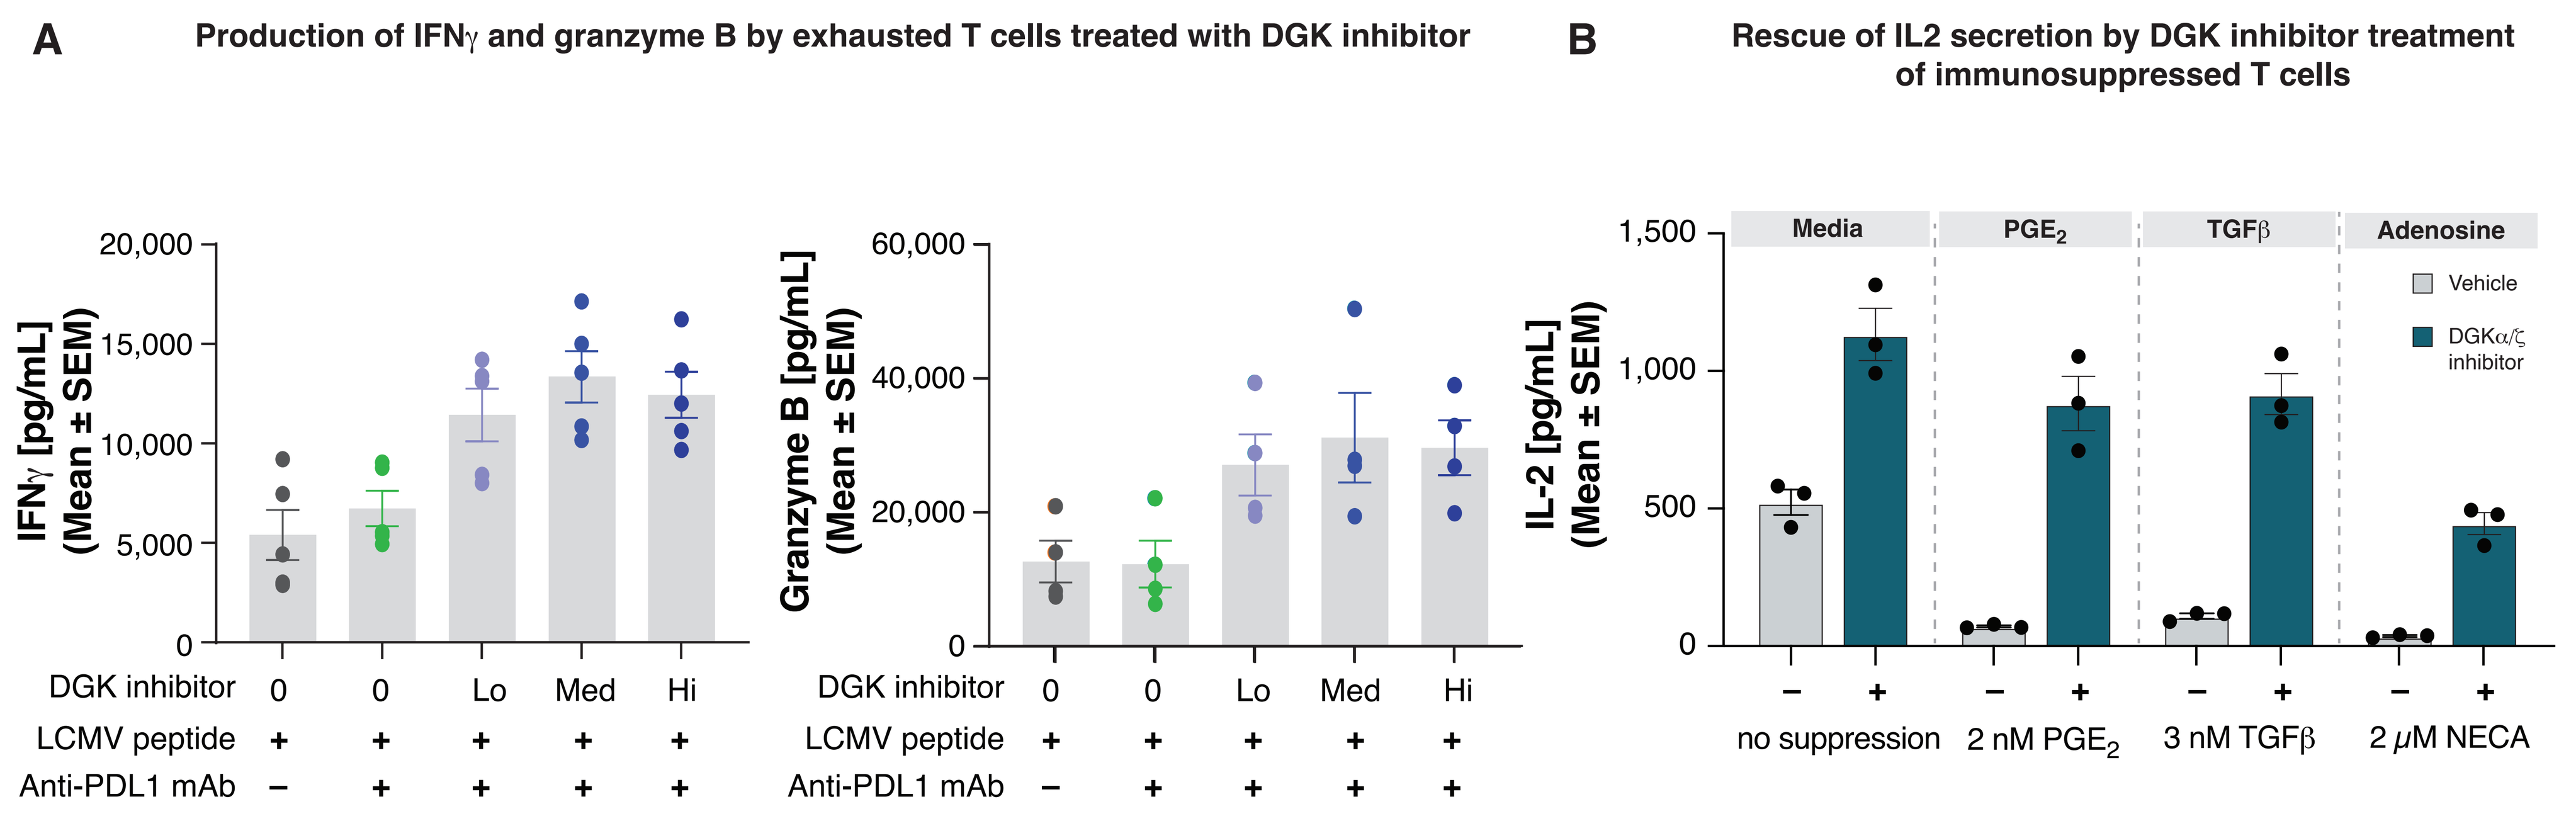
A
Production of IFNγ and granzyme B by exhausted T cells treated with DGK inhibitor
IFNγ [pg/mL] (Mean ± SEM)
Granzyme B [pg/mL] (Mean ± SEM)
DGK inhibitor
LCMV peptide
Anti-PDL1 mAb
B
Rescue of IL2 secretion by DGK inhibitor treatment of immunosuppressed T cells
IL-2 [pg/mL] (Mean ± SEM)
Media
PGE2
TGFβ
Adenosine
Vehicle
DGKα/ζ inhibitor
no suppression
2 nM PGE2
3 nM TGFβ
2 μM NECA

Splenocytes from LCMV clone 13-infected mice isolated at day 30 post-infection were stimulated *ex vivo* with LCMV peptide pool (5 μg/mL GP61-81 and 2 μg/mL GP33-41, NP396-404 and GP276-286) in the presence of vehicle or DGKα/ζ inhibitor, and with or without anti-PDL1 mAb (25 μg/mL). **A**. Production of IFNγ and granzyme B was measured in the culture supernatants at day 5, by MSD and ELISA, respectively. **B**. Purified human CD3+ T cells were cultured with media or in the presence of PGE_2 (2 nM), TGFβ (3 nM) or the adenosine receptor agonist NECA (2 μM), with or without DGKα/ζ inhibitor. IL2 production was measured in the culture supernatants at day 3 using ELISA.

Figure 5. DGKα/ζ inhibitor enhances T cell cytotoxicity and *in vitro* killing of tumor cells

- Inhibition of DGKα/ζ enhances OT-I cell lytic potential and promotes robust *in vitro* killing of MC38-OVA tumor cells at different effector:target cell ratios

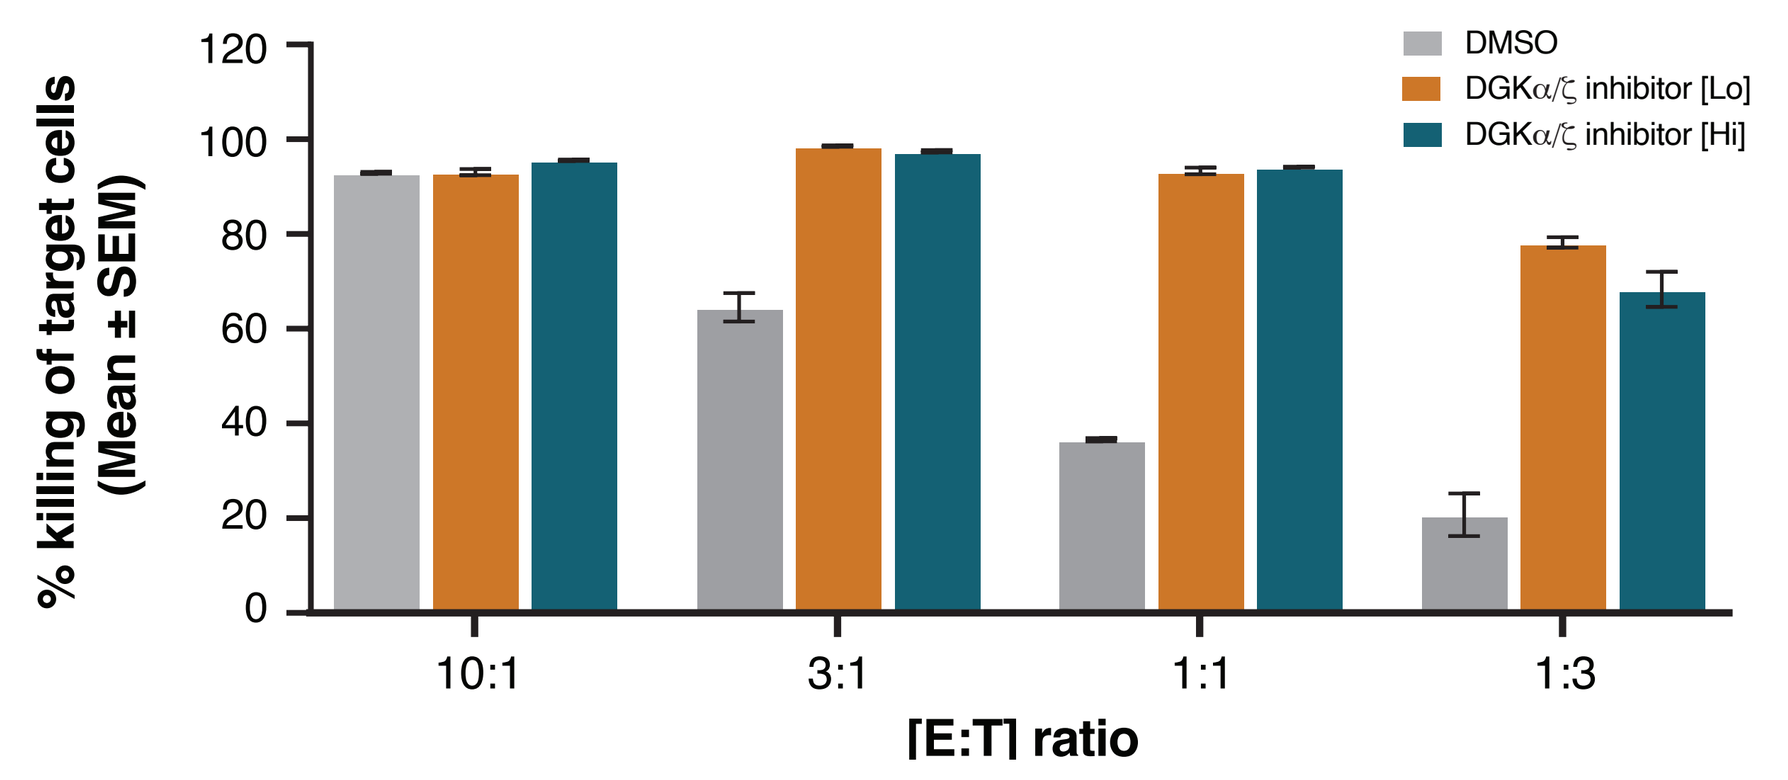
% killing of target cells (Mean ± SEM)
DMSO
DGKα/ζ inhibitor [Lo]
DGKα/ζ inhibitor [Hi]
10:1
3:1
1:1
1:3
[E:T] ratio

OT-I splenocyte (effectors, E) were activated with OT-I peptide (50 ng/mL) in the presence of vehicle, or DGKα/ζ inhibitor at low [Lo] or high [Hi] concentration, for 4 days. Syngeneic MC38-OVA tumor cells (target cells, T) were co-cultured with effector cells at the indicated E:T ratios. Target cell death was measured after 24 hours of E:T cell co-culture.

Figure 6. DGKα/ζ dual inhibitor demonstrates robust *in vivo* efficacy in the MC38 mouse tumor model

- DGKα/ζ inhibitor combined with α-PD1 mAb demonstrates dose-dependent efficacy in the MC38 model (**Figure 6B**)
- Medium and high doses of the DGK inhibitor result in complete tumor regression observed in >70% of animals implanted with MC38 cells (**Figure 6B**)
- Mice with sustained tumor regression (up to 100 days post initial tumor implantation) eradicate new tumors after being rechallenged with 10-fold more MC38 cells in the absence of additional treatments (**Figure 6C**)

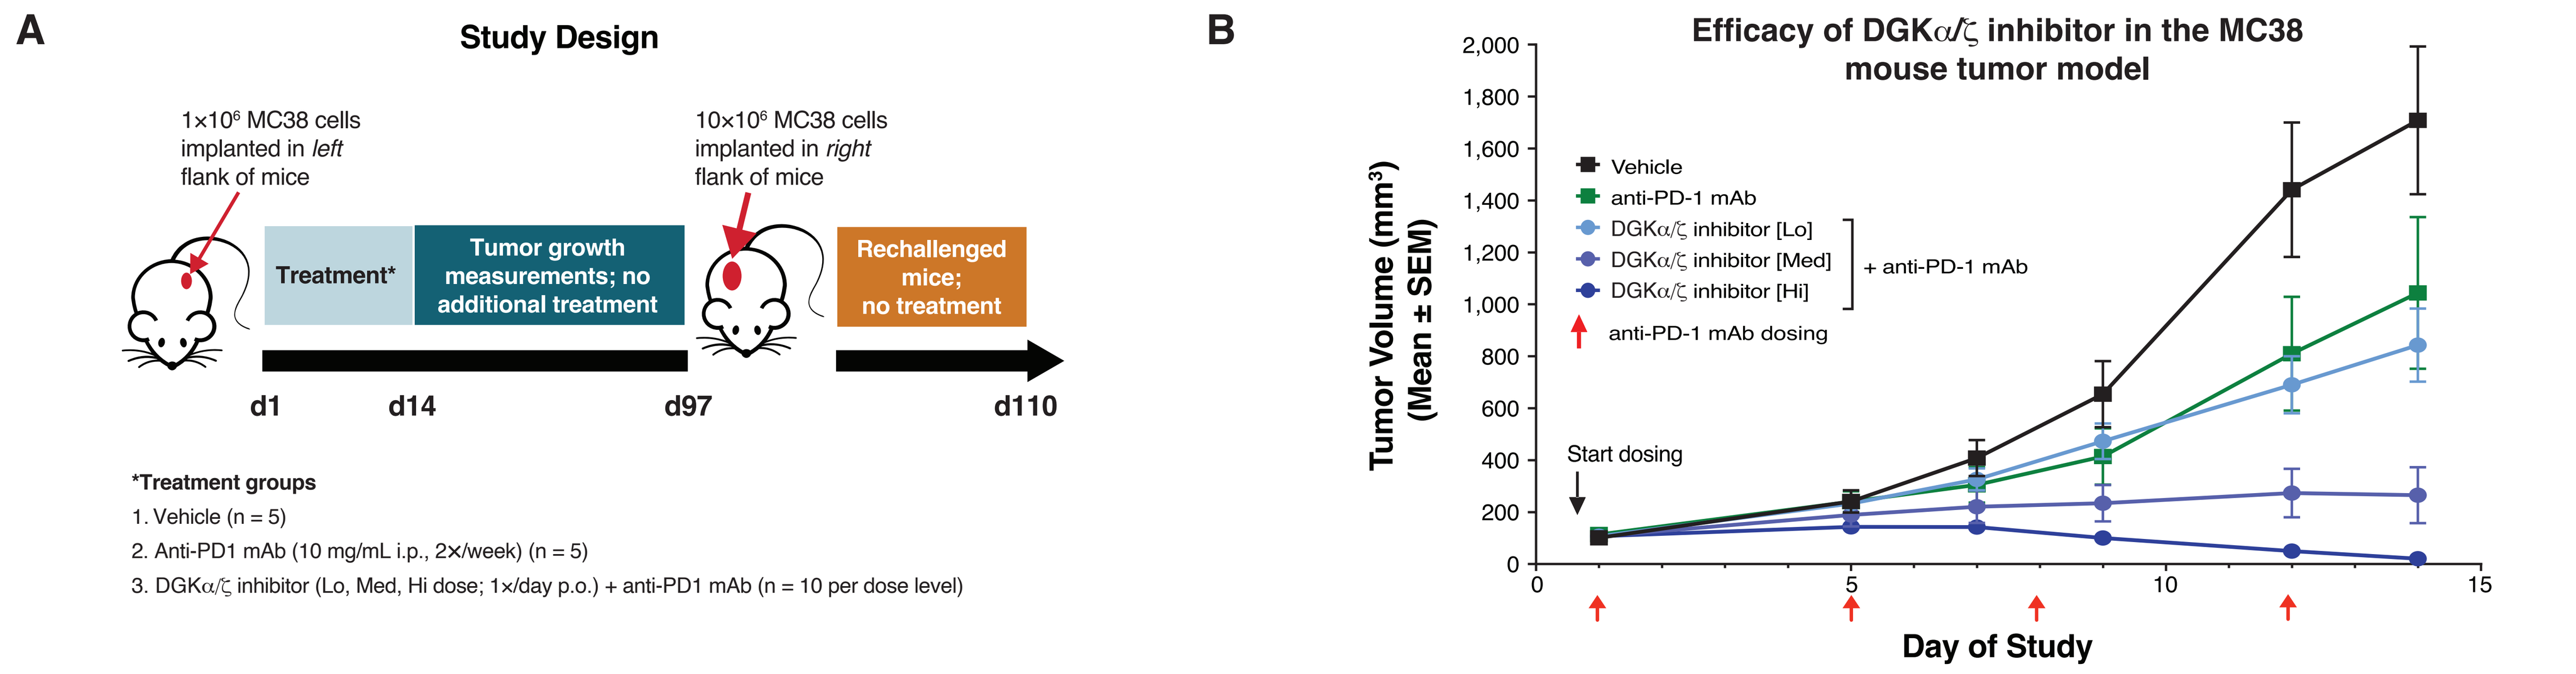
A
Study Design
1×10⁶ MC38 cells implanted in *left* flank of mice
Treatment*
Tumor growth measurements; no additional treatment
10×10⁶ MC38 cells implanted in *right* flank of mice
Rechallenged mice; no treatment
d1
d14
d97
d110
*Treatment groups
1. Vehicle (n = 5)
2. Anti-PD1 mAb (10 mg/mL i.p., 2×/week) (n = 5)
3. DGKα/ζ inhibitor (Lo, Med, Hi dose; 1×/day p.o.) + anti-PD1 mAb (n = 10 per dose level)
B
Efficacy of DGKα/ζ inhibitor in the MC38 mouse tumor model
Tumor Volume (mm³) (Mean ± SEM)
Vehicle
anti-PD-1 mAb
DGKα/ζ inhibitor [Lo]
DGKα/ζ inhibitor [Med]
DGKα/ζ inhibitor [Hi]
+ anti-PD-1 mAb
anti-PD-1 mAb dosing
Start dosing
Day of Study

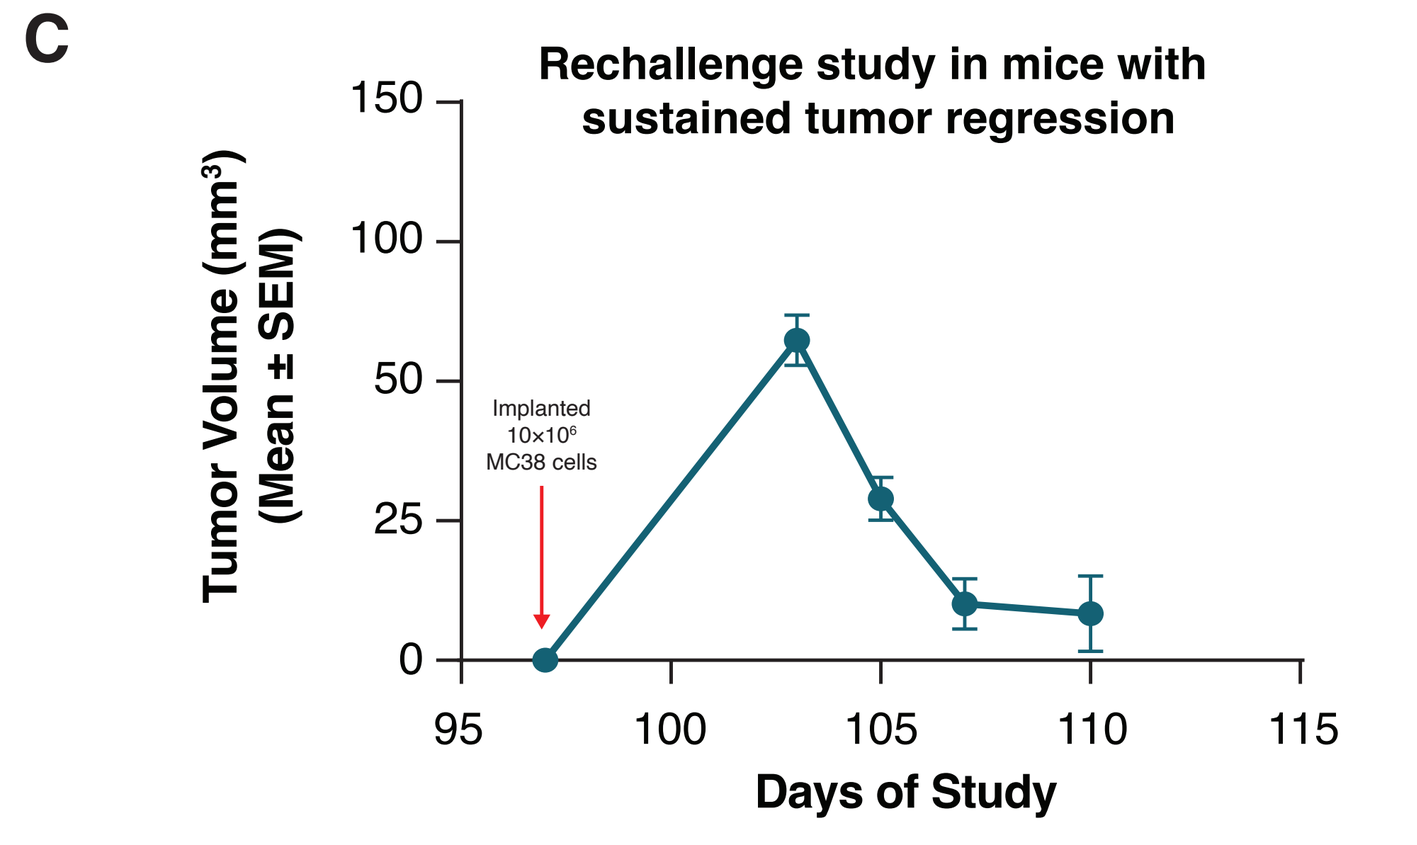
C
Rechallenge study in mice with sustained tumor regression
Tumor Volume (mm³) (Mean ± SEM)
Implanted 10×10⁶ MC38 cells
Days of Study

A. Efficacy study in the MC38 murine colon carcinoma model. N = 40 mice implanted subcutaneously with 1×10^6 MC38 cells were treated as stated. The DGK inhibitor was administered for 14 consecutive days starting when tumor size reached ~100 mm^3 (d1). Animals with complete tumor regression remained in study for 97 days (d97) for rechallenge experiments. At d97, mice were rechallenged with 10×10^6 MC38 cells in the opposite flank to the original tumor, without additional treatments. **B.** Evaluation of efficacy of DGKα/ζ inhibitor administered at a low [Lo], medium [Med] or high [Hi] dose. **C.** Tumor growth in animals (n = 14) with sustained tumor regression rechallenged at d97 in absence of additional DGK inhibitor or anti-PD1 mAb treatment.

SUMMARY AND CONCLUSIONS

- **Here, we demonstrated that DGKα and DGKζ synergistically regulate T cell activity**
- **Inhibition of both isoforms with dual DGKα/ζ inhibitors developed by Gossamer Bio invigorates CD8 T cell responses to weak tumor antigens, reverts exhaustion, overcomes immunosuppression, and enhances CD8 T cell cytotoxic activity**
- **Treatment of mice with DGKα/ζ inhibitors promoted robust tumor growth inhibition that led to complete tumor regression and long-lasting anti-tumor memory responses in the MC38 syngeneic tumor model**
- **Our data suggest that the combination of a DGKα/ζ dual inhibitor with anti-PD1 therapy represents a promising therapeutic approach that could potentially provide long-lasting anti-tumor immunity for the treatment of cancer**

REFERENCES

1. Mérida I, Avila-Flores A, Merino E. *Biochem J.* 2008; 409(1):1-18.
2. Krishna S, Zhong X. *Crit Rev Immunol.* 2013; 33(2):97-118.
3. Guo R, Wan C-K, Carpenter JH, et al. *Proc Natl Acad Sci* U S A. 2008; 105(33):11909-14.
4. Riese MJ, Grewal J, Das J, et al. *J Biol Chem.* 2011; 286(7):5254-65.
5. Prinz PU, Mendler AN, Masouris I, et al. *J Immunol.* 2012; 188(12):5990-6000 2012.
6. Zheng Y, Zha Y, Gajewski TF. *EMBO Rep.* 2008; 9(1):50-5.

ACKNOWLEDGEMENTS

The authors gratefully acknowledge the Gossamer Bio employees Anthony Casarez, Rachel Rowlands, Charlotte James, David Guimond, Alex Catoire, Beili Zhang , Maurice Marsini, Mark Rose, and Martin Rowbottom for their contributions to the work presented in this poster.

Presented at the 37th Annual Meeting of the Society for Immunotherapy of Cancer, November 8 – 12, 2022